129

Evaluating the Impact of Switching From High-Sodium Oxybate to Low-Sodium Oxybate on Ambulatory Blood Pressure in People With Narcolepsy

Virend K. Somers, MD, PhD[1]; Richard J. Kovacs, MD[2]; Jessica K. Alexander, PhD[3]; Christine Baranak, MA[3]; Deborah A. Nichols, MS[4]; Jing Dai, PhD[4]; Marisa Whalen, PharmD[4]; Akinyemi Ajayi, MD[5]; Barbara Hutchinson, MD, PhD[6]; Yves Dauvilliers, MD[7]; William B. White, MD[8]

[1]Mayo Clinic, Rochester, MN, USA; [2]Division of Cardiovascular Medicine, Indiana University School of Medicine, Indianapolis, IN, USA; [3]Jazz Pharmaceuticals, Philadelphia, PA, USA; [4]Jazz Pharmaceuticals, Palo Alto, CA, USA; [5]Children's Lung, Asthma and Sleep Specialists, Winter Park, FL, USA; [6]Chesapeake Cardiac Care, Bowie, MD, USA; [7]University of Montpellier, Montpellier, France; [8]Calhoun Cardiology Center, University of Connecticut School of Medicine, Farmington, CT, USA

Introduction

- Excess sodium intake, consumed by ≈90% of Americans, is strongly linked to elevated blood pressure (BP) and cardiovascular disease (CVD)[1-6]
- People living with narcolepsy are at increased risk for hypertension and CVD[7-11]
- Scientific bodies, qualified experts, and governments around the world recommend a total daily sodium intake upper limit of 2300 mg with an ideal limit of <1500 mg/day for most adults to improve BP and heart health[1,12-17]
- High-sodium oxybate (SXB; Xyrem®) and low-sodium oxybate (LXB; Xywav®) are both approved by the US Food and Drug Administration to treat excessive daytime sleepiness or cataplexy in patients ≥7 years of age with narcolepsy; LXB is also approved to treat idiopathic hypersomnia in adults[18,19]
 – LXB has the same active moiety as SXB but with 92% less sodium[7,18-23]

Table 1. Approximate Sodium Amounts at the Recommended Nightly Dosage of SXB or LXB

Oxybate	Recommended Adult Dosage	Sodium Amount per Nightly Exposure	% of 2300 mg Upper Daily Recommended Sodium Intake[12]
SXB[19]	6–9 g/night	1100–1640 mg	48–71%
LXB[7,18,23]	6–9 g/night	87–131 mg	4–6%

LXB, low-sodium oxybate; SXB, high-sodium oxybate.

Objective

- XYLO (NCT05869773) evaluated changes in ambulatory and seated resting ("office") BP changes in study participants with narcolepsy after switching from twice-nightly SXB to LXB

Methods

Figure 1. Study Design

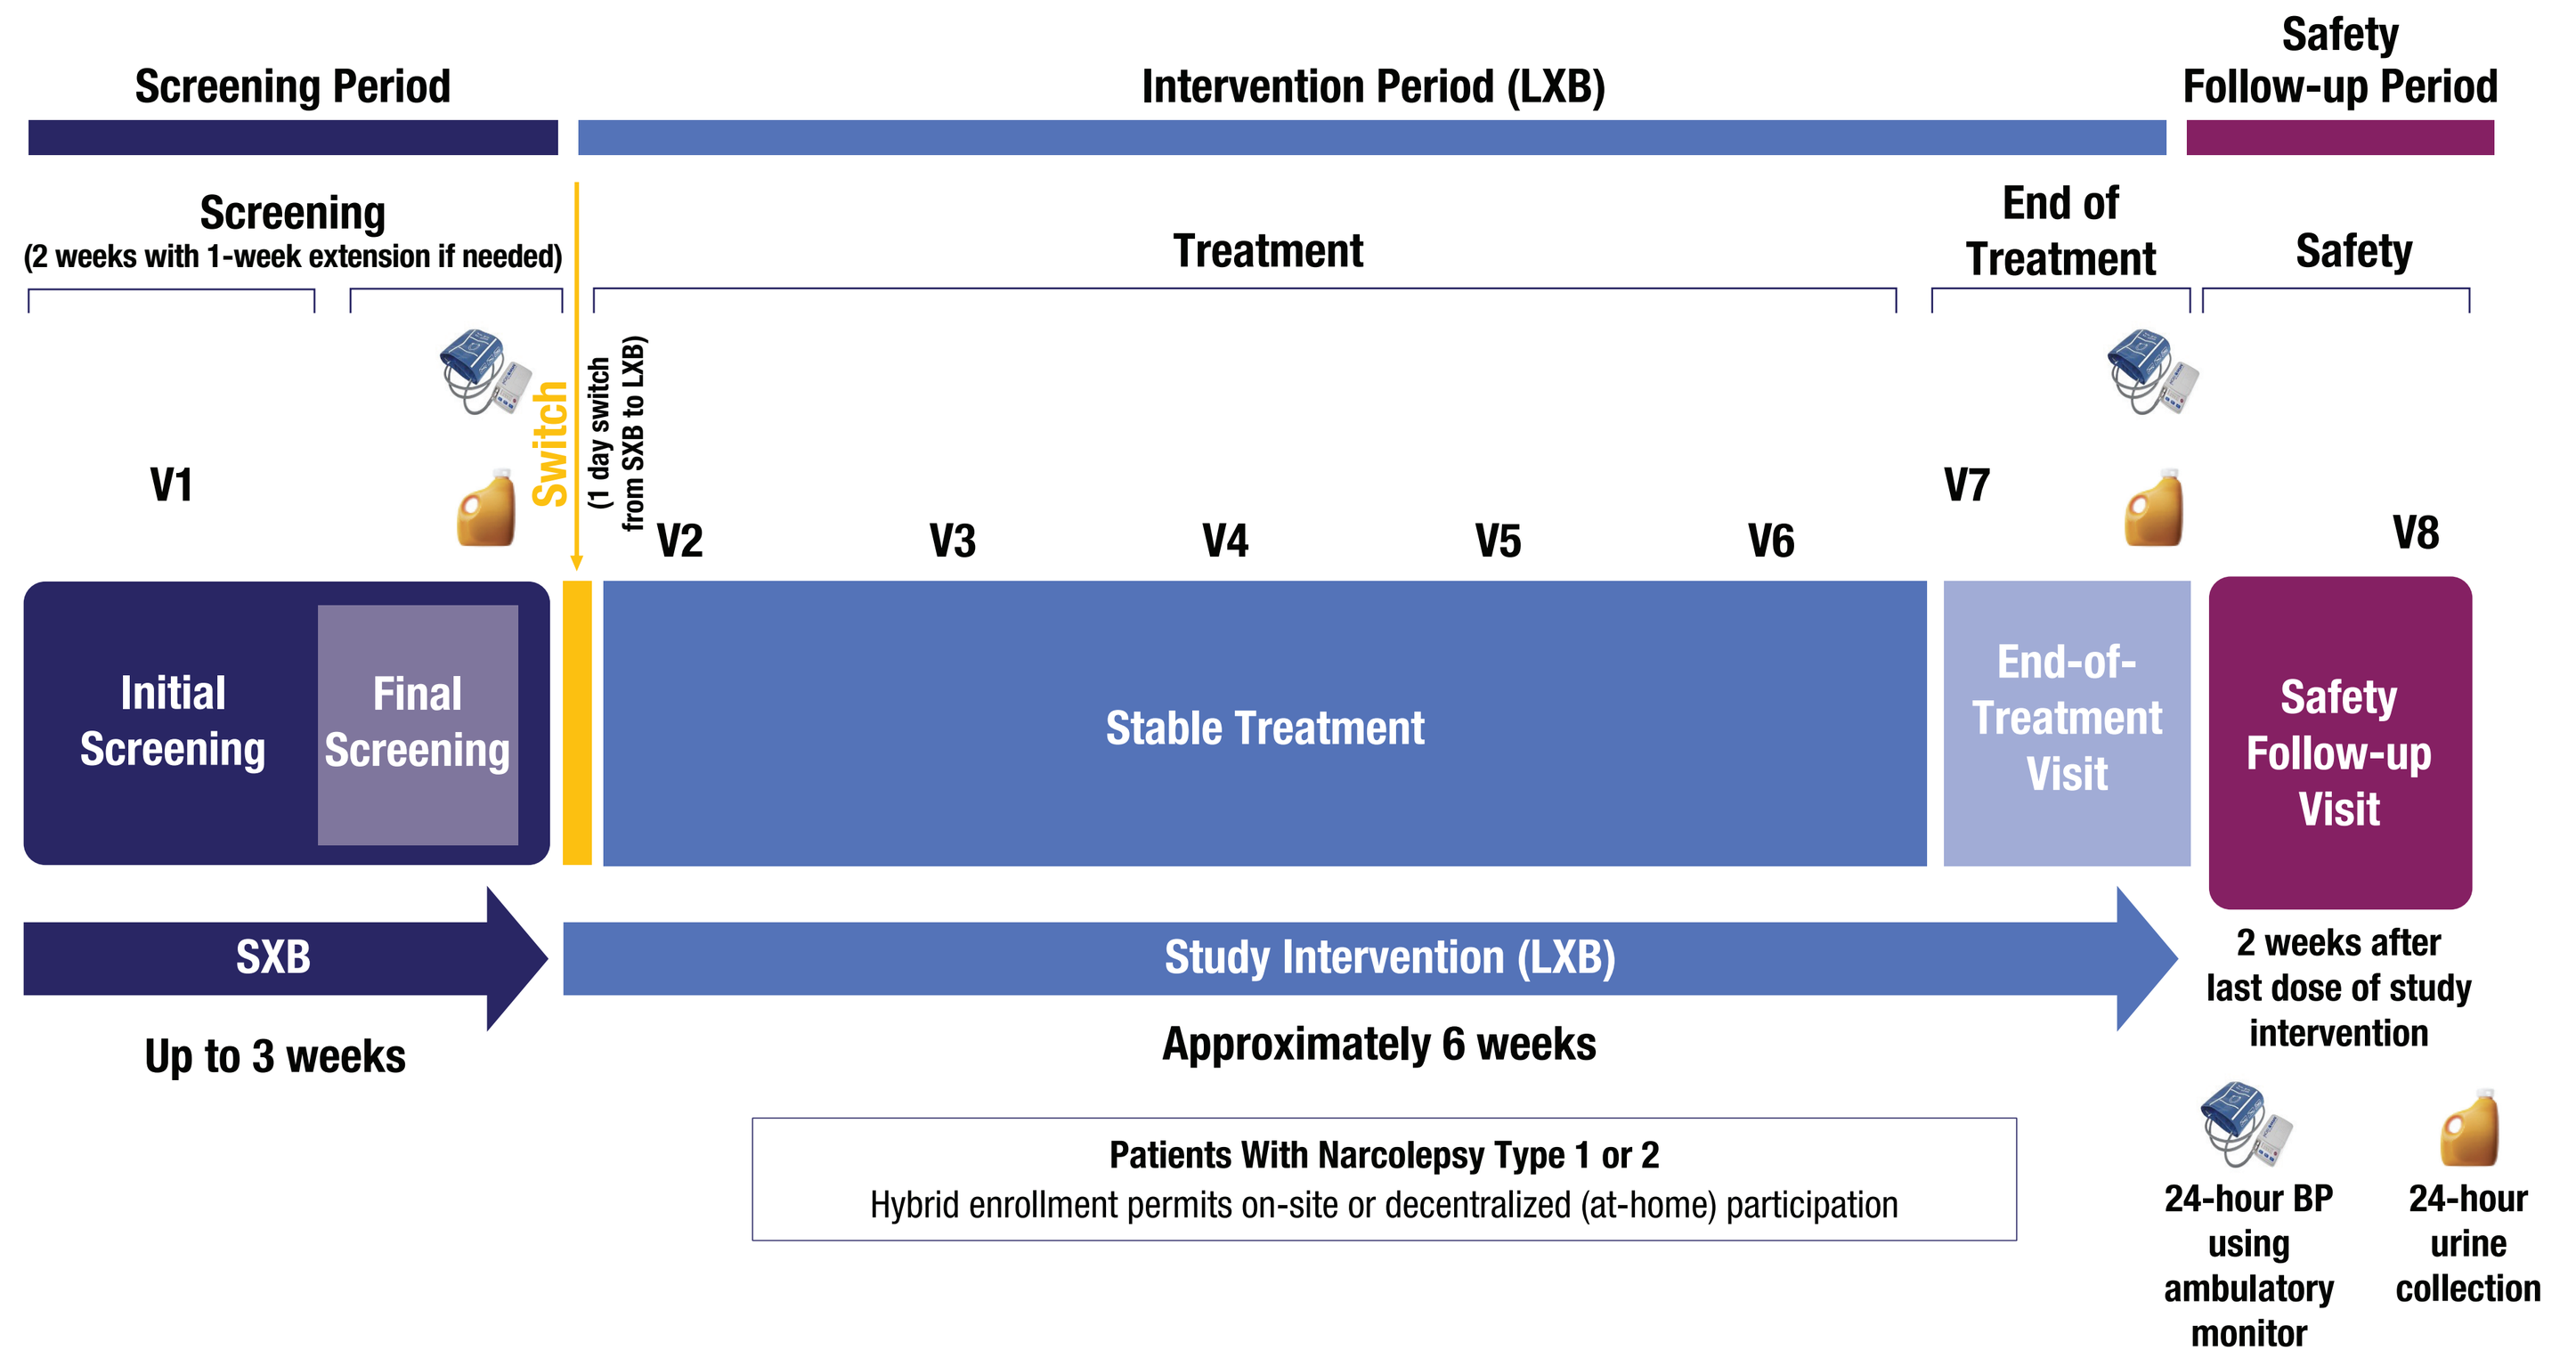

Screening Period
Intervention Period (LXB)
Safety Follow-up Period
Screening
(2 weeks with 1-week extension if needed)
Treatment
End of Treatment
Safety
Switch
(1 day switch from SXB to LXB)
V1
V2
V3
V4
V5
V6
V7
V8
Initial Screening
Final Screening
Stable Treatment
End-of-Treatment Visit
Safety Follow-up Visit
SXB
Up to 3 weeks
Study Intervention (LXB)
Approximately 6 weeks
2 weeks after last dose of study intervention
Patients With Narcolepsy Type 1 or 2
Hybrid enrollment permits on-site or decentralized (at-home) participation
24-hour BP using ambulatory monitor
24-hour urine collection

BP, blood pressure; LXB, low-sodium oxybate; SXB, high-sodium oxybate; V, visit.

- XYLO was an open-label, single-arm, multicenter, switch study conducted in the US and Europe, enrolling participants 18 to 70 years of age with narcolepsy (type 1 or 2) taking 6 to 9 g/night of SXB for ≥6 weeks at study entry
- A hybrid study design allowed enrollment either at an investigational site or at home (decentralized; with in-person visits from study site staff)
- Eligibility criteria included an average screening seated resting ("office") systolic blood pressure (SBP) between 130 and 155 mmHg (inclusive) and diastolic blood pressure (DBP) ≤95 mmHg (3 BP measurements at 1-minute intervals)
- Patients with severe hypertension and target organ damage were excluded
- The primary endpoint was change from baseline (on SXB) to end of treatment (EOT; on LXB) in mean 24-hour ambulatory SBP
- Key secondary endpoints were change from baseline (SXB) to EOT (LXB) in mean daytime ambulatory, seated resting ("office"), and mean nighttime ambulatory SBP
- Exploratory endpoints included changes in mean 24-hour ambulatory, daytime ambulatory, seated resting ("office"), and nighttime mean DBP, and change in 24-hour urinary sodium
- Additional details on statistical analyses can be found in the supplemental material, available by scanning the QR code in the lower right corner of this poster

Results

Figure 2. Reductions in 24-Hour Urinary Sodium From Baseline (on SXB) to EOT (on LXB) Paralleled Reduced Sodium Intake (Completer Population[a])

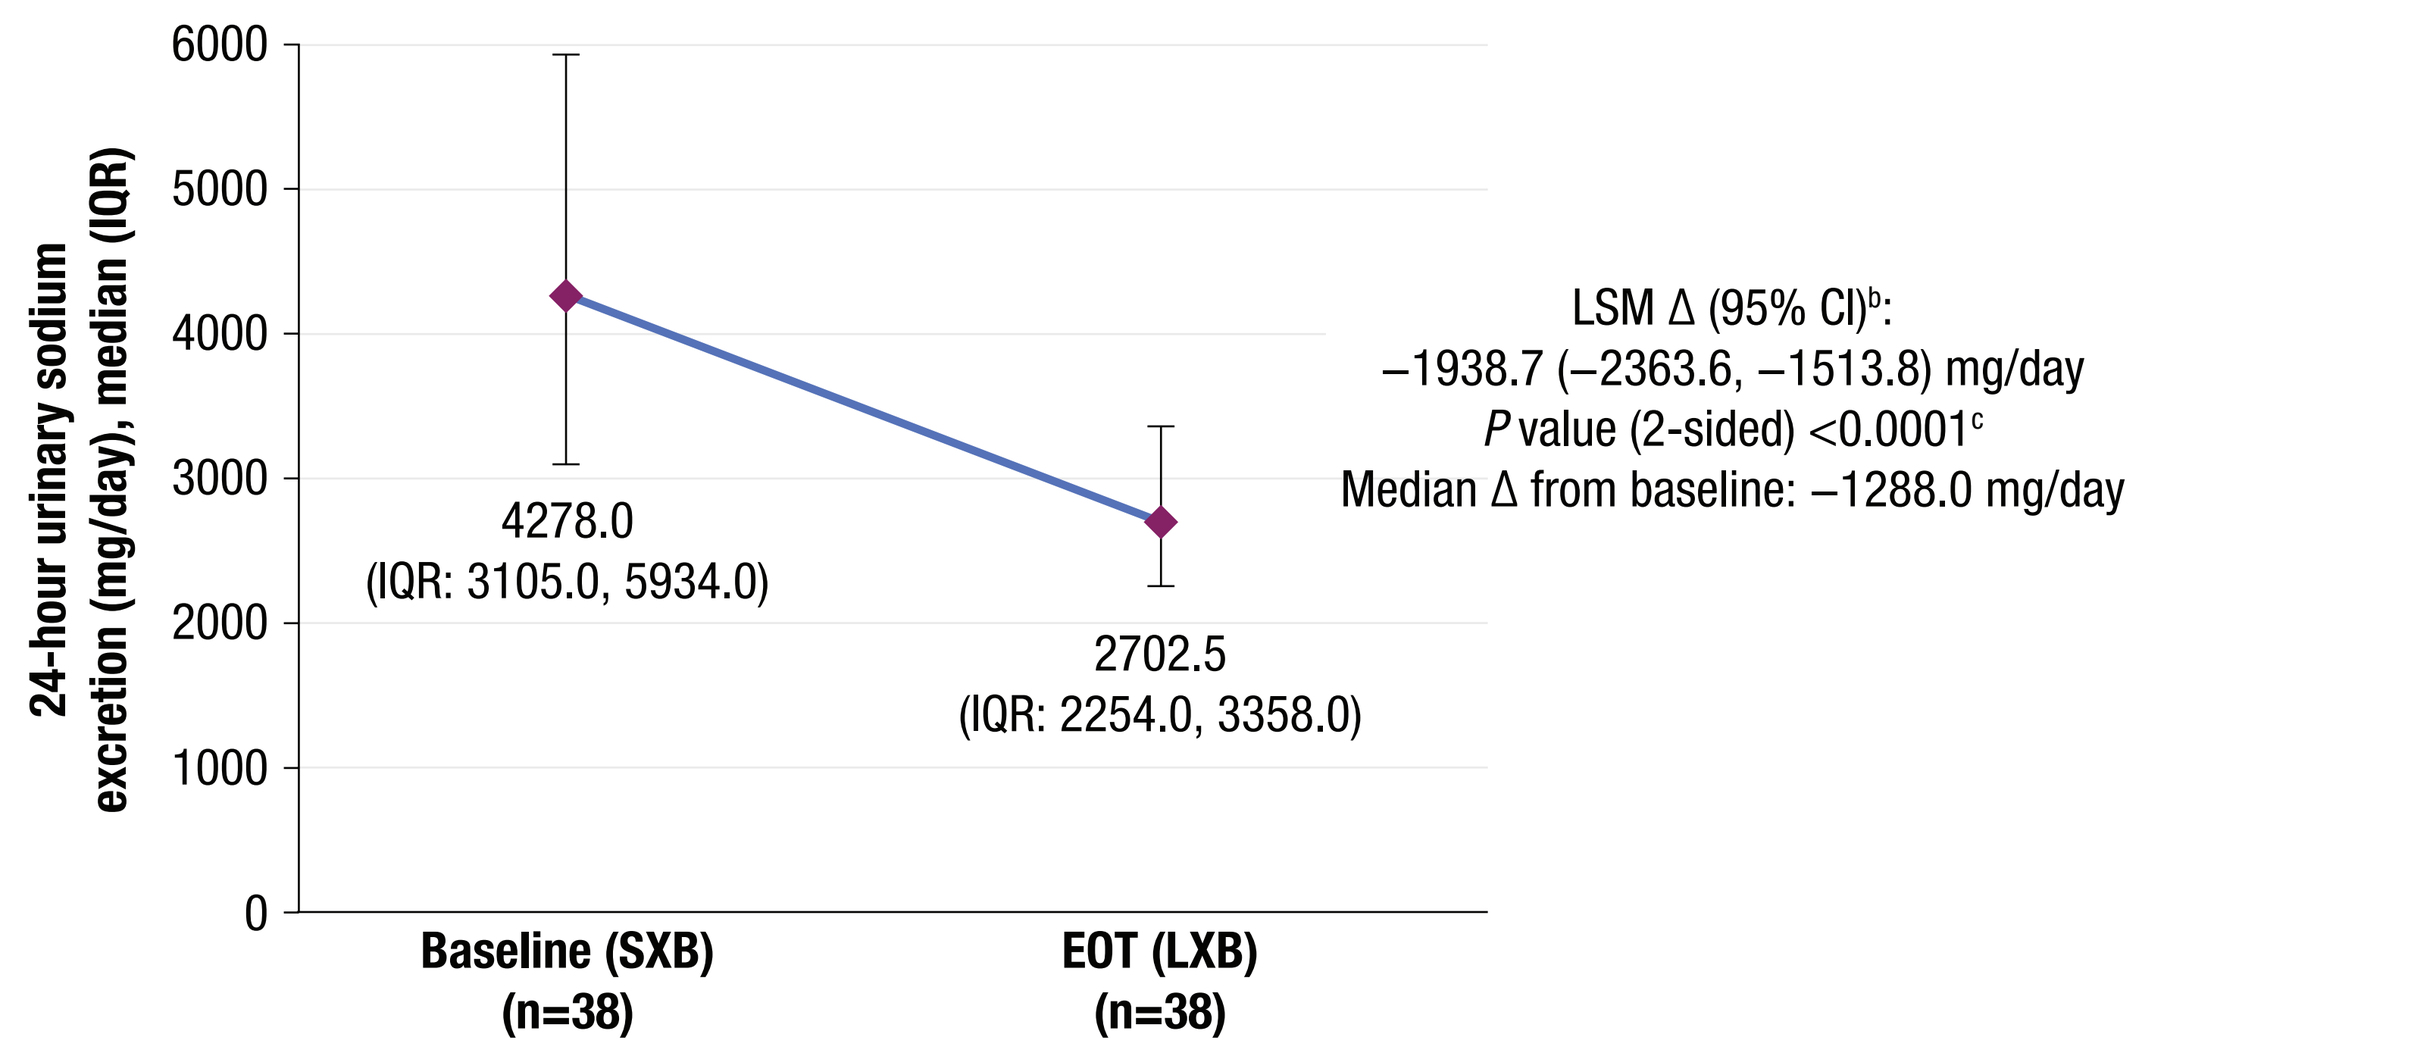

24-hour urinary sodium excretion (mg/day), median (IQR)
6000
5000
4000
3000
2000
1000
0
4278.0
(IQR: 3105.0, 5934.0)
2702.5
(IQR: 2254.0, 3358.0)
Baseline (SXB)
(n=38)
EOT (LXB)
(n=38)
LSM Δ (95% CI)[b]:
–1938.7 (–2363.6, –1513.8) mg/day
P value (2-sided) <0.0001[c]
Median Δ from baseline: –1288.0 mg/day

In the line chart, the marker is the median; error bars indicate IQR.
[a]Completer population includes all participants who completed the EOT visit after 6 weeks of LXB with a valid 24-hour blood pressure recording. [b]Baseline-adjusted change from baseline to EOT.
[c]P value is nominal.
CI, confidence interval; EOT, end of treatment; IQR, interquartile range; LSM, least squares mean; LXB, low-sodium oxybate; SXB, high-sodium oxybate.

- Mean (SD) total SXB and LXB dosages were 8.0 (1.1) and 8.1 (1.1) g/night, respectively, representing a mean (SD) sodium content of 1456.5 (206.2) and 117.8 (16.3) mg (mean [SD] difference of 1338.8 [190.7] mg sodium)

Table 2. Demographics and Baseline Characteristics (Completer Population[a])

Parameter	Overall (N=43)
Age, years, mean (SD)	45.0 (11.3)
Female, n (%)	28 (65.1)
Race, n (%)	
White	37 (86.0)
Black or African-American	3 (7.0)
Asian	1 (2.3)
Unknown	2 (4.7)
Ethnicity, n (%)	
Hispanic or Latino	3 (7.0)
Not Hispanic or Latino	40 (93.0)
BMI,[b] kg/m², mean (SD)	30.3 (7.5)
Seated resting ("office") SBP at screening, mmHg, mean (SD)	138.0 (5.7)
Seated resting ("office") DBP at screening, mmHg, mean (SD)	85.2 (6.6)
Use of antihypertensive medications, n (%)	14 (32.6)
Narcolepsy type, n (%)	
Type 1	21 (48.8)
Type 2	22 (51.2)

[a]Completer population includes all participants who completed the EOT visit after 6 weeks of LXB with a valid 24-hour blood pressure recording. [b]BMI data was available for 41 participants.
BMI, body mass index; DBP, diastolic blood pressure; EOT, end of treatment; LXB, low-sodium oxybate; SBP, systolic blood pressure; SD, standard deviation.

- Per the prespecified study design, 43 participants with valid EOT 24-hour ambulatory SBP recordings were analyzed; mean age was 45 years, 65% were female, 86% were White, and 33% were on stable antihypertensives

Figure 3. Clinically Meaningful Blood Pressure Reductions Were Observed From Baseline (on SXB) to EOT (on LXB) (Completer Population[a])

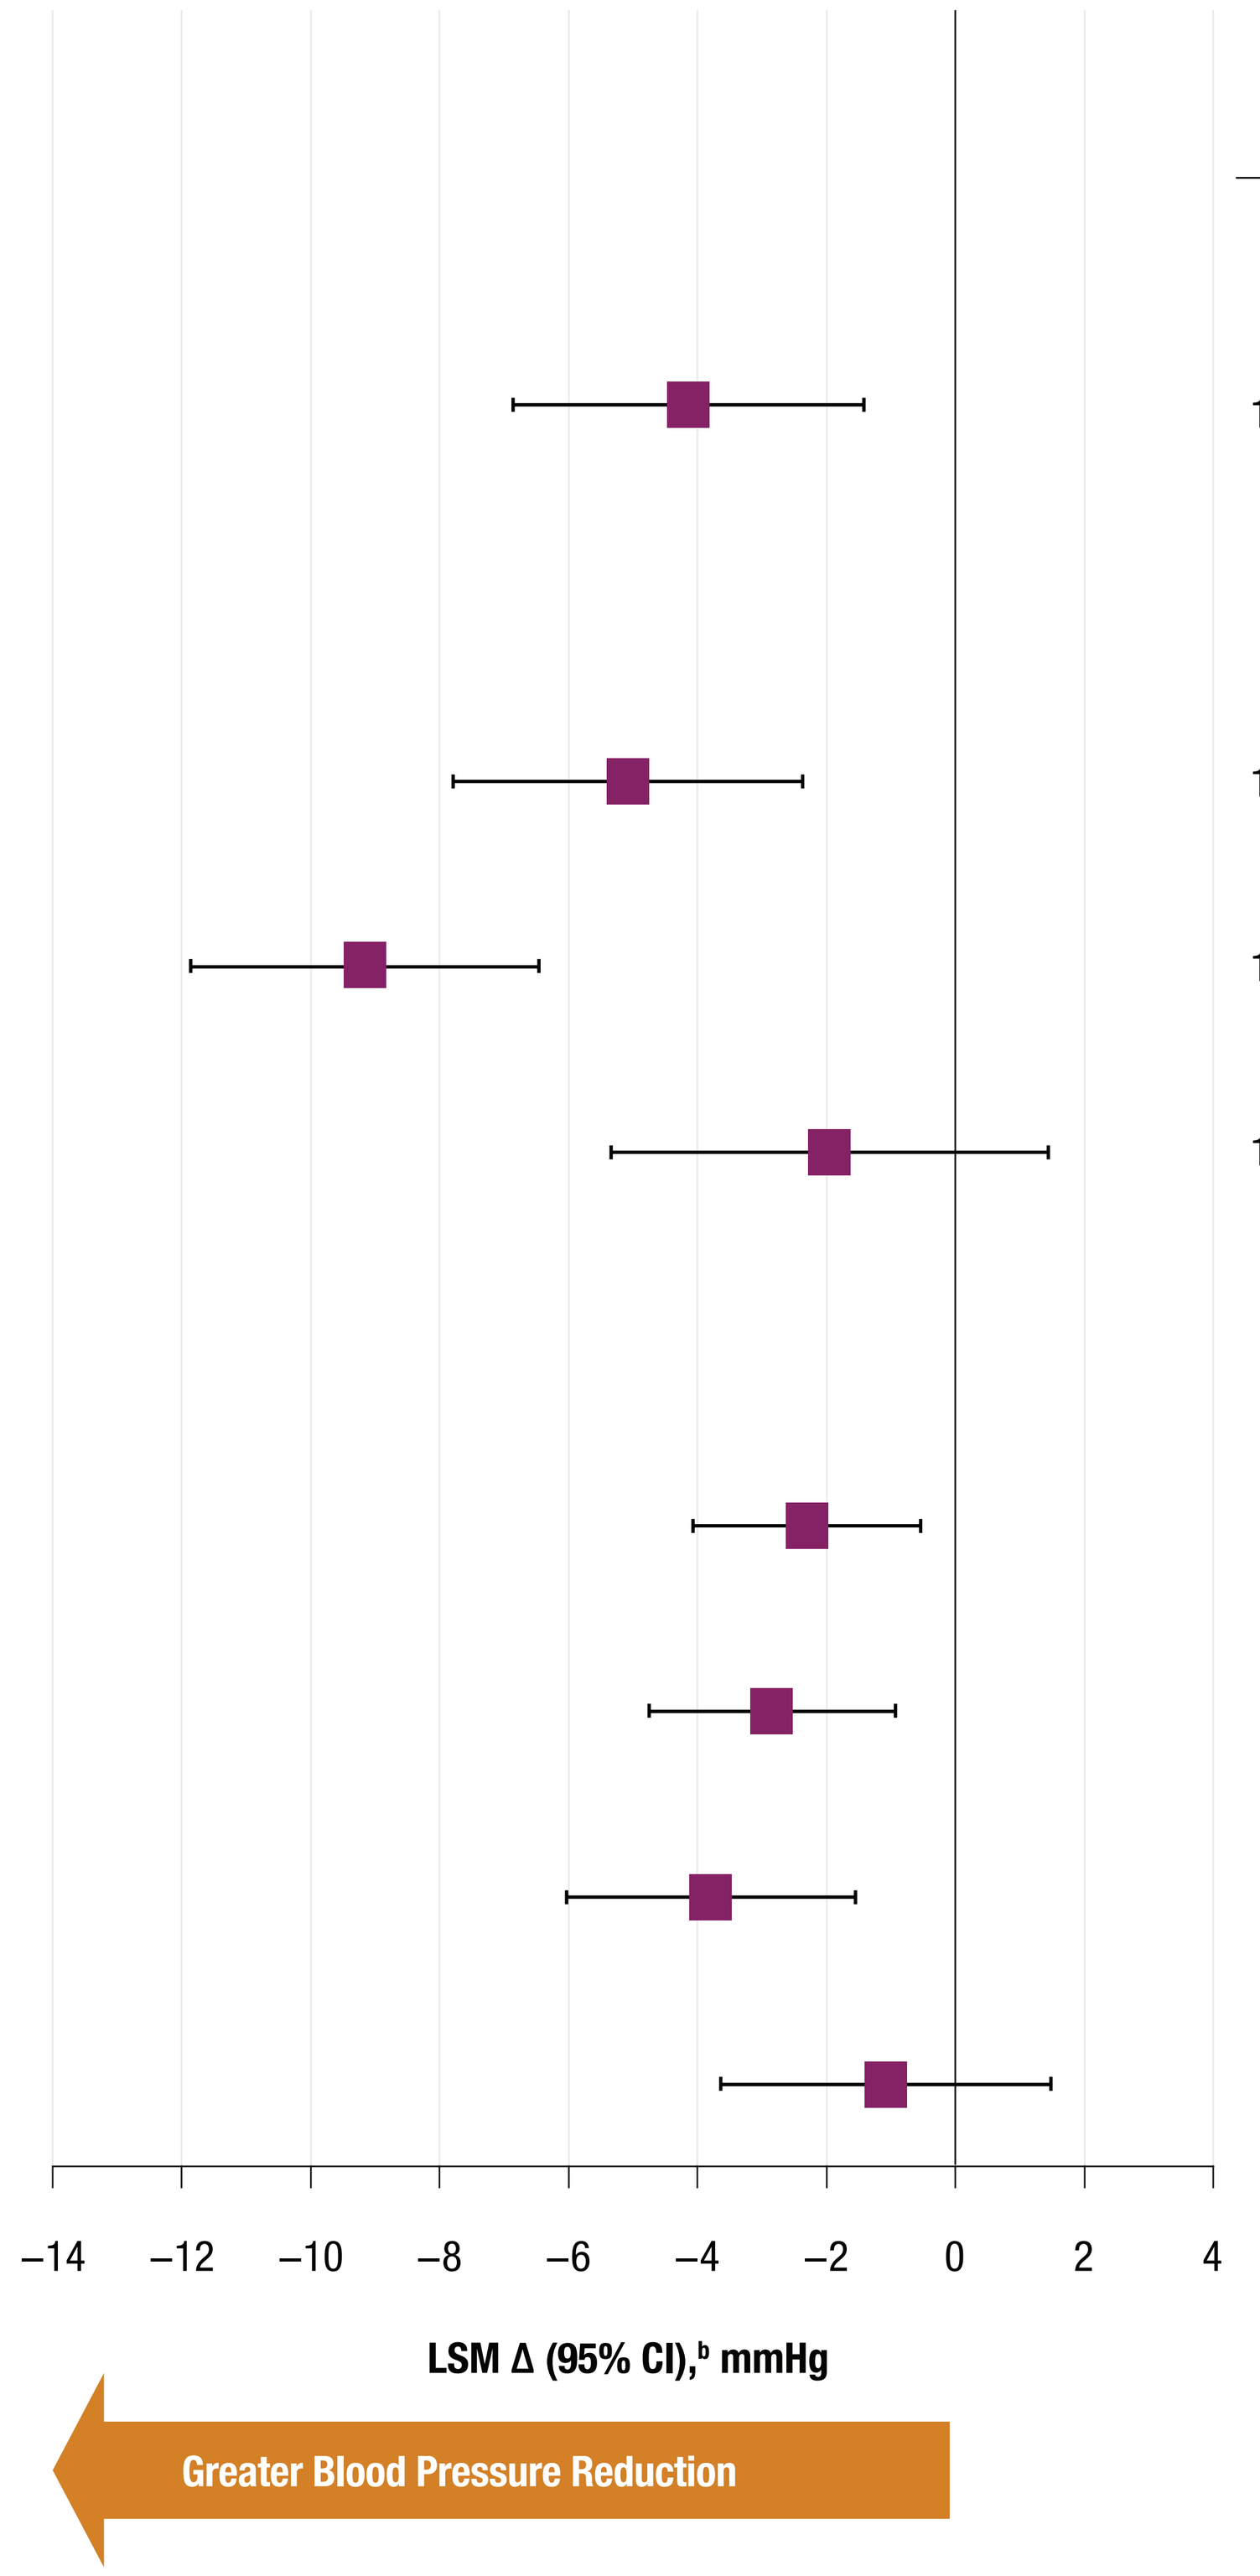

–14
–12
–10
–8
–6
–4
–2
0
2
4
LSM Δ (95% CI),[b] mmHg
Greater Blood Pressure Reduction

Endpoint	BL (SXB) (N=43) Mean (SE), mmHg	EOT (LXB) (N=43) Mean (SE), mmHg	LSM Δ (95% CI),[b] mmHg	P value 1-sided	P value 2-sided
Primary Endpoint					
24-hour ambulatory SBP	132.3 (1.8)	128.2 (1.8)	−4.1 (−6.9, −1.4)	0.0019*	0.0038
Secondary Endpoints					
Daytime ambulatory SBP	139.1 (1.9)	134.1 (1.9)	−5.1 (−7.8, −2.4)	0.0003*	0.0005
Seated resting ("office") SBP	138.0 (0.9)	128.8 (1.4)	−9.2 (−11.9, −6.5)	<0.0001*	<0.0001
Nighttime ambulatory SBP	116.9 (2.1)	114.9 (2.4)	−2.0 (−5.3, 1.4)	0.1265	0.2530
Exploratory Endpoints					
24-hour ambulatory DBP	77.3 (1.6)	75.0 (1.4)	−2.3 (−4.1, −0.5)		0.0118[c]
Daytime ambulatory DBP	82.7 (1.6)	79.8 (1.5)	−2.8 (−4.8, −0.9)		0.0045[c]
Seated resting ("office") DBP	85.2 (1.0)	81.4 (1.5)	−3.8 (−6.0, −1.6)		0.0014[c]
Nighttime ambulatory DBP	65.2 (1.8)	64.1 (1.9)	−1.1 (−3.6, 1.5)		0.3993[c]

The marker is the LSM change, and the error bars indicate the 95% CI.
[a]Completer population includes all participants who completed the EOT visit after 6 weeks of LXB with a valid 24-hour blood pressure recording. [b]Baseline-adjusted change in BP from BL (on SXB) to EOT (on LXB). [c]P values are nominal. *Indicates statistical significance.
BL, baseline; BP, blood pressure; CI, confidence interval; DBP, diastolic blood pressure; EOT, end of treatment; LSM, least squares mean; LXB, low-sodium oxybate; SBP, systolic blood pressure; SE, standard error; SXB, high-sodium oxybate.

- LSM (95% CI) change in 24-hour ambulatory SBP (primary endpoint) was −4.1 (−6.9, −1.4; P=0.0019) mmHg, achieving the interim (75%) prespecified efficacy stopping criteria
- The proportion of participants with office SBP <130 mmHg at baseline was 0% (per eligibility criterion) and increased to 47% at EOT; the proportion of participants with 24-hour ambulatory SBP <130 mmHg also increased from baseline (35%) to EOT (56%)

Table 3. Treatment-Emergent Adverse Events (Safety Population)

Parameter, n (%)	Overall (N=67)
TEAE[a]	27 (40.3)
Mild	21 (31.3)
Moderate	6 (9.0)
TEAE related to study drug	10 (14.9)
Serious TEAE	0[b]
Participants with ≥1 TEAE leading to discontinuation of LXB	0
Participants with ≥1 TEAE leading to dose change of LXB	0
Common TEAEs (occurring in ≥1 participant)	
Upper respiratory tract infection	3 (4.5)
Dysgeusia	2 (3.0)
Gastroenteritis	2 (3.0)
Nausea	2 (3.0)
Vomiting	2 (3.0)

[a]No TEAEs were severe, life-threatening, or fatal. [b]A serious adverse event of "hospitalization-unknown cause" was reported for 1 participant who was lost to follow-up and which occurred after the last known dose of study drug, which did not meet the definition of a TEAE.
LXB, low-sodium oxybate; TEAE, treatment-emergent adverse event.

- Adverse events occurred in 40.3% of participants in the safety population
 – All adverse events were mild or moderate in severity; all adverse events considered related to the study drug were mild in severity

Conclusions

- Switching from SXB to LXB in the XYLO study reduced daily treatment-related sodium intake by mean (SD) of 1338.8 (190.7) mg, which was paralleled by 24-hour urinary sodium reduction and associated with clinically meaningful reductions in 24-hour ambulatory and seated resting ("office") SBP
- Limitations of XYLO include the open-label, single-arm design, which affects ability to establish causality
- Reducing sodium intake from medications has important implications for BP, consistent with the robust and well-established published body of evidence on dietary and medication-related sodium reduction[1-4,12,13,24-28]
- Given the increased risk and burden of CV conditions for people with narcolepsy,[7-11] these BP results help inform individuals living with narcolepsy and their healthcare providers when assessing oxybate treatment options

References: 1. Jones DW, et al. *Hypertension.* 2025;82:e00–e00 [Online ahead of print]. **2.** National Academies of Sciences, Engineering, and Medicine (2019). Dietary reference intakes for sodium and potassium. Washington, DC: The National Academies Press. https://doi.org/10.17226/25353. **3.** US Food and Drug Administration. Voluntary sodium reduction goals: Target mean and upper bound concentrations for sodium in commercially processed, packaged, and prepared foods (Edition 2): Guidance for industry. 2024. https://www.fda.gov/regulatory-information/search-fda-guidance-documents/draft-guidance-industry-voluntary-sodium-reduction-goals-edition-2. **4.** Strazzullo P, et al. *BMJ.* 2009;339:b4567. **5.** Mayne ST, et al. *JAMA.* 2021;326(17):1675-1676. **6.** FDA.gov. Sodium Reduction in the Food Supply. https://www.fda.gov/food/nutrition-food-labeling-and-critical-foods/sodium-reduction-food-supply. **7.** Bogan RK, et al. *Sleep.* 2021;44(3):zsaa206. **8.** Black J, et al. *Sleep Med.* 2017;33:13-18. **9.** Ben-Joseph RH, et al. *Sleep.* 2023;46(10):zsad161. **10.** Bosco A, et al. *Neurology.* 2018;90(6):e479-e491. **11.** Jennum PJ, et al. *Sleep Med Rev.* 2021;58:101440. **12.** US Department of Agriculture, US Department of Health and Human Services. Dietary guidelines for Americans, 2020-2025. 2020. https://www.dietaryguidelines.gov/resources/2020-2025-dietary-guidelines-online-materials. **13.** US Food and Drug Administration. Sodium reduction. 2021. https://www.fda.gov/food/food-additives-petitions/sodium-reduction. **14.** American Heart Association. How much sodium should I eat per day? 2024. https://www.heart.org/en/healthy-living/healthy-eating/eat-smart/sodium/how-much-sodium-should-i-eat-per-day. **15.** Arnett DK, et al. *J Am Coll Cardiol.* 2019;74(10):e177-e232. **16.** World Health Organization. Guideline: sodium intake for adults and children. 2012. https://www.who.int/publications/i/item/9789241504836. **17.** World Health Organization. WHO global report on sodium intake reduction. 2023. https://www.who.int/publications/i/item/9789240069985. **18.** Xywav® (calcium, magnesium, potassium, and sodium oxybates) oral solution, CIII [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **19.** Xyrem® (sodium oxybate) oral solution, CIII [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **20.** Szarfman A, et al. *N Engl J Med.* 1995;333(19):1291. **21.** US Food and Drug Administration. Clinical review for Binosto, NDA 202344. 2012. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2012/202344Orig1s000MedR.pdf. **22.** US Food and Drug Administration. Quantitative labeling of sodium, potassium, and phosphorus for human over-the-counter and prescription drug products. Guidance for industry. 2022. https://www.fda.gov/regulatoryinformation/search-fda-guidance-documents/quantitative-labeling-sodium-potassium-and-phosphorus-human-over-counter-and-prescription-drug. **23.** Chen C, et al. *Clin Transl Sci.* 2021;14(6):2278-2287. **24.** Ma Y, et al. *N Engl J Med.* 2022;386(3):252-263. **25.** Filippini T, et al. *Circulation.* 2021;143(16):1542-1567. **26.** Mozaffarian D, et al. *N Engl J Med.* 2014;371(7):624-634. **27.** Benitez-Camps M, et al. *J Hypertens.* 2018;36(8):1656-1662. **28.** Ubeda A, et al. *Pharmacoepidemiol Drug Saf.* 2009;18(5):417–419.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees Kalpana Vijayan, PhD, and Stephanie Phan, PharmD, provided medical writing support and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: VK Somers is a consultant for ApniMed, Axsome, Lilly, Mineralys Therapeutics, and Jazz Pharmaceuticals, and serves on the Sleep Number Scientific Advisory Board. **RJ Kovacs** is a consultant to Lilly and Jazz Pharmaceuticals. He serves on data and safety monitoring boards for Immunovant and Relmada and on clinical events committees for Cook and Zydus. **JK Alexander, C Baranak, DA Nichols, J Dai,** and **M Whalen** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **A Ajayi** is a consultant to Jazz Pharmaceuticals. He has participated in scientific and marketing advisory boards for Avadel and Jazz Pharmaceuticals. Dr Ajayi has been a site principal investigator for narcolepsy and studies sponsored by Avadel, Centessa, Harmony Biosciences, Jazz Pharmaceuticals, and Takeda. **B Hutchinson** is a consultant to CardioOne and Jazz Pharmaceuticals. **Y Dauvilliers** is a consultant for and has participated in advisory boards for Avadel, Bioprojet, Centessa, Harmony Biosciences, Idorsia, Jazz Pharmaceuticals, and Takeda. **WB White** is a cardiovascular safety consultant to Jazz Pharmaceuticals.

Scan this code to access this poster online. This code is not for promotional purposes.

Presented at Psych Congress 2025; September 17–25, 2025; San Diego, CA, USA

129

Evaluating the Impact of Switching From High-Sodium Oxybate to Low-Sodium Oxybate on Ambulatory Blood Pressure in People With Narcolepsy

Virend K. Somers, MD, PhD[1]; Richard J. Kovacs, MD[2]; Jessica K. Alexander, PhD[3]; Christine Baranak, MA[3]; Deborah A. Nichols, MS[4]; Jing Dai, PhD[4]; Marisa Whalen, PharmD[4]; Akinyemi Ajayi, MD[5]; Barbara Hutchinson, MD, PhD[6]; Yves Dauvilliers, MD[7]; William B. White, MD[8]

[1]Mayo Clinic, Rochester, MN, USA; [2]Division of Cardiovascular Medicine, Indiana University School of Medicine, Indianapolis, IN, USA; [3]Jazz Pharmaceuticals, Philadelphia, PA, USA; [4]Jazz Pharmaceuticals, Palo Alto, CA, USA; [5]Children's Lung, Asthma and Sleep Specialists, Winter Park, FL, USA; [6]Chesapeake Cardiac Care, Bowie, MD, USA; [7]University of Montpellier, Montpellier, France; [8]Calhoun Cardiology Center, University of Connecticut School of Medicine, Farmington, CT, USA

Supplemental Methods

- Statistical analyses
 - A 2-stage group sequential design with an adaptive sample size was used, targeting a sample size of 57 completers for 90% power to detect a mean decrease in 24-hour SBP of 3.5 mmHg with a standard deviation of 8.0 mmHg from baseline (SXB) to EOT (LXB)
 - An interim analysis was planned after 43 (75%) participants completed a valid 24-hour ambulatory SBP recording at the EOT visit after 6 weeks of LXB and the study recruitment would end if the primary endpoint significance level (O'Brien-Fleming one-sided alpha level of 0.00998) was met
 - Least squares mean (LSM), 95% confidence interval (CI), and *P* values were obtained from an analysis of covariance model of the change for each endpoint from the baseline to the EOT, adjusted for the baseline value
 - To account for multiplicity secondary to multiple endpoints, a fixed hierarchical testing method was prespecified and tested sequentially for change in mean SBP following 6 weeks of LXB after switching from SXB in the following order: 24-hour ambulatory SBP, daytime ambulatory SBP, seated resting ("office") SBP, and nighttime ambulatory SBP; other endpoints were not adjusted for multiplicity and are therefore considered nominal
 - One-sided *P* values were reported for baseline-adjusted SBP LSM (95% CI) changes
 - Two-sided *P* values were reported for baseline-adjusted DBP and 24-hour urinary sodium LSM (95% CI) changes
- Study populations:
 - **Completer population:** all participants who completed a valid (ie, meeting minimal data standards) 24-hour ambulatory BP recording at the EOT visit after 6 weeks of LXB
 - **Safety population:** all participants who took ≥1 dose of LXB by the conclusion of the study