166

Nighttime Symptom Prevalence and 24-Hour Symptom Burden in Patients With Narcolepsy: Findings From the Adelphi Real World Narcolepsy Disease Specific Programme™

Lionel Pinto, BPharm, MS[1]; Caroleen Drachenberg, PhD, MSPH[1]; Sarah Cotton, MA[2]; Lucy Hargreaves, BSc[2]; Rebecca Storm, BSc[2]; Sarah C. Markt, ScD, MPH[1]; Jessica K. Alexander, PhD[1]; Marisa Whalen, PharmD[3]; Silky Beaty, PharmD, MSPH[1]; Richard K. Bogan, MD, FCCP, FAASM[4]

[1]Jazz Pharmaceuticals, Palo Alto, CA, USA; [2]Adelphi Real World, Bollington, UK; [3]Jazz Pharmaceuticals, Philadelphia, PA, USA; [4]University of South Carolina School of Medicine, Columbia, SC, USA

Objective

- To assess the prevalence of specific nighttime sleep complaints and describe their association with daytime symptoms, work productivity, and quality of life in individuals with narcolepsy

Conclusions

- This real-world, multistakeholder study found that patients with evaluated nighttime sleep complaints (shallow nighttime sleep, frequent nighttime awakenings, and/or early awakenings) were numerically more likely than those without these complaints to experience daytime symptoms such as brain fog, inattention, poor memory, and moderate or severe excessive daytime sleepiness
- Patients experiencing ≥1 evaluated nighttime sleep complaint also reported numerically greater work impairment and numerically lower health-related quality of life than those without these complaints, suggesting a higher overall disease burden across the day among those experiencing nighttime sleep complaints
- Since no statistical comparisons were conducted, findings are descriptive and cannot be interpreted causally; in addition, reports of nighttime sleep complaints were provided by patients' healthcare providers rather than by the patients themselves
- These findings highlight the 24-hour burden of narcolepsy; further research into the impact of nighttime sleep complaints and relationship with daytime burden is required to optimize treatment and management of patients with narcolepsy

References: **1.** Kornum BR, et al. *Nat Rev Dis Primers.* 2017;3:16100. **2.** Ortiz LE, et al. *CNS Drugs.* 2025;39(Suppl 1):23-36. **3.** Anderson P, et al. *Curr Med Res Opin.* 2023;39(12):1707-1715. **4.** Babineaux SM, et al. *BMJ Open.* 2016;6(8):e010352. **5.** Higgins V, et al. *Diabetes Metab Syndr Obes.* 2016;9:371-380. **6.** Rabin R, de Charro F. *Ann Med.* 2001;33(5):337-343. **7.** Reilly MC, et al. *Pharmacoeconomics.* 1993;4(5):353-365.

Support and Acknowledgments: The Adelphi Real World Narcolepsy Disease Specific Programme™ is a wholly owned Adelphi Real World Product. Jazz Pharmaceuticals is one of multiple subscribers to the Disease Specific Programme™. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees, Aeja Jackson, PhD, MS, and Eleanor Bush, MA, provided medical writing support and Katie Herman provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: L Pinto, C Drachenberg, SC Markt, JK Alexander, M Whalen, and **S Beaty** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **S Cotton, L Hargreaves,** and **R Storm** are employees of Adelphi Real World. **RK Bogan** is a shareholder of Watermark Medical and Healthy Humming; serves on the board of directors for Watermark; is a medical consultant to Alkermes, Avadel, Harmony Biosciences, Jazz Pharmaceuticals, Oventus, and Takeda; has conducted industry-funded research for Avadel, Axsome, Balance, Bayer, Bresotec, Eisai, Fresca, Idorsia, Jazz Pharmaceuticals, LivaNova, Merck, NLS, Noctrix, Philips, Roche, Sanofi, Sommetrics, Suven, Takeda, and Vanda; and is on speakers bureaus for Axsome, Eisai, Harmony Biosciences, and Jazz Pharmaceuticals

Scan this code to access a plain language summary of the published abstract. This code is not for promotional purposes.

Scan this code to access this poster online. This code is not for promotional purposes.

Presented at Psych Congress 2026; September 15–19, 2026; New Orleans, LA, USA

Introduction

- Narcolepsy is a chronic neurological sleep disorder characterized by symptoms including excessive daytime sleepiness (EDS), disrupted nighttime sleep, sleep paralysis, hypnagogic/hypnopompic hallucinations, and cataplexy (narcolepsy type 1 only); these symptoms can substantially impair daily functioning and quality of life[1]
- A recent US patient survey highlighted the substantial burden of narcolepsy, with most patients reporting that narcolepsy had an extremely severe or very severe impact on their daily lives[2]
- However, the relationship between nighttime sleep complaints and daytime symptom burden remains poorly understood

Methods

- Data were drawn from the Adelphi Real World Narcolepsy Disease Specific Programme™ (DSP), a real-world, cross-sectional survey of patients and their treating physicians in the United States from August 2025 to March 2026
 - The DSP methodology has been previously described, validated, and shown to be representative and consistent over time[3-5]
- The healthcare provider (HCP) survey was completed by primary care physicians, neurologists, psychiatrists, and pulmonologists
 - HCPs provided data on patient demographics, clinical characteristics, current Epworth Sleepiness Scale (ESS) scores (normal daytime sleepiness, 6–10; mild EDS, 11–12; moderate EDS, 13–15; severe EDS, 16–24), and current daytime symptoms and nighttime sleep complaints
 - Evaluated nighttime sleep complaints comprised shallow nighttime sleep, frequent nighttime awakenings, and early morning awakenings
- Patients completed the Work Productivity and Activity Impairment–Specific Health Problem (WPAI-SHP) and EuroQol Visual Analogue Scale (EQ-VAS; score range: 0–100, where 0 = the worst health imaginable and 100 = the best health imaginable) questionnaires,[3,4] along with de novo questions about the impact of narcolepsy on quality of life
- Findings are descriptive; numerical differences are reported for the study sample, stratified by evaluated nighttime sleep complaints as reported by HCPs

Results

Figure 1. HCP-Reported Patient Demographic and Clinical Characteristics (N=637)

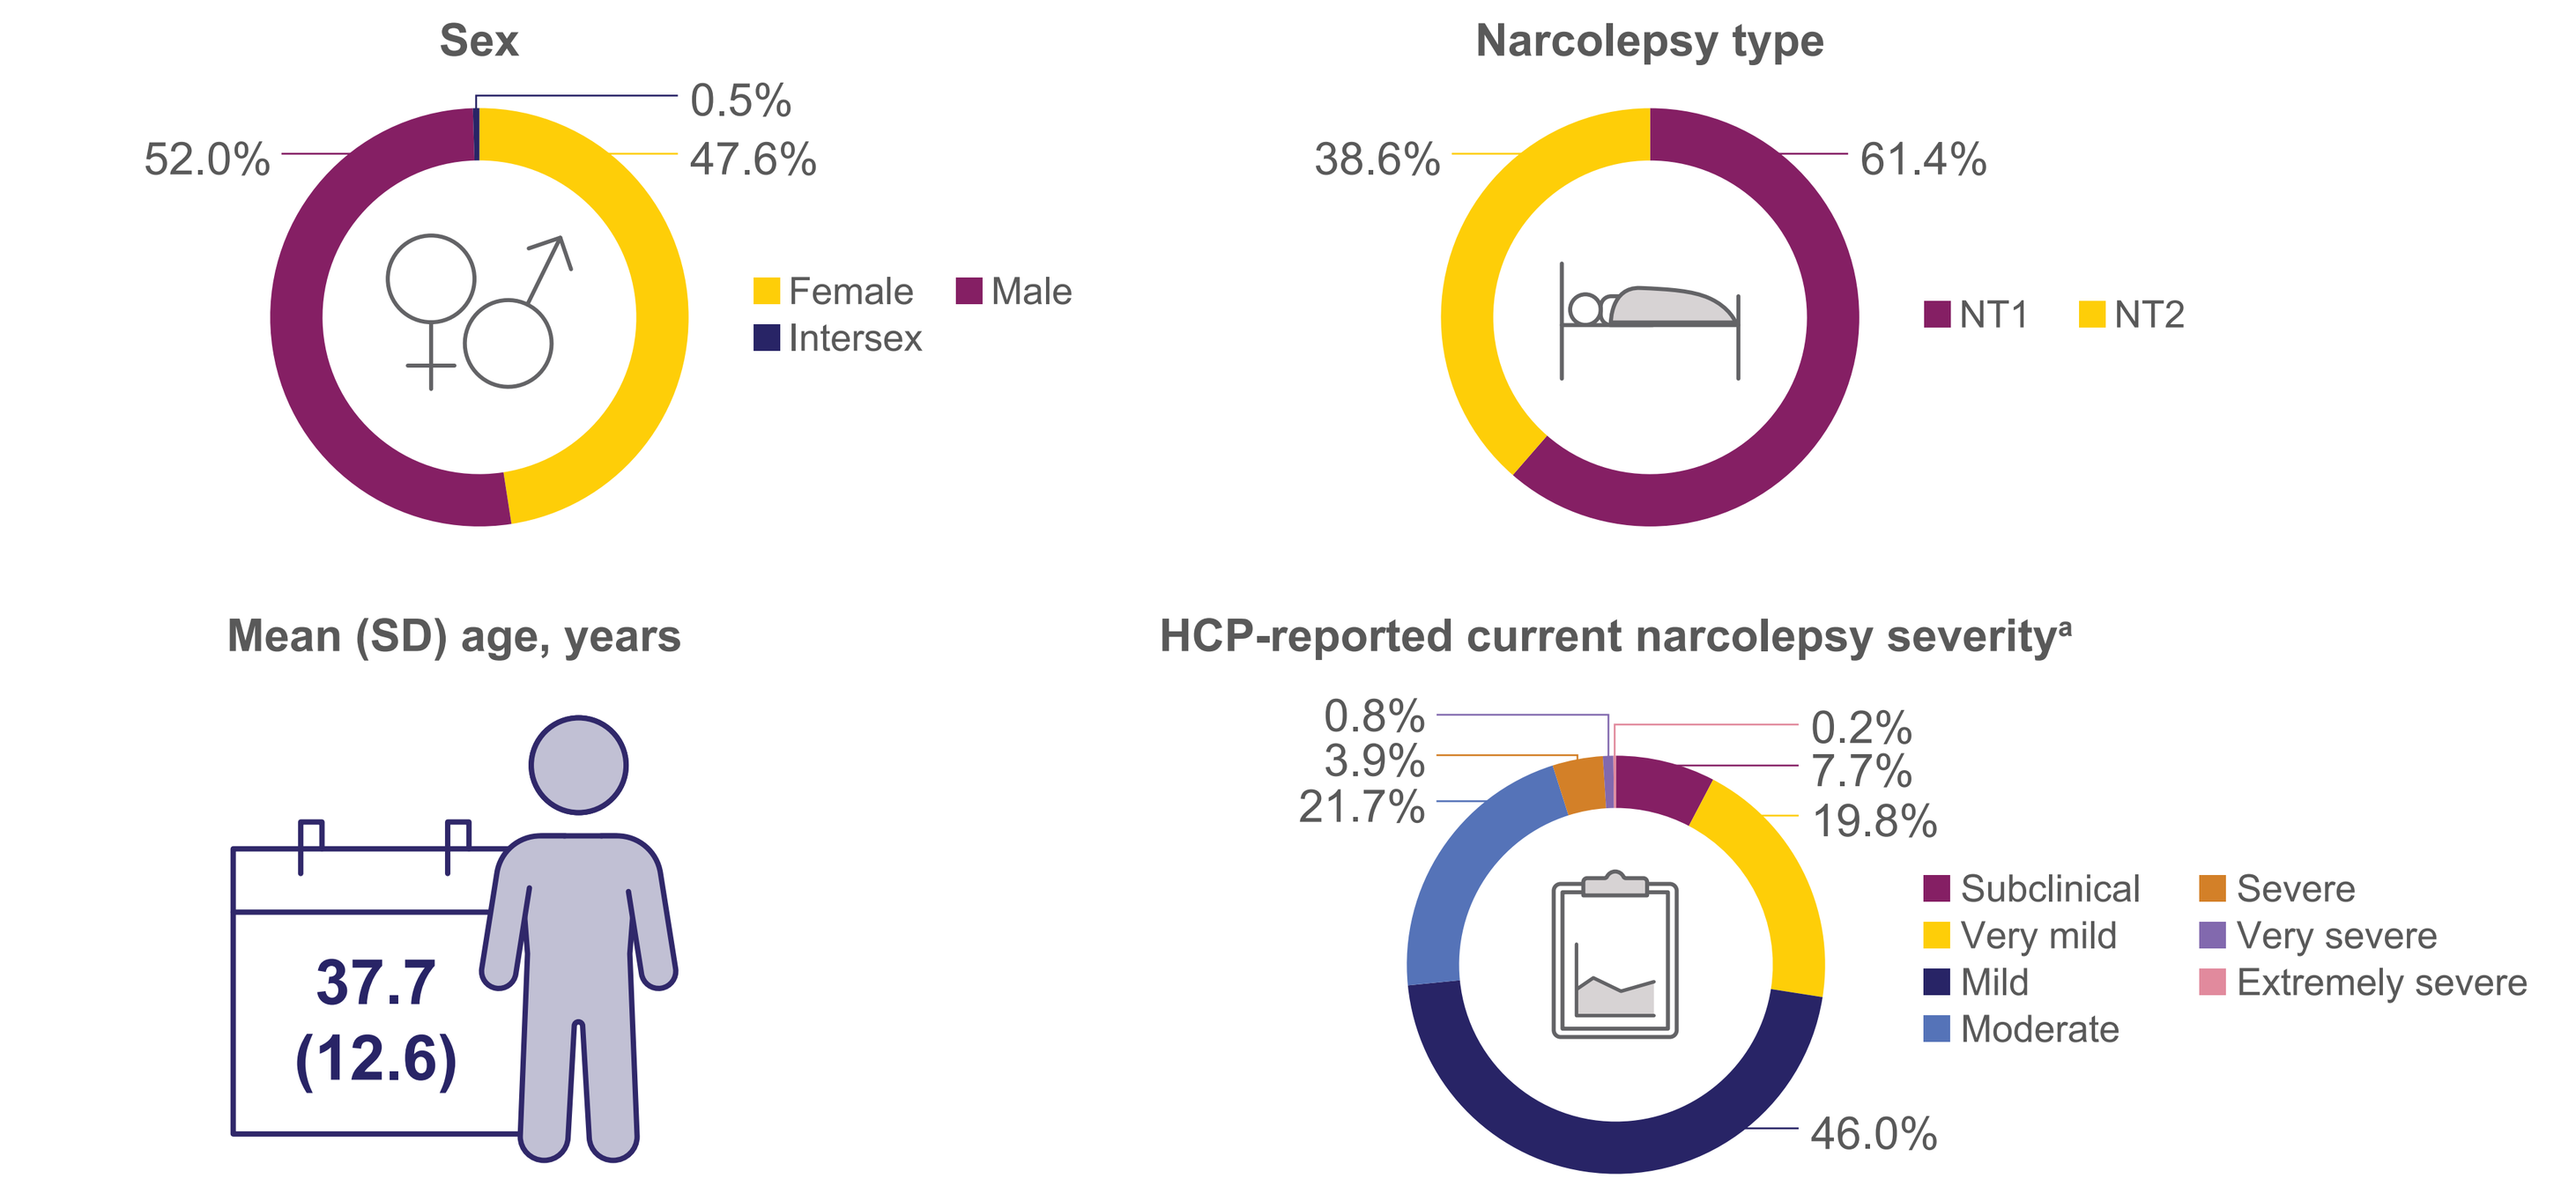

Sex
52.0%
0.5%
47.6%
Female
Male
Intersex
Narcolepsy type
38.6%
61.4%
NT1
NT2
Mean (SD) age, years
37.7 (12.6)
HCP-reported current narcolepsy severity[a]
0.8%
3.9%
21.7%
0.2%
7.7%
19.8%
46.0%
Subclinical
Very mild
Mild
Moderate
Severe
Very severe
Extremely severe

[a]De novo question.
HCP, healthcare provider; NT1, narcolepsy type 1; NT2, narcolepsy type 2; SD, standard deviation.

- HCP-recorded data were available for 637 patients
- The mean (SD) patient age was 37.7 (12.6) years, 47.6% were female, and 61.4% had narcolepsy type 1
- Mild narcolepsy severity was reported by HCPs for 46.0% of patients

Figure 2. HCP-Reported Prevalence of Shallow Nighttime Sleep, Frequent Nighttime Awakenings, and/or Early Morning Awakenings, Among Those With ≥1 Evaluated Nighttime Sleep Complaint

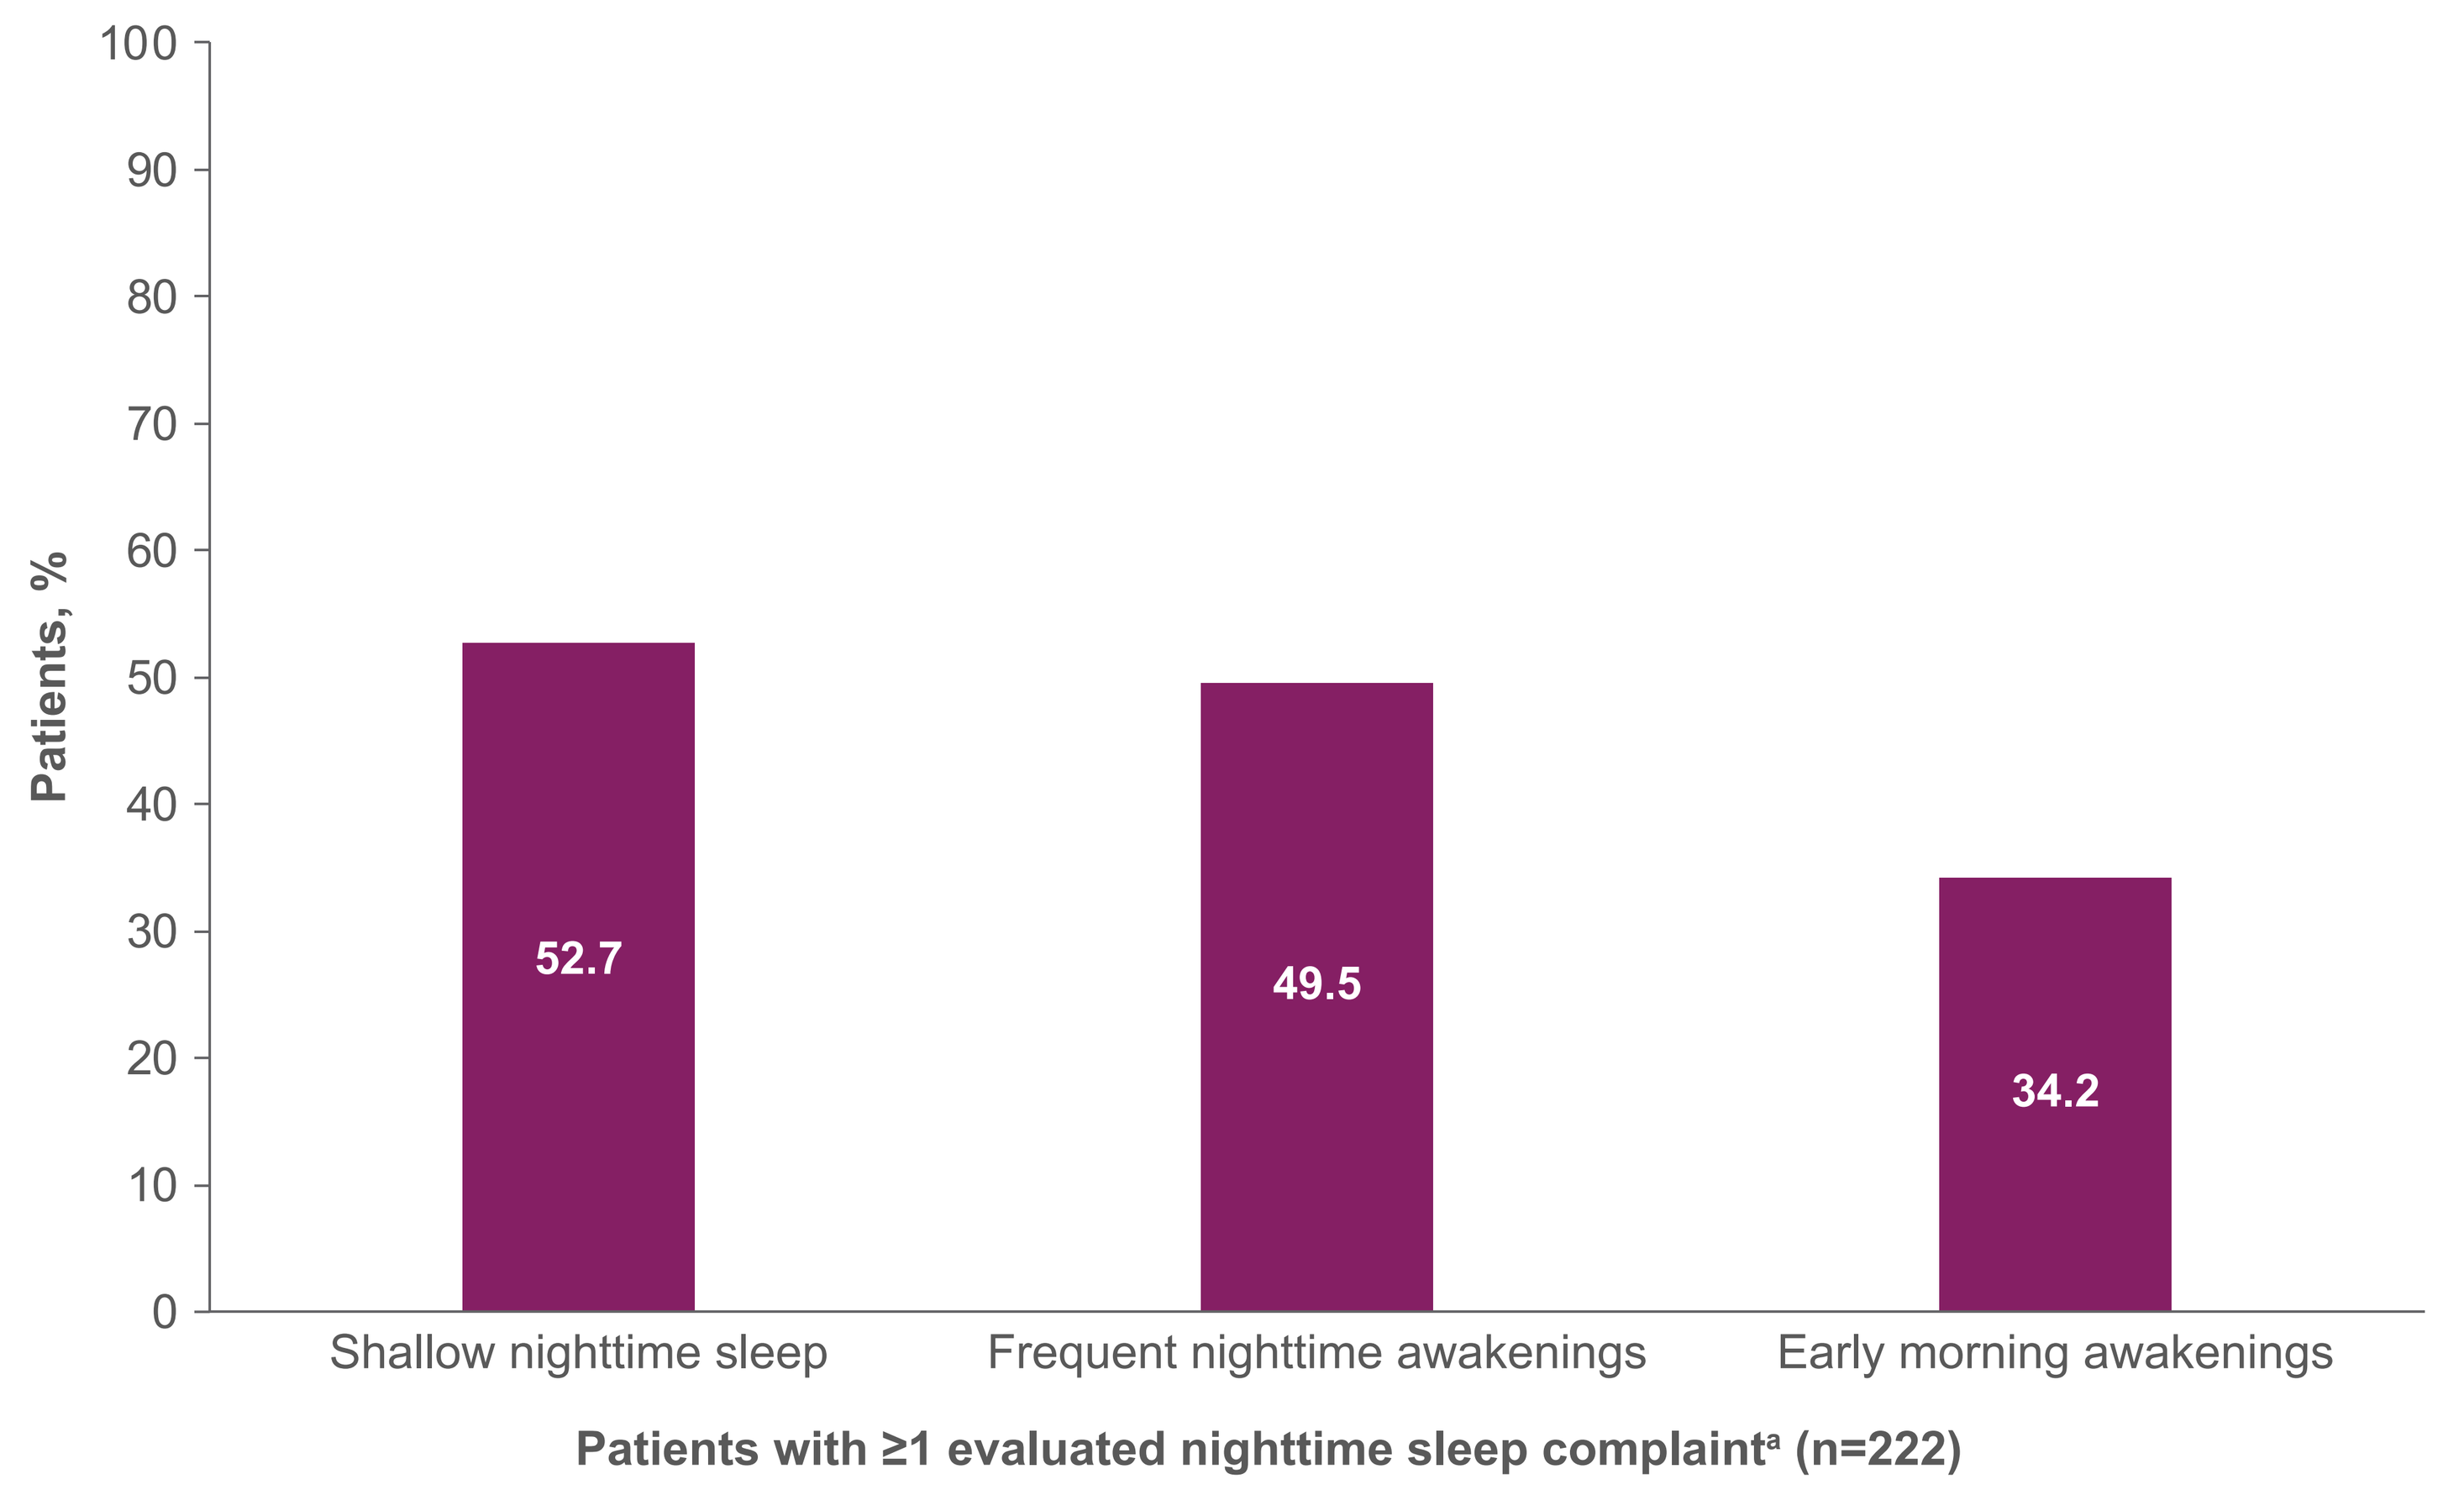

Patients, %
100
90
80
70
60
50
40
30
20
10
0
52.7
49.5
34.2
Shallow nighttime sleep
Frequent nighttime awakenings
Early morning awakenings
Patients with ≥1 evaluated nighttime sleep complaint[a] (n=222)

[a]Evaluated nighttime sleep complaints comprised frequent nighttime awakenings, shallow nighttime sleep, and early morning awakenings, as reported by HCPs.
HCP, healthcare provider.

- Of the 637 patients, HCPs reported that 222 patients (34.9%) had ≥1 evaluated nighttime sleep complaint at the time of data collection; 64.7% of these patients were reported to experience nighttime sleep complaints several nights per week or more frequently
- Of the 3 nighttime sleep complaints evaluated, shallow sleep was the most frequently recorded (52.7%, n=117), followed by frequent nighttime awakenings (49.5%, n=110) and early morning awakenings (34.2%, n=76)

Figure 3. Patients With vs Without ≥1 Evaluated Nighttime Sleep Complaint Were Numerically More Likely to Also Have Moderate or Severe EDS (HCP-Reported Data; n=291)[a]

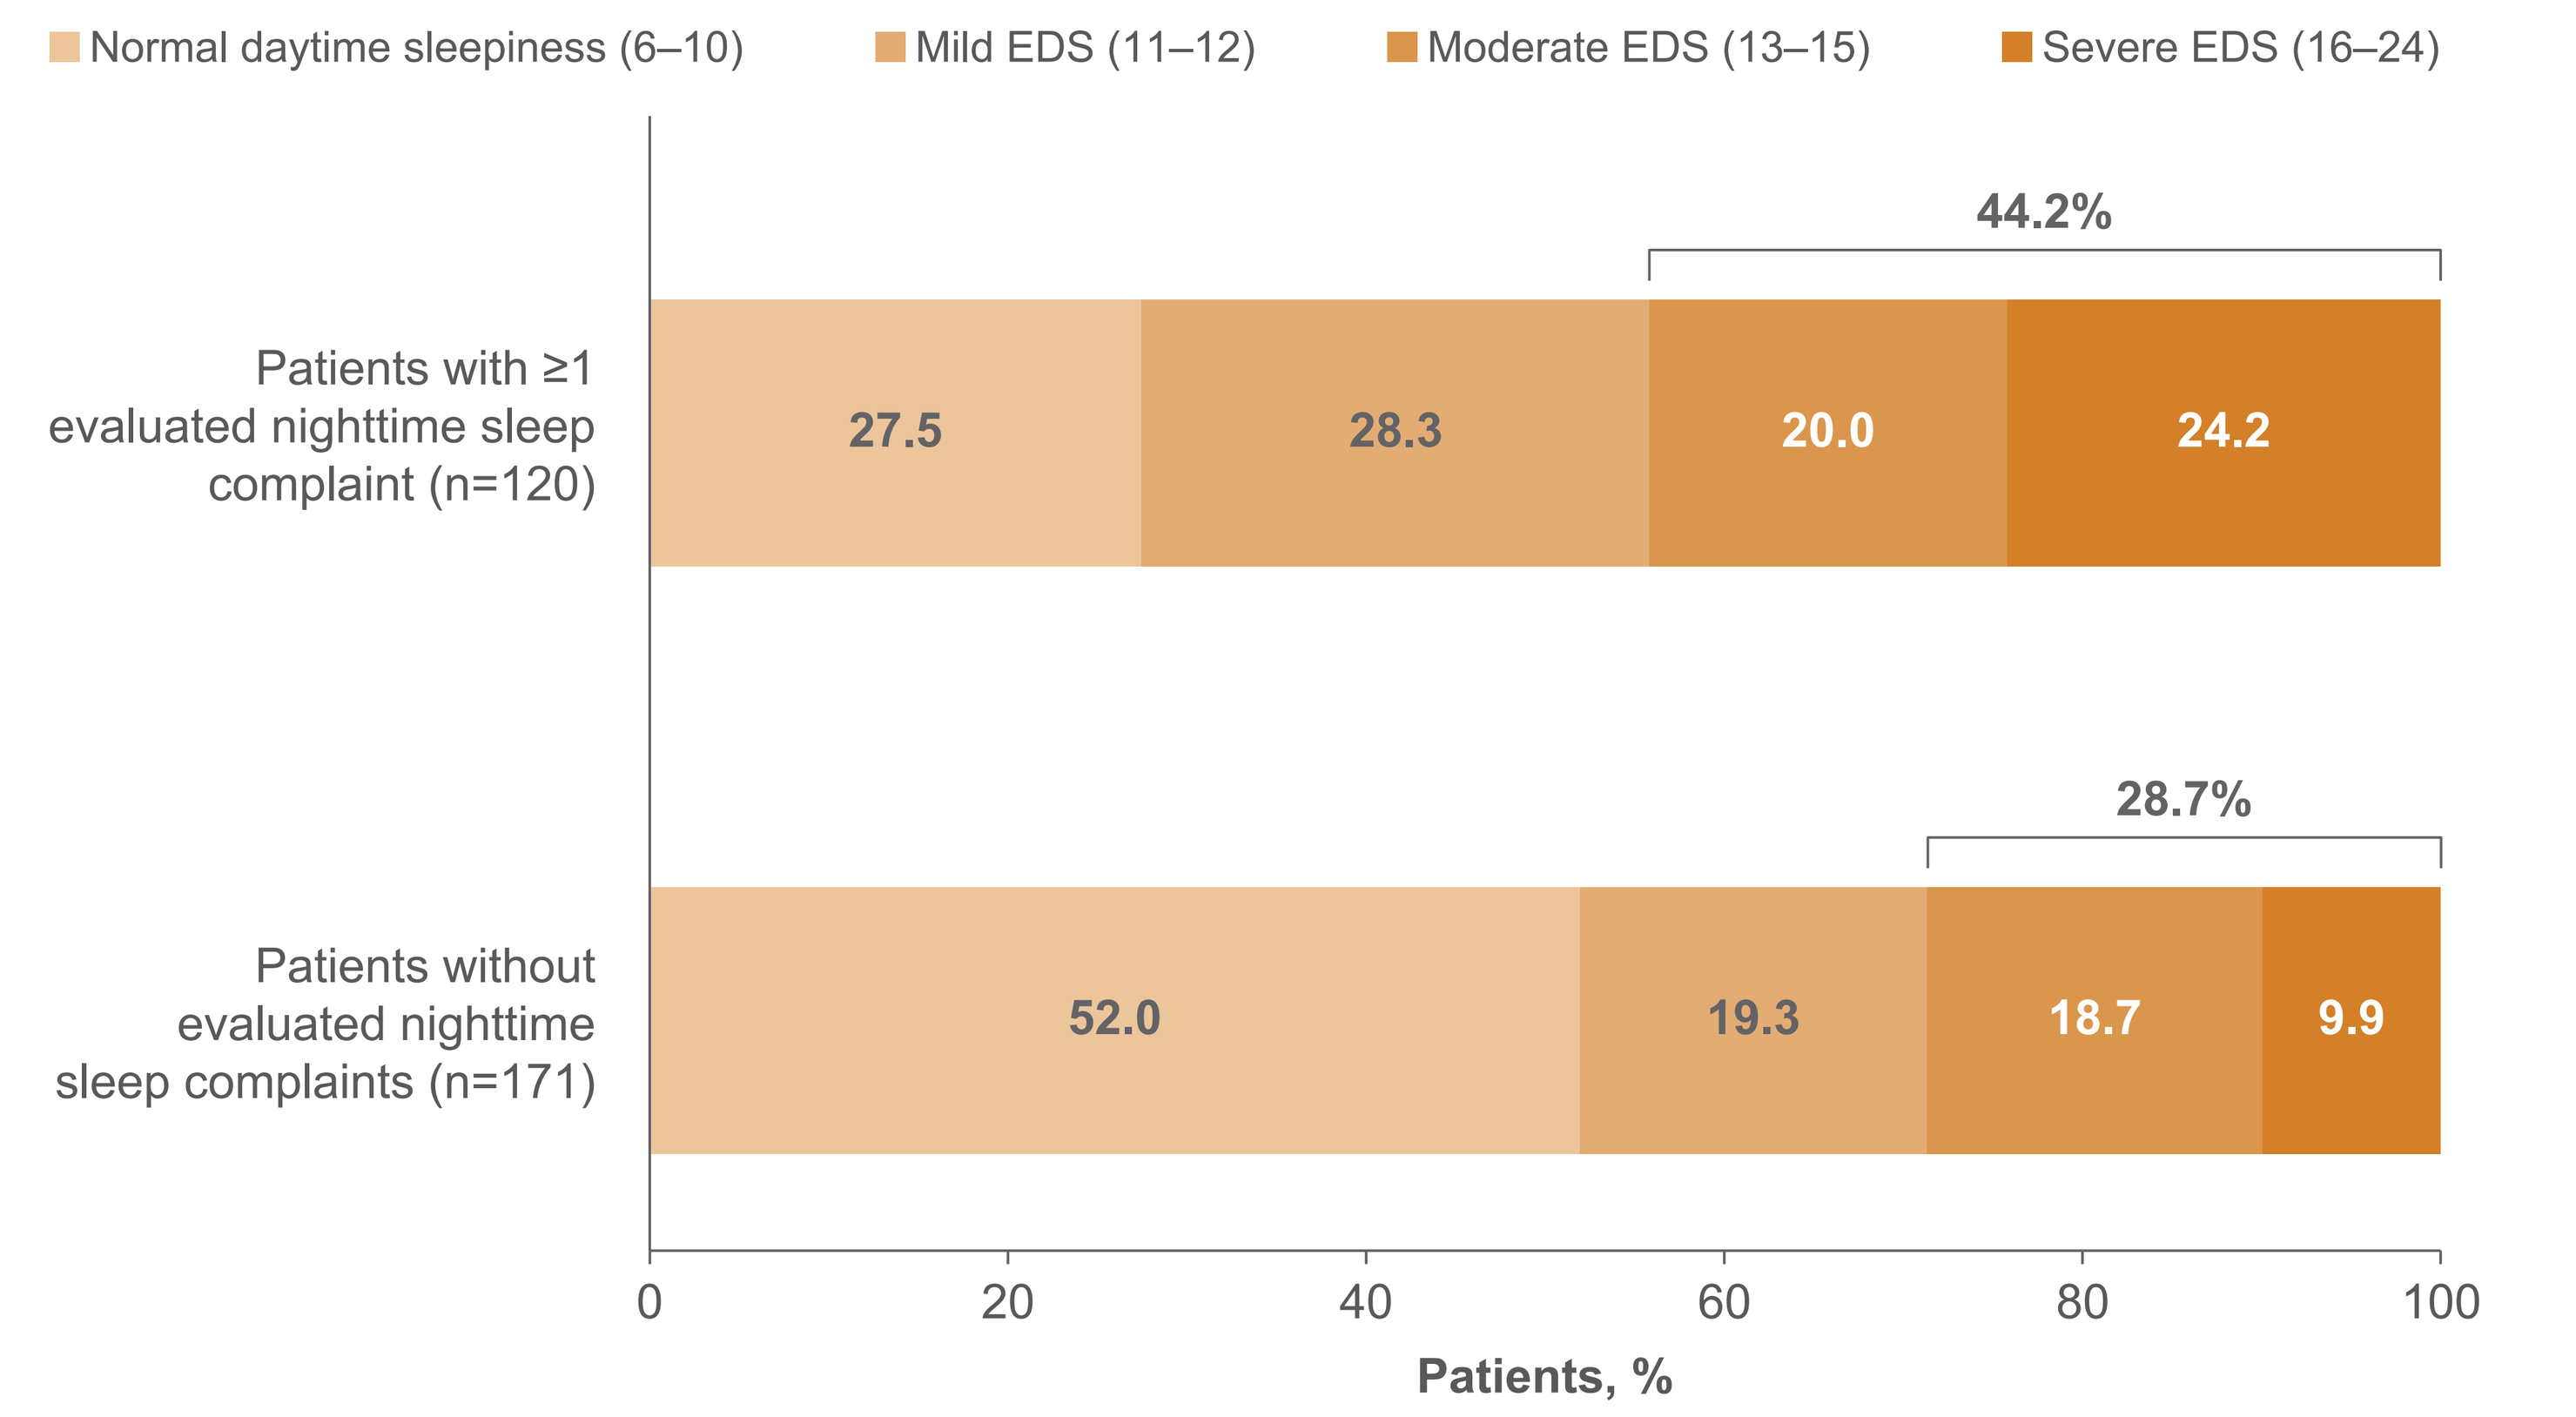

Normal daytime sleepiness (6–10)
Mild EDS (11–12)
Moderate EDS (13–15)
Severe EDS (16–24)
Patients with ≥1 evaluated nighttime sleep complaint (n=120)
27.5
28.3
20.0
24.2
44.2%
Patients without evaluated nighttime sleep complaints (n=171)
52.0
19.3
18.7
9.9
28.7%
0
20
40
60
80
100
Patients, %

[a]EDS as defined by HCP-reported ESS score; ESS scores were not available for all patients. A further 38 patients had ESS scores of 0 to 5, indicating lower normal daytime sleepiness; data not shown. Evaluated nighttime sleep complaints comprised frequent nighttime awakenings, shallow nighttime sleep, and early morning awakenings, as reported by HCPs. Proportions have been rounded.
EDS, excessive daytime sleepiness; ESS, Epworth Sleepiness Scale; HCP, healthcare provider.

- Numerically higher proportions of patients with vs without ≥1 evaluated nighttime sleep complaint had ESS scores indicating moderate or severe EDS (44.2% vs 28.7%)

Figure 4. Patients With vs Without ≥1 Evaluated Nighttime Sleep Complaint[a] Were Numerically More Likely to Have Cognitive Daytime Symptoms Recorded (HCP-Reported Data; n=637)

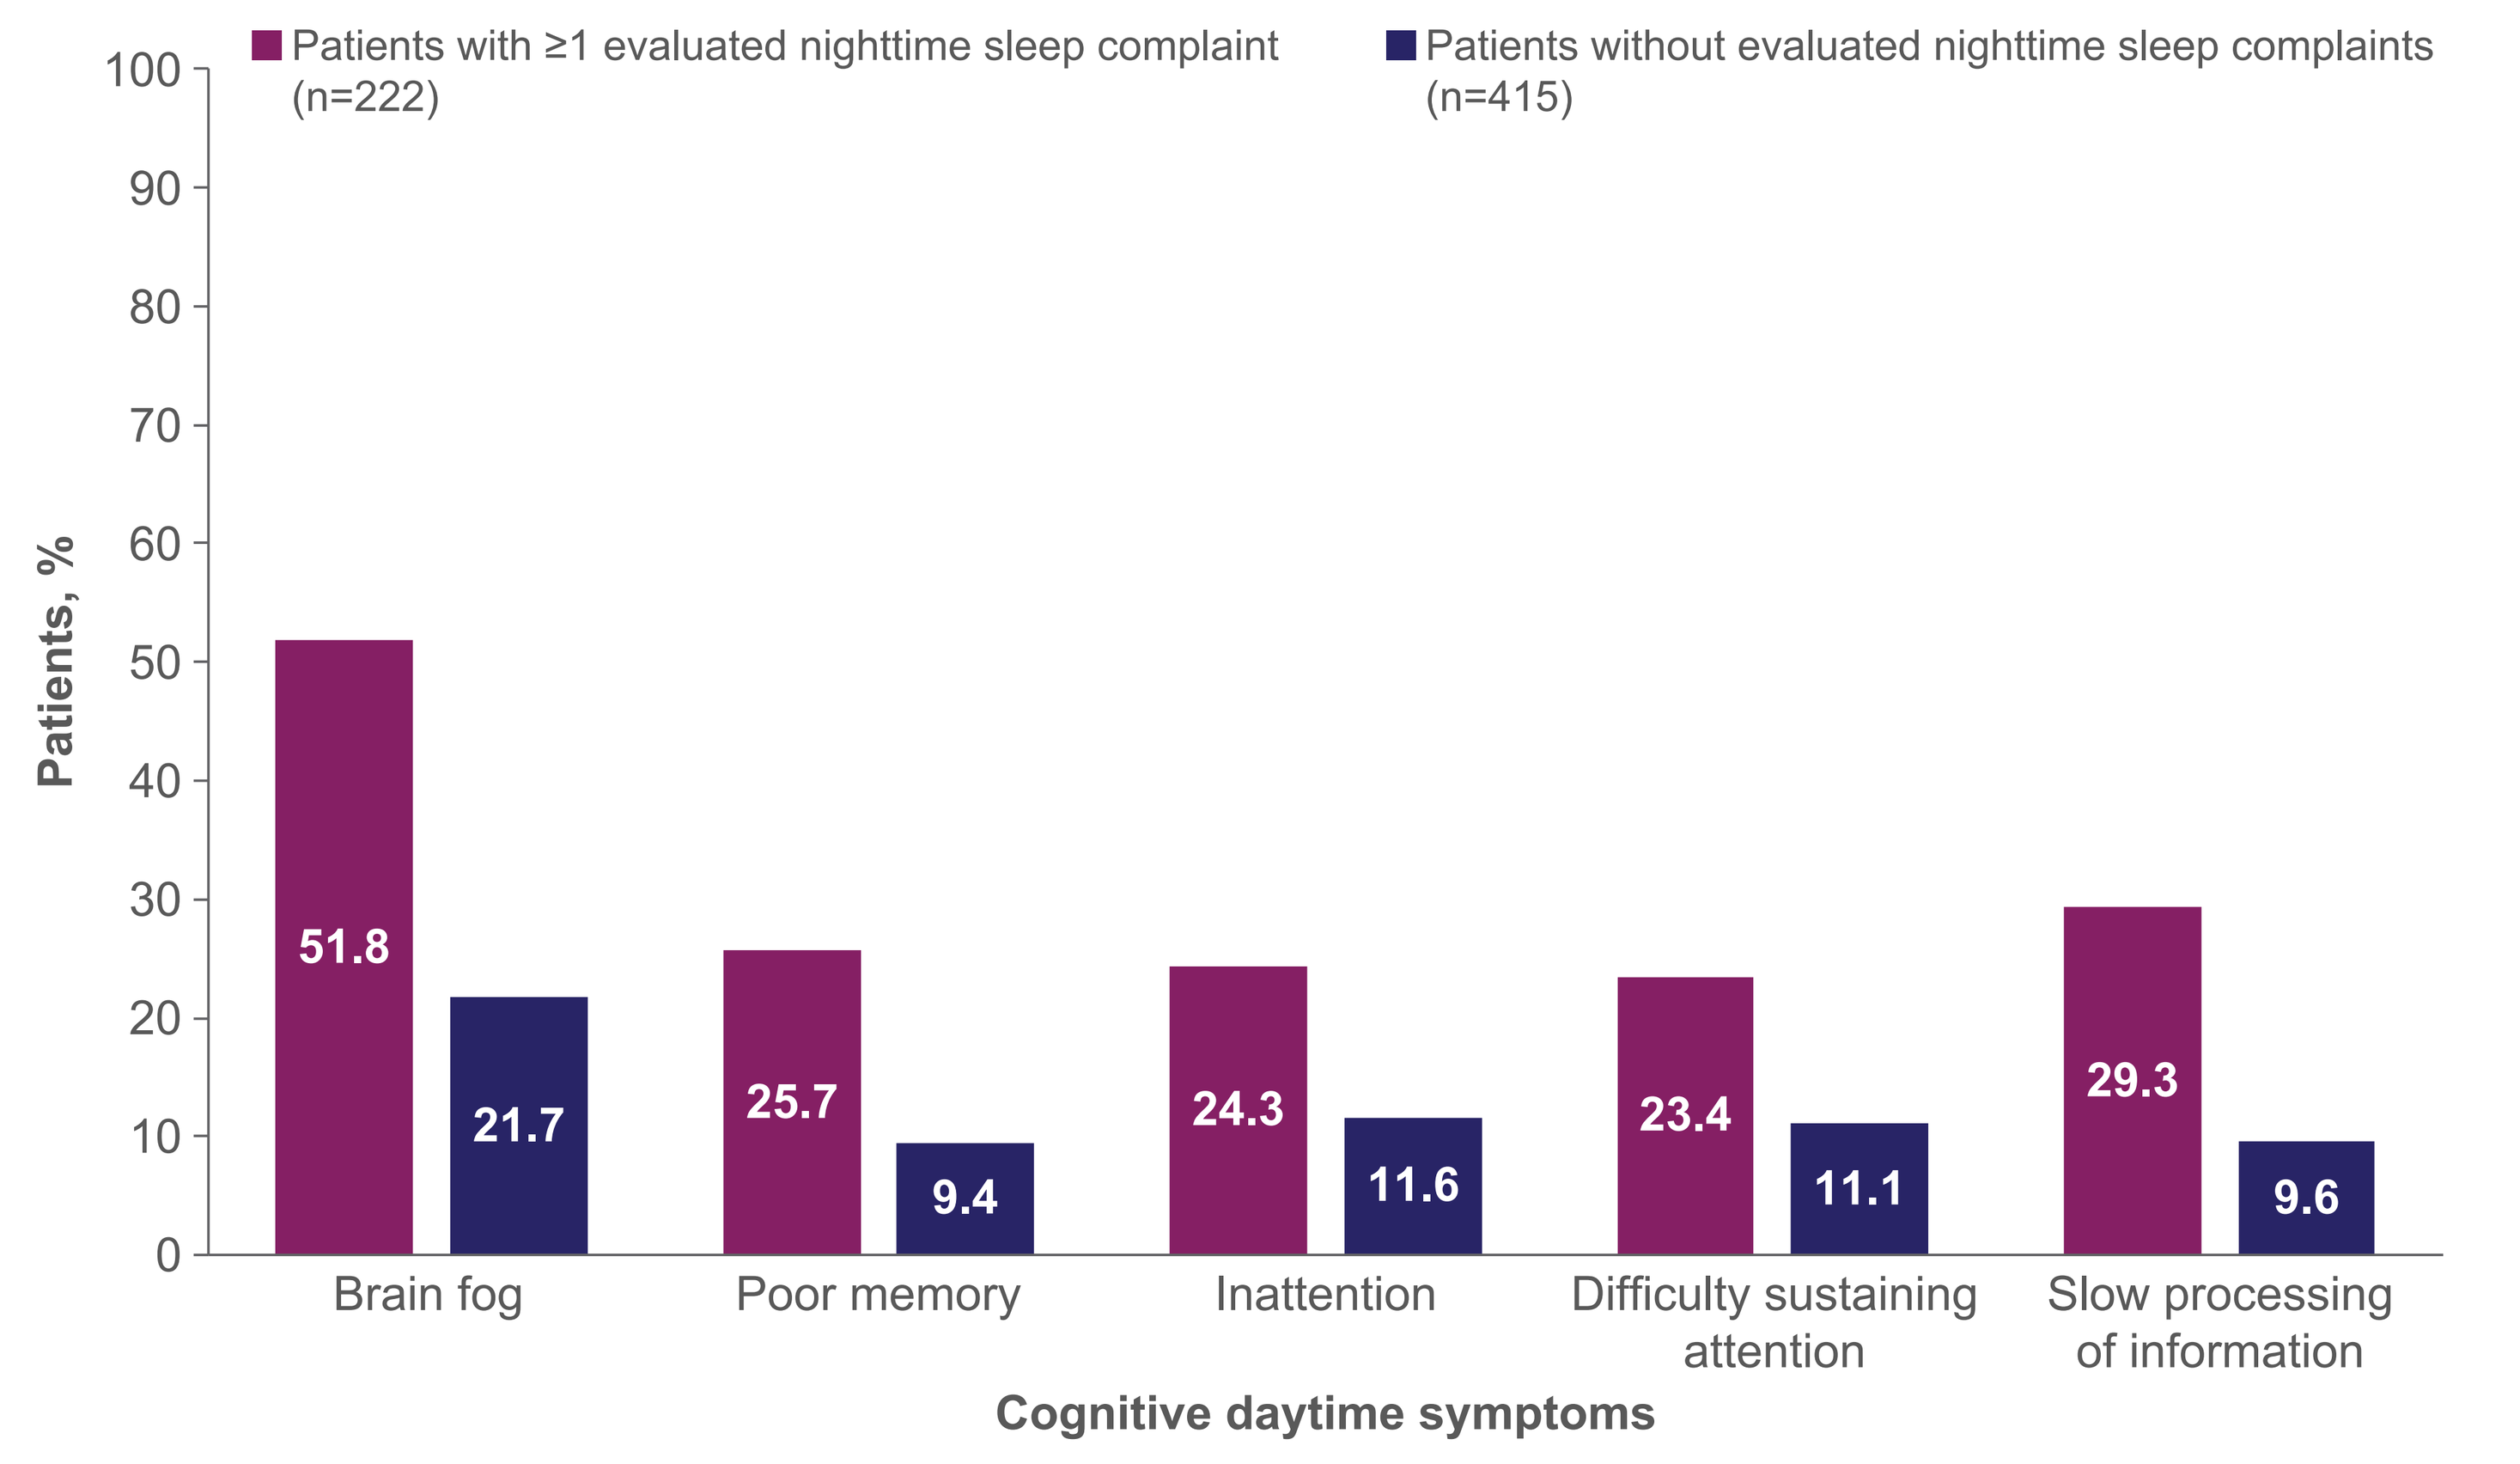

Patients with ≥1 evaluated nighttime sleep complaint (n=222)
Patients without evaluated nighttime sleep complaints (n=415)
Patients, %
100
90
80
70
60
50
40
30
20
10
0
51.8
21.7
25.7
9.4
24.3
11.6
23.4
11.1
29.3
9.6
Brain fog
Poor memory
Inattention
Difficulty sustaining attention
Slow processing of information
Cognitive daytime symptoms

[a]Evaluated nighttime sleep complaints comprised frequent nighttime awakenings, shallow nighttime sleep, and early morning awakenings, as reported by HCPs.
HCP, healthcare provider.

- Numerically greater proportions of patients with vs without ≥1 evaluated nighttime sleep complaint also had cognitive daytime symptoms recorded, including brain fog, poor memory, and inattention

Figure 5. Patients With vs Without ≥1 Evaluated Nighttime Sleep Complaint[a] Reported Numerically Lower Health Status and Greater Work Impairment (Patient Survey; n=184)

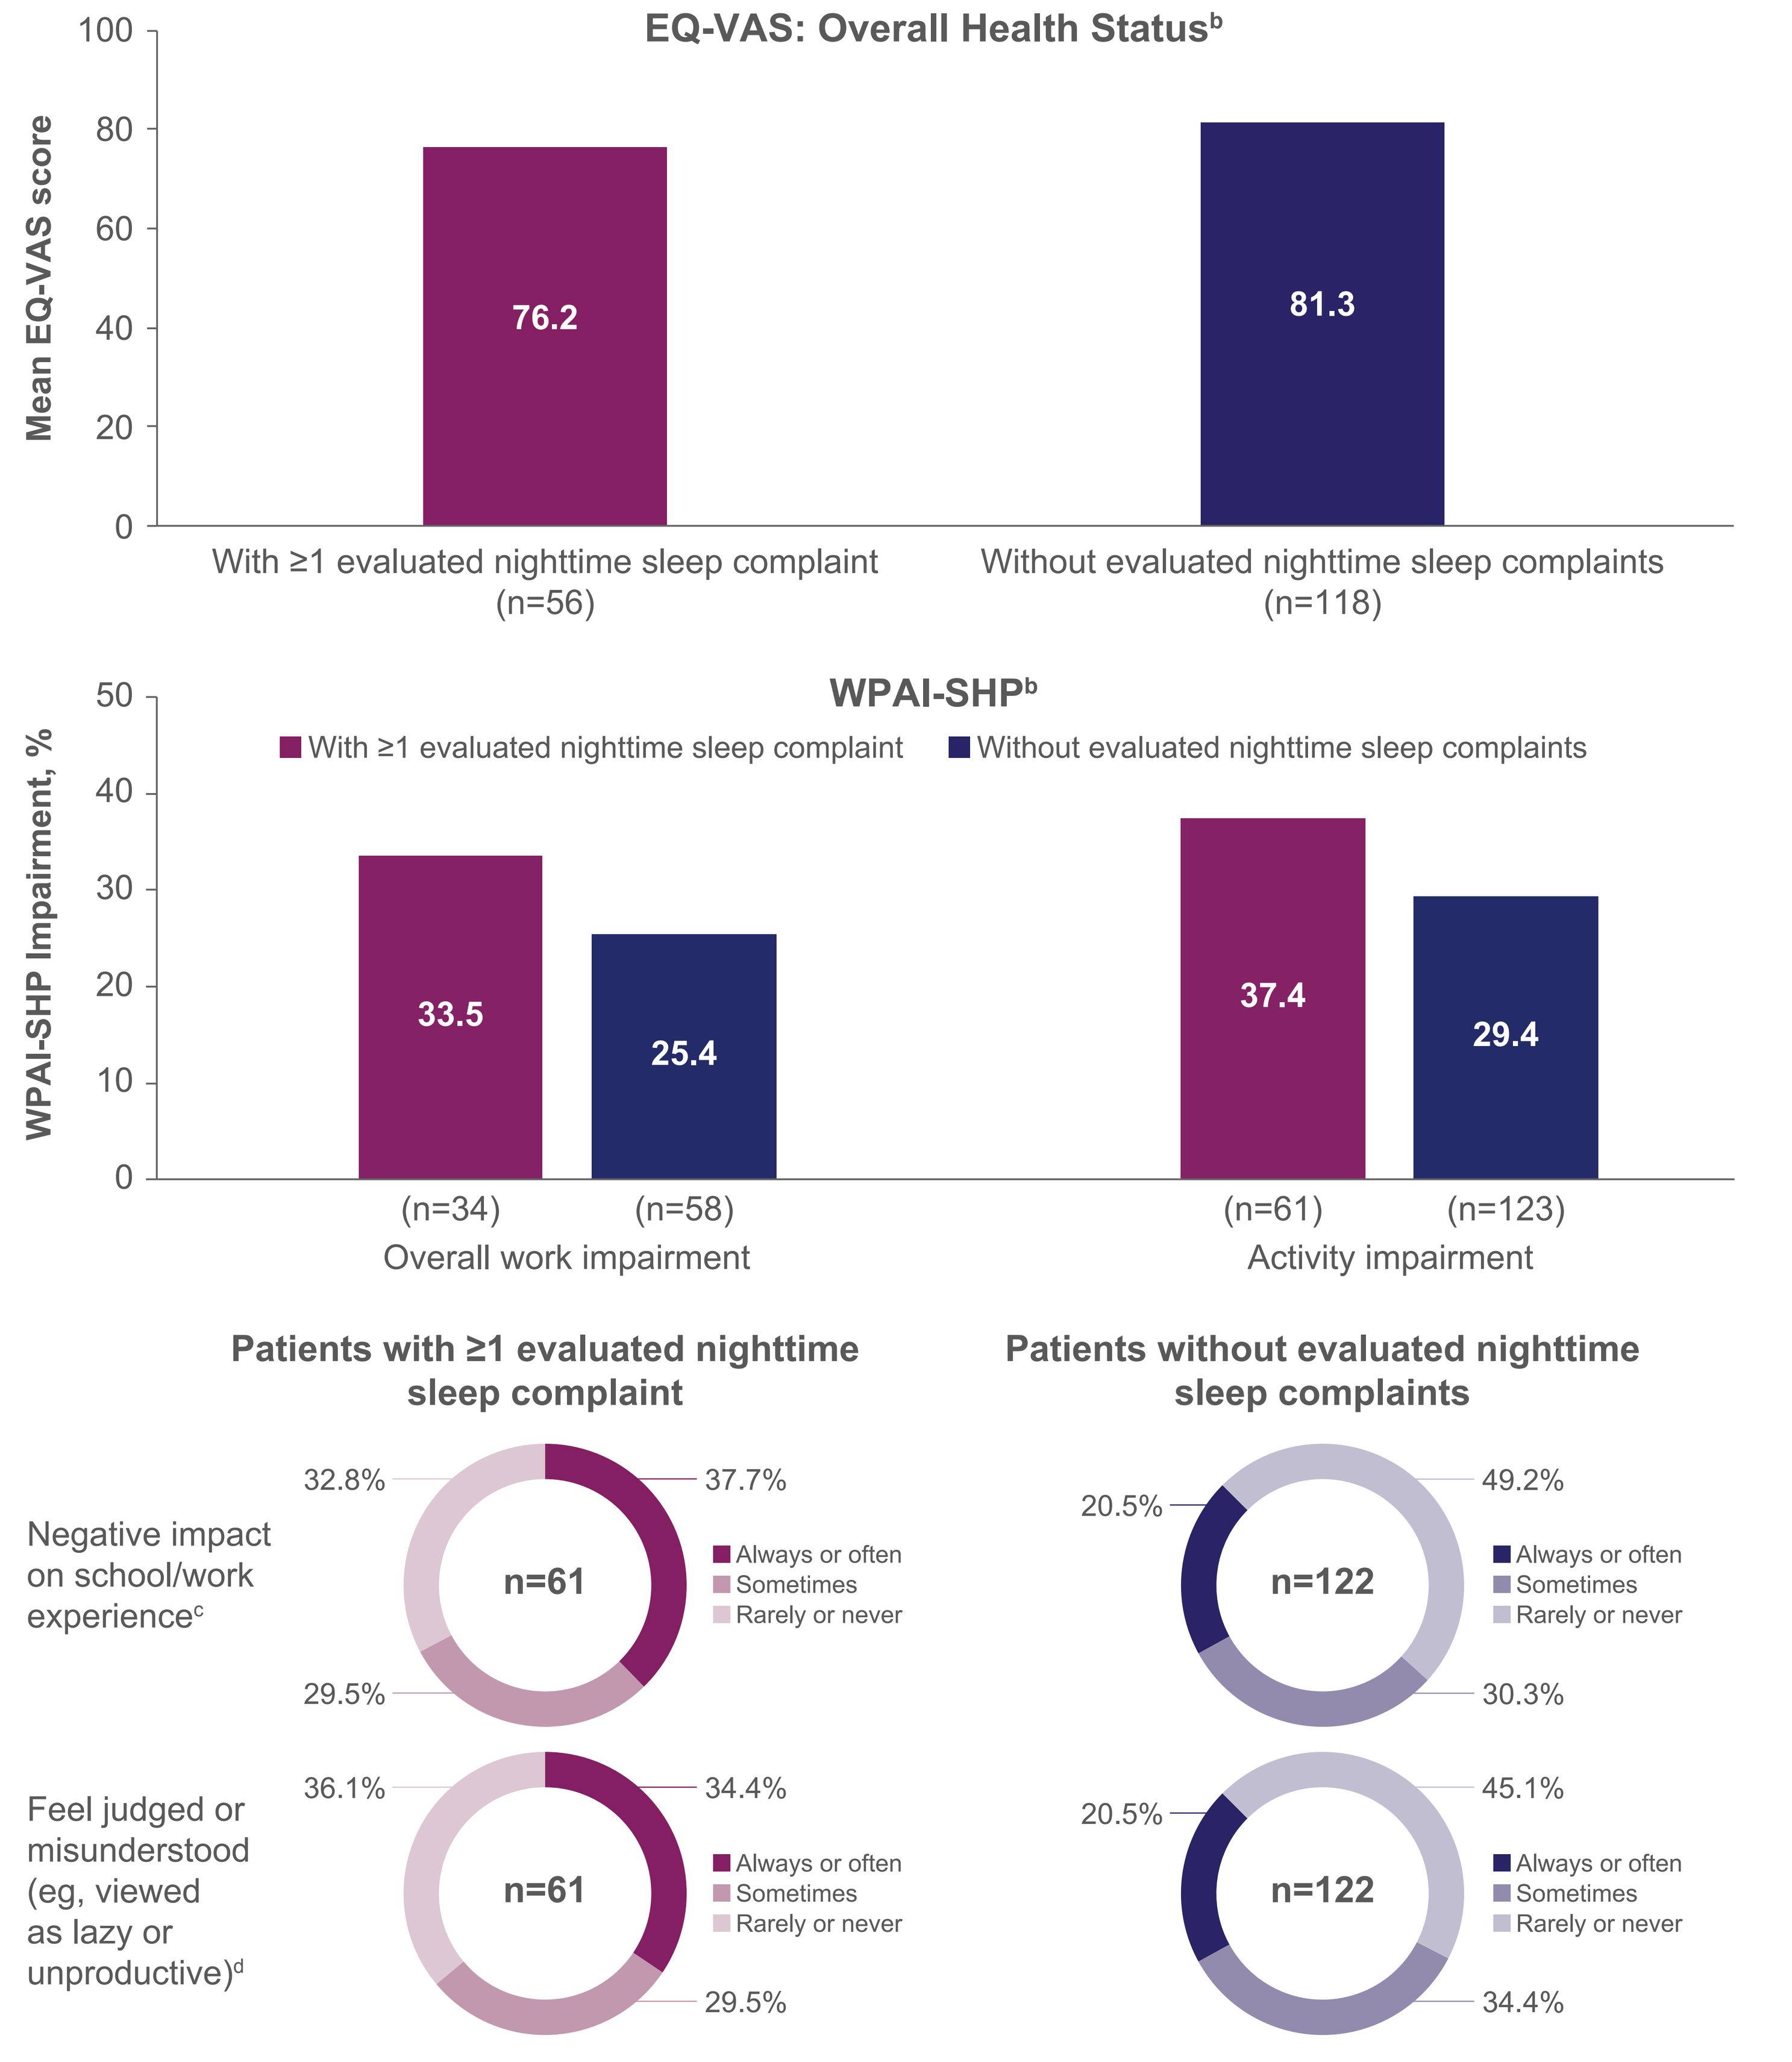

EQ-VAS: Overall Health Status[b]
Mean EQ-VAS score
100
80
60
40
20
0
76.2
81.3
With ≥1 evaluated nighttime sleep complaint (n=56)
Without evaluated nighttime sleep complaints (n=118)
WPAI-SHP[b]
With ≥1 evaluated nighttime sleep complaint
Without evaluated nighttime sleep complaints
WPAI-SHP Impairment, %
50
40
30
20
10
0
33.5
25.4
37.4
29.4
(n=34)
(n=58)
Overall work impairment
(n=61)
(n=123)
Activity impairment
Patients with ≥1 evaluated nighttime sleep complaint
Patients without evaluated nighttime sleep complaints
Negative impact on school/work experience[c]
32.8%
37.7%
29.5%
n=61
Always or often
Sometimes
Rarely or never
20.5%
49.2%
30.3%
n=122
Always or often
Sometimes
Rarely or never
Feel judged or misunderstood (eg, viewed as lazy or unproductive)[d]
36.1%
34.4%
29.5%
n=61
Always or often
Sometimes
Rarely or never
20.5%
45.1%
34.4%
n=122
Always or often
Sometimes
Rarely or never

[a]Evaluated nighttime sleep complaints comprised frequent nighttime awakenings, shallow nighttime sleep, and early morning awakenings, as reported by HCPs. [b]Higher EQ-VAS scores indicate better health-related quality of life; lower WPAI-SHP scores indicate less impairment. No minimal important difference has been established for the EQ-VAS or WPAI-SHP in narcolepsy. [c]Patients were asked, "How often do you feel that your narcolepsy negatively impacts your school/work experience?" [d]Patients were asked, "How often do you feel judged or misunderstood by others due to your narcolepsy (eg, being viewed as lazy or unproductive)?"
EQ-VAS, EuroQol Visual Analog Scale; WPAI-SHP, Work Productivity and Activity Impairment–Specific Health Problem.

- Patient-reported data were available for 184 patients, of whom 61 (33.2%) experienced ≥1 evaluated nighttime sleep complaint (as reported by HCPs)
- Numerically lower EQ-VAS scores were reported among patients with vs without ≥1 evaluated nighttime sleep complaint (76.2 vs 81.3), indicating lower health-related quality of life
- Patients with vs without ≥1 evaluated nighttime sleep complaint reported numerically greater overall work impairment (33.5% vs 25.4%) and activity impairment (37.4% vs 29.4%) via the WPAI-SHP
- Patients with vs without ≥1 evaluated nighttime sleep complaint were numerically more likely to report "always" or "often" experiencing a negative impact of narcolepsy on work or school (37.7% vs 20.5%), as well as feeling judged or misunderstood (34.4% vs 20.5%) due to narcolepsy