113

Clinical Characteristics and Productivity Losses Among Individuals Diagnosed With Narcolepsy or Idiopathic Hypersomnia

Adeniyi T. Togun, MD, PhD[1]; Silky Beaty, PharmD, MSPH[1]; Caroleen Drachenberg, PhD, MSPH[1]; Jessica K. Alexander, PhD[1]; Marisa Whalen, PharmD[1]; Jing Dai, PhD[1]; Weiyi Ni, PhD[1]; Sarah C. Markt, ScD, MPH[1]; Jed Black, MD[1,2]

[1]Jazz Pharmaceuticals, Palo Alto, CA, USA; [2]Stanford University Center for Sleep Sciences and Medicine, Palo Alto, CA, USA

Introduction

- Narcolepsy and idiopathic hypersomnia are central disorders of hypersomnolence primarily characterized by excessive daytime sleepiness[1]
 - Other narcolepsy symptoms include cataplexy (narcolepsy type 1), disrupted nighttime sleep, sleep-related hallucinations, and sleep paralysis[1]
 - Idiopathic hypersomnia is often accompanied by severe sleep inertia, long and unrefreshing naps, prolonged nighttime sleep, autonomic complaints, and cognitive impairment[1]
- While narcolepsy and idiopathic hypersomnia are associated with higher comorbidity burden and increased healthcare resource utilization (HCRU) compared with those without narcolepsy or idiopathic hypersomnia,[2-4] the full spectrum of the clinical and economic burden posed by these disorders has seldom been comprehensively or comparatively explored

Objective

- This study described clinical characteristics and productivity losses in individuals with narcolepsy or idiopathic hypersomnia compared with individuals without narcolepsy or idiopathic hypersomnia

Methods

- Merative MarketScan® Research Database (MMRD) linked to the Health and Productivity Management (HPM) Database (01/01/2017–12/31/2023) were used to identify individuals with narcolepsy or idiopathic hypersomnia aged 18 to 64 years with continuous medical and pharmacy enrollment in the 365-day pre- and post-index periods
 - The index date was the first diagnosis of narcolepsy or idiopathic hypersomnia
 - For analyses using the HPM Database, 365 days of continuous HPM data availability in both the pre- and post-index periods was also required
- Individuals without narcolepsy or idiopathic hypersomnia were matched 2:1 to those with narcolepsy or idiopathic hypersomnia using propensity-score risk-set matching on age (within 1 year), gender, census region, relationship to employee, and cohort entry date
- Outcomes
 - Demographic and clinical characteristics, including comorbidities, in the pre- and post-index periods in the full MMRD population were descriptively analyzed
 - Productivity characteristics: number and percentage of individuals with a short-term disability (STD), long-term disability (LTD), workers' compensation (WC), and workplace absence (WA) claim were assessed descriptively in the post-index period for employees with available HPM data
 - Among those with a claim, number of days of STD, LTD, WC, and WA absence were calculated
 - Indirect costs per person, per year were calculated among individuals with STD, LTD, WC, and WA claims, multiplying days absent by the median weekly earnings of full-time workers ($1145) in the fourth quarter of 2023

Results

Table 1. Demographic Characteristics in the MMRD Population

	Narcolepsy (n=18,231)	Matched Non-narcolepsy (n=36,462)	Idiopathic Hypersomnia (n=7156)	Matched Non-idiopathic Hypersomnia (n=14,312)
Age, mean (SD), years	40.0 (13.1)	40.4 (13.1)	41.1 (12.8)	41.5 (12.8)
Gender, n (%)				
Female	11,714 (64.3)	23,428 (64.3)	4725 (66.0)	9449 (66.0)
Male	6517 (35.7)	13,034 (35.7)	2431 (34.0)	4863 (34.0)
Region				
Northeast	1945 (10.7)	3889 (10.7)	773 (10.8)	1547 (10.8)
North Central	5139 (28.2)	10,276 (28.2)	1962 (27.4)	3922 (27.4)
South	9461 (51.9)	18,923 (51.9)	3496 (48.9)	6993 (48.9)
West	1667 (9.1)	3335 (9.1)	917 (12.8)	1834 (12.8)
Unknown	19 (0.1)	39 (0.1)	8 (0.1)	16 (0.1)

MMRD, Merative MarketScan Research Database; SD, standard deviation.

- The study sample included 18,231 individuals with narcolepsy, matched to 36,462 individuals without narcolepsy; the mean (standard deviation [SD]) age in the cohorts was 40.0 (13.1) and 40.4 (13.1) years, respectively, and 64.3% were female
- The idiopathic hypersomnia cohort comprised 7156 individuals, matched to 14,312 individuals without idiopathic hypersomnia; the mean (SD) age in the cohorts was 41.1 (12.8) and 41.5 (12.8) years, respectively, and 66.0% were female

Figure 1. Prevalence[a] of (A) Sleep-related/Neurologic and Psychiatric Comorbidities and Alerting Agent Use and (B) Cardiovascular, Cardiometabolic, and Renal Comorbidities in Individuals With and Without Narcolepsy

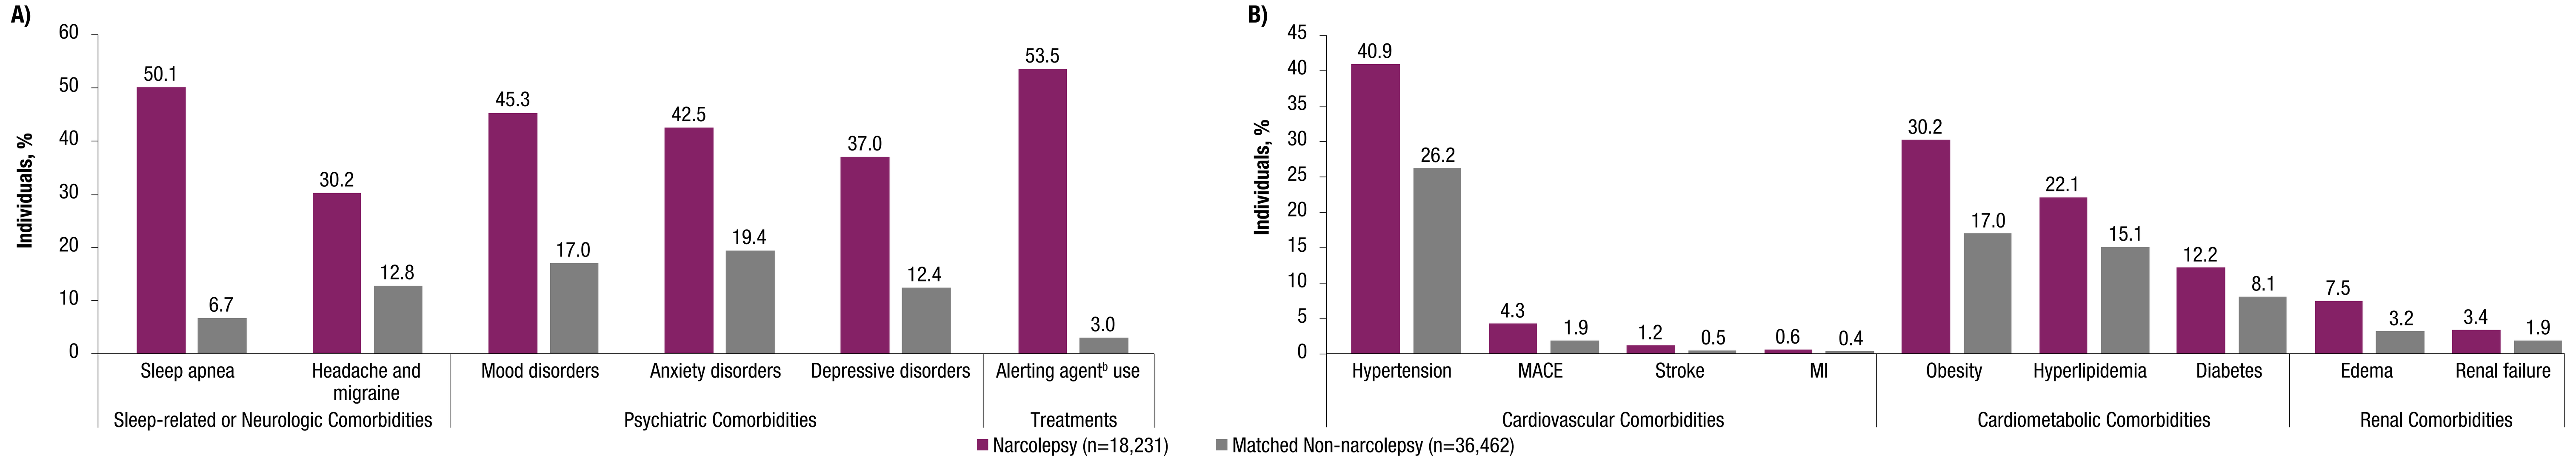

A)
Individuals, %
50.1
6.7
30.2
12.8
45.3
17.0
42.5
19.4
37.0
12.4
53.5
3.0
Sleep apnea
Headache and migraine
Mood disorders
Anxiety disorders
Depressive disorders
Alerting agent[b] use
Sleep-related or Neurologic Comorbidities
Psychiatric Comorbidities
Treatments
B)
40.9
26.2
4.3
1.9
1.2
0.5
0.6
0.4
30.2
17.0
22.1
15.1
12.2
8.1
7.5
3.2
3.4
1.9
Hypertension
MACE
Stroke
MI
Obesity
Hyperlipidemia
Diabetes
Edema
Renal failure
Cardiovascular Comorbidities
Cardiometabolic Comorbidities
Renal Comorbidities
Narcolepsy (n=18,231)
Matched Non-narcolepsy (n=36,462)

[a]Proportions have been rounded. [b]Alerting agents were defined as stimulants (ie, amphetamines or methylphenidates) or wake-promoting agents (eg, modafinil, solriamfetol, or pitolisant).
MACE, major adverse cardiovascular event; MI, myocardial infarction.

- Individuals with versus without narcolepsy had a higher prevalence of sleep-related/neurologic (eg, sleep apnea), psychiatric (eg, mood disorders), cardiovascular (eg, hypertension), cardiometabolic (eg, diabetes), and renal (eg, edema) comorbidities, as well as alerting agent use

Figure 2. Prevalence[a] of (A) Sleep-related/Neurologic and Psychiatric Comorbidities and Alerting Agent Use and (B) Cardiovascular, Cardiometabolic, and Renal Comorbidities in Individuals With and Without Idiopathic Hypersomnia

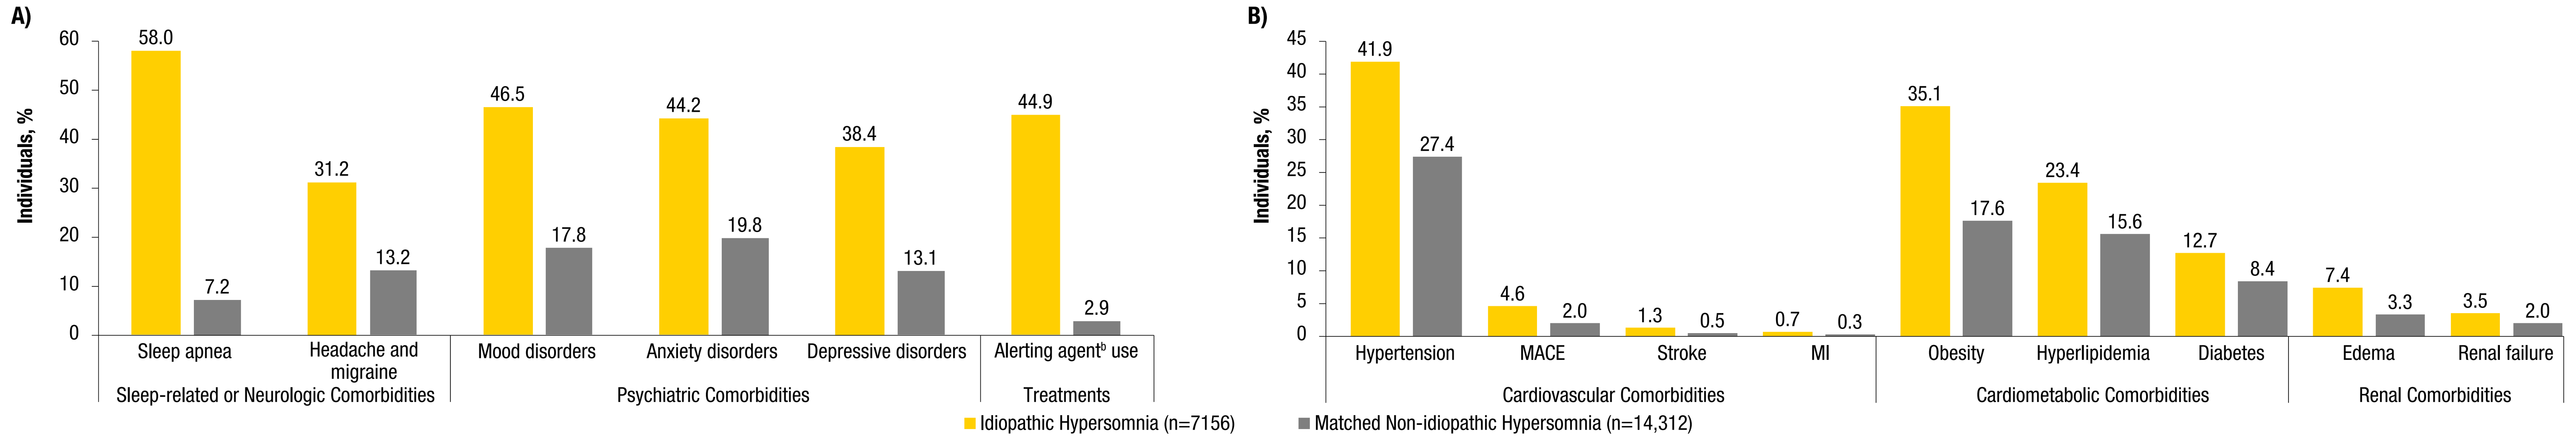

A)
Individuals, %
58.0
7.2
31.2
13.2
46.5
17.8
44.2
19.8
38.4
13.1
44.9
2.9
Sleep apnea
Headache and migraine
Mood disorders
Anxiety disorders
Depressive disorders
Alerting agent[b] use
Sleep-related or Neurologic Comorbidities
Psychiatric Comorbidities
Treatments
B)
41.9
27.4
4.6
2.0
1.3
0.5
0.7
0.3
35.1
17.6
23.4
15.6
12.7
8.4
7.4
3.3
3.5
2.0
Hypertension
MACE
Stroke
MI
Obesity
Hyperlipidemia
Diabetes
Edema
Renal failure
Cardiovascular Comorbidities
Cardiometabolic Comorbidities
Renal Comorbidities
Idiopathic Hypersomnia (n=7156)
Matched Non-idiopathic Hypersomnia (n=14,312)

[a]Proportions have been rounded. [b]Alerting agents were defined as stimulants (ie, amphetamines or methylphenidates) or wake-promoting agents (eg, modafinil, solriamfetol, or pitolisant).
MACE, major adverse cardiovascular event; MI, myocardial infarction.

- Individuals with versus without idiopathic hypersomnia had a higher prevalence of sleep-related/neurologic (eg, sleep apnea), psychiatric (eg, mood disorders), cardiovascular (eg, hypertension), cardiometabolic (eg, diabetes), and renal (eg, edema) comorbidities, as well as alerting agent use

Table 2. Productivity Losses In Individuals With and Without Narcolepsy and Idiopathic Hypersomnia

	Narcolepsy	Matched Non-narcolepsy	Idiopathic Hypersomnia	Matched Non-idiopathic Hypersomnia
Short-term Disability,[a] n	**2553**	**5106**	**950**	**1900**
STD claimants, n (%)	351 (13.7)	404 (7.9)	99 (10.4)	151 (7.9)
Workdays absent,[b] n	79	61	63	62
Long-term Disability,[a] n	**2553**	**5106**	**950**	**1899**
LTD claimants, n (%)	16 (0.6)	28 (0.5)	7 (0.7)	8 (0.4)
Workdays absent,[b] n	117	87	84	78
Workers' Compensation,[a] n	**1812**	**3624**	**597**	**1194**
WC claimants, n (%)	56 (3.1)	111 (3.1)	23 (3.9)	34 (2.8)
Workdays absent,[b] n	89	95	81	67
Workplace Absence,[a] n	**407**	**814**	**179**	**358**
WA claimants, n (%)	205 (50.4)	509 (62.5)	140 (78.2)	243 (67.9)
Workdays absent,[b] n	4.3	3.8	5	4

[a]Number in HPM database with available data. [b]Average number of workdays absent among those with ≥1 claim.
HPM, Health and Productivity Management; LTD, long-term disability; STD, short-term disability; WA, workplace absence; WC, workers' compensation.

- Higher proportions of individuals with versus without narcolepsy had at least 1 claim for STD and LTD; among those with STD or LTD claims, individuals with narcolepsy recorded more STD and LTD workdays absent
 - Similar proportions in the narcolepsy and non-narcolepsy cohort had WC claims, but among those with WC claims, individuals with narcolepsy recorded fewer WC workdays absent
 - A smaller proportion of individuals with versus without narcolepsy had WA claims, but among those with WA claims, individuals with narcolepsy recorded more WA workdays absent
- Higher proportions of individuals with versus without idiopathic hypersomnia had claims for STD, LTD, WC, and WA
 - Among those with claims, individuals with idiopathic hypersomnia had more STD, LTD, WC, and WA workdays absent

Figure 3. Indirect Costs of Productivity Losses in Individuals With and Without (A) Narcolepsy and (B) Idiopathic Hypersomnia

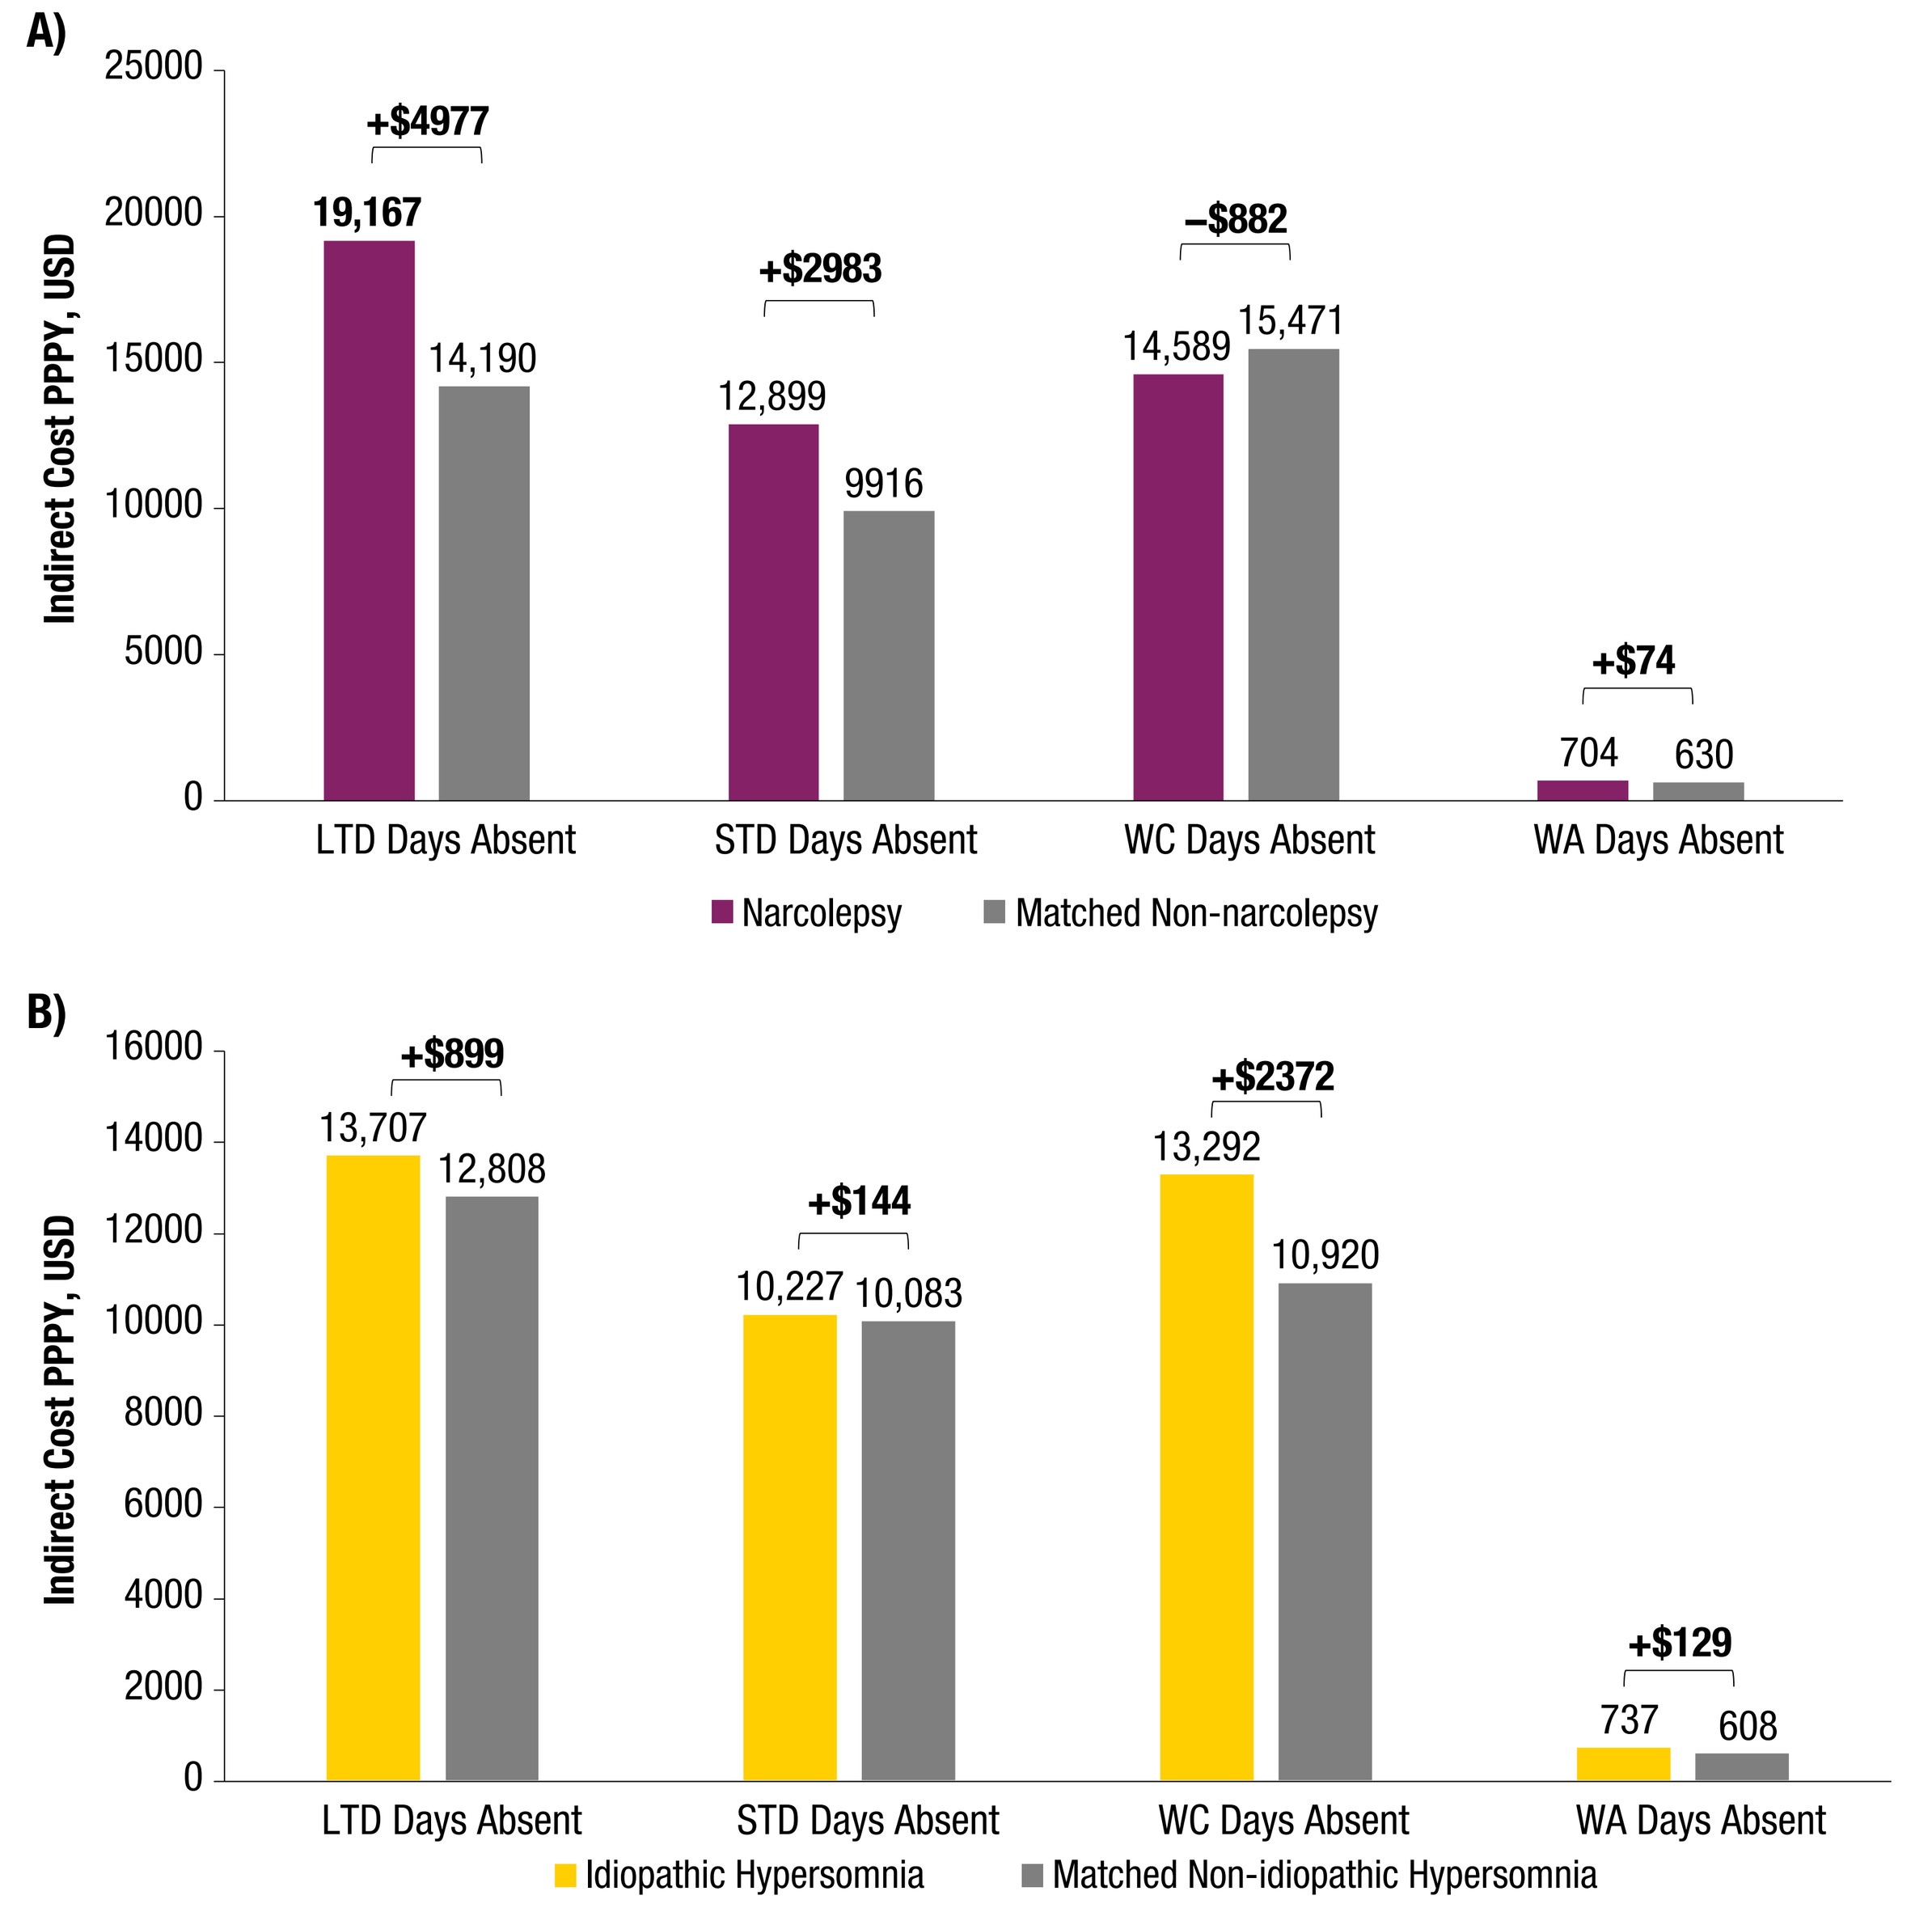

A)
Indirect Cost PPPY, USD
+$4977
19,167
14,190
+$2983
12,899
9916
−$882
14,589
15,471
+$74
704
630
LTD Days Absent
STD Days Absent
WC Days Absent
WA Days Absent
Narcolepsy
Matched Non-narcolepsy
B)
+$899
13,707
12,808
+$144
10,227
10,083
+$2372
13,292
10,920
+$129
737
608
Idiopathic Hypersomnia
Matched Non-idiopathic Hypersomnia

LTD, long-term disability; PPPY, per person per year; STD, short-term disability; USD, United States dollar; WA, workplace absence; WC, workers' compensation.

- The total indirect costs of LTD and STD days absent were higher for individuals with narcolepsy versus those without narcolepsy by a difference of $4977 and $2983, respectively
- Individuals with idiopathic hypersomnia had higher indirect costs than those without idiopathic hypersomnia for LTD and STD days by a difference of $899 and $144, respectively

Conclusions

- Individuals with narcolepsy or idiopathic hypersomnia had higher comorbidity burden, productivity losses, and indirect costs compared with individuals without narcolepsy or idiopathic hypersomnia
- Limitations of the study include the small sample sizes for the HPM subsets and lack of information about reasons for disability-related days missed from work
- Findings underscore the high comorbidity burden, productivity losses, and indirect costs in these populations and need for more holistic management of narcolepsy and idiopathic hypersomnia to mitigate excess comorbidity-related, personal, and societal impact

References: 1. American Academy of Sleep Medicine. *International Classification of Sleep Disorders – Third Edition, Text Revision*. Darien, IL: American Academy of Sleep Medicine; 2023. **2.** Saad R, et al. *Nat Sci Sleep*. 2025;17:1743-1755. **3.** Black J, et al. *Sleep Med*. 2014;15(5):522-529. **4.** Black J, et al. *Sleep Med*. 2017;33:13-18.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees Aeja Jackson, PhD, MS, and Eleanor Bush, MA, provided medical writing support, and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: AT Togun, S Beaty, C Drachenberg, JK Alexander, M Whalen, J Dai, W Ni, and **SC Markt** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **J Black** is a part-time employee of Jazz Pharmaceuticals and shareholder of Jazz Pharmaceuticals, plc.

Scan this code to access this poster online.
This code is not for promotional purposes.

Presented at Psych Congress 2025; September 17–21, 2025; San Diego, CA, USA