149

The Patient Experience of Sleep Inertia in Idiopathic Hypersomnia: A Qualitative Study

Mia Wraight, BS[1]; Cristina Casstevens, PhD[1]; Sarah C. Markt, ScD, MPH[1]; Meagan Farrell, PhD[2]; Samantha Linton, PhD[2]; Mariel Alcantara, MD[2]; Christina A. Graham, BS, BA[2]; David T. Plante, MD, PhD[3]

[1]Jazz Pharmaceuticals, Palo Alto, CA, USA; [2]IQVIA, New York, NY, USA; [3]Department of Psychiatry, University of Wisconsin-Madison, Madison, WI, USA

Objective

- This qualitative study was conducted to better understand the lived experience of sleep inertia among individuals with idiopathic hypersomnia and to develop a conceptual model of sleep inertia that comprehensively organizes the most important symptom components and impacts

Conclusions

- This patient-centric study highlights sleep inertia as a complex, multidimensional aspect of idiopathic hypersomnia that spans physical, cognitive, emotional, and behavioral domains
 - Participants commonly identified "difficulty emerging from sleep" and "difficulty moving body after waking" as among the most bothersome components
- Participants reported that sleep inertia profoundly affected multiple aspects of life, including work or school, finances, emotional health, and social relationships, with difficulties in social activities and relationships identified as particularly bothersome impacts
- A deeper understanding of the lived experience of sleep inertia in idiopathic hypersomnia may improve recognition of its clinical burden and inform more patient-centered assessment and management strategies

References: 1. American Academy of Sleep Medicine. *International Classification of Sleep Disorders: ICSD-3-Text revision.* Darien, IL: American Academy of Sleep Medicine; 2023. **2.** Evangelista E, et al. *Sleep.* 2022;45(1). **3.** Arnulf I, et al. *Sleep Med Rev.* 2023;69:101766. **4.** Trotti LM, et al. *Sleep Med.* 2020;75:343-349. **5.** Casstevens C, et al. Presented at: Psych Congress; October 29–November 2, 2024; Boston, MA, USA.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees, Aeja Jackson, PhD, MS, and Eleanor Bush, MA, provided medical writing support and Katie Herman provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: M Wraight, C Casstevens, and **SC Markt** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **M Farrell, S Linton, M Alcantara,** and **CA Graham** are full-time employees of IQVIA, which was contracted by Jazz Pharmaceuticals to conduct this study. **DT Plante** is a consultant and advisory board member for Jazz Pharmaceuticals; served as a consultant/advisory board member for Alkermes, Centessa, Harmony Biosciences, and Takeda; is on an advisory board for Apnimed, Eisai, and Lundbeck; and is a consultant for Aditum Bio and Teva Pharmaceuticals (Australia).

Scan this code to access a plain language summary of the published abstract. This code is not for promotional purposes.

Scan this code to access this poster and supplemental material online. This code is not for promotional purposes.

Presented at Psych Congress 2026; September 15–19, 2026; New Orleans, LA, USA

Introduction

- Idiopathic hypersomnia is a multisymptom, neurologic, sleep disorder characterized by excessive daytime sleepiness (EDS); prolonged, nonrestorative sleep; and severe sleep inertia[1]
- Sleep inertia, the transitional state between sleep and wakefulness, is a highly complex symptom that can affect daily functioning and quality of life[2,3]
- Despite its high prevalence in idiopathic hypersomnia, sleep inertia remains inconsistently defined in the literature, and there is limited evidence about sleep inertia from the perspective of people living with idiopathic hypersomnia[4]

Methods

- Prior concept elicitation (CE) interviews exploring the adult patient experience of idiopathic hypersomnia (n=20)[5] were reanalyzed to extract insights into the patient experience of sleep inertia and to develop a preliminary conceptual model (PCM)
- The PCM informed development of a semi-structured interview guide used in additional CE interviews with adults diagnosed with idiopathic hypersomnia (n=15) to further explore the experience and impact of sleep inertia from the patient perspective
 - Open-ended and probing questions were used to elicit key sleep inertia components and impacts, and factors that influence the severity of sleep inertia in idiopathic hypersomnia
 - A final conceptual model of sleep inertia in idiopathic hypersomnia was developed, illustrating the key components and associated impacts on daily life
- Key requirements for participant inclusion: age 18–64 years at time of screening, documented diagnosis of idiopathic hypersomnia, current experience with EDS and sleep inertia, US resident, fluent in English, and consent to the interview process
- Participants were excluded if they had a diagnosis of another medical condition or medication associated with EDS, untreated or inadequately treated sleep-disordered breathing or periodic limb movement disorder resulting in daytime sleepiness (sleep apnea permitted if adequately treated), average total sleep time of <7 hours/night over the preceding 6 months, or an occupation that required nighttime or variable shift work
- Bothersomeness ratings (0–10) of sleep inertia components and impacts were collected during interviews
 - Frequency of report and bothersomeness ratings were used to evaluate salience (reported by ≥33.3% of participants; mean bothersomeness rating ≥5)
 - Participants' 3 most bothersome sleep inertia components and impacts and 3 most important components to improve were also collected
- Interview data were coded and analyzed using MAXQDA software

Results

Figure 1. Participant-Reported Demographic Information

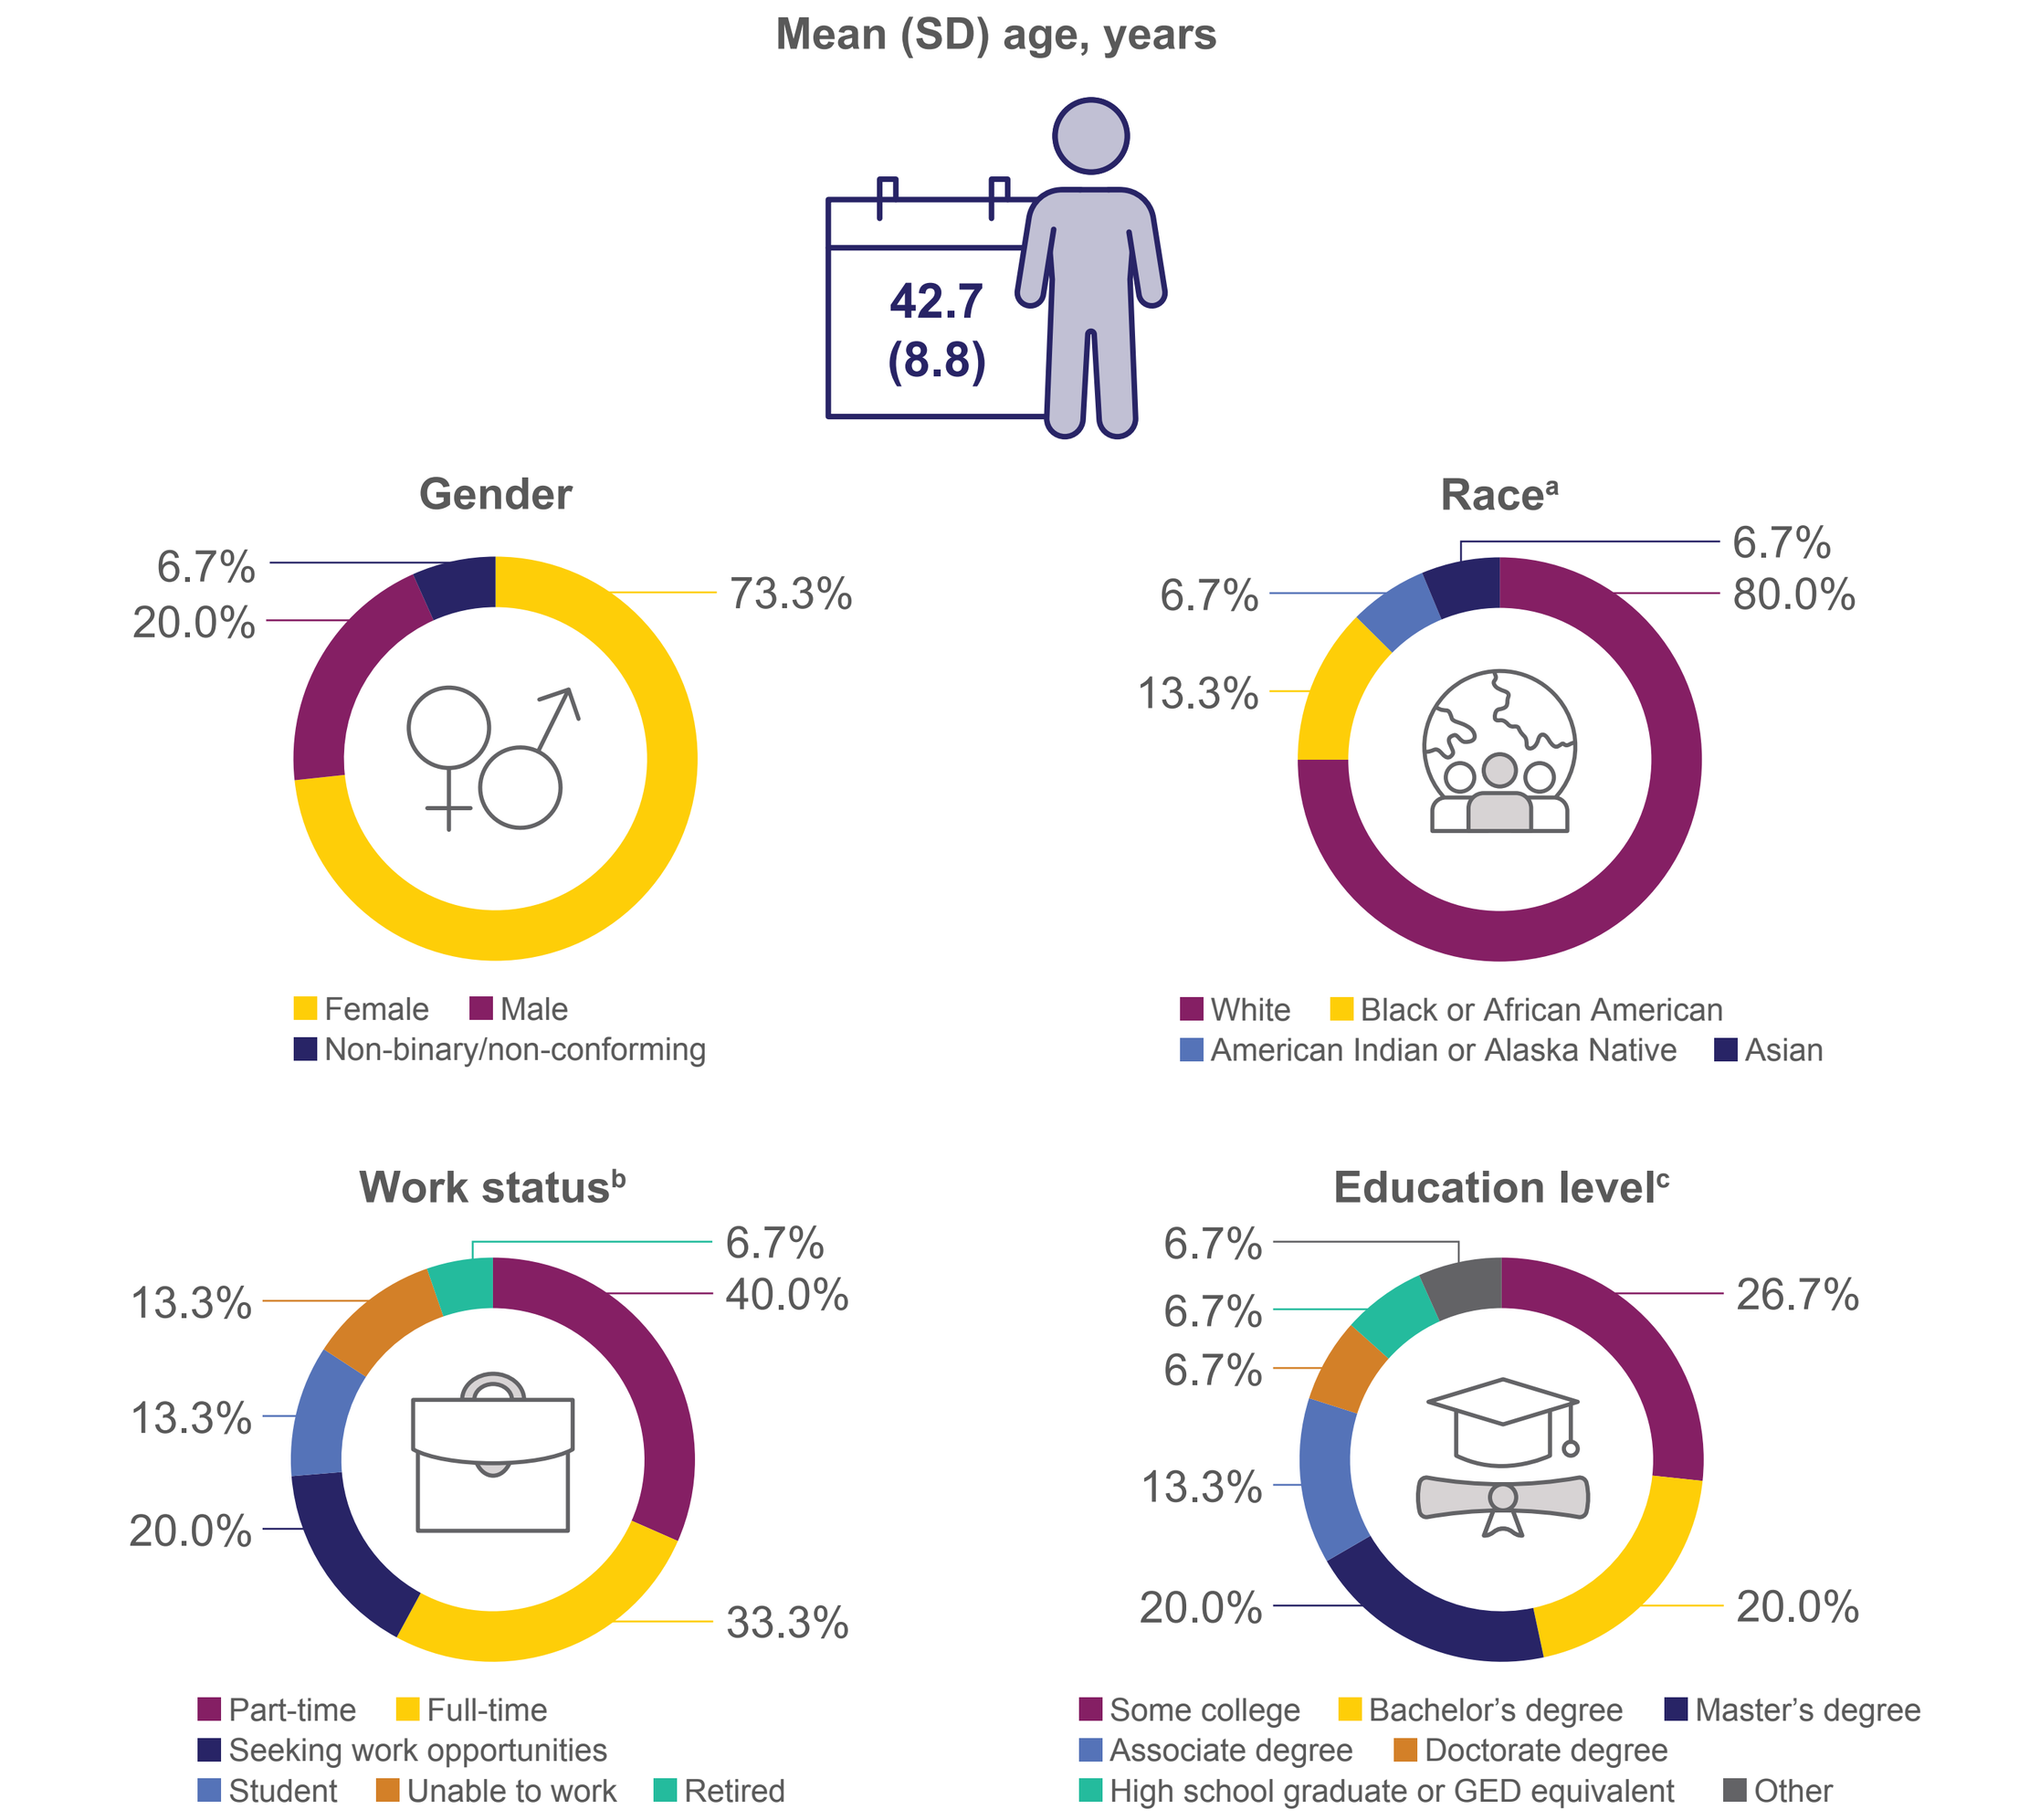

Mean (SD) age, years
42.7 (8.8)
Gender
6.7%
20.0%
73.3%
Female
Male
Non-binary/non-conforming
Race[a]
6.7%
80.0%
6.7%
13.3%
White
Black or African American
American Indian or Alaska Native
Asian
Work status[b]
6.7%
40.0%
13.3%
13.3%
20.0%
33.3%
Part-time
Full-time
Seeking work opportunities
Student
Unable to work
Retired
Education level[c]
6.7%
6.7%
6.7%
13.3%
20.0%
26.7%
20.0%
Some college
Bachelor's degree
Master's degree
Associate degree
Doctorate degree
High school graduate or GED equivalent
Other

[a]One participant reported their race as both Asian and White. [b]Two participants reported both working part-time and student; 1 participant reported both retired and unable to work; 1 participant reported both working part-time and seeking work opportunities. [c]One participant selected Other and reported "trade school." GED, General Educational Development; SD, standard deviation.

- Among 15 participants, the mean (SD) age was 42.7 (8.8) years
- Most participants were female (73.3%), White (80.0%), employed either part-time (40.0%) or full-time (33.3%), and had some college education or higher educational attainment (86.7%)

Figure 2. Sleep Inertia Occurred Daily for Most Participants and Lasted at Least 1 Hour

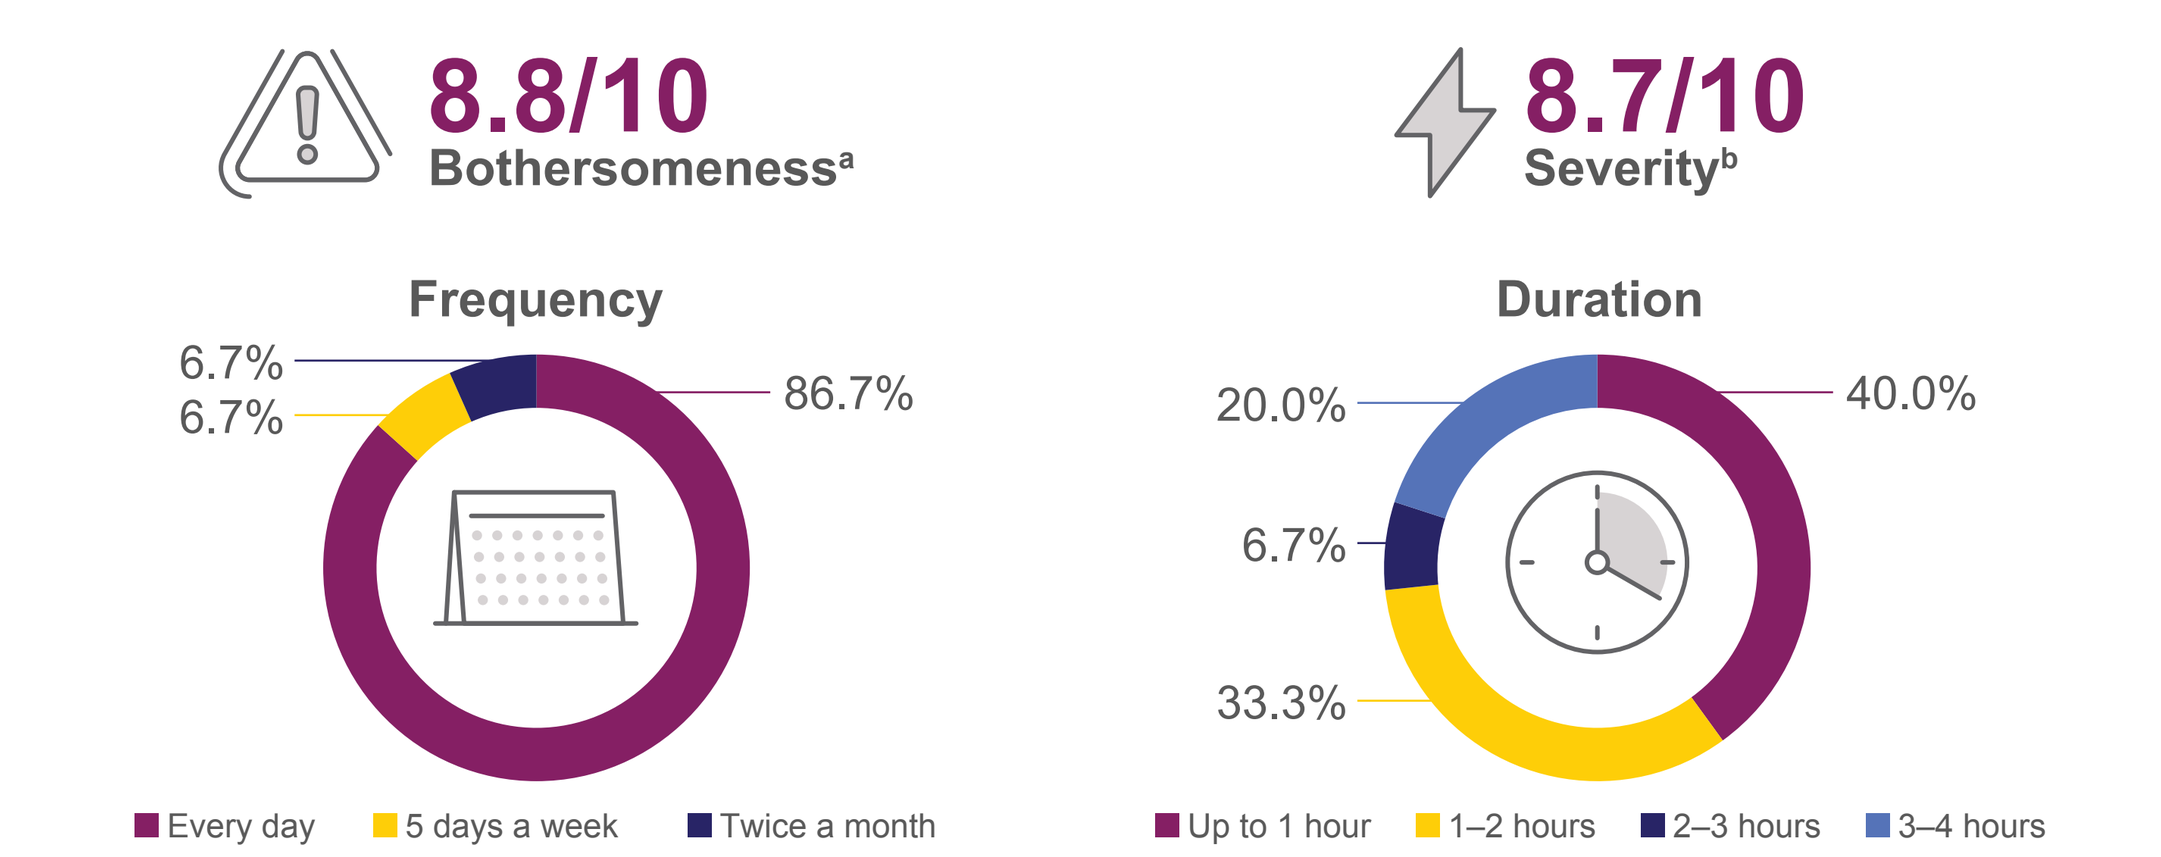

8.8/10
Bothersomeness[a]
8.7/10
Severity[b]
Frequency
6.7%
6.7%
86.7%
Every day
5 days a week
Twice a month
Duration
20.0%
6.7%
33.3%
40.0%
Up to 1 hour
1–2 hours
2–3 hours
3–4 hours

[a]Bothersomeness ratings were provided by participants when asked: "On a scale of 0 to 10, 0 meaning 'it does not bother me at all,' and 10 meaning 'you are extremely bothered by it,' how would you rate the degree to which [sleep inertia] bothers you currently? Please explain." [b]Severity ratings were provided when asked: "On a scale of 0 to 10, 0 meaning 'it is not severe at all,' and 10 meaning 'it is extremely severe,' how severe (how bad) is [sleep inertia] when it is at its worst? Please explain."

- Mean (SD) sleep inertia bothersomeness and severity ratings, both rated on a 0–10 scale, were 8.8 (1.7) and 8.7 (1.4), respectively
- Sleep inertia lasting 3 to 4 hours was reported by 1 in 5 participants
- Factors worsening sleep inertia included napping (46.7%), medications (33.3%), and too much/too little sleep the night before (40.0%) (see Supplemental Figure S1, accessed via the QR code)

Figure 3. Sleep Inertia Components

A) Of 16 Sleep Inertia Components Reported, 13 Were Deemed Salient[a,b]

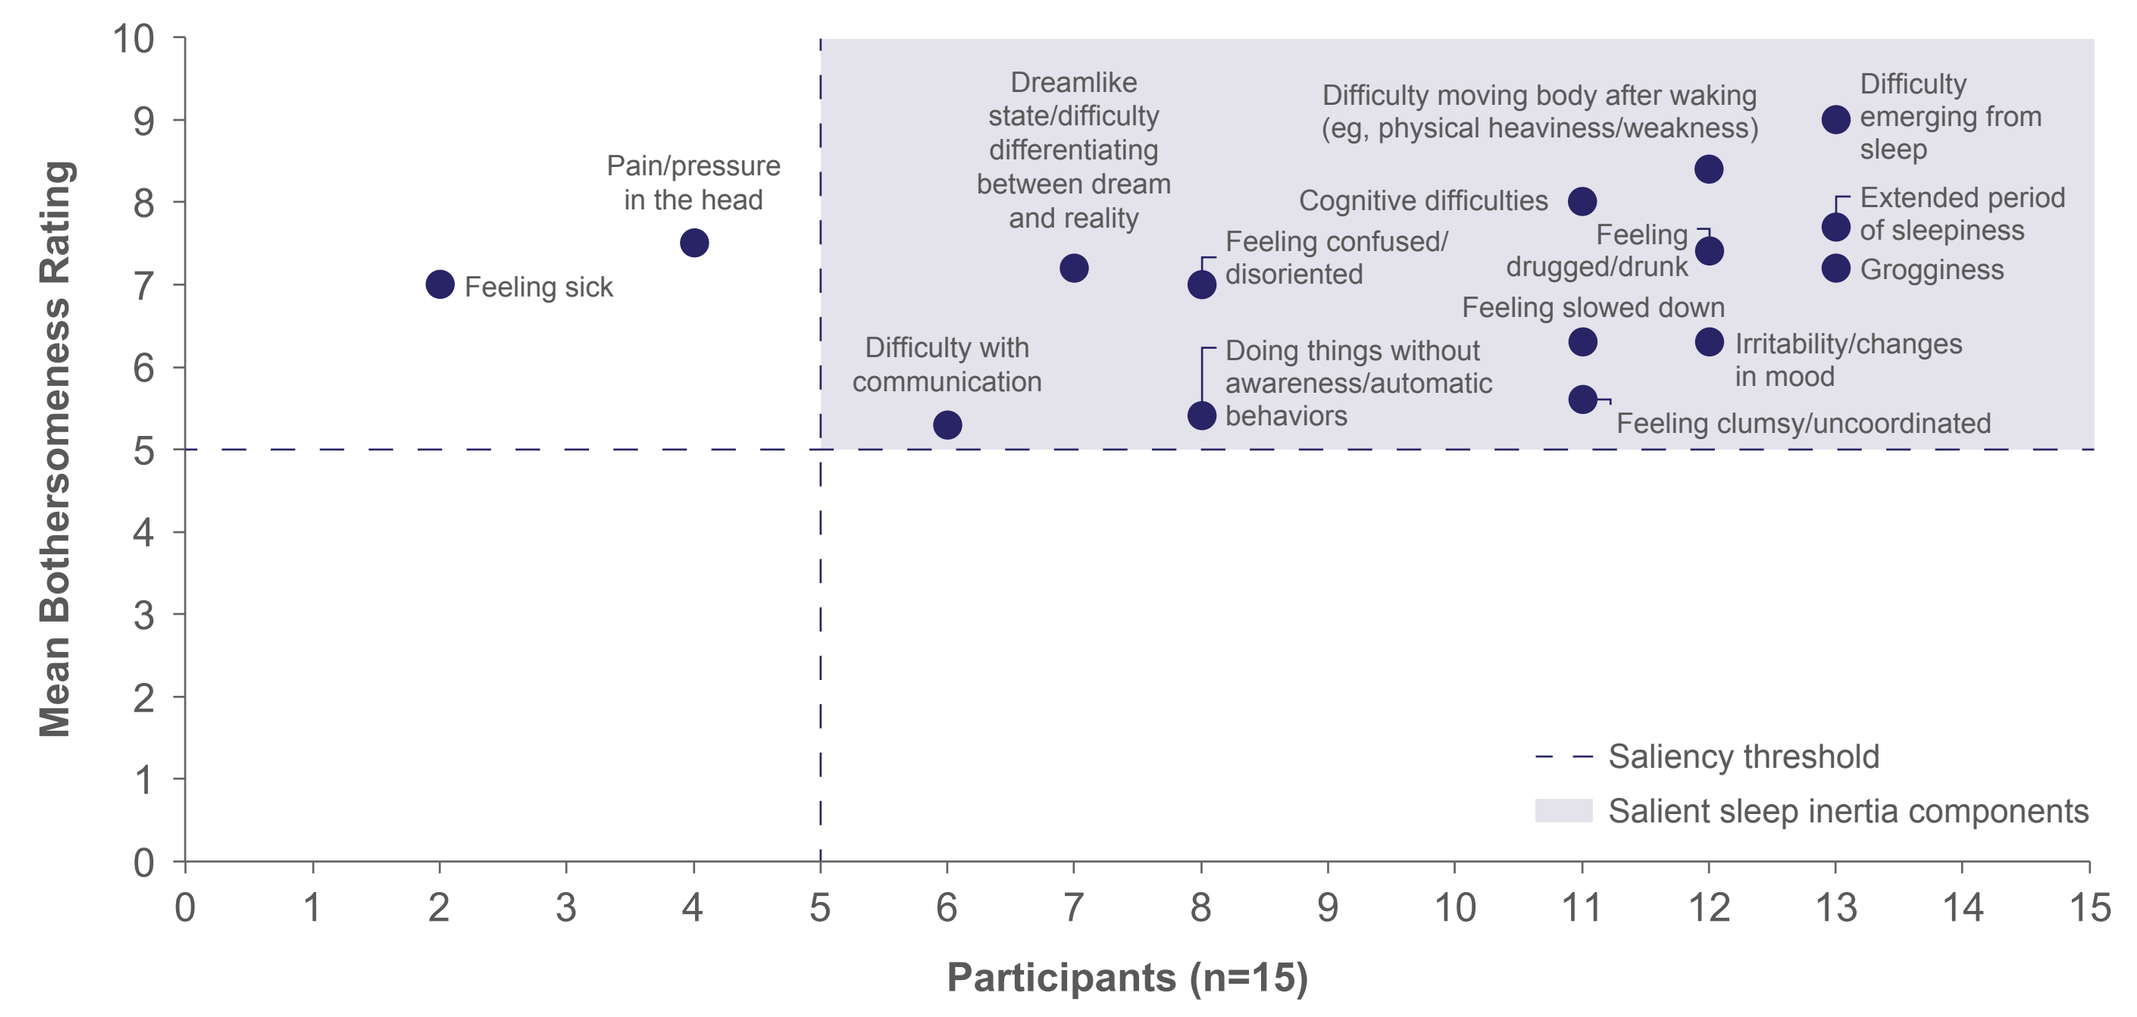

Mean Bothersomeness Rating
Participants (n=15)
Feeling sick
Pain/pressure in the head
Dreamlike state/difficulty differentiating between dream and reality
Difficulty with communication
Feeling confused/disoriented
Doing things without awareness/automatic behaviors
Cognitive difficulties
Difficulty moving body after waking (eg, physical heaviness/weakness)
Feeling drugged/drunk
Feeling slowed down
Irritability/changes in mood
Feeling clumsy/uncoordinated
Difficulty emerging from sleep
Extended period of sleepiness
Grogginess
Saliency threshold
Salient sleep inertia components

B) "Difficulty Emerging From Sleep" and "Difficulty Moving Body After Waking" Were Commonly Identified Among the Most Bothersome and Most Important Sleep Inertia Components to Improve

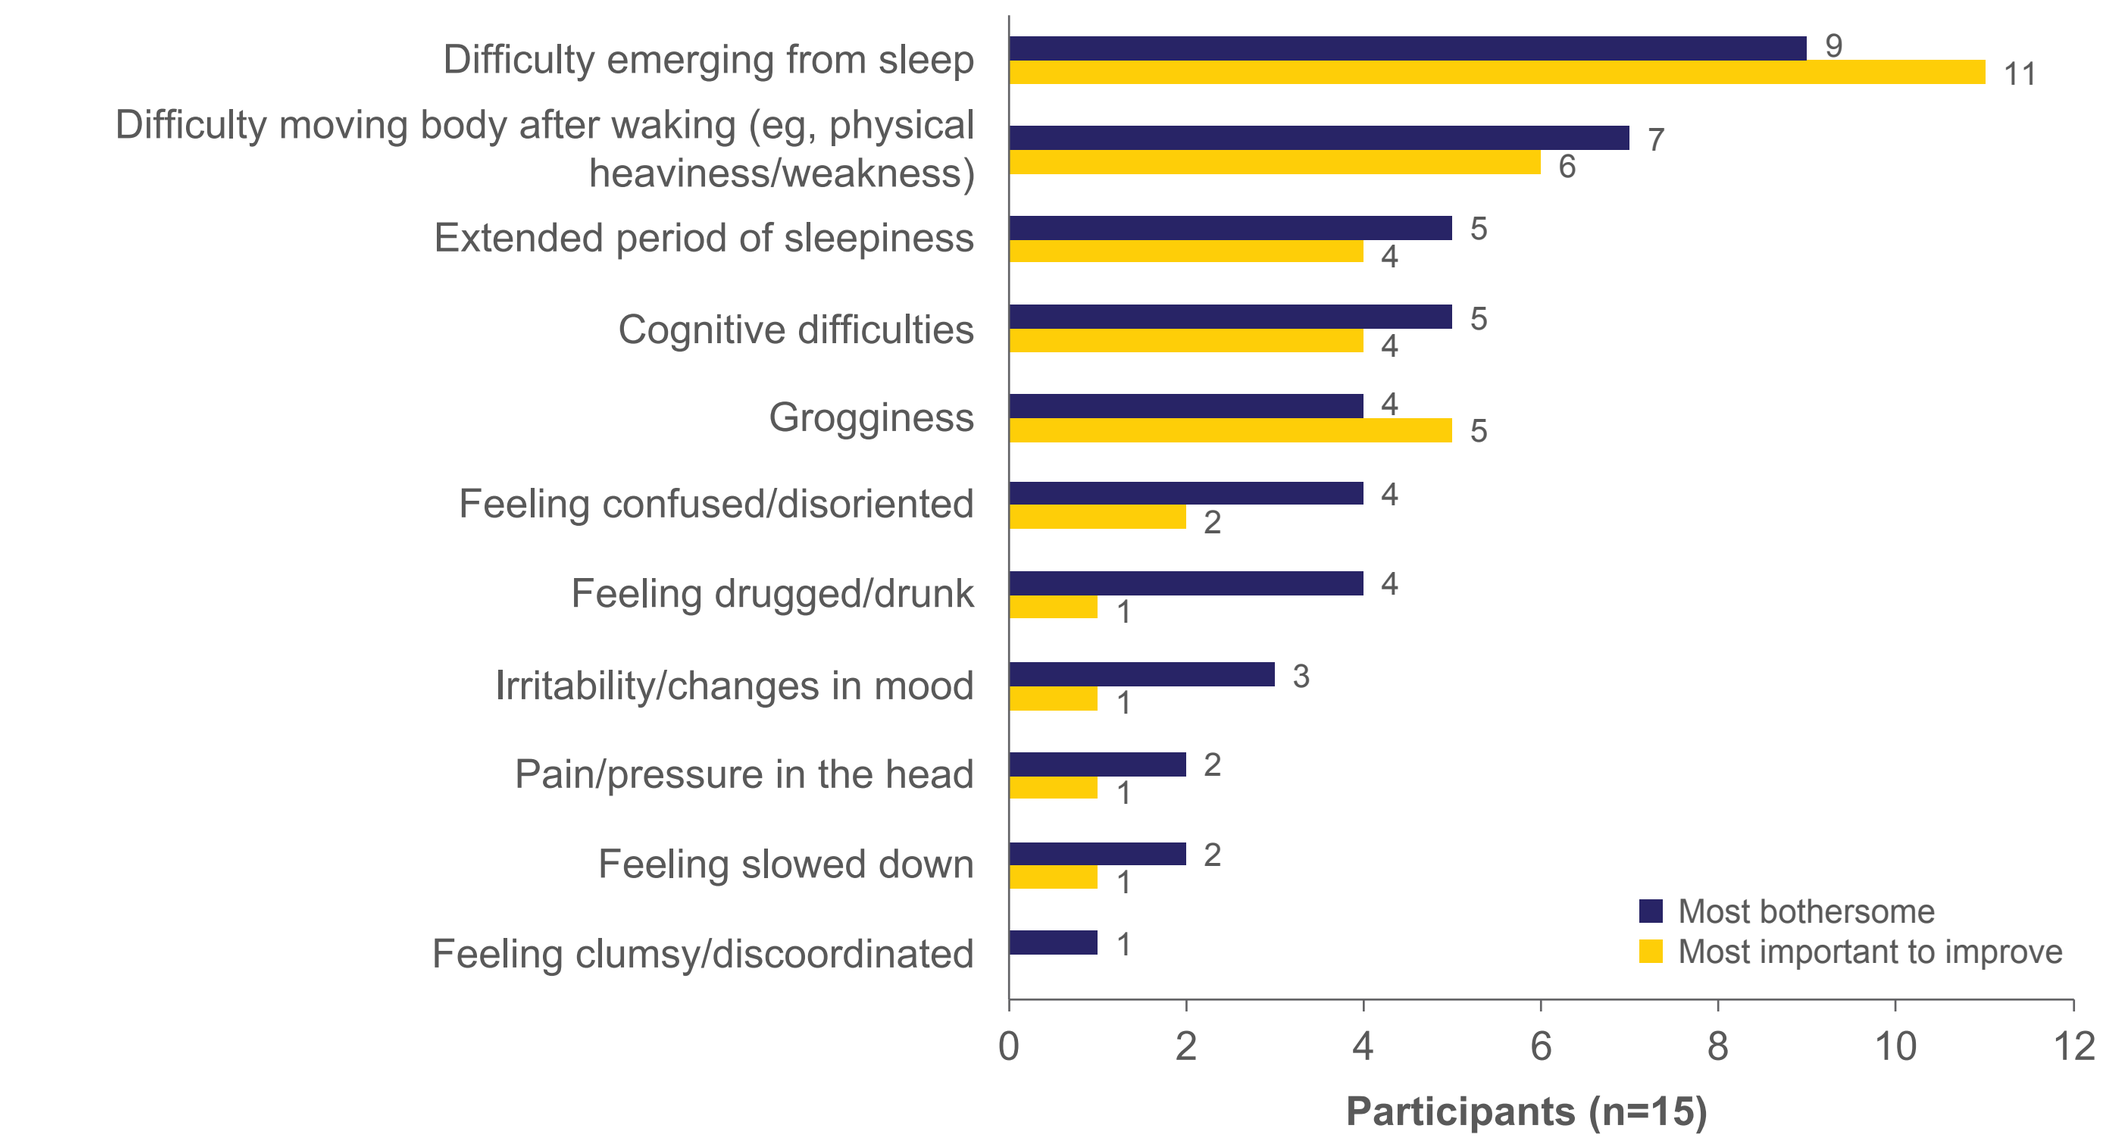

Difficulty emerging from sleep 9 11
Difficulty moving body after waking (eg, physical heaviness/weakness) 7 6
Extended period of sleepiness 5 4
Cognitive difficulties 5 4
Grogginess 4 5
Feeling confused/disoriented 4 2
Feeling drugged/drunk 4 1
Irritability/changes in mood 3 1
Pain/pressure in the head 2 1
Feeling slowed down 2 1
Feeling clumsy/discoordinated 1
Most bothersome
Most important to improve
Participants (n=15)

C) Participants Described Lived Experiences With Sleep Inertia

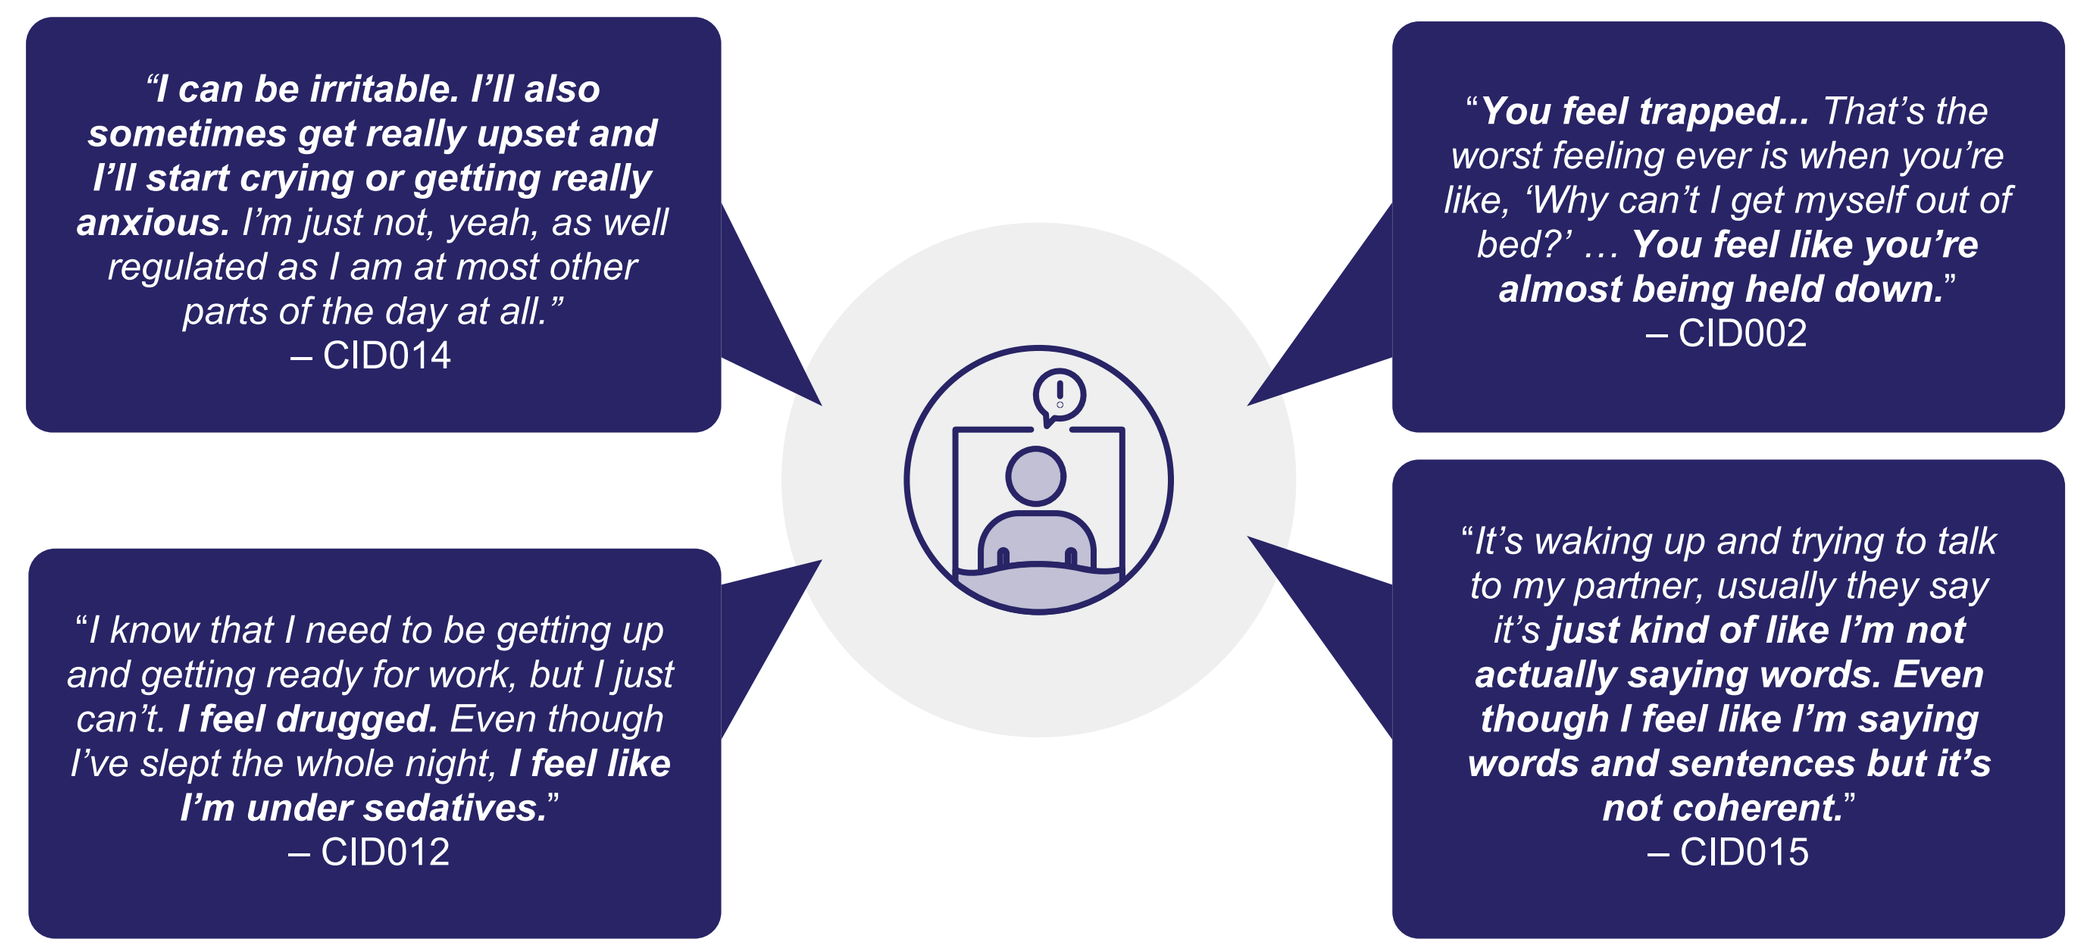

"I can be irritable. I'll also sometimes get really upset and I'll start crying or getting really anxious. I'm just not, yeah, as well regulated as I am at most other parts of the day at all." – CID014
"You feel trapped... That's the worst feeling ever is when you're like, 'Why can't I get myself out of bed?' … You feel like you're almost being held down." – CID002
"I know that I need to be getting up and getting ready for work, but I just can't. I feel drugged. Even though I've slept the whole night, I feel like I'm under sedatives." – CID012
"It's waking up and trying to talk to my partner, usually they say it's just kind of like I'm not actually saying words. Even though I feel like I'm saying words and sentences but it's not coherent." – CID015

[a]Two participants reported dizziness as a sleep inertia component but did not provide a bothersomeness rating. [b]Salience was defined as being reported by at least 33.3% of participants and having an average bothersomeness rating of 5 or higher.

- Sleep inertia components spanned physical, cognitive, emotional, and behavioral dimensions, and included "difficulty emerging from sleep" (86.7% frequency; mean [SD] bothersomeness rating: 9.0 [1.3]); "difficulty moving body after waking (eg, physical heaviness/weakness)" (80.0%; 8.4 [2.0]); "irritability/changes in mood" (80.0%; 6.3 [2.1]); and cognitive difficulties (73.3%; 8.0 [1.9])

Figure 4. Sleep Inertia Impacts

A) Of 31 Sleep Inertia Impacts Reported, 16 Were Deemed Salient[a,b]

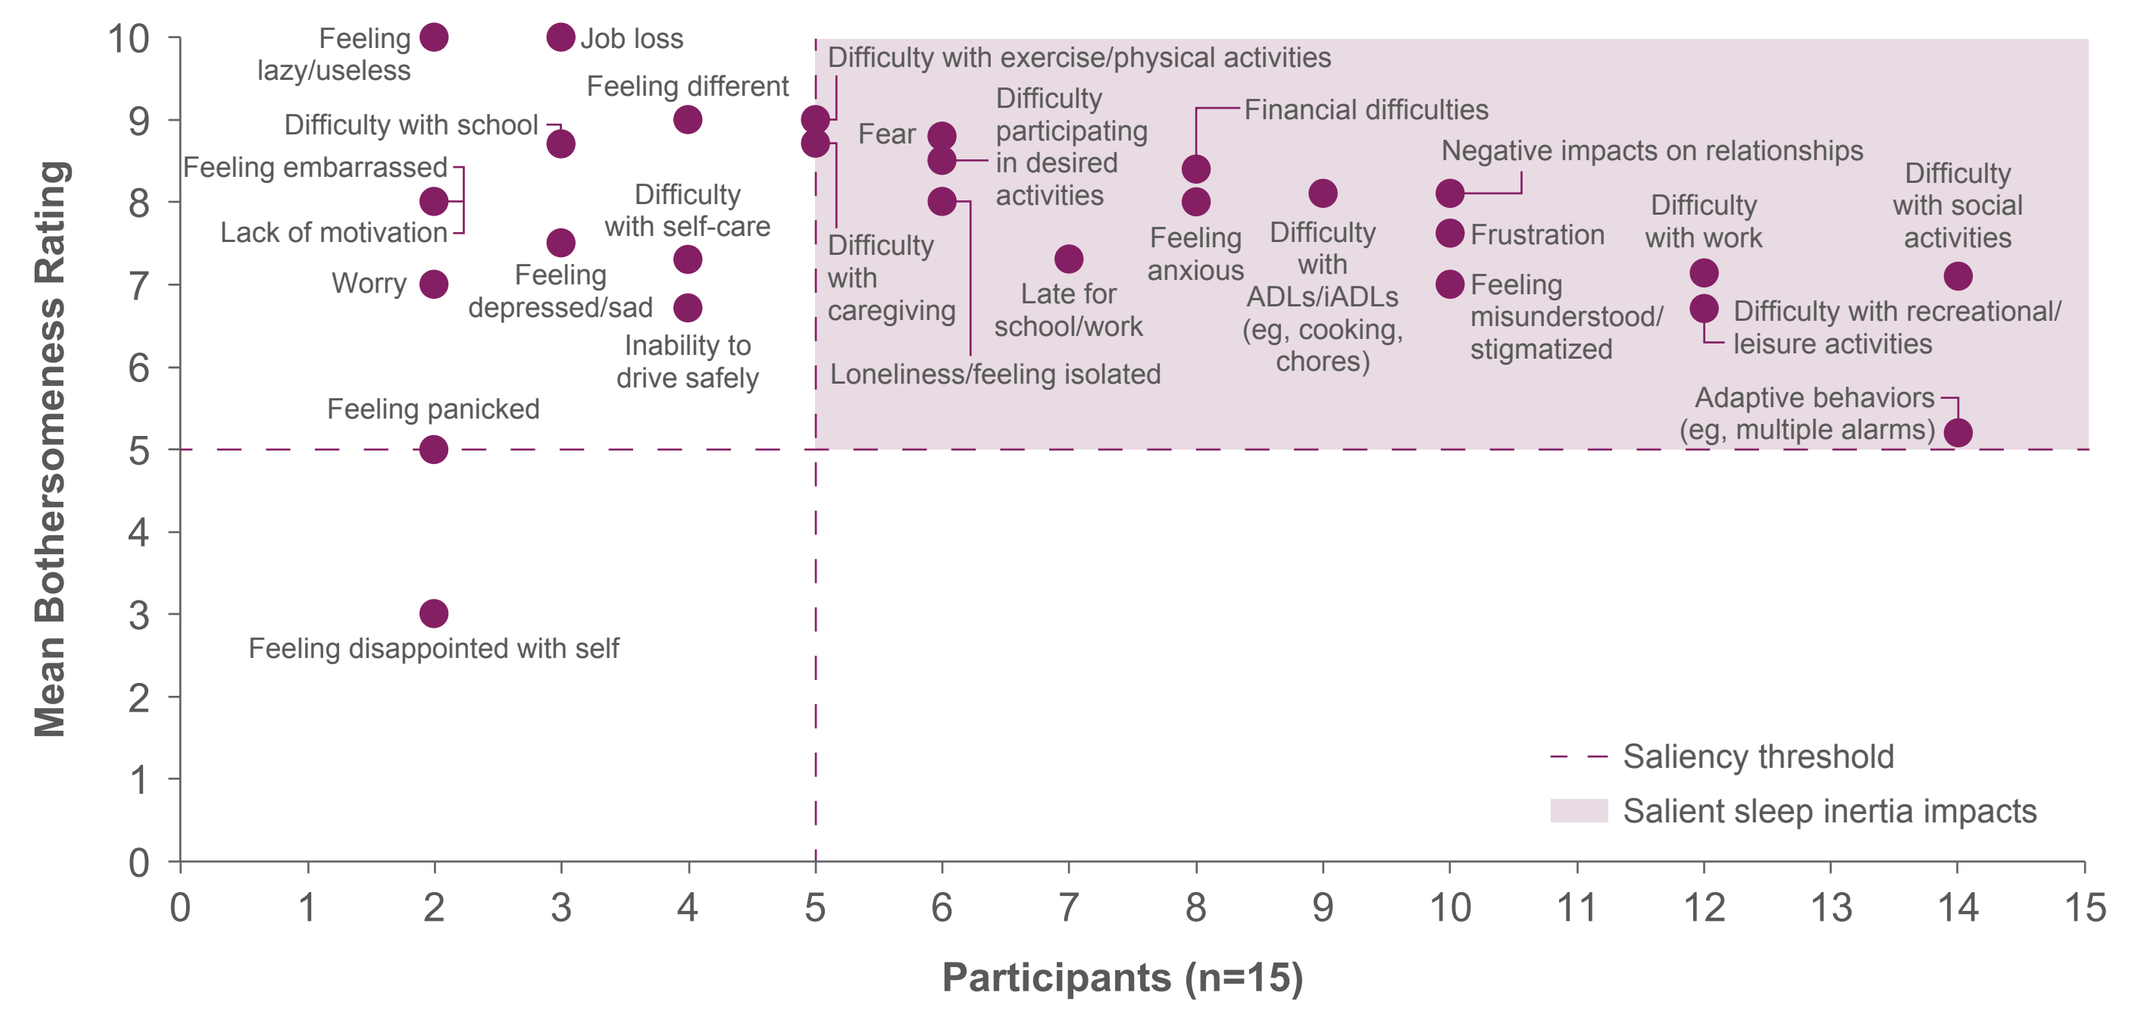

Mean Bothersomeness Rating
Participants (n=15)
Feeling lazy/useless
Job loss
Feeling different
Difficulty with school
Feeling embarrassed
Lack of motivation
Difficulty with self-care
Worry
Feeling depressed/sad
Inability to drive safely
Feeling panicked
Feeling disappointed with self
Difficulty with exercise/physical activities
Fear
Difficulty participating in desired activities
Difficulty with caregiving
Loneliness/feeling isolated
Late for school/work
Financial difficulties
Feeling anxious
Difficulty with ADLs/iADLs (eg, cooking, chores)
Negative impacts on relationships
Frustration
Feeling misunderstood/stigmatized
Difficulty with work
Difficulty with social activities
Difficulty with recreational/leisure activities
Adaptive behaviors (eg, multiple alarms)
Saliency threshold
Salient sleep inertia impacts

B) "Difficulty With Social Activities" and "Negative Impacts on Relationships" Were Commonly Identified Among the Most Bothersome Impacts

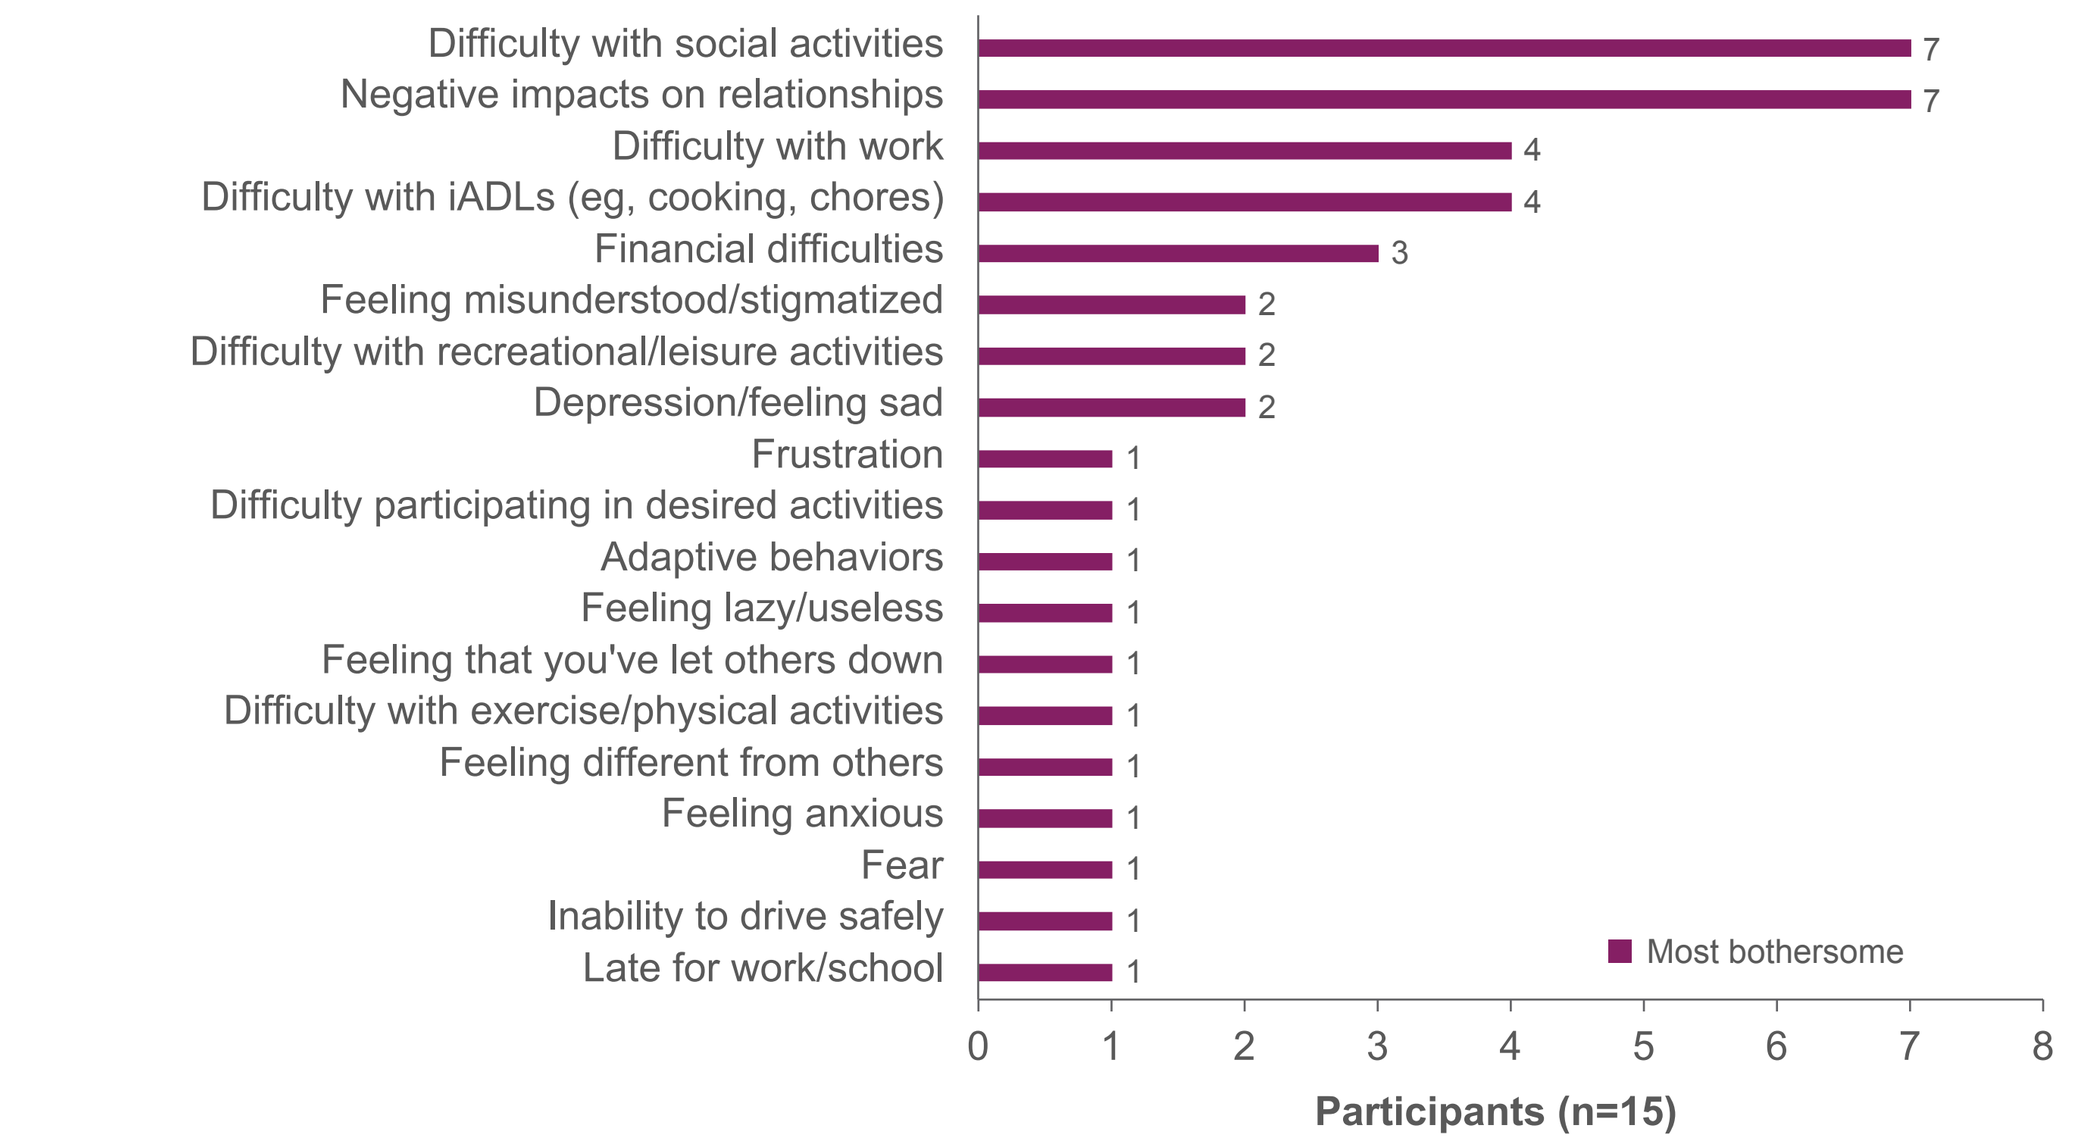

Difficulty with social activities 7
Negative impacts on relationships 7
Difficulty with work 4
Difficulty with iADLs (eg, cooking, chores) 4
Financial difficulties 3
Feeling misunderstood/stigmatized 2
Difficulty with recreational/leisure activities 2
Depression/feeling sad 2
Frustration 1
Difficulty participating in desired activities 1
Adaptive behaviors 1
Feeling lazy/useless 1
Feeling that you've let others down 1
Difficulty with exercise/physical activities 1
Feeling different from others 1
Feeling anxious 1
Fear 1
Inability to drive safely 1
Late for work/school 1
Most bothersome
Participants (n=15)

C) Participants Described How Improving Sleep Inertia Would Affect Function and Quality of Life

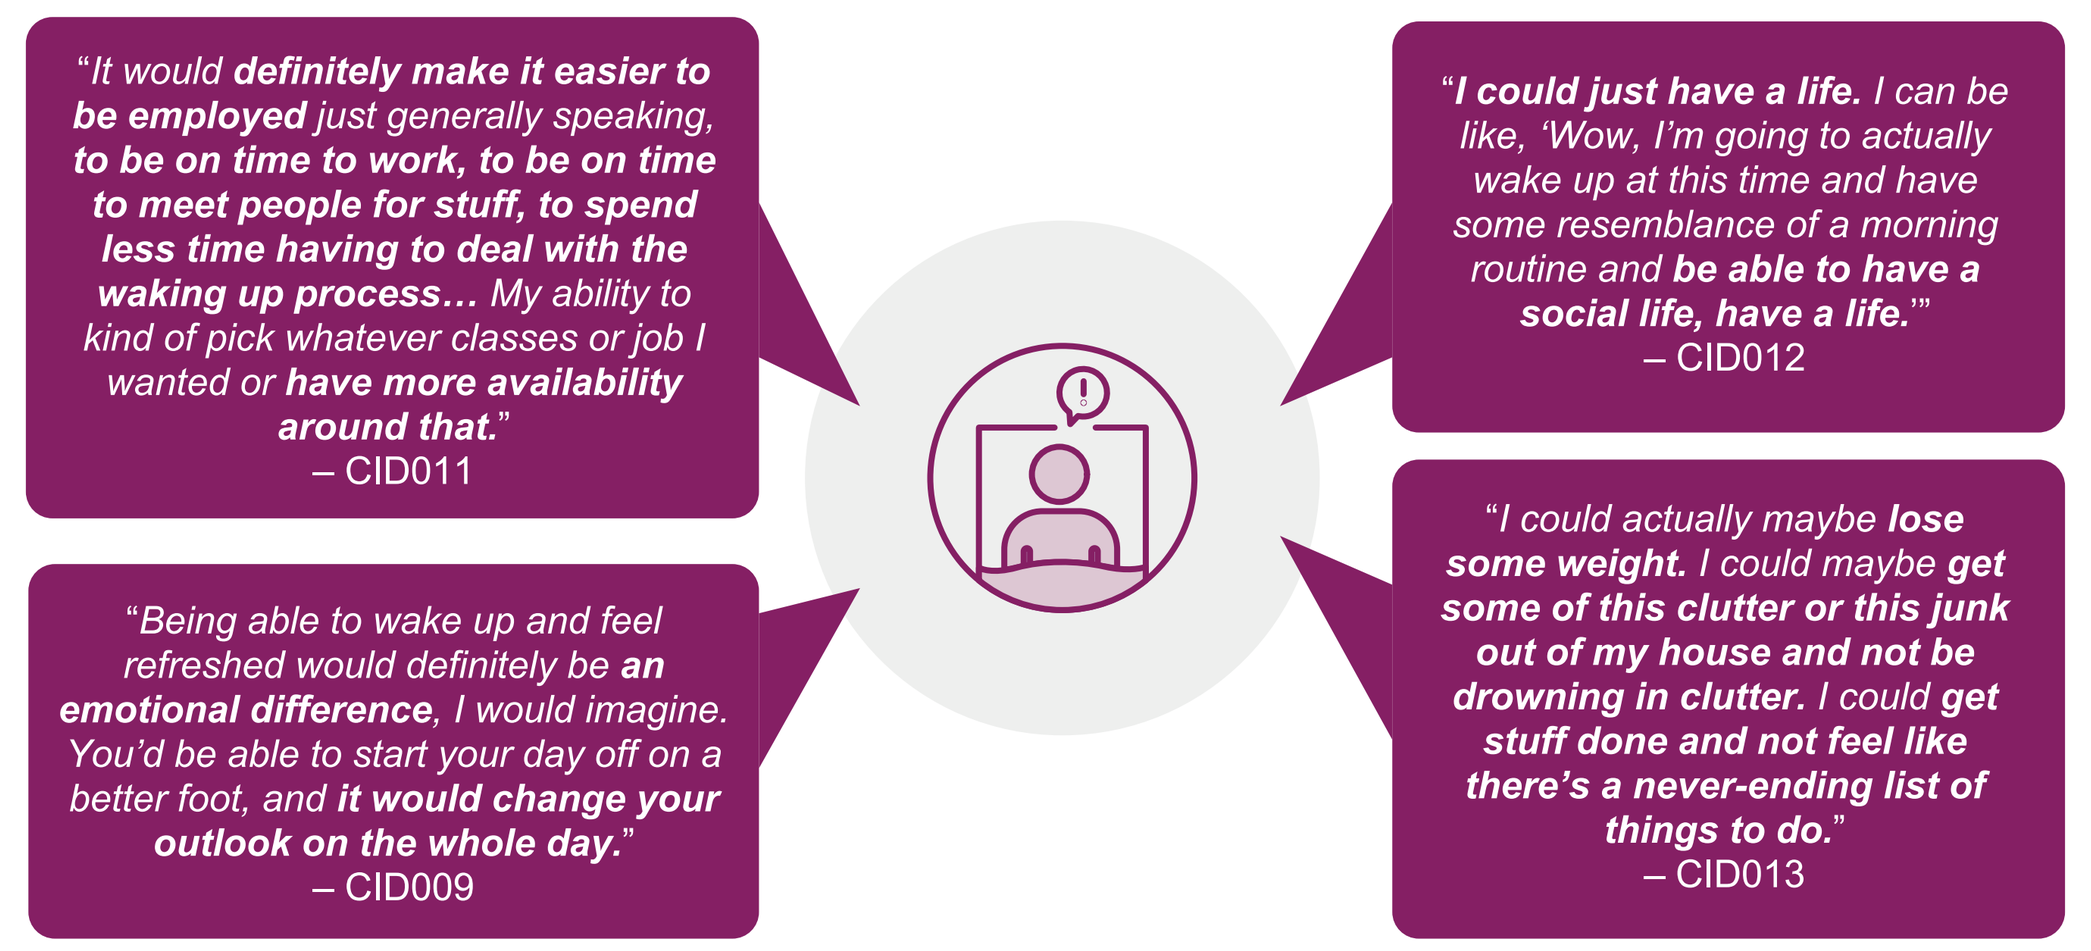

"It would definitely make it easier to be employed just generally speaking, to be on time to work, to be on time to meet people for stuff, to spend less time having to deal with the waking up process… My ability to kind of pick whatever classes or job I wanted or have more availability around that." – CID011
"I could just have a life. I can be like, 'Wow, I'm going to actually wake up at this time and have some resemblance of a morning routine and be able to have a social life, have a life.'" – CID012
"Being able to wake up and feel refreshed would definitely be an emotional difference, I would imagine. You'd be able to start your day off on a better foot, and it would change your outlook on the whole day." – CID009
"I could actually maybe lose some weight. I could maybe get some of this clutter or this junk out of my house and not be drowning in clutter. I could get stuff done and not feel like there's a never-ending list of things to do." – CID013

[a]Two participants reported "stress" as an impact of sleep inertia but did not provide a bothersomeness rating; likewise, "feeling dependent on others" and "feeling that you've let others down" were each reported by 1 participant but did not have associated bothersomeness ratings. [b]Salience was defined as being reported by at least 33.3% of participants and having an average bothersomeness rating of 5 or higher.
ADL, activity of daily living; iADL, instrumental activity of daily living.

- Sleep inertia impacts spanned 9 domains, including social, financial, behavioral/emotional, and relational domains
- Participants reported that improving sleep inertia would improve general quality of life (80.0%), as well as social functioning (26.7%), activities of daily living (26.7%), behavioral/emotional impacts (20.0%), and school/work impacts (20.0%)

Figure 5. Final Conceptual Model of Sleep Inertia in Idiopathic Hypersomnia

Sleep Inertia Components[a]

- Difficulty emerging from sleep
- Extended period of sleepiness
- Grogginess
- Difficulty moving body after waking (eg, physical heaviness/weakness)
- Feeling drugged/drunk
- Irritability/mood changes
- Feeling slowed down
- Cognitive difficulties
- Feeling clumsy/uncoordinated
- Doing things without awareness/automatic behaviors
- Feeling confused/disoriented
- Dreamlike state/difficulty differentiating between dream and reality
- Difficulty with communication

Sleep Inertia Impacts

Adaptive behaviors
- Adaptive behaviors (eg, setting multiple alarms/calendar reminders)

Behavioral/emotional
- Fear
- Feeling anxious
- Feeling misunderstood/stigmatized
- Frustration
- Loneliness/feeling isolated

ADLs/iADLs
- Difficulty with iADLs (eg, cooking, daily routine, chores)
- Difficulty with caregiving

School/work
- Difficulty with work
- Late for school

Recreation/leisure
- Difficulty with recreational/leisure activities
- Difficulty participating in desired activities

Financial
- Financial difficulties

Social
- Difficulty with social activities

Relationships
- Negative impacts on relationships

Physical functioning
- Difficulty with exercise/physical activities

Factors that worsen sleep inertia[a]:
- Taking naps
- Amount of sleep the previous night (too much/little)
- Medications
- Poor sleep quality
- High activity the previous day
- Menstrual cycle
- Sickness
- Nothing worsens sleep inertia

Factors that improve sleep inertia[a]:
- Medications
- Good sleep hygiene

[a]Concepts are ordered from highest to lowest frequency of participant report during interviews. Factors influencing sleep inertia were included if they were reported by at least 2 participants.
ADL, activity of daily living; iADL, instrumental activity of daily living.

149

The Patient Experience of Sleep Inertia in Idiopathic Hypersomnia: A Qualitative Study

Mia Wraight, BS[1]; Cristina Casstevens, PhD[1]; Sarah C. Markt, ScD, MPH[1]; Meagan Farrell, PhD[2]; Samantha Linton, PhD[2]; Mariel Alcantara, MD[2]; Christina A. Graham, BS, BA[2]; David T. Plante, MD, PhD[3]

[1]Jazz Pharmaceuticals, Palo Alto, CA, USA; [2]IQVIA, New York, NY, USA; [3]Department of Psychiatry, University of Wisconsin-Madison, Madison, WI, USA

Scan this code to access a plain language summary of the published abstract. This code is not for promotional purposes.

Scan this code to access this poster and supplemental material online. This code is not for promotional purposes.

Presented at Psych Congress 2026; September 15–19, 2026; New Orleans, LA, USA

Supplemental Material

Supplemental Figure S1. Factors[a] Reported to Worsen or Improve Sleep Inertia Severity

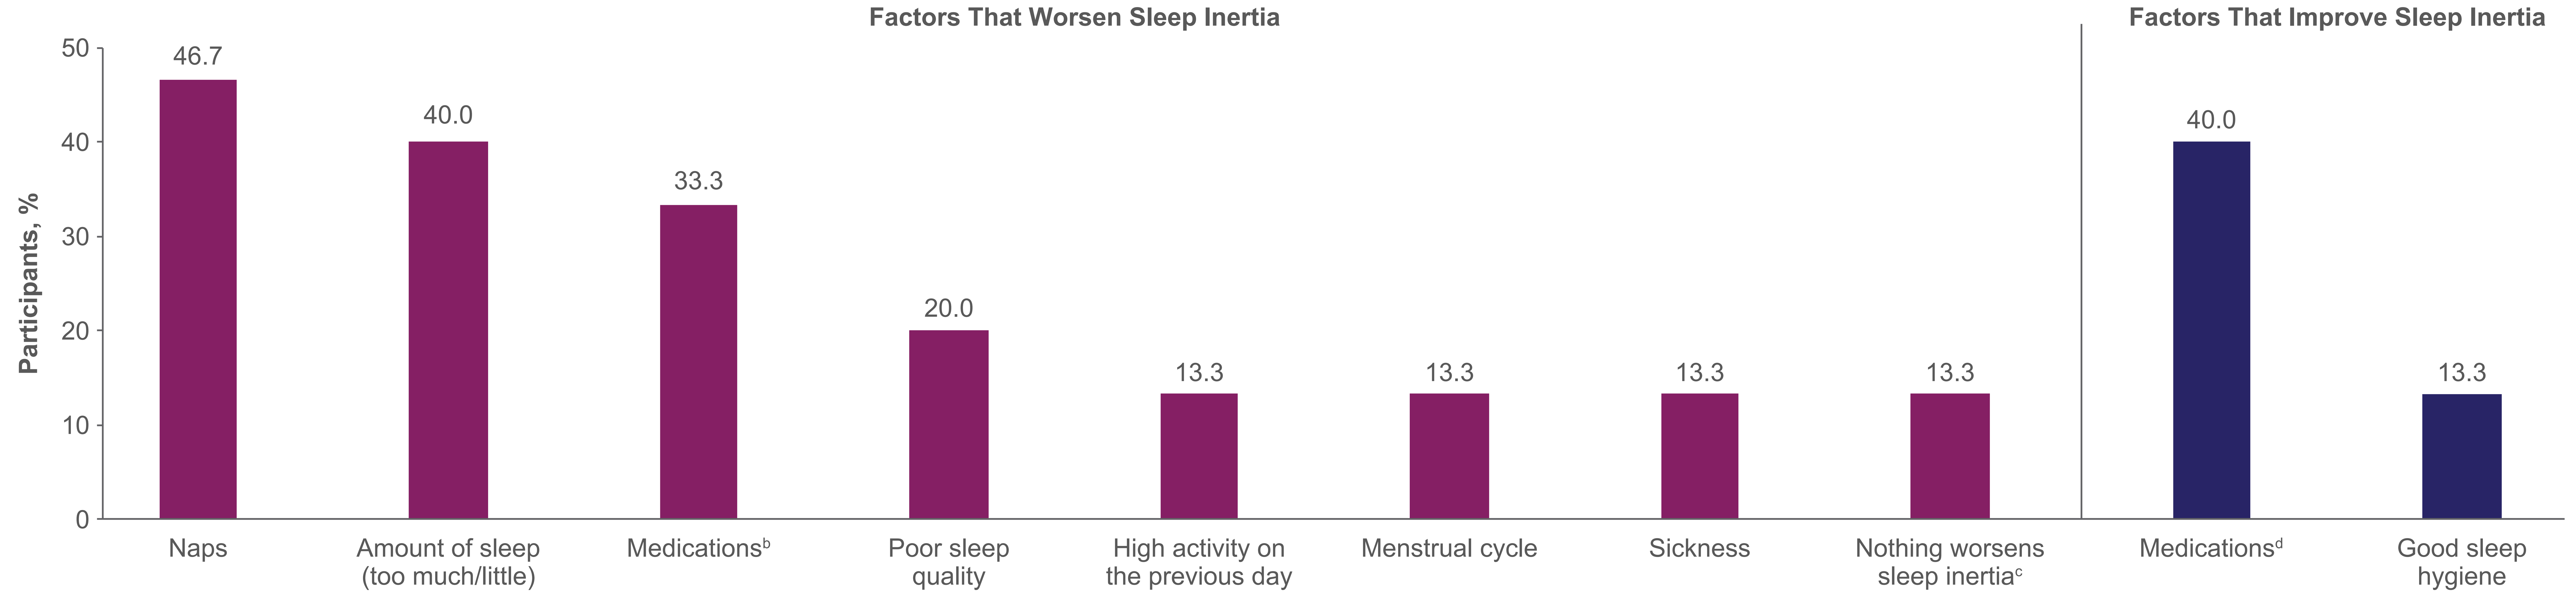
Factors That Worsen Sleep Inertia
Factors That Improve Sleep Inertia
Participants, %
0
10
20
30
40
50
46.7
40.0
33.3
20.0
13.3
13.3
13.3
13.3
40.0
13.3
Naps
Amount of sleep (too much/little)
Medications[b]
Poor sleep quality
High activity on the previous day
Menstrual cycle
Sickness
Nothing worsens sleep inertia[c]
Medications[d]
Good sleep hygiene

[a]The semi-structured interview guide included questions designed to elicit information about factors that improved or worsened sleep inertia; the factors presented are those that were reported during interviews by at least 2 participants. [b]Medications that participants reported worsened their sleep inertia included Adderall®, Excedrin PM®, and melatonin. [c]Participants had not noticed anything that worsened their sleep inertia. [d]Medications that participants reported improved their sleep inertia included Adderall®, Prozac®, and Xywav®.