128 Impact of Sleep Inertia on Cognition in People With Idiopathic Hypersomnia

Sarah C. Markt, ScD, MPH[1]; Jessica K. Alexander, PhD[1]; Marisa Whalen, PharmD[2]; Maureen H. Carlyle, MPH[3]; John C. White, DPM, MS[3]; Laura L. Dick, MPH[3]; Cristina Casstevens, PhD[1]; Silky Beaty, PharmD, MSPH[1]; Alyssa Cairns, PhD[2]; Caroleen Drachenberg, PhD, MSPH[1]; Chad M. Ruoff, MD[4]

[1]Jazz Pharmaceuticals, Palo Alto, CA; [2]Jazz Pharmaceuticals, Philadelphia, PA; [3]Optum, Eden Prairie, MN; [4]Mayo Clinic, Scottsdale, AZ

Introduction

- Idiopathic hypersomnia is a central disorder of hypersomnolence characterized by several key clinical features, including excessive daytime sleepiness, patient-reported nonrestorative sleep which is often prolonged, and sleep inertia[1,2]
- Sleep inertia is characterized by difficulty waking up with repeated returns to sleep, irritability, automatic behavior, and cognitive impairments including confusion[1,3,4]
- The cognitive impacts of sleep inertia among people with idiopathic hypersomnia remain underexplored

Objective

- To describe the impacts of sleep inertia on cognition in people with idiopathic hypersomnia

Methods

- This observational, cross-sectional, direct-to-patient survey used administrative claims from the Optum Research Database (1/1/2019–5/31/2024) to identify commercially insured adults
- Eligible participants were adults aged ≥18 years with at least 1 medical claim with a diagnosis code for idiopathic hypersomnia (International Classification of Diseases, Tenth Revision, Clinical Modification [ICD-10-CM]: G47.11, G47.12) during the identification period
 - Individuals with a medical claim with a diagnosis for narcolepsy (ICD-10-CM: G47.411, G47.421) on or after their most recent idiopathic hypersomnia claim; or who had a medical claim with a diagnosis for cognitive impairment (ICD-10-CM: R41.9) within 6 months before sample identification; or who were not enrolled in a health plan that allowed for patient identification were excluded
- Eligible individuals were mailed a survey comprising validated assessments and de novo questions
 - Findings from this survey related to the prevalence and severity of sleep inertia have been previously presented[5]; this analysis focuses on the cognitive impacts of sleep inertia
- Data were analyzed using descriptive statistics

Table 1. Participant Survey Assessments

Assessment	Description	Rating Scale for Each Item	Total Score Range	Severity Categories
IHSS	• 14 items; validated[6]	• 0–3 or 0–4	• 0–50	• Mild (0–12) • Moderate (13–25) • Severe (26–38) • Very severe (39–50)
SIQ	• 21 rated items[a]; validated[b,7] • 4 subscales: behavioral, physiological, cognitive, and emotional[8]	• 1 ("Not at all") to 5 ("All the time")	• 21–105 • SI is defined as an SIQ score ≥42	• No SI (21–41) • Mild (42–61) • Moderate (62–72) • Severe (73–84) • Very severe (85–105)
Impact of SI on Daily Life	• De novo	• 0 ("No impact") to 4 ("High impact")	• NA	• NA

[a]Two non-scored items are included for descriptive purposes, and ask, "How long does it take you to 'come to' in the morning?" and "How many days per week is this the case?" [b]The SIQ was validated in depression[7]; SIQ has been assessed in idiopathic hypersomnia and has been found to distinguish between individuals with hypersomnolence and healthy controls.[8]
IHSS, Idiopathic Hypersomnia Severity Scale; NA, not applicable; SI, sleep inertia; SIQ, Sleep Inertia Questionnaire.

References: 1. American Academy of Sleep Medicine. *International Classification of Sleep Disorders – Third Edition*, Text Revision. Darien, IL: American Academy of Sleep Medicine; 2023. **2.** Trotti LM, Arnulf I. *Neurotherapeutics*. 2021;18(1):20-31. **3.** Arnulf I, et al. *Sleep Med Rev*. 2023;69:101766. **4.** Stevens J, et al. *Nat Sci Sleep*. 2023;15:593-606. **5.** Alexander J, et al. Prevalence and Severity of Sleep Inertia Among Individuals With Idiopathic Hypersomnia. presented at: SLEEP 2025, the 39th Annual Meeting of the Associated Professional Sleep Societies, LLC (APSS); 2025; Seattle. **6.** Dauvilliers Y, et al. *Neurology*. 2019;92(15):e1754-e1762. **7.** Kanady JC, Harvey AG. *Cognit Ther Res*. 2015;39(5):601-612. **8.** Sung ER, et al. *Sleep Med*. 2024;121:352-358. **9.** Carlyle M, et al. *Value Health*. 2023;26(6 suppl):S295. **10.** Rassu AL, et al. *J Clin Sleep Med*. 2022;18(2):617-629.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees Joseph Mansonet, MPH, and Eleanor Bush, MA, provided medical writing support and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: SC Markt, JK Alexander, M Whalen, C Casstevens, S Beaty, and **C Drachenberg** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **MH Carlyle, JC White,** and **LL Dick** are full-time employees of Optum who were contracted by Jazz to implement the study described in this abstract. **A Cairns** is a full-time contract employee and advisory board member for Jazz Pharmaceuticals, plc. **CM Ruoff** has served as an advisory board member for Alkermes, Eisai, Jazz Pharmaceuticals, and Takeda, and has received grant funding from Jazz Pharmaceuticals.

Results

Table 2. Participant-reported Demographic Characteristics

Characteristic	Participants (N=242)
Age at survey completion, years, mean (SD)	41.9 (13.1)
Gender, n (%)	
Female	191 (78.9)
Male	47 (19.4)
Nonbinary	3 (1.2)
Prefer not to answer	1 (0.4)
Race,[a] n (%)	
American Indian or Alaska Native	2 (0.8)
Asian	3 (1.2)
Native Hawaiian or Pacific Islander	0
Black or African American	11 (4.6)
White	227 (93.8)
Other[b]	6 (2.5)
Ethnicity, n (%) (n=239)	
Hispanic or Latino/a	12 (5.0)
Not Hispanic or Latino/a	227 (95.0)
Marital status, n (%)	
Single, never married	59 (24.4)
Living with partner	25 (10.3)
Separated	2 (0.8)
Married	129 (53.3)
Divorced	24 (9.9)
Widowed	3 (1.2)
Highest education level, n (%)	
Some high school	0
High school graduated or GED equivalent	17 (7.0)
Some college, but no degree	49 (20.2)
2-year college	25 (10.3)
4-year college	86 (35.5)
Graduate school	65 (26.9)
Employment status,[a] n (%) (n=241)	
Working full time	171 (71.0)
Working part time	38 (15.8)
Disability or medical leave	3 (1.2)
Student	20 (8.3)
Retired	10 (4.1)
Unemployed	15 (6.2)
Calculated BMI categories, n (%)	
Underweight (<18.5 kg/m²)	4 (1.7)
Normal weight (18.5 to <25.0 kg/m²)	75 (31.0)
Overweight (25.0 to <30.0 kg/m²)	85 (35.1)
Obese (≥30 kg/m²)	78 (32.2)

[a]Participants may select >1 option. [b]Includes multiple races.
BMI, body mass index; GED, General Education Development; SD, standard deviation.

- Of 1292 eligible individuals who were mailed a survey, 242 completed it; the overall response rate was 18.9%[9]

Table 3. Participant-reported Idiopathic Hypersomnia Diagnosis Information

Characteristic	Participants (N=242)
Age when first told by HCP that you have IH, years, mean (SD) (n=211)	32.2 (11.5)
Number of years since first IH diagnosis, years, mean (SD) (n=211)	8.7 (7.4)
Prescription medicine use within last 30 days to treat IH,[a] n (%)	180 (74.4)
Tests used to diagnose IH,[b] n (%) (n=228)	
Both PSG and an MSLT	165 (72.4)
24-hour sleep study (at home or in a sleep lab)	49 (21.5)
Sleep monitoring device worn on wrist for ≥7 days (actigraphy)	15 (6.6)
PSG (in a sleep lab)	204 (89.5)
MSLT (in a sleep lab)	168 (73.7)
Either PSG or an MSLT	207 (90.8)
Sleep diary	50 (21.9)
Another test or tool	23 (10.1)
No tests or tools were used for diagnosis	7 (3.1)
ESS score, mean (SD) (n=240)	13.4 (5.3)
IHSS score, mean (SD)	29.5 (9.6)
IHSS severity category, n (%)	
Mild	16 (6.6)
Moderate	58 (24.0)
Severe	124 (51.2)
Very severe	44 (18.2)
SIQ score, mean (SD)	69.0 (18.8)
SIQ severity category, n (%)	
None	22 (9.1)
Mild	55 (22.7)
Moderate	56 (23.1)
Severe	55 (22.7)
Very severe	54 (22.3)

[a]The most common treatment classes were stimulants (48.3%), antidepressants (45.0%), and armodafinil or modafinil (40.6%). [b]More than 1 response could be selected.
ESS, Epworth Sleepiness Scale; HCP, health care provider; IH, idiopathic hypersomnia; IHSS, Idiopathic Hypersomnia Severity Scale; MSLT, multiple sleep latency test; PSG, polysomnography; SD, standard deviation; SIQ, Sleep Inertia Questionnaire.

- Most participants (n=220; 90.9%) reported experiencing sleep inertia (defined as Sleep Inertia Questionnaire [SIQ] score ≥42)
- Nearly half of participants (n=109; 49.5%) reported severe or very severe sleep inertia (SIQ score ≥73)
- The mean (SD) ESS score was 13.4 (5.3; n=240) and mean (SD) IHSS score was 29.5 (9.6; n=242)

Figure 1. Impact of Hypersomnolence on Intellectual Functioning per IHSS Item 11, Overall and by Sleep Inertia Severity[a]

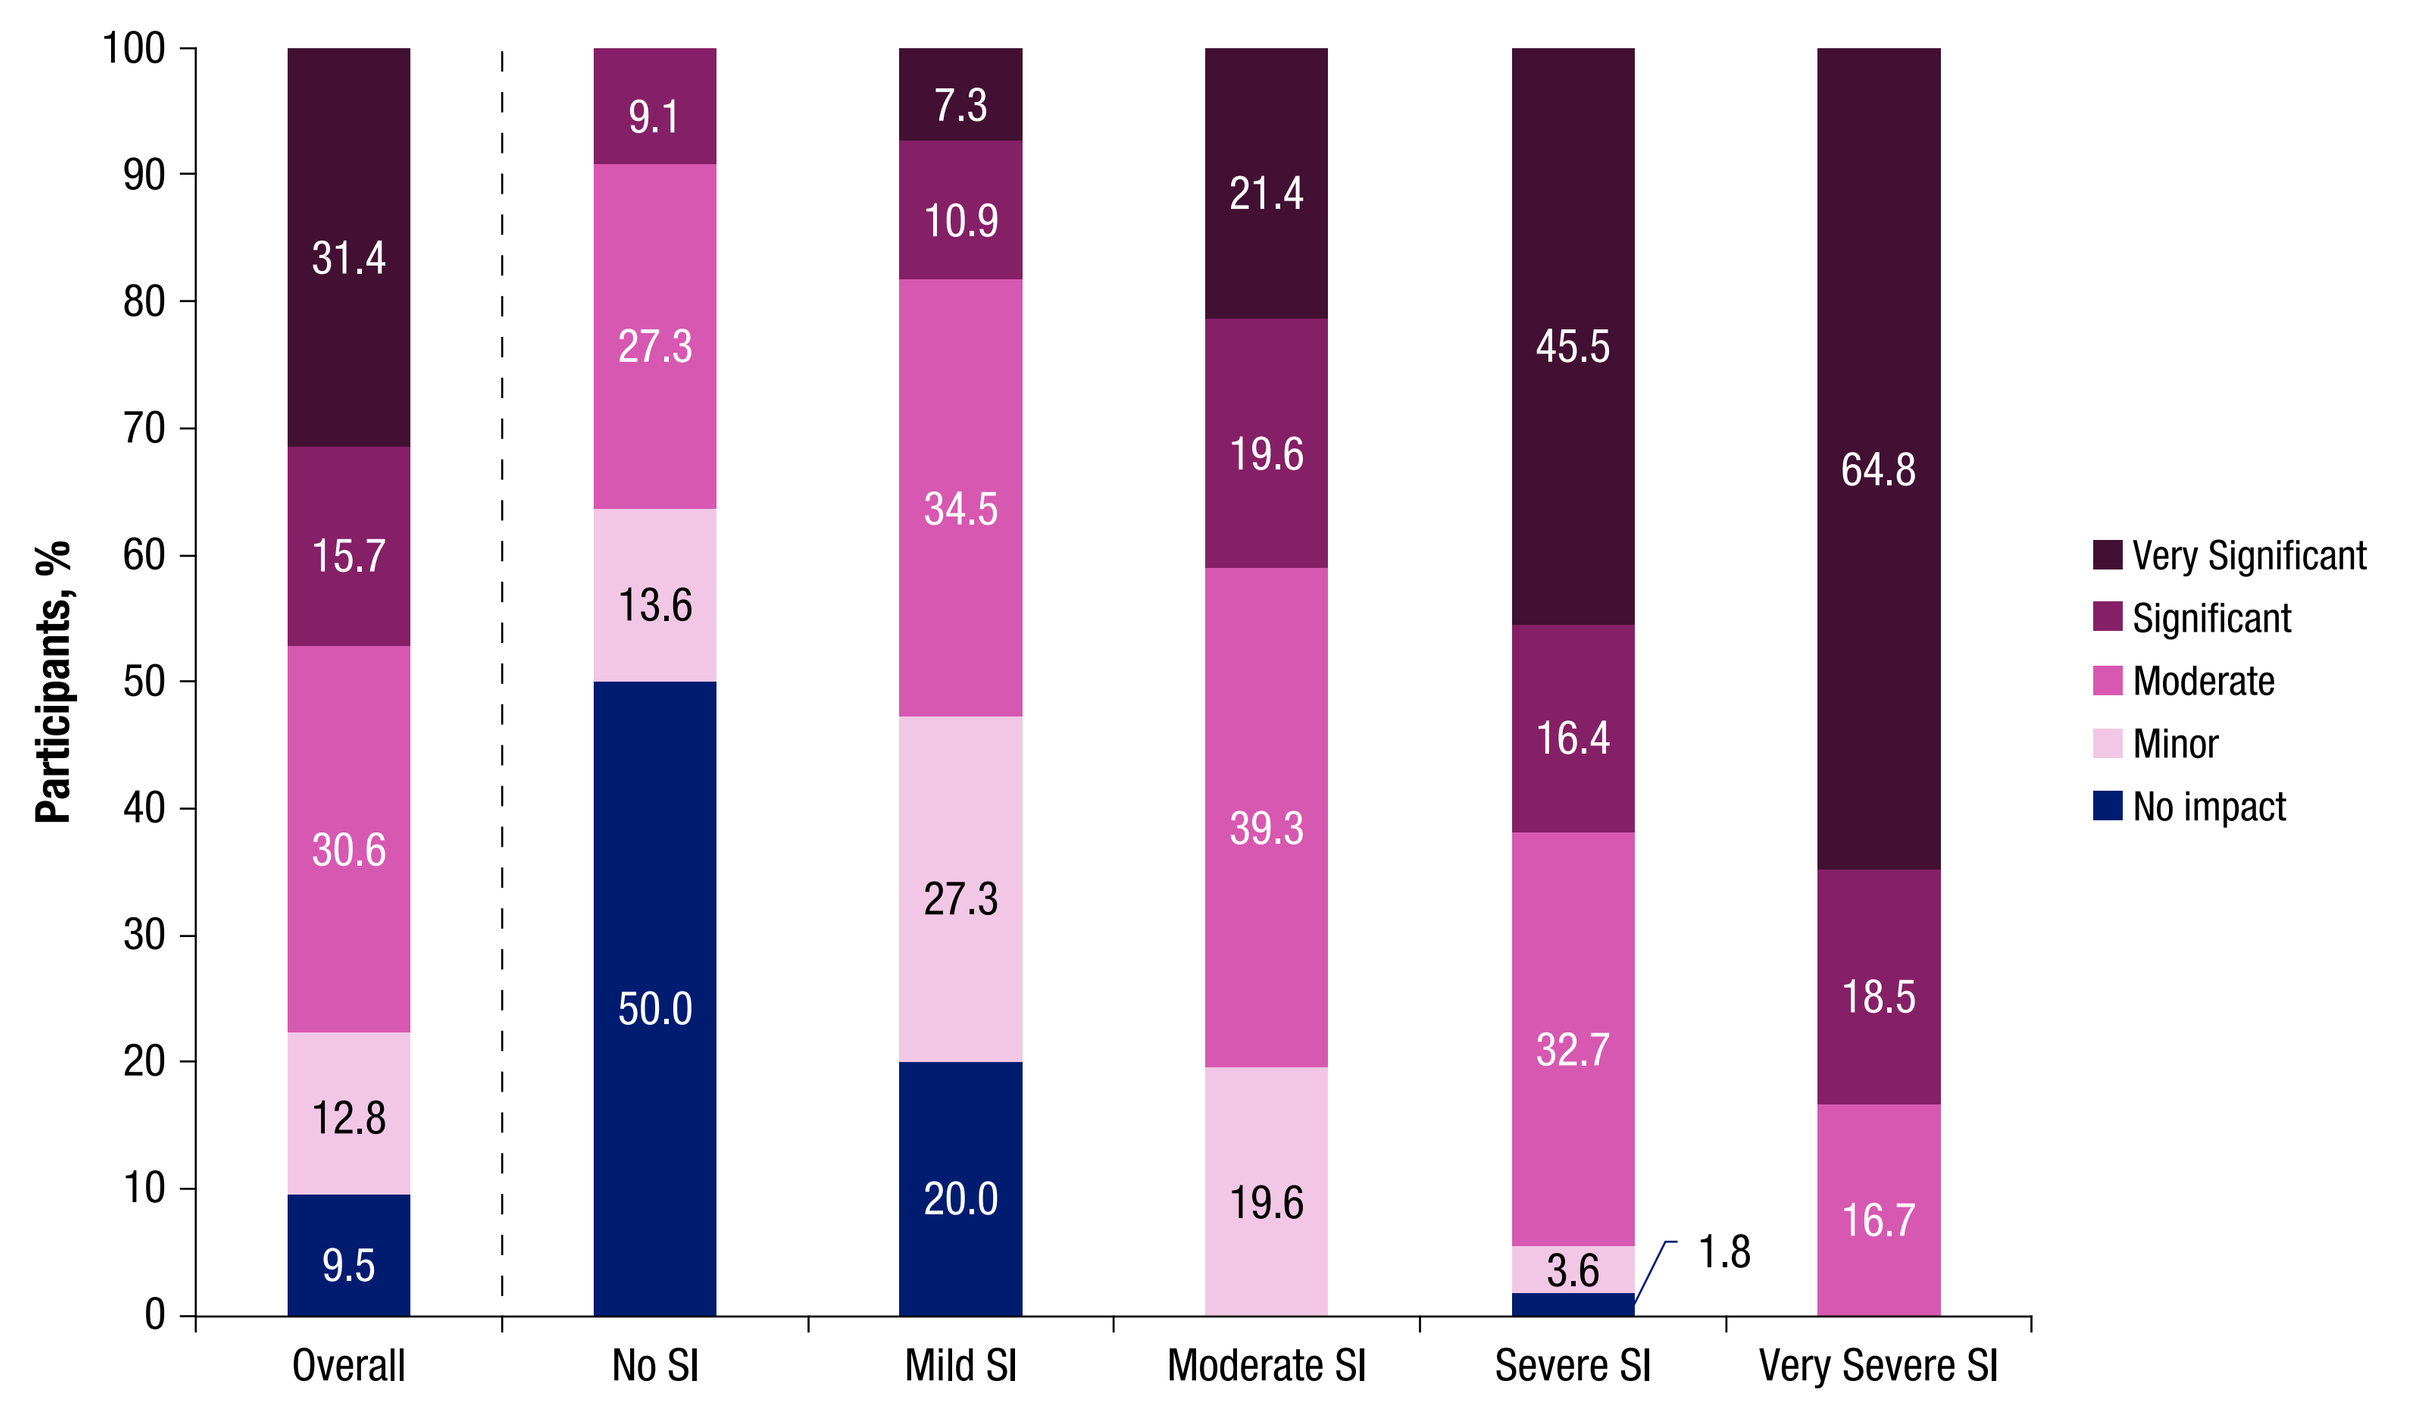
Participants, %
Very Significant
Significant
Moderate
Minor
No impact
Overall
No SI
Mild SI
Moderate SI
Severe SI
Very Severe SI

[a]As measured by IHSS item 11 asking: "Do you consider that your hypersomnolence is a problem in terms of your proper intellectual functioning (ie, problems with concentration, memory problems, decrease in your intellectual performance)?"[10]
IHSS, Idiopathic Hypersomnia Severity Scale; SI, sleep inertia.

- Overall, 47.1% of all participants and 72.5% of participants with severe or very severe sleep inertia (SIQ scores ≥73) reported that hypersomnolence significantly or very significantly impacted intellectual functioning

Figure 2. Impact of Sleep Inertia on Ability to Concentrate (A) and Performance at School or Work (B)[a]

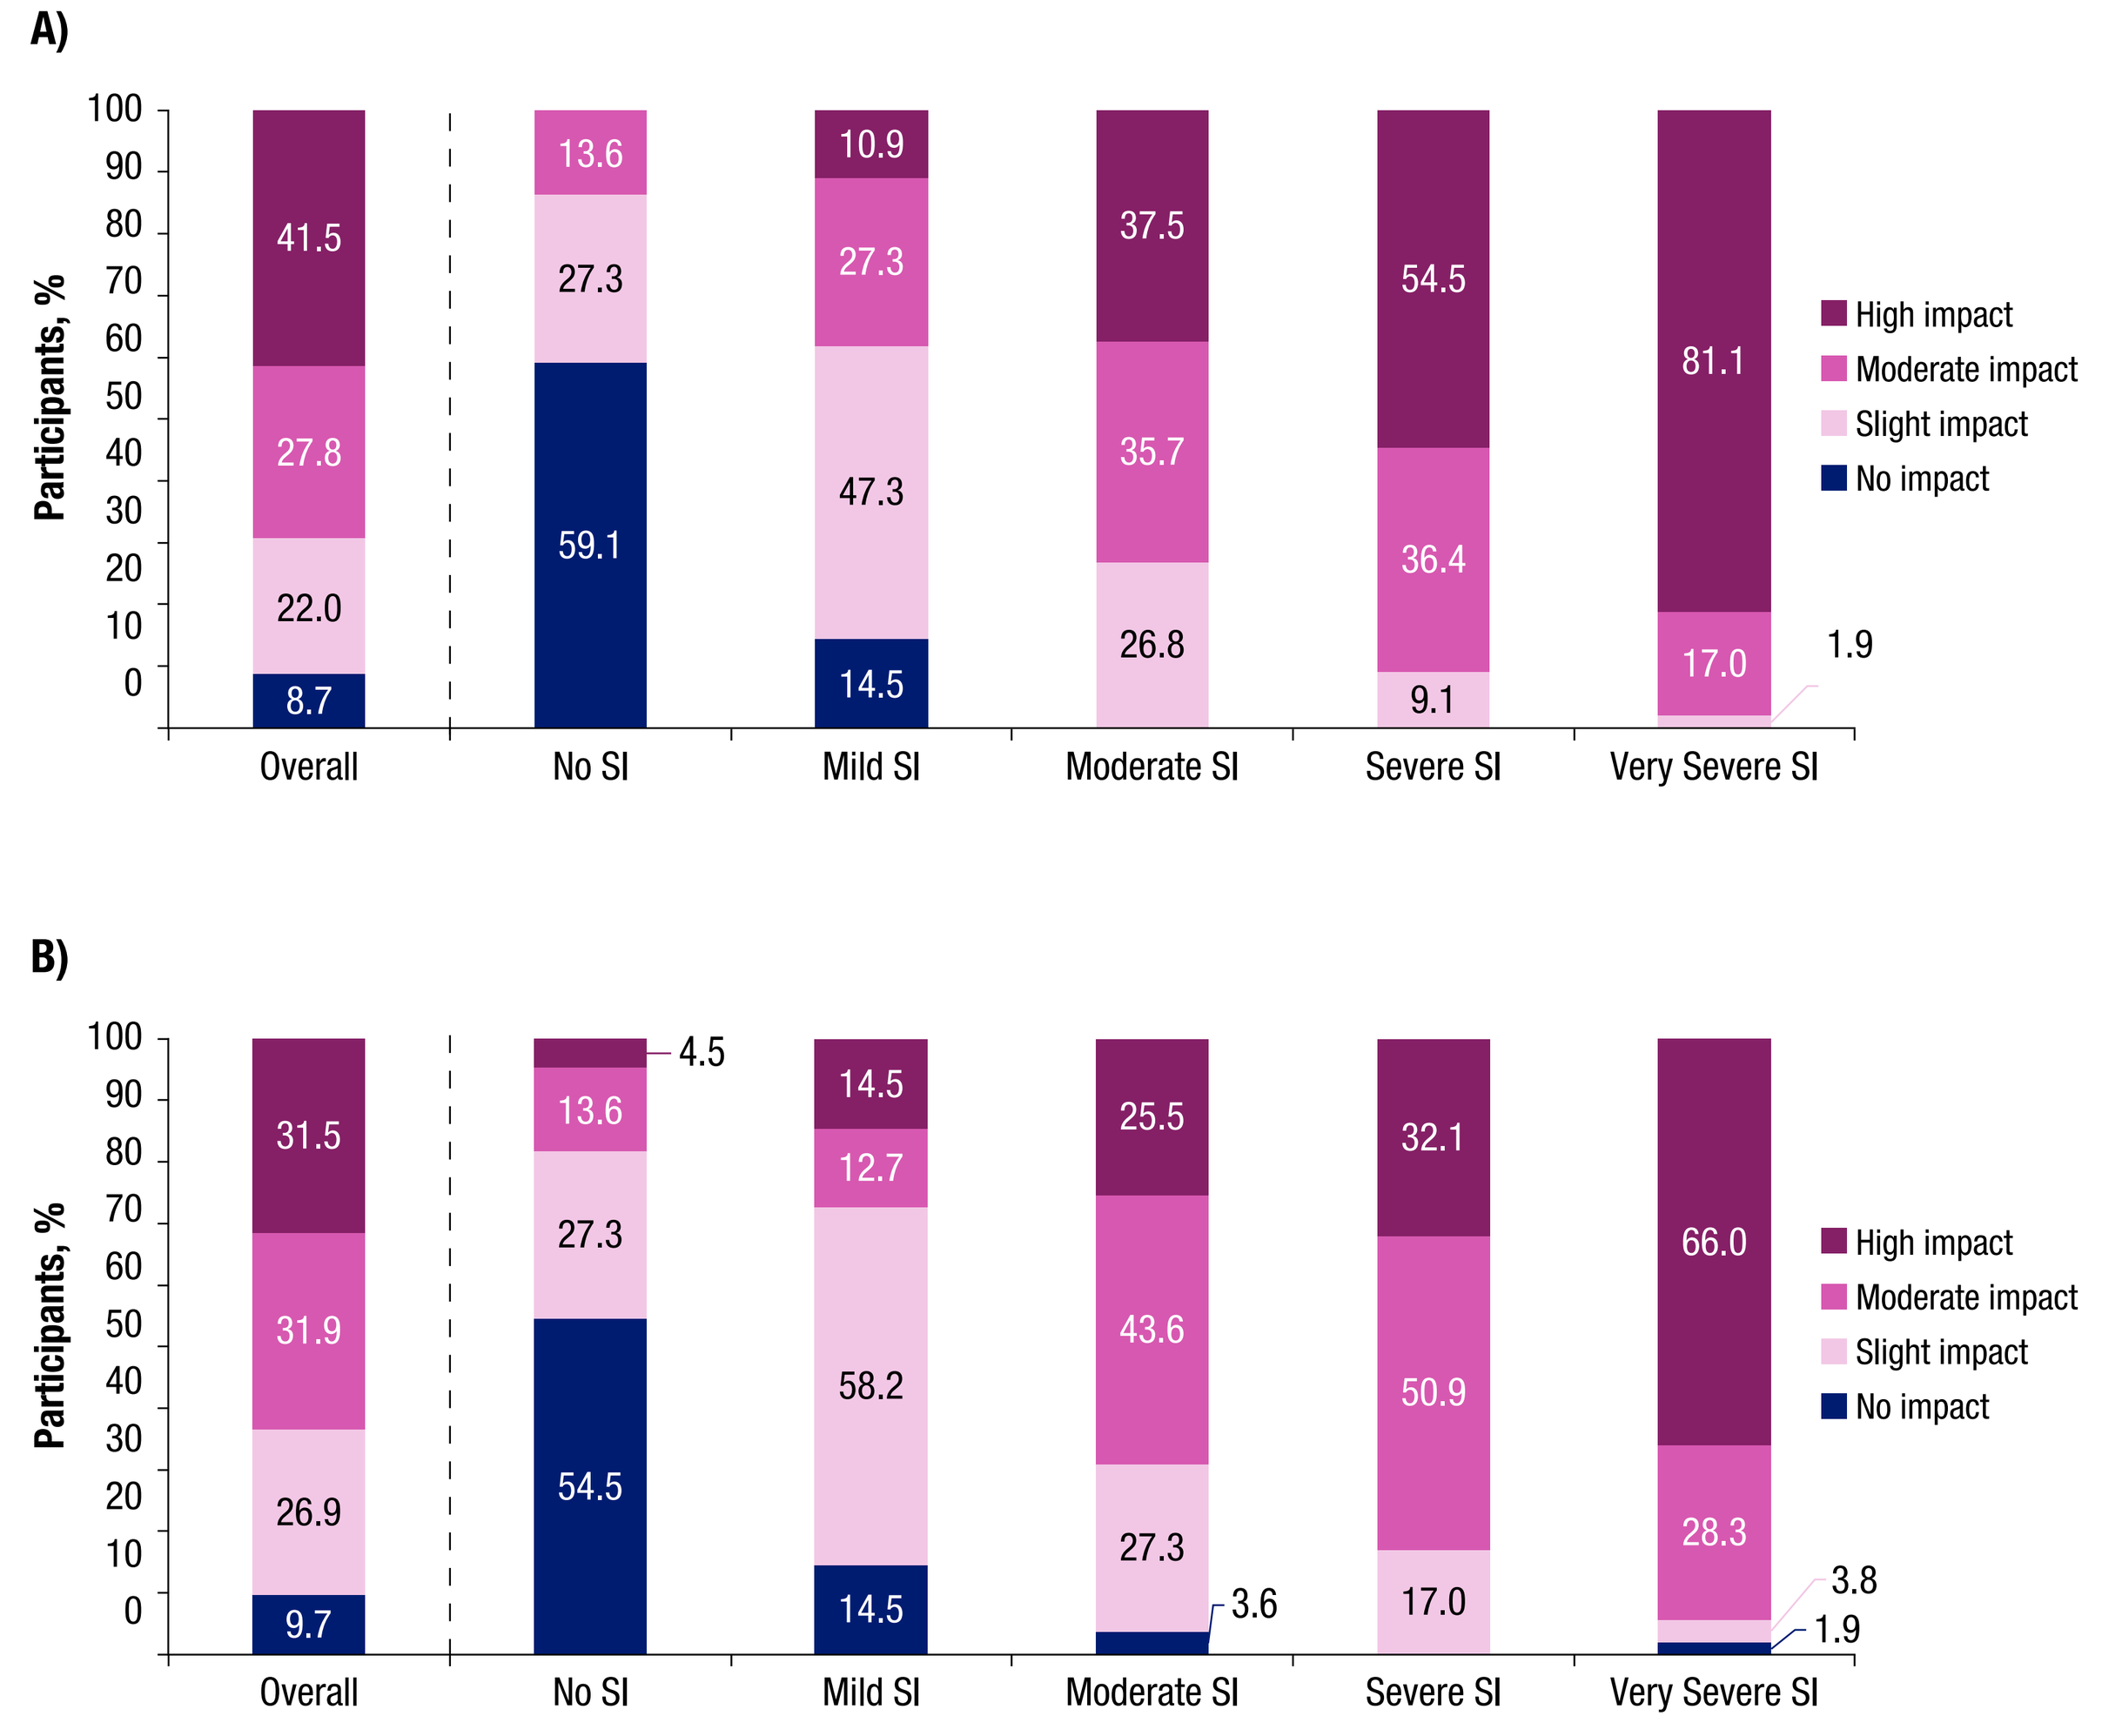
A)
B)
Participants, %
High impact
Moderate impact
Slight impact
No impact
Overall
No SI
Mild SI
Moderate SI
Severe SI
Very Severe SI

[a]As measured by a de novo question asking: "Please check the box that most closely describes the impact sleep inertia has on areas of your life."
SI, sleep inertia.

- Over 60% of participants reported that sleep inertia had a moderate or high impact on concentration (69.3%) and school/work performance (63.4%)
 - The proportion increased with increasing sleep inertia severity (98.1% and 94.3%, respectively, among those with very severe sleep inertia)

Figure 3. Idiopathic Hypersomnia Symptoms Related to Cognitive Impairment,[a] Overall (A) and Stratified by Sleep Inertia Severity (B)

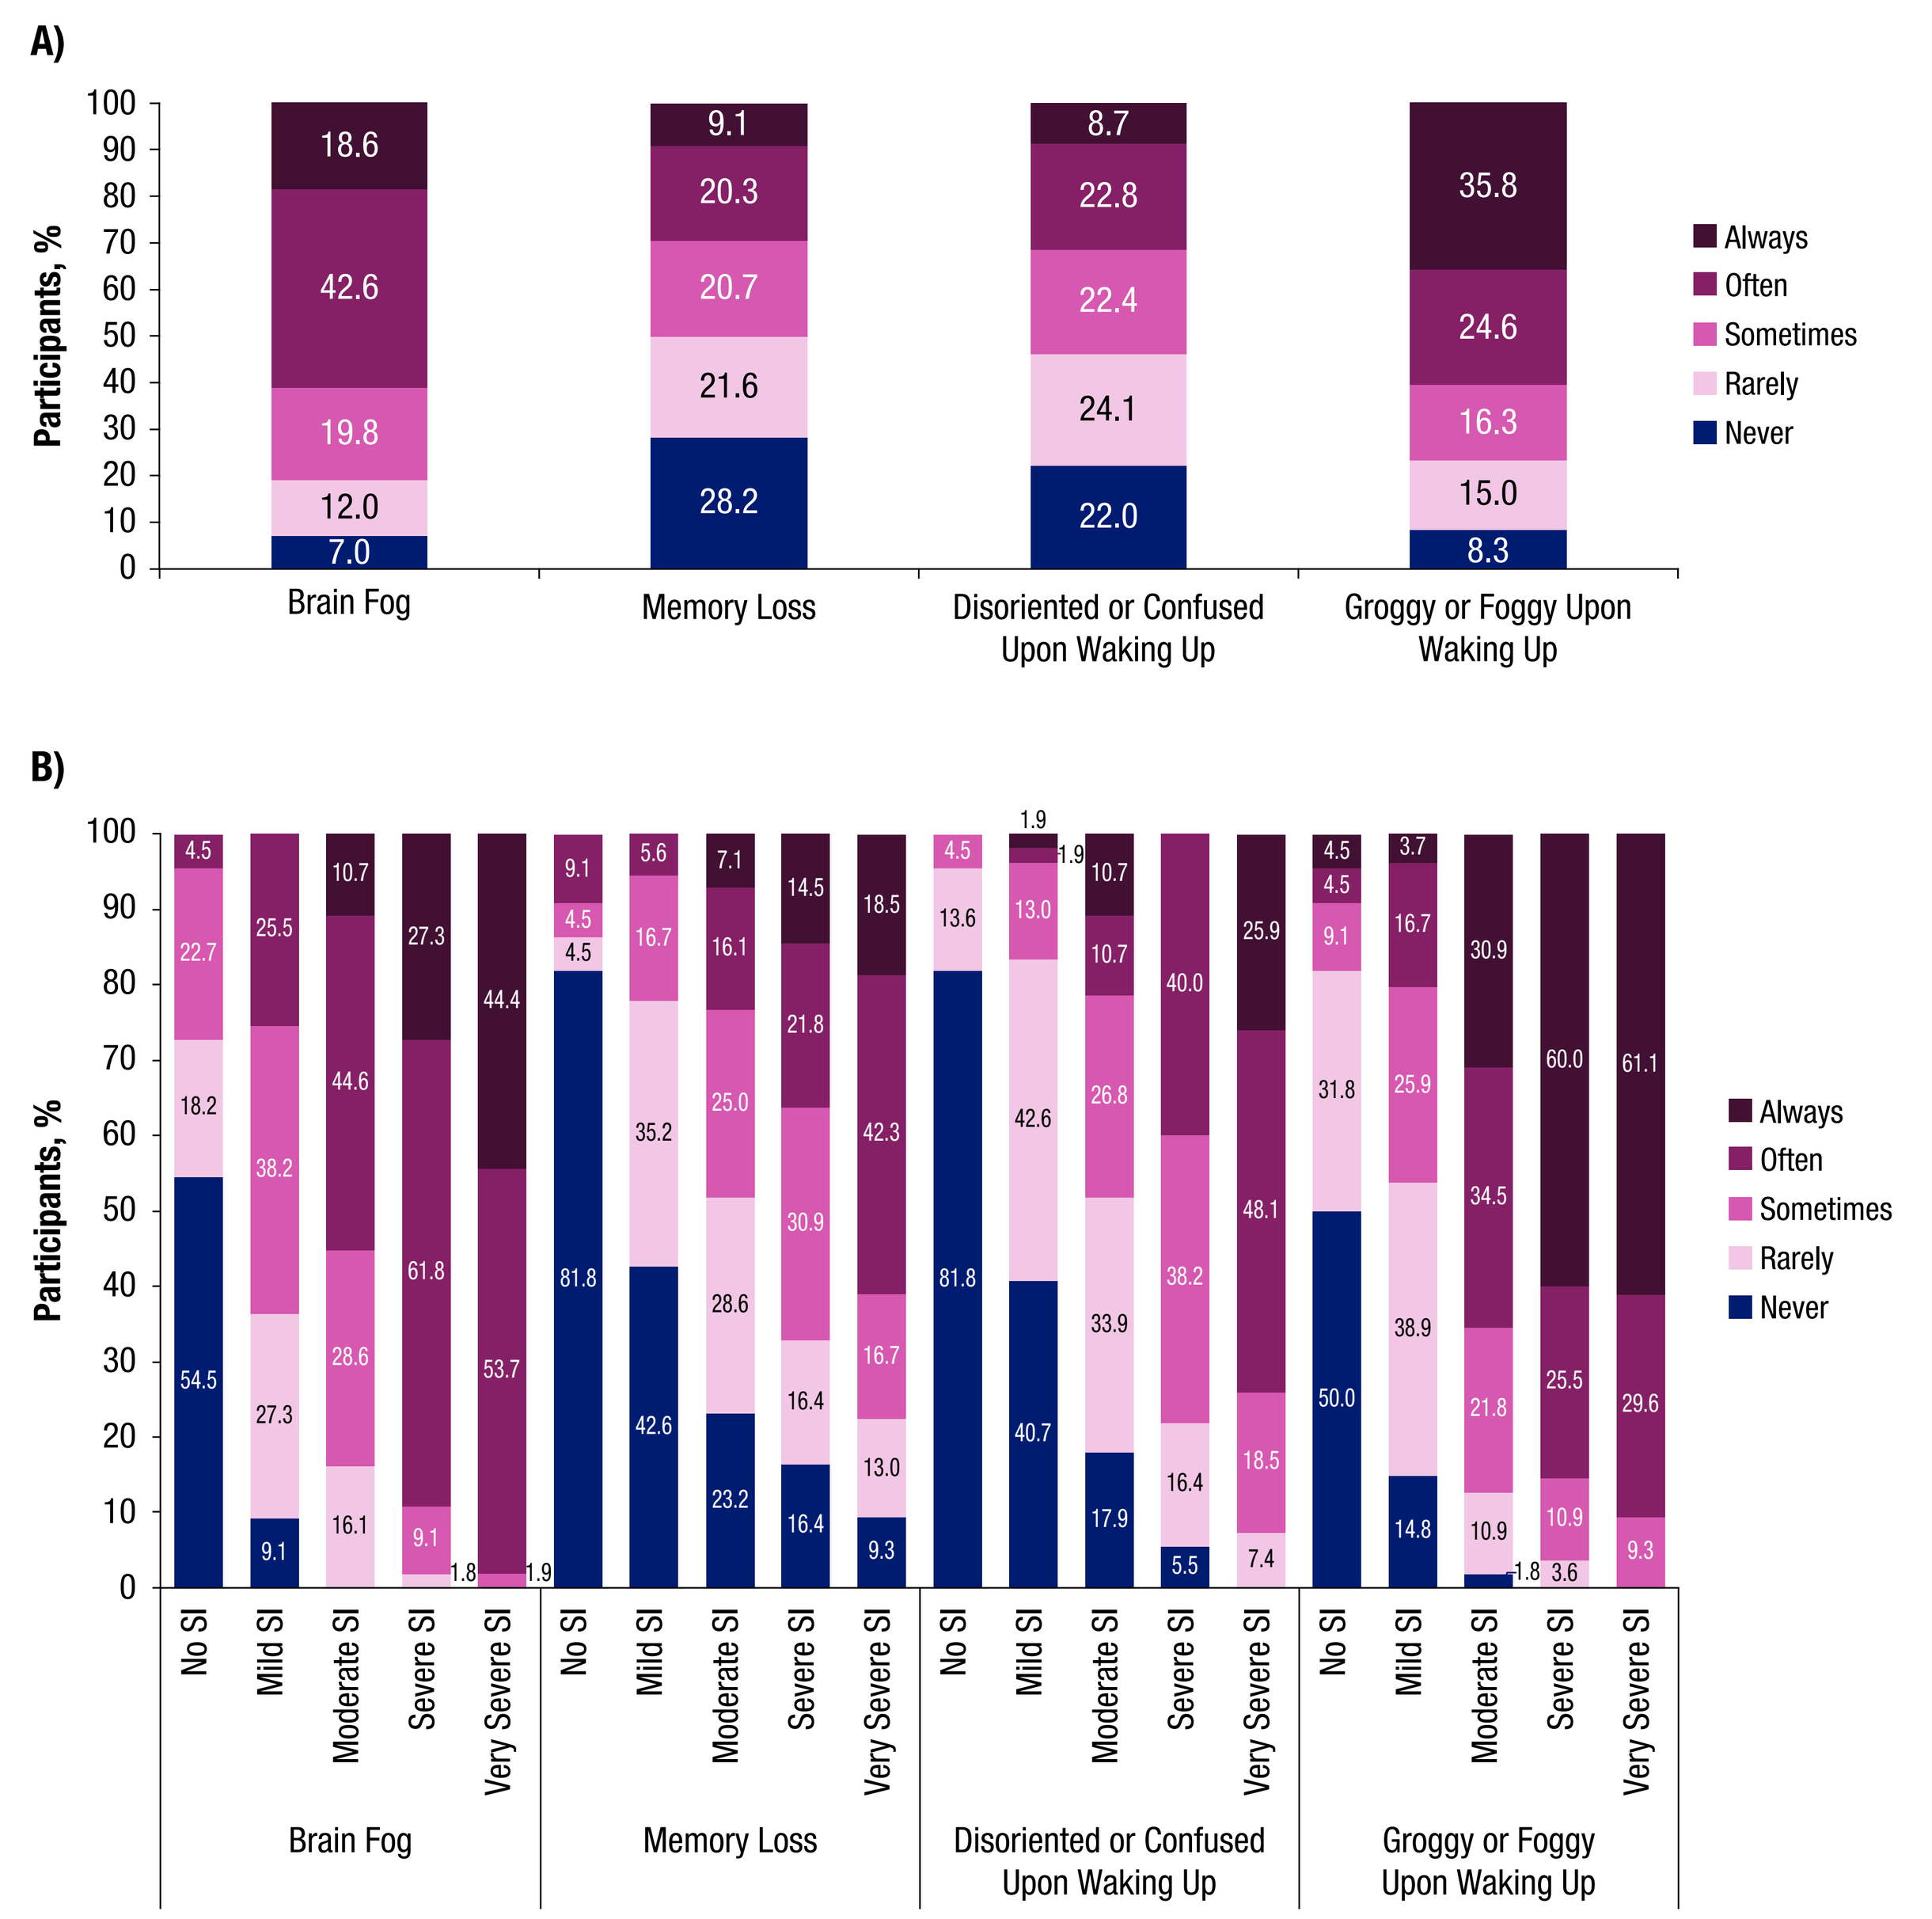
A)
B)
Participants, %
Always
Often
Sometimes
Rarely
Never
Brain Fog
Memory Loss
Disoriented or Confused Upon Waking Up
Groggy or Foggy Upon Waking Up
No SI
Mild SI
Moderate SI
Severe SI
Very Severe SI

[a]As measured by a de novo question asking: "How often do you experience the following idiopathic hypersomnia symptoms?"
SI, sleep inertia.

- Over 60% of participants reported that they often or always experienced brain fog (61.2%) and grogginess/fogginess on waking (60.4%)
- As sleep inertia severity increased, a higher proportion of participants reported more frequent cognitive symptoms

Figure 4. Impact of Sleep Inertia on Cognition, as Measured by the SIQ[a]

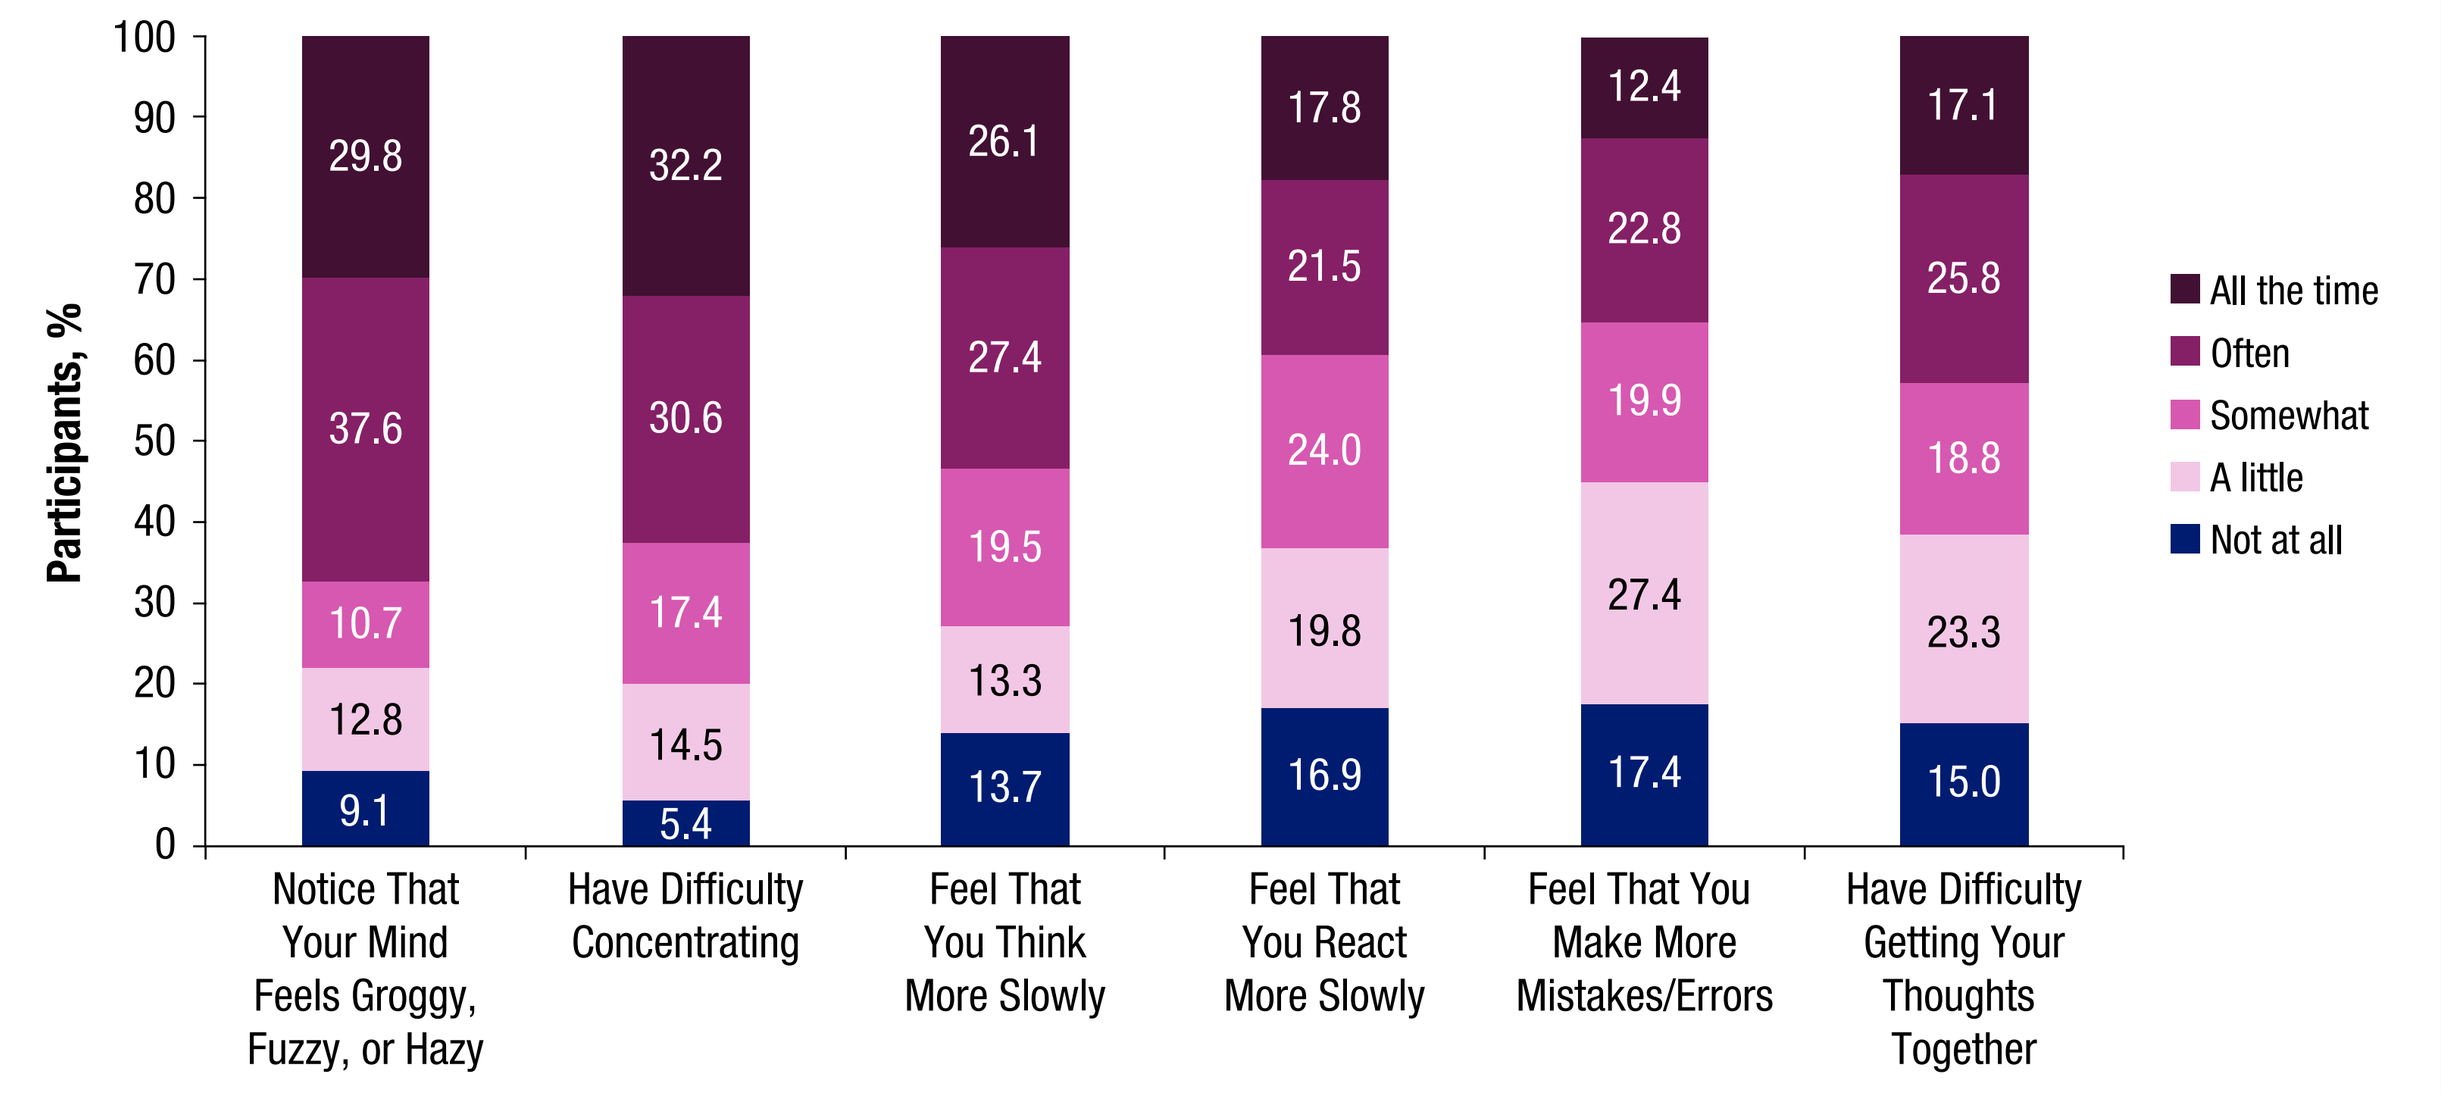
Participants, %
All the time
Often
Somewhat
A little
Not at all
Notice That Your Mind Feels Groggy, Fuzzy, or Hazy
Have Difficulty Concentrating
Feel That You Think More Slowly
Feel That You React More Slowly
Feel That You Make More Mistakes/Errors
Have Difficulty Getting Your Thoughts Together

[a]As measured by the SIQ items 9, 16, 17, 18, 19, and 21.[7]
SIQ, Sleep Inertia Questionnaire.

- On the SIQ, most participants (>55%) reported that they were somewhat affected to affected all the time by cognitive symptoms upon waking in the past week
- A majority reported that, upon waking, they noticed their minds feeling groggy, fuzzy, or hazy (67.4%), and had difficulty concentrating (62.8%) often or all the time

Figure 5. Prevalence (A) and Duration (B) of Cognitive Symptoms Upon Waking, Stratified by Sleep Inertia Severity

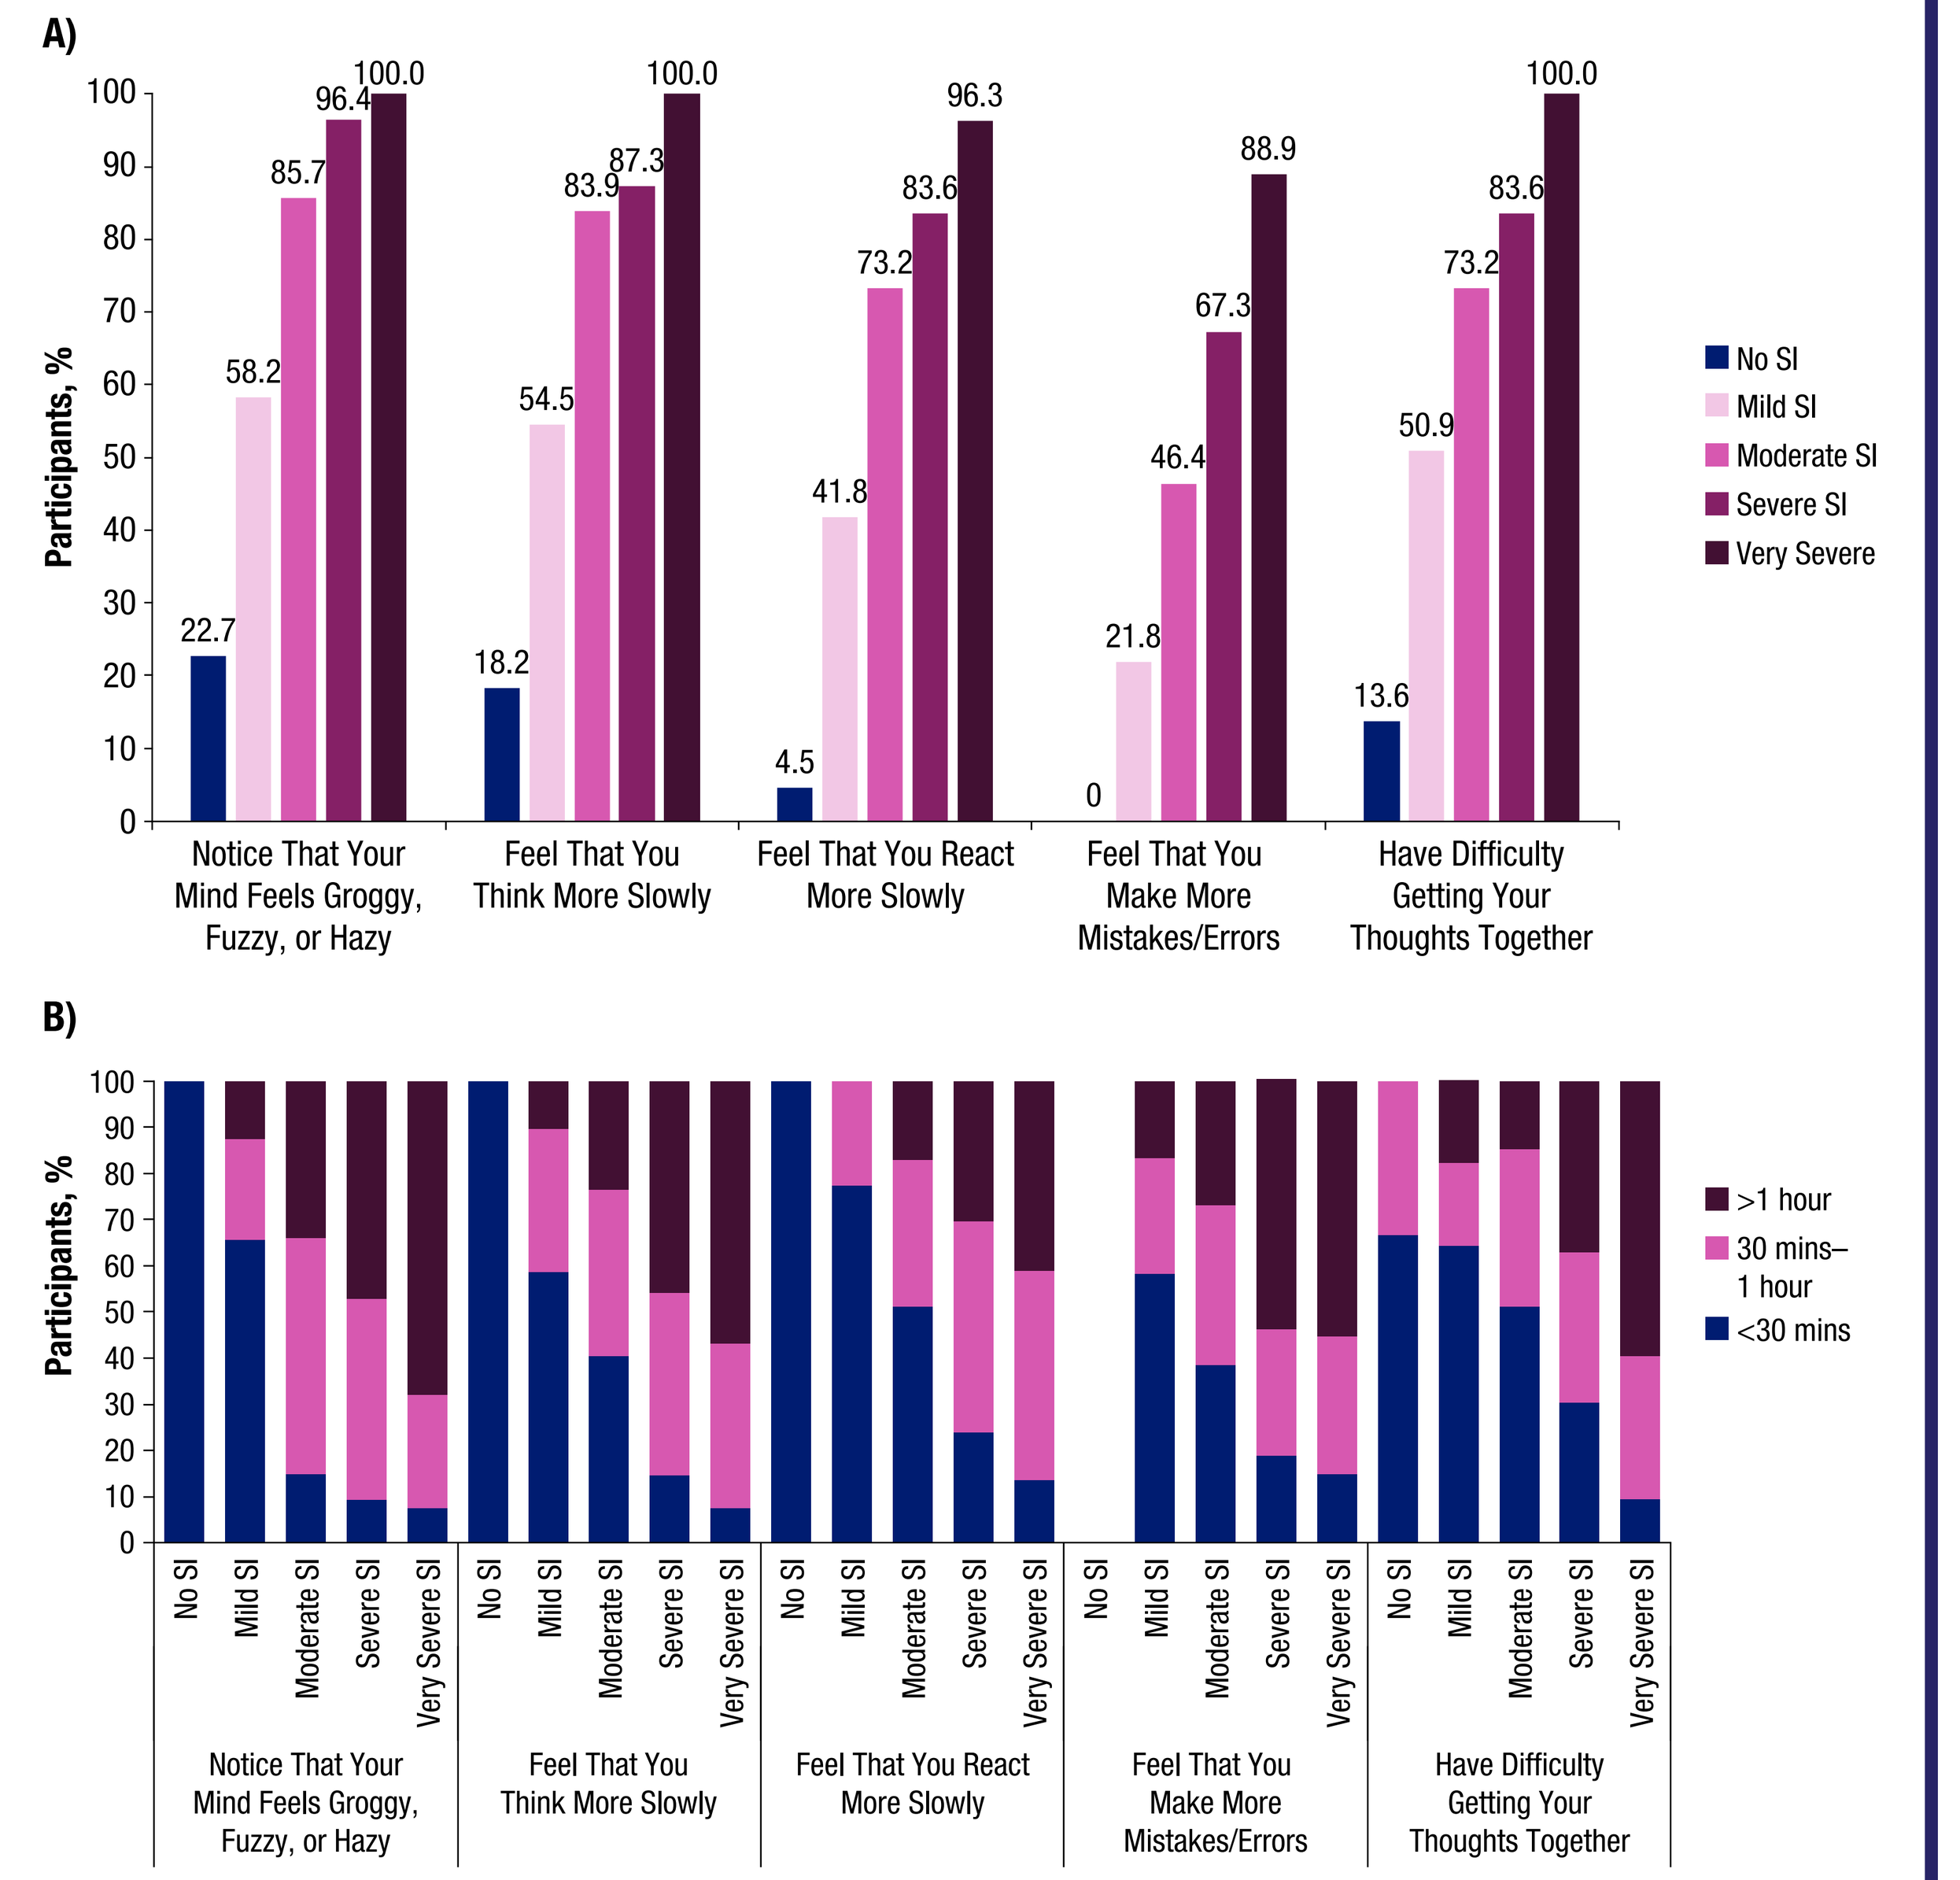
A)
B)
Participants, %
No SI
Mild SI
Moderate SI
Severe SI
Very Severe
>1 hour
30 mins–1 hour
<30 mins
Notice That Your Mind Feels Groggy, Fuzzy, or Hazy
Feel That You Think More Slowly
Feel That You React More Slowly
Feel That You Make More Mistakes/Errors
Have Difficulty Getting Your Thoughts Together

SI, sleep inertia.

- The prevalence and duration of cognitive symptoms upon waking increased with the severity of sleep inertia
- Every participant with very severe sleep inertia (SIQ score ≥85; n=54) reported that, upon waking, they thought more slowly, had difficulty getting their thoughts together, and noticed their minds felt groggy, fuzzy, or hazy
 - Over half reported that these symptoms persisted for >1 hour after waking

Conclusions

- Participants with idiopathic hypersomnia reported that sleep inertia greatly affected multiple aspects of cognition, including experiencing brain fog, memory loss, and difficulty concentrating
- Cognitive impairment across several domains was reported to persist for an extended period of time after waking, with degree of impairment increasing with sleep inertia severity
- Limitations include the cross-sectional study design and self-selection of participants who may not be representative of the broader population with idiopathic hypersomnia
- The profound impact on cognition underscores the need for clinical vigilance of sleep inertia when diagnosing and treating people with idiopathic hypersomnia

Scan this code to access this poster online.
This code is not for promotional purposes.

Presented at Psych Congress 2025; September 17–21, 2025; San Diego, CA, USA