150

Patient and Healthcare Provider Perspectives on Symptom Burden and Treatment Decision-Making in Narcolepsy and Idiopathic Hypersomnia

Caroleen Drachenberg, PhD, MSPH[1]; Sarah C. Markt, ScD, MPH[1]; Samantha Linton, PhD[2]; Meagan Farrell, PhD[2]; Jennifer D'Souza, PhD[2]; Jennifer N. Hill, MA[2]; Christina A. Graham, BS, BA[2]; Jessica K. Alexander, PhD[1]; Marisa Whalen, PharmD[3]; Silky Beaty, PharmD, MSPH[1]; Barbara Hutchinson, MD, PhD, FACC[4]; Shane Drahos, MD[5]

[1]Jazz Pharmaceuticals, Palo Alto, CA, USA; [2]IQVIA, Boston, MA, USA; [3]Jazz Pharmaceuticals, Philadelphia, PA, USA; [4]Chesapeake Cardiac Care, Bowie, MD, USA; [5]Premier Sleep Medicine, Winter Garden, FL, USA

Objectives

- To characterize nighttime sleep and daytime symptom burden and identify unmet needs in patients with narcolepsy (PwN) and patients with idiopathic hypersomnia (PwIH), from the perspectives of both healthcare providers (HCPs) and patients
- To describe the symptoms and treatment attributes considered in treatment decision-making, as reported by HCPs, PwN, and PwIH

Conclusions

- RECITALS captured real-world perspectives from HCPs, as well as PwN and PwIH, providing a comprehensive view of care delivery and lived impact
 - Study limitations included a small sample size that resulted in exploratory findings, a cross-sectional design, and self-reported information; future research could provide additional insight into these findings
- Real-world, HCP-reported decision-making was primarily focused on addressing EDS as the most prevalent symptom; however, patients report their experience as a broader 24-hour burden of bothersome symptoms that profoundly affect their daily lives
- Results highlight potential opportunities to expand the HCP–patient dialogue and incorporate a broader range of nighttime and daytime sleep symptoms into shared decision-making to support comprehensive, patient-centered care

References: 1. Barateau L, et al. *Handb Clin Neurol.* 2019;161:353-367. **2.** Dauvilliers Y, et al. *Nat Rev Neurol.* 2014;10(7):386-395. **3.** Ruoff C, Rye D. *Curr Med Res Opin.* 2016;32(10):1611-1622. **4.** Sateia MJ. *Chest.* 2014;146(5):1387-1394.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees, Eleanor Bush, MA, provided medical writing support and Katie Herman provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: C Drachenberg, JK Alexander, S Beaty, SC Markt, and **M Whalen** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **S Linton, J D'Souza, JN Hill, C Graham,** and **M Farrell** are full-time employees of IQVIA, which received funding from Jazz Pharmaceuticals to conduct the study. **B Hutchinson** is a consultant to CardioOne and Jazz Pharmaceuticals. **S Drahos** is a paid speaker for Jazz Pharmaceuticals.

Scan this code to access a plain language summary of the published abstract. This code is not for promotional purposes.

Scan this code to access this poster and supplemental material online. This code is not for promotional purposes.

Presented at Psych Congress 2026; September 15–19, 2026; New Orleans, LA, USA

Introduction

- Narcolepsy and idiopathic hypersomnia (IH) are central disorders of hypersomnolence associated with a range of symptoms that impact sleep and daytime functioning, including disrupted nighttime sleep, nonrestorative/ineffective sleep, sleep inertia, and excessive daytime sleepiness (EDS)[1-4]
- The patient perspective on symptoms and treatment attributes, as well as how these factors are considered during treatment decision-making, are not well characterized in the real world
- Understanding what matters most to HCPs and patients with narcolepsy and IH may facilitate shared decision-making and improved patient treatment experiences

Methods

- Real-world survEys on Clinical practIce and Treatment decision-mAking in narcoLepsy and idiopathic hyperSomnia (RECITALS) was a cross-sectional US study exploring real-world symptom burden and treatment considerations using HCP and patient (PwN and PwIH) online surveys
- Participants met several key eligibility criteria, including:
 - HCPs: At least 5 years' experience managing the care of PwN and PwIH; care management of >5 PwN and/or PwIH over the past 6 months
 - PwN and PwIH: At least 18 years old; current confirmed diagnosis of narcolepsy type 1 (NT1), narcolepsy type 2 (NT2), or IH
 - Targeted sampling was based on NT1/NT2, the presence or absence of long sleep time among participants with IH, treatment status, and demographic characteristics
- Two surveys were administered between December 2025 and April 2026: 1 for HCPs and 1 for PwN and PwIH
 - The HCP survey included de novo items assessing perspectives on symptom burden, care coordination and access to care, treatment decision-making, and understanding of and experience managing narcolepsy and IH
 - The patient survey included standardized patient-reported outcome instruments (Epworth Sleepiness Scale [ESS], Idiopathic Hypersomnia Severity Scale for PwIH) and de novo items assessing disease experiences, symptom burden, and treatment decision-making
- Descriptive analyses were conducted; odds ratios quantified associations between symptom bothersomeness and functional impacts of narcolepsy and IH on daily life

Results

Figure 1. Characteristics of participating HCPs, PwN, and PwIH

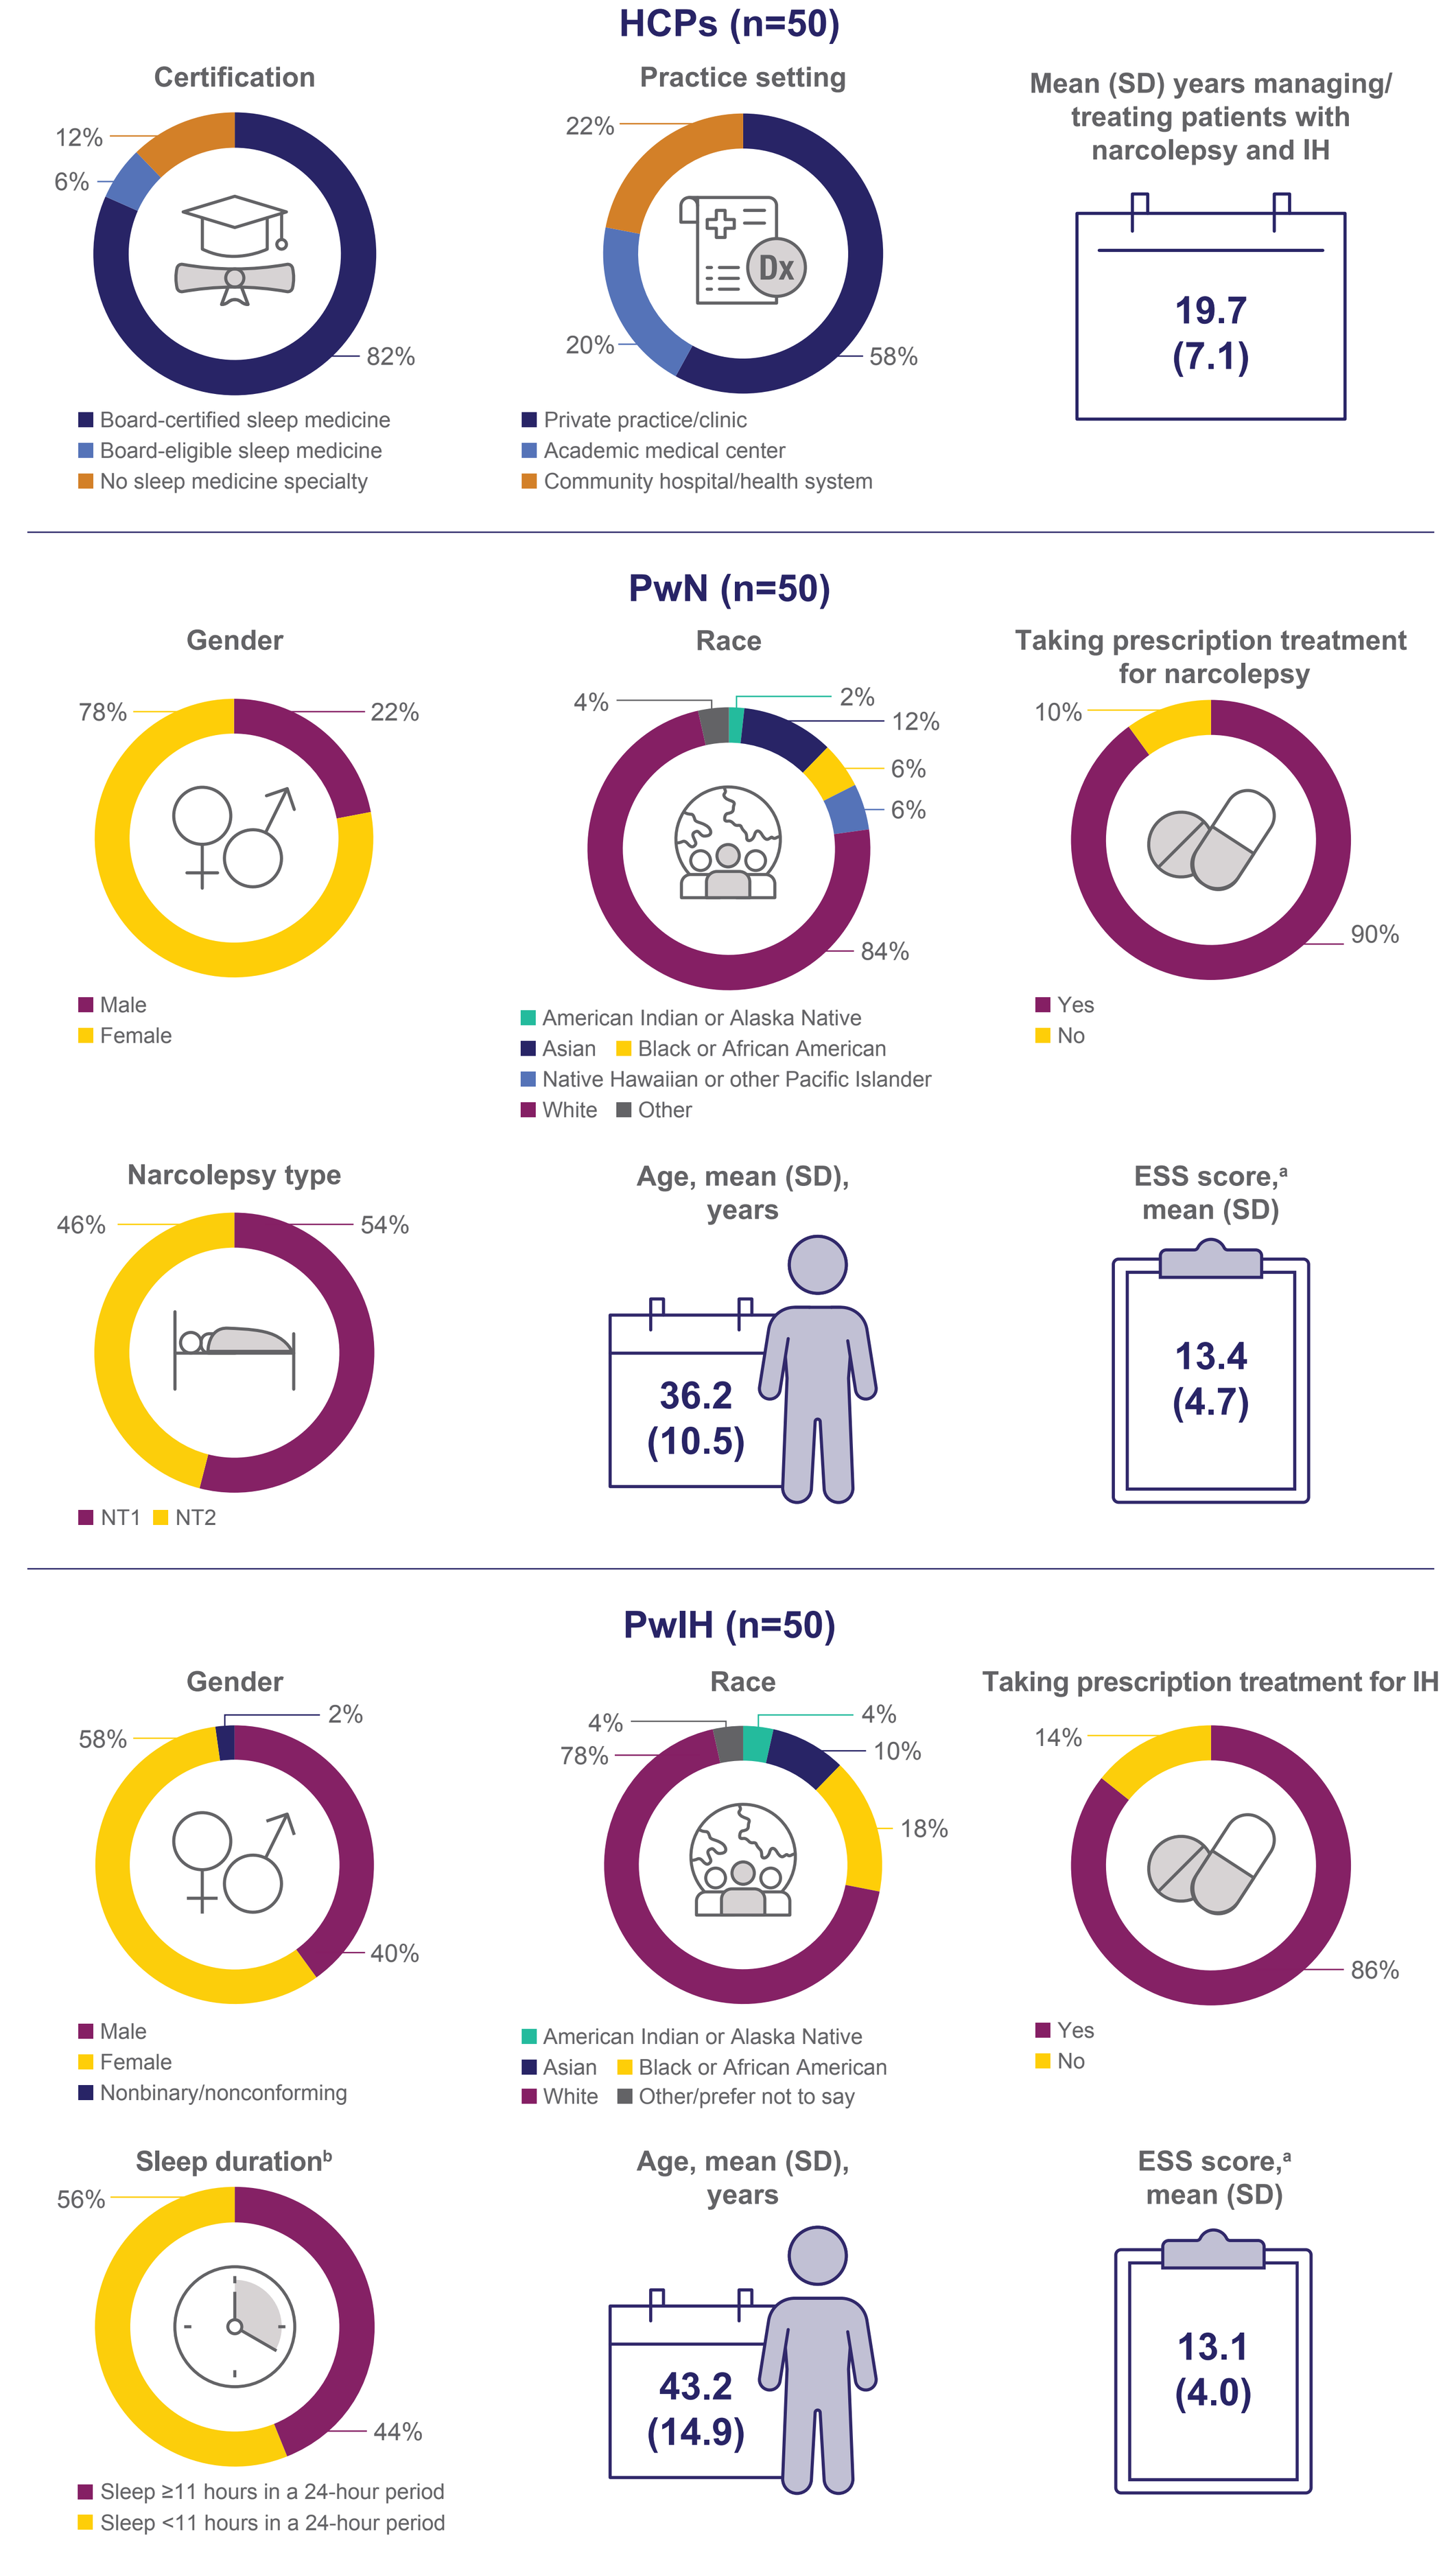
HCPs (n=50)
Certification
12%
6%
82%
Board-certified sleep medicine
Board-eligible sleep medicine
No sleep medicine specialty
Practice setting
22%
20%
58%
Private practice/clinic
Academic medical center
Community hospital/health system
Mean (SD) years managing/treating patients with narcolepsy and IH
19.7 (7.1)
PwN (n=50)
Gender
78%
22%
Male
Female
Race
4%
2%
12%
6%
6%
84%
American Indian or Alaska Native
Asian
Black or African American
Native Hawaiian or other Pacific Islander
White
Other
Taking prescription treatment for narcolepsy
10%
90%
Yes
No
Narcolepsy type
46%
54%
NT1
NT2
Age, mean (SD), years
36.2 (10.5)
ESS score,a mean (SD)
13.4 (4.7)
PwIH (n=50)
Gender
2%
58%
40%
Male
Female
Nonbinary/nonconforming
Race
4%
78%
4%
10%
18%
American Indian or Alaska Native
Asian
Black or African American
White
Other/prefer not to say
Taking prescription treatment for IH
14%
86%
Yes
No
Sleep durationb
56%
44%
Sleep ≥11 hours in a 24-hour period
Sleep <11 hours in a 24-hour period
Age, mean (SD), years
43.2 (14.9)
ESS score,a mean (SD)
13.1 (4.0)

[a]Higher scores indicate increased severity of daytime sleepiness. [b]Sleep duration is the actual total sleep time reported by PwIH.
ESS, Epworth Sleepiness Scale; HCP, healthcare provider; IH, idiopathic hypersomnia; NT1, narcolepsy type 1; NT2, narcolepsy type 2; PwIH, patients with idiopathic hypersomnia; PwN, patients with narcolepsy; SD, standard deviation.

- Overall, most HCPs surveyed were board-certified sleep medicine specialists (82%), delivering care primarily in private practice settings (58%)
- The majority of PwN and PwIH were White and female; mean age was 36.2 years and 43.2 years for PwN and PwIH, respectively
- Most PwN and PwIH were currently taking prescription medication for their narcolepsy or IH and had moderate to severe EDS based on ESS scores
 - The mean (SD) IHSS score for PwIH was 30.8 (7.2)

Figure 2. Symptoms Reported by HCPs as Most Prevalent[a] and Most Considered When Determining Treatment[b] in (A) NT1, (B) NT2, and (C) IH

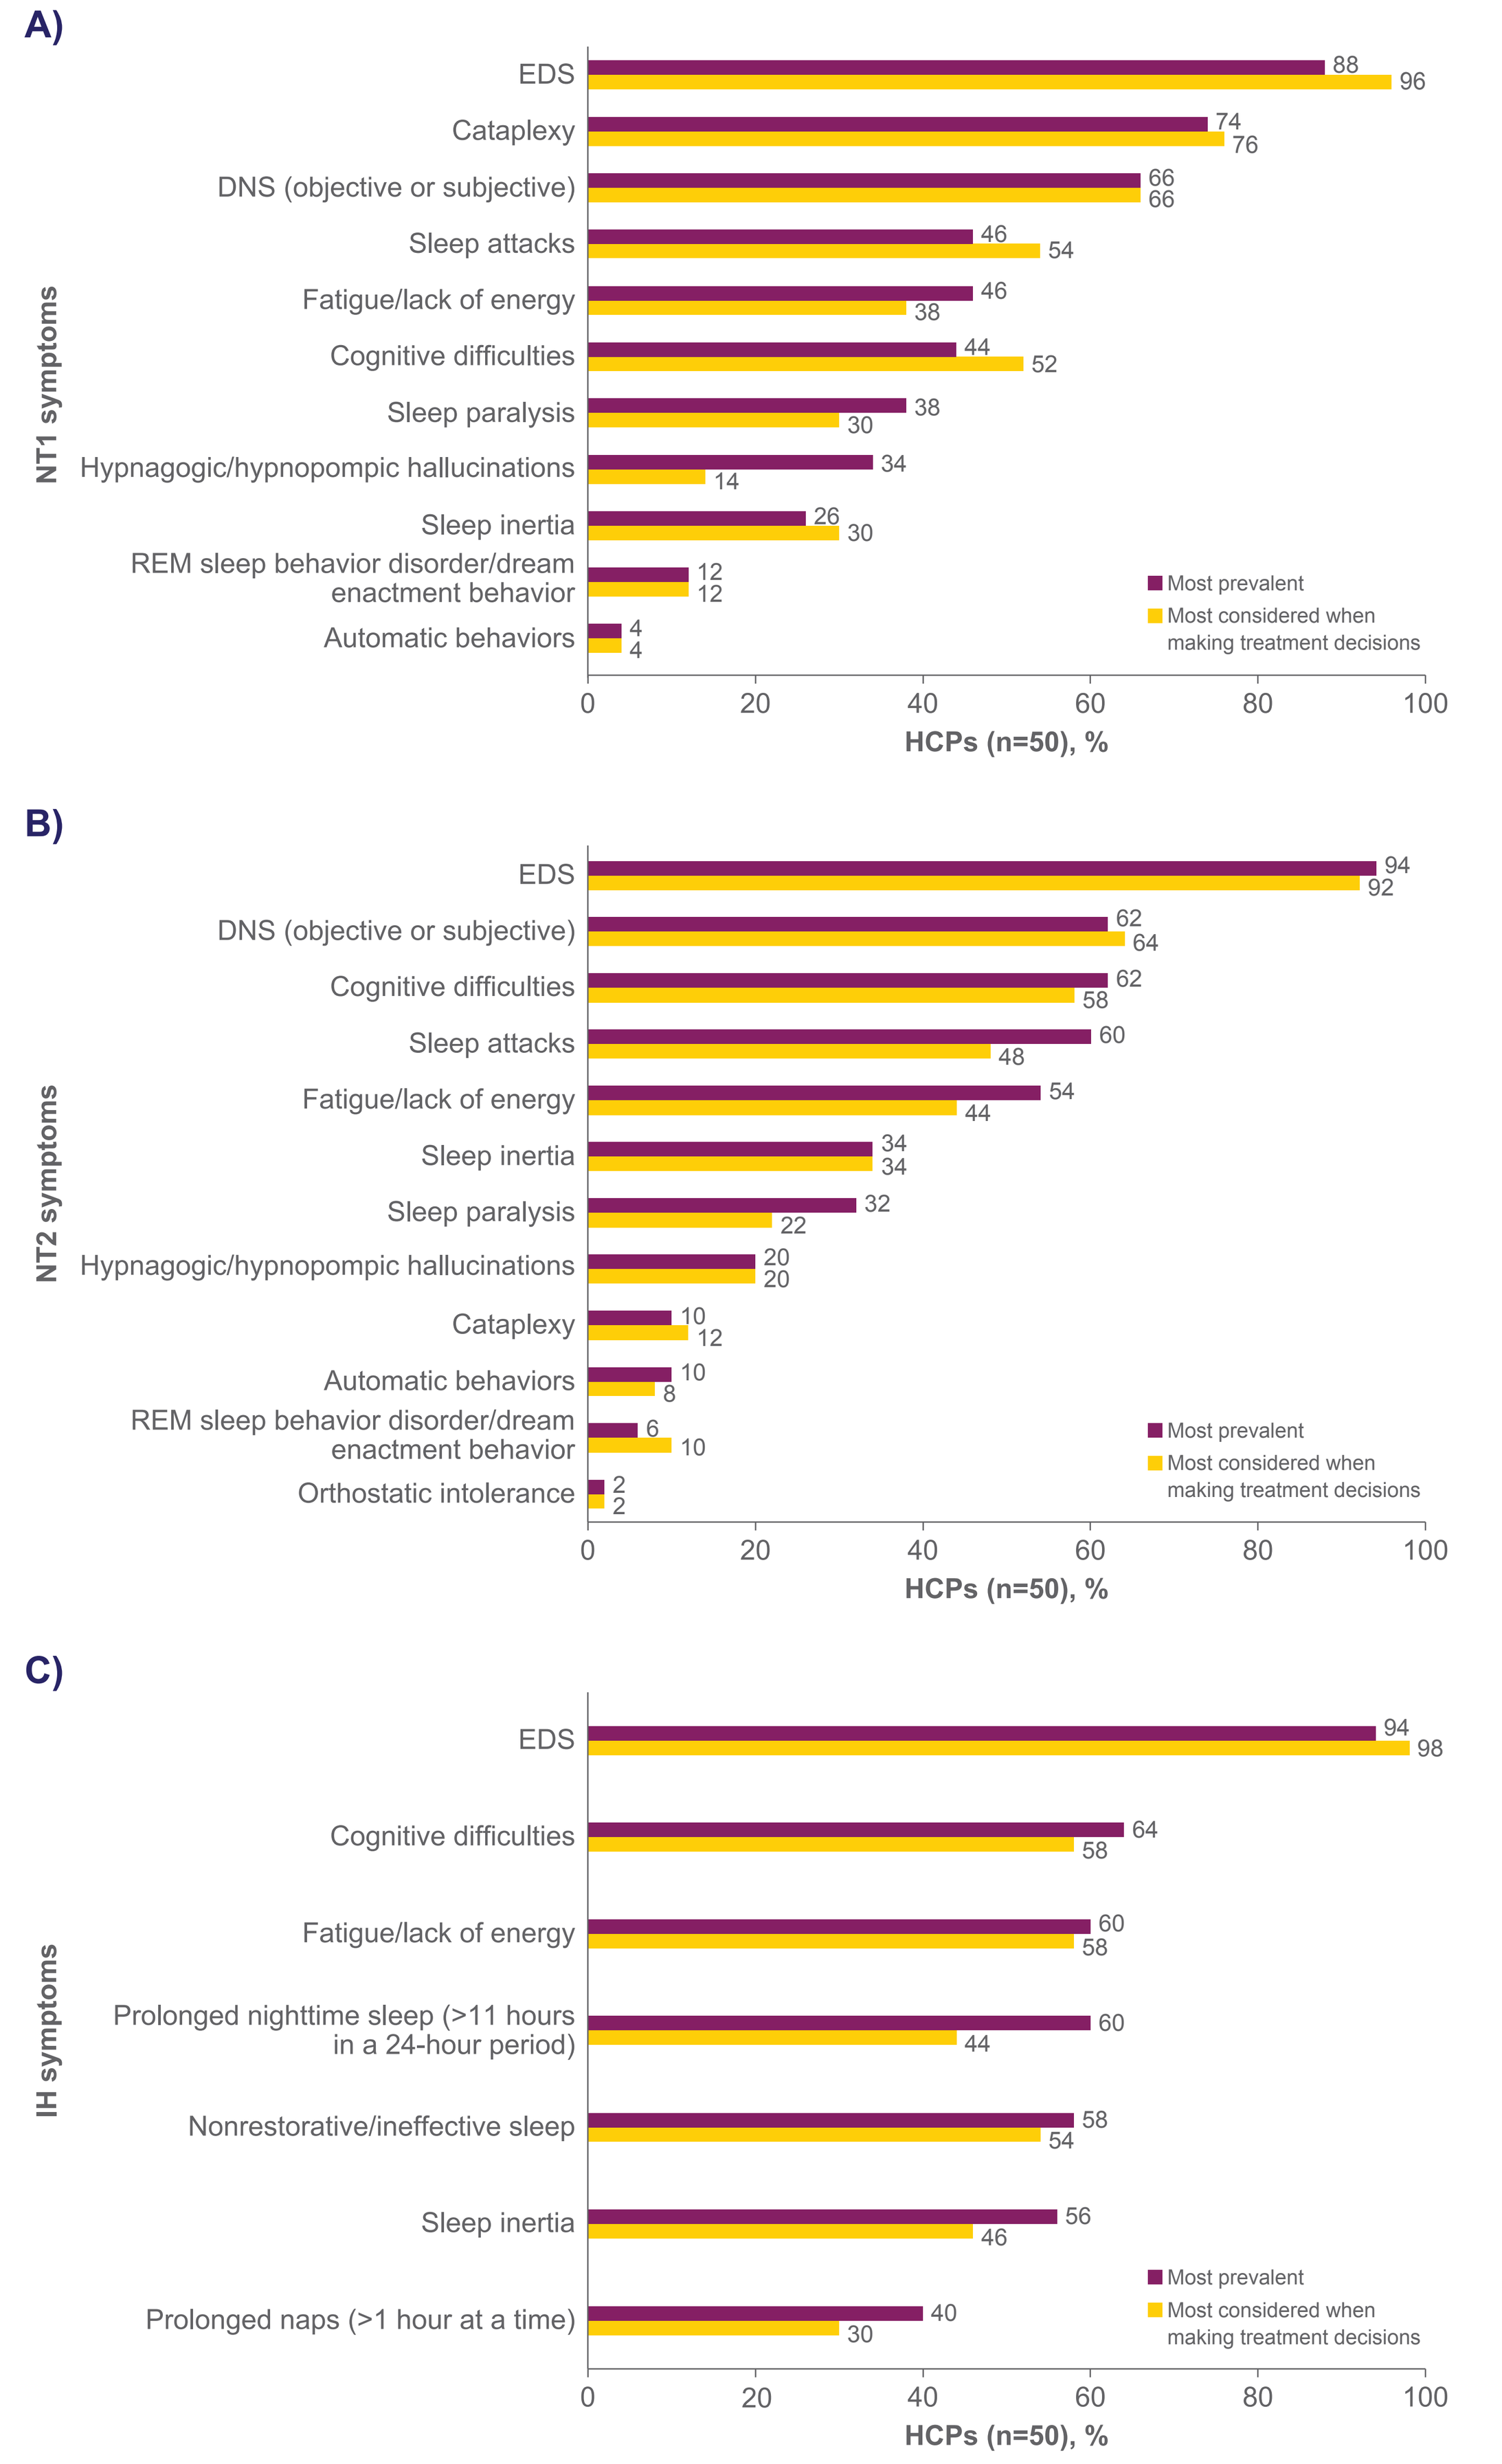
A)
NT1 symptoms
EDS 88 96
Cataplexy 74 76
DNS (objective or subjective) 66 66
Sleep attacks 46 54
Fatigue/lack of energy 46 38
Cognitive difficulties 44 52
Sleep paralysis 38 30
Hypnagogic/hypnopompic hallucinations 34 14
Sleep inertia 26 30
REM sleep behavior disorder/dream enactment behavior 12 12
Automatic behaviors 4 4
HCPs (n=50), %
B)
NT2 symptoms
EDS 94 92
DNS (objective or subjective) 62 64
Cognitive difficulties 62 58
Sleep attacks 60 48
Fatigue/lack of energy 54 44
Sleep inertia 34 34
Sleep paralysis 32 22
Hypnagogic/hypnopompic hallucinations 20 20
Cataplexy 10 12
Automatic behaviors 10 8
REM sleep behavior disorder/dream enactment behavior 6 10
Orthostatic intolerance 2 2
HCPs (n=50), %
C)
IH symptoms
EDS 94 98
Cognitive difficulties 64 58
Fatigue/lack of energy 60 58
Prolonged nighttime sleep (>11 hours in a 24-hour period) 60 44
Nonrestorative/ineffective sleep 58 54
Sleep inertia 56 46
Prolonged naps (>1 hour at a time) 40 30
HCPs (n=50), %
Most prevalent
Most considered when making treatment decisions
0 20 40 60 80 100

[a]HCPs were asked, "What are the most prevalent symptoms reported by patients with [NT1/NT2/IH]? Please select up to 5 top symptoms." [b]HCPs were asked, "Which symptoms do you most consider when determining potential treatment(s) for [NT1/NT2/IH]? Please select up to 5 top symptoms." The graphs present endorsed symptoms.
DNS, disrupted nighttime sleep; EDS, excessive daytime sleepiness; HCP, healthcare provider; IH, idiopathic hypersomnia; NT1, narcolepsy type 1; NT2, narcolepsy type 2; REM, rapid eye movement.

- More than 88% of HCPs identified EDS as the most prevalent symptom in narcolepsy and IH; nearly all (92%–98%) considered EDS as a top symptom when determining potential treatment approaches
 - Efficacy for treating EDS was among the most important factors influencing HCP treatment decisions for NT1, NT2, and IH; additional factors are available in the supplement, accessed via the QR code
- HCPs cited nighttime sleep-related symptoms less frequently than EDS as key treatment decision drivers, including disrupted nighttime sleep in NT1 (66%) and NT2 (62%), and nonrestorative/ineffective sleep in IH (54%)

Figure 3. (A) PwN and (B) PwIH Rated a Broad Spectrum of Nighttime Sleep and Daytime Symptoms as "Extremely Bothersome"[a]

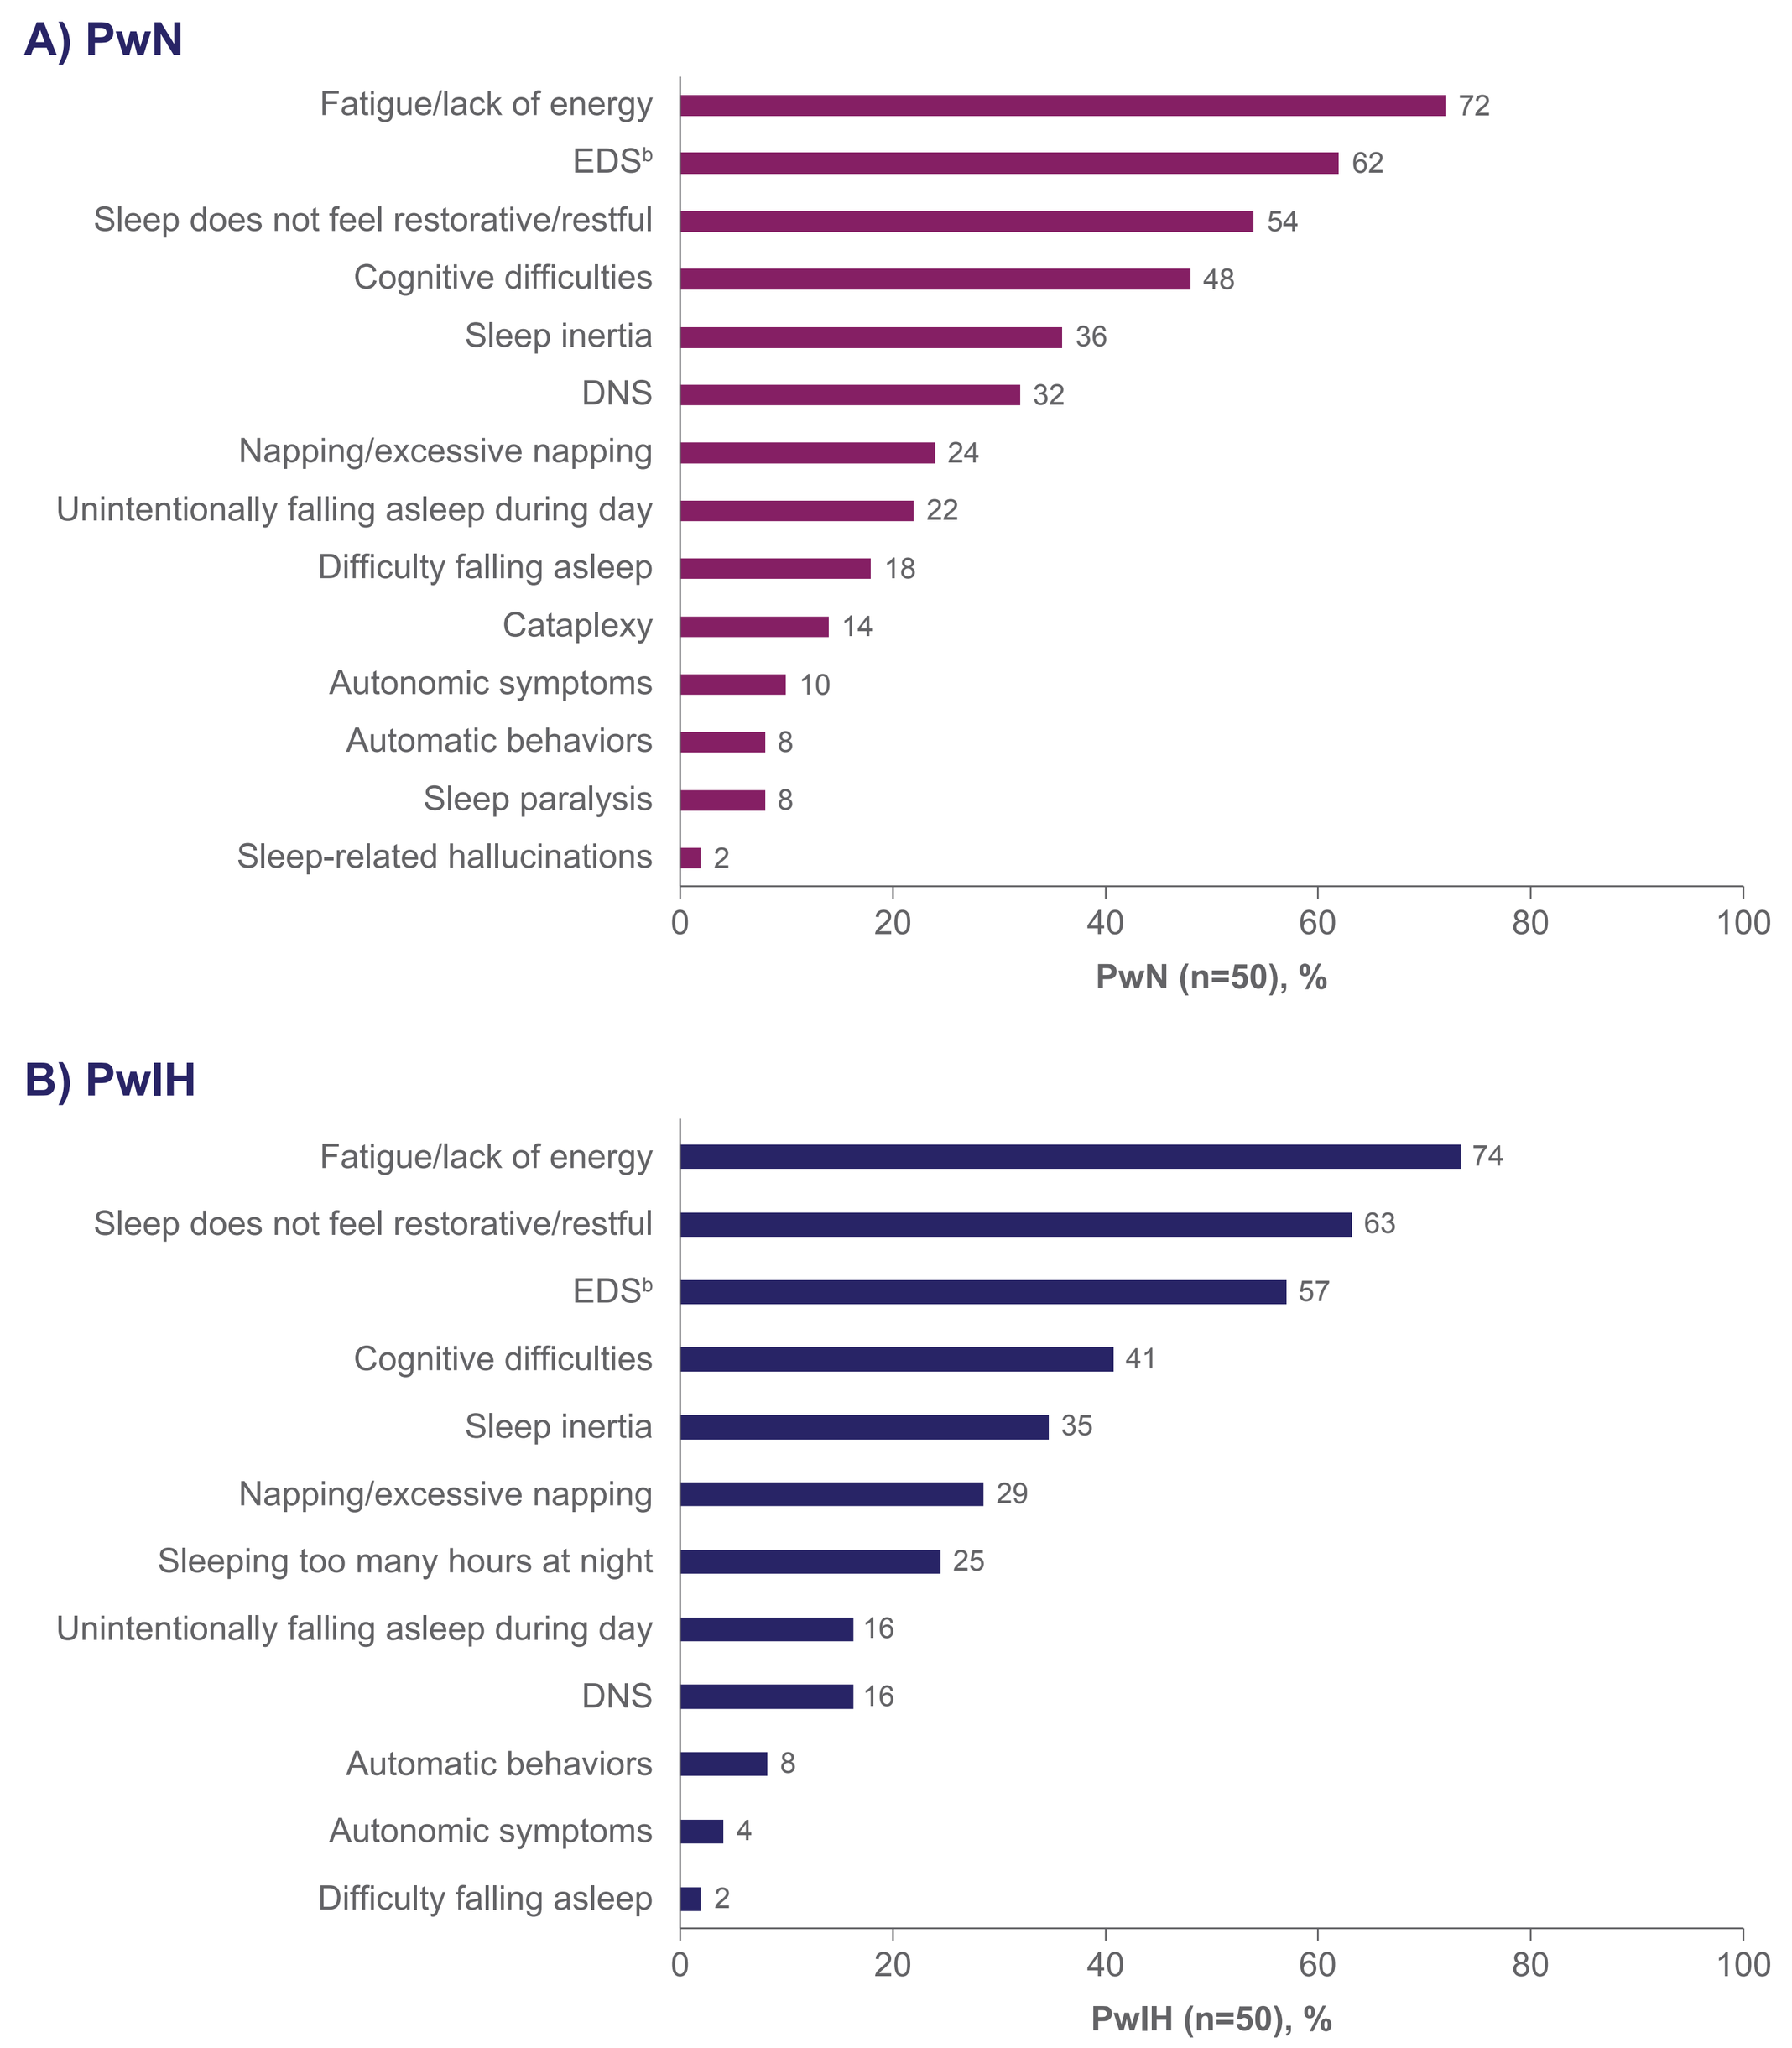
A) PwN
Fatigue/lack of energy 72
EDSb 62
Sleep does not feel restorative/restful 54
Cognitive difficulties 48
Sleep inertia 36
DNS 32
Napping/excessive napping 24
Unintentionally falling asleep during day 22
Difficulty falling asleep 18
Cataplexy 14
Autonomic symptoms 10
Automatic behaviors 8
Sleep paralysis 8
Sleep-related hallucinations 2
PwN (n=50), %
B) PwIH
Fatigue/lack of energy 74
Sleep does not feel restorative/restful 63
EDSb 57
Cognitive difficulties 41
Sleep inertia 35
Napping/excessive napping 29
Sleeping too many hours at night 25
Unintentionally falling asleep during day 16
DNS 16
Automatic behaviors 8
Autonomic symptoms 4
Difficulty falling asleep 2
PwIH (n=50), %
0 20 40 60 80 100

[a]Patient participants rated each symptom from 1–5, where 1 indicated that the symptom was not at all bothersome and 5 that the symptom was extremely bothersome. Participants could also indicate that they did not experience the symptom. The graphs present the proportion of patient participants rating symptoms as "extremely bothersome." [b]EDS was presented as "daytime sleepiness" in the patient survey.
DNS, disrupted nighttime sleep; EDS, excessive daytime sleepiness; PwIH, patients with idiopathic hypersomnia; PwN, patients with narcolepsy.

- PwN and PwIH reported a broad symptom burden, frequently rating fatigue (PwN: 72%; PwIH: 74%) and nonrestorative sleep (PwN: 54%; PwIH: 63%) as extremely bothersome, comparable to EDS (PwN: 62%; PwIH: 57%)

Figure 4. (A) PwN and (B) PwIH Reporting Nighttime Sleep and Daytime Symptoms as Extremely Bothersome Were More Likely to Report Extreme Impacts on Daily Life[a]

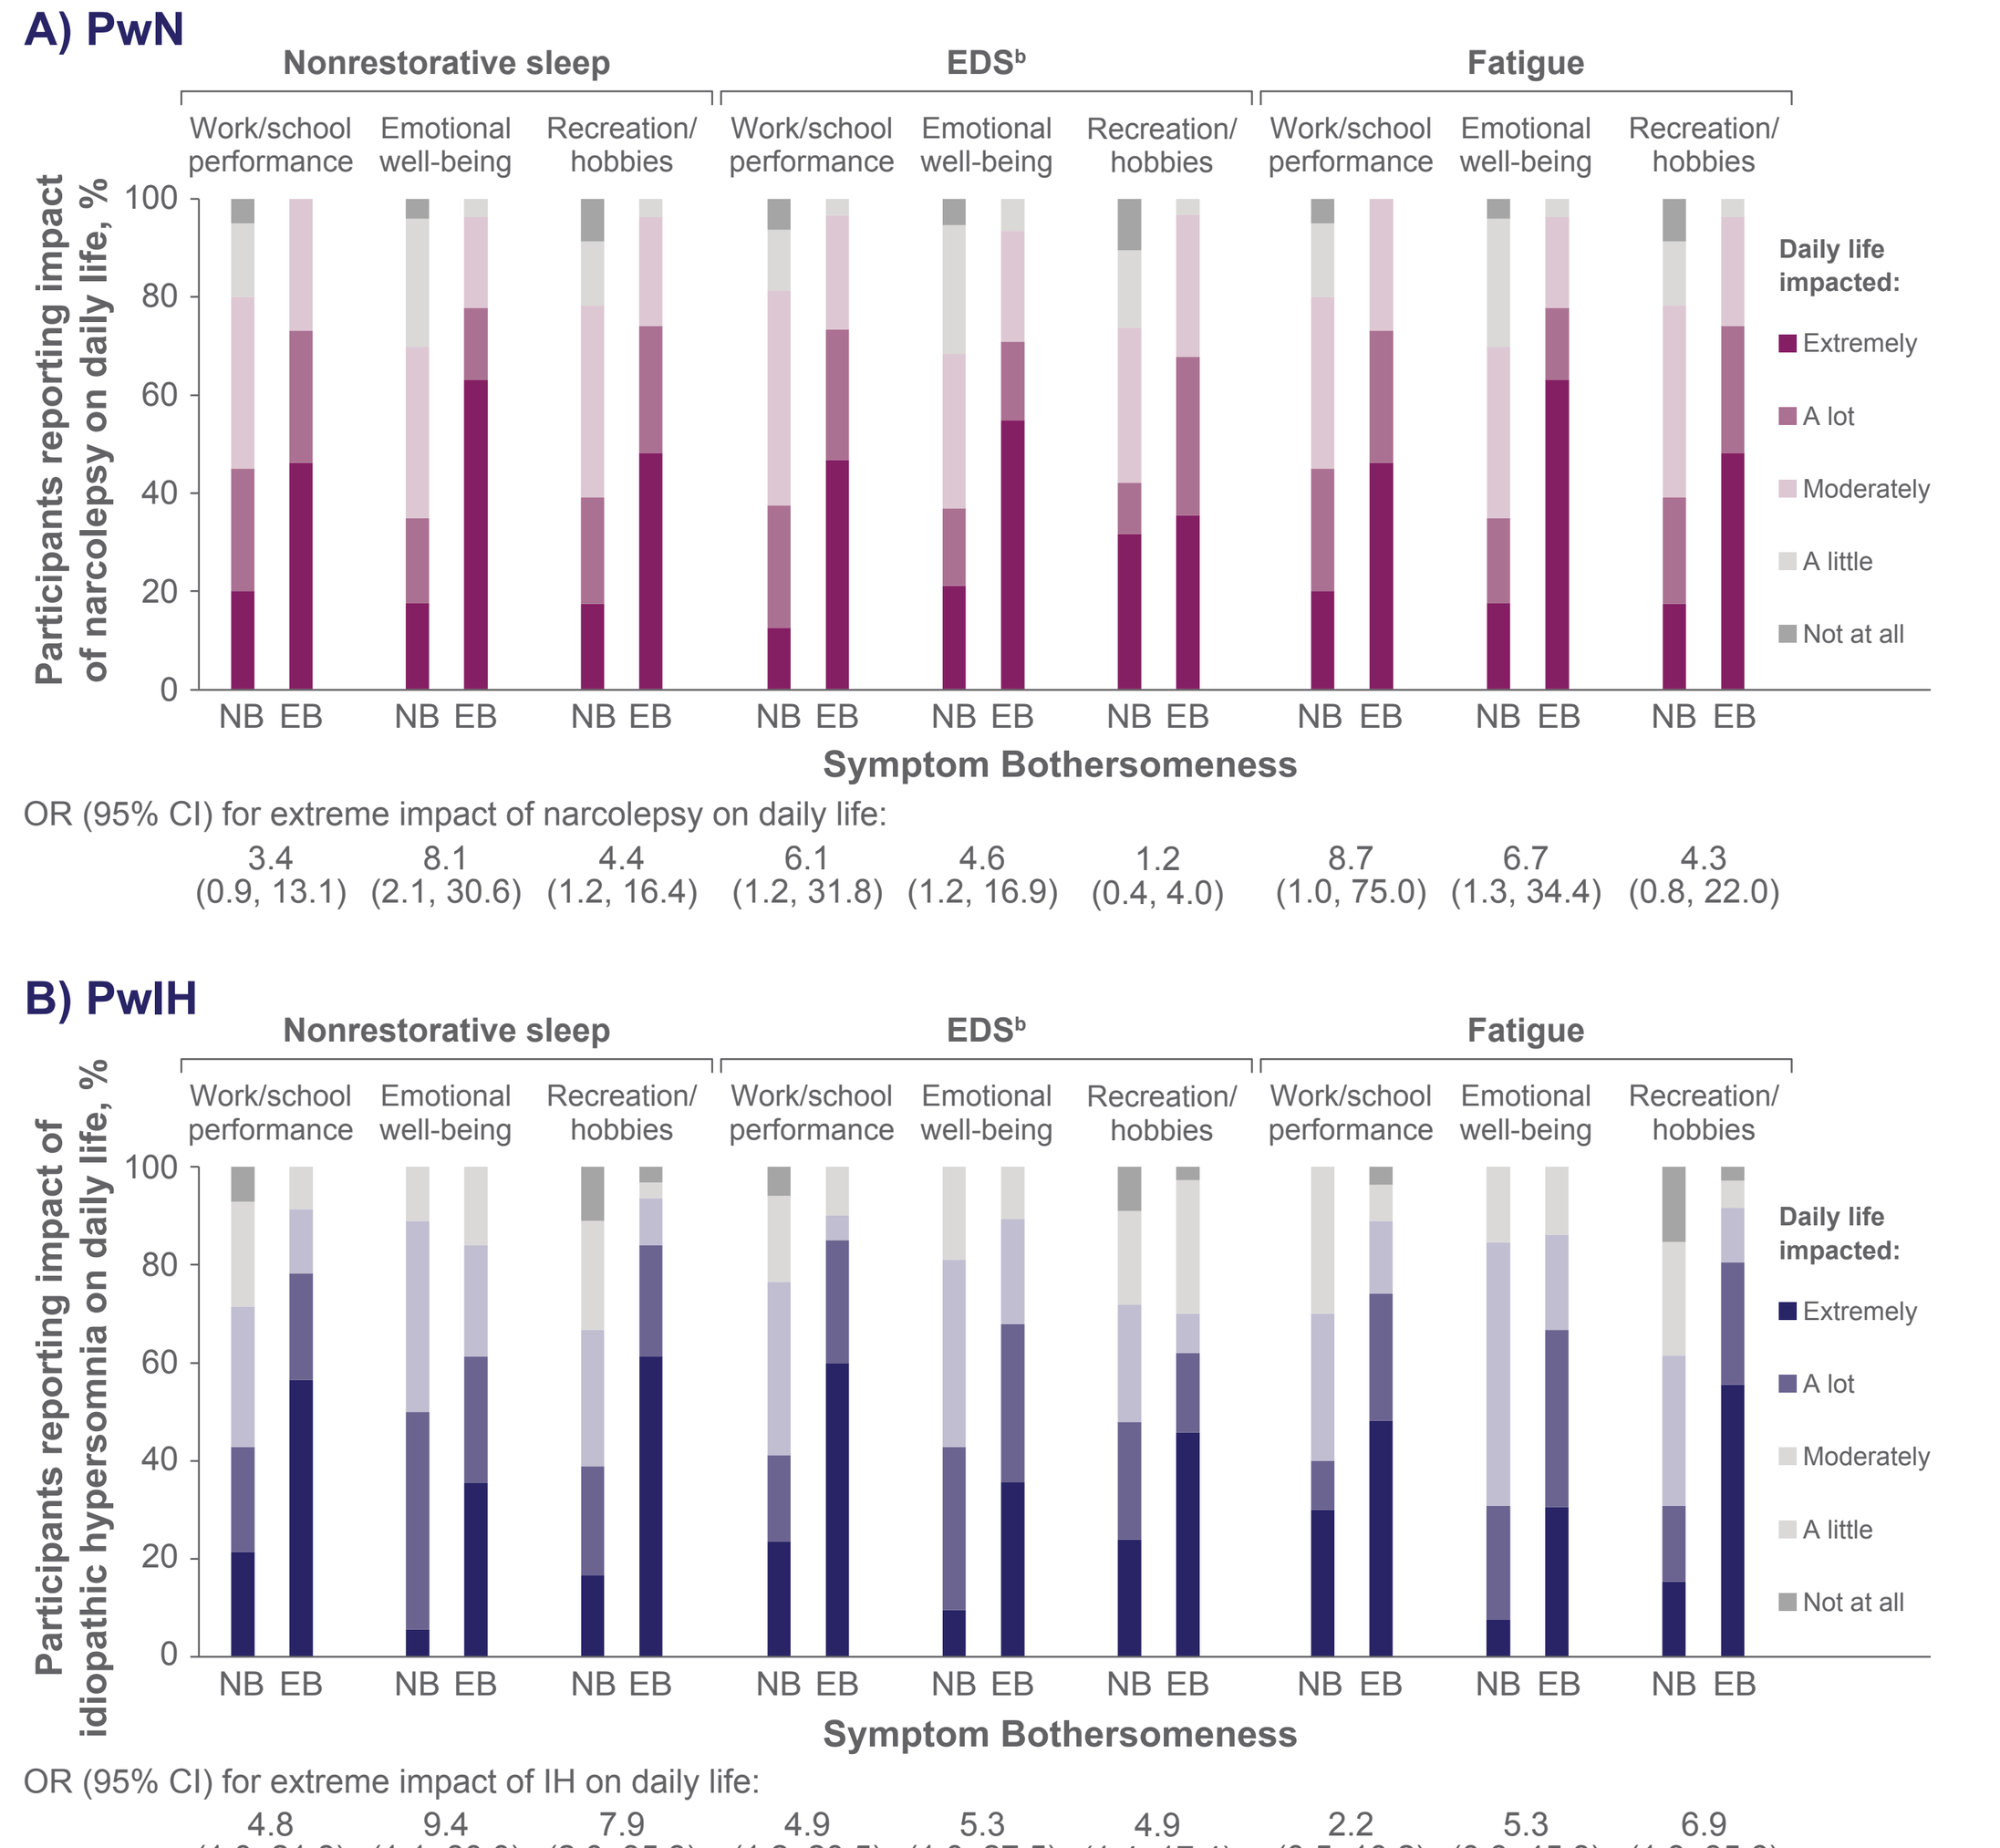
A) PwN
Nonrestorative sleep
EDSb
Fatigue
Work/school performance
Emotional well-being
Recreation/hobbies
Participants reporting impact of narcolepsy on daily life, %
NB EB
Symptom Bothersomeness
Daily life impacted:
Extremely
A lot
Moderately
A little
Not at all
OR (95% CI) for extreme impact of narcolepsy on daily life:
3.4 (0.9, 13.1)
8.1 (2.1, 30.6)
4.4 (1.2, 16.4)
6.1 (1.2, 31.8)
4.6 (1.2, 16.9)
1.2 (0.4, 4.0)
8.7 (1.0, 75.0)
6.7 (1.3, 34.4)
4.3 (0.8, 22.0)
B) PwIH
Participants reporting impact of idiopathic hypersomnia on daily life, %
OR (95% CI) for extreme impact of IH on daily life:
4.8 (1.0, 21.8)
9.4 (1.1, 80.0)
7.9 (2.0, 35.3)
4.9 (1.2, 20.5)
5.3 (1.0, 27.5)
4.9 (1.4, 17.4)
2.2 (0.5, 10.2)
5.3 (0.6, 45.8)
6.9 (1.3, 35.6)

[a]Patient participants were asked, "How bothersome do you find each of the following symptoms related to your [narcolepsy/IH]? Please rate each symptom on a scale from 1 (not at all bothersome) to 5 (extremely bothersome)" and "To what extent does [narcolepsy/IH] currently (ie, within the past month) impact the following aspects of your life?" Odds ratios compare the odds of reporting an extreme impact on daily life domains among those reporting a symptom as extremely bothersome vs those not reporting the symptom as extremely bothersome. Participants who reported "Not applicable" for impacts were excluded. Sample sizes for PwN who were extremely bothered by each symptom shown ranged from 26 to 36; sample sizes for PwIH who were extremely bothered by each symptom shown ranged from 20 to 36. [b]EDS was presented as "daytime sleepiness" in the patient survey.
CI, confidence interval; EB, extremely bothersome; EDS, excessive daytime sleepiness; IH, idiopathic hypersomnia; NB, not extremely bothersome; OR, odds ratio; PwIH, patients with idiopathic hypersomnia; PwN, patients with narcolepsy.

- Many PwN and PwIH who reported being extremely bothered by nonrestorative sleep, EDS, and/or fatigue also reported extreme impacts of narcolepsy or IH on work/school performance, emotional well-being, and recreation/hobbies
- Nonrestorative sleep may be associated with extreme impacts on daily life to a similar or greater extent than EDS and fatigue, underscoring the burden of nighttime sleep complaints in addition to daytime symptoms experienced by PwN and PwIH

Figure 5. Top 3 Most Important Treatment Attributes[a] for (A) PwN and (B) PwIH

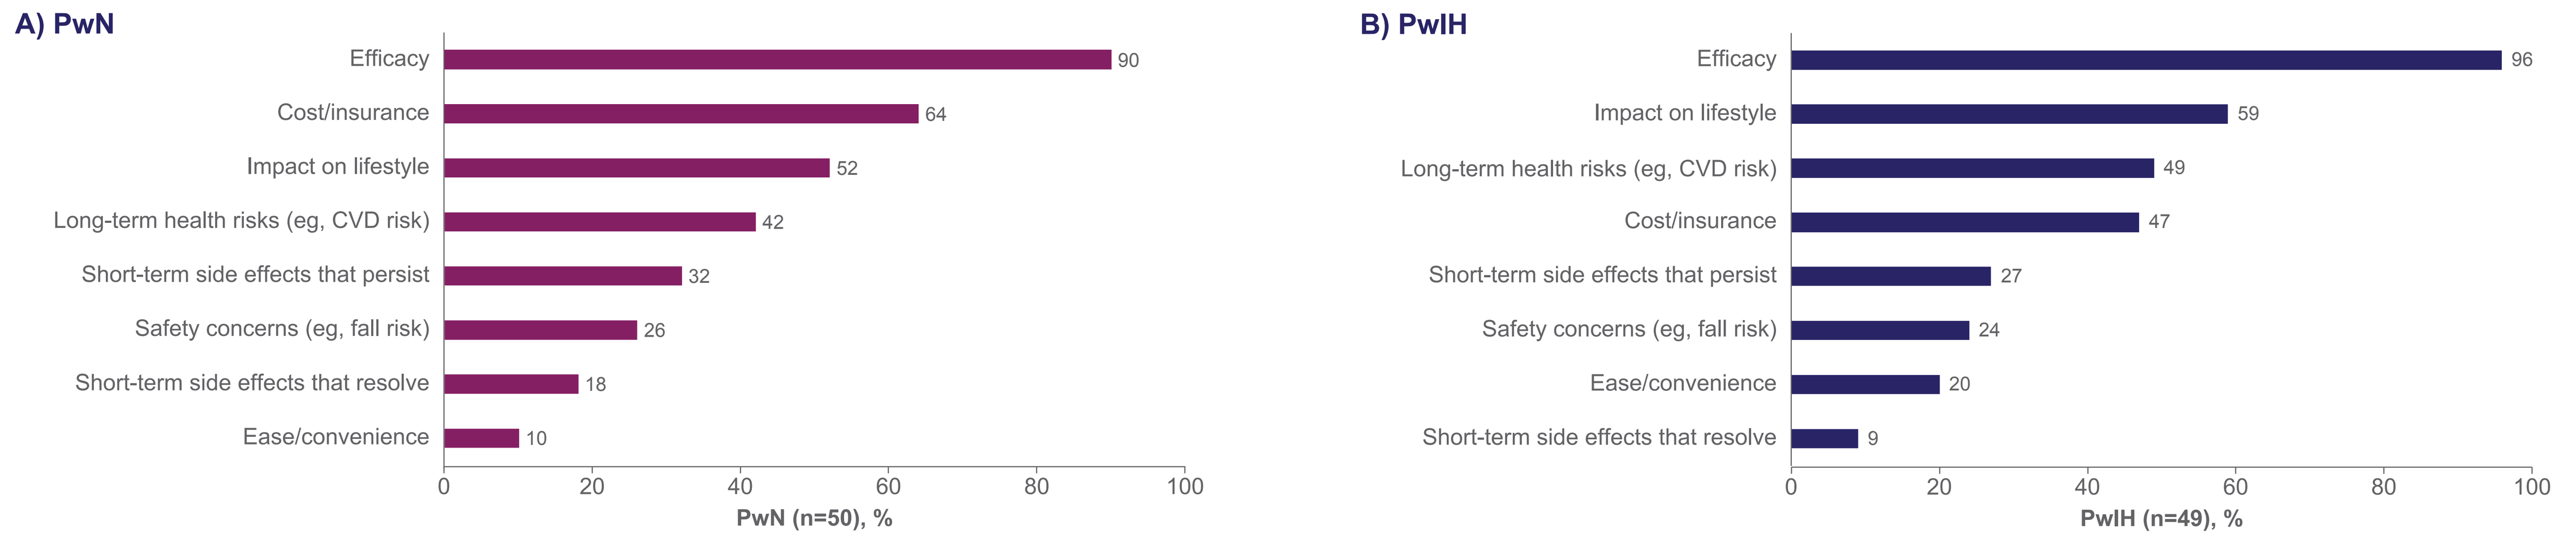
A) PwN
Efficacy 90
Cost/insurance 64
Impact on lifestyle 52
Long-term health risks (eg, CVD risk) 42
Short-term side effects that persist 32
Safety concerns (eg, fall risk) 26
Short-term side effects that resolve 18
Ease/convenience 10
PwN (n=50), %
B) PwIH
Efficacy 96
Impact on lifestyle 59
Long-term health risks (eg, CVD risk) 49
Cost/insurance 47
Short-term side effects that persist 27
Safety concerns (eg, fall risk) 24
Ease/convenience 20
Short-term side effects that resolve 9
PwIH (n=49), %
0 20 40 60 80 100

[a]Patient participants ranked the most important treatment attributes from 1–8; they could select the same rank for different attributes. The graphs present the proportion of patient participants ranking treatment attributes among the top 3 out of 8.
CVD, cardiovascular disease; PwIH, patients with idiopathic hypersomnia; PwN, patients with narcolepsy.

- Efficacy was highly ranked as an important treatment attribute by both PwN (90%) and PwIH (96%)
- PwIH and PwN further emphasized cost/insurance, impact on lifestyle, and long-term health risks (eg, cardiovascular disease risk) as important attributes that factor into treatment decision-making

150

Patient and Healthcare Provider Perspectives on Symptom Burden and Treatment Decision-Making in Narcolepsy and Idiopathic Hypersomnia

Caroleen Drachenberg, PhD, MSPH[1]; Sarah C. Markt, ScD, MPH[1]; Samantha Linton, PhD[2]; Meagan Farrell, PhD[2]; Jennifer D'Souza, PhD[2]; Jennifer N. Hill, MA[2]; Christina A. Graham, BS, BA[2]; Jessica K. Alexander, PhD[1]; Marisa Whalen, PharmD[3]; Silky Beaty, PharmD, MSPH[1]; Barbara Hutchinson, MD, PhD, FACC[4]; Shane Drahos, MD[5]

[1]Jazz Pharmaceuticals, Palo Alto, CA, USA; [2]IQVIA, Boston, MA, USA; [3]Jazz Pharmaceuticals, Philadelphia, PA, USA; [4]Chesapeake Cardiac Care, Bowie, MD, USA; [5]Premier Sleep Medicine, Winter Garden, FL, USA

Supplemental Material

Supplemental Figure S1. Factors Influencing HCP Treatment Decisions[a] for (A) NT1, (B) NT2, and (C) IH

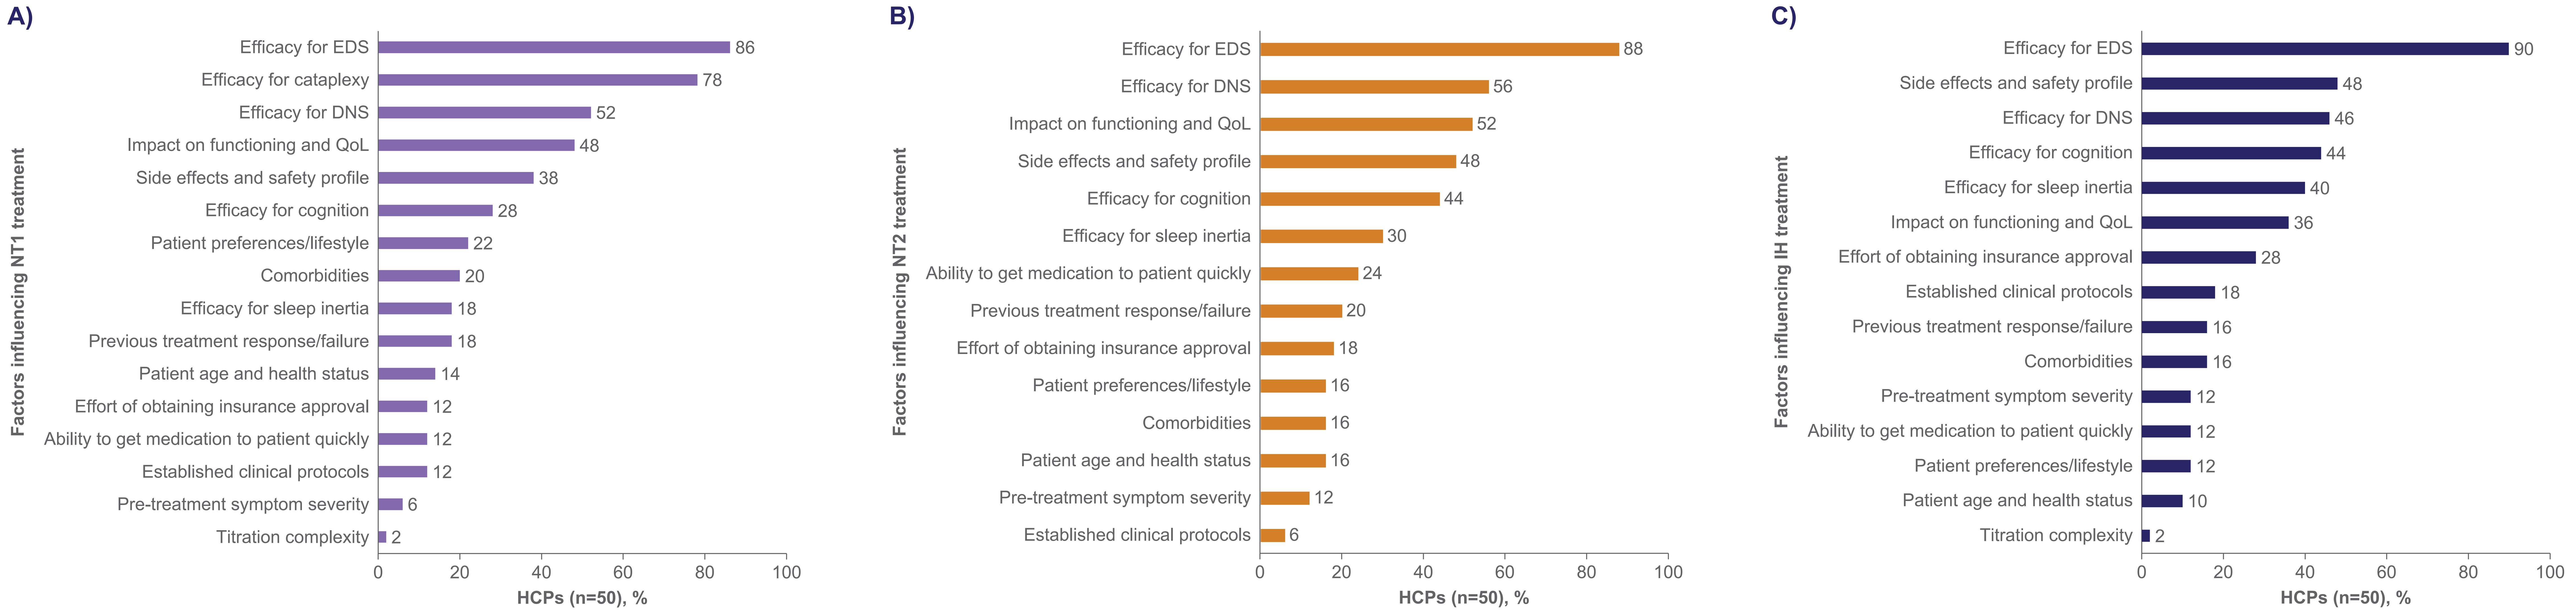

A)
Factors influencing NT1 treatment
Efficacy for EDS 86
Efficacy for cataplexy 78
Efficacy for DNS 52
Impact on functioning and QoL 48
Side effects and safety profile 38
Efficacy for cognition 28
Patient preferences/lifestyle 22
Comorbidities 20
Efficacy for sleep inertia 18
Previous treatment response/failure 18
Patient age and health status 14
Effort of obtaining insurance approval 12
Ability to get medication to patient quickly 12
Established clinical protocols 12
Pre-treatment symptom severity 6
Titration complexity 2
0 20 40 60 80 100
HCPs (n=50), %
B)
Factors influencing NT2 treatment
Efficacy for EDS 88
Efficacy for DNS 56
Impact on functioning and QoL 52
Side effects and safety profile 48
Efficacy for cognition 44
Efficacy for sleep inertia 30
Ability to get medication to patient quickly 24
Previous treatment response/failure 20
Effort of obtaining insurance approval 18
Patient preferences/lifestyle 16
Comorbidities 16
Patient age and health status 16
Pre-treatment symptom severity 12
Established clinical protocols 6
0 20 40 60 80 100
HCPs (n=50), %
C)
Factors influencing IH treatment
Efficacy for EDS 90
Side effects and safety profile 48
Efficacy for DNS 46
Efficacy for cognition 44
Efficacy for sleep inertia 40
Impact on functioning and QoL 36
Effort of obtaining insurance approval 28
Established clinical protocols 18
Previous treatment response/failure 16
Comorbidities 16
Pre-treatment symptom severity 12
Ability to get medication to patient quickly 12
Patient preferences/lifestyle 12
Patient age and health status 10
Titration complexity 2
0 20 40 60 80 100
HCPs (n=50), %

[a]HCP participants were asked, "What factors influence your choice in prescribing treatments for [NT1/NT2/IH]? Please select up to 5 top factors."
DNS, disrupted nighttime sleep; EDS, excessive daytime sleepiness; HCP, healthcare provider; IH, idiopathic hypersomnia; NT1, narcolepsy type 1; NT2, narcolepsy type 2; QoL, quality of life.

- Efficacy for treating excessive daytime sleepiness was a chief factor for healthcare providers when making treatment decisions for patients with narcolepsy type 1 (86%), narcolepsy type 2 (88%), and idiopathic hypersomnia (90%)

Scan this code to access a plain language summary of the published abstract. This code is not for promotional purposes.

Scan this code to access this poster and supplemental material online. This code is not for promotional purposes.

Presented at Psych Congress 2026; September 15–19, 2026; New Orleans, LA, USA