123

Effectiveness and Safety of Low-Sodium Oxybate in Participants With Idiopathic Hypersomnia With or Without Psychiatric Comorbidities: Results From the Phase 4 DUET Study

David T. Plante, MD, PhD[1]; Chad M. Ruoff, MD[2]; Deborah A. Nichols, MS[3]; Teresa L. Steininger, PhD[3]; Jing Dai, PhD[3]; M. Todd Kirby, PhD[4,5]; Alyssa Cairns, PhD[4]; Marisa Whalen, PharmD[4]; Logan D. Schneider, MD[6]

[1]Department of Psychiatry, University of Wisconsin–Madison, Madison, WI, USA; [2]Center for Sleep Medicine, Mayo Clinic, Scottsdale, AZ, USA; [3]Jazz Pharmaceuticals, Palo Alto, CA, USA; [4]Jazz Pharmaceuticals, Philadelphia, PA, USA; [5]Department of Psychiatry & Health Behavior, Medical College of Georgia at Augusta University, Augusta, GA, USA; [6]Stanford University Center for Sleep Sciences and Medicine, Stanford, CA, USA

Introduction

- Low-sodium oxybate (LXB; Xywav®)[1-4] is approved by the US Food and Drug Administration for the treatment of cataplexy or excessive daytime sleepiness in patients 7 years of age or older with narcolepsy and the treatment of idiopathic hypersomnia in adults
- Individuals with idiopathic hypersomnia commonly have psychiatric comorbidities,[5-7] and potential psychiatric adverse events are important considerations in idiopathic hypersomnia treatment[1, 8-11]
 - Evaluating the effectiveness and safety of LXB in people with idiopathic hypersomnia with psychiatric comorbidities will help guide clinicians in treating patients with these comorbidities
- Jazz DUET (**D**evelop hypersomnia **U**nderstanding by **E**valuating low-sodium oxybate **T**reatment) was a phase 4, prospective, open-label study (NCT05875974) to assess effectiveness of LXB on sleep and daytime symptoms in participants with narcolepsy (type 1 or 2) or idiopathic hypersomnia

Objective

- To evaluate the effectiveness and safety of LXB treatment in participants with idiopathic hypersomnia with or without psychiatric comorbidities

Methods

Figure 1. Study Design

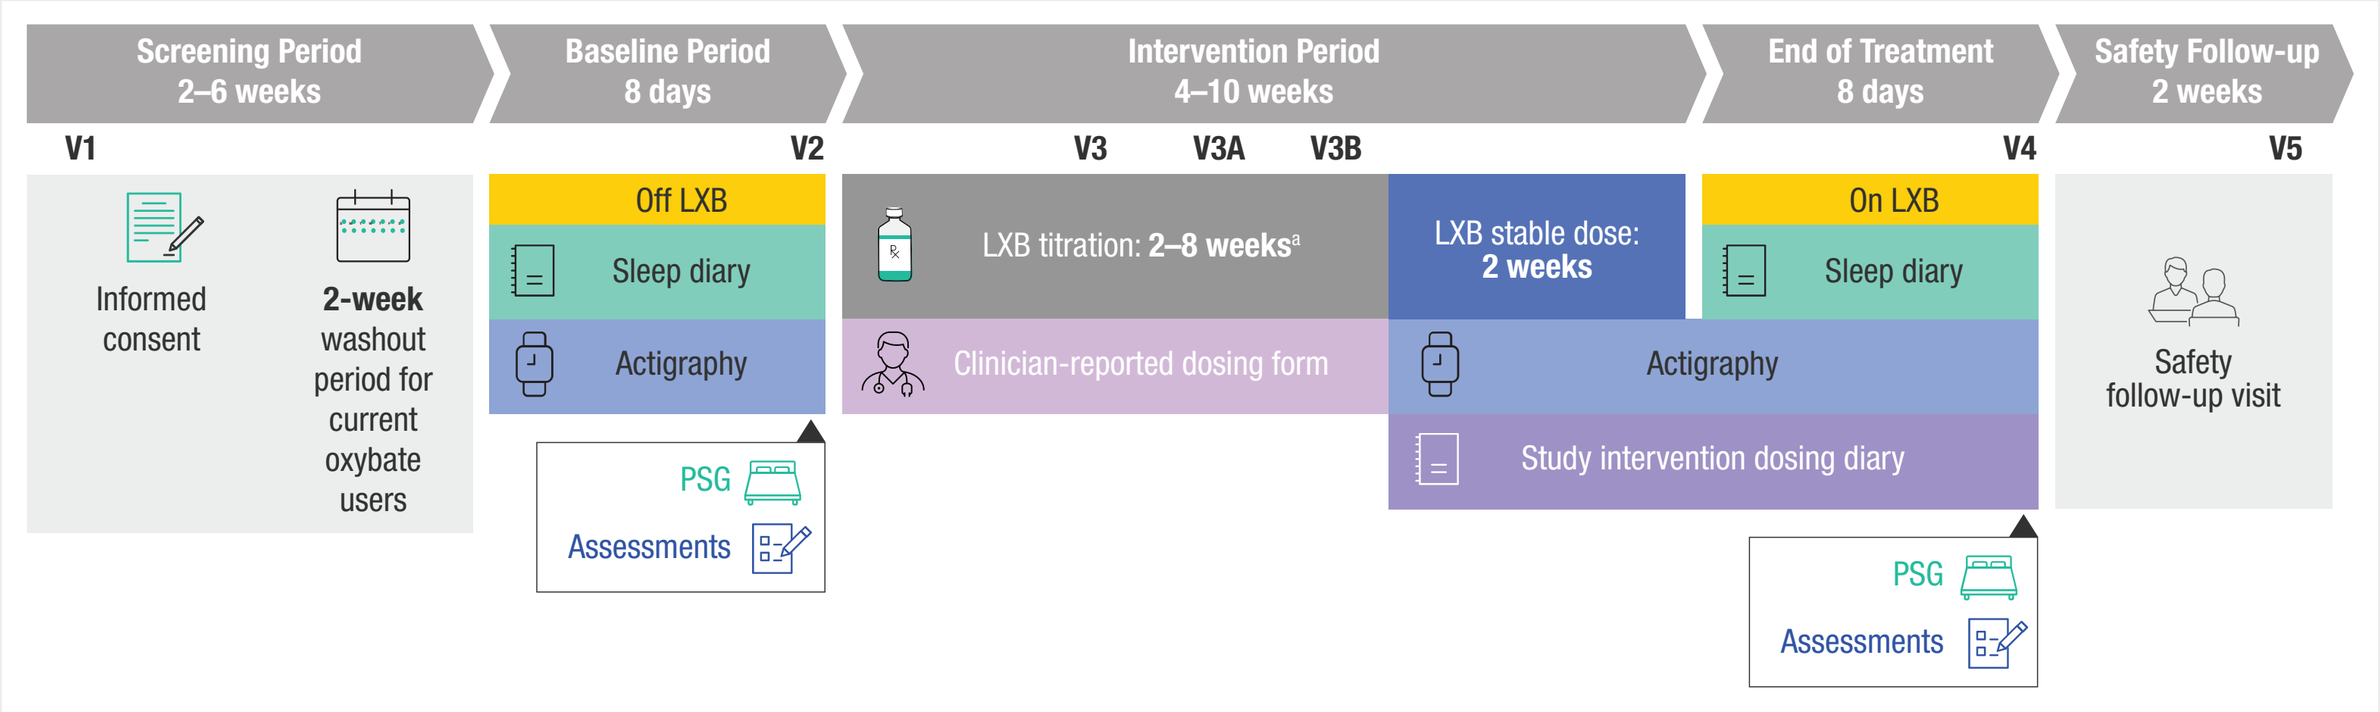
Screening Period 2–6 weeks
Baseline Period 8 days
Intervention Period 4–10 weeks
End of Treatment 8 days
Safety Follow-up 2 weeks
V1
V2
V3
V3A
V3B
V4
V5
Informed consent
2-week washout period for current oxybate users
Off LXB
Sleep diary
Actigraphy
PSG
Assessments
LXB titration: 2–8 weeks[a]
Clinician-reported dosing form
LXB stable dose: 2 weeks
Actigraphy
Study intervention dosing diary
On LXB
Sleep diary
PSG
Assessments
Safety follow-up visit

Adapted from Nichols DA, et al. *Neurol Ther.* 2025;14:1705–727. http://creativecommons.org/licenses/by-nc/4.0/.
[a]Weekly titration visits were by teleconference. Visit 3 occurred on titration day 14. Titration could take between 2 and 8 weeks. Additional in-clinic visits were scheduled for day 35 (visit 3A) and day 56 (visit 3B), as needed. The investigator could optimize participant dosage and move to SDP at visit 3, 3A, or 3B, but not during intervening weekly teleconferences.
LXB, low-sodium oxybate; PSG, polysomnography; SDP, stable-dose period; V, visit.

- DUET included a screening period (with a washout period for oxybate users), a baseline (BL) period, a titration period (participants began LXB treatment with individualized dosing adjustments to achieve their optimal dose), a stable-dose period (SDP; at optimal LXB dose), an end-of-treatment (EOT) period, and a safety follow-up
 - Participants with idiopathic hypersomnia could be treated with a once- or twice-nightly LXB regimen based on the investigator's discretion (per US prescribing label)[1]
- The primary outcome was change in Epworth Sleepiness Scale (ESS) from BL to EOT
- Secondary endpoints reported here include the change from BL to EOT in the Idiopathic Hypersomnia Severity Scale (IHSS) score, the Patient Global Impression of Severity (PGI-S), and the Patient Global Impression of Change (PGI-C) assessed at EOT
- Exploratory endpoints included change from BL to EOT in the Patient Health Questionnaire-9 (PHQ-9, score range 0 to 27) and the Patient Health Questionnaire-2 (PHQ-2; scored from the first 2 items of the PHQ-9 [range 0 to 6], and used to assess mood/anhedonia unrelated to sleep[12])
- Least squares (LS) mean differences (95% confidence interval [CI]) between EOT and BL were estimated using an analysis of covariance model adjusted for the BL value
 - *P* values were not adjusted for multiple comparisons
- Safety endpoints included incidence and severity of treatment-emergent adverse events (TEAEs) and the Columbia-Suicide Severity Rating Scale (C-SSRS) administered at every in-clinic visit
- The safety analysis set includes all participants who enrolled in the study and took their prescribed LXB regimen for ≥1 night after the BL period; the completer analysis set includes all participants who enrolled in the study, took their prescribed LXB regimen for ≥1 night after the BL period, completed the SDP, and completed the polysomnography EOT visit

Figure 2. Inclusion/Exclusion Criteria

Key Inclusion Criteria
- 18–75 years of age with primary diagnosis of idiopathic hypersomnia (per ICSD-3 criteria)
 - Individuals with and without sleep time were included[a]
- ESS score >10[b]
- Participants were allowed to continue taking concomitant alerting agents (stimulants or wake-promoting agents), but had to have been taking the same dosage for ≥1 month before screening visit 1 with no plan to adjust dosage during the study

Key Exclusion Criteria
- Untreated/inadequately treated sleep-disordered breathing (AHI >10)[c]
- History/presence of:
 - Other untreated/inadequately treated sleep disorder
 - Unstable/clinically significant medical condition
 - Behavioral/psychiatric disorder (such as active suicidal ideation or a current or past major depressive episode within the past year)
 - Bipolar disorder, bipolar-related disorder, schizophrenia, schizophrenia spectrum disorders
 - Other psychotic disorders according to DSM-5 criteria, neurologic disorder, or surgical history that might affect the participant's safety or interfere with study conduct

[a]Analyses were performed for the complete idiopathic hypersomnia cohort, with no distinction made between those with and without long sleep time. [b]At screening visit 1 or an ESS score >10 after the washout period, if taking an oxybate medication. [c]Defined as apnea-hypopnea index >10, with hypopnea definition including a ≥4% desaturation as per Rule 1B of *The AASM Manual for the Scoring of Sleep and Associated Events.*[13]
AASM, American Academy of Sleep Medicine; AHI, apnea-hypopnea index; DSM-5, *Diagnostic and Statistical Manual of Mental Disorders, 5th edition*; ESS, Epworth Sleepiness Scale; PSG, polysomnography.

Results

Table 1. Concurrent Psychiatric Disorders[a,b]

System Organ Class, n (%)[c] Preferred Term, n (%)[c]	Idiopathic Hypersomnia (N=46)
Psychiatric disorders	26 (56.5)
Depression	11 (23.9)
Anxiety	10 (21.7)
Attention deficit hyperactivity disorder	9 (19.6)
Major depression	2 (4.3)
Anxiety disorder	1 (2.2)
Autism spectrum disorder	1 (2.2)
Insomnia	1 (2.2)
Bruxism	1 (2.2)
Obsessive-compulsive disorder	1 (2.2)
Stress	1 (2.2)

[a]Safety analysis set. [b]Concurrent medical conditions/diseases are medical conditions/diseases that were ongoing as of the screening date, per medical history. [c]Participants reporting a condition/disease more than once within a system organ class/preferred term were counted only once for that system organ class/preferred term.
BL, baseline; LXB, low-sodium oxybate.

- Twenty-six enrolled participants had concurrent psychiatric comorbidities, most commonly depression (23.9%), anxiety (21.7%), and attention deficit hyperactivity disorder (19.6%)

Table 2. Demographics and Baseline Characteristics[a]

Characteristic	With Psychiatric Comorbidities (n=26)	Without Psychiatric Comorbidities (n=20)	Total Idiopathic Hypersomnia Cohort (N=46)
Age, years			
Mean (SD)	38.5 (12.1)	37.6 (11.7)	38.1 (11.8)
Median (min, max)	38.0 (20, 65)	34.0 (21, 68)	37.5 (20, 68)
Sex at birth, n (%)			
Male	4 (15.4)	5 (25.0)	9 (19.6)
Female	22 (84.6)	15 (75.0)	37 (80.4)
Gender identity, n (%)			
Male (including transgender man)	4 (15.4)	6 (30.0)	10 (21.7)
Female (including transgender woman)	22 (84.6)	14 (70.0)	36 (78.3)
Race, n (%)			
White	22 (84.6)	17 (85.0)	39 (84.8)
Black or African American	2 (7.7)	1 (5.0)	3 (6.5)
Asian	1 (3.8)	1 (5.0)	2 (4.3)
Native Hawaiian or other Pacific Islander	1 (3.8)	0	1 (2.2)
Multiple[b]	0	1 (5.0)	1 (2.2)
Ethnicity, n (%)			
Hispanic or Latino	3 (11.5)	7 (35.0)	10 (21.7)
Not Hispanic or Latino	23 (88.5)	12 (60.0)	35 (76.1)
Body mass index, kg/m²			
Mean (SD)	28.2 (6.8)	28.9 (6.0)	28.5 (6.4)
Median (min, max)	27.2 (19.3, 45.1)	28.2 (17.1, 41.4)	28.2 (17.1, 45.1)
Oxybate type at study entry, n (%)			
Naive[c]	20 (76.9)	17 (85.0)	37 (80.4)
Low-sodium oxybate	6 (23.1)	3 (15.0)	9 (19.6)
Sodium oxybate	0	0	0
Once-nightly sodium oxybate	0	0	0
Concomitant alerting agents, n (%)	17 (65.4)	2 (10.0)	19 (41.3)

[a]Safety analysis set. [b]Participant with more than one race. [c]No oxybate use within 2 weeks of entering the study.
Max, maximum; min, minimum; SD, standard deviation.

- Forty-six participants with idiopathic hypersomnia enrolled; 40 completed the study

Table 3. Mean Nightly LXB Dosage During Stable-Dose Period[a]

	With Psychiatric Comorbidities (n=21)[a]	Without Psychiatric Comorbidities (n=20)[a]	Total Idiopathic Hypersomnia Cohort (n=41)[a]
Total once nightly dosage, g/night	n=9	n=6	n=15
Mean (SD)	4.8 (1.0)	4.7 (1.3)	4.8 (1.1)
Total twice-nightly dosage, g/night	n=12	n=14	n=26
Mean (SD)	8.0 (1.3)	7.4 (1.1)	7.7 (1.2)
First nightly LXB dose	4.2 (0.7)	3.9 (0.9)	4.0 (0.8)
Second nightly LXB dose	3.8 (0.8)	3.5 (0.8)	3.6 (0.8)

[a]Includes participants from the safety analysis set who reached the SDP.
LXB, low-sodium oxybate; SD, standard deviation; SDP, stable-dose period.

- Fifteen (36.6%) participants took once-nightly LXB and 26 (63.4%) participants took twice-nightly LXB

Figure 3. ESS Scores Improved From BL to EOT in Participants With or Without Psychiatric Comorbidities[a]

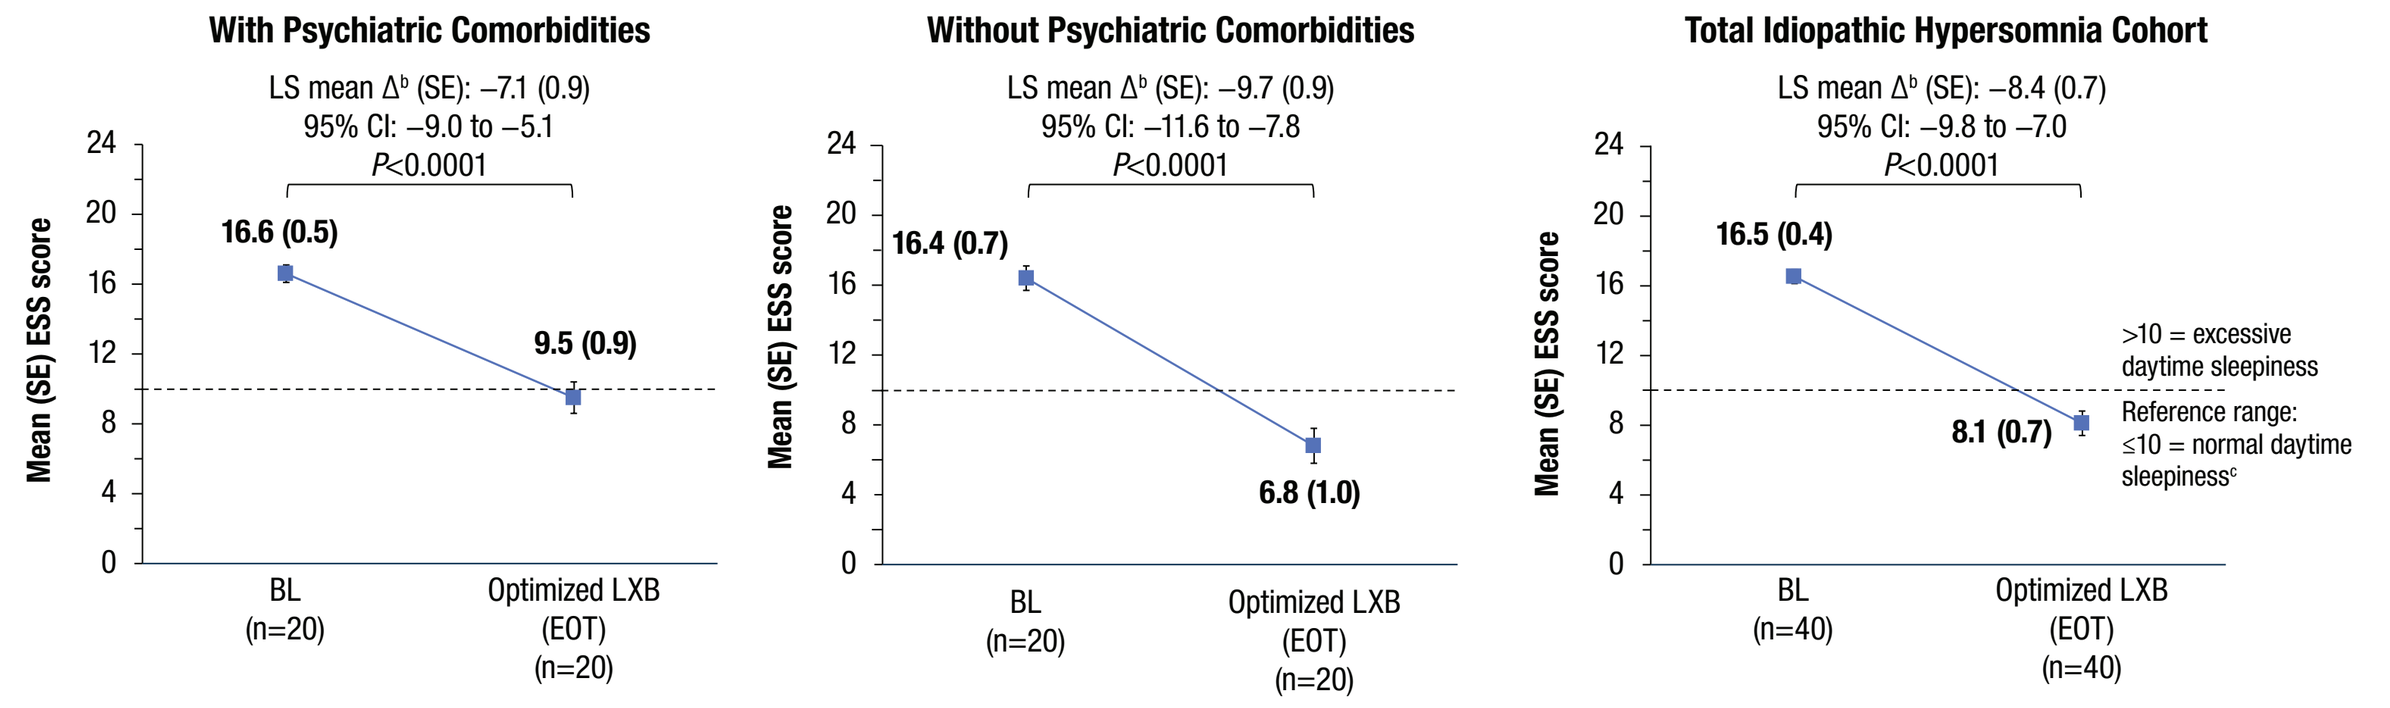
With Psychiatric Comorbidities
LS mean Δ[b] (SE): −7.1 (0.9)
95% CI: −9.0 to −5.1
P<0.0001
16.6 (0.5)
9.5 (0.9)
Without Psychiatric Comorbidities
LS mean Δ[b] (SE): −9.7 (0.9)
95% CI: −11.6 to −7.8
P<0.0001
16.4 (0.7)
6.8 (1.0)
Total Idiopathic Hypersomnia Cohort
LS mean Δ[b] (SE): −8.4 (0.7)
95% CI: −9.8 to −7.0
P<0.0001
16.5 (0.4)
8.1 (0.7)
>10 = excessive daytime sleepiness
Reference range: ≤10 = normal daytime sleepiness[c]
Mean (SE) ESS score
BL (n=20)
Optimized LXB (EOT) (n=20)
BL (n=40)
Optimized LXB (EOT) (n=40)

[a]Completer analysis set. [b]Difference between BL and EOT. [c]Established categories consider scores up to 10 normal.[14] The MCID for ESS was defined as 2 points according to an AASM systematic review and meta-analysis.[15]
AASM, American Academy of Sleep Medicine; BL, baseline; CI, confidence interval; EOT, end of treatment; ESS, Epworth Sleepiness Scale; LS, least squares; LXB, low sodium-oxybate; MCID, minimum clinically important difference; SE, standard error.

Table 4. Treatment-Emergent Adverse Events[a]

Participants, n (%)	With Psychiatric Comorbidities (n=26)	Without Psychiatric Comorbidities (n=20)	Total Idiopathic Hypersomnia Cohort (N=46)
With ≥1 TEAE	18 (69.2)	16 (80.0)	34 (73.9)
With ≥1 serious TEAE	0	1 (5.0)	0
With ≥1 TEAE related to treatment	14 (53.8)	16 (80.0)	30 (65.2)
With ≥1 TEAE leading to discontinuation	1 (3.8)	0	1 (2.2)
TEAEs occurring in ≥5% of participants			
Nausea	6 (23.1)	3 (15.0)	9 (19.6)
Dizziness	6 (23.1)	2 (10.0)	8 (17.4)
Headache	4 (15.4)	4 (20.0)	8 (17.4)
Vomiting	4 (15.4)	1 (5.0)	5 (10.9)
Middle insomnia	2 (7.7)	2 (10.0)	4 (8.7)
Somnolence	2 (7.7)	1 (5.0)	3 (6.5)
Anxiety	2 (7.7)	1 (5.0)	3 (6.5)
Enuresis	2 (7.7)	1 (5.0)	3 (6.5)
Decreased appetite	1 (3.8)	2 (10.0)	3 (6.5)
Sleep paralysis	2 (7.7)	0	2 (4.3)
Tremor	2 (7.7)	0	2 (4.3)
COVID-19	1 (3.8)	1 (5.0)	2 (4.3)
Influenza	1 (3.8)	1 (5.0)	2 (4.3)
Bruxism	1 (3.8)	1 (5.0)	2 (4.3)
Pain in extremity	1 (3.8)	1 (5.0)	2 (4.3)
Hyperhidrosis	1 (3.8)	1 (5.0)	2 (4.3)
Sinusitis	0	2 (10.0)	2 (4.3)
Pollakiuria	0	2 (10.0)	2 (4.3)
Cough	0	2 (10.0)	2 (4.3)
Nasal congestion	0	2 (10.0)	2 (4.3)
Brain fog	0	1 (5.0)	1 (2.2)
Hypersomnia	0	1 (5.0)	1 (2.2)
Dyspepsia	0	1 (5.0)	1 (2.2)
Hunger	0	1 (5.0)	1 (2.2)
Medical device site rash	0	1 (5.0)	1 (2.2)
Pyrexia	0	1 (5.0)	1 (2.2)
Confusional arousal	0	1 (5.0)	1 (2.2)
Mental status change	0	1 (5.0)	1 (2.2)
Hypoxia	0	1 (5.0)	1 (2.2)
Nocturnal dyspnea	0	1 (5.0)	1 (2.2)
Tendonitis	0	1 (5.0)	1 (2.2)
Blood iron decreased	0	1 (5.0)	1 (2.2)

[a]Safety analysis set.
TEAE, treatment-emergent adverse event.

- Overall, 18 participants (69.2%) with psychiatric comorbidities and 16 (80.0%) participants without psychiatric comorbidities reported ≥1 TEAE
 - One participant without psychiatric comorbidities reported a serious TEAE of hypoxia (concurrent with influenza), which was of moderate severity, determined by the investigator to be unrelated to the study drug, and resolved
 - One participant with medical history of anxiety and post-traumatic stress disorder and no history of depression or antidepressant use, discontinued LXB 3 days after initiation due to a TEAE of depression, which resolved after study drug discontinuation
- No participants endorsed suicidal ideation or behavior on the C-SSRS at any point during the study
 - During screening, 4 participants with idiopathic hypersomnia reported a history of suicidal ideation or behavior at some point in their lifetime, but there were no reports in the past 6 months

Figure 4. IHSS Scores Improved From BL to EOT in Participants With or Without Psychiatric Comorbidities[a]

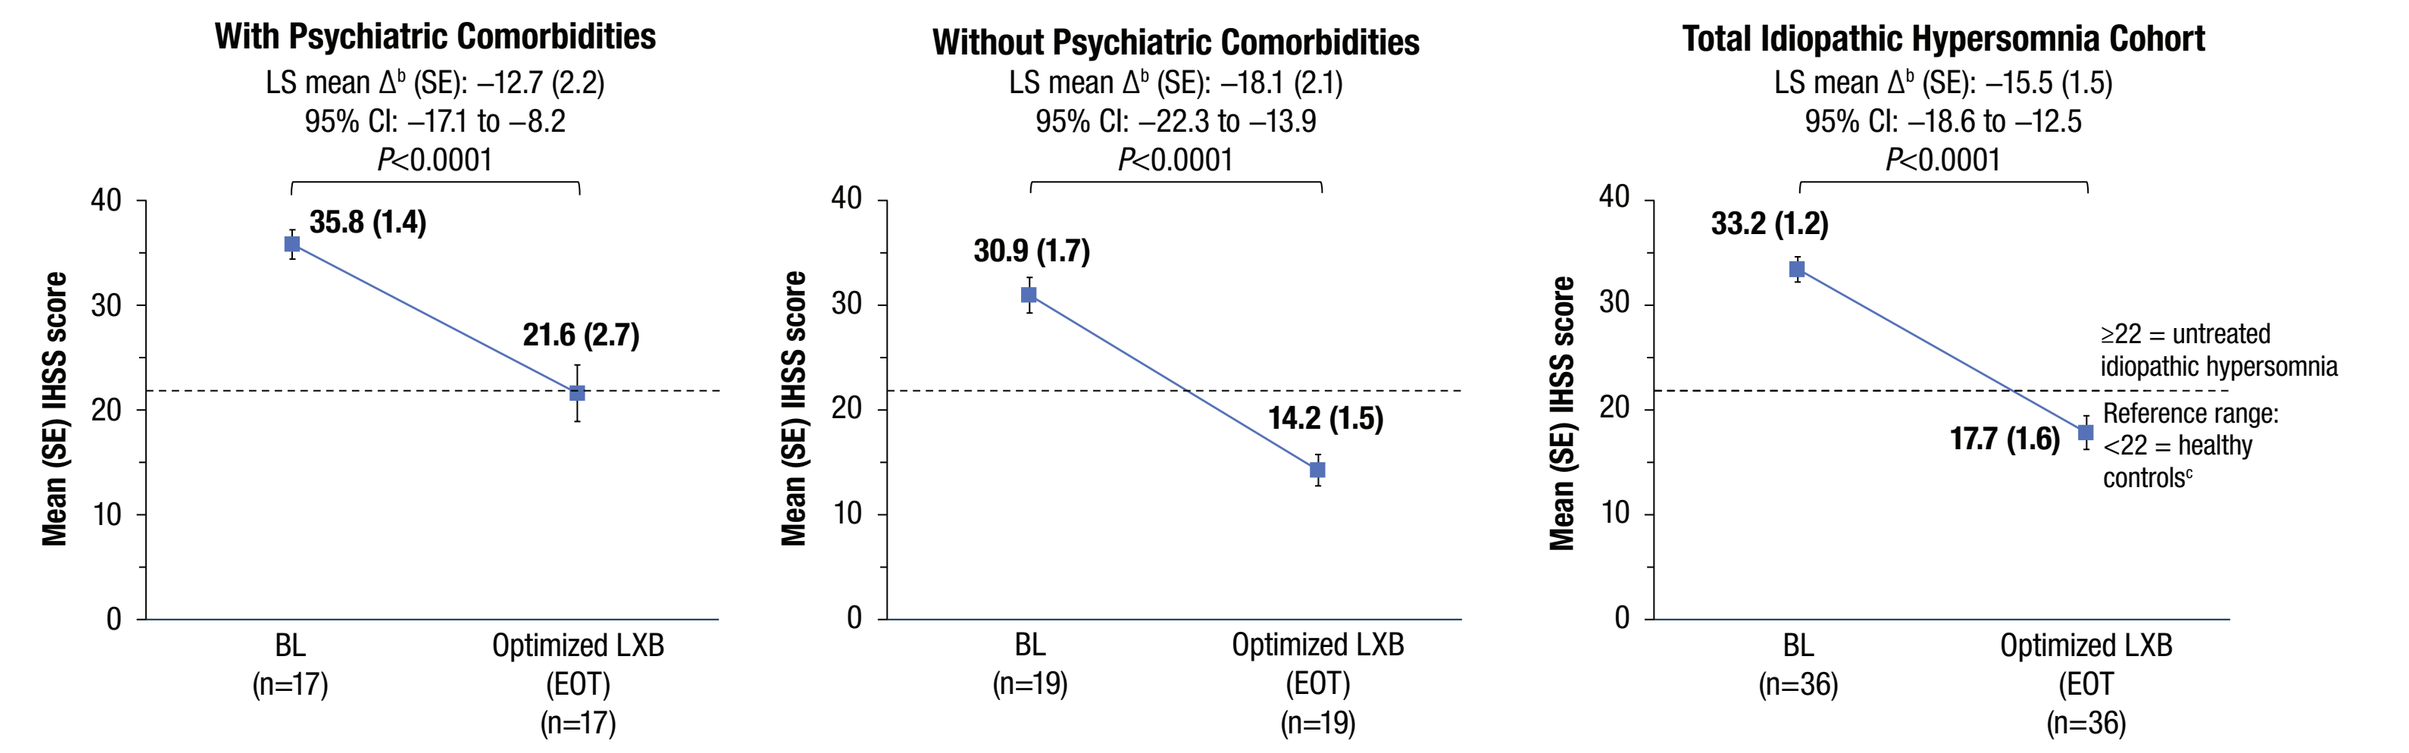
With Psychiatric Comorbidities
LS mean Δ[b] (SE): −12.7 (2.2)
95% CI: −17.1 to −8.2
P<0.0001
35.8 (1.4)
21.6 (2.7)
BL (n=17)
Optimized LXB (EOT) (n=17)
Without Psychiatric Comorbidities
LS mean Δ[b] (SE): −18.1 (2.1)
95% CI: −22.3 to −13.9
P<0.0001
30.9 (1.7)
14.2 (1.5)
BL (n=19)
Optimized LXB (EOT) (n=19)
Total Idiopathic Hypersomnia Cohort
LS mean Δ[b] (SE): −15.5 (1.5)
95% CI: −18.6 to −12.5
P<0.0001
33.2 (1.2)
17.7 (1.6)
BL (n=36)
Optimized LXB (EOT (n=36)
≥22 = untreated idiopathic hypersomnia
Reference range: <22 = healthy controls[c]
Mean (SE) IHSS score

[a]Completer analysis set. [b]Difference between BL and EOT. [c]An IHSS score of 22 is the optimal cutoff for distinguishing between untreated patients with idiopathic hypersomnia and community-dwelling controls.[16] The MCID for IHSS was defined as 4 points according to a cross-sectional analysis.[17]
BL, baseline; CI, confidence interval; EOT, end of treatment; IHSS, Idiopathic Hypersomnia Severity Scale; LS, least squares; LXB, low sodium-oxybate; SE, standard error.

Figure 5. PHQ-9 (A) and PHQ-2 (B) Scores Improved From BL to EOT in Participants With or Without Psychiatric Comorbidities[a]

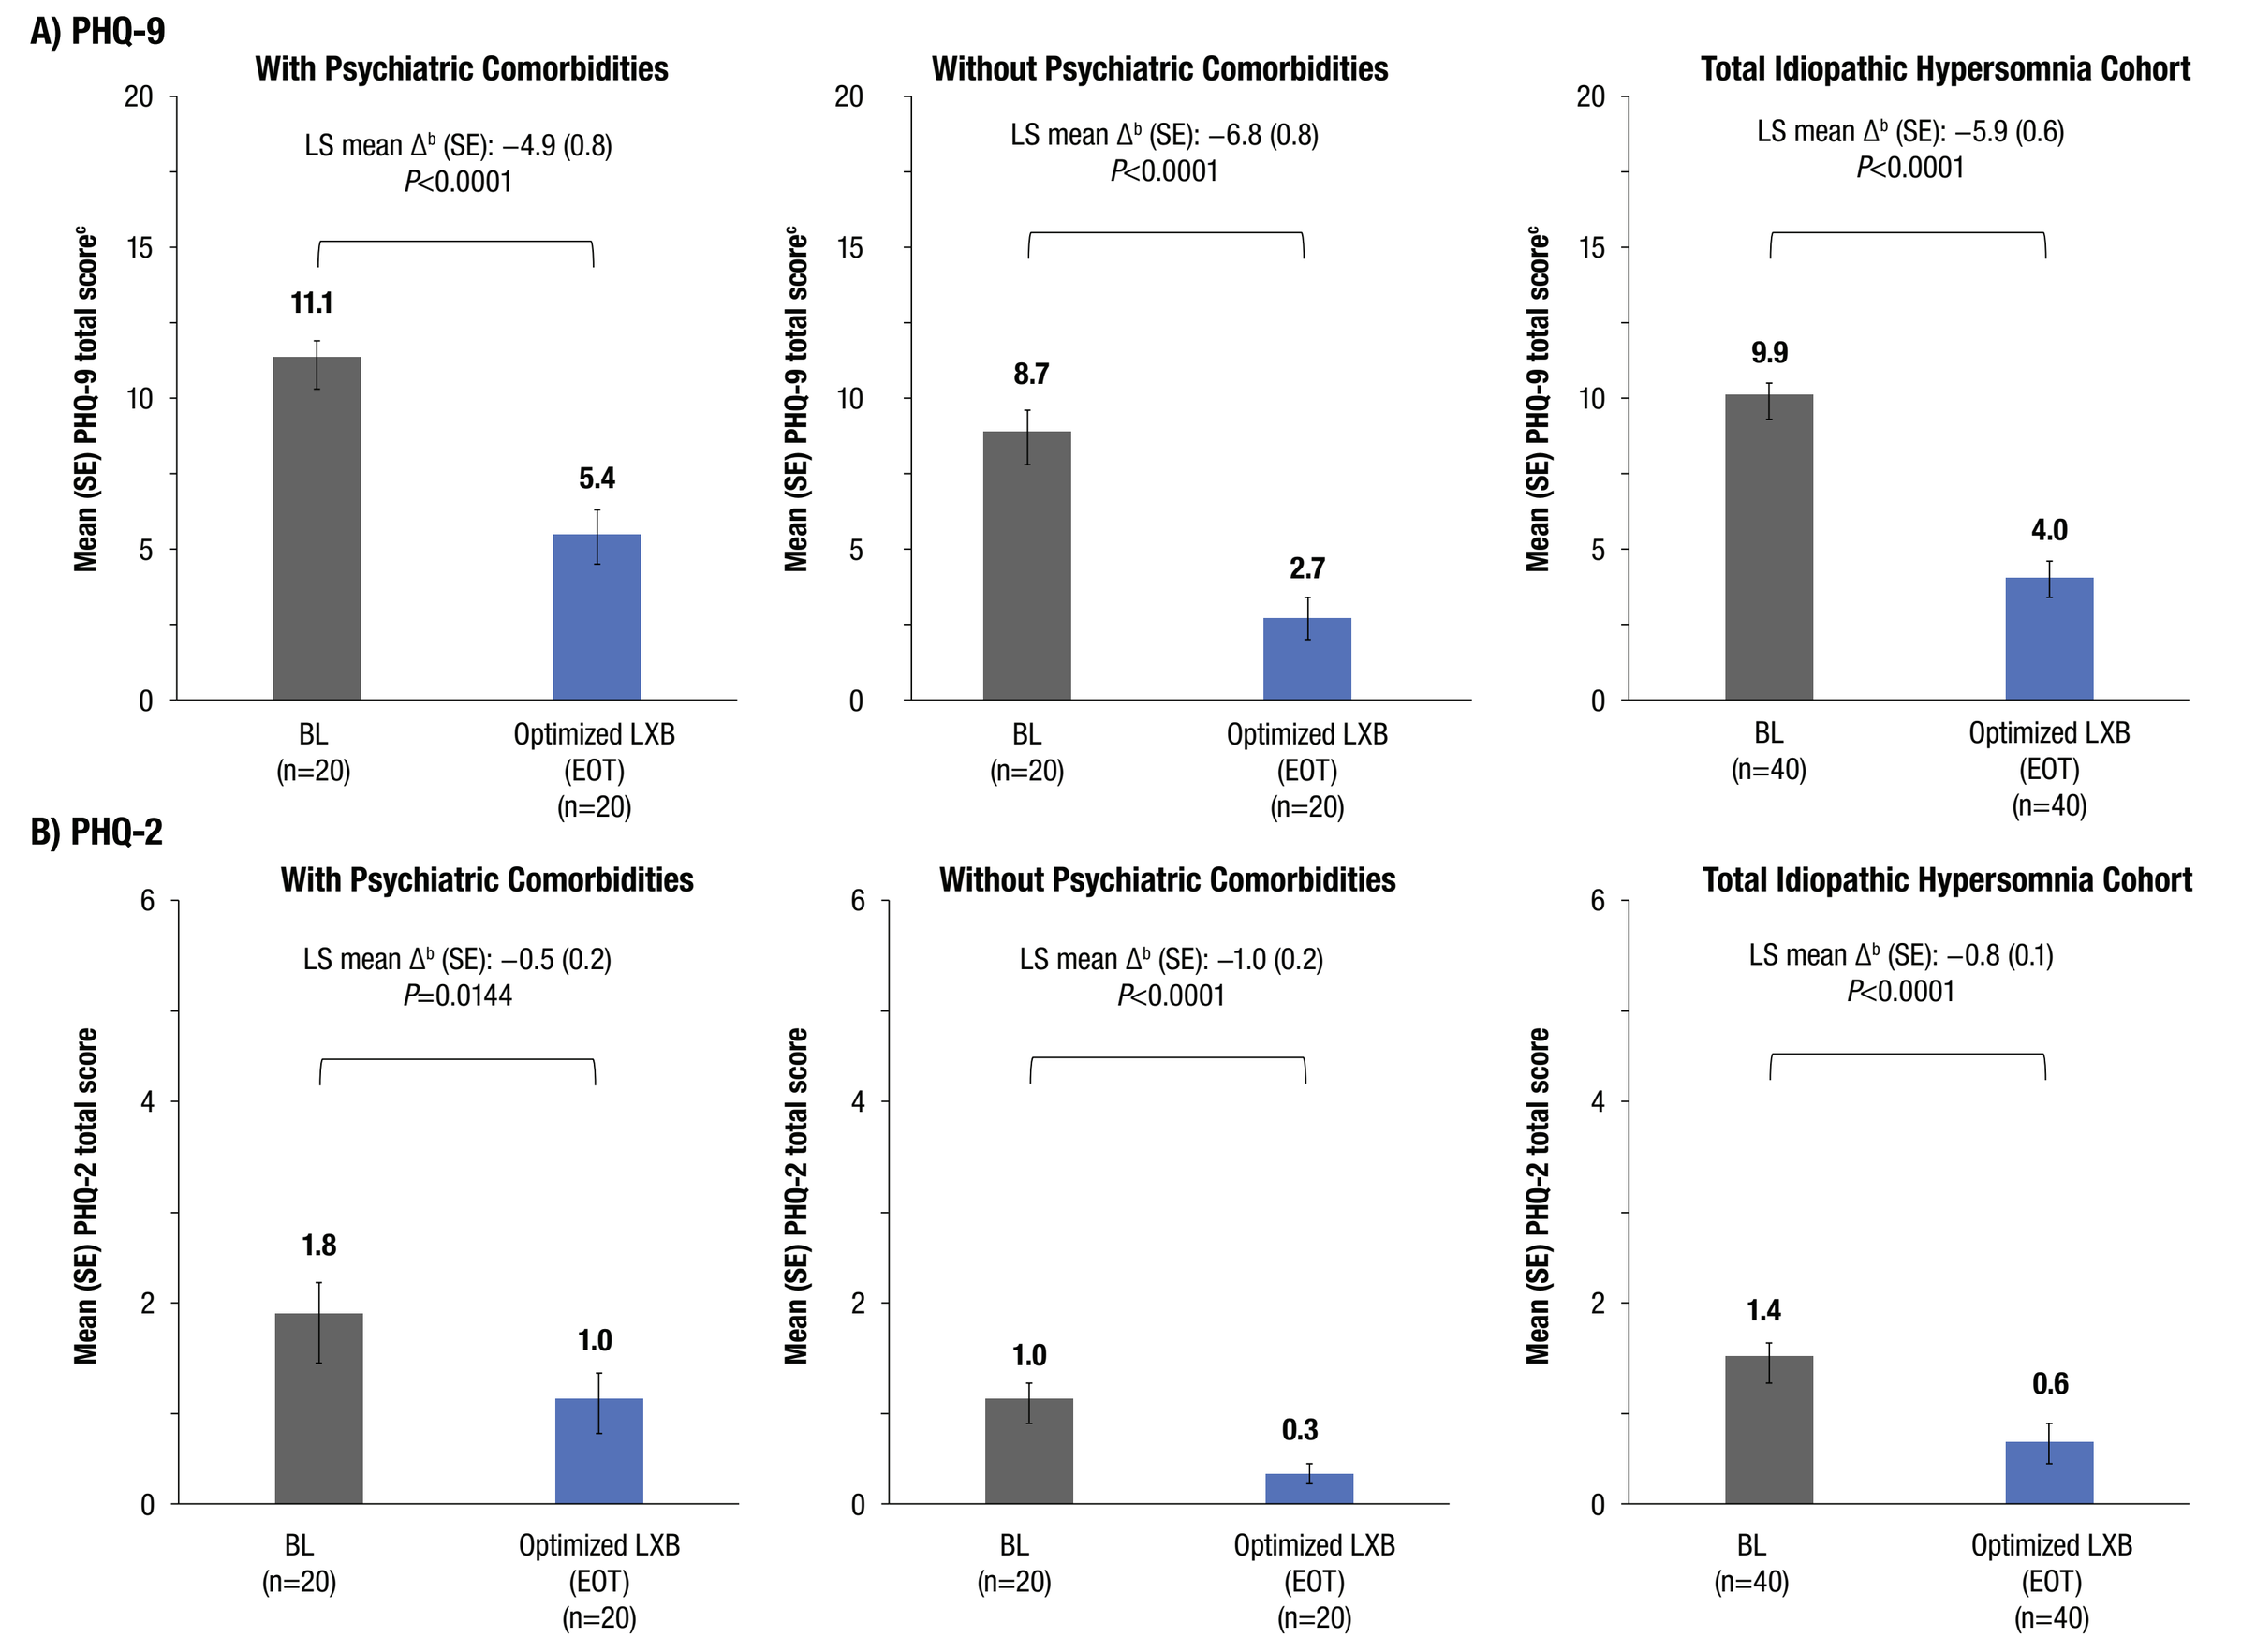
A) PHQ-9
With Psychiatric Comorbidities
LS mean Δ[b] (SE): −4.9 (0.8)
P<0.0001
11.1
5.4
Without Psychiatric Comorbidities
LS mean Δ[b] (SE): −6.8 (0.8)
P<0.0001
8.7
2.7
Total Idiopathic Hypersomnia Cohort
LS mean Δ[b] (SE): −5.9 (0.6)
P<0.0001
9.9
4.0
Mean (SE) PHQ-9 total score[c]
B) PHQ-2
With Psychiatric Comorbidities
LS mean Δ[b] (SE): −0.5 (0.2)
P=0.0144
1.8
1.0
Without Psychiatric Comorbidities
LS mean Δ[b] (SE): −1.0 (0.2)
P<0.0001
1.0
0.3
Total Idiopathic Hypersomnia Cohort
LS mean Δ[b] (SE): −0.8 (0.1)
P<0.0001
1.4
0.6
Mean (SE) PHQ-2 total score
BL (n=20)
Optimized LXB (EOT) (n=20)
BL (n=40)
Optimized LXB (EOT) (n=40)

[a]Completer analysis set. [b]Difference between BL and EOT. [c]Level of depressive symptom severity per PHQ-9 score: minimal, 0–4; mild, 5–9; moderate, 10–14; moderately severe, 15–19; severe, 20–27.[18]
BL, baseline; EOT, end of treatment; LS, least squares; LXB, low-sodium oxybate; PHQ, Patient Health Questionnaire; SE, standard error.

Figure 6. Proportions of Participants Reporting "None to Minimal" Severity of Depressive Symptoms on the PHQ-9 Increased From BL to EOT in Participants With or Without Psychiatric Comorbidities[a]

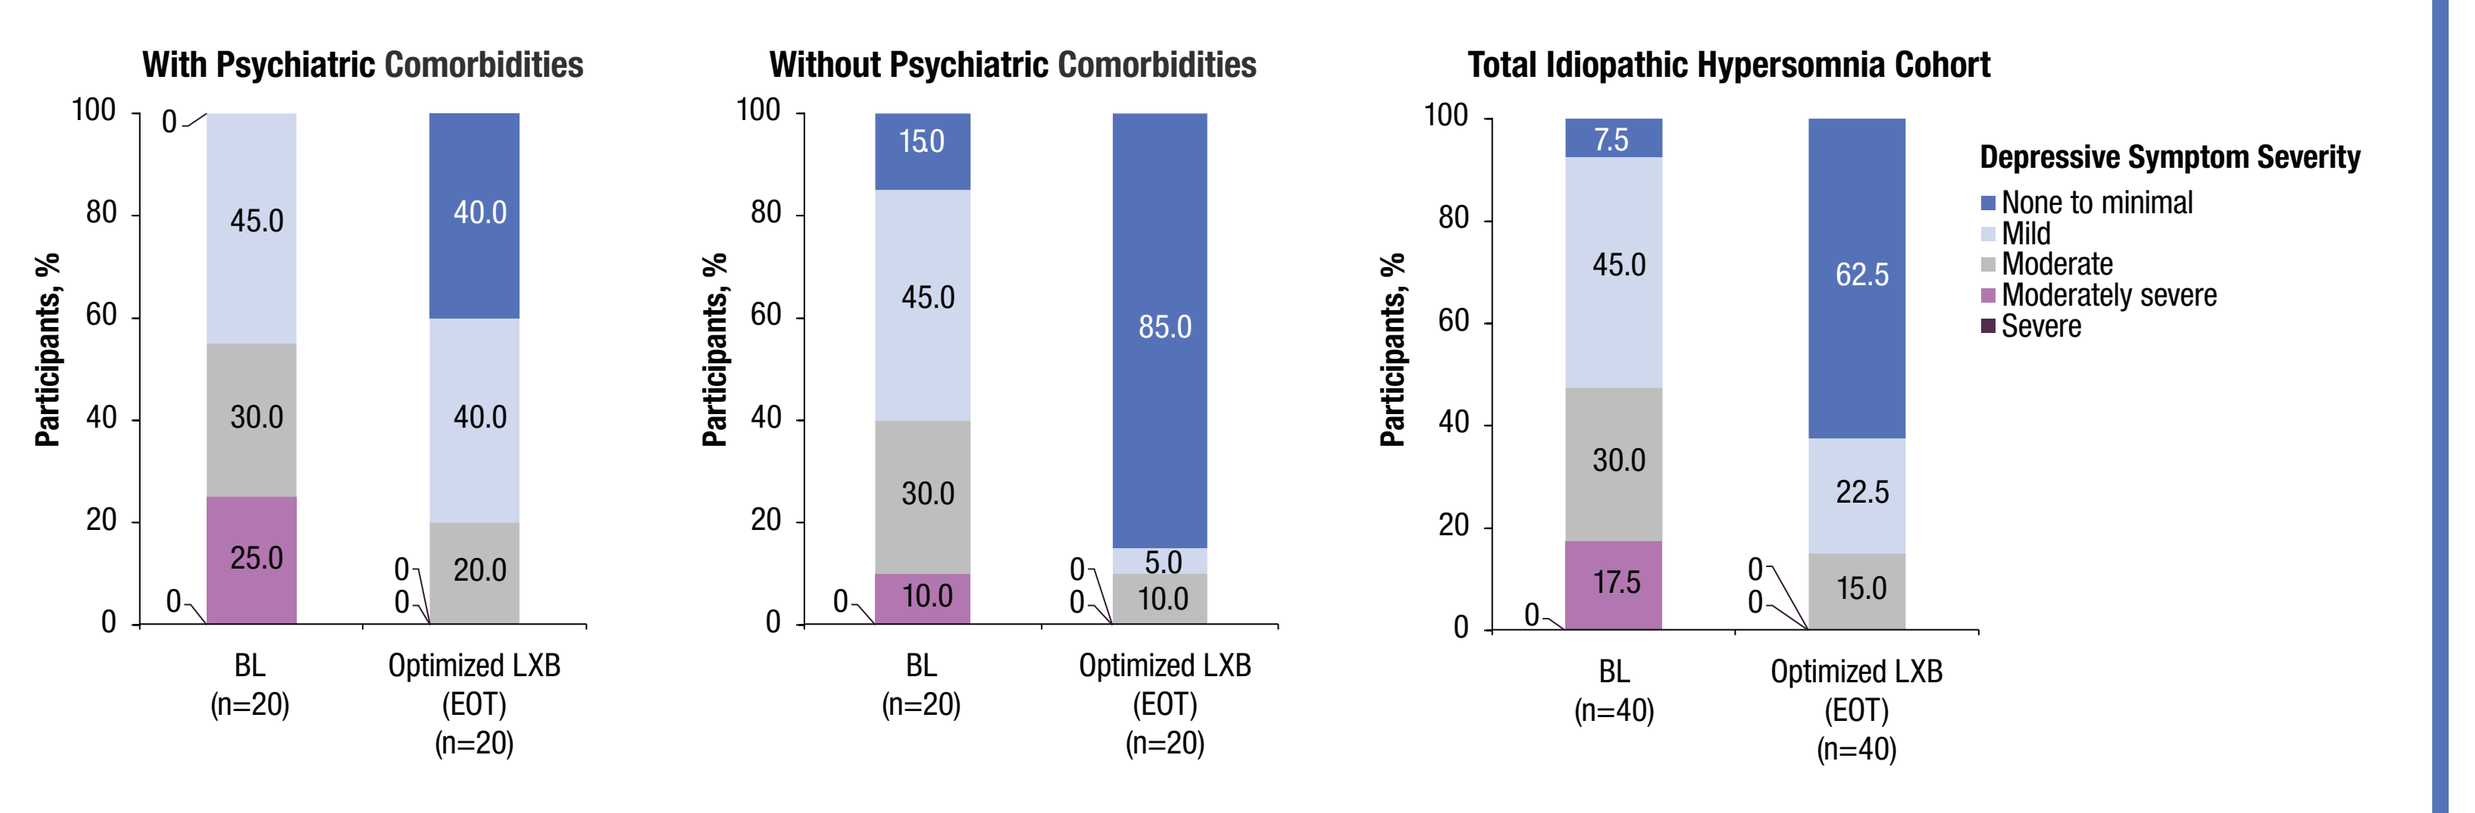
With Psychiatric Comorbidities
Without Psychiatric Comorbidities
Total Idiopathic Hypersomnia Cohort
Participants, %
Depressive Symptom Severity
None to minimal
Mild
Moderate
Moderately severe
Severe
BL (n=20)
Optimized LXB (EOT) (n=20)
BL (n=40)
Optimized LXB (EOT) (n=40)

[a]Completer analysis set.
BL, baseline; EOT, end of treatment; LXB, low-sodium oxybate; PHQ, Patient Health Questionnaire.

Figure 7. Participants With or Without Psychiatric Comorbidities Reported Improvement in Overall Idiopathic Hypersomnia Disease on the Patient Global Impression of Severity (A) and the Patient Global Impression of Change (B) Scales[a]

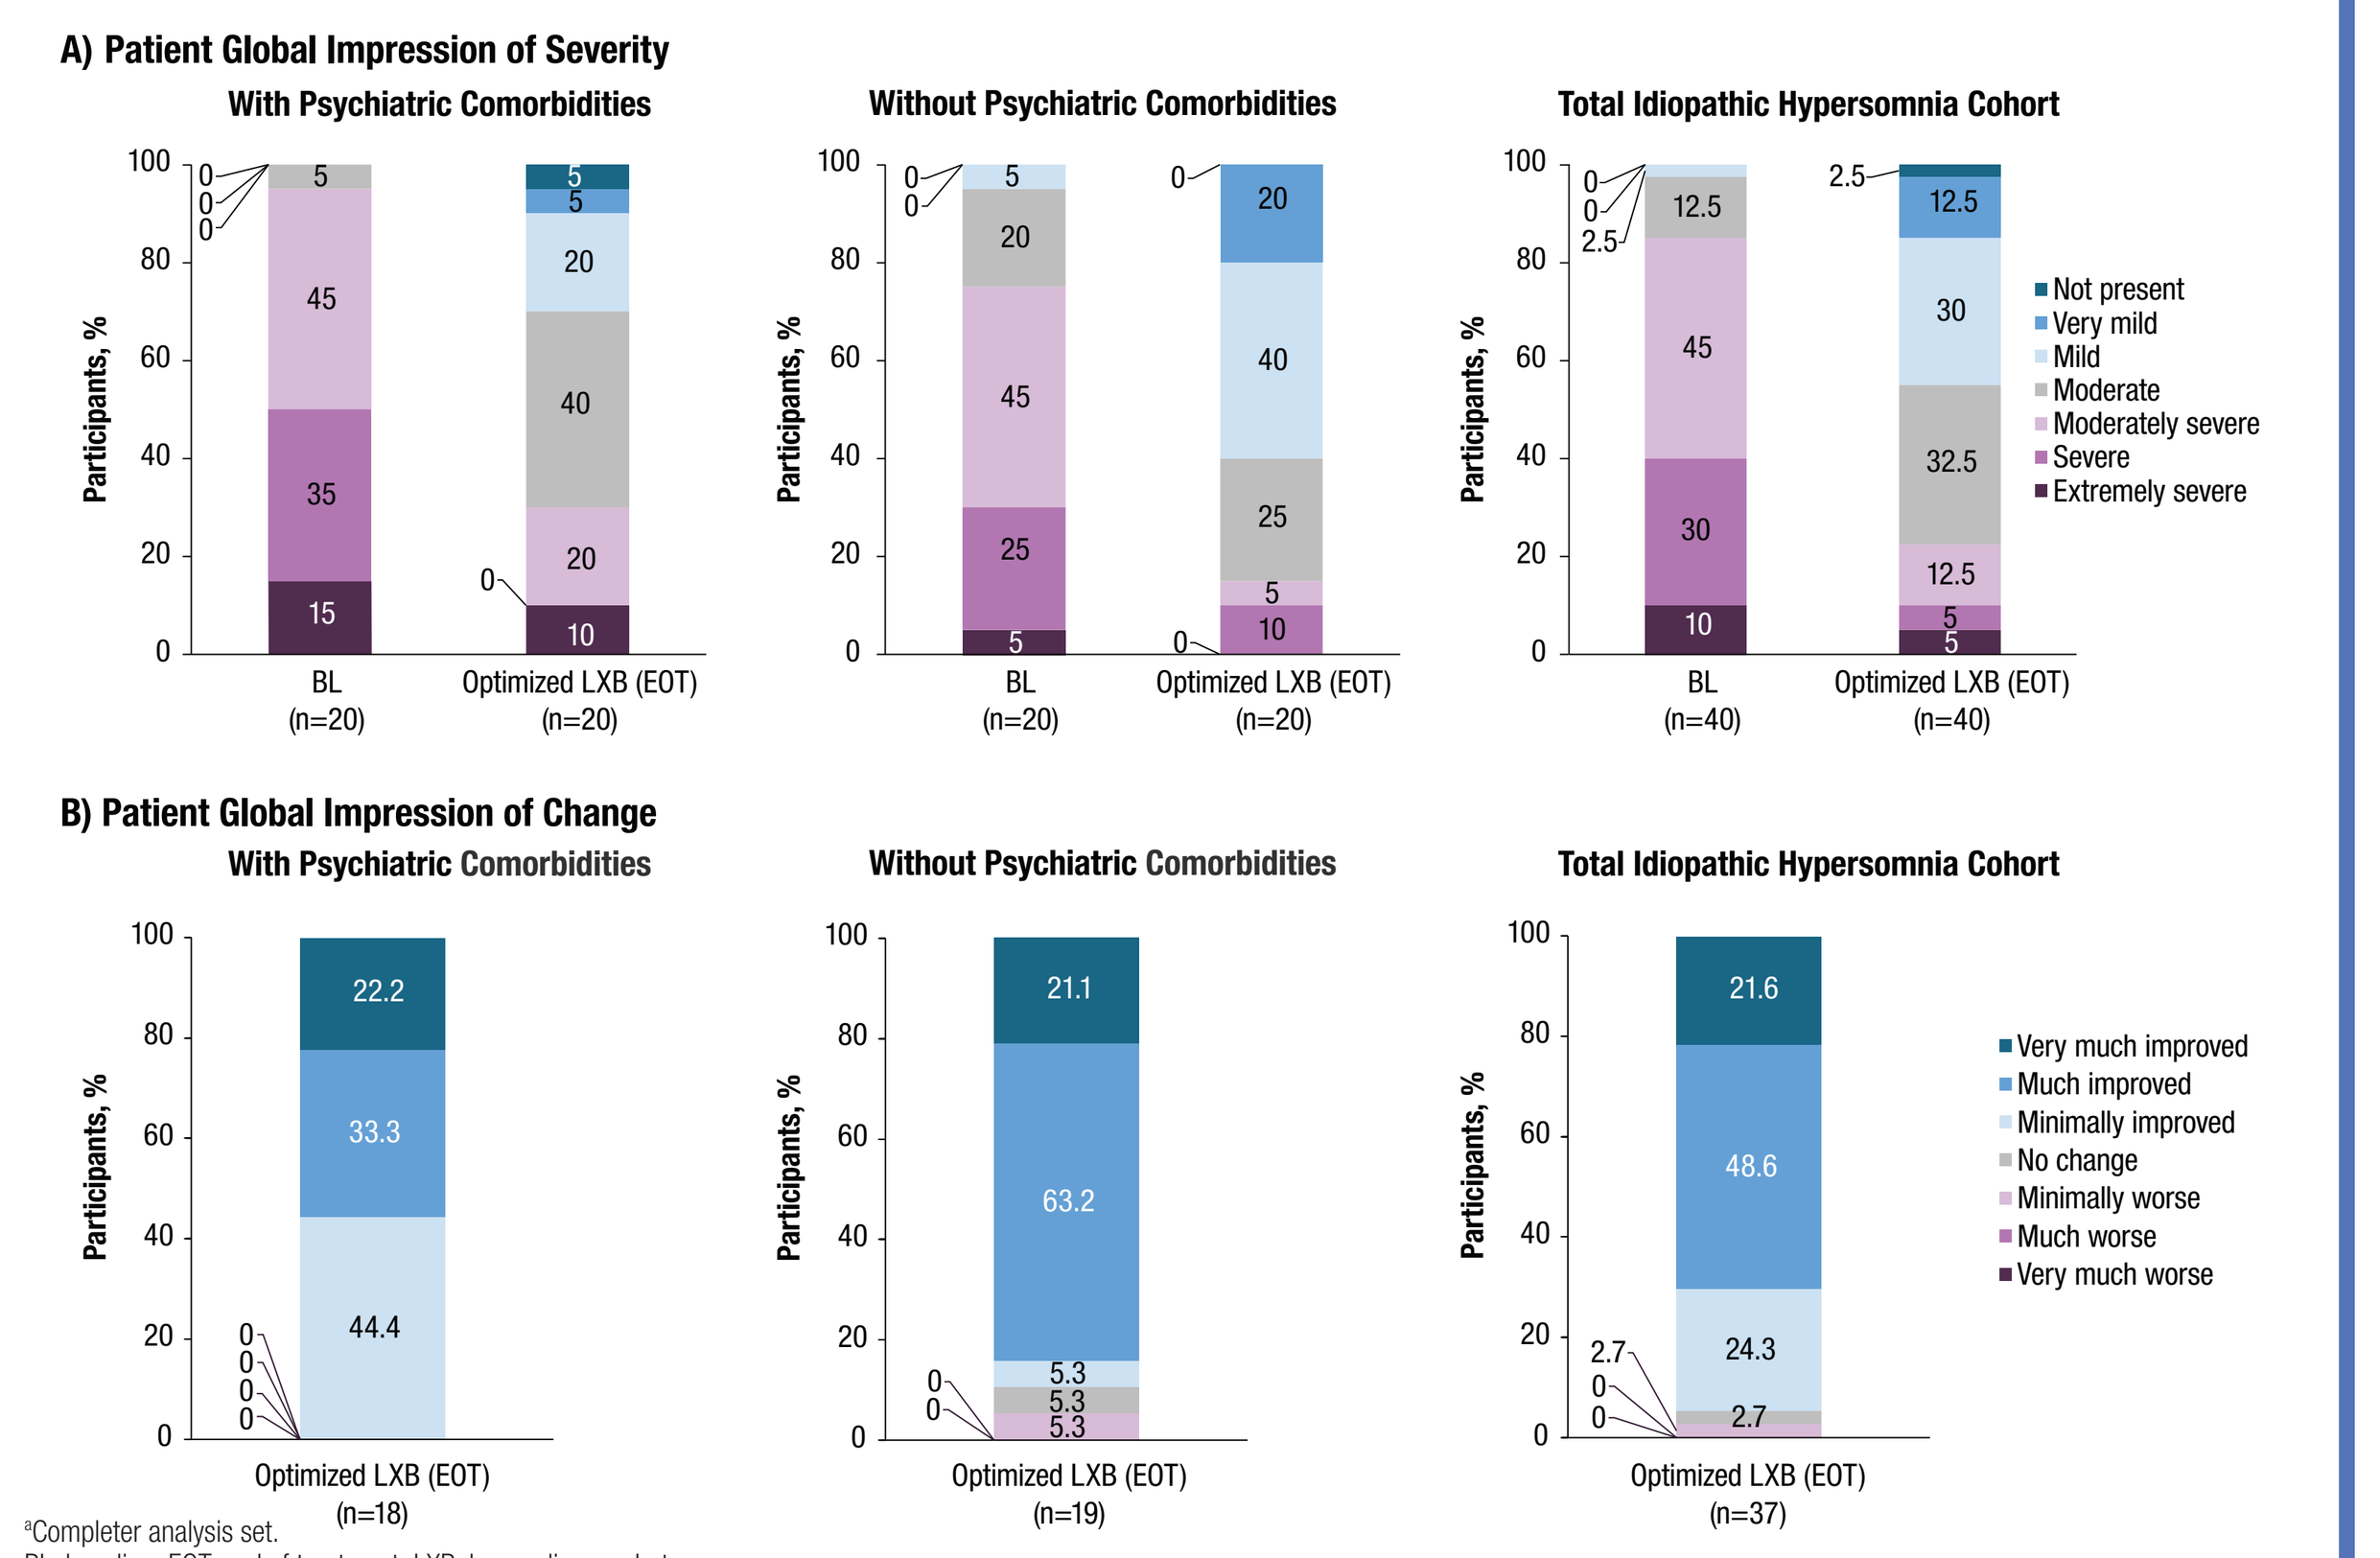
A) Patient Global Impression of Severity
With Psychiatric Comorbidities
Without Psychiatric Comorbidities
Total Idiopathic Hypersomnia Cohort
Participants, %
Not present
Very mild
Mild
Moderate
Moderately severe
Severe
Extremely severe
BL (n=20)
Optimized LXB (EOT) (n=20)
BL (n=40)
Optimized LXB (EOT) (n=40)
B) Patient Global Impression of Change
Very much improved
Much improved
Minimally improved
No change
Minimally worse
Much worse
Very much worse
Optimized LXB (EOT) (n=18)
Optimized LXB (EOT) (n=19)
Optimized LXB (EOT) (n=37)

[a]Completer analysis set.
BL, baseline; EOT, end of treatment; LXB, low-sodium oxybate.

Conclusions

- Results demonstrated the effectiveness of LXB in participants with idiopathic hypersomnia, both with and without psychiatric comorbidities
 - Of note, participants with psychiatric comorbidities had numerically greater mean IHSS, PHQ-9, and PHQ-2 scores at baseline than those without psychiatric comorbidities
 - Participants in both subgroups showed improvements in daytime sleepiness (ESS scores), idiopathic hypersomnia symptoms (IHSS scores), and overall disease (PGI-C and PGI-S scores) with optimized LXB treatment compared with BL (off-LXB); notably, mean changes in ESS and IHSS scores from BL to EOT in both subgroups exceeded MCID thresholds[15, 17]
 - Participants in both subgroups showed improvements in PHQ-9 and PHQ-2 scores with optimized LXB treatment compared with BL, demonstrating reduced depressive symptoms
- TEAEs were consistent with the known LXB safety profile
- Study limitations include the open-label and single-arm design; causality cannot be established. Additionally, analyses were based on the completer analysis set of participants who reached a stable LXB dosage and may not represent the experience of all individuals starting LXB treatment

References: **1.** Xywav® (calcium, magnesium, potassium, and sodium oxybates) oral solution, CIII [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc.; 2023. **2.** Szarfman A, et al. *N Engl J Med.* 1995;333:1291. **3.** US Food and Drug Administration. Clinical review for Binosto, NDA 202344. 2012. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2012/202344Orig1s000MedR.pdf. **4.** US Food and Drug Administration. Quantitative labeling of sodium, potassium, and phosphorus for human over-the-counter and prescription drug products. Guidance for industry. 2022. https://www.fda.gov/regulatory-information/search-fda-guidance-documents/quantitative-labeling-sodium-potassium-and-phosphorus-human-over-counter-and-prescription-drug. **5.** Saad R, et al. *Sleep Epidemiol.* 2023;3:100059. **6.** Lillaney P, et al. Presented at: Annual Meeting of the Associated Professional Sleep Societies; June 3-7, 2023; Indianapolis, IN. **7.** Chepke C, et al. Presented at: Annual Psych Congress; September 6-10, 2023; Nashville, TN. **8.** Provigil [package insert]. North Wales, PA: Teva Pharmaceuticals; 2018. **9.** Biaxin [package insert]. North Chicago, IL: AbbVie; 2013. **10.** Wakix [package insert]. Plymouth Meeting, PA: Harmony Biosciences; 2022. **11.** Ritalin [package insert]. East Hanover, NJ: Novartis Pharmaceuticals Corporation; 2019. **12.** Kroenke K, et al. *Med Care.* 2003;41:1284-92. **13.** Berry RB, et al. The AASM Manual for the Scoring of Sleep and Associated Events: Rules, Terminology and Technical Specifications, Version 3. Darien, IL: American Academy of Sleep Medicine; 2023. **14.** Johns MW. *Sleep.* 1991;14:540-5. **15.** Maski K, et al. *J Clin Sleep Med.* 2021;17:1895-945. **16.** Dauvilliers Y, et al. *Neurology.* 2019;92:e1754-e62. **17.** Rassu AL, et al. *J Clin Sleep Med.* 2022;18(2):617-629. **18.** Kroenke K, et al. *J Gen Intern Med.* 2001;16:606-13.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees Martina Kusi-Mensah, PharmD, and Emily Bruggeman, PhD, provided medical writing support and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: **DT Plante** is a consultant and advisory board member for Jazz Pharmaceuticals; an advisory board member for Apnimed; a consultant for Aditum Bio and Teva Pharmaceuticals (Australia); and has served as a consultant/advisory board member for Alkermes, Centessa, Harmony Biosciences, and Takeda. **C Ruoff** has served as an advisory board member for Alkermes, Eisai, Jazz Pharmaceuticals, and Takeda and has received grant funding from Jazz Pharmaceuticals. **DA Nichols, J Dai, MT Kirby,** and **M Whalen** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **TL Steininger** is a former full-time employee and current contract worker for Jazz Pharmaceuticals who has received shares of Jazz Pharmaceuticals, plc. **A Cairns** is a full-time contract worker and advisory board member for Jazz Pharmaceuticals. **LD Schneider** has received personal compensation for serving on advisory boards and speakers bureaus for Avadel, Eisai, and Jazz Pharmaceuticals.

Scan this code to access this poster online. This code is not for promotional purposes.

Presented at Psych Congress 2025; September 17–21, 2025; San Diego, CA, USA