560

IMforte: Quality-Adjusted Time Without Symptoms or Toxicity (Q-TWiST) Analysis of First-Line Maintenance Treatment With Lurbinectedin + Atezolizumab vs Atezolizumab in Extensive-Stage Small Cell Lung Cancer

Hossein Borghaei[1], Luis Paz-Ares[2], Martin Reck[3], Roy S Herbst[4], Solange Peters[5], Kamalnayan Bhatt[6], Xiaoyan Wang[7], Jonathon Gable[7], Siobhan Connor-Ahmad[8], Carla Mamolo[9], Ya-Chen Lin[9], Stephen V Liu[10]

[1]Department of Hematology and Oncology, Fox Chase Cancer Center, Philadelphia, PA, USA; [2]Department of Medical Oncology, Hospital Universitario 12 de Octubre, H12O-CNIO Lung Cancer Unit, Universidad Complutense and Ciberonc, Madrid, Spain; [3]Department of Thoracic Oncology, Lung Clinic Grosshansdorf, Airway Research Center North, German Center of Lung Research, Grosshansdorf, Germany; [4]Department of Medical Oncology and Hematology, Yale School of Medicine, New Haven, CT, USA; [5]Department of Oncology, Centre Hospitalier Universitaire Vaudois, Lausanne, Switzerland; [6]Jazz Pharmaceuticals, Philadelphia, PA, USA; [7]Jazz Pharmaceuticals, Palo Alto, CA, USA; [8]Roche Products Ltd, Welwyn, UK; [9]Genentech, a Member of the Roche Group, South San Francisco, CA, USA; [10]Department of Medical Oncology, Lombardi Comprehensive Cancer Center, Georgetown University, Washington, DC, USA

Objective

- Here, we describe a post hoc Q-TWiST analysis using IMforte data to further elucidate the benefit-risk profile of lurbinectedin + atezolizumab in first-line (1L) maintenance therapy for patients with extensive-stage small cell lung cancer (ES-SCLC)

Conclusions

- Patients receiving lurbinectedin + atezolizumab spent an average of 3.4 more months without symptoms or toxicity, which represented a higher proportion of their survival time (9.8/14.8 months; 66%), compared to patients receiving atezolizumab alone (6.4/12.9 months; 50%)
- This analysis showed superior quality-adjusted survival for lurbinectedin + atezolizumab as a 1L maintenance treatment for ES-SCLC compared to atezolizumab alone
 - Patients treated with lurbinectedin + atezolizumab showed a clinically important 14.9% gain in quality-adjusted survival relative to those treated with atezolizumab, further supporting a favorable benefit-risk profile of lurbinectedin + atezolizumab as 1L maintenance treatment for ES-SCLC
- Higher utility weights in time without symptoms or toxicity (TWiST) and toxicity (TOX) health states than the relapsed (REL) state underscore the importance of delaying disease progression with 1L maintenance therapy to improve quality-of-life (QoL) outcomes in patients with ES-SCLC

References: 1. PDQ® Adult Treatment Editorial Board. PDQ small cell lung cancer treatment. National Cancer Institute. Updated May 14, 2025. Accessed September 10, 2025. https://www.cancer.gov/types/lung/hp/small-cell-lung-treatment-pdq. **2.** Paz-Ares L, et al. *Lancet*. 2025;405:2129-43. **3.** Herbst RS, et al. Poster presented at: IASLC/ASCO 2025 North America Conference on Lung Cancer; December 5-7, 2025; Chicago, IL; P01.144. **4.** Jazz Pharmaceuticals, Inc. ZEPZELCA® (lurbinectedin) for injection, for intravenous use. Updated October 2025. Accessed October 30, 2025. https://pp.jazzpharma.com/pi/zepzelca.en.USPI.pdf. **5.** Swissmedic Swiss Public Assessment Report. Extension of therapeutic indication Zepzelca. Updated January 27, 2026. Accessed March 3, 2026. https://www.swissmedic.ch/dam/swissmedic/de/dokumente/zulassung/swisspar/67729-zepzelca-02-swisspar-20260127.pdf.download.pdf/SwissPAR%20Zepzelca.pdf. **6.** Referenced with permission from the NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for small cell lung cancer. V2.2026. © National Comprehensive Cancer Network, Inc. 2026. All rights reserved. Accessed January 15, 2026. To view the most recent and complete version of the guideline, go online to NCCN.org. **7.** Bogart E, et al. *Comput Methods Programs Biomed*. 2016;125:79-87. **8.** Revicki DA, et al. *Qual Life Res*. 2006;15:411-23. **9.** Wan Y, et al. *Value Health*. 2016;19:A385.

Support and Acknowledgements: This study was funded by F. Hoffmann-La Roche and Jazz Pharmaceuticals. Under direction of the authors, Adam Schroer, MS, PhD, of Red Nucleus, provided medical writing and editorial support, which were funded by Jazz Pharmaceuticals in accordance with Good Publication Practice (GPP 2022) guidelines (https://www.ismpp.org/gpp-2022). Jazz Pharmaceuticals and F. Hoffmann-La Roche also reviewed and edited the poster for scientific accuracy. NCCN makes no warranties of any kind whatsoever regarding their content, use or application, and disclaims any responsibility for their application or use in any way. The authors would like to thank all patients and their families, investigators, clinical trial researchers, personnel, and staff who contributed to or participated in the trial.

Disclosures: Dr Hossein Borghaei reported receiving grants or contracts (to their institution) from Amgen, Bristol Myers Squibb, and Eli Lilly; consulting or advisory fees from AbbVie, Amgen, AstraZeneca, Axiom, Bayer, BeiGene, BerGenBio, BioNTech, Bristol Myers Squibb, Boehringer Ingelheim, Daiichi Sankyo, Eli Lilly, EMD Serono, Genentech, Genmab, Gilead, Grid Therapeutics, Guardant, Iobiotech, iTEO, Janssen, Jazz Pharmaceuticals, Merck, Mirati, Natera, Novartis, Novocure, Oncocyte, Pfizer, PharmaMar, Puma, RAPT, Regeneron, Takeda, and Summit; receiving honoraria from Amgen, Daiichi Sankyo, Janssen, Jazz Pharmaceuticals, Pfizer, and Regeneron; receiving support for attending meetings and/or travel from Amgen, Bristol Myers Squibb, Eli Lilly, EMD Serono, Genentech, Jazz Pharmaceuticals, Merck, Mirati, and Regeneron; participating in a data safety monitoring board or advisory board for Incyte, Novartis, Takeda, University of Pennsylvania CAR T Program, and Springworks; holding stock options from Inspirna, Nucleai, and Sonnetbio; and receiving medical writing services funded by Amgen, AstraZeneca, Bristol Myers Squibb, F. Hoffmann-La Roche, Jazz Pharmaceuticals, Merck, and Mirati.

Corresponding author: Dr Hossein Borghaei
hossein.borghaei@fccc.edu

Poster presented at the 2026 American Society of Clinical Oncology Annual Meeting; May 29–June 2, 2026; Chicago, IL, and online.

Copies of this presentation obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission from the authors.

Introduction

- ES-SCLC is an aggressive and highly lethal disease, with median survival of 6 to 12 months in clinical practice[1]
- IMforte (NCT05091567) is the first global, phase 3 maintenance study to demonstrate statistically significant and clinically meaningful improvements in progression-free survival (PFS; stratified hazard ratio [HR], 0.54) and overall survival (OS; stratified HR, 0.73) in patients with ES-SCLC receiving 1L maintenance treatment with lurbinectedin + atezolizumab vs atezolizumab alone[2]
- The combination was generally well tolerated despite increased incidence of adverse events (AEs) compared with atezolizumab alone[2]
 - Patients did not report increased treatment burden with the combination, and treatment discontinuation rates were low[2,3]
- 1L maintenance treatment with lurbinectedin + atezolizumab is now approved in multiple countries, including the US and Switzerland, for adults with ES-SCLC whose disease has not progressed after 1L induction therapy with atezolizumab, carboplatin, and etoposide and is a preferred treatment option in the National Comprehensive Cancer Network® (NCCN®) Clinical Practice Guidelines in Oncology®[4,5,6]
- **We conducted a Q-TWiST analysis to evaluate whether the OS benefit with the combination regimen in the IMforte study was associated with superior quality-adjusted survival**
 - The Q-TWiST method combines measures of efficacy and safety into a single output (quality-adjusted survival) to compare treatment groups in a clinical trial[7,8]

Methods

- Patients without progressive disease after 1L induction treatment with atezolizumab, carboplatin, and etoposide were randomized 1:1 to 1L maintenance treatment every 3 weeks with atezolizumab 1200 mg intravenously (IV) ± lurbinectedin 3.2 mg/m^2 IV until disease progression or unacceptable toxicity (**Figure 1**)
- Survival time was partitioned into 3 health states per standard Q-TWiST methodology (**Figure 1**)[7,8]
 - **TOX**: time after randomization with **unresolved grade ≥3 AEs** (Common Terminology Criteria for Adverse Events [CTCAE]) before disease progression; patients who did not die or experience disease progression were censored at their PFS-censoring date
 - A sensitivity analysis used time with unresolved grade ≥2 AEs as the TOX state
 - **TWiST**: time after randomization without TOX before disease progression (**PFS − TOX**); patients who did not die or experience disease progression were censored at their PFS-censoring date
 - **REL**: time from disease progression to death (**OS − PFS**); surviving patients were censored at the time of last contact
- The mean time spent in each health state was estimated using restricted mean survival times, calculated as area under the Kaplan-Meier curve
- Utility weights were derived from patient-reported QoL data via the EQ-5D-5L questionnaires, which was collected on day 1 of cycles 1 to 6 and every other cycle thereafter as well as at the treatment-discontinuation visit and 3- and 6-month post-treatment follow-up
- Q-TWiST was the sum of the mean time in each health state adjusted by their respective utility weights
 - Q-TWiST = (TOX time × TOX utility weight) + (TWiST time × TWiST utility weight) + (REL time × REL utility weight)
- A threshold analysis was performed to evaluate the robustness of the benefits with respect to variability in QoL, ranging from 0 to 1 for TOX and REL utility weights, while maintaining TWiST utility weight at 1
- Nonparametric bootstrapping with replacement and 1000 replications was used to generate standard errors and construct 95% CIs[7]

Figure 1. IMforte study design[2] and Q-TWiST analysis

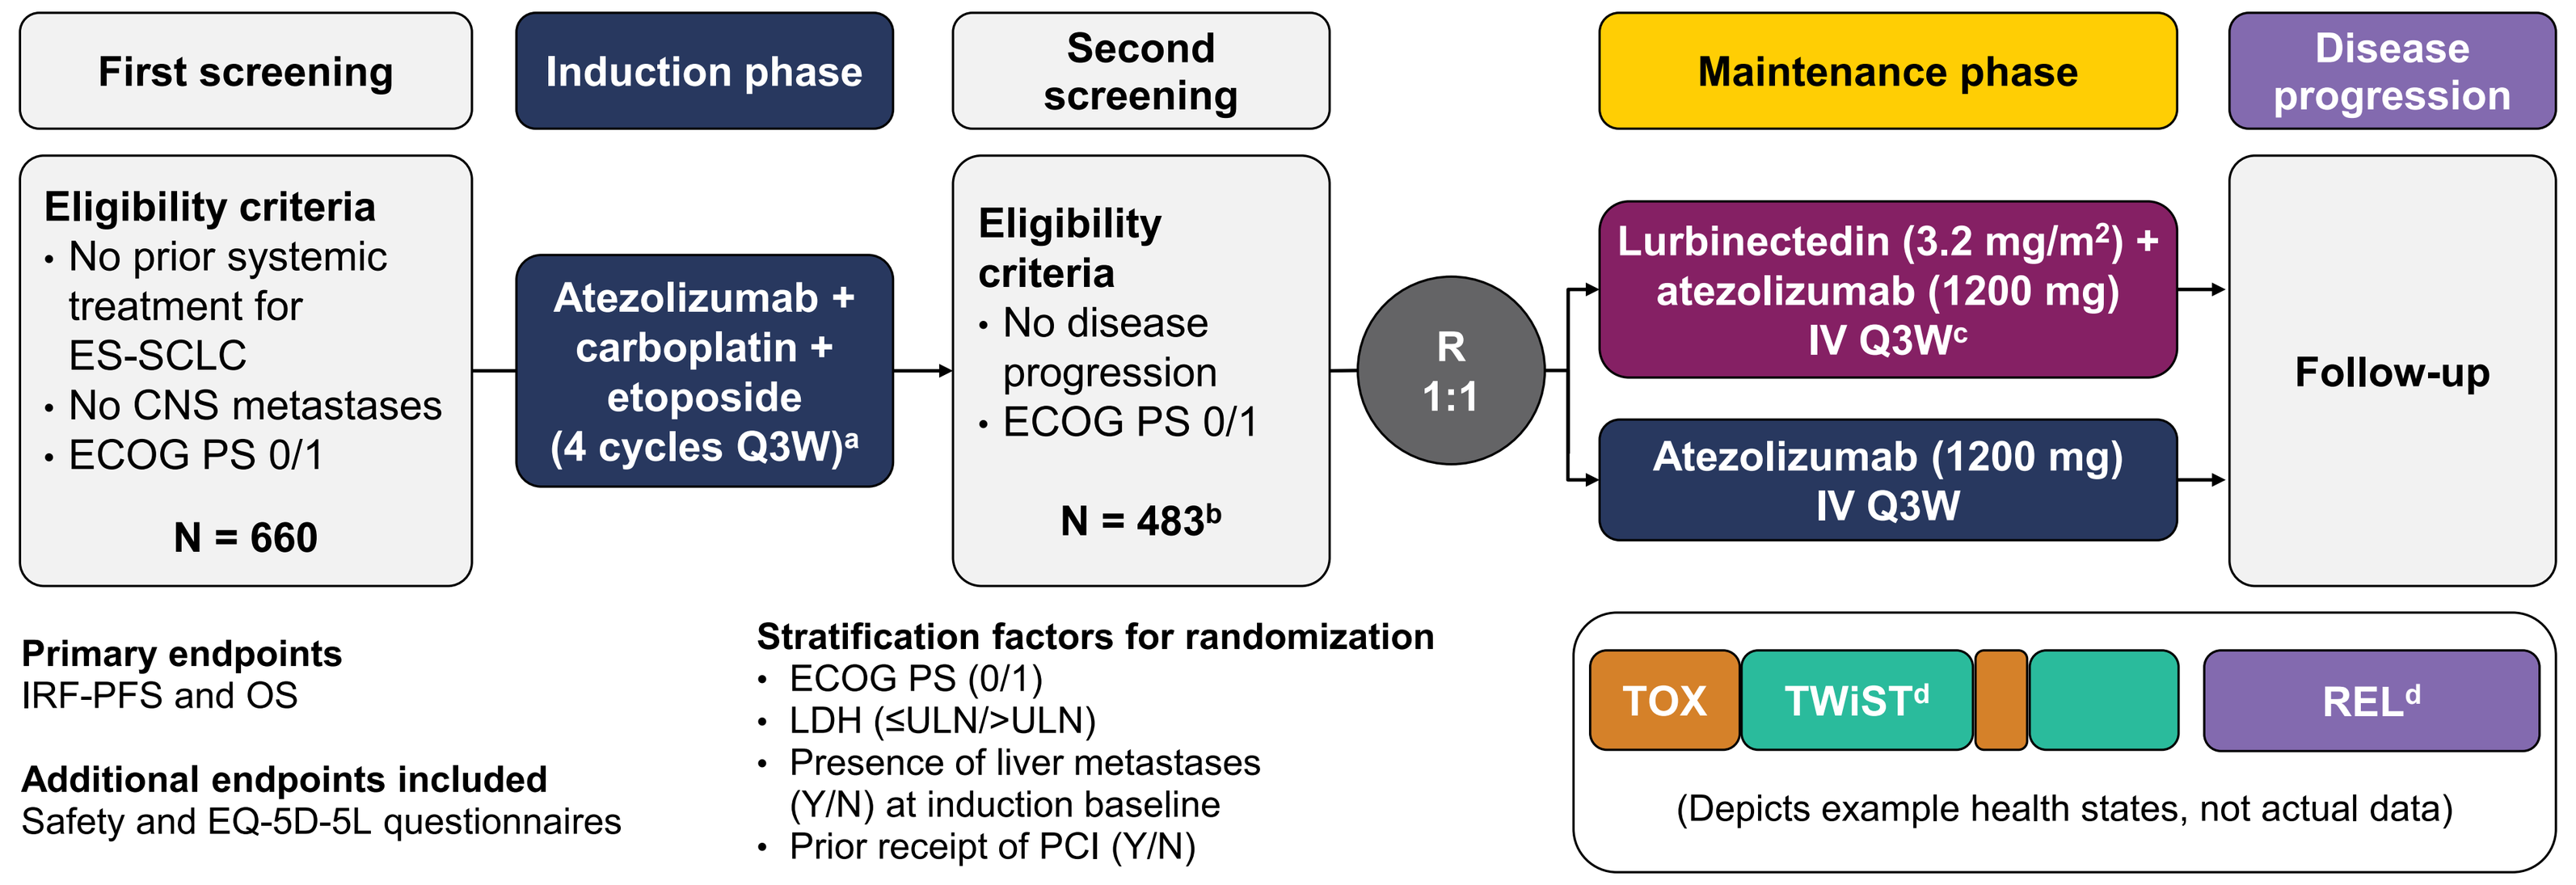

First screening
Induction phase
Second screening
Maintenance phase
Disease progression
Eligibility criteria
• No prior systemic treatment for ES-SCLC
• No CNS metastases
• ECOG PS 0/1
N = 660
Atezolizumab + carboplatin + etoposide (4 cycles Q3W)a
Eligibility criteria
• No disease progression
• ECOG PS 0/1
N = 483b
R 1:1
Lurbinectedin (3.2 mg/m2) + atezolizumab (1200 mg) IV Q3Wc
Atezolizumab (1200 mg) IV Q3W
Follow-up
Primary endpoints
IRF-PFS and OS
Additional endpoints included
Safety and EQ-5D-5L questionnaires
Stratification factors for randomization
• ECOG PS (0/1)
• LDH (≤ULN/>ULN)
• Presence of liver metastases (Y/N) at induction baseline
• Prior receipt of PCI (Y/N)
TOX
TWiSTd
RELd
(Depicts example health states, not actual data)

[a]Administered per standard dose. [b]73% of patients continued from induction to maintenance. [c]With prophylactic granulocyte colony-stimulating factor and antiemetics. [d]Patients who were alive by the clinical cutoff date or who were lost to follow-up were censored at the time of the last tumor assessment or last contact.
CNS, central nervous system; ECOG PS, Eastern Cooperative Oncology Group Performance Status; ES-SCLC, extensive-stage small cell lung cancer; IRF, independent review facility; IV, intravenously; LDH, lactate dehydrogenase; OS, overall survival; PCI, prophylactic cranial irradiation; PFS, progression-free survival; Q3W, every 3 weeks; R, randomization; REL, relapse; TOX, toxicity; TWiST, time without symptoms or toxicity; ULN, upper limit of normal; Y/N, yes/no.

Results

Table 1. QoL EQ-5D-5L utility weights for each health state by treatment

Health state	Utility weight, mean (95% CI)[a] Lurbi + atezo n = 242	Atezo n = 241
TOX	0.83 (0.80, 0.87)	0.83 (0.77, 0.89)
TWiST	0.84 (0.82, 0.86)	0.83 (0.80, 0.85)
REL	0.78 (0.74, 0.82)	0.76 (0.73, 0.80)

[a]Based on regression coefficients of a linear mixed model with health state (TOX, TWiST, REL), planned treatment, and health state × planned treatment interaction as fixed effects and intercept and slope as random effects. An autoregressive covariance matrix was used to model the correlation within patients.
atezo, atezolizumab; lurbi, lurbinectedin; QoL, quality of life; REL, relapse; TOX, toxicity; TWiST, time without symptoms or toxicity.

- At the data cutoff date of July 29, 2024, the median follow-up was 15.0 months
- In the lurbinectedin + atezolizumab arm, 174 (72%) patients had disease progression or died, and in the atezolizumab arm, 202 (84%) had disease progression or died
- QoL utility weights for each health state were similar across arms and were numerically lower in the REL health state compared with the TWiST and TOX health states

Figure 2. Partitioned survival curves presenting the relative time and proportion of patients in each health state

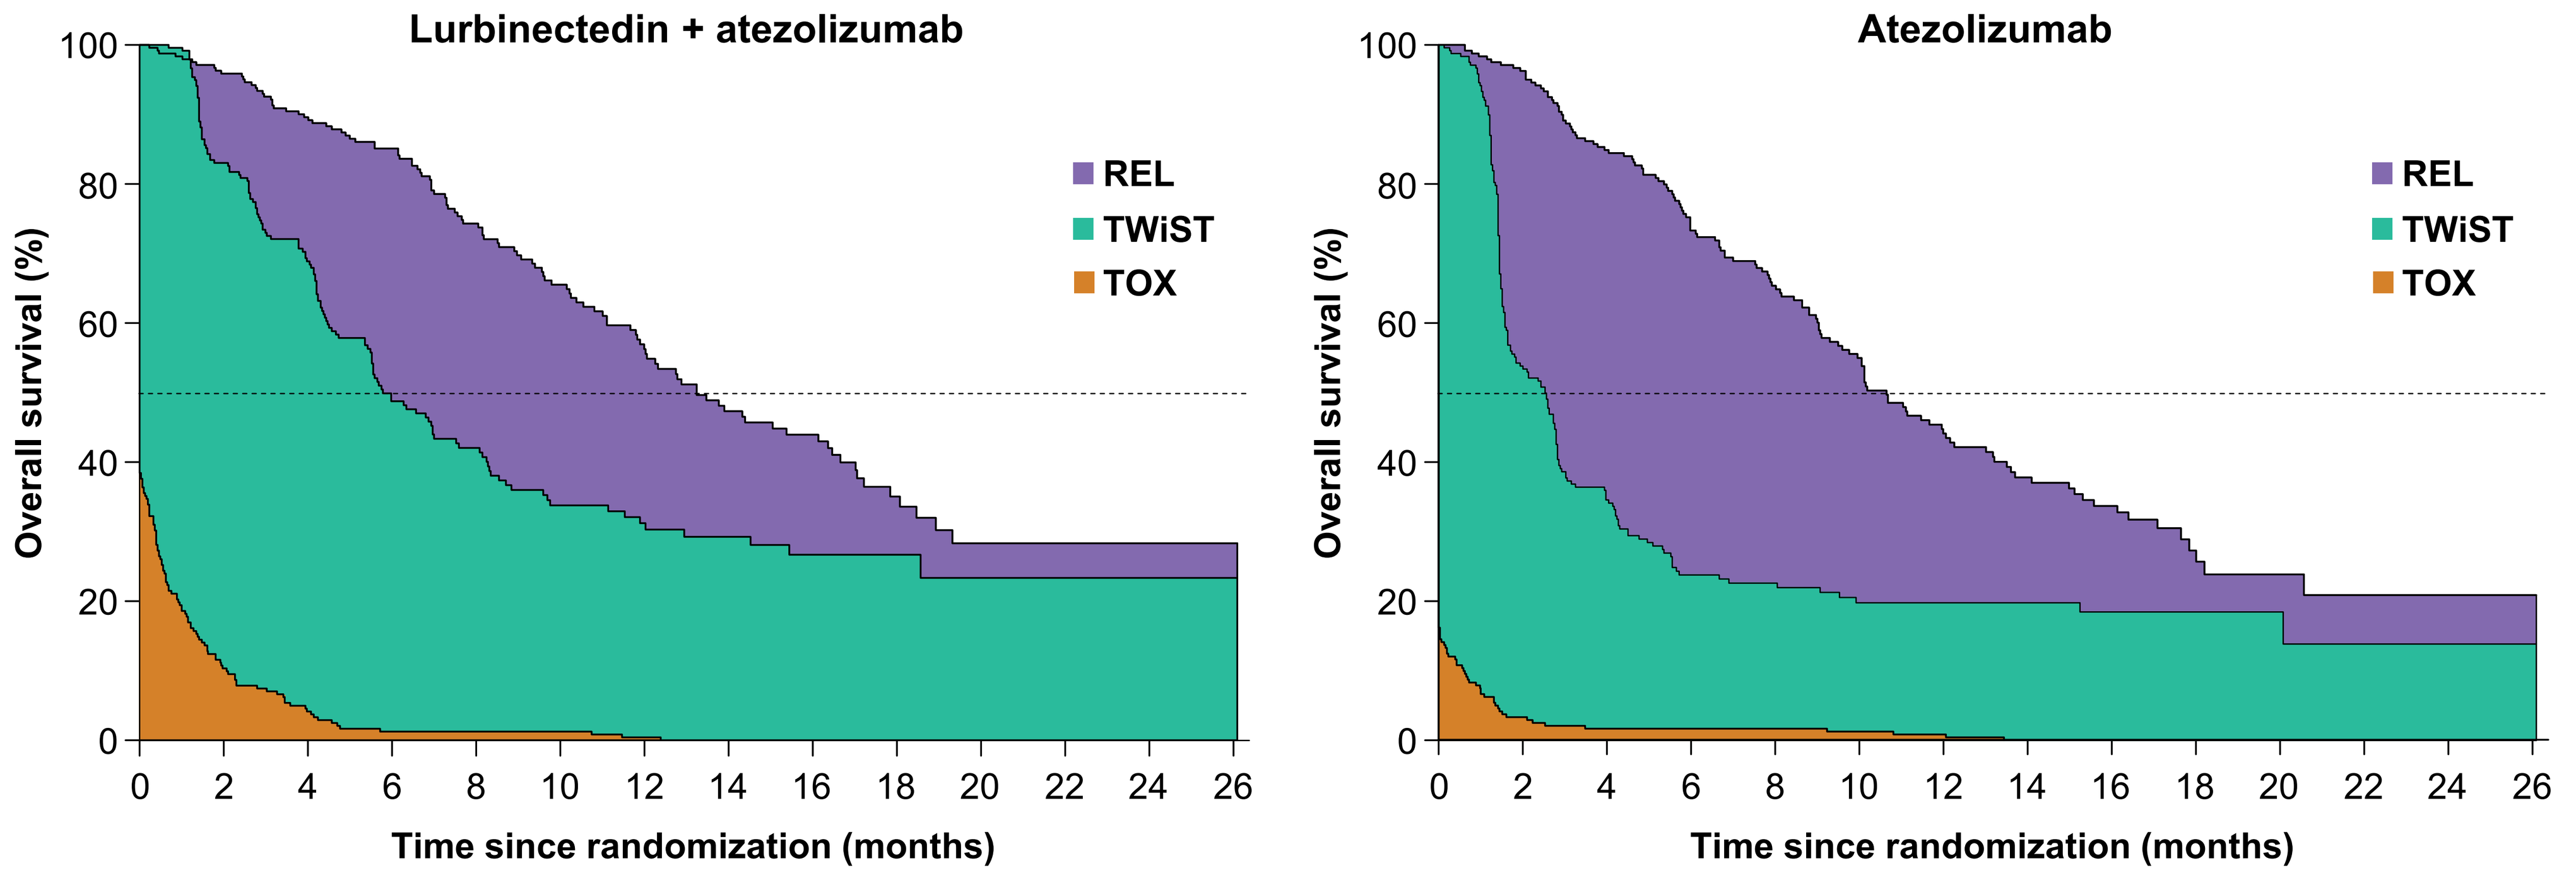

Lurbinectedin + atezolizumab
Atezolizumab
REL
TWiST
TOX
Overall survival (%)
0
20
40
60
80
100
0 2 4 6 8 10 12 14 16 18 20 22 24 26
Time since randomization (months)

REL, relapse; TOX, toxicity; TWiST, time without symptoms or toxicity.

Table 2. Mean duration of PFS, OS, each health state, and Q-TWiST

Month, mean (95% CI)	Lurbi + atezo n = 242	Atezo n = 241	Difference	Relative gain[a]
PFS[b]	10.5 (9.1, 11.9)	6.8 (5.4, 8.1)	3.7 (1.8, 5.6)	
OS[b]	14.8 (13.5, 16.1)	12.9 (11.6, 14.2)	2.0 (0.1, 3.8)	
TWiST	9.8 (8.4, 11.2)	6.4 (5.2, 7.7)	3.4 (1.5, 5.2)	
% survival time	66%	50%	16%	
TOX	0.7 (0.5, 0.9)	0.3 (0.1, 0.5)	0.4 (0.1, 0.6)	
% survival time	5%	2%	2%	
REL	4.4 (2.8, 6.0)	6.1 (4.6, 7.6)	−1.8 (−4.0, 0.4)	
% survival time	29%	47%	−18%	
Q-TWiST	**12.2 (11.1, 13.2)**	**10.2 (9.2, 11.3)**	**1.9 (0.5, 3.4)**	**14.9%**

[a]Calculated by dividing the difference by mean OS of the atezo arm. [b]Median PFS and OS were 5.4 and 13.2 months, respectively, in the lurbi + atezo arm and 2.1 and 10.6 months, respectively, in the atezo arm.
atezo, atezolizumab; lurbi, lurbinectedin; OS, overall survival; PFS, progression-free survival; Q-TWiST, quality-adjusted TWiST; REL, relapse; TOX, toxicity; TWiST, time without symptoms or toxicity.

- **The lurbinectedin + atezolizumab arm had a longer mean Q-TWiST vs atezolizumab (12.2 vs 10.2 months), representing a 14.9% gain in quality-adjusted survival, typically considered a clinically important difference[6]**
- In a sensitivity analysis, which included grade ≥2 AEs in the TOX health state, the Q-TWiST duration remained substantially longer in the lurbinectedin + atezolizumab vs atezolizumab arm, with a mean difference of 1.9 months (95% CI: 0.5, 3.4), representing a 15.0% relative gain in quality-adjusted survival

Figure 3. Q-TWiST difference and relative gain[a] over follow-up time

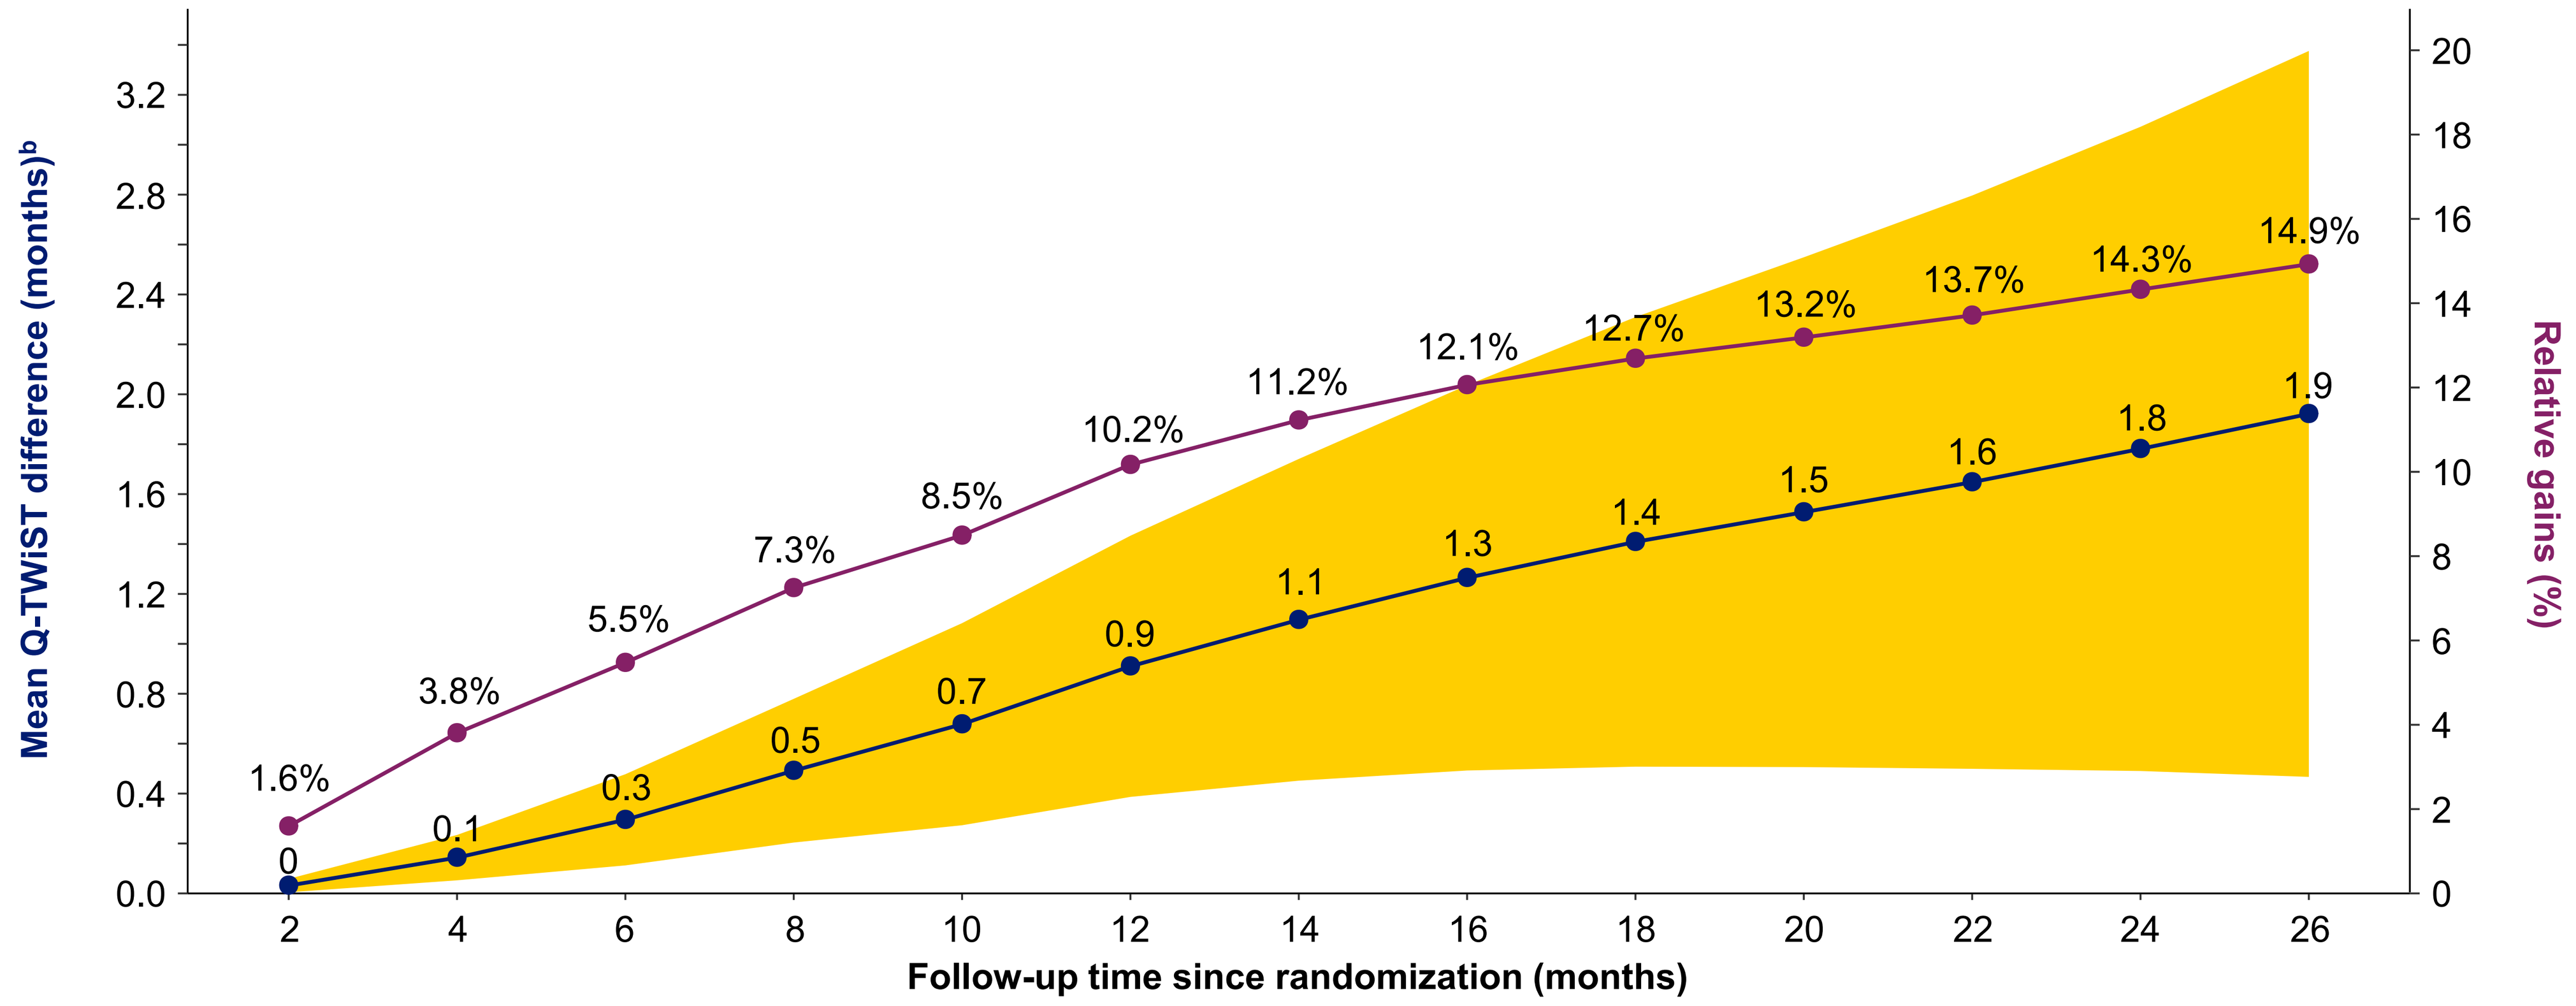

Mean Q-TWiST difference (months)b
Relative gains (%)
Follow-up time since randomization (months)
1.6%
3.8%
5.5%
7.3%
8.5%
10.2%
11.2%
12.1%
12.7%
13.2%
13.7%
14.3%
14.9%
0
0.1
0.3
0.5
0.7
0.9
1.1
1.3
1.4
1.5
1.6
1.8
1.9

[a]Reported as the difference between the Q-TWiSTs for lurbinectedin + atezolizumab and atezolizumab alone. Gains are reported relative to the atezolizumab mean overall survival. [b]Gold shading represents 95% CI for mean difference.
Q-TWiST, quality-adjusted time without symptoms or toxicity.

- The largest gain in Q-TWiST for lurbinectedin + atezolizumab over atezolizumab alone was observed at the maximum follow-up of 26.1 months
- A positive relationship was observed between the amount of follow-up time and the relative gain in Q-TWiST for lurbinectedin + atezolizumab compared to atezolizumab alone

Figure 4. Threshold utility analysis

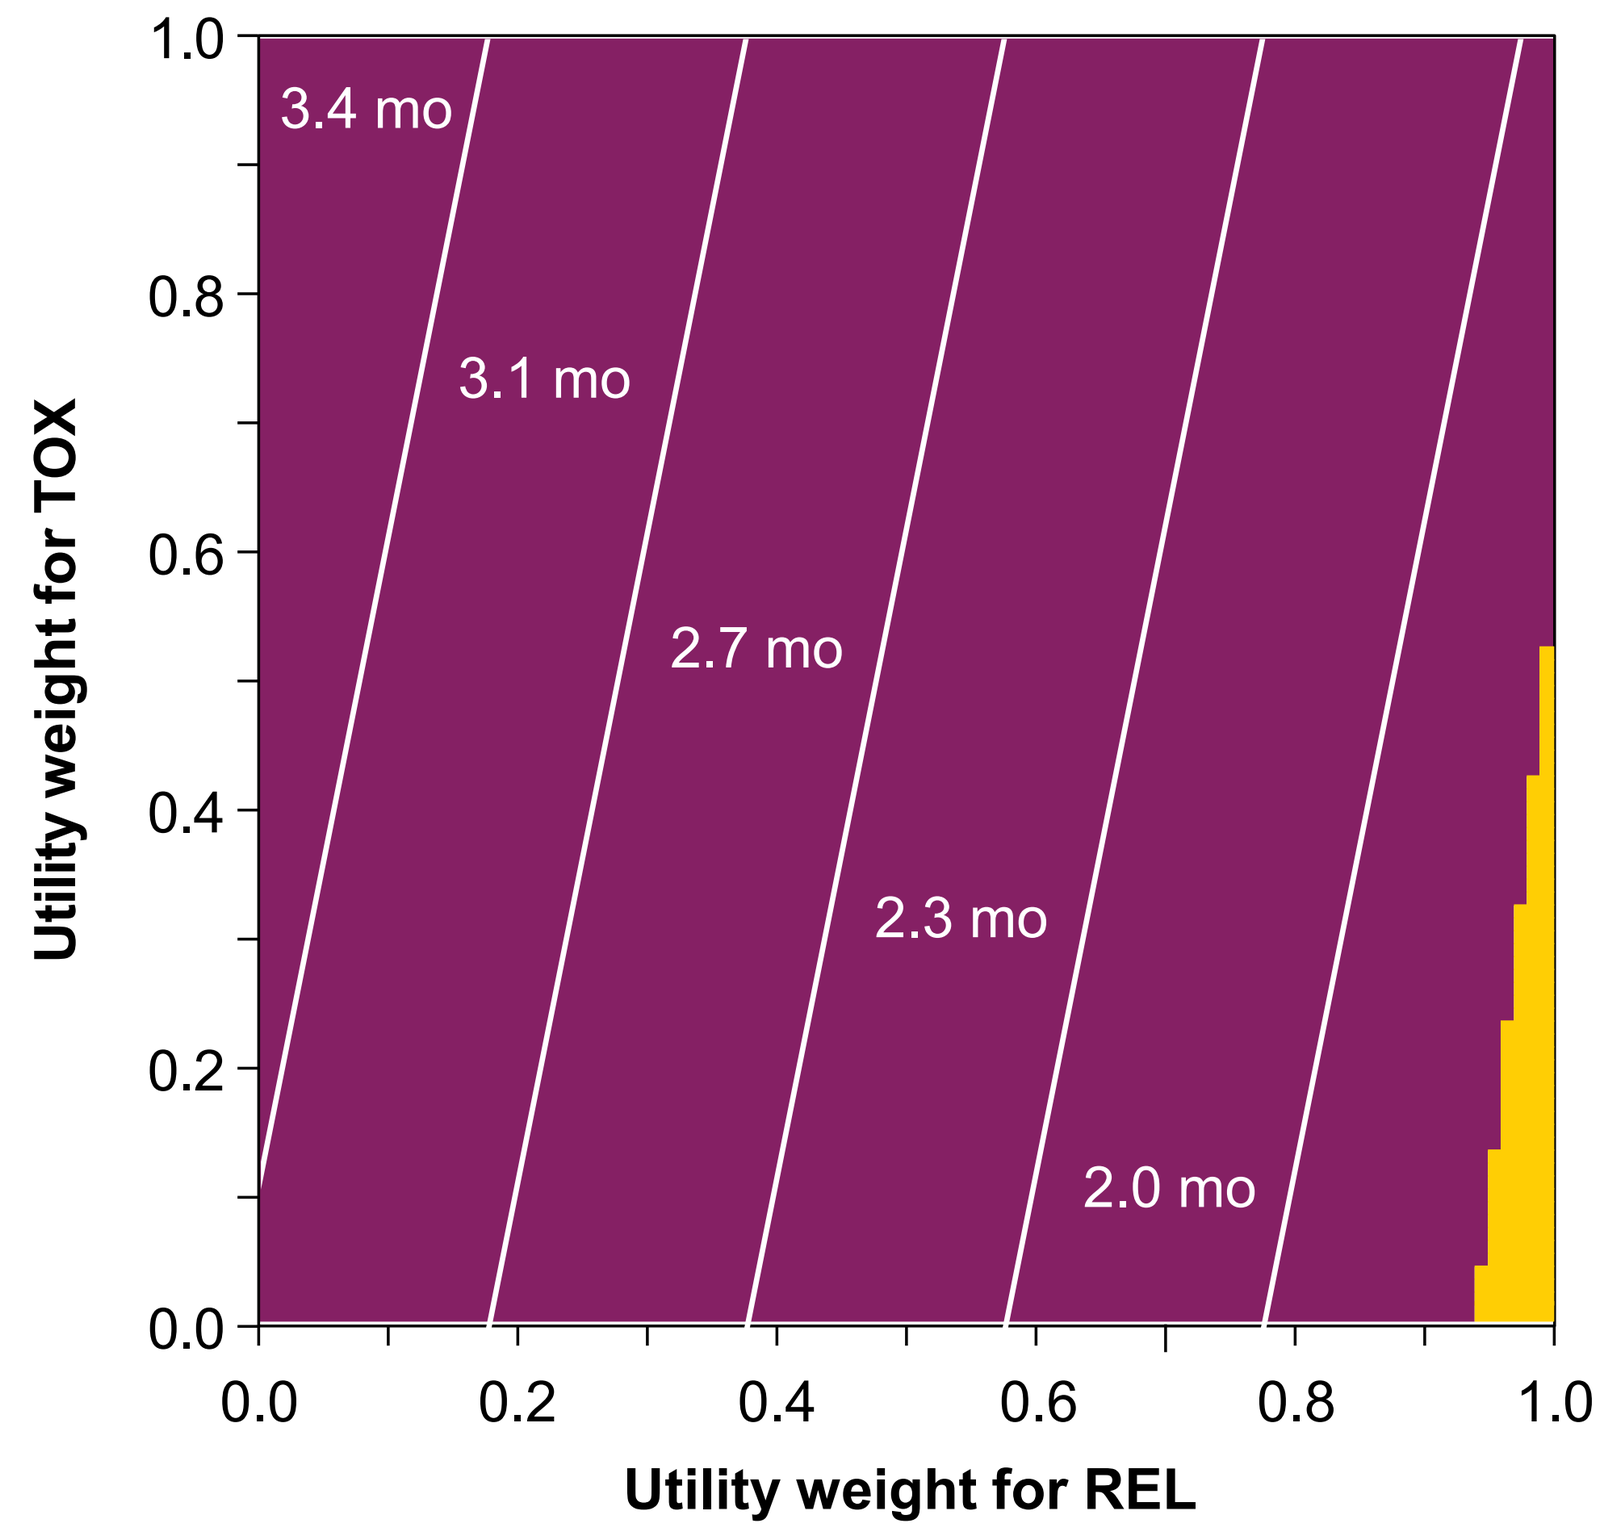

3.4 mo
3.1 mo
2.7 mo
2.3 mo
2.0 mo
Utility weight for TOX
Utility weight for REL

Lurbi + atezo is significantly longer than atezo

No significant difference between lurbi + atezo and atezo

Diagonal white lines show difference in months between the Q-TWiSTs for lurbi + atezo and atezo alone

- Across all possible QoL utility weight combinations, Q-TWiST differences favored lurbinectedin + atezolizumab over atezolizumab alone
- When using the common standardized utility weight (TOX, 0.5; TWiST, 1.0; REL, 0.5),[9] the mean Q-TWiST was 2.7 (95% CI: 1.2, 4.2) months longer in patients receiving lurbinectedin + atezolizumab vs atezolizumab, representing a 21% relative gain

The maximum follow-up of 26.1 months was used, and the TWiST utility weight was assumed to be 1.0. Significance was determined by a nominal $P < 0.05$.
atezo, atezolizumab; lurbi, lurbinectedin; Q-TWiST, quality-adjusted TWiST; REL, relapse; TOX, toxicity; TWiST, time without symptoms or toxicity.

Limitations

- While utility weights derived from patient-reported QoL data represent the gold-standard in Q-TWiST analyses,[8] questionnaires may not fully capture the key features of the health states from patients' points of view
- The utility threshold analysis is biased toward the TWiST state by assuming a utility weight of 1.0, as per standard methodology[7,8]; nevertheless, benefits of lurbinectedin + atezolizumab maintenance therapy vs atezolizumab alone were supported by analyses using patient-derived health metrics across various sensitivity analyses

560

IMforte: Quality-Adjusted Time Without Symptoms or Toxicity (Q-TWiST) Analysis of First-Line Maintenance Treatment With Lurbinectedin + Atezolizumab vs Atezolizumab in Extensive-Stage Small Cell Lung Cancer

Hossein Borghaei[1], Luis Paz-Ares[2], Martin Reck[3], Roy S Herbst[4], Solange Peters[5], Kamalnayan Bhatt[6], Xiaoyan Wang[7], Jonathon Gable[7], Siobhan Connor-Ahmad[8], Carla Mamolo[9], Ya-Chen Lin[9], Stephen V Liu[10]

[1]Department of Hematology and Oncology, Fox Chase Cancer Center, Philadelphia, PA, USA; [2]Department of Medical Oncology, Hospital Universitario 12 de Octubre, H12O-CNIO Lung Cancer Unit, Universidad Complutense and Ciberonc, Madrid, Spain; [3]Department of Thoracic Oncology, Lung Clinic Grosshansdorf, Airway Research Center North, German Center of Lung Research, Grosshansdorf, Germany; [4]Department of Medical Oncology and Hematology, Yale School of Medicine, New Haven, CT, USA; [5]Department of Oncology, Centre Hospitalier Universitaire Vaudois, Lausanne, Switzerland; [6]Jazz Pharmaceuticals, Philadelphia, PA, USA; [7]Jazz Pharmaceuticals, Palo Alto, CA, USA; [8]Roche Products Ltd, Welwyn, UK; [9]Genentech, a Member of the Roche Group, South San Francisco, CA, USA; [10]Department of Medical Oncology, Lombardi Comprehensive Cancer Center, Georgetown University, Washington, DC, USA

Corresponding author: Dr Hossein Borghaei
hossein.borghaei@fccc.edu

Poster presented at the 2026 American Society of Clinical Oncology Annual Meeting; May 29–June 2, 2026; Chicago, IL, and online.

Copies of this presentation obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission from the authors.

Lay summary

- **Why did we perform this research?**
 - Extensive-stage small cell lung cancer (ES-SCLC) is a deadly disease: without treatment, at least half of patients die within 2 to 4 months
 - Most people treated for ES-SCLC have good results over the first 3 months of treatment with potent chemotherapy plus an immunotherapy
 - After those first 3 months, treatment continues with an immunotherapy alone, but most patients’ disease gets worse within 6 months, so it is important to identify new ways to extend the benefits of those initial treatments
 - IMforte was a phase 3 study that showed that adding a drug called lurbinectedin to an immunotherapy, atezolizumab, after patients finished 3 months of initial treatment with chemotherapy and atezolizumab, helped patients who have ES-SCLC live longer, although this drug combination had more side effects than atezolizumab alone
 - We conducted this study to understand how much time patients spent alive and feeling well, without being bothered by cancer symptoms or treatment side effects. By using the Q-TWiST method, we could measure not just how long the drug extended life but also the quality of that extra time
- **How did we perform this research?**
 - Patients in the IMforte study answered questionnaires that asked how they felt throughout the study
 - We measured the total time each patient lived and then adjusted that time based on how they felt during the study using the results from the questionnaires
 - We gave more “weight” to time spent feeling healthy and less “weight” to time spent feeling unwell, resulting in a single score that represents the overall quality of survival time
- **What were the results of this research?**
 - When adjusting for the quality of their time alive, lurbinectedin and atezolizumab lengthened patients’ lives by around 15% from when they began treatment compared to patients who received atezolizumab alone
 - Compared to those who received atezolizumab alone, patients who received the combination of lurbinectedin and atezolizumab spent about 3 and half more months without being significantly burdened by side effects and without their disease getting worse