153

Clinical Responders to Low-Sodium Oxybate in Narcolepsy and Idiopathic Hypersomnia: Evaluation of Epworth Sleepiness Scale and Idiopathic Hypersomnia Severity Scale Established Thresholds and Normal Scores

Chad M. Ruoff, MD[1]; R. Bart Sangal, MD[2,3]; Deborah A. Nichols, MS[4]; Teresa L. Steininger, PhD[4]; Jing Dai, PhD[4]; Shawn Vahabzadeh, PharmD[5]; Marisa Whalen, PharmD[5]; Sarah C. Markt, ScD, MPH[4]; Laura B. Herpel, MD[6,7]; Nancy Foldvary-Schaefer, DO, MS[8]

[1]Mayo Clinic, Scottsdale, AZ, USA; [2]Obesity and Sleep Medicine, Bloomfield Hills, MI, USA; [3]Oakland University William Beaumont School of Medicine, Rochester Hills, MI, USA; [4]Jazz Pharmaceuticals, Palo Alto, CA, USA; [5]Jazz Pharmaceuticals, Philadelphia, PA, USA; [6]Bogan Sleep Consultants, Columbia, SC, USA; [7]Medical University of South Carolina, Charleston, SC, USA; [8]Sleep Disorders Center, Neurological Institute, Cleveland Clinic, Cleveland, OH, USA

Objective

- To describe the proportions of DUET participants with narcolepsy or idiopathic hypersomnia who achieved a clinically important response to low-sodium oxybate (LXB) treatment based on established thresholds and normal score ranges on the Epworth Sleepiness Scale (ESS; both cohorts) and Idiopathic Hypersomnia Severity Scale (IHSS; idiopathic hypersomnia cohort only)

Conclusions

- The majority of DUET participants met the American Academy of Sleep Medicine ≥2-point ESS clinical improvement criterion (narcolepsy, 82.4%; idiopathic hypersomnia, 95.0%) and achieved an ESS score within normal range (≤10; narcolepsy, 70.6%; idiopathic hypersomnia, 70.0%) after optimization of LXB treatment
- The majority of participants with idiopathic hypersomnia achieved clinically meaningful IHSS improvement (reduction of ≥4 points; 94.4%) as well as an IHSS score within normal range (<22; 69.4%) after optimization of LXB treatment
- Clinically meaningful improvements were observed in the majority of participants with narcolepsy or idiopathic hypersomnia, including in those with and without concomitant alerting agents use
- These findings suggest a consistent and clinically important response to LXB treatment compared with baseline in people with narcolepsy or idiopathic hypersomnia

References: 1. American Academy of Sleep Medicine. *International Classification of Sleep Disorders – Third Edition, Text Revision.* Darien, IL: American Academy of Sleep Medicine; 2023. **2.** Johns MW. *Sleep.* 1991;14(6):540-545. **3.** Dauvilliers Y, et al. *Neurology.* 2019;92(15):e1754-e1762. **4.** Rassu AL, et al. *J Clin Sleep Med.* 2022;18(2):617-629. **5.** Xywav (calcium, magnesium, potassium, and sodium oxybates) oral solution, CIII [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **6.** Schneider LD, et al. *Neurol Ther.* 2026;15:1613–1641. **7.** Plante DT, et al. *CNS Drugs.* 2026;40:803-822. **8.** Nichols DA, et al. *Neurol Ther.* 2025;14(4):1705-1727. **9.** Maski K, et al. *J Clin Sleep Med.* 2021;17(9):1895-1945.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company) employees Martina Kusi-Mensah, PharmD, and Emily Bruggeman, PhD, provided medical writing support and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: CM Ruoff has served as an advisory board member for Jazz Pharmaceuticals, Eisai, Alkermes, and Takeda, and has received grant funding from Jazz Pharmaceuticals. **RB Sangal** has received research support from Alkermes, Avadel, Apnimed, Centessa, Incannex, Jazz Pharmaceuticals, Zevra, Harmony Biosciences, and Suven Life Sciences. **DA Nichols**, **J Dai**, **S Vahabzadeh**, **M Whalen**, and **SC Markt** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **TL Steininger** is a former full-time employee and current contract worker for Jazz Pharmaceuticals who previously has received shares of Jazz Pharmaceuticals, plc. **LB Herpel** is an affiliate assistant professor at the Medical University of South Carolina; participates in clinical research for Alkermes, Axsome Therapeutics, Eisai, Eli Lilly, Harmony Biosciences, Idorsia, Jazz Pharmaceuticals, LivaNova/OSPREY, Merck, Noctrix, Roche, Samsung, Sanofi, Suven, Takeda, and Vanda; served as an advisor to Avadel, Alkermes, Axsome, Eli Lilly, Harmony Biosciences, and Jazz Pharmaceuticals; and serves on the speaker bureau for Alkermes, Harmony Biosciences, and Takeda as well as sponsored educational presentations for Jazz Pharmaceuticals. **N Foldvary-Schaefer** has received research grants from Avadel, Harmony Biosciences, Jazz Pharmaceuticals, Suven, Takeda, and Vanda; served on a publication committee and advisory committees for Jazz Pharmaceuticals; served on advisory boards for Takeda and Alkermes; and received royalites from UpToDate and Oxford University Press.

Scan this code to access a plain language summary of the published abstract. This code is not for promotional purposes.

Scan this code to access this poster and supplemental material online. This code is not for promotional purposes.

Presented at Psych Congress 2026; September 15–19, 2026; New Orleans, LA, USA

Introduction

- Narcolepsy and idiopathic hypersomnia are central disorders of hypersomnolence that are characterized by a constellation of symptoms that impact nighttime sleep and daytime functioning, including excessive daytime sleepiness (EDS)[1]
- The Epworth Sleepiness Scale (ESS) is a validated subjective assessment of EDS[2]; the Idiopathic Hypersomnia Severity Scale (IHSS) is a validated subjective measure of the severity, frequency, and functional impact of idiopathic hypersomnia symptoms[3,4]
- Low-sodium oxybate (LXB; Xywav®) is approved by the US Food and Drug Administration for the treatment of cataplexy or EDS in people with narcolepsy ≥7 years of age and for the treatment of idiopathic hypersomnia in adults[5]
- LXB has demonstrated clinically meaningful improvements in EDS in both narcolepsy and idiopathic hypersomnia, and clinically meaningful improvements in idiopathic hypersomnia symptom severity based on IHSS[6,7]

Methods

- DUET was a phase 4, prospective, single-arm, open-label study (NCT05875974)

Figure 1. DUET Study Design

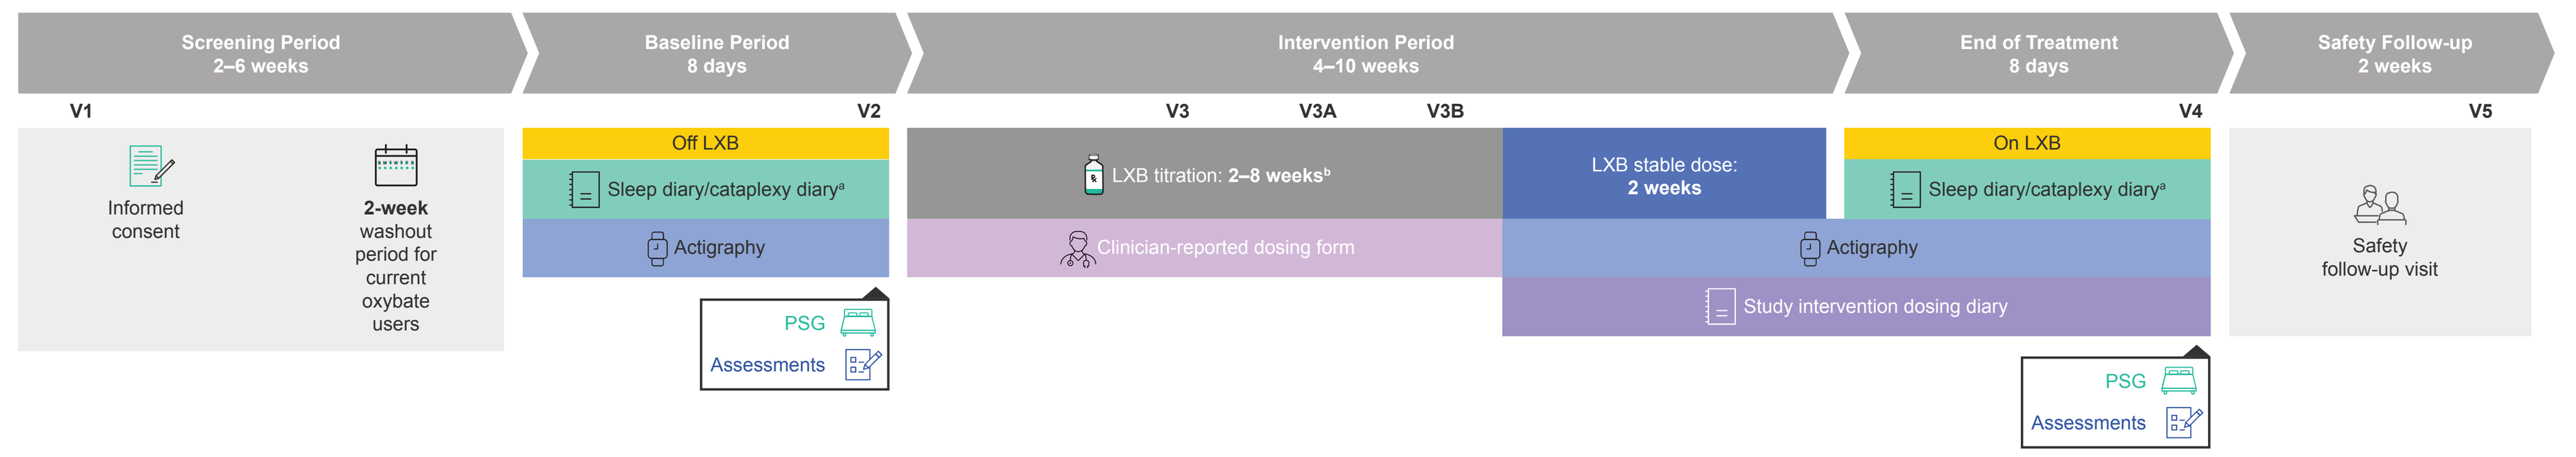
Screening Period
2–6 weeks
Baseline Period
8 days
Intervention Period
4–10 weeks
End of Treatment
8 days
Safety Follow-up
2 weeks
V1
V2
V3
V3A
V3B
V4
V5
Informed consent
2-week washout period for current oxybate users
Off LXB
Sleep diary/cataplexy diary[a]
Actigraphy
PSG
Assessments
LXB titration: 2–8 weeks[b]
Clinician-reported dosing form
LXB stable dose: 2 weeks
Study intervention dosing diary
On LXB
Safety follow-up visit

Figure adapted from Nichols DA, et. al.[8] http://creativecommons.org/licenses/by-nc/4.0/.
[a]Cataplexy diary in narcolepsy type 1 only. [b]Weekly titration visits were by teleconference. Visit 3 occurred on titration day 14. Titration could take between 2 and 8 weeks. Additional in-clinic visits were scheduled for day 35 (visit 3A) and day 56 (visit 3B), as needed. Investigator could optimize participant dosage and move to stable-dose period at visit 3, 3A, or 3B, but not during intervening weekly teleconferences.
DUET, Develop hypersomnia Understanding by Evaluating low-sodium oxybate Treatment; LXB, low-sodium oxybate; PSG, polysomnography; V, visit.

- DUET included participants 18 to 75 years of age with a primary diagnosis of idiopathic hypersomnia or narcolepsy, ESS score >10, and stable use of concomitant anticatapletics or alerting agents allowed
 - Additional study details can be found in the Supplement, accessed via the QR code
- Effectiveness outcomes evaluated were changes from BL to EOT in ESS scores (both cohorts) and IHSS score (idiopathic hypersomnia cohort)
 - ESS assesses daytime sleepiness (8 items on a 4-point scale, total score range 0–24)[2]
 - The minimal clinically important difference (MCID) for ESS per the American Academy of Sleep Medicine is a ≥2-point decrease.[9] Normal scores are ≤10 points[2]
 - IHSS assesses idiopathic hypersomnia symptoms and impacts (14 items, total score range 0–50 points)[3]
 - A previously validated MCID for IHSS is a ≥4-point decrease.[4] Normal scores are <22 points[3]
- Proportions of participants who responded to treatment were descriptively analyzed as follows:
 - MCID response (ESS): Participants who had a ≥2-point decrease on ESS
 - Normal range (ESS): Participants who achieved an ESS score ≤10 points at EOT
 - MCID response (IHSS): Participants (idiopathic hypersomnia only) who had a ≥4-point decrease on IHSS
 - Normal range (IHSS): Participants (idiopathic hypersomnia only) who had an IHSS score <22 points at EOT

Results

Figure 2. Mean Epworth Sleepiness Scale Scores Improved (A, B) and Most Participants With Narcolepsy (C) or Idiopathic Hypersomnia (D) Achieved Clinically Meaningful Improvement and ESS Scores Within Normal Range (≤10) With Optimized LXB Treatment[a]

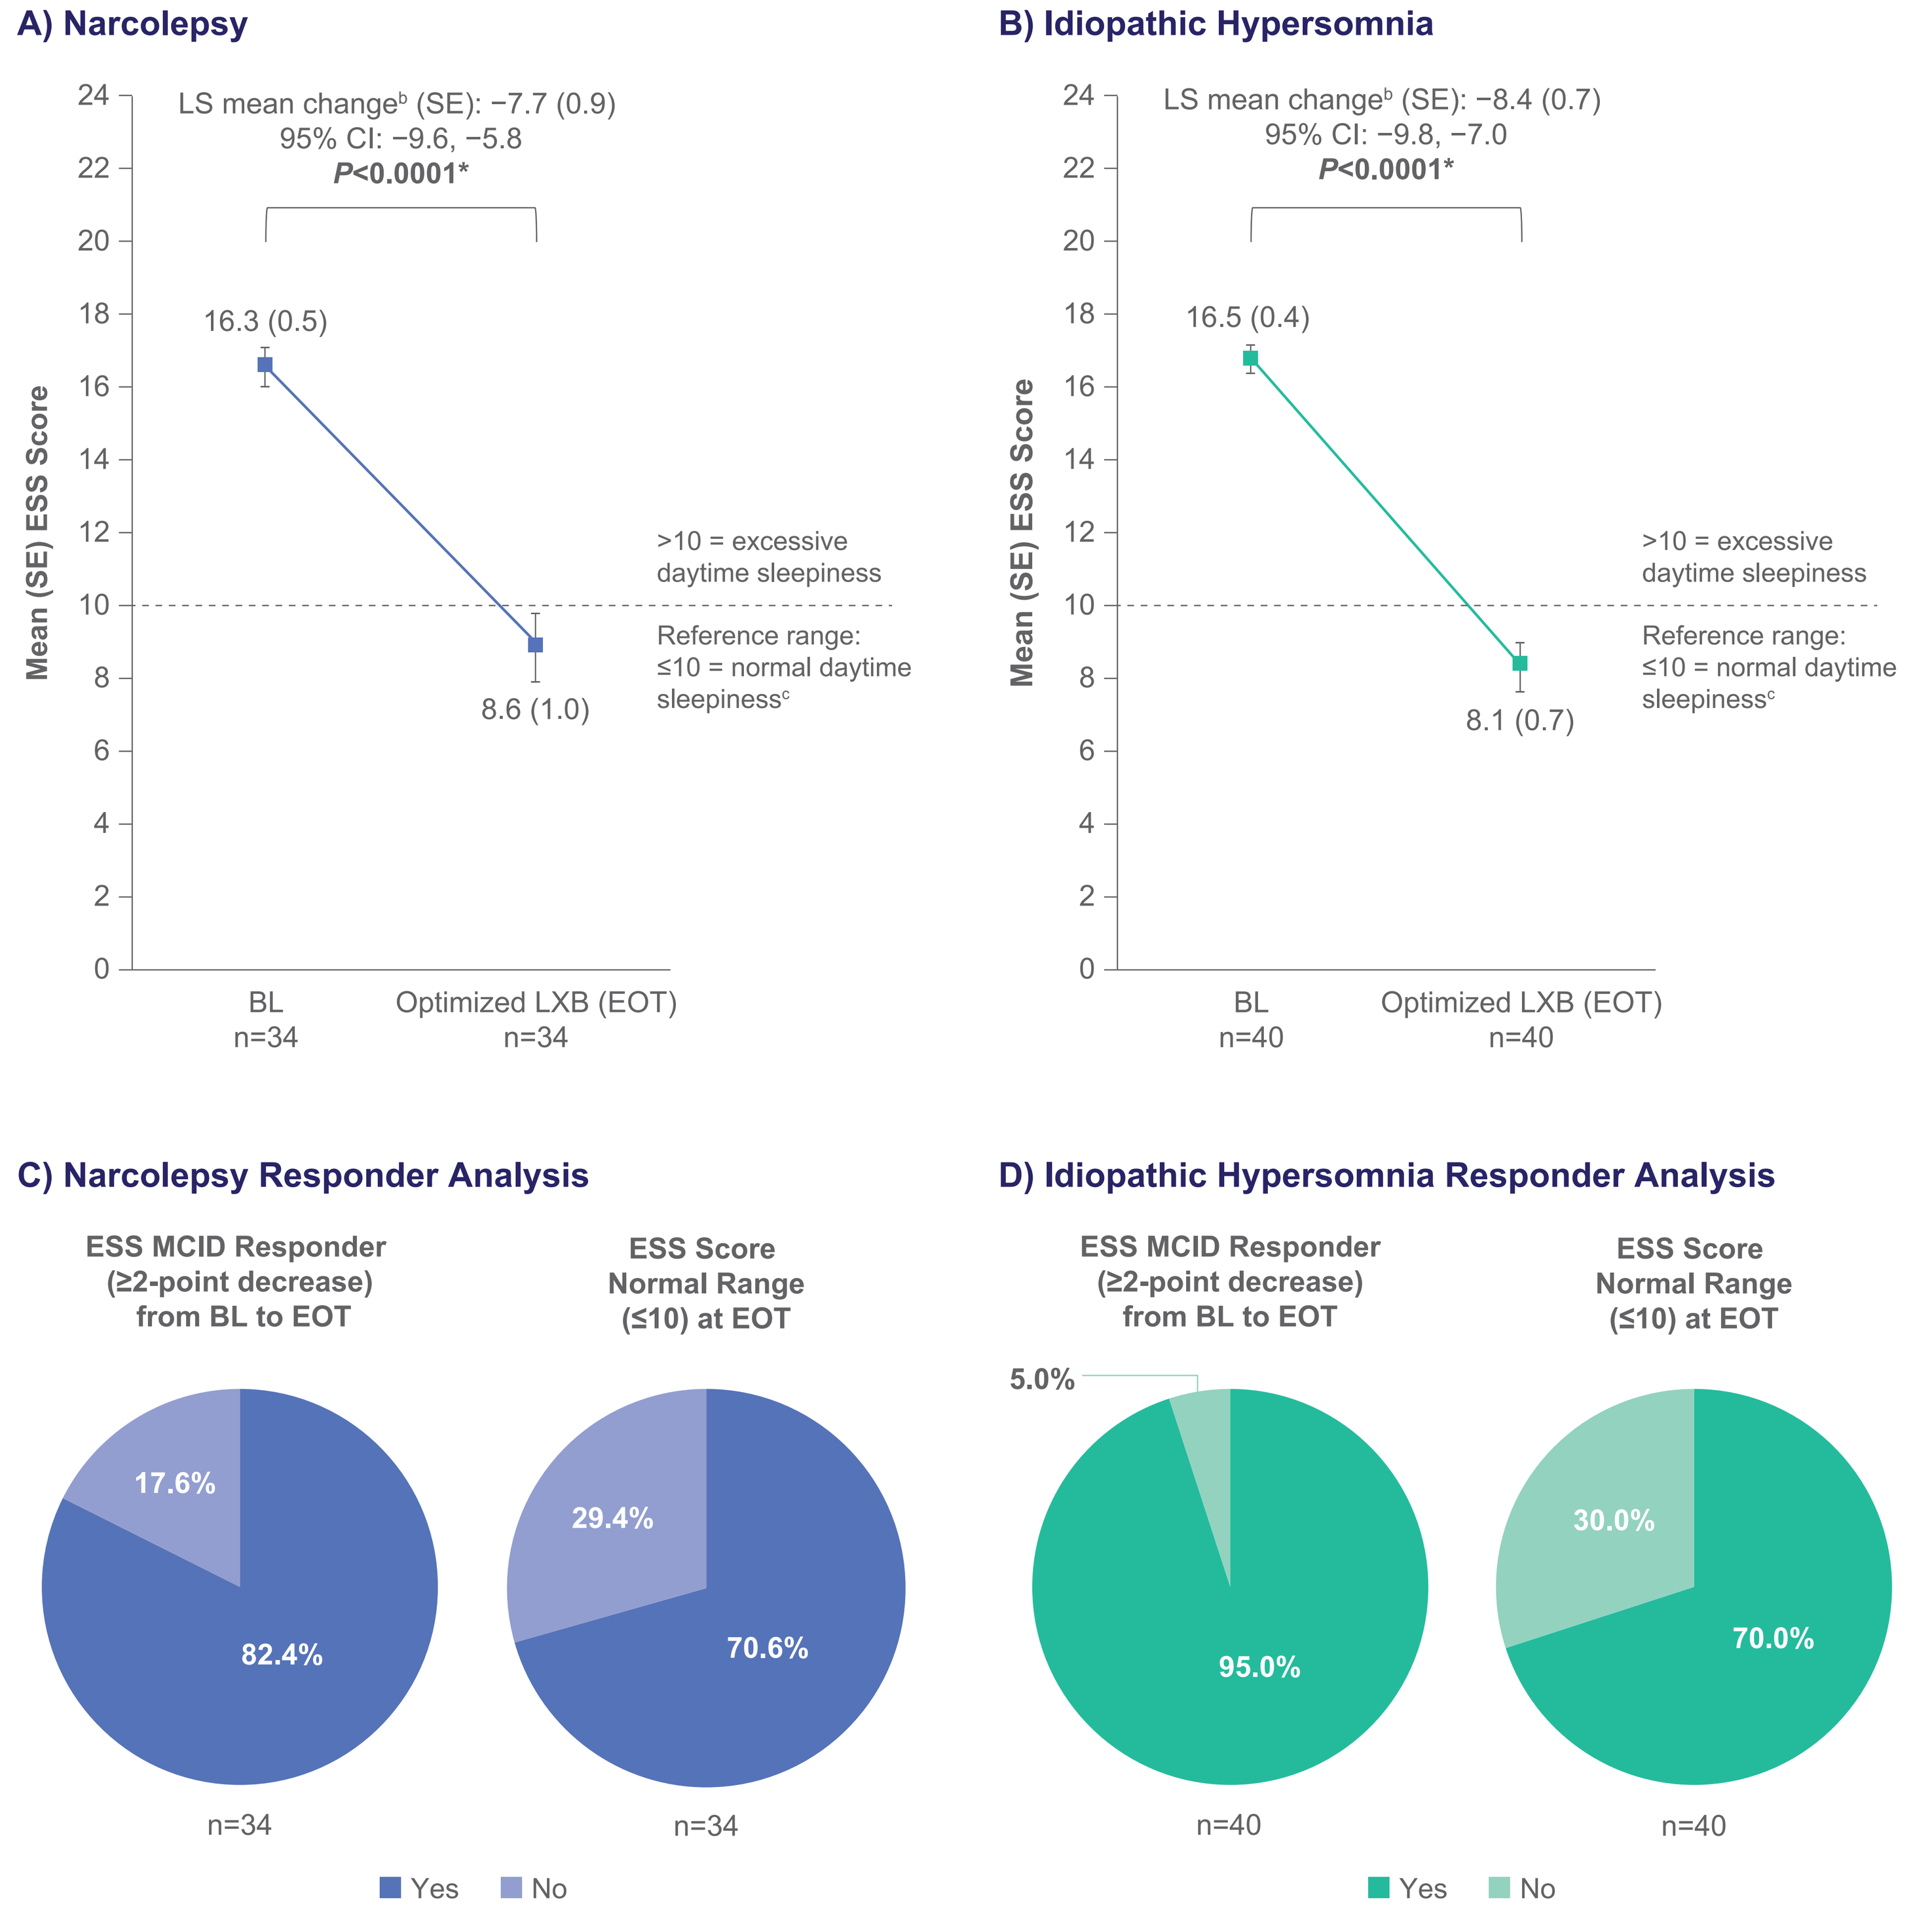
A) Narcolepsy
LS mean change[b] (SE): −7.7 (0.9)
95% CI: −9.6, −5.8
P<0.0001*
16.3 (0.5)
8.6 (1.0)
>10 = excessive daytime sleepiness
Reference range: ≤10 = normal daytime sleepiness[c]
Mean (SE) ESS Score
BL
n=34
Optimized LXB (EOT)
n=34
B) Idiopathic Hypersomnia
LS mean change[b] (SE): −8.4 (0.7)
95% CI: −9.8, −7.0
P<0.0001*
16.5 (0.4)
8.1 (0.7)
>10 = excessive daytime sleepiness
Reference range: ≤10 = normal daytime sleepiness[c]
BL
n=40
Optimized LXB (EOT)
n=40
C) Narcolepsy Responder Analysis
ESS MCID Responder (≥2-point decrease) from BL to EOT
ESS Score Normal Range (≤10) at EOT
17.6%
82.4%
29.4%
70.6%
n=34
n=34
Yes
No
D) Idiopathic Hypersomnia Responder Analysis
ESS MCID Responder (≥2-point decrease) from BL to EOT
ESS Score Normal Range (≤10) at EOT
5.0%
95.0%
30.0%
70.0%
n=40
n=40
Yes
No

Figure A from Schneider LD, et al.[6] http://creativecommons.org/licenses/by-nc/4.0/. Figure B (with color modification) from Plante DT, et al.[7] 2026. http://creativecommons.org/licenses/by-nc/4.0/.
[a]Completer analysis set. [b]Difference between EOT and BL. LS mean and 95% CI are provided at EOT visit using an ANCOVA model adjusted for the BL value. [c]Established categories consider scores up to 10 normal.[2]
*Controlled for multiplicity in prespecified sequential testing. *P*<0.05 is statistically significant.
ANCOVA, analysis of covariance; BL, baseline; CI, confidence interval; EOT, end of treatment; ESS, Epworth Sleepiness Scale; LS, least squares; LXB, low-sodium oxybate; MCID, minimal clinically important difference; SE, standard error.

- The majority of participants with narcolepsy (82.4%; n=28/34) met MCID criteria for ESS; 70.6% (n=24/34) achieved normal range ESS scores with optimized LXB at EOT
- Most participants with idiopathic hypersomnia (95.0%; n=38/40) met MCID criteria for ESS; 70.0% (n=28/40) achieved normal range ESS scores with optimized LXB at EOT
- Demographics and BL characteristics of participants based on ESS response can be found in the Supplement, accessed via the QR code

Figure 3. Mean Idiopathic Hypersomnia Severity Scale Scores Improved (A) and Most Participants With Idiopathic Hypersomnia Achieved Clinically Meaningful Improvement and IHSS Scores Within Normal Range (<22) (B) With Optimized LXB Treatment[a]

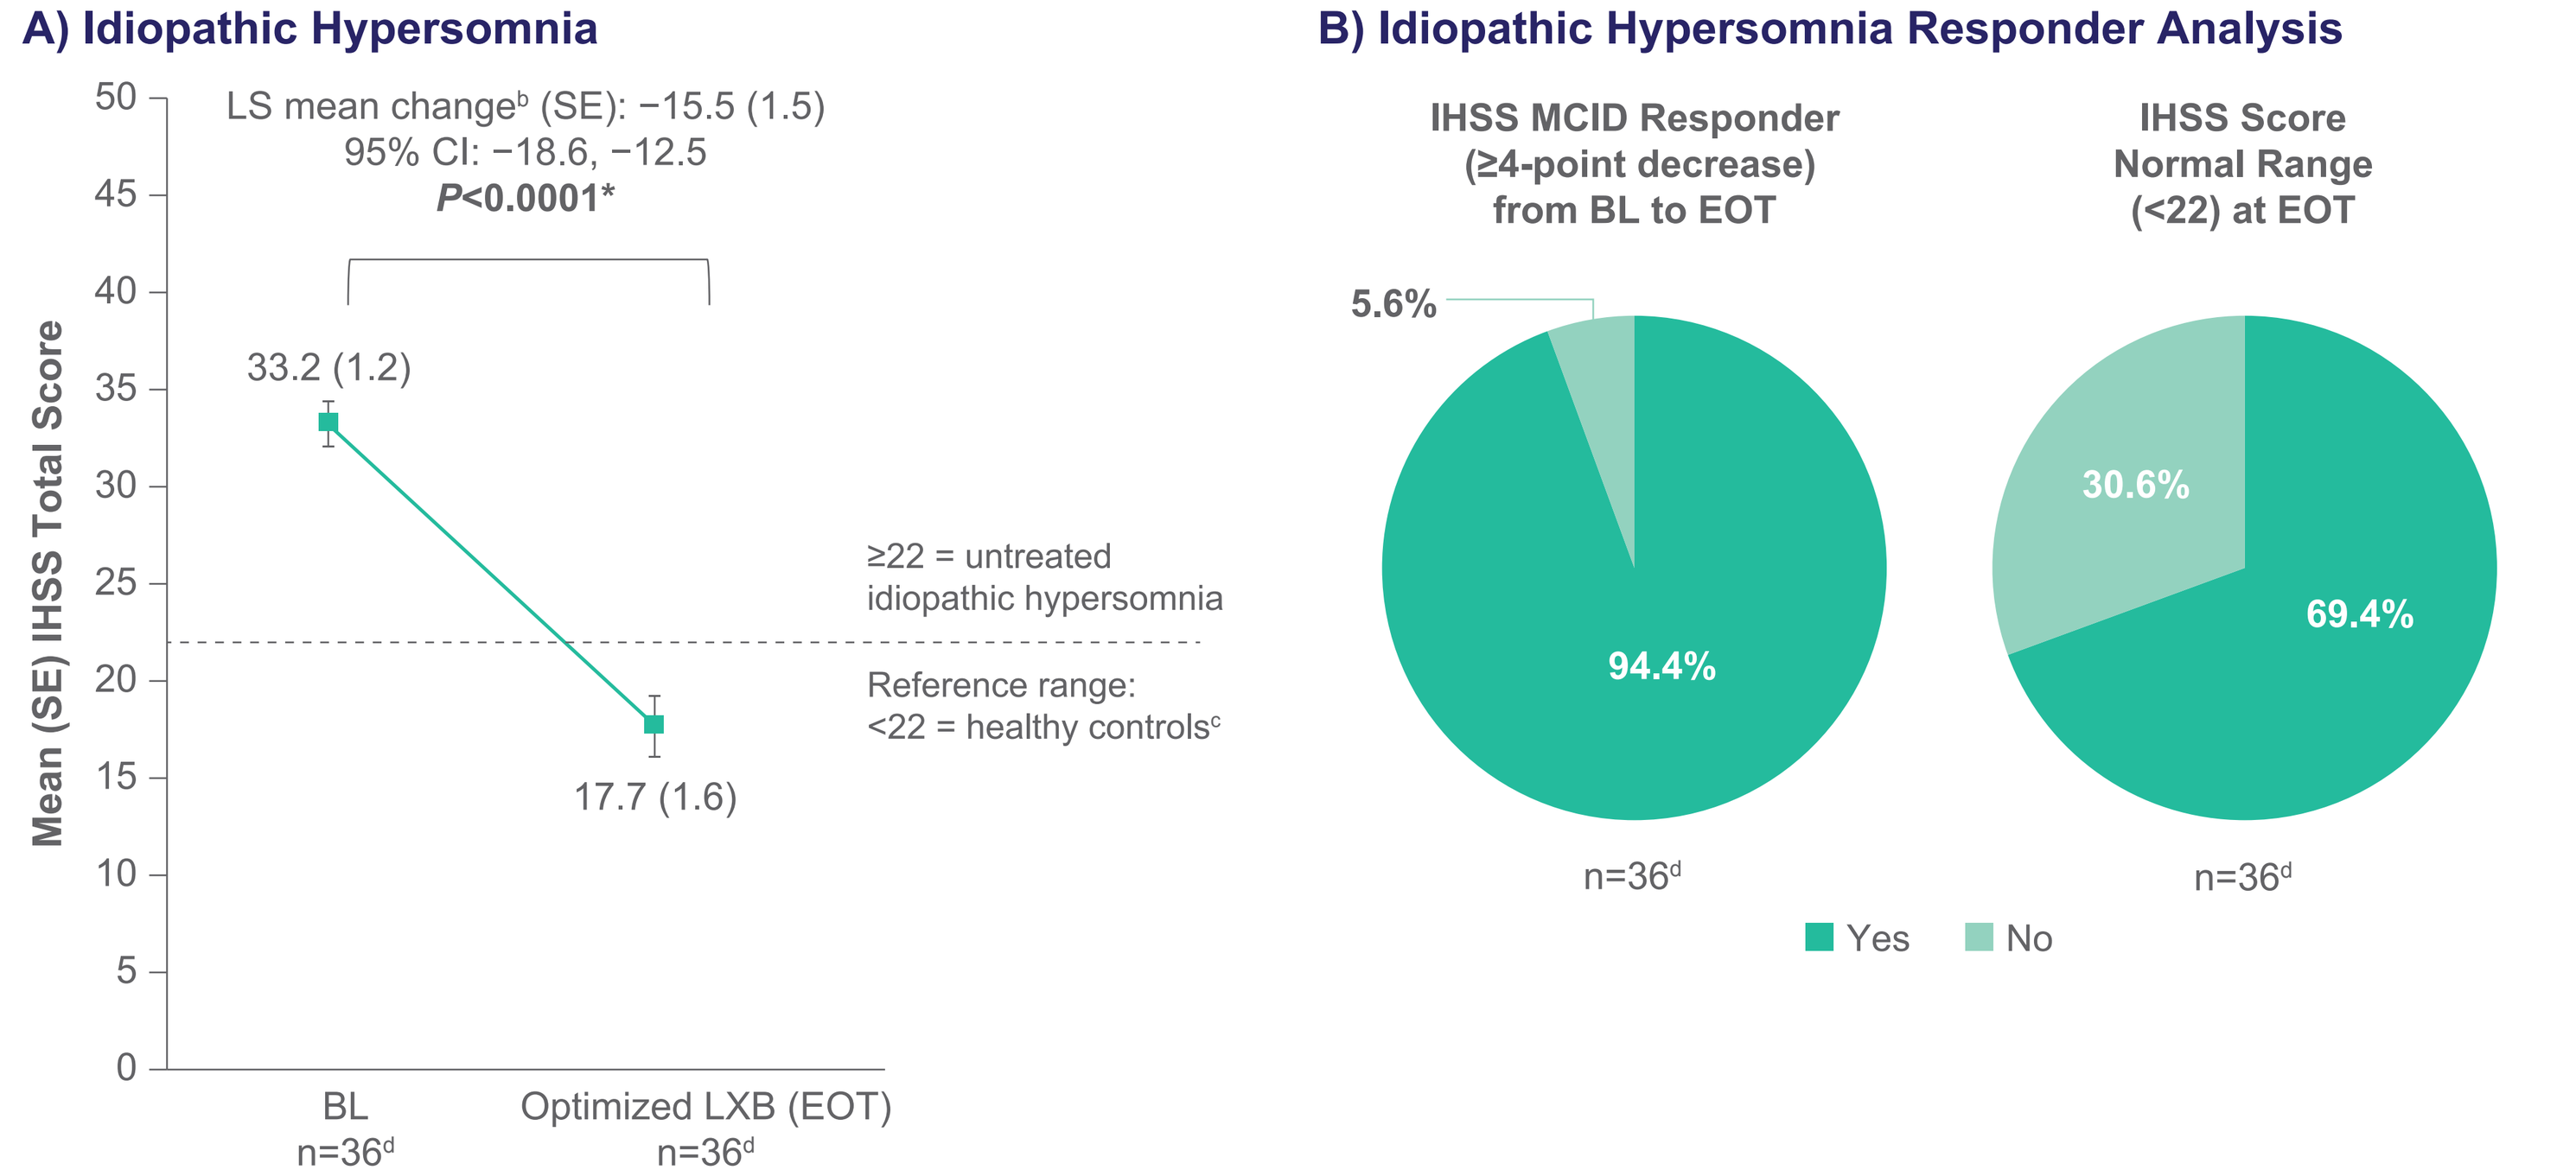
A) Idiopathic Hypersomnia
LS mean change[b] (SE): −15.5 (1.5)
95% CI: −18.6, −12.5
P<0.0001*
33.2 (1.2)
17.7 (1.6)
≥22 = untreated idiopathic hypersomnia
Reference range: <22 = healthy controls[c]
Mean (SE) IHSS Total Score
BL
n=36[d]
Optimized LXB (EOT)
n=36[d]
B) Idiopathic Hypersomnia Responder Analysis
IHSS MCID Responder (≥4-point decrease) from BL to EOT
IHSS Score Normal Range (<22) at EOT
5.6%
94.4%
30.6%
69.4%
n=36[d]
n=36[d]
Yes
No

Figure A (with color modification) from Plante DT, et al.[7] 2026. http://creativecommons.org/licenses/by-nc/4.0/.
[a]Completer analysis. [b]Difference between EOT and BL. [c]An IHSS score of 22 is the optimal cutoff for distinguishing between untreated patients with idiopathic hypersomnia and community-dwelling controls.[3] [d]Not all participants completed all assessments.
*Controlled for multiplicity.
BL, baseline; CI, confidence interval; EOT, end of treatment; IHSS, Idiopathic Hypersomnia Severity Scale; LS, least squares; LXB, low-sodium oxybate; SE, standard error.

- Mean IHSS score decreased from 33.2 at BL to 17.7 (normal range) at EOT[7]
- Most participants with idiopathic hypersomnia (94.4%; n=34/36) achieved MCID for IHSS; 69.4% (n=25/36) achieved a normal IHSS score with optimized LXB at EOT
- Demographics and BL characteristics of participants with idiopathic hypersomnia based on IHSS response can be found in the Supplement, accessed via the QR code

Figure 4. Most Participants With NT1 and NT2 Achieved Clinically Meaningful Improvement and ESS Scores Within Normal Range (≤10) With Optimized LXB Treatment at EOT[a]

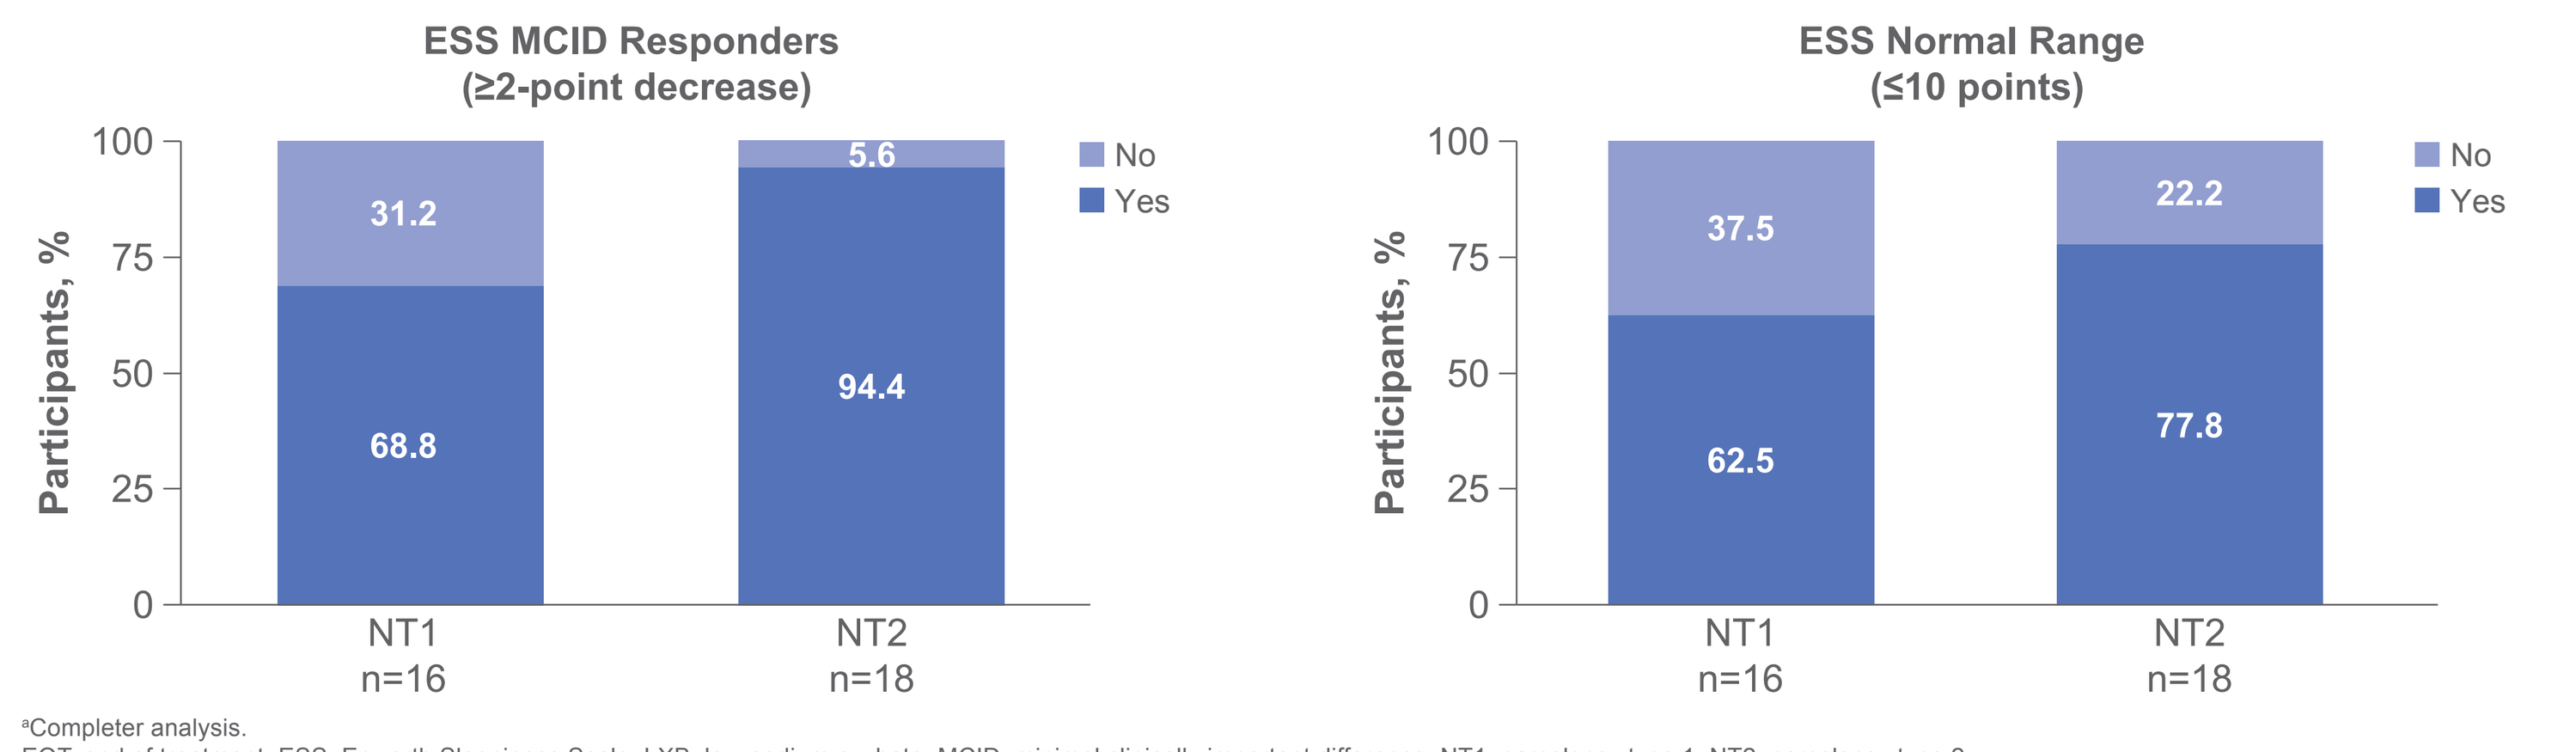
ESS MCID Responders (≥2-point decrease)
Participants, %
31.2
68.8
5.6
94.4
No
Yes
NT1
n=16
NT2
n=18
ESS Normal Range (≤10 points)
37.5
62.5
22.2
77.8
NT1
n=16
NT2
n=18

[a]Completer analysis.
EOT, end of treatment; ESS, Epworth Sleepiness Scale; LXB, low-sodium oxybate; MCID, minimal clinically important difference; NT1, narcolepsy type 1; NT2, narcolepsy type 2.

- At BL, mean (SE) ESS scores were 16.6 (0.8) for participants with narcolepsy type 1 (NT1) and 15.9 (0.8) for participants with narcolepsy type 2 (NT2)[6]
- Among participants with NT1, 68.8% were ESS MCID responders after optimization with LXB treatment; 62.5% achieved normal range ESS scores
- Among participants with NT2, 94.4% were ESS MCID responders; 77.8% achieved normal range ESS scores

Figure 5. Most Participants With Narcolepsy Achieved Clinically Meaningful Improvement and ESS Scores Within Normal Range (≤10) With Optimized LXB Treatment, Regardless of Concomitant Alerting Agent Use[a,b,c]

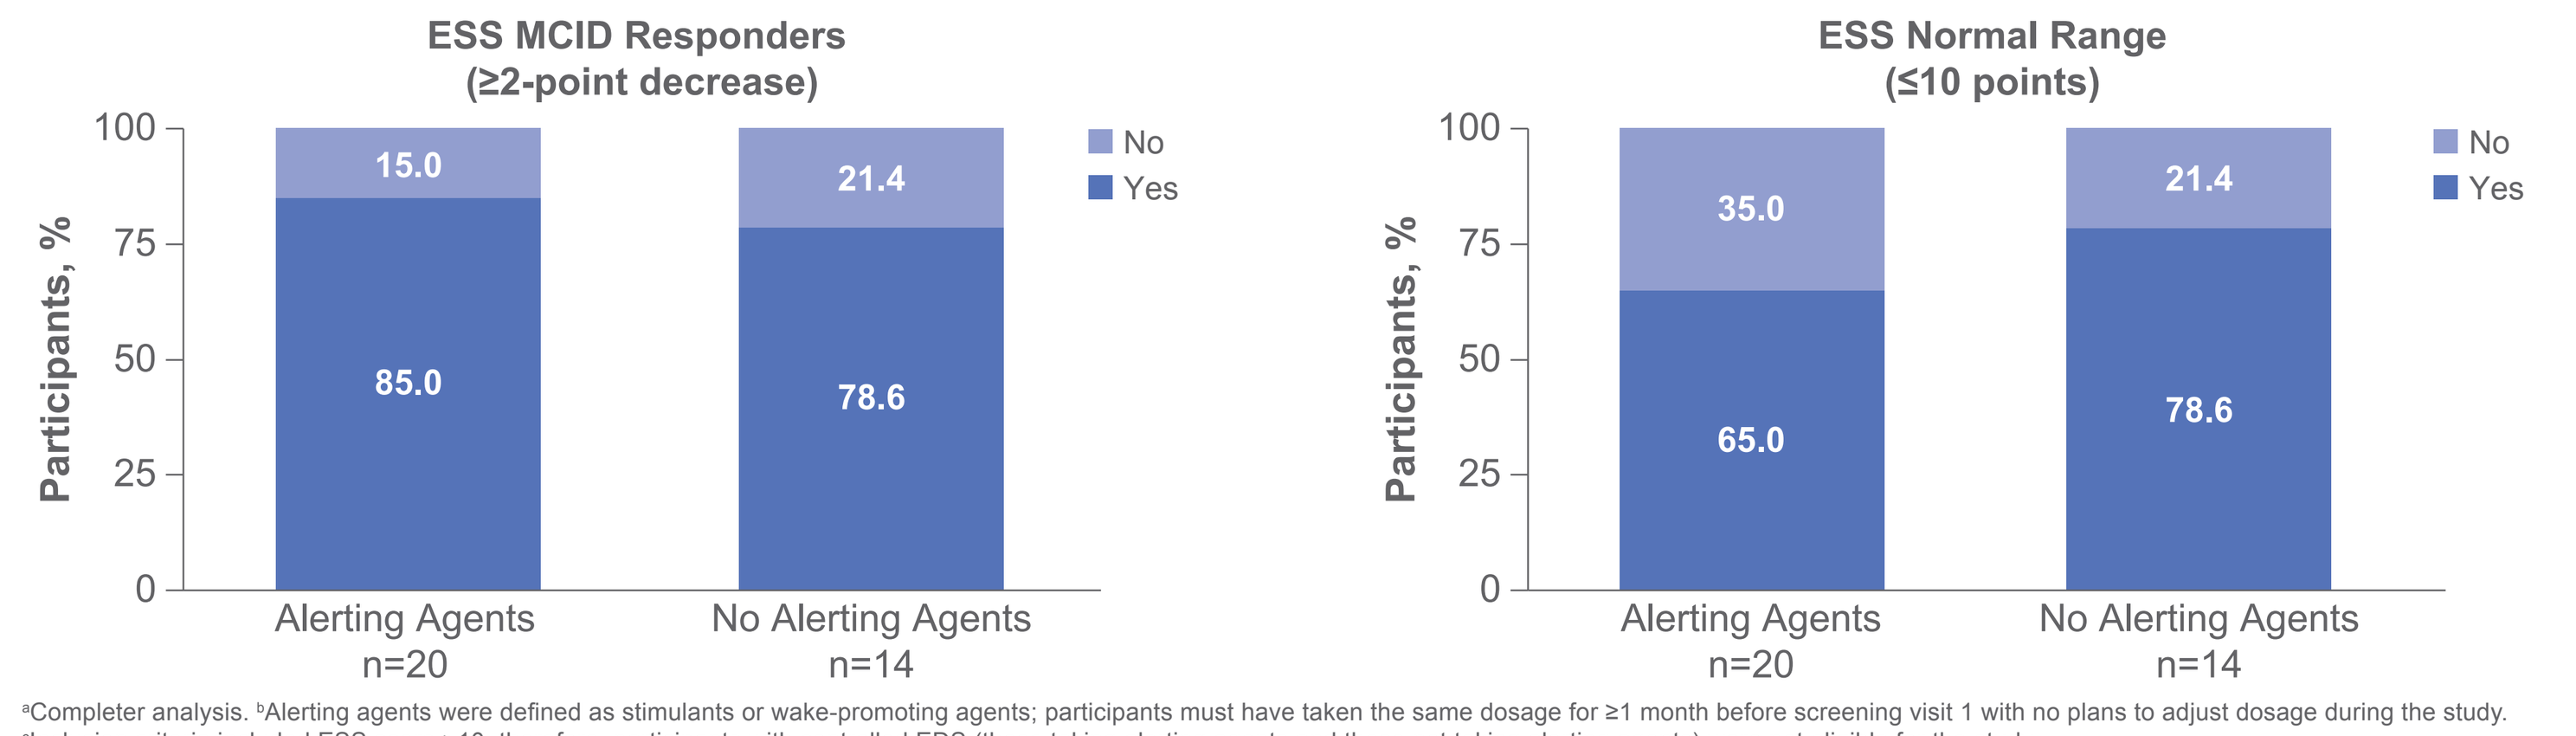
ESS MCID Responders (≥2-point decrease)
Participants, %
15.0
85.0
21.4
78.6
No
Yes
Alerting Agents
n=20
No Alerting Agents
n=14
ESS Normal Range (≤10 points)
35.0
65.0
21.4
78.6
Alerting Agents
n=20
No Alerting Agents
n=14

[a]Completer analysis. [b]Alerting agents were defined as stimulants or wake-promoting agents; participants must have taken the same dosage for ≥1 month before screening visit 1 with no plans to adjust dosage during the study.
[c]Inclusion criteria included ESS score >10; therefore, participants with controlled EDS (those taking alerting agents and those not taking alerting agents) were not eligible for the study.
EDS, excessive daytime sleepiness; EOT, end of treatment; ESS, Epworth Sleepiness Scale; LXB, low-sodium oxybate; MCID, minimal clinically important difference.

Figure 6. Most Participants With Idiopathic Hypersomnia Achieved Clinically Meaningful Improvement and ESS Scores Within Normal Range (≤10) With Optimized LXB Treatment, Regardless of Concomitant Alerting Agent Use[a,b,c]

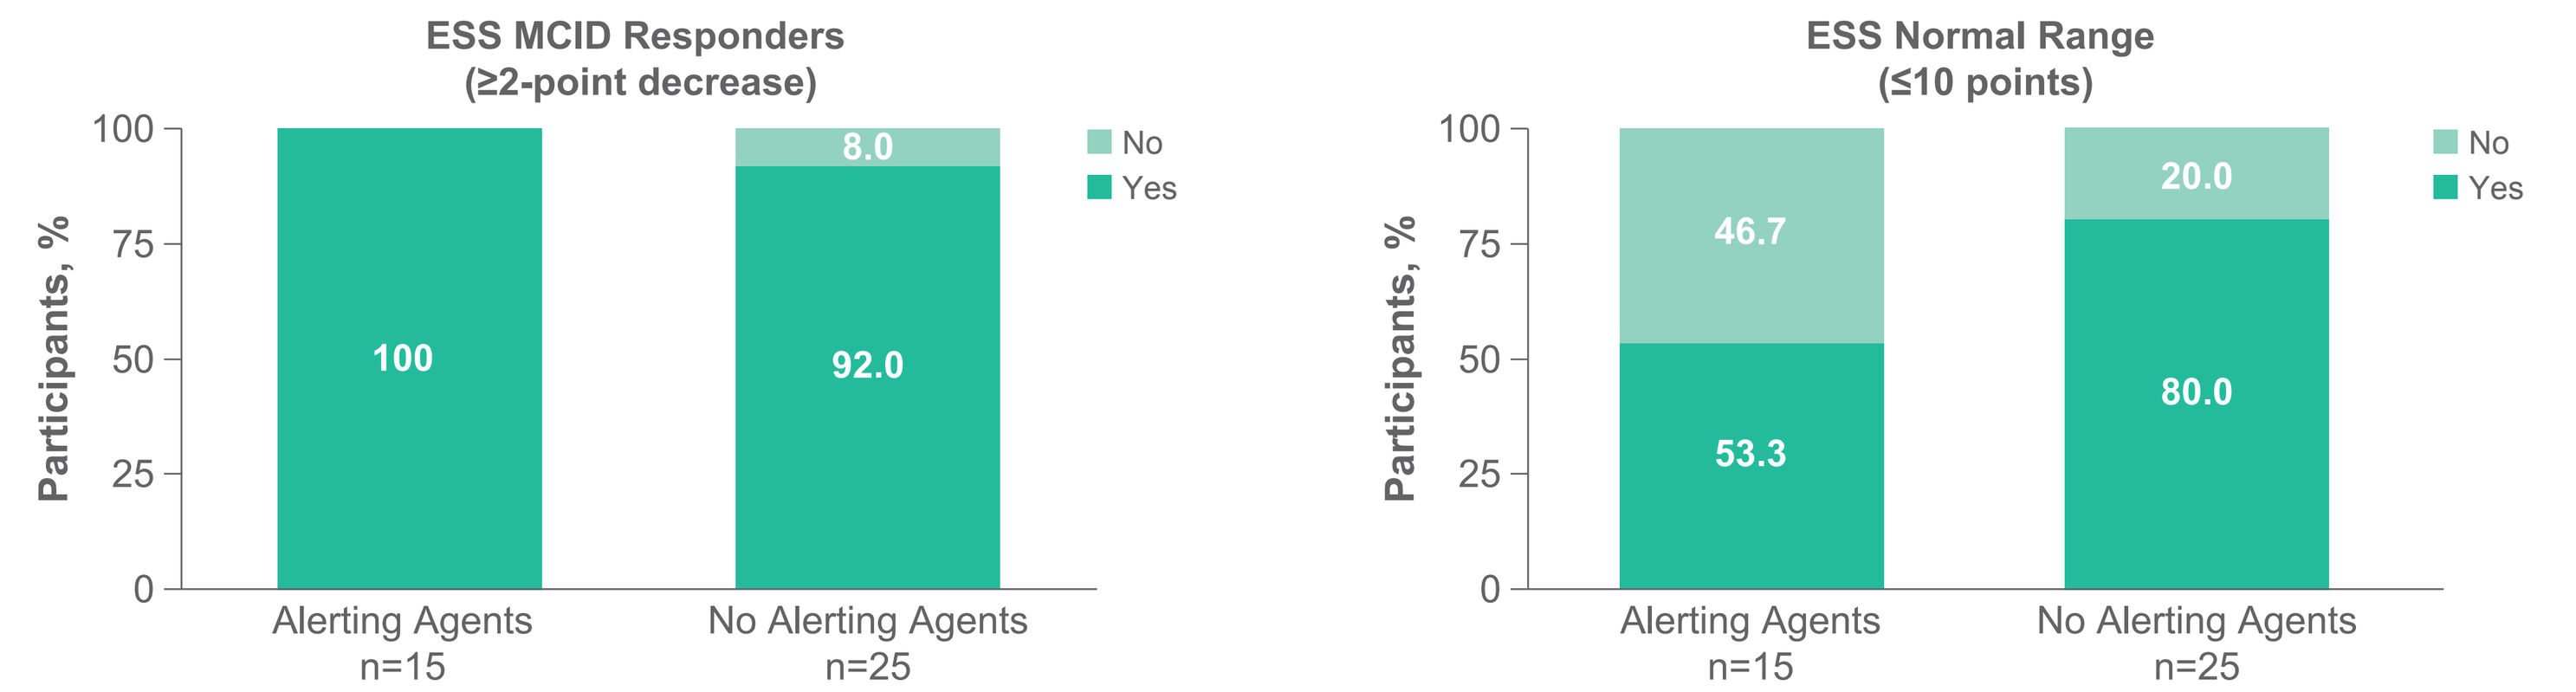
ESS MCID Responders (≥2-point decrease)
Participants, %
100
8.0
92.0
No
Yes
Alerting Agents
n=15
No Alerting Agents
n=25
ESS Normal Range (≤10 points)
46.7
53.3
20.0
80.0
Alerting Agents
n=15
No Alerting Agents
n=25

[a]Completer analysis. [b]Alerting agents were defined as stimulants or wake-promoting agents; participants must have taken the same dosage for ≥1 month before screening visit 1 with no plans to adjust dosage during the study.
[c]Inclusion criteria included ESS score >10; therefore, participants with controlled EDS (those taking alerting agents and those not taking alerting agents) were not eligible for the study.
EDS, excessive daytime sleepiness; EOT, end of treatment; ESS, Epworth Sleepiness Scale; LXB, low-sodium oxybate.

Figure 7. Most Participants With Idiopathic Hypersomnia Achieved Clinically Meaningful Improvement and IHSS Scores Within Normal Range (<22) With Optimized LXB Treatment, Regardless of Concomitant Alerting Agent Use[a,b,c]

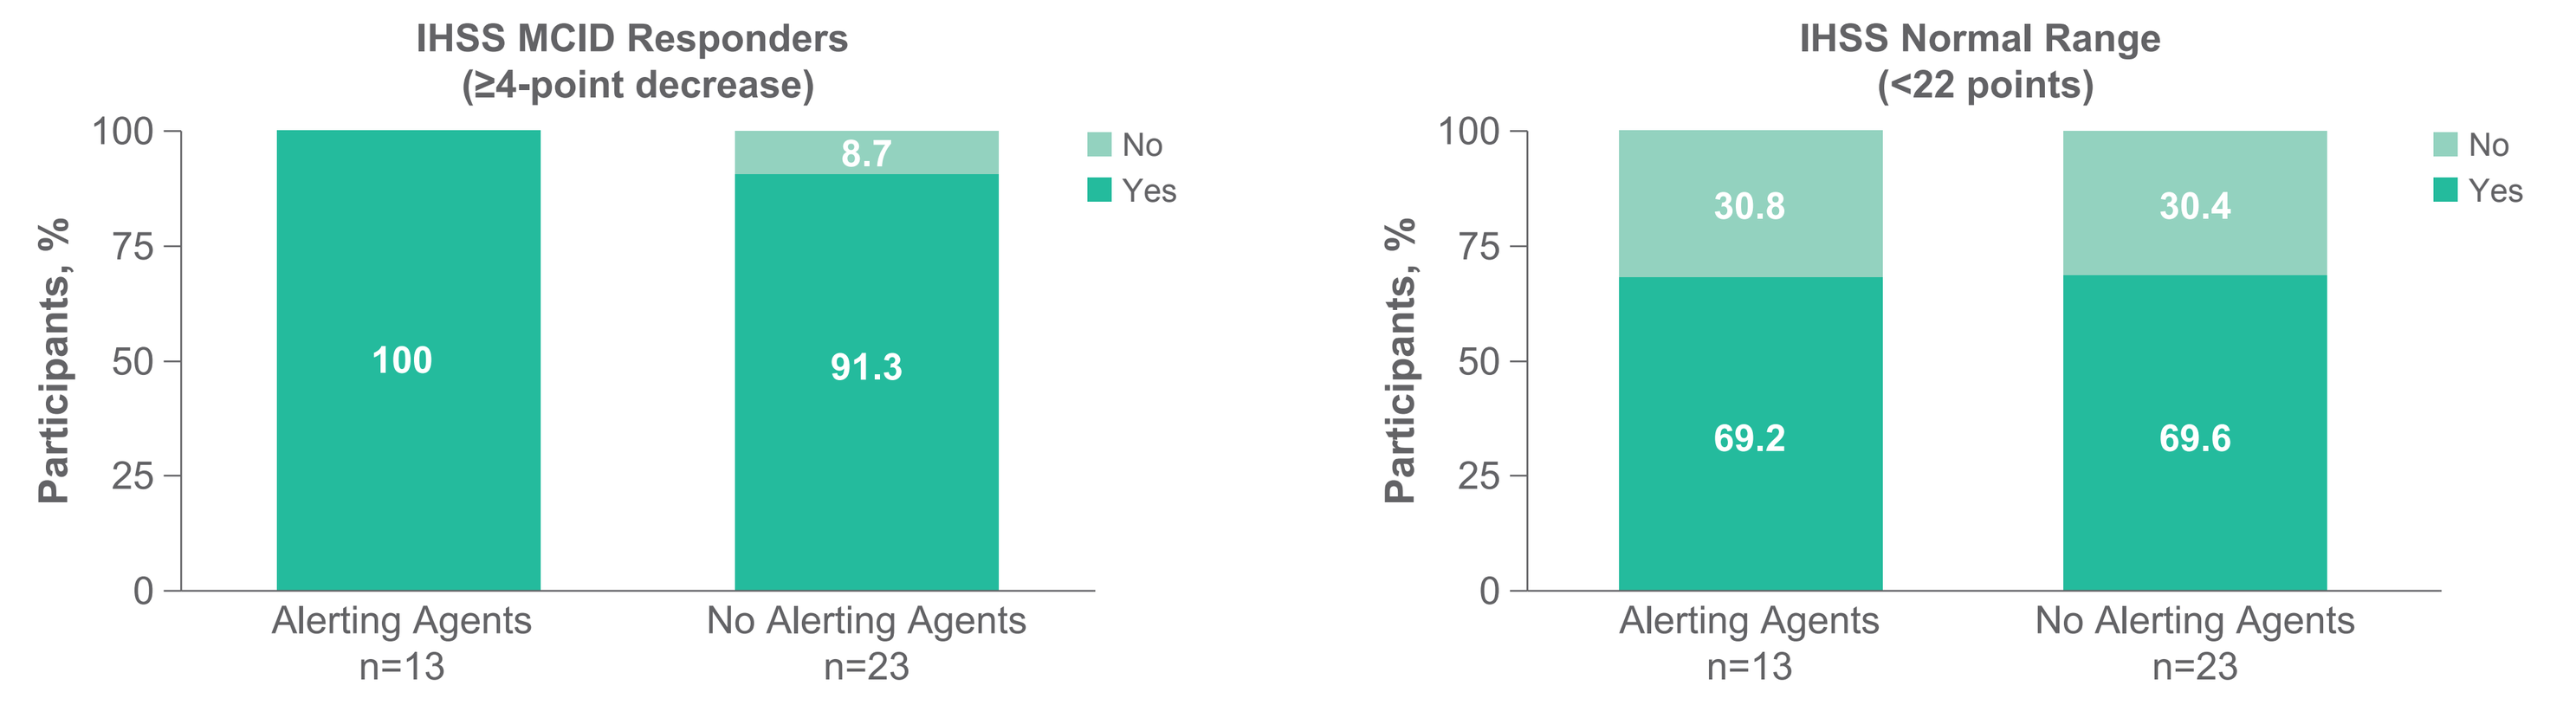
IHSS MCID Responders (≥4-point decrease)
Participants, %
100
8.7
91.3
No
Yes
Alerting Agents
n=13
No Alerting Agents
n=23
IHSS Normal Range (<22 points)
30.8
69.2
30.4
69.6
Alerting Agents
n=13
No Alerting Agents
n=23

[a]Completer analysis. [b]Alerting agents were defined as stimulants or wake-promoting agents; participants must have taken the same dosage for ≥1 month before screening visit 1 with no plans to adjust dosage during the study.
[c]Inclusion criteria included ESS score >10; therefore, participants with controlled EDS (those taking alerting agents and those not taking alerting agents) were not eligible for the study.
EDS, excessive daytime sleepiness; EOT, end of treatment; IHSS, Idiopathic Hypersomnia Severity Scale; LXB, low-sodium oxybate.

153

Clinical Responders to Low-Sodium Oxybate in Narcolepsy and Idiopathic Hypersomnia: Evaluation of Epworth Sleepiness Scale and Idiopathic Hypersomnia Severity Scale Established Thresholds and Normal Scores

Chad M. Ruoff, MD[1]; R. Bart Sangal, MD[2,3]; Deborah A. Nichols, MS[4]; Teresa L. Steininger, PhD[4]; Jing Dai, PhD[4]; Shawn Vahabzadeh, PharmD[5]; Marisa Whalen, PharmD[5]; Sarah C. Markt, ScD, MPH[4]; Laura B. Herpel, MD[6,7]; Nancy Foldvary-Schaefer, DO, MS[8]

[1]Mayo Clinic, Scottsdale, AZ, USA; [2]Obesity and Sleep Medicine, Bloomfield Hills, MI, USA; [3]Oakland University William Beaumont School of Medicine, Rochester Hills, MI, USA; [4]Jazz Pharmaceuticals, Palo Alto, CA, USA; [5]Jazz Pharmaceuticals, Philadelphia, PA, USA; [6]Bogan Sleep Consultants, Columbia, SC, USA; [7]Medical University of South Carolina, Charleston, SC, USA; [8]Sleep Disorders Center, Neurological Institute, Cleveland Clinic, Cleveland, OH, USA

Scan this code to access a plain language summary of the published abstract. This code is not for promotional purposes.

Scan this code to access this poster and supplemental material online. This code is not for promotional purposes.

Presented at Psych Congress 2026; September 15–19, 2026; New Orleans, LA, USA

Supplemental Material

Analyses Sets

- The safety analysis set includes all participants who enrolled in the study and took their prescribed LXB regimen for ≥1 night after the BL period (evaluated for demographics/BL characteristics, dosing, treatment-emergent adverse events); the completer analysis set includes all participants who enrolled in the study, took the prescribed LXB regimen for ≥1 night after the BL period, completed the stable-dose period, and completed the polysomnography EOT visit (evaluated for ESS, IHSS)
- Data analyses were completed January 2026

Supplemental Figure S1. Inclusion/Exclusion Criteria

Inclusion Criteria

- Adults 18–75 with a primary diagnosis of NT1 or NT2 (ICSD-3 or DSM-5) or idiopathic hypersomnia (ICSD-3)[a]
- ESS score >10[b]
- Stable use of concomitant anticataplectics or alerting agents allowed[c]

Exclusion Criteria

- Untreated/inadequately treated sleep-disordered breathing (AHI >10)[d]
- History/presence of other untreated, inadequately treated, or unstable disorder or condition (eg, sleep, medical, behavioral/psychiatric, neurologic[e])
- Medication(s) contraindicated, with known drug-drug interaction, or similar EEG effects as LXB or known to have clinically significant CNS sedating effects

[a]Diagnosis of idiopathic hypersomnia and analyses performed for the idiopathic hypersomnia cohort make no distinction between those with and without long sleep time. [b]At screening visit 1 or, if taking an oxybate medication at screening, at visit 2. [c]Alerting agents were defined as stimulants or wake-promoting agents; participants must have taken the same dosage for ≥1 month before screening visit 1 with no plan to adjust dosage during the study. [d]Hypopnea definition included a ≥4% desaturation per Rule 1B of *The AASM Manual for the Scoring of Sleep and Associated Events*, as assessed during the baseline PSG visit. [e]As determined by the investigator.
AASM, American Academy of Sleep Medicine; AHI, apnea-hypopnea index; CNS, central nervous system; DSM-5, *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition*; EEG, electroencephalogram; ESS, Epworth Sleepiness Scale; ICSD-3, *International Classification of Sleep Disorders – Third Edition*; LXB, low-sodium oxybate; NT1, narcolepsy type 1; NT2, narcolepsy type 2; PSG, polysomnography.

Supplemental Table S1. Demographics/Baseline Characteristics for Narcolepsy Cohort Based on ESS Response[a]

	ESS Score				
Characteristic	MCID-Responders: ≥2-point Decrease n=28	Non–MCID-Responders: <2-point Decrease n=6	Normal Range: ≤10 at EOT n=24	Not Within Normal Range: >10 at EOT n=10	Total Narcolepsy Cohort n=34
Age (years)					
Mean (SD)	34.3 (14.5)	42.7 (11.7)	35.3 (15.4)	36.8 (11.6)	35.7 (14.2)
Female at birth, n (%)	20 (71.4)	5 (83.3)	18 (75.0)	7 (70.0)	25 (73.5)
Race, n (%)					
White	23 (82.1)	4 (66.7)	19 (79.2)	8 (80.0)	27 (79.4)
Black or African American	4 (14.3)	0	3 (12.5)	1 (10.0)	4 (11.8)
Asian	1 (3.6)	1 (16.7)	1 (4.2)	1 (10.0)	2 (5.9)
Unknown	0	1 (16.7)	1 (4.2)	0	1 (2.9)
Ethnicity, n (%)					
Hispanic or Latino	2 (7.1)	1 (16.7)	2 (8.3)	1 (10.0)	3 (8.8)
Not Hispanic or Latino	26 (92.9)	5 (83.3)	22 (91.7)	9 (90.0)	31 (91.2)
Body mass index (kg/m^2), mean (SD)	28.6 (5.7)	31.4 (8.3)	27.0 (5.1)	34.1 (5.7)	29.1 (6.2)
Oxybate type at study entry, n (%)					
Naive[b]	21 (75.0)	5 (83.3)	20 (83.3)	6 (60.0)	26 (76.5)
Low-sodium oxybate	4 (14.3)	0	2 (8.3)	2 (20.0)	4 (11.8)
Sodium oxybate	2 (7.1)	1 (16.7)	1 (4.2)	2 (20.0)	3 (8.8)
Once-nightly sodium oxybate	1 (3.6)	0	1 (4.2)	0	1 (2.9)
Comorbid OSA, n (%)	4 (14.3)	1 (16.7)	3 (12.5)	2 (20.0)	5 (14.7)
Without comorbid OSA, n (%)	24 (85.7)	5 (83.3)	21 (87.5)	8 (80.0)	29 (85.3)

[a]Completer analysis set. [b]No oxybate use within 2 weeks before entering the study.
BL, baseline; EOT, end of treatment; ESS, Epworth Sleepiness Scale; MCID, minimal clinically important difference; NT1, narcolepsy type 1; NT2, narcolepsy type 2; OSA, obstructive sleep apnea; SD, standard deviation.

Supplemental Table S2. Demographics/Baseline Characteristics for Idiopathic Hypersomnia Cohort Based on ESS Response[a]

	ESS Score				
Characteristics	MCID-Responders: ≥2-point Decrease n=38	Non–MCID-Responders: <2-point Decrease n=2	Normal Range: ≤10 at EOT n=28	Not Within Normal Range: >10 at EOT n=12	Total Idiopathic Hypersomnia Cohort n=40
Age (years)					
Mean (SD)	38.7 (12.5)	38.5 (10.6)	37.5 (12.0)	41.3 (13.0)	38.7 (12.3)
Female at birth, n (%)	30 (78.9)	1 (50.0)	22 (78.6)	9 (75.0)	31 (77.5)
Race, n (%)					
White	32 (84.2)	2 (100)	24 (85.7)	10 (83.3)	34 (85.0)
Black or African American	2 (5.3)	0	1 (3.6)	1 (8.3)	2 (5.0)
Asian	2 (5.3)	0	2 (7.1)	0	2 (5.0)
Native Hawaiian or other Pacific Islander	1 (2.6)	0	0	1 (8.3)	1 (2.5)
Multiple[b]	1 (2.6)	0	1 (3.6)	0	1 (2.5)
Ethnicity, n (%)					
Hispanic or Latino	8 (21.1)	1 (50.0)	7 (25.0)	2 (16.7)	9 (22.5)
Not Hispanic or Latino	29 (76.3)	1 (50.0)	20 (71.4)	10 (83.3)	30 (75.0)
Body mass index (kg/m^2), mean (SD)	29.1 (6.5)	31.3 (7.9)	28.9 (6.6)	29.8 (6.4)	29.2 (6.5)
Oxybate type at study entry, n (%)					
Naive[c]	30 (78.9)	2 (100)	24 (85.7)	8 (66.7)	32 (80.0)
Low-sodium oxybate	8 (21.1)	0	4 (14.3)	4 (33.3)	8 (20.0)
Comorbid OSA, n (%)	10 (26.3)	0	6 (21.4)	4 (33.3)	10 (25.0)
Without comorbid OSA, n (%)	28 (73.7)	2 (100)	22 (78.6)	8 (66.7)	30 (75.0)
Long sleep need[d]					
>9 hours	19 (50.0)	1 (50.0)	16 (57.1)	4 (33.3)	20 (50.0)
≤9 hours	18 (47.4)	1 (50.0)	12 (42.9)	7 (58.3)	19 (47.5)
LXB regimen, n (%)					
Once-nightly LXB	15 (39.5)	0	12 (42.9)	3 (25.0)	15 (37.5)
Twice-nightly LXB	23 (60.5)	2 (100)	16 (57.1)	9 (75.0)	25 (62.5)

[a]Completer analysis set. [b]Participant reported >1 race. [c]No oxybate use within 2 weeks before entering the study. [d]Based on participants' response to Item 1 on the IHSS at BL. Not all participants completed the IHSS assessment at BL.
BL, baseline; EOT, end of treatment; ESS, Epworth Sleepiness Scale; IHSS, Idiopathic Hypersomnia Severity Scale; LXB, low-sodium oxybate; MCID, minimal clinically important difference; OSA, obstructive sleep apnea; SD, standard deviation.

Supplemental Table S3. Demographics/Baseline Characteristics for Idiopathic Hypersomnia Cohort Based on IHSS Response[a]

	IHSS Score				
Characteristics	MCID-Responders: ≥4-point Decrease n=34	Non–MCID-Responders: <4-point Decrease n=2	Normal Range: <22 at EOT n=25	Not Within Normal Range: ≥22 at EOT n=11	Total Idiopathic Hypersomnia Cohort n=36
Age (years)					
Mean (SD)	38.0 (12.3)	34.5 (16.3)	39.6 (13.1)	33.9 (9.6)	37.8 (12.3)
Female at birth, n (%)	28 (82.4)	1 (50.0)	21 (84.0)	8 (72.7)	29 (80.6)
Race, n (%)					
White	29 (85.3)	1 (50.0)	22 (88.0)	8 (72.7)	30 (83.3)
Black or African American	1 (2.9)	1 (50.0)	1 (4.0)	1 (9.1)	2 (5.6)
Asian	2 (5.9)	0	1 (4.0)	1 (9.1)	2 (5.6)
Native Hawaiian or other Pacific Islander	1 (2.9)	0	0	1 (9.1)	1 (2.8)
Multiple[b]	1 (2.9)	0	1 (4.0)	0	1 (2.8)
Ethnicity, n (%)					
Hispanic or Latino	9 (26.5)	0	7 (28.0)	2 (18.2)	9 (25.0)
Not Hispanic or Latino	24 (70.6)	2 (100)	17 (68.0)	9 (81.8)	26 (72.2)
Body mass index (kg/m^2), mean (SD)	29.5 (6.6)	31.3 (8.0)	29.4 (5.4)	29.9 (9.0)	29.6 (6.6)
Oxybate type at study entry, n (%)					
Naive[c]	28 (82.4)	1 (50.0)	21 (84.0)	8 (72.7)	29 (80.6)
Low-sodium oxybate	6 (17.6)	1 (50.0)	4 (16.0)	3 (27.3)	7 (19.4)
Long sleep need[d]					
>9 hours	19 (55.9)	0	13 (52.0)	6 (54.5)	19 (52.8)
≤9 hours	15 (44.1)	2 (100)	12 (48.0)	5 (45.5)	17 (47.2)
LXB regimen, n (%)					
Once-nightly LXB	13 (38.2)	0	9 (36.0)	4 (36.4)	13 (36.1)
Twice-nightly LXB	21 (61.8)	2 (100)	16 (64.0)	7 (63.6)	23 (63.9)

[a]Completer analysis set. [b]Participant reported >1 race. [c]No oxybate use within 2 weeks before entering the study. [d]Based on participants' response to Item 1 on the IHSS at BL. Not all participants completed the IHSS assessment at BL.
BL, baseline; EOT, end of treatment; ESS, Epworth Sleepiness Scale; IHSS, Idiopathic Hypersomnia Severity Scale; LXB, low-sodium oxybate; MCID, minimal clinically important difference; OSA, obstructive sleep apnea; SD, standard deviation.

Supplemental Table S4. Treatment-Emergent Adverse Events[a] (Occurring in ≥10% of Participants)

Preferred term, n (%)	Idiopathic Hypersomnia N=46	Narcolepsy N=55
Nausea	9 (19.6)	13 (23.6)
Dizziness	8 (17.4)	8 (14.5)
Headache	8 (17.4)	7 (12.7)
Vomiting	5 (10.9)	6 (10.9)
Somnolence	—[b]	6 (10.9)

[a]Safety analysis set. [b]Reported by <10% of participants.