36

Real-World Treatment Patterns and Overall Survival in Patients With HER2-Positive Advanced or Metastatic Gastroesophageal Adenocarcinomas in the US

Farshid Dayyani[1], Xiaozhou Fan[2], Joan Zape[2], Roderick Murphy[3], Keith A Betts[4], Yan Wang[4], Shuang Wang[5], Ariel Chao[4], Wayne Su[2], Douglas S Fuller[2], Javier Sabater[3], Michael K Gibson[6], Peter Enzinger[7,8]

[1]Chao Family Comprehensive Cancer Center, University of California Irvine, Orange, CA, USA; [2]Jazz Pharmaceuticals, Philadelphia, PA, USA; [3]Jazz Pharmaceuticals, London, UK; [4]Analysis Group, Inc., Los Angeles, CA, USA; [5]Analysis Group, Inc., Boston, MA, USA; [6]Vanderbilt-Ingram Cancer Center, Nashville, TN, USA; [7]Dana-Farber Cancer Institute, Boston, MA, USA; [8]Harvard Medical School, Boston, MA, USA

Objective

- To describe real-world (rw) treatment patterns, progression-free survival (PFS), and overall survival (OS) among adults with human epidermal growth factor receptor 2–positive (HER2+) advanced or metastatic gastroesophageal adenocarcinoma (mGEA) in the US following Food and Drug Administration (FDA) approval of first-line (1L) immunotherapy (IO)-containing regimens for HER2+ mGEA

Conclusions

- Despite the availability of HER2-targeted therapy, a significant unmet need remains, as we found that 40% of patients with HER2+ mGEA did not receive 1L HER2-targeted therapy
- Treatment patterns across lines of therapy in patients with HER2+ mGEA highlight potential non-adherence to treatment guidelines
- Among patients who received 1L therapy, numerically longer rwPFS and OS were observed in the programmed death-ligand 1–positive (PD-L1+) subgroup treated with HER2-targeted therapy in combination with IO and chemotherapy relative to the PD-L1+ subgroup that did not receive IO
- These findings reinforce the importance of delivering biomarker-guided and guideline-concordant treatment approaches in HER2+ mGEA to ultimately improve survival outcomes in the rw

References: 1. Siegel RL, et al. *CA Cancer J Clin.* 2025;75(1):10-45. 2. Bonomi M, et al. *Int J Mol Sci.* 2024;25(7):3876. 3. Janjigian YY, et al. *Lancet.* 2023;402(10418):2197-208. 4. Elimova E, et al. *J Clin Oncol.* 2026;44(suppl 2):LBA285. 5. Janjigian YY, et al. *N Engl J Med.* 2024;391(14):1360-2.

Support and Acknowledgments: This research was funded by Jazz Pharmaceuticals. Under direction of the authors, Marita Chakhtoura, MS, PhD, of Red Nucleus, provided medical writing and editorial support, which were funded by Jazz Pharmaceuticals in accordance with Good Publication Practice (GPP 2022) guidelines (https://www.ismpp.org/gpp-2022). Jazz Pharmaceuticals also reviewed and edited the poster for scientific accuracy.

Disclosures: Farshid Dayyani reports consulting or advisory roles for AstraZeneca, Eisai, and Jazz Pharmaceuticals; speakers' bureau for Astellas Pharma, BeOne Medicines, Ipsen, Sirtex Medical, and Takeda; research funding from AstraZeneca (Inst), Bristol Myers Squibb (Inst), Exelixis (Inst), Genentech (Inst), Ipsen (Inst), Merck (Inst), and Taiho Pharmaceutical (Inst).

Corresponding author: Farshid Dayyani, MD
fdayyani@hs.uci.edu

Poster presented at the American Society of Clinical Oncology Annual Meeting; May 29–June 2, 2026; Chicago, IL, USA, and online.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster.

Introduction

- GEA accounts for more than 50,000 new cases annually and is among the leading causes of cancer-related mortality in the US[1]
- Roughly 20% of mGEA cases are HER2+[2]
- The 1L standard of care for patients with HER2+ mGEA is HER2-targeted therapy plus platinum- and fluoropyrimidine-based chemotherapy. The addition of IO to this regimen has further improved survival outcomes in patients with HER2+ PD-L1+ disease[3]
 - Median OS was 26.4 months with zanidatamab + IO + chemotherapy vs 24.4 months with zanidatamab + chemotherapy alone in HER2+ mGEA, regardless of PD-L1 status[4]
 - Median OS was 20.1 months with trastuzumab + IO + chemotherapy vs 15.7 months with trastuzumab + chemotherapy alone in HER2+ PD-L1+ mGEA[5]

Figure 1. Study design

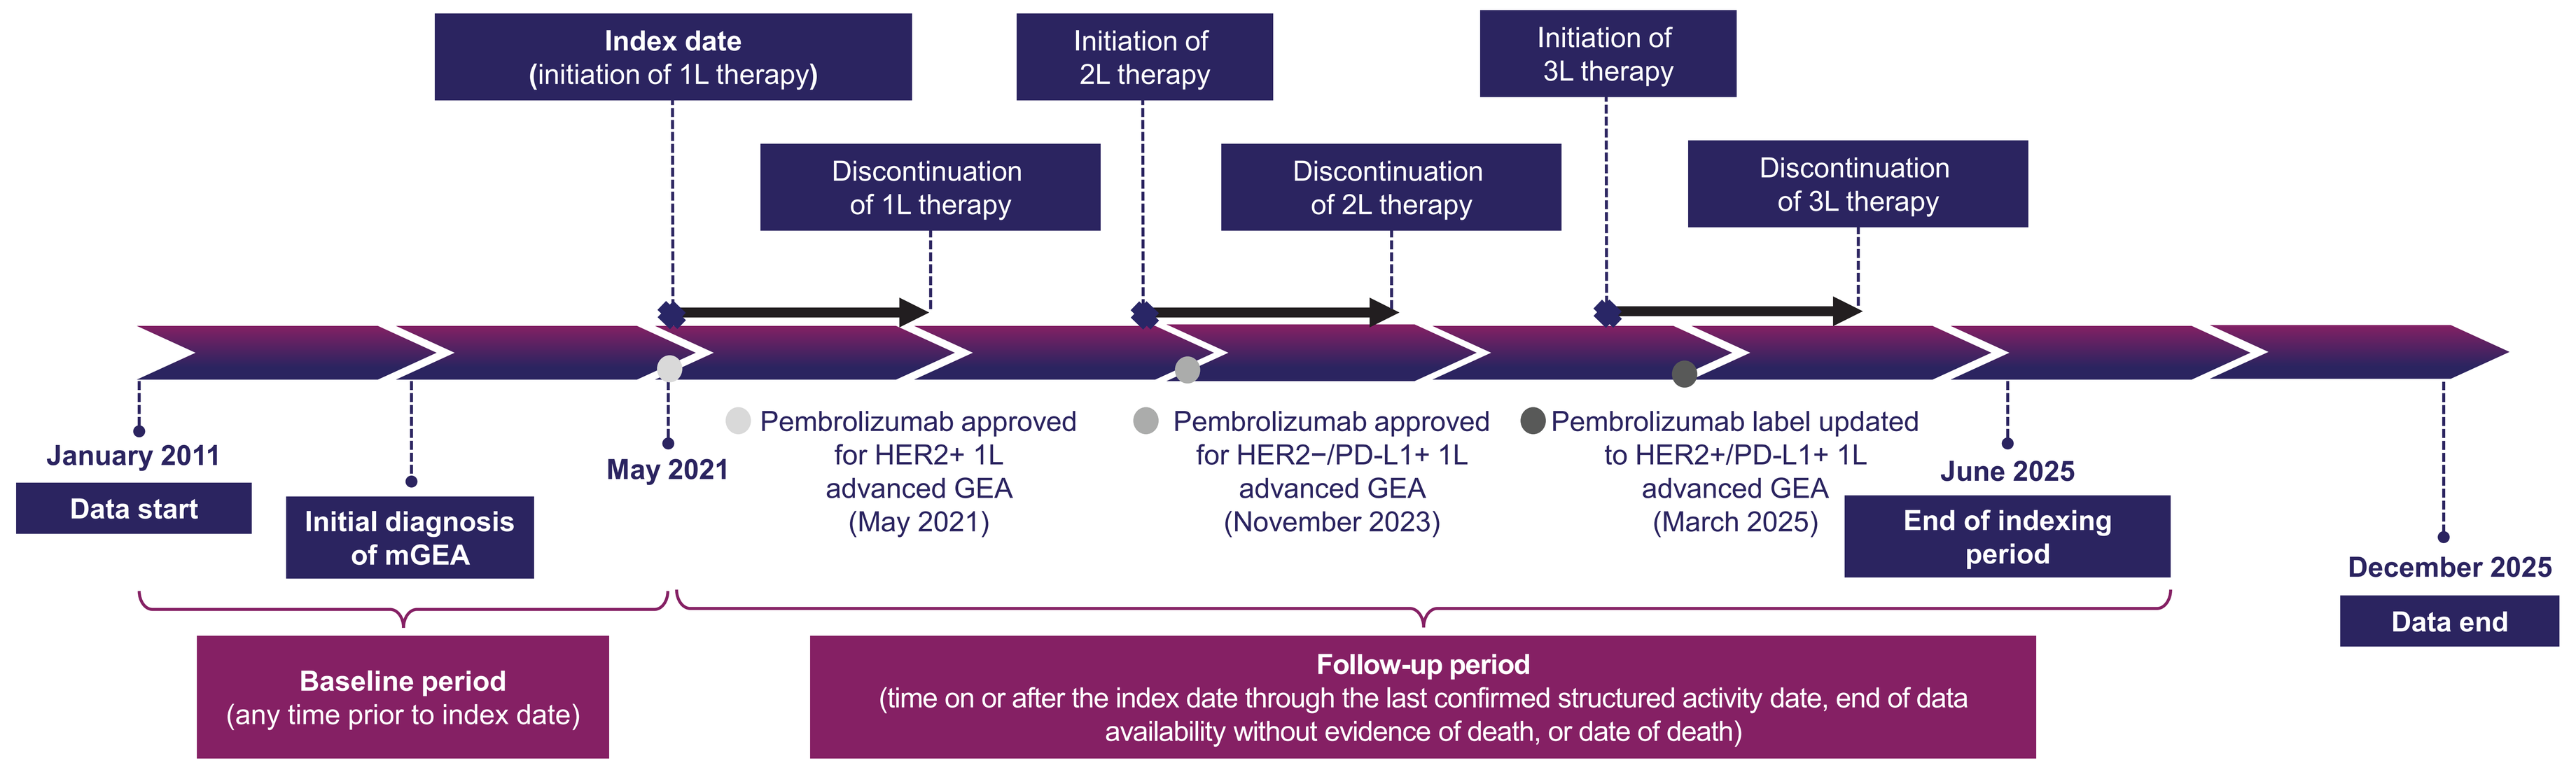

Index date (initiation of 1L therapy)
Initiation of 2L therapy
Initiation of 3L therapy
Discontinuation of 1L therapy
Discontinuation of 2L therapy
Discontinuation of 3L therapy
January 2011
Data start
Initial diagnosis of mGEA
May 2021
Pembrolizumab approved for HER2+ 1L advanced GEA (May 2021)
Pembrolizumab approved for HER2−/PD-L1+ 1L advanced GEA (November 2023)
Pembrolizumab label updated to HER2+/PD-L1+ 1L advanced GEA (March 2025)
June 2025
End of indexing period
December 2025
Data end
Baseline period (any time prior to index date)
Follow-up period (time on or after the index date through the last confirmed structured activity date, end of data availability without evidence of death, or date of death)

1L, first-line; 2L, second-line; 3L, third-line; GEA, gastroesophageal adenocarcinoma; HER2, human epidermal growth factor receptor 2; mGEA, advanced or metastatic GEA; PD-L1, programmed death-ligand 1.

Methods

Data source

- This is a retrospective observational study using the US Flatiron Health Electronic Health Record (EHR) deidentified database between January 2011 and December 2025
- Adults with HER2+ mGEA who received 1L treatment following the FDA approval of 1L IO (between May 2021 and June 2025) were selected, with the 1L initiation date defined as the index date
 - Generally, 1L therapy was defined as the first eligible drug episode plus other drugs administered within 28 days. The name of the regimen for that line of therapy is the combination of therapies in that line, unless otherwise noted
- HER2+ was defined by immunohistochemistry (IHC) test with staining intensity 3+, IHC 2+ with erb-b2 receptor tyrosine kinase 2 gene (*ERBB2*; gene encoding HER2) amplification, or clinician documentation, assessed any time prior to the index date or within 14 days post-index
- Patients were further assessed by PD-L1 status (PD-L1+ or PD-L1−); PD-L1+ was defined by a combined positive score ≥1 or clinician documentation, assessed at any time prior to or within 14 days post-index (no additional tests for PD-L1 status were identified from the database)
 - Additional treatment-specific subgroups were defined based on 1L HER2-targeted therapy + chemotherapy ± IO use among patients with PD-L1+ or PD-L1− disease

Outcomes

- Real-world PFS was defined as the time from the index date to the first occurrence of rw progression, initiation of second-line (2L) treatment, or death due to any cause, whichever occurred first
 - Patients who did not experience these events were censored on the date of last structured activity
- OS was defined as the time from the date of 1L initiation to death due to any cause
 - Patients who had no documented death were censored on the date of last structured activity

Statistical analysis

- Treatment regimen distributions from 1L to third-line (3L) were described using frequency counts and proportions
- Common treatment sequences across the first 3 lines were described using a Sankey diagram among the overall patients with HER2+ disease, regardless of PD-L1 status
- Time from 1L treatment initiation to rwPFS and OS was estimated using the Kaplan-Meier method in the overall HER2+ population, PD-L1 subgroups, and treatment-specific by PD-L1 status

Results

Table 1. Real-world baseline characteristics in adult patients with HER2+ mGEA who received 1L systemic therapies

	HER2+ N = 489	HER2+/PD-L1+ n = 256	HER2+/PD-L1− n = 56
Demographics as of the index date			
Age (years), mean ± SD	66.1 ± 11.7	66.4 ± 12.6	65.1 ± 10.0
Female, n (%)	96 (19.6)	54 (21.1)	12 (21.4)
Race/ethnicity, n (%)			
White	313 (64.0)	165 (64.5)	34 (60.7)
Hispanic or Latino	46 (9.4)	29 (11.3)	8 (14.3)
Black or African American	34 (7.0)	19 (7.4)	7 (12.5)
Asian	5 (1.0)	1 (0.4)	0
Other race	18 (3.7)	6 (2.3)	1 (1.8)
Missing	73 (14.9)	36 (14.1)	6 (10.7)
Practice type, n (%)			
Academic	100 (20.4)	51 (19.9)	16 (28.6)
Community	364 (74.4)	197 (77.0)	36 (64.3)
Missing	25 (5.1)	8 (3.1)	4 (7.1)
Clinical characteristics during the baseline period			
ECOG PS, n (%)			
0 or 1	325 (66.5)	171 (66.8)	40 (71.4)
≥2	50 (10.2)	31 (12.1)	3 (5.4)
Missing	114 (23.3)	54 (21.1)	13 (23.2)
Stage at initial diagnosis, n (%)			
Stage I	10 (2.0)	3 (1.2)	2 (3.6)
Stage II	29 (5.9)	18 (7.0)	1 (1.8)
Stage III	61 (12.5)	26 (10.2)	6 (10.7)
Stage IV	310 (63.4)	165 (64.5)	41 (73.2)
Missing/unknown	79 (16.2)	44 (17.2)	6 (10.7)
Primary site of disease, n (%)			
Gastric	123 (25.2)	67 (26.2)	20 (35.7)
GEJ	117 (23.9)	59 (23.0)	10 (17.9)
Esophageal	249 (50.9)	130 (50.8)	26 (46.4)
HER2 IHC, n (%)			
IHC 3+	344 (70.3)	184 (71.9)	42 (75.0)
IHC 2+/*ERBB2*-amplified	128 (26.2)	65 (25.4)	14 (25.0)
Per physician's notes	17 (3.5)	7 (2.7)	0
CPS, n (%)			
<1	53 (10.8)	0	51 (91.1)
1 to <5	118 (24.1)	103 (40.2)	0
5 to <10	70 (14.3)	62 (24.2)	0
≥10	91 (18.6)	82 (32.0)	0
Missing/unknown	157 (32.1)	9 (3.5)	5 (8.9)

1L, first-line; CPS, combined positive score; ECOG PS, Eastern Cooperative Oncology Group Performance Status; *ERBB2*, erb-b2 receptor tyrosine kinase 2 gene; GEJ, gastroesophageal junction; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; mGEA, advanced or metastatic gastroesophageal adenocarcinoma; PD-L1, programmed death-ligand 1; SD, standard deviation.

- Among 2335 patients with known HER2 status, 489 (20.9%) had HER2+ disease
 - Of patients with HER2+ disease, 344 (70.3%) had IHC 3+ tumors, 128 (26.2%) had IHC 2+/*ERBB2*-amplified tumors, and 17 (3.5%) had documented HER2+ disease without testing results
- Among 312 patients with HER2+ mGEA with known PD-L1 status, 256 (82.1%) had PD-L1+ and 56 (17.9%) had PD-L1− disease
- In the overall HER2+ cohort, mean (standard deviation) age was 66.1 (11.7) years, 19.6% of patients were female, and 66.5% had an Eastern Cooperative Oncology Group Performance Status of 0 or 1
- The most common tumor site was the esophagus (50.9%), followed by the stomach (25.2%) and gastroesophageal junction (23.9%)

Table 2. Real-world 1L to 3L treatment regimens in adults with HER2+ mGEA

	HER2+	HER2+/PD-L1+	HER2+/PD-L1−
Treatment regimen in 1L, n (%)	N = 489	n = 256	n = 56
HER2-targeted therapy	293 (59.9)	156 (60.9)	42 (75.0)
HER2-targeted therapy + IO + chemotherapy	174 (35.6)	106 (41.4)	16 (28.6)
HER2-targeted therapy + chemotherapy	104 (21.3)	41 (16.0)	23 (41.1)
HER2-targeted therapy only	8 (1.6)	4 (1.6)	2 (3.6)
Other[a]	7 (1.4)	5 (2.0)	1 (1.8)
IO therapy	47 (9.6)	31 (12.1)	1 (1.8)
IO + chemotherapy	31 (6.3)	20 (7.8)	1 (1.8)
IO only	16 (3.3)	11 (4.3)	0
Other non–HER2-targeted therapy[b]	8 (1.6)	3 (1.2)	3 (5.4)
Chemotherapy only	141 (28.8)	66 (25.8)	10 (17.9)
Agents of special interest in 1L, n (%)			
Pembrolizumab	183 (37.4)	116 (45.3)	16 (28.6)
Nivolumab	49 (10.0)	29 (11.3)	1 (1.8)
FOLFOX	299 (61.1)	153 (59.8)	41 (73.2)
Subsequent lines of therapy, n (%)			
Patients who received 2L treatment	n = 242	n = 127	n = 31
HER2-targeted therapy	169 (69.8)	91 (71.7)	25 (80.6)
Fam-trastuzumab deruxtecan-nxki	66 (27.3)	35 (27.6)	15 (48.4)
Chemotherapy only	30 (12.4)	13 (10.2)	2 (6.5)
Patients who received 3L treatment	n = 103	n = 54	n = 13
HER2-targeted therapy	51 (49.5)	30 (55.6)	6 (46.2)
Chemotherapy only	14 (13.6)	4 (7.4)	3 (23.1)

[a]Includes HER2-targeted therapy + IO, or HER2-targeted therapy + other non-HER2-target therapy + chemotherapy.
[b]Other non–HER2-targeted therapy (all anti-cancer treatments) administered as monotherapy in the 1L setting included anastrozole, apalutamide, bicalutamide, carfilzomib, cetuximab, crizotinib, enzalutamide, erlotinib, exemestane, fulvestrant, futibatinib, goserelin, ibrutinib, isatuximab-irfc, lenalidomide, letrozole, leuprolide, nilotinib, olaparib, osimertinib, palbociclib, ramucirumab, ribociclib, rituximab, tamoxifen, venetoclax, and zanubrutinib.
1L, first-line; 2L, second-line; 3L, third-line; FOLFOX, leucovorin + fluorouracil + oxaliplatin; HER2, human epidermal growth factor receptor 2; IO, immunotherapy; mGEA, advanced or metastatic gastroesophageal adenocarcinoma; PD-L1, programmed death-ligand 1.

- Among 489 treated patients with HER2+ disease, only 293 (59.9%) received HER2-targeted therapy in 1L
 - HER2-targeted therapy was most commonly combined with IO and chemotherapy in 174 (35.6%) patients or with chemotherapy alone in 104 (21.3%) patients
- One hundred forty-one (28.8%) patients received chemotherapy alone, and 47 (9.6%) received IO regimens without HER2-targeted therapy
- One hundred and eighty-three (37.4%) patients received pembrolizumab as 1L therapy, and 49 (10.0%) received nivolumab
- FOLFOX was the most commonly used 1L chemotherapy, received by 299 (61.1%) patients

Figure 2. Sankey diagram of 1L to 3L treatment patterns in patients with HER2+ mGEA

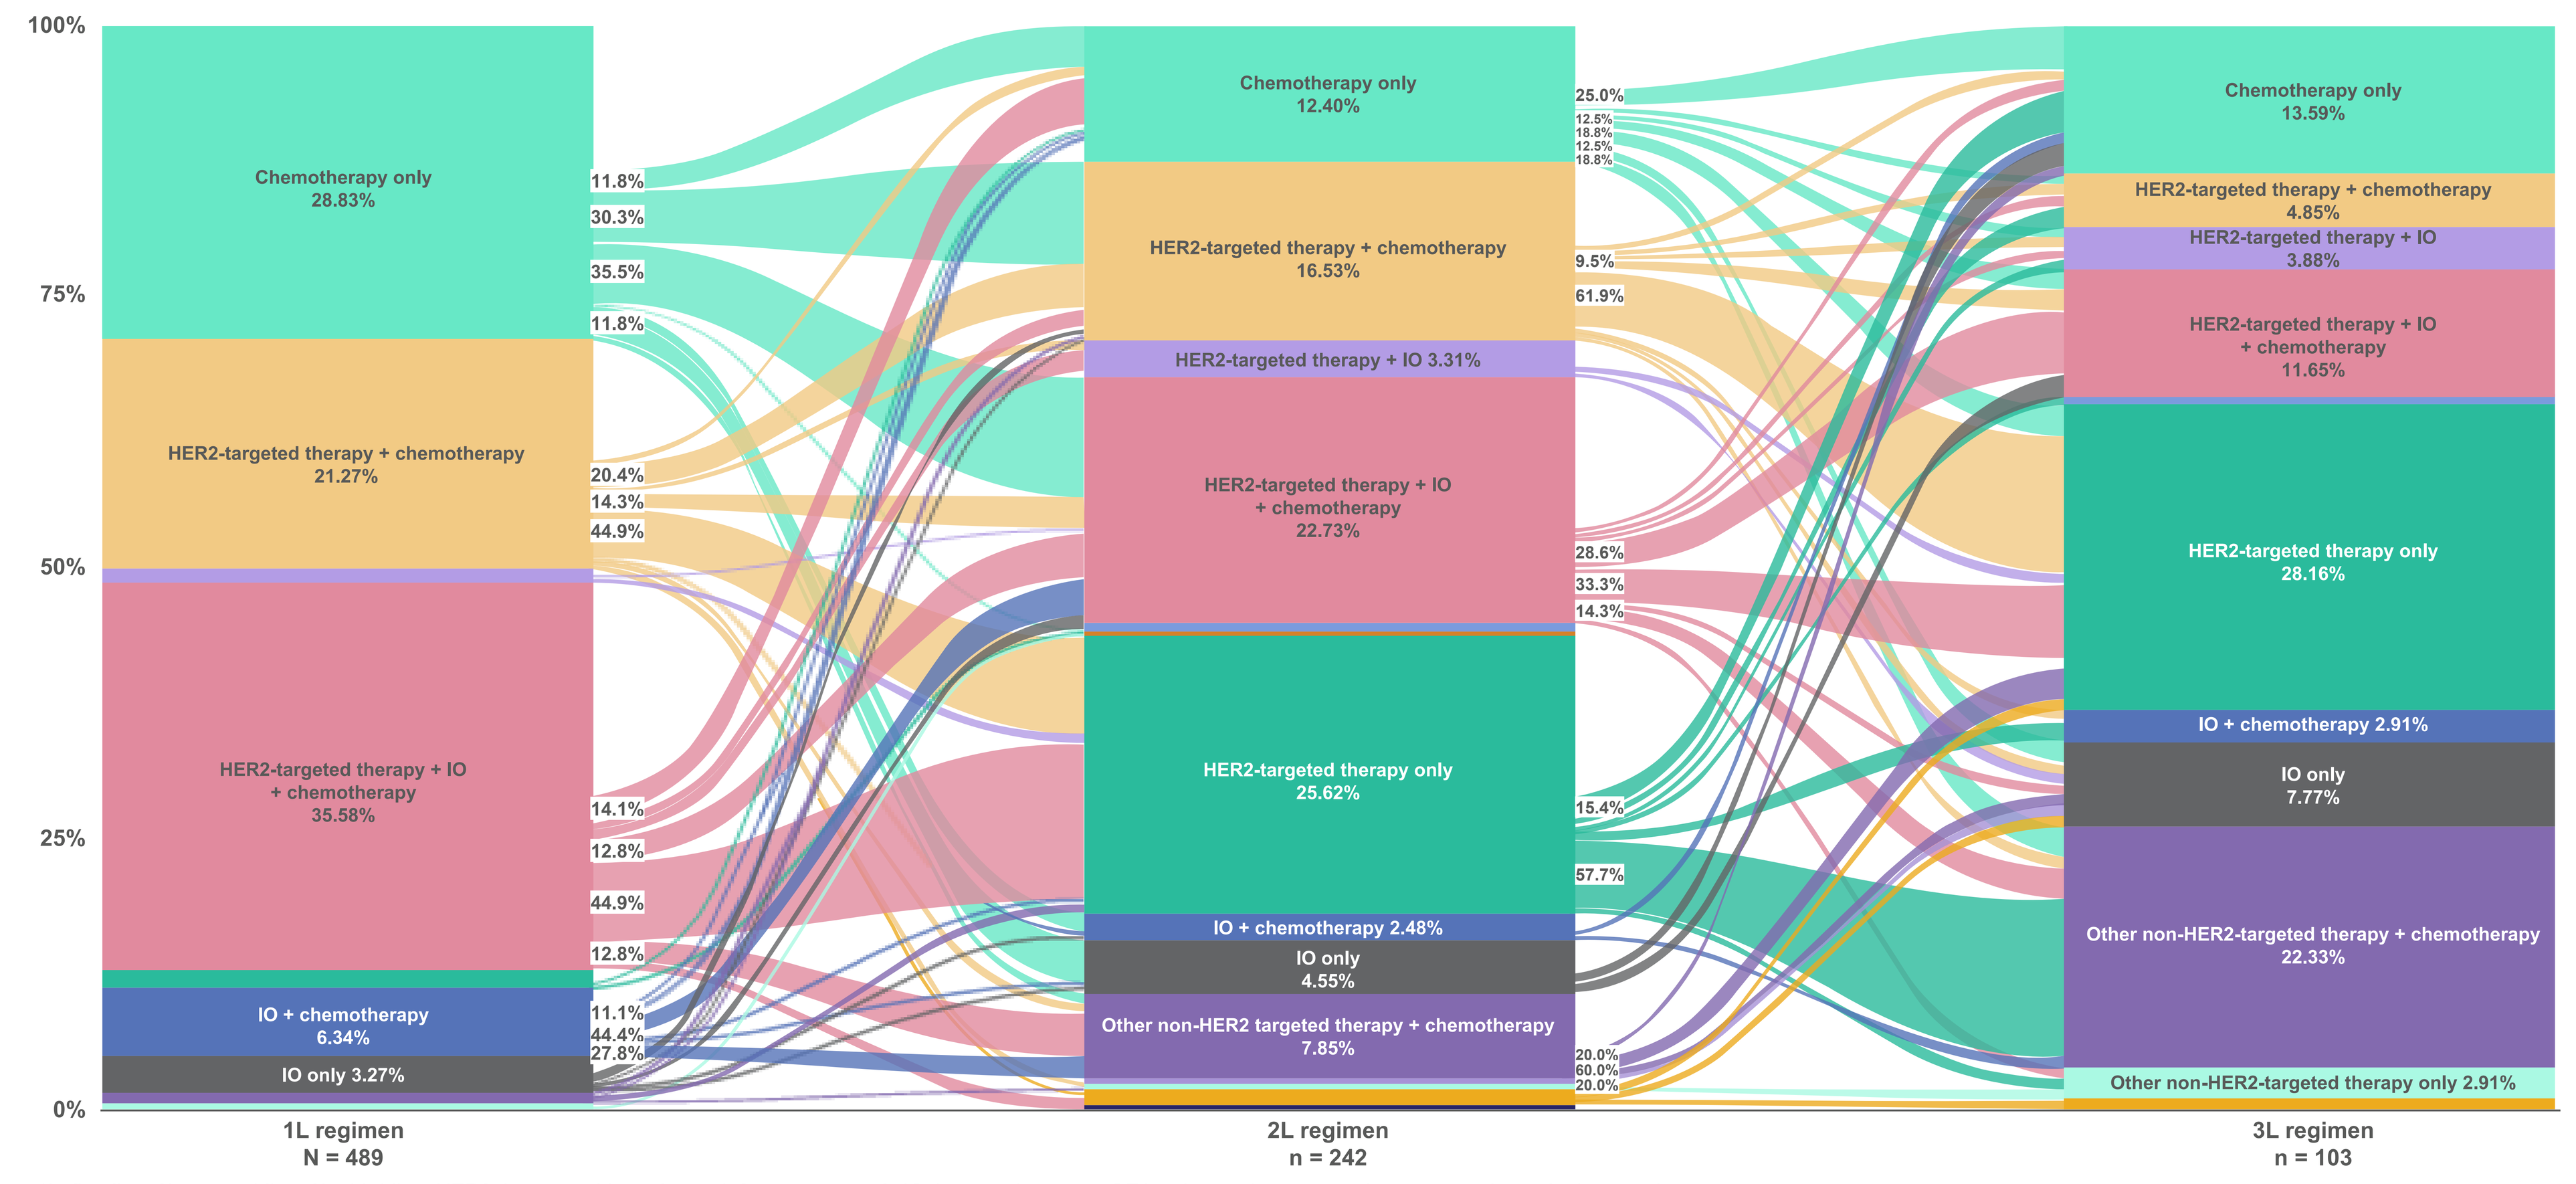

100%
75%
50%
25%
0%
Chemotherapy only 28.83%
HER2-targeted therapy + chemotherapy 21.27%
HER2-targeted therapy + IO + chemotherapy 35.58%
IO + chemotherapy 6.34%
IO only 3.27%
1L regimen N = 489
Chemotherapy only 12.40%
HER2-targeted therapy + chemotherapy 16.53%
HER2-targeted therapy + IO 3.31%
HER2-targeted therapy + IO + chemotherapy 22.73%
HER2-targeted therapy only 25.62%
IO + chemotherapy 2.48%
IO only 4.55%
Other non-HER2 targeted therapy + chemotherapy 7.85%
2L regimen n = 242
Chemotherapy only 13.59%
HER2-targeted therapy + chemotherapy 4.85%
HER2-targeted therapy + IO 3.88%
HER2-targeted therapy + IO + chemotherapy 11.65%
HER2-targeted therapy only 28.16%
IO + chemotherapy 2.91%
IO only 7.77%
Other non-HER2-targeted therapy + chemotherapy 22.33%
Other non-HER2-targeted therapy only 2.91%
3L regimen n = 103

1L, first-line; 2L, second-line; 3L, third-line; HER2, human epidermal growth factor receptor 2; IO, immunotherapy; mGEA, advanced or metastatic gastroesophageal adenocarcinoma.

- Among the 489 patients who were treated in the 1L setting, 242 (49.5%) received 2L therapy and 103 (21.1%) received 3L therapy
 - In the 2L setting, 169 (69.8%) patients received HER2-targeted therapy alone or in combination with other agents and 62 (25.6%) received HER2-targeted monotherapy; among these, 64 (38.4%) were HER2-naïve in the 1L setting, having received chemotherapy or IO-based regimens without HER2-targeted therapy
 - In the 3L setting, 51 (49.5%) patients received HER2-targeted therapy and 29 (28.2%) received HER2-targeted monotherapy

rwPFS

Figure 3. Kaplan-Meier curves for rwPFS in patients with HER2+ mGEA in the 1L setting

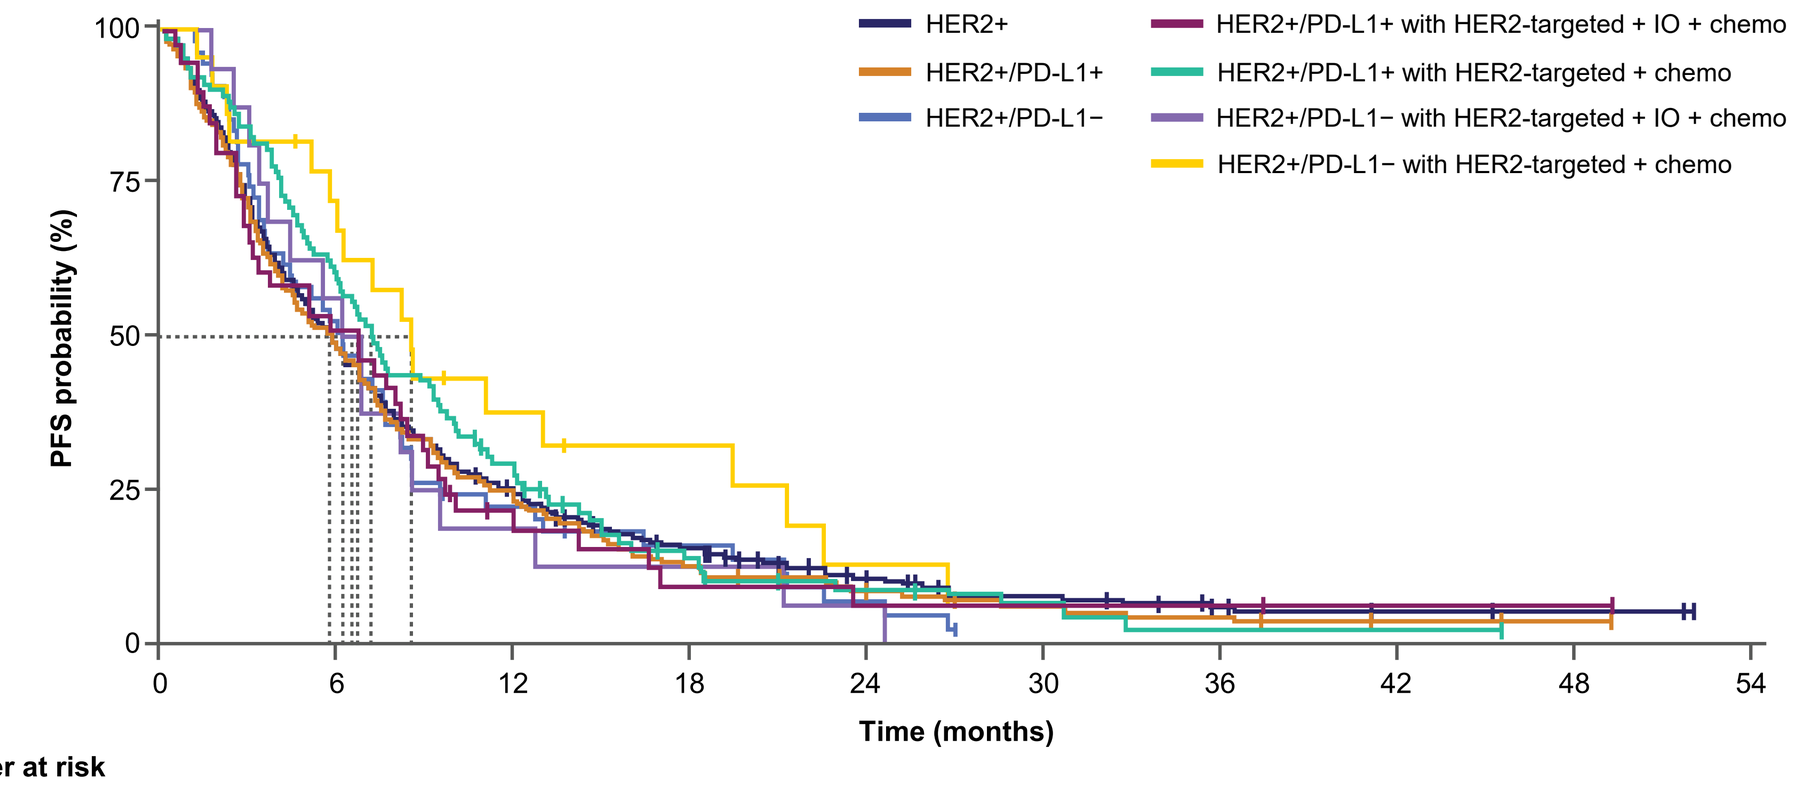

PFS probability (%)
Time (months)
HER2+
HER2+/PD-L1+
HER2+/PD-L1−
HER2+/PD-L1+ with HER2-targeted + IO + chemo
HER2+/PD-L1+ with HER2-targeted + chemo
HER2+/PD-L1− with HER2-targeted + IO + chemo
HER2+/PD-L1− with HER2-targeted + chemo

Number at risk	0	6	12	18	24	30	36	42	48	54
HER2+	489	225	102	56	28	16	9	5	3	
HER2+/PD-L1+	256	119	51	25	13	7	5	2	1	
HER2+/PD-L1−	56	28	11	7	3	0	0	0	0	
HER2+/PD-L1+ with HER2-targeted + IO + chemo	106	62	26	11	5	3	1	1	0	
HER2+/PD-L1+ with HER2-targeted + chemo	41	21	6	3	2	2	2	1	1	
HER2+/PD-L1− with HER2-targeted + IO + chemo	16	9	3	2	1	0	0	0	0	
HER2+/PD-L1− with HER2-targeted + chemo	23	15	7	5	2	0	0	0	0	

	Total patients	Patients with rwP, advancement to 2L treatment, or death, n (%)	Median rwPFS (95% CI), months	rwPFS at landmark time points: Month 6	Month 12	Month 18	Month 24	Month 30	Month 36
HER2+	489	416 (85.1)	5.8 (5.1, 6.6)	48.2%	24.6%	15.4%	10.1%	7.2%	5.7%
HER2+/PD-L1+	256	224 (87.5)	5.7 (4.6, 6.7)	48.2%	23.5%	12.8%	8.4%	6.1%	4.3%
HER2+/PD-L1−	56	51 (91.1)	6.2 (4.4, 8.2)	52.6%	22.4%	16.0%	6.9%	NA	NA
HER2+/PD-L1+ with HER2-targeted + IO + chemo	106	92 (86.8)	7.2 (6.0, 9.4)	59.6%	28.4%	13.9%	8.4%	6.3%	2.1%
HER2+/PD-L1+ with HER2-targeted + chemo	41	37 (90.2)	6.7 (3.3, 8.9)	51.2%	18.6%	9.3%	6.2%	6.2%	6.2%
HER2+/PD-L1− with HER2-targeted + IO + chemo	16	16 (100)	6.5 (3.7, 12.7)	56.3%	18.8%	12.5%	6.3%	0	0
HER2+/PD-L1− with HER2-targeted + chemo	23	18 (78.3)	8.5 (6.2, 22.5)	72.2%	37.9%	32.5%	13.0%	NA	NA

1L, first-line; 2L, second-line; chemo, chemotherapy; HER2, human epidermal growth factor receptor 2; IO, immunotherapy; mGEA, advanced or metastatic gastroesophageal adenocarcinoma; NA, not available; PD-L1, programmed death-ligand 1; rwP, real-world progression; rwPFS, real-world progression-free survival.

- Median PFS was comparable across treatment groups, and the 24-month PFS rate was ≤10%
- Among patients with HER2+/PD-L1+ disease, the median rwPFS was numerically longer in those receiving HER2-targeted therapy in combination with IO and chemotherapy compared with those receiving HER2-targeted therapy in combination with chemotherapy alone (7.2 vs 6.7 months, respectively)

OS

Figure 4. Kaplan-Meier curves for OS in patients with HER2+ mGEA in the 1L setting

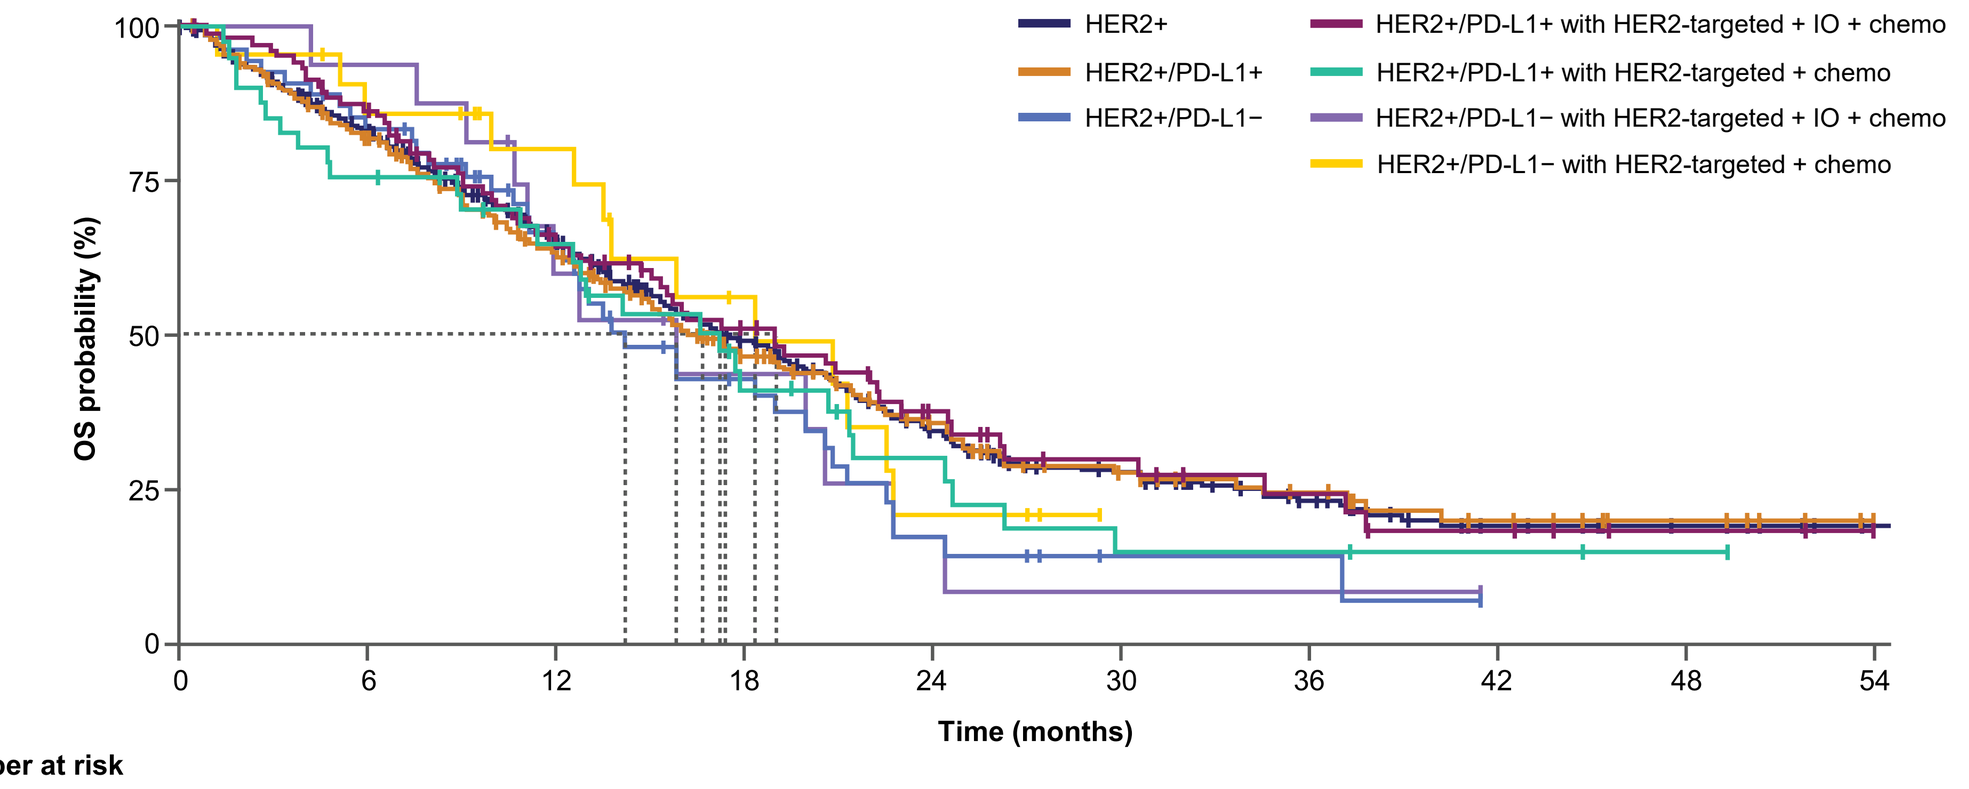

OS probability (%)
Time (months)
HER2+
HER2+/PD-L1+
HER2+/PD-L1−
HER2+/PD-L1+ with HER2-targeted + IO + chemo
HER2+/PD-L1+ with HER2-targeted + chemo
HER2+/PD-L1− with HER2-targeted + IO + chemo
HER2+/PD-L1− with HER2-targeted + chemo

Number at risk	0	6	12	18	24	30	36	42	48	54
HER2+	489	382	262	167	89	56	34	19	10	1
HER2+/PD-L1+	256	199	133	82	47	28	20	12	7	0
HER2+/PD-L1−	56	45	28	16	6	2	2	0	0	0
HER2+/PD-L1+ with HER2-targeted + IO + chemo	106	89	58	37	21	12	8	5	2	0
HER2+/PD-L1+ with HER2-targeted + chemo	41	31	23	13	8	4	4	2	1	0
HER2+/PD-L1− with HER2-targeted + IO + chemo	16	15	8	5	2	1	1	0	0	0
HER2+/PD-L1− with HER2-targeted + chemo	23	18	14	8	3	0	0	0	0	0

	Total patients	Patients died, n (%)	Median OS (95% CI), months	OS at landmark time points: Month 6	Month 12	Month 18	Month 24	Month 30	Month 36
HER2+	489	289 (59.1)	17.4 (15.7, 19.9)	82.9%	65.6%	49.0%	34.4%	27.8%	23.2%
HER2+/PD-L1+	256	151 (59.0)	16.7 (14.7, 20.9)	81.9%	63.6%	46.9%	35.9%	27.8%	24.4%
HER2+/PD-L1−	56	38 (67.9)	14.2 (12.2, 20.8)	83.5%	64.5%	43.1%	17.4%	14.5%	14.5%
HER2+/PD-L1+ with HER2-targeted + IO + chemo	106	62 (58.5)	19.0 (15.4, 24.5)	86.5%	66.6%	51.2%	37.8%	30.0%	24.5%
HER2+/PD-L1+ with HER2-targeted + chemo	41	29 (70.7)	17.2 (12.6, 24.4)	75.6%	64.9%	41.2%	30.2%	15.1%	15.1%
HER2+/PD-L1− with HER2-targeted + IO + chemo	16	12 (75.0)	15.8 (11.1, –)	93.8%	60.2%	43.9%	17.6%	8.8%	8.8%
HER2+/PD-L1− with HER2-targeted + chemo	23	13 (56.5)	18.3 (13.8, –)	85.9%	80.2%	56.2%	21.1%	NA	NA

1L, first-line; chemo, chemotherapy; HER2, human epidermal growth factor receptor 2; IO, immunotherapy; mGEA, advanced or metastatic gastroesophageal adenocarcinoma; NA, not available; OS, overall survival; PD-L1, programmed death-ligand 1.

- Across treatment groups, median OS remained <2 years, and the 24-month survival rate was <40%
- Among patients with HER2+/PD-L1+ disease, the median OS was numerically longer in those receiving HER2-targeted therapy in combination with IO and chemotherapy compared with those receiving HER2-targeted therapy in combination with chemotherapy alone (19.0 vs 17.2 months, respectively)

Limitations

- The Flatiron Health EHR data are primarily sourced from US community oncology networks, which may limit the generalizability of the findings for patients treated in academic settings or outside of the US
- Although Flatiron Health EHR data provide comprehensive patient-level information from structured data elements and unstructured clinical notes, the EHR data derived from routine data collection is subject to missing or incomplete data capture, which may cause disease and/or treatment misclassification