124

Improvement in Bothersome Mood, Cognitive, and Functional Impacts of Idiopathic Hypersomnia After Low-Sodium Oxybate Treatment in the DUET Study

David T. Plante, MD, PhD[1]; Logan D. Schneider, MD[2]; Deborah A. Nichols, MS[3]; Teresa L. Steininger, PhD[3]; Jing Dai, PhD[3]; M. Todd Kirby, PhD[4,5]; Marisa Whalen, PharmD[4]; Cristina Casstevens, PhD[3]; Alyssa Cairns, PhD[4]; Chad M. Ruoff, MD[6]

[1]Department of Psychiatry, University of Wisconsin–Madison, Madison, WI, USA; [2]Stanford University Center for Sleep Sciences and Medicine, Stanford, CA, USA; [3]Jazz Pharmaceuticals, Palo Alto, CA, USA; [4]Jazz Pharmaceuticals, Philadelphia, PA, USA; [5]Department of Psychiatry & Health Behavior, Medical College of Georgia at Augusta University, Augusta, GA, USA; [6]Mayo Clinic, Scottsdale, AZ, USA

Introduction

- Idiopathic hypersomnia is a central disorder of hypersomnolence characterized by excessive daytime sleepiness, sleep inertia, long and unrefreshing naps, and subjective cognitive impairment; some patients experience prolonged nighttime sleep[1-3]
- A previous report of patient interviews identified the most bothersome impacts of idiopathic hypersomnia, which included mood disturbances, cognitive complaints, and personal/professional difficulties[4]
 - Data on idiopathic hypersomnia treatment effects on these impacts may help guide treatment decisions[4]
- Low-sodium oxybate (LXB; calcium, magnesium, potassium, and sodium oxybates; Xywav®) is approved by the US Food and Drug Administration for treating idiopathic hypersomnia in adults and excessive daytime sleepiness or cataplexy in patients ≥7 years of age with narcolepsy[5]
- Jazz DUET (**D**evelop hypersomnia **U**nderstanding by **E**valuating low-sodium oxybate **T**reatment) was a phase 4, prospective, open-label, multiple-cohort study (NCT05875974) evaluating the effectiveness of LXB on daytime and nighttime symptoms and functional outcomes in participants with idiopathic hypersomnia or narcolepsy

Objective

- This analysis evaluated the effectiveness of LXB on functional outcomes determined to be most bothersome to patients, including mood, cognitive, and work productivity and activity impairments, in DUET study participants with idiopathic hypersomnia

Methods

Figure 1. Study Design

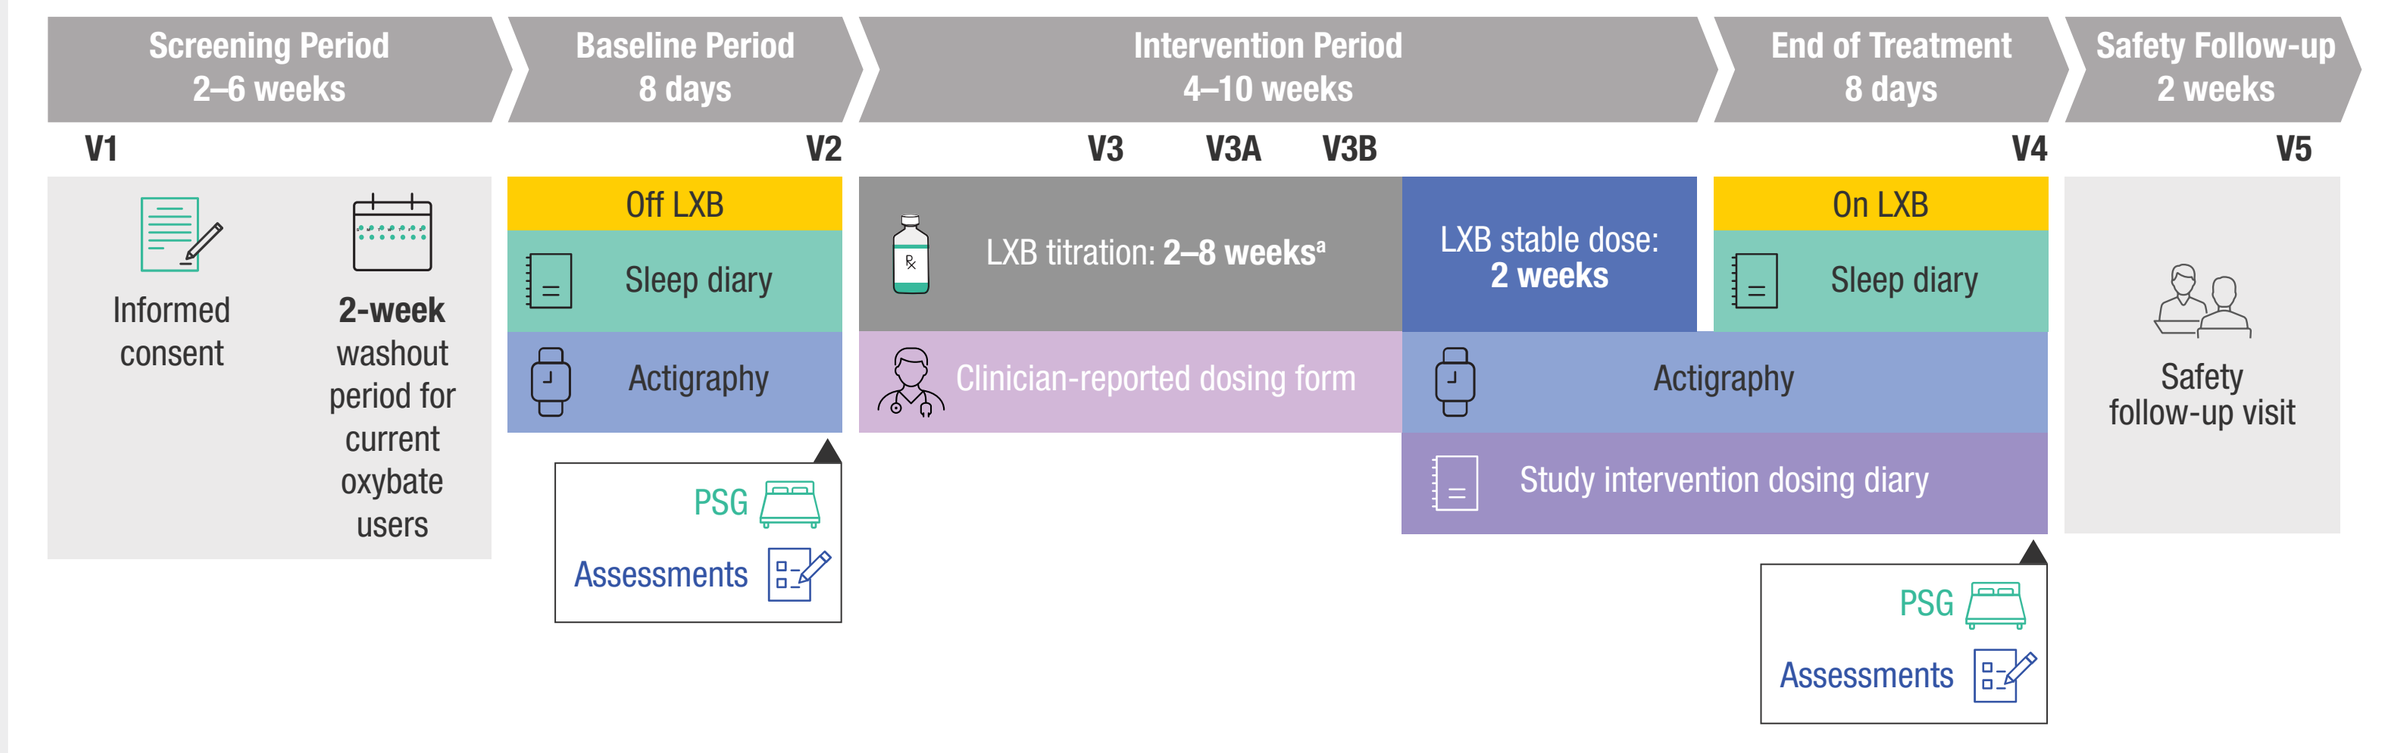

Screening Period 2–6 weeks
Baseline Period 8 days
Intervention Period 4–10 weeks
End of Treatment 8 days
Safety Follow-up 2 weeks
V1
V2
V3 V3A V3B
V4
V5
Informed consent
2-week washout period for current oxybate users
Off LXB
Sleep diary
Actigraphy
PSG
Assessments
LXB titration: 2–8 weeks[a]
Clinician-reported dosing form
LXB stable dose: 2 weeks
Actigraphy
Study intervention dosing diary
On LXB
Sleep diary
Safety follow-up visit

Adapted from Nichols DA, et al. *Neurol Ther.* 2025;14:1705–727. http://creativecommons.org/licenses/by-nc/4.0/.
[a]Weekly titration visits were by teleconference. Visit 3 occurred on titration day 14. Titration could take between 2 and 8 weeks. Additional in-clinic visits were scheduled for day 35 (visit 3A) and day 56 (visit 3B), as needed. Investigator could optimize participant dosage and move participant to SDP at visit 3, 3A, or 3B, but not during intervening weekly teleconferences.
LXB, low-sodium oxybate; PSG, polysomnography; SDP, stable-dose period; V, visit.

- Individuals were screened at DUET study sites. Eligible participants were 18 to 75 years of age and diagnosed with idiopathic hypersomnia (per *International Classification of Sleep Disorders – Third Edition* [ICSD-3] criteria[6]) and had an Epworth Sleepiness Scale[7] score >10
 - Individuals with and without long sleep time were included
 - Participants with untreated or inadequately treated sleep-disordered breathing (apnea-hypopnea index >10) or history/presence of other untreated sleep disorder or other condition that might interfere with study conduct were excluded
- Participants were not taking LXB at study baseline (BL). Participants taking alerting agents (stimulants or wake-promoting agents) were allowed to continue during the study period at a stable dosage
- During the BL period, assessments were administered. These included the Patient Health Questionnaire-9 (PHQ-9), British Columbia Cognitive Complaints Inventory (BC-CCI), and Work Productivity and Activity Impairment Questionnaire: Specific Health Problem (WPAI:SHP); all were exploratory outcomes
- During the titration period, participants began LXB treatment with individualized dosing adjustments to achieve their optimal dose. Once the optimal LXB dosage was achieved, participants remained on a stable dose through the end-of-treatment (EOT) period
 - Participants could be treated with a once- or twice-nightly LXB dosage based on the investigator's discretion in the idiopathic hypersomnia cohort (per the US prescribing label)[5]
- All BL assessments were administered again at EOT (on optimized LXB dosage); a safety follow-up visit occurred at the end of the study

Figure 2. Self-Reported Assessments and Rating Scales

Functional Outcome Identified as Bothersome by Patients

Mood (PHQ-9[8])

PHQ-9: Assessment of depressive symptoms; 9-item scale

- Scored using a 4-point Likert scale for each item (0 ["not at all"] to 3 ["nearly every day"])
- Total score range is 0–27 (higher scores indicate greater severity)
- Categorical scoring is as follows: 0–4, none or minimal; 5–9, mild; 10–14, moderate; 15–19, moderately severe; 20–27, severe

Cognition (BC-CCI[9])

BC-CCI: Assessment of cognitive complaints (problems with concentration, memory, trouble expressing thoughts, word finding, slow thinking, and difficulty solving problems[a]); 6-item measure

- Scored using a 4-point Likert scale for each item (0 ["not at all"] to 3 ["very much"])
- Total score range is 0–18 (higher scores indicate more cognitive impairment)
- Categorical scoring is as follows: 0–4, none or minimal; 5–8, mild; 9–14, moderate; 15–18, severe

Work Productivity and Activity Impairment (WPAI:SHP[b,10])

WPAI:SHP: Measures the effect of a specified health problem (ie, idiopathic hypersomnia) on work productivity and non-work activities; 6-item questionnaire

- Work time missed (absenteeism), impairment while working (presenteeism), overall work impairment (absenteeism + presenteeism), and impairment in regular daily activities other than work are each expressed as a percentage (0–100%)
- Higher percentages indicate greater reduction in productivity

[a]Three additional items that asked about how these symptoms impact work, relationships, and social/recreational activities were also included. [b]Items regarding work productivity were completed by employed participants only.
BC-CCI, British Columbia Cognitive Complaints Inventory; PHQ-9, Patient Health Questionnaire-9; WPAI:SHP, Work Productivity and Activity Impairment Questionnaire: Specific Health Problem.

- The safety analysis set included all participants who enrolled in the study and took their prescribed LXB regimen for ≥1 night after the BL period. The completer analysis set included all participants who enrolled in the study, took their prescribed LXB regimen for ≥1 night after the BL period, completed the SDP, and completed the visit 4 polysomnography EOT visit
- LS mean (SE) changes, 95% CIs, and *P* values are provided at EOT using analysis of covariance models adjusted for BL values

Results

Table 1. Demographics and Baseline Characteristics for Enrolled Participants With Idiopathic Hypersomnia[a]

Characteristic	Total (N=46)
Age (years), mean (SD)	38.1 (11.8)
Sex at birth, n (%)	
Male	9 (19.6)
Female	37 (80.4)
Race, n (%)	
White	39 (84.8)
Black or African American	3 (6.5)
American Indian or Alaska Native	0
Asian	2 (4.3)
Native Hawaiian or other Pacific Islander	1 (2.2)
Multiple[b]	1 (2.2)
Body mass index (kg/m²), mean (SD)	28.5 (6.4)
Oxybate type at study entry,[c] n (%)	
Naive[d]	37 (80.4)
Low-sodium oxybate	9 (19.6)
Sodium oxybate	0
Once-nightly sodium oxybate	0
Oxybate total nightly dosage at screening[e] (g)	
Mean (SD)	6.8 (2.2)
Median (min, max)	6.8 (3.8, 9.0)
Concomitant alerting agents, n (%)	19 (41.3)

[a]Safety analysis set, which includes all participants who enrolled in the study and took their prescribed LXB regimen for ≥1 night after the BL period. [b]Participant reported >1 race. [c]Screening period included a 2-week washout for current oxybate users. [d]No oxybate use within 2 weeks of entering the study. [e]For the participants who were taking an oxybate at screening.
BL, baseline; LXB, low-sodium oxybate; max, maximum; min, minimum; SD, standard deviation.

- Forty-six people with idiopathic hypersomnia enrolled and took LXB for ≥1 night; 40 completed the study
- Most enrolled participants were female (80.4%) and White (84.8%); the mean (SD) age was 38.1 (11.8) years

Table 2. Treatment-Emergent Adverse Events (Safety Analysis Set)

TEAEs occurring in ≥5% of participants, n (%)	Total (N=46)
Nausea	9 (19.6)
Dizziness	8 (17.4)
Headache	8 (17.4)
Vomiting	5 (10.9)
Middle insomnia	4 (8.7)
Anxiety	3 (6.5)
Decreased appetite	3 (6.5)
Enuresis	3 (6.5)
Somnolence	3 (6.5)

TEAE, treatment-emergent adverse event.

- One serious adverse event was reported in the idiopathic hypersomnia cohort (hypoxia [concurrent with influenza] that was of moderate severity, determined to be unrelated to study drug in the opinion of the investigator, and resolved)

Figure 3. PHQ-9 Total Scores and Severity Categories at BL and EOT (Completer Analysis Set)

A) Total Score

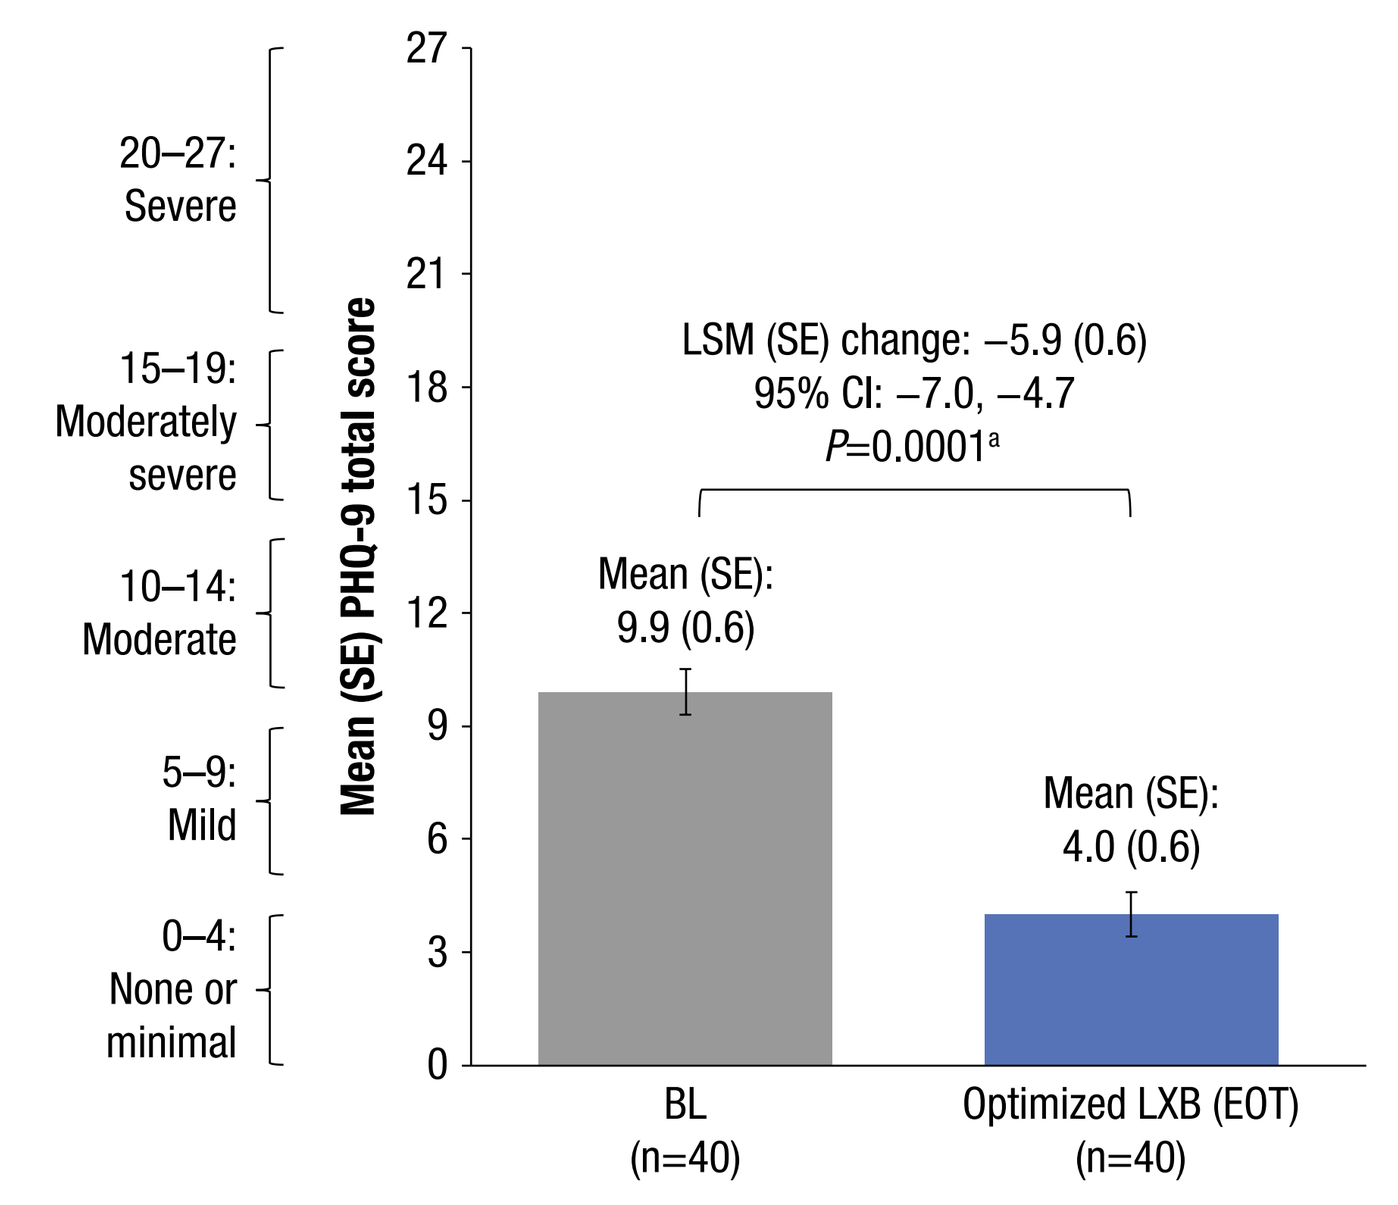

20–27: Severe
15–19: Moderately severe
10–14: Moderate
5–9: Mild
0–4: None or minimal
Mean (SE) PHQ-9 total score
LSM (SE) change: −5.9 (0.6)
95% CI: −7.0, −4.7
P=0.0001[a]
Mean (SE): 9.9 (0.6)
Mean (SE): 4.0 (0.6)
BL (n=40)
Optimized LXB (EOT) (n=40)

B) Depressive Symptom Severity Categories

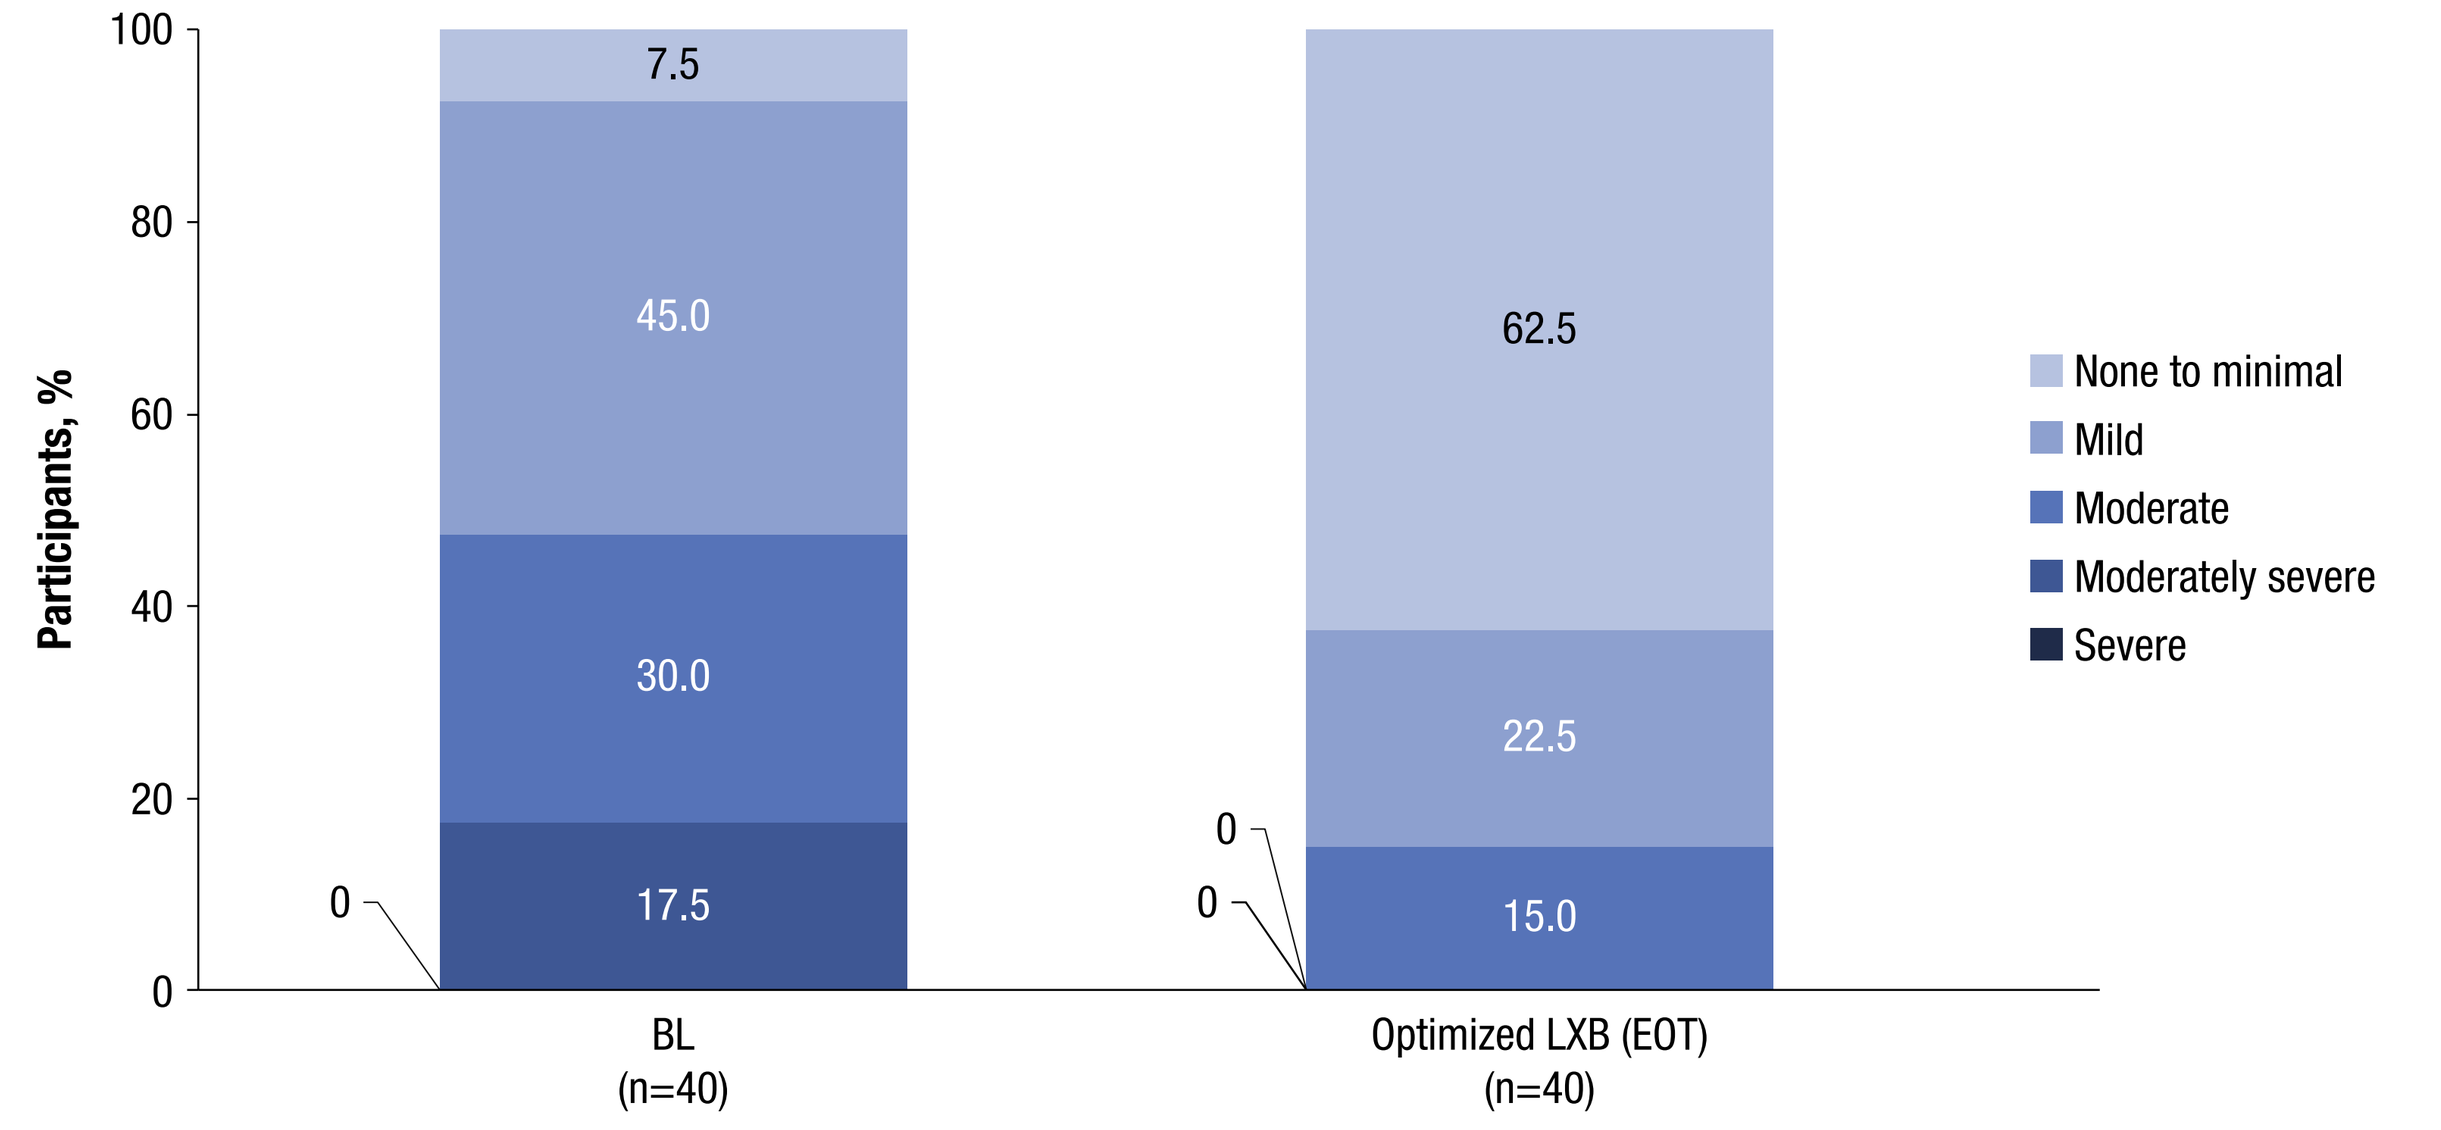

Participants, %
7.5
45.0
30.0
17.5
0
62.5
22.5
15.0
0
0
BL (n=40)
Optimized LXB (EOT) (n=40)
None to minimal
Mild
Moderate
Moderately severe
Severe

[a]*P* value is nominal.
BL, baseline; CI, confidence interval; EOT, end of treatment; LSM, least squares mean; LXB, low-sodium oxybate; PHQ-9, Patient Health Questionnaire-9; SE, standard error.

- PHQ-9 total scores decreased from BL to EOT, indicating improvement in depressive symptom severity
- The proportion of participants rating their depressive symptoms as none to minimal increased from 7.5% at BL to 62.5% at EOT
- The proportion of participants rating their depressive symptoms as moderately severe declined from 17.5% at BL to 0% at EOT
 - No participants rated their depressive symptoms as severe at BL or EOT

Figure 4. BC-CCI Scores and Categories at BL and EOT (Completer Analysis Set)

A) Total Score

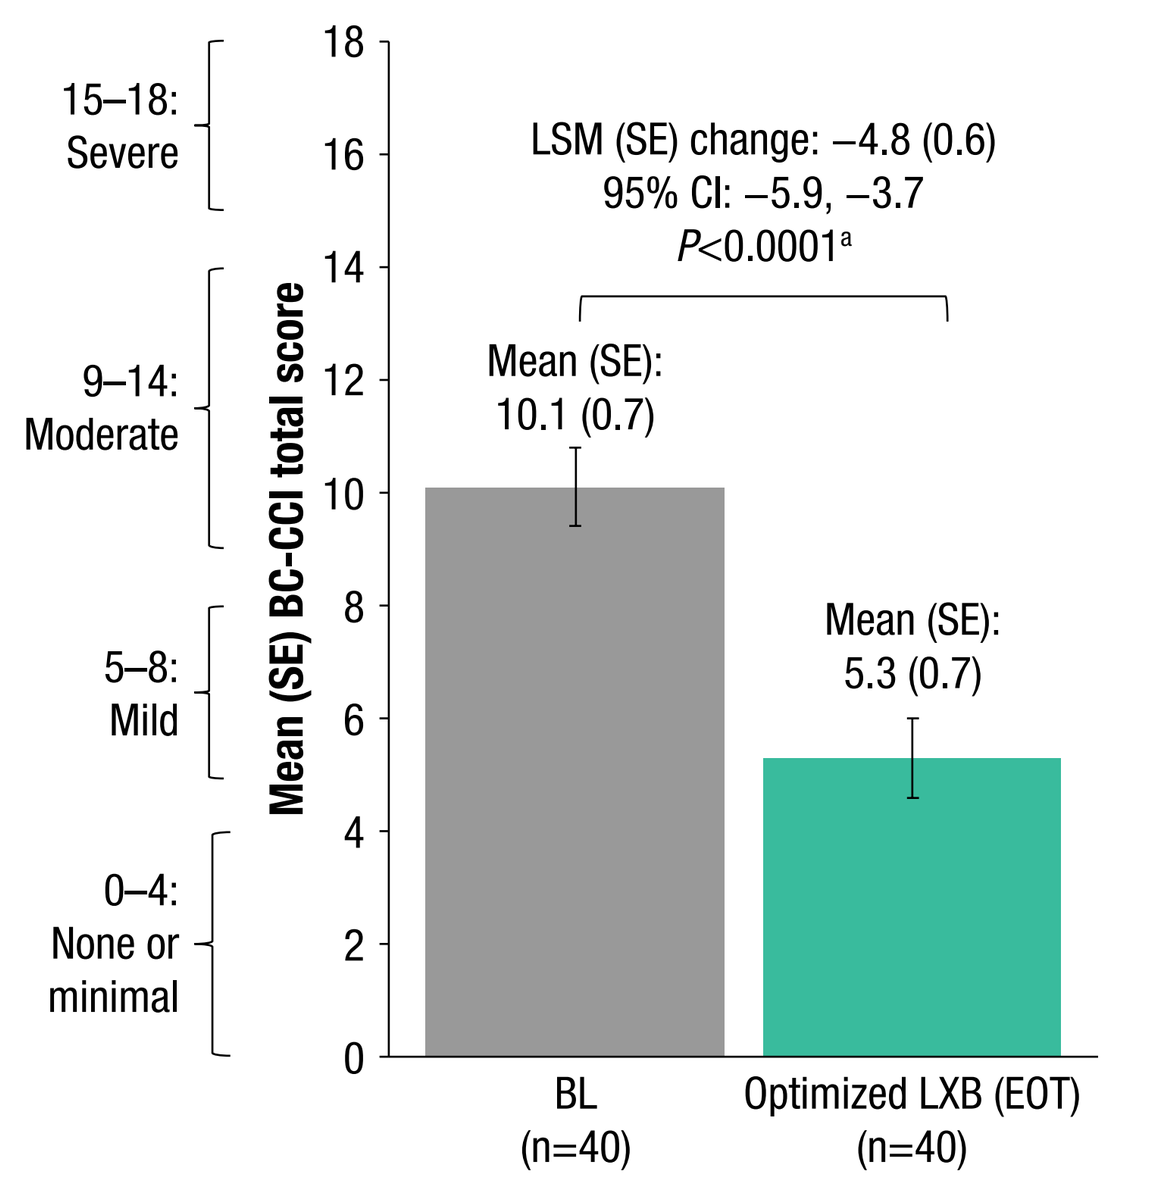

15–18: Severe
9–14: Moderate
5–8: Mild
0–4: None or minimal
Mean (SE) BC-CCI total score
LSM (SE) change: −4.8 (0.6)
95% CI: −5.9, −3.7
P<0.0001[a]
Mean (SE): 10.1 (0.7)
Mean (SE): 5.3 (0.7)
BL (n=40)
Optimized LXB (EOT) (n=40)

B) Cognitive Complaint Severity Categories

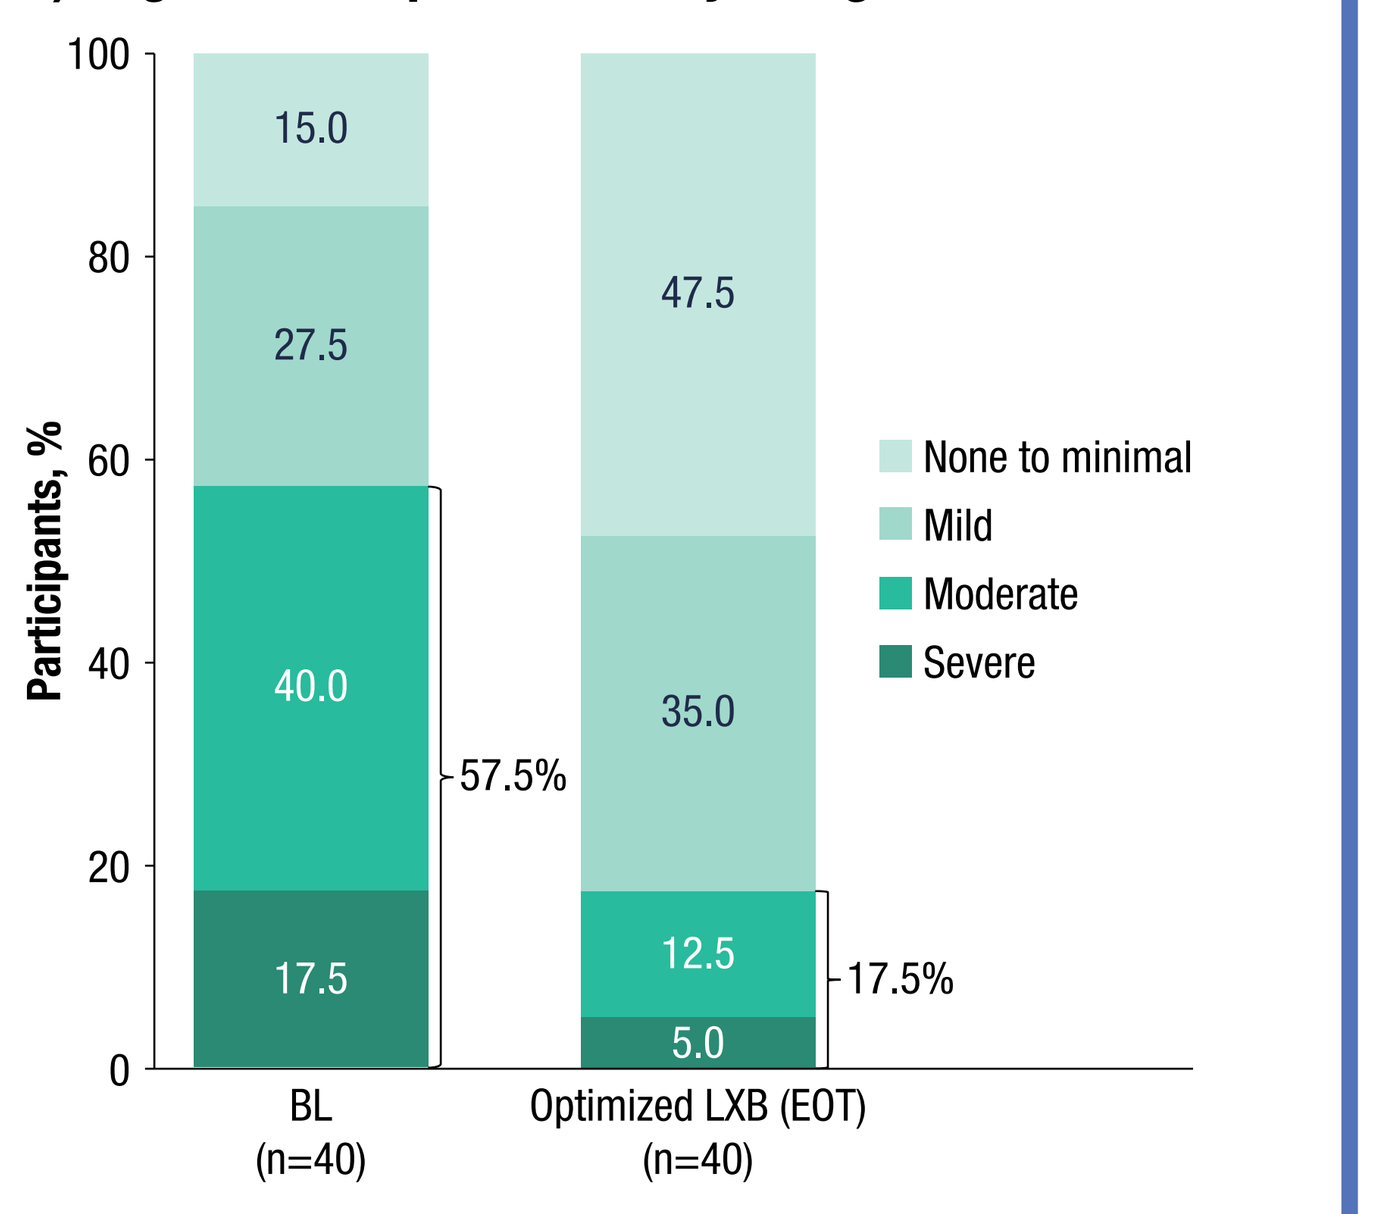

Participants, %
15.0
27.5
40.0
17.5
57.5%
47.5
35.0
12.5
5.0
17.5%
BL (n=40)
Optimized LXB (EOT) (n=40)
None to minimal
Mild
Moderate
Severe

C) Functional Impacts of Cognitive Complaints[b]

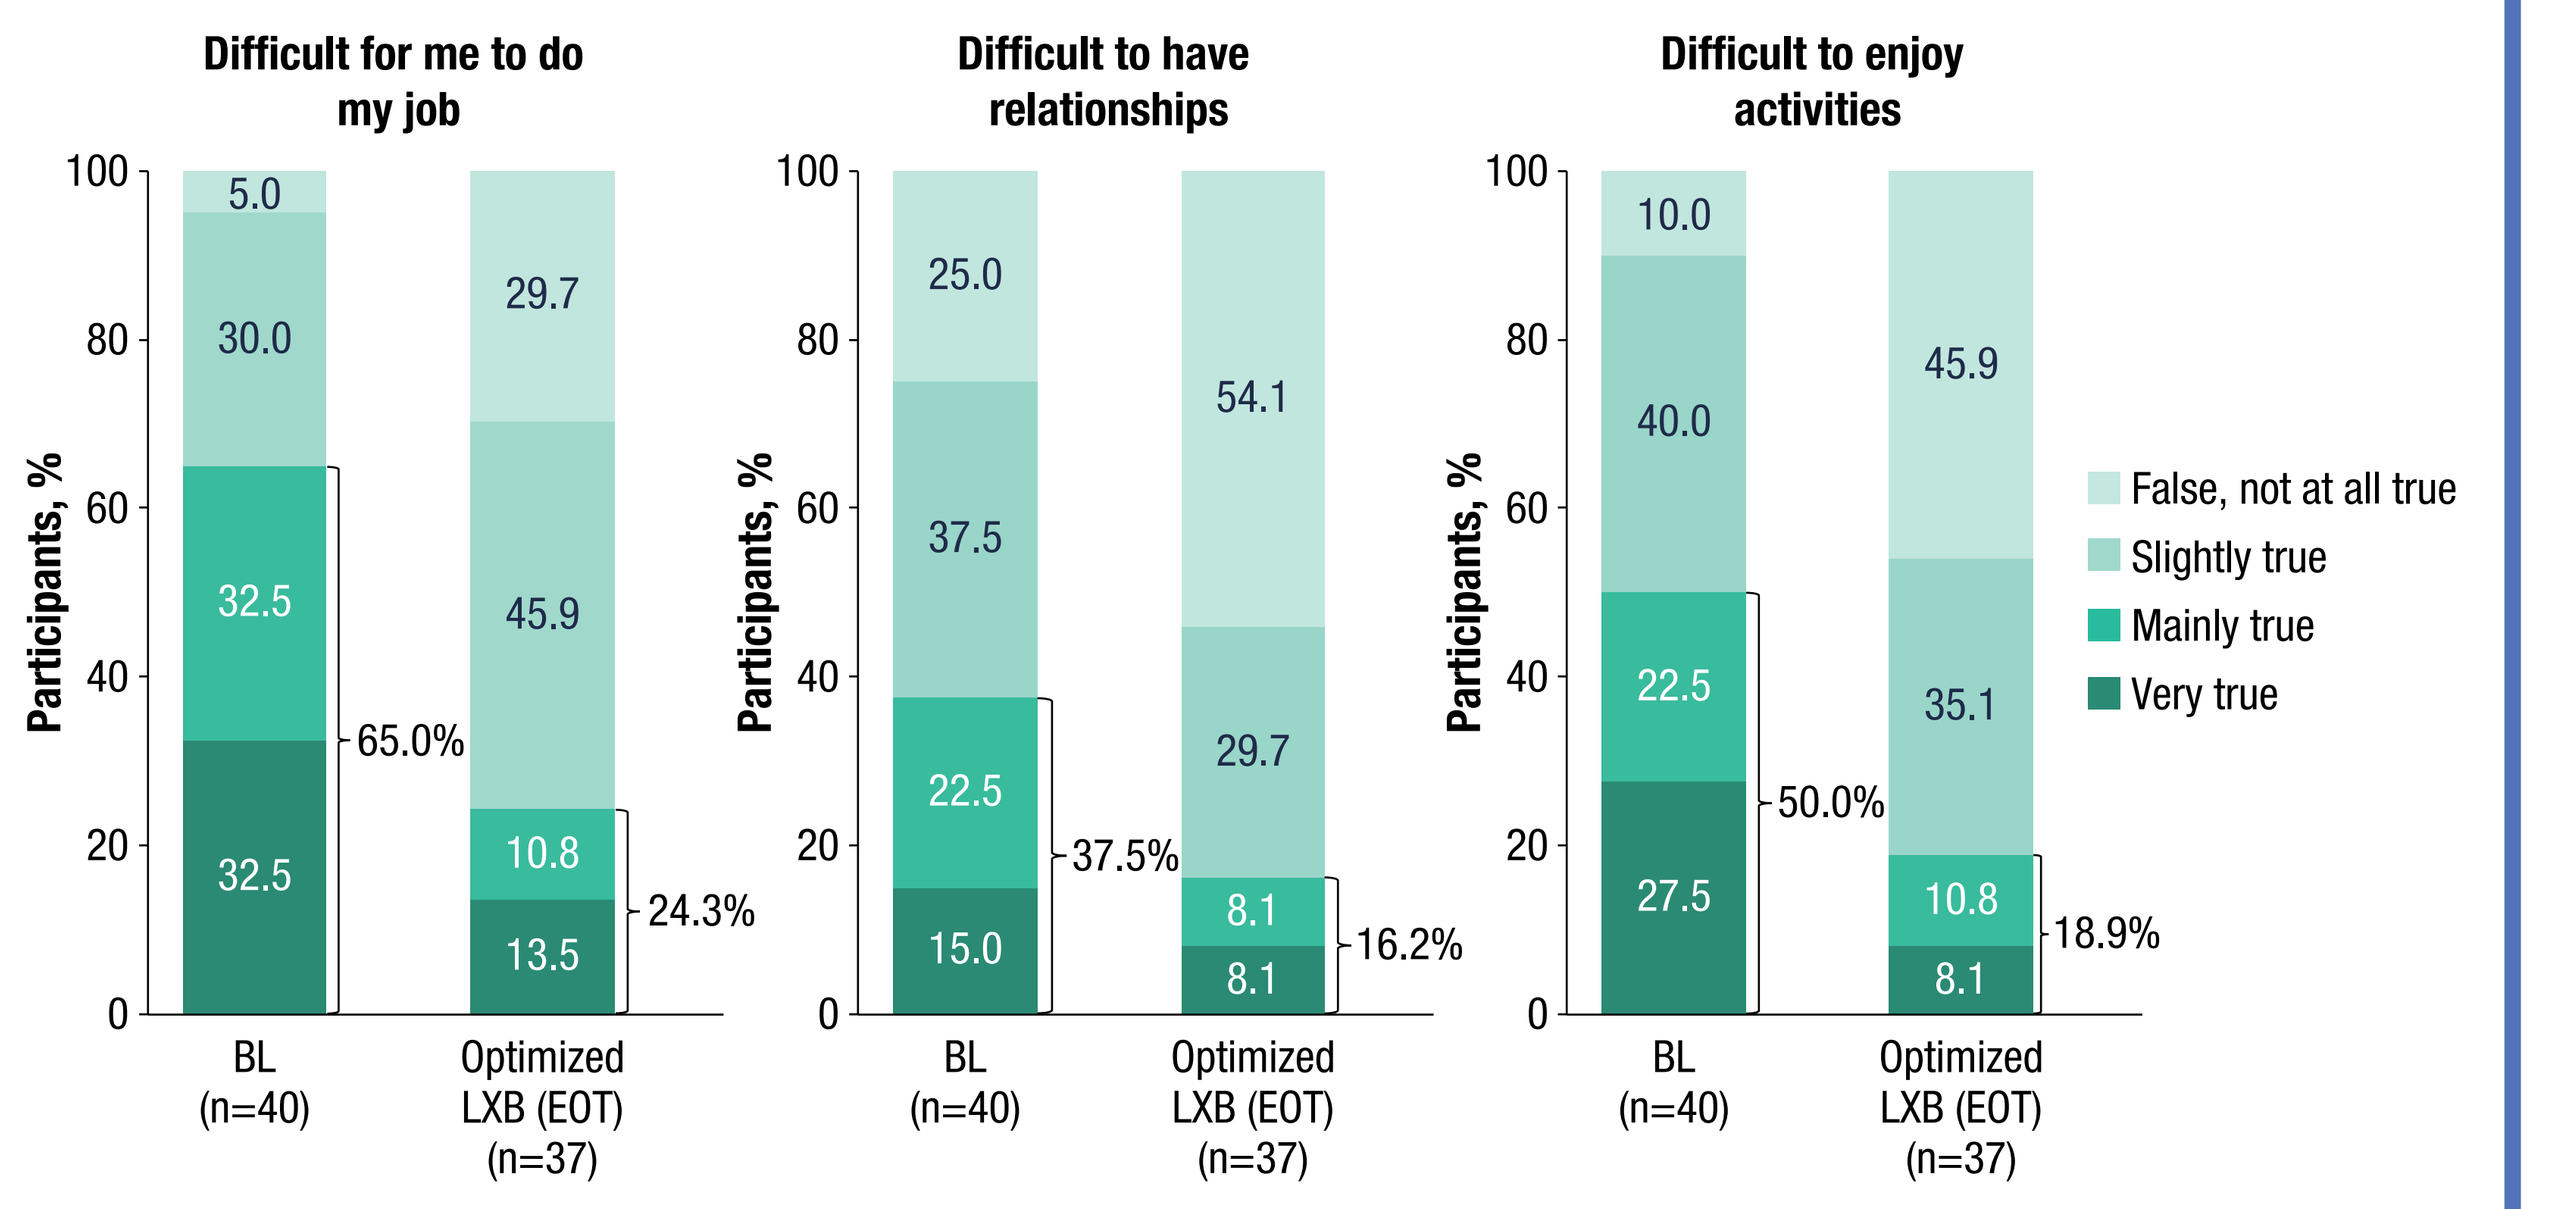

Difficult for me to do my job
5.0
30.0
32.5
32.5
65.0%
29.7
45.9
10.8
13.5
24.3%
Difficult to have relationships
25.0
37.5
22.5
15.0
37.5%
54.1
29.7
8.1
8.1
16.2%
Difficult to enjoy activities
10.0
40.0
22.5
27.5
50.0%
45.9
35.1
10.8
8.1
18.9%
Participants, %
BL (n=40)
Optimized LXB (EOT) (n=37)
False, not at all true
Slightly true
Mainly true
Very true

[a]*P* value is nominal. [b]BC-CCI Supplemental Items.
BC-CCI, British Columbia Cognitive Complaints Inventory; BL, baseline; CI, confidence interval; EOT, end of treatment; LSM, least squares mean; LXB, low-sodium oxybate; SE, standard error.

- BC-CCI total scores decreased from BL to EOT, indicating improvement in overall cognitive function
- The proportion of participants reporting moderate or severe cognitive complaints declined from 57.5% at BL to 17.5% at EOT
- There were also decreases from BL to EOT in the proportion of participants rating the following as "mainly true" or "very true": cognitive symptoms made it difficult to work (from 65.0% to 24.3%), have relationships (from 37.5% to 16.2%), or enjoy activities (from 50.0% to 18.9%)

Figure 5. WPAI:SHP Ratings of Work Productivity and Activity Impairment Due to Idiopathic Hypersomnia at BL and EOT (Completer Analysis Set[a])

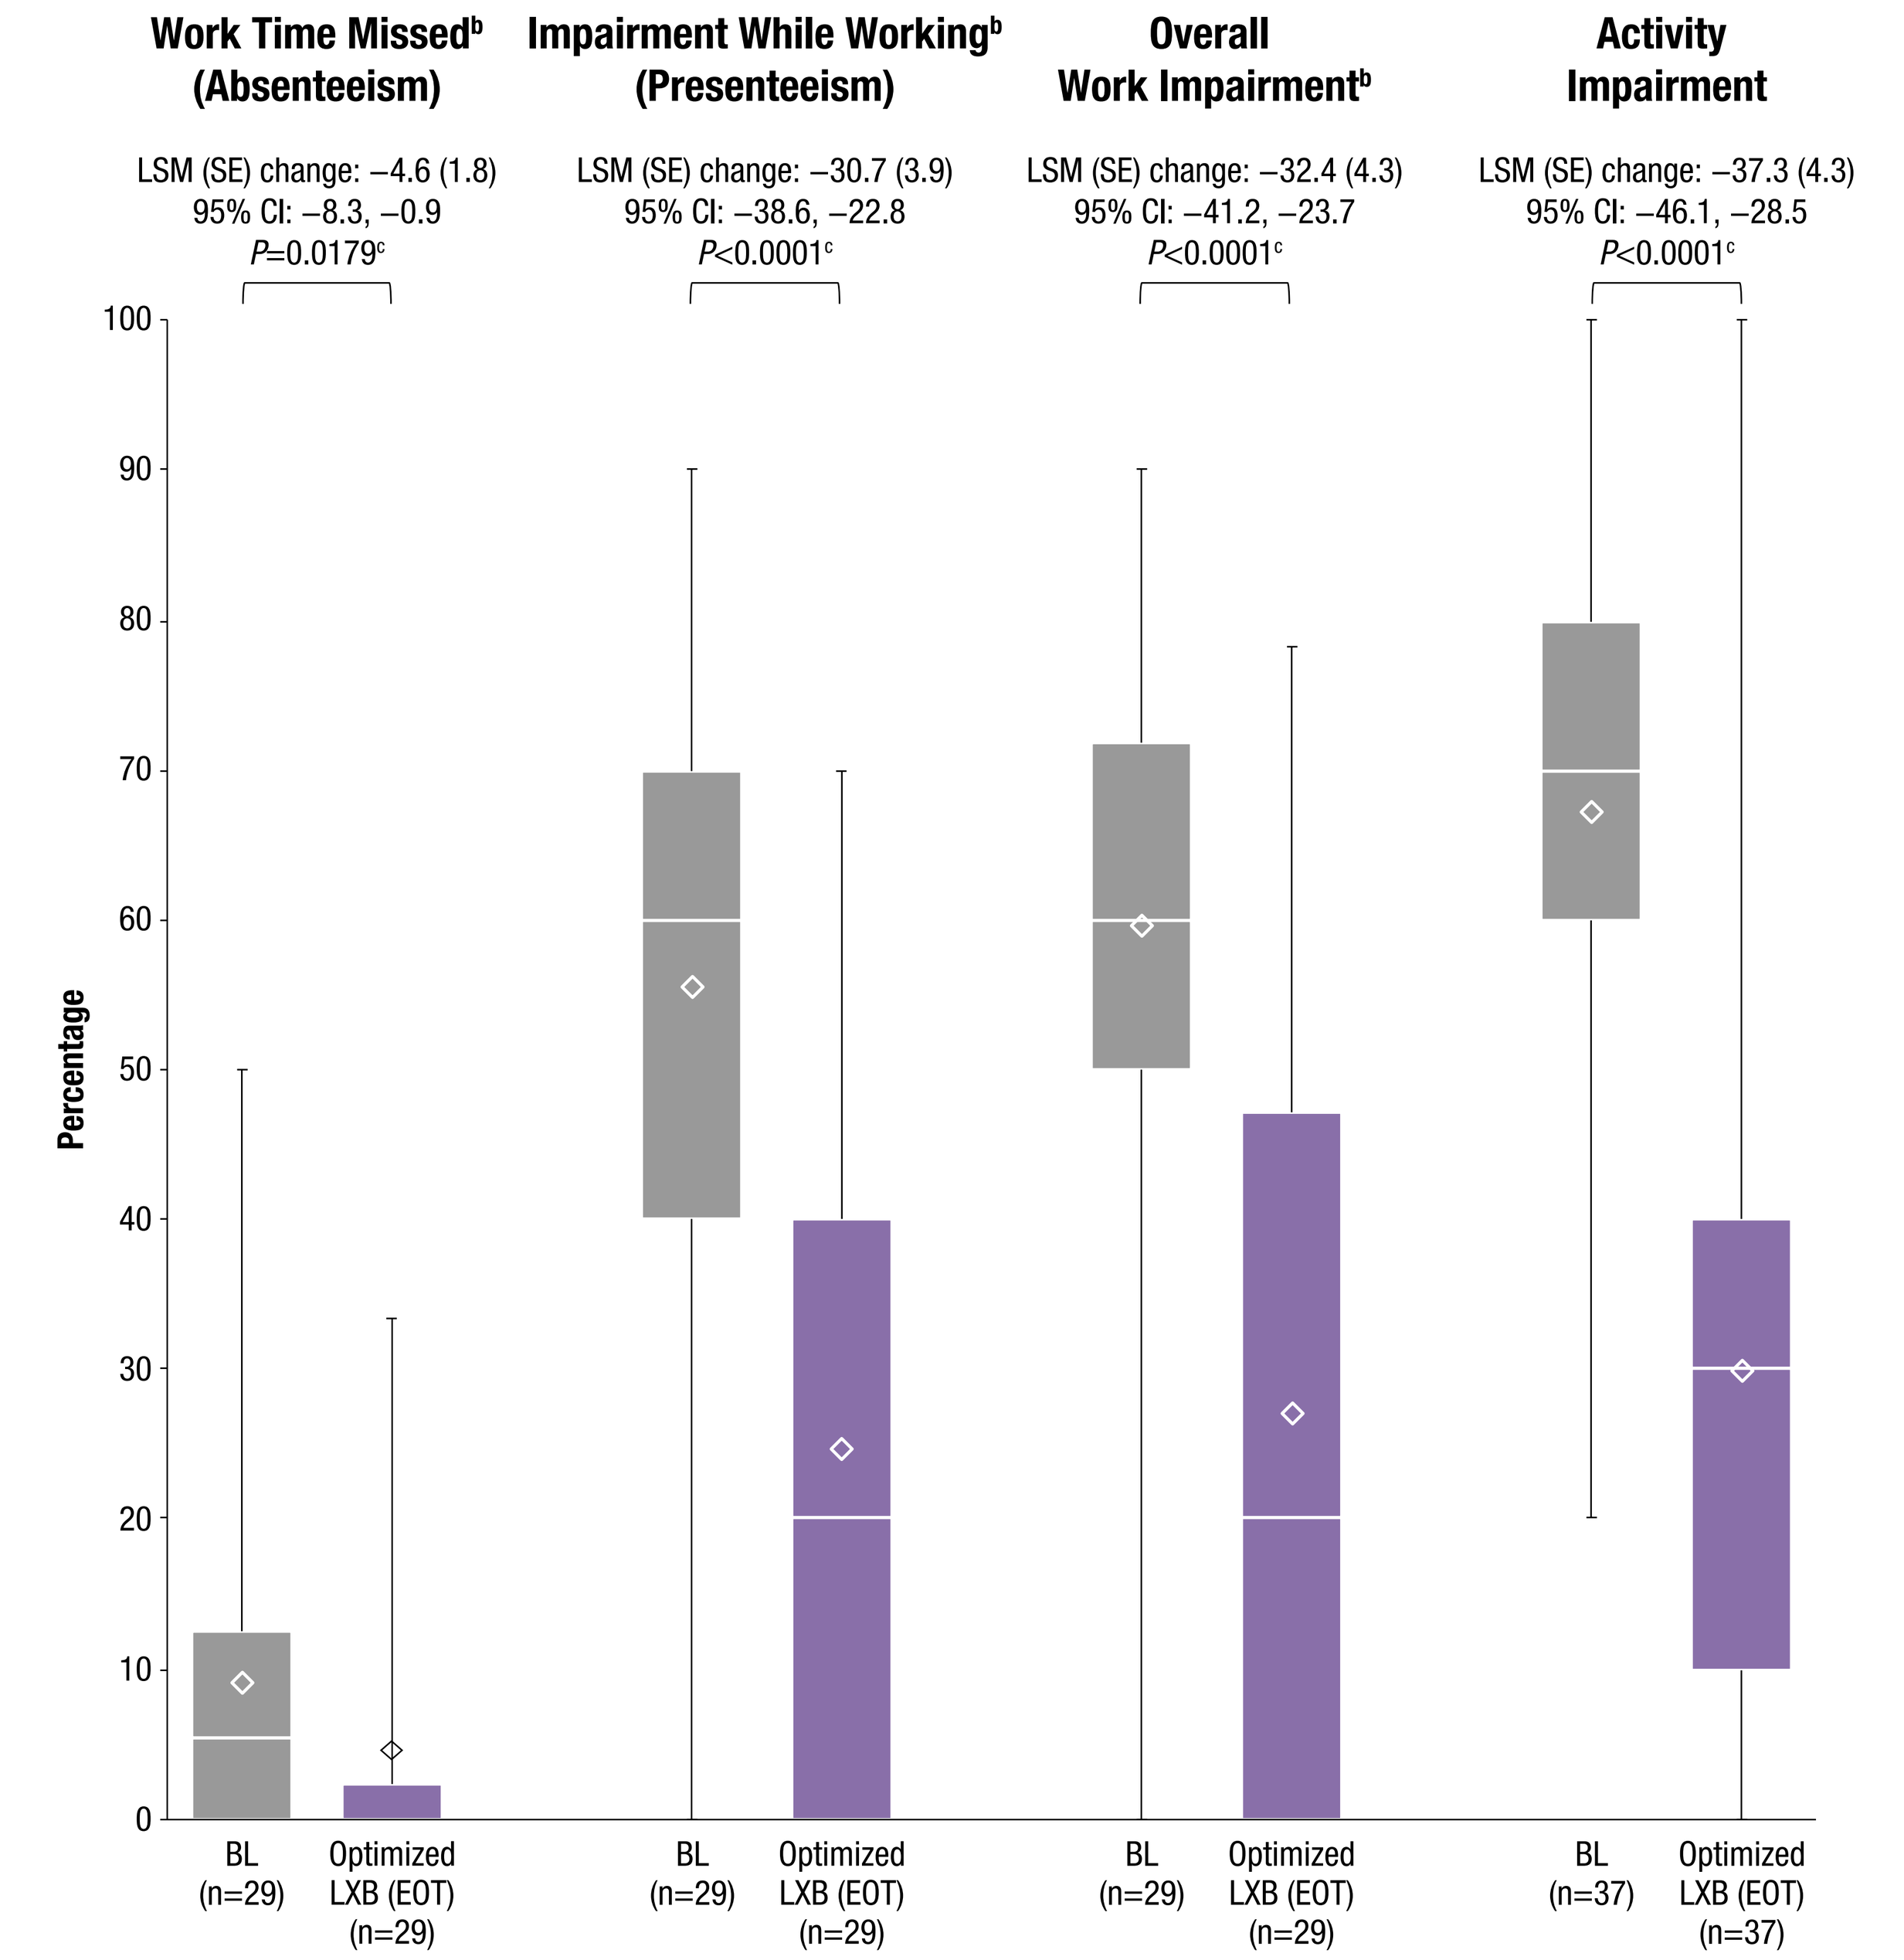

Work Time Missed[b] (Absenteeism)
LSM (SE) change: −4.6 (1.8)
95% CI: −8.3, −0.9
P=0.0179[c]
Impairment While Working[b] (Presenteeism)
LSM (SE) change: −30.7 (3.9)
95% CI: −38.6, −22.8
P<0.0001[c]
Overall Work Impairment[b]
LSM (SE) change: −32.4 (4.3)
95% CI: −41.2, −23.7
P<0.0001[c]
Activity Impairment
LSM (SE) change: −37.3 (4.3)
95% CI: −46.1, −28.5
P<0.0001[c]
Percentage
BL (n=29)
Optimized LXB (EOT) (n=29)
BL (n=37)
Optimized LXB (EOT) (n=37)

[a]Including participants completing both BL and EOT assessments. [b]Items pertaining to work impairment were completed by employed participants only. The bottom and top edges of the box indicate the first and third quartiles, the line inside the box is the median, and the diamond marker inside the box is the mean. The whiskers extending from the box indicate the minimum and maximum. [c]*P* value is nominal.
BL, baseline; CI, confidence interval; EOT, end of treatment; LSM, least squares mean; LXB, low-sodium oxybate; SE, standard error; WPAI:SHP, Work Productivity and Activity Impairment Questionnaire: Specific Health Problem.

- Absenteeism (mean [SE] % work time missed) declined from BL (9.1 [2.3]) to EOT (4.5 [1.8])
- Likewise, study participants had reductions in presenteeism (% impairment while working) and overall work impairment (absenteeism + presenteeism) due to idiopathic hypersomnia, as well as reduced activity impairment (overall daily activity other than work)

Conclusions

- Participants with idiopathic hypersomnia taking open-label LXB showed substantial[11] improvements in symptom impacts that were previously identified as highly bothersome through qualitative patient interviews,[4] including mood disturbances (ie, depressive symptoms), cognitive complaints, and personal/professional difficulties (ie, work productivity and activity impairments)
- DUET is the first study to assess changes in depressive symptoms or cognitive complaints in people with idiopathic hypersomnia following treatment with LXB
- Improvement in WPAI:SHP following LXB treatment is consistent with findings of a previous study of LXB in idiopathic hypersomnia[12]
- Limitations of this study include the open-label and single-arm design, which limits the ability to causally attribute findings to LXB
- Treatment-emergent adverse events were consistent with the known safety profile of LXB[5]

References: 1. American Academy of Sleep Medicine. *International Classification of Sleep Disorders – Third Edition, Text Revision.* Darien, IL: American Academy of Sleep Medicine; 2023. **2.** Arnulf I, et al. *Sleep Med Rev.* 2023;69:101766. **3.** Vernet C, et al. *J Sleep Res.* 2010;19(4):525-534. **4.** Casstevens C, et al. Presented at: Annual Psych Congress; October 29-November 2, 2024; Boston, MA. **5.** Xywav® (calcium, magnesium, potassium, and sodium oxybates) oral solution, CIII [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **6.** American Academy of Sleep Medicine. *International Classification of Sleep Disorders – Third Edition.* Darien, IL: American Academy of Sleep Medicine; 2014. **7.** Johns MW. *Sleep.* 1991;14(6):540-545. **8.** Kroenke K, et al. *J Gen Intern Med.* 2001;16(9):606-613. **9.** Iverson GL, Lam RW. *Ann Clin Psych.* 2013;25(2):135-140. **10.** Reilly MC, et al. *PharmacoEconomics.* 1993;4(5):353-365. **11.** Löwe B, et al. *Med Care.* 2004;42(12):1194-1201. **12.** Dauvilliers Y, et al. *Lancet Neurol.* 2022;21(1):53-65.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees Karyn Liu, PhD, and Stephanie Phan, PharmD, provided medical writing support and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: DT Plante is a consultant and advisory board member for Jazz Pharmaceuticals; an advisory board member for Apnimed; a consultant for Aditum Bio, LLC, and Teva Pharmaceuticals (Australia); and has served as a consultant/advisory board member for Alkermes, Centessa, Harmony Biosciences, and Takeda. **LD Schneider** is a compensated member of advisory boards and speakers bureaus for Jazz Pharmaceuticals, Eisai, and Harmony Biosciences. **DA Nichols, J Dai, MT Kirby, M Whalen,** and **C Casstevens** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **TL Steininger** is a former full-time employee and current contract worker for Jazz Pharmaceuticals who has received shares of Jazz Pharmaceuticals, plc. **A Cairns** is a full-time contract employee and advisory board member for Jazz Pharmaceuticals. **CM Ruoff** has served as an advisory board member for Alkermes, Eisai, Jazz Pharmaceuticals, and Takeda, and has received grant funding from Jazz Pharmaceuticals.

Scan this code to access this poster online.
This code is not for promotional purposes.

Presented at Psych Congress 2025; September 17–21, 2025; San Diego, CA, USA