120

Effectiveness and Safety of Low-Sodium Oxybate in Participants With Narcolepsy With or Without Psychiatric Comorbidities: Results From the Phase 4 DUET Study

DUET STUDY 407

Chad M. Ruoff, MD[1]; David T. Plante, MD, PhD[2]; Deborah A. Nichols, MS[3]; Teresa L. Steininger, PhD[3]; Jing Dai, PhD[3]; M. Todd Kirby, PhD[4,5]; Alyssa Cairns, PhD[4]; Marisa Whalen, PharmD[4]; Logan D. Schneider, MD[6]

[1]Center for Sleep Medicine, Mayo Clinic, Scottsdale, AZ, USA; [2]Department of Psychiatry, University of Wisconsin–Madison, Madison, WI, USA; [3]Jazz Pharmaceuticals, Palo Alto, CA, USA; [4]Jazz Pharmaceuticals, Philadelphia, PA, USA; [5]Department of Psychiatry & Health Behavior, Medical College of Georgia at Augusta University, Augusta, GA, USA; [6]Stanford University Center for Sleep Sciences and Medicine, Stanford, CA, USA.

Introduction

- Low-sodium oxybate (LXB; Xywav®) is approved by the US Food and Drug Administration for the treatment of cataplexy or excessive daytime sleepiness in patients 7 years of age or older with narcolepsy and the treatment of idiopathic hypersomnia in adults[1-4]
- Psychiatric comorbidities are common in people with narcolepsy[5-10]
 - Evaluating the effectiveness and safety of LXB in people with narcolepsy with psychiatric comorbidities will help guide clinicians in treating patients with these comorbidities
- Jazz DUET (**D**evelop hypersomnia **U**nderstanding by **E**valuating low-sodium oxybate **T**reatment) was a phase 4, prospective, open-label study (NCT05875974) to assess effectiveness of LXB on sleep and daytime symptoms in participants with narcolepsy (type 1 [NT1] or 2 [NT2]) or idiopathic hypersomnia

Objective

- To evaluate the effectiveness and safety of LXB treatment in participants with narcolepsy with or without psychiatric comorbidities

Methods

Figure 1. Study Design

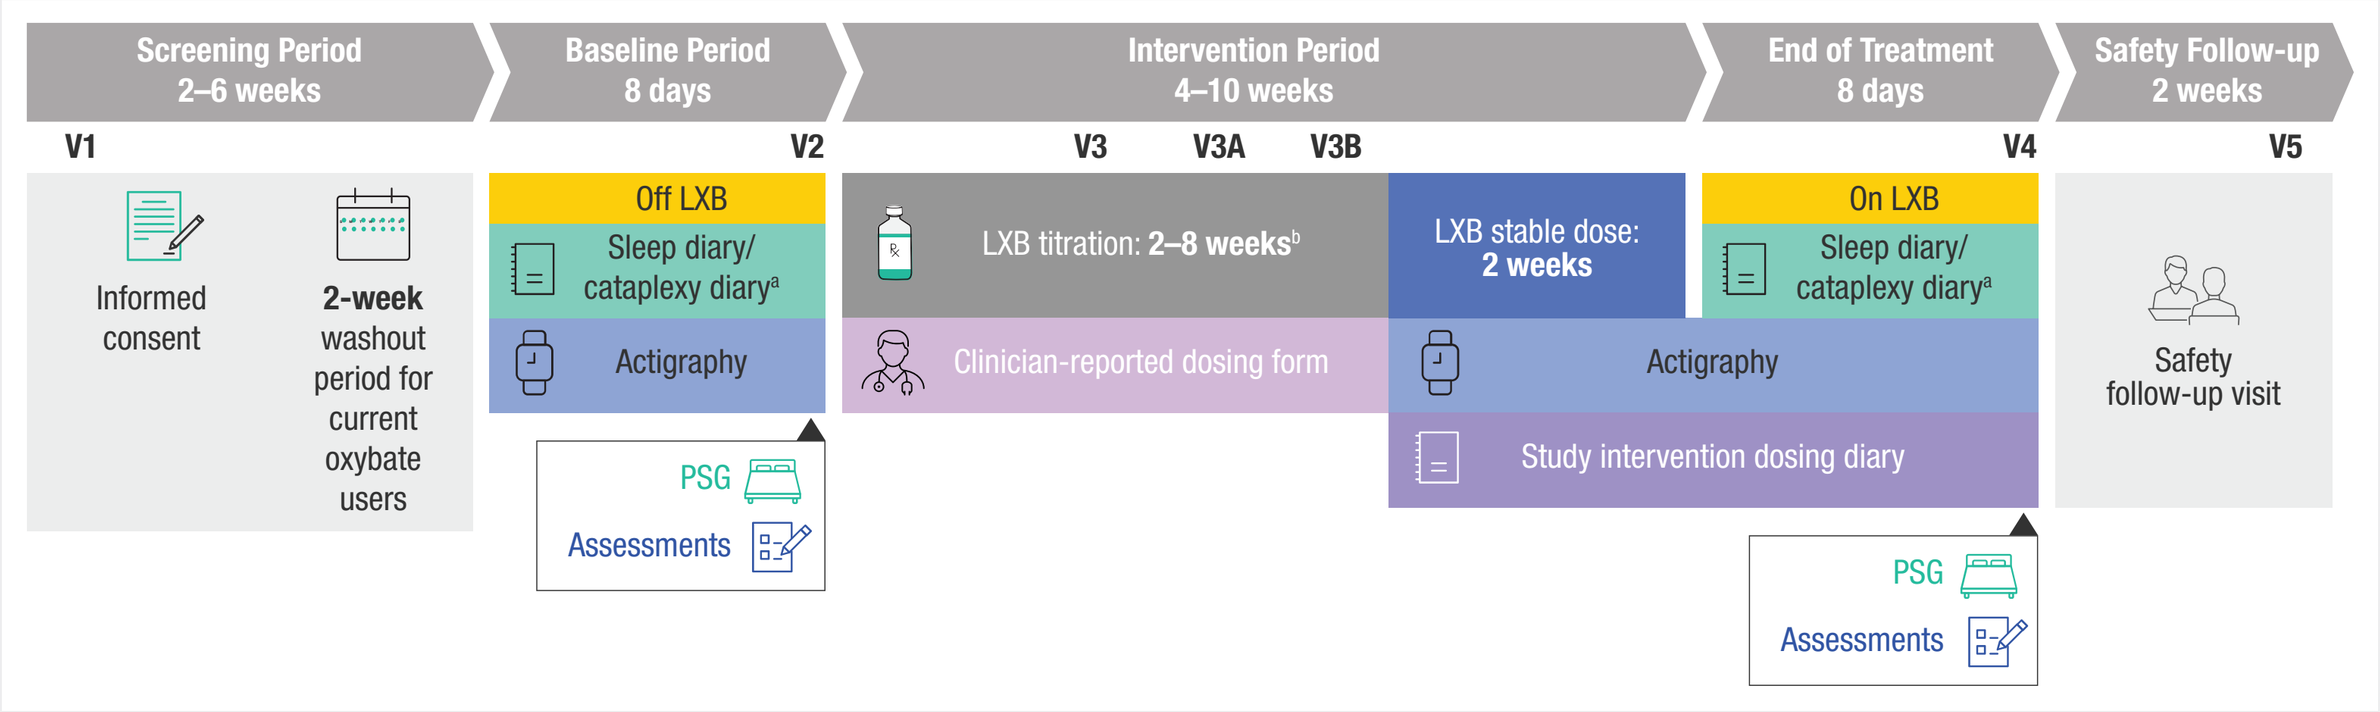

Screening Period 2–6 weeks
Baseline Period 8 days
Intervention Period 4–10 weeks
End of Treatment 8 days
Safety Follow-up 2 weeks
V1
V2
V3
V3A
V3B
V4
V5
Informed consent
2-week washout period for current oxybate users
Off LXB
Sleep diary/ cataplexy diary[a]
Actigraphy
PSG
Assessments
LXB titration: 2–8 weeks[b]
Clinician-reported dosing form
LXB stable dose: 2 weeks
Study intervention dosing diary
On LXB
Safety follow-up visit

Adapted from Nichols DA, et al. *Neurol Ther.* 2025;14:1705–727. http://creativecommons.org/licenses/by-nc/4.0/.
[a]Cataplexy diary in narcolepsy type 1 only. [b]Weekly titration visits were by teleconference. Visit 3 occurred on titration day 14. Titration could take between 2 and 8 weeks. Additional in-clinic visits were scheduled for day 35 (visit 3A) and day 56 (visit 3B), as needed. Investigator could optimize participant dosage and move to SDP at visit 3, 3A, or 3B, but not during intervening weekly teleconferences.
LXB, low-sodium oxybate; PSG, polysomnography; SDP, stable-dose period; V, visit.

- DUET included a screening period (with a washout period for oxybate users), a baseline (BL) period, a titration period (participants began LXB treatment with individualized dosing adjustments to achieve their optimal dose), a stable-dose period (SDP; at optimal LXB dose), an end-of-treatment (EOT) period, and a safety follow-up
 - Participants with narcolepsy could be treated with LXB twice nightly (per US prescribing label)[1]
- The primary outcome was change in Epworth Sleepiness Scale (ESS) score from BL to EOT
- Secondary endpoints reported here include the change from BL to EOT in the Patient Global Impression of Severity (PGI-S) and the Patient Global Impression of Change (PGI-C) assessed at EOT
- Exploratory endpoints included change from BL to EOT in the Narcolepsy Severity Scale (NSS) score (the NSS-2 version, excluding questions on cataplexy, was administered in participants with NT2[11]), the Patient Health Questionnaire-9 (PHQ-9, score range 0 to 27), and the Patient Health Questionnaire-2 (PHQ-2; scored from the first 2 items of the PHQ-9 [range 0 to 6], and used to assess mood/anhedonia unrelated to sleep[12])
- Least squares (LS) mean differences (95% confidence interval) between EOT and BL were estimated using an analysis of covariance model adjusted for the BL value
 - *P* values were not adjusted for multiple comparisons
- Safety endpoints included incidence and severity of treatment-emergent adverse events (TEAEs) and the Columbia-Suicide Severity Rating Scale (C-SSRS) administered at every in-clinic visit
- The safety analysis set includes all participants who enrolled in the study and took their prescribed LXB regimen for ≥1 night after the BL period; the completer analysis set includes all participants who enrolled in the study, took their prescribed LXB regimen for ≥1 night after the BL period, completed the SDP, and completed the polysomnography EOT visit

Figure 2. Inclusion/Exclusion Criteria

Key Inclusion Criteria
- 18–75 years of age with primary diagnosis of NT1 or NT2 (per ICSD-3 or DSM-5 criteria)
- ESS score >10[a]
- Participants were allowed to continue taking concomitant alerting agents (stimulants or wake-promoting agents), but had to have been taking the same dosage for ≥1 month before screening visit 1 with no plan to adjust dosage during the study

Key Exclusion Criteria
- Untreated/inadequately treated sleep-disordered breathing (AHI >10)[b]
- History/presence of:
 - Other untreated/inadequately treated sleep disorder
 - Unstable/clinically significant medical condition
 - Behavioral/psychiatric disorder (such as active suicidal ideation or a current or past major depressive episode within the past year)
 - Bipolar disorder, bipolar-related disorder, schizophrenia, schizophrenia spectrum disorders
 - Other psychotic disorders according to DSM-5 criteria, neurologic disorder, or surgical history that might affect the participant's safety or interfere with study conduct

[a]At screening visit 1 or an ESS score >10 after the washout period, if taking an oxybate medication. [b]Defined as apnea-hypopnea index >10, with hypopnea definition including a ≥4% desaturation as per Rule 1B of *The AASM Manual for the Scoring of Sleep and Associated Events.*[13]
AASM, American Academy of Sleep Medicine; AHI, apnea-hypopnea index; DSM-5, *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition*; ESS, Epworth Sleepiness Scale; ICSD-3, *The International Classification of Sleep Disorders – Third Edition*; NT1, narcolepsy type 1; NT2, narcolepsy type 2.

Results

Table 1. Concurrent Psychiatric Disorders[a,b]

System Organ Class, n (%)[c] Preferred Term, n (%)[c]	Total Narcolepsy Cohort (N=55)
Psychiatric disorders	**27 (49.1)**
Depression	12 (21.8)
Attention deficit hyperactivity disorder	9 (16.4)
Anxiety	7 (12.7)
Anxiety disorder	2 (3.6)
Post-traumatic stress disorder	2 (3.6)
Autism spectrum disorder	1 (1.8)
Generalized anxiety disorder	1 (1.8)
Insomnia	1 (1.8)
Major depression	1 (1.8)
Social anxiety disorder	1 (1.8)

[a]Safety analysis set. [b]Concurrent medical conditions/diseases are medical conditions/diseases that were ongoing as of the screening date, per medical history. [c]Participants reporting a condition/disease more than once within a system organ class/preferred term were counted only once for that system organ class/preferred term.

Table 2. Demographics and Baseline Characteristics[a]

Characteristic	With Psychiatric Comorbidities (n=27)	Without Psychiatric Comorbidities (n=28)	Total Narcolepsy Cohort (N=55)
Age, years			
Mean (SD)	34.5 (14.7)	32.4 (11.0)	33.4 (12.9)
Median (min, max)	31.0 (18, 75)	28.5 (20, 62)	29.0 (18, 75)
Sex at birth, n (%)			
Male	6 (22.2)	9 (32.1)	15 (27.3)
Female	21 (77.8)	19 (67.9)	40 (72.7)
Gender identity, n (%)			
Male (including transgender man)	6 (22.2)	9 (32.1)	15 (27.3)
Female (including transgender woman)	21 (77.8)	19 (67.9)	40 (72.7)
Race, n (%)			
White	25 (92.6)	19 (67.9)	44 (80.0)
Black or African American	0	7 (25.0)	7 (12.7)
Asian	0	2 (7.1)	2 (3.6)
Multiple[b]	1 (3.7)	0	0
Unknown	1 (3.7)	0	1 (1.8)
Ethnicity, n (%)			
Hispanic or Latino	3 (11.1)	0	3 (5.5)
Not Hispanic or Latino	24 (88.9)	28 (100)	52 (94.5)
Body mass index, kg/m²			0
Mean (SD)	28.6 (6.6)	30.27 (6.9)	29.45 (6.7)
Median (min, max)	25.8 (20, 42)	31.6 (21, 44)	27.5 (20, 44)
Oxybate type at study entry, n (%)			
Naive[c]	20 (74.1)	22 (78.6)	42 (76.4)
Low-sodium oxybate	1 (3.7)	5 (17.9)	6 (10.9)
Sodium oxybate	5 (18.5)	0	5 (9.1)
Once-nightly sodium oxybate	1 (3.7)	1 (3.6)	2 (3.6)
Concomitant alerting agents, n (%)	15 (57.7)	16 (55.2)	31 (56.4)

[a]Safety analysis set. [b]Participant reported >1 race. [c]No oxybate use within 2 weeks of entering the study.
Max, maximum; min, minimum; SD, standard deviation.

- Fifty-five narcolepsy participants enrolled, and 34 completed the study
 - Thirteen participants transferred to a separate >9 g narcolepsy cohort (after titrating to 9 g and being identified as participants who may benefit from continued titration to a dosage >9 g in the opinion of the investigator; results reported elsewhere), and 8 participants discontinued the study

Figure 3. ESS Scores Improved From BL to EOT in Participants With or Without Psychiatric Comorbidities[a]

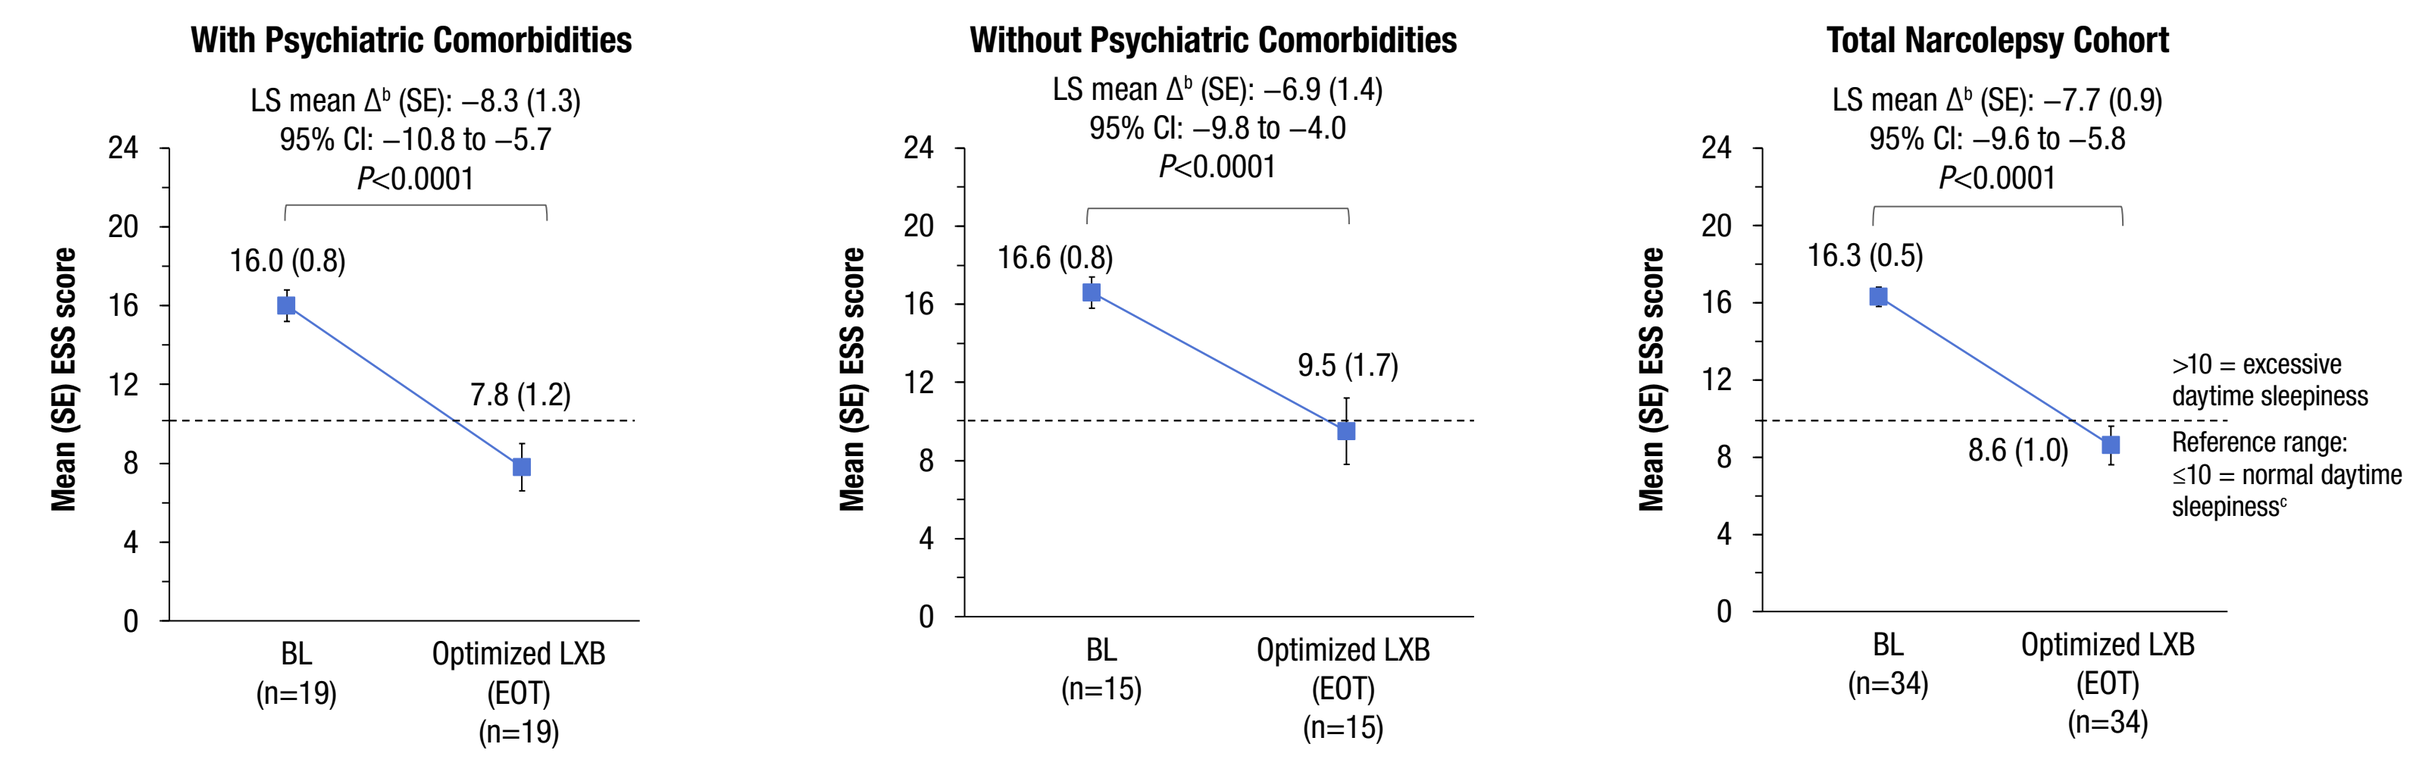

With Psychiatric Comorbidities
LS mean Δ[b] (SE): −8.3 (1.3)
95% CI: −10.8 to −5.7
P<0.0001
16.0 (0.8)
7.8 (1.2)
BL (n=19)
Optimized LXB (EOT) (n=19)
Without Psychiatric Comorbidities
LS mean Δ[b] (SE): −6.9 (1.4)
95% CI: −9.8 to −4.0
P<0.0001
16.6 (0.8)
9.5 (1.7)
BL (n=15)
Optimized LXB (EOT) (n=15)
Total Narcolepsy Cohort
LS mean Δ[b] (SE): −7.7 (0.9)
95% CI: −9.6 to −5.8
P<0.0001
16.3 (0.5)
8.6 (1.0)
BL (n=34)
Optimized LXB (EOT) (n=34)
>10 = excessive daytime sleepiness
Reference range: ≤10 = normal daytime sleepiness[c]
Mean (SE) ESS score

[a]Completer analysis set. [b]Difference between BL and EOT. [c]Established categories consider scores up to 10 normal.[14] The MCID for ESS was defined as 2 points according to an AASM systematic review and meta-analysis.[15]
AASM, American Academy of Sleep Medicine; BL, baseline; CI, confidence interval; EOT, end of treatment; ESS, Epworth Sleepiness Scale; LS, least squares; LXB, low sodium-oxybate; MCID, minimum clinically important difference; SE, standard error.

Figure 4. NSS (A) and NSS-2 (B) Scores Improved From BL to EOT in Participants With or Without Psychiatric Comorbidities[a]

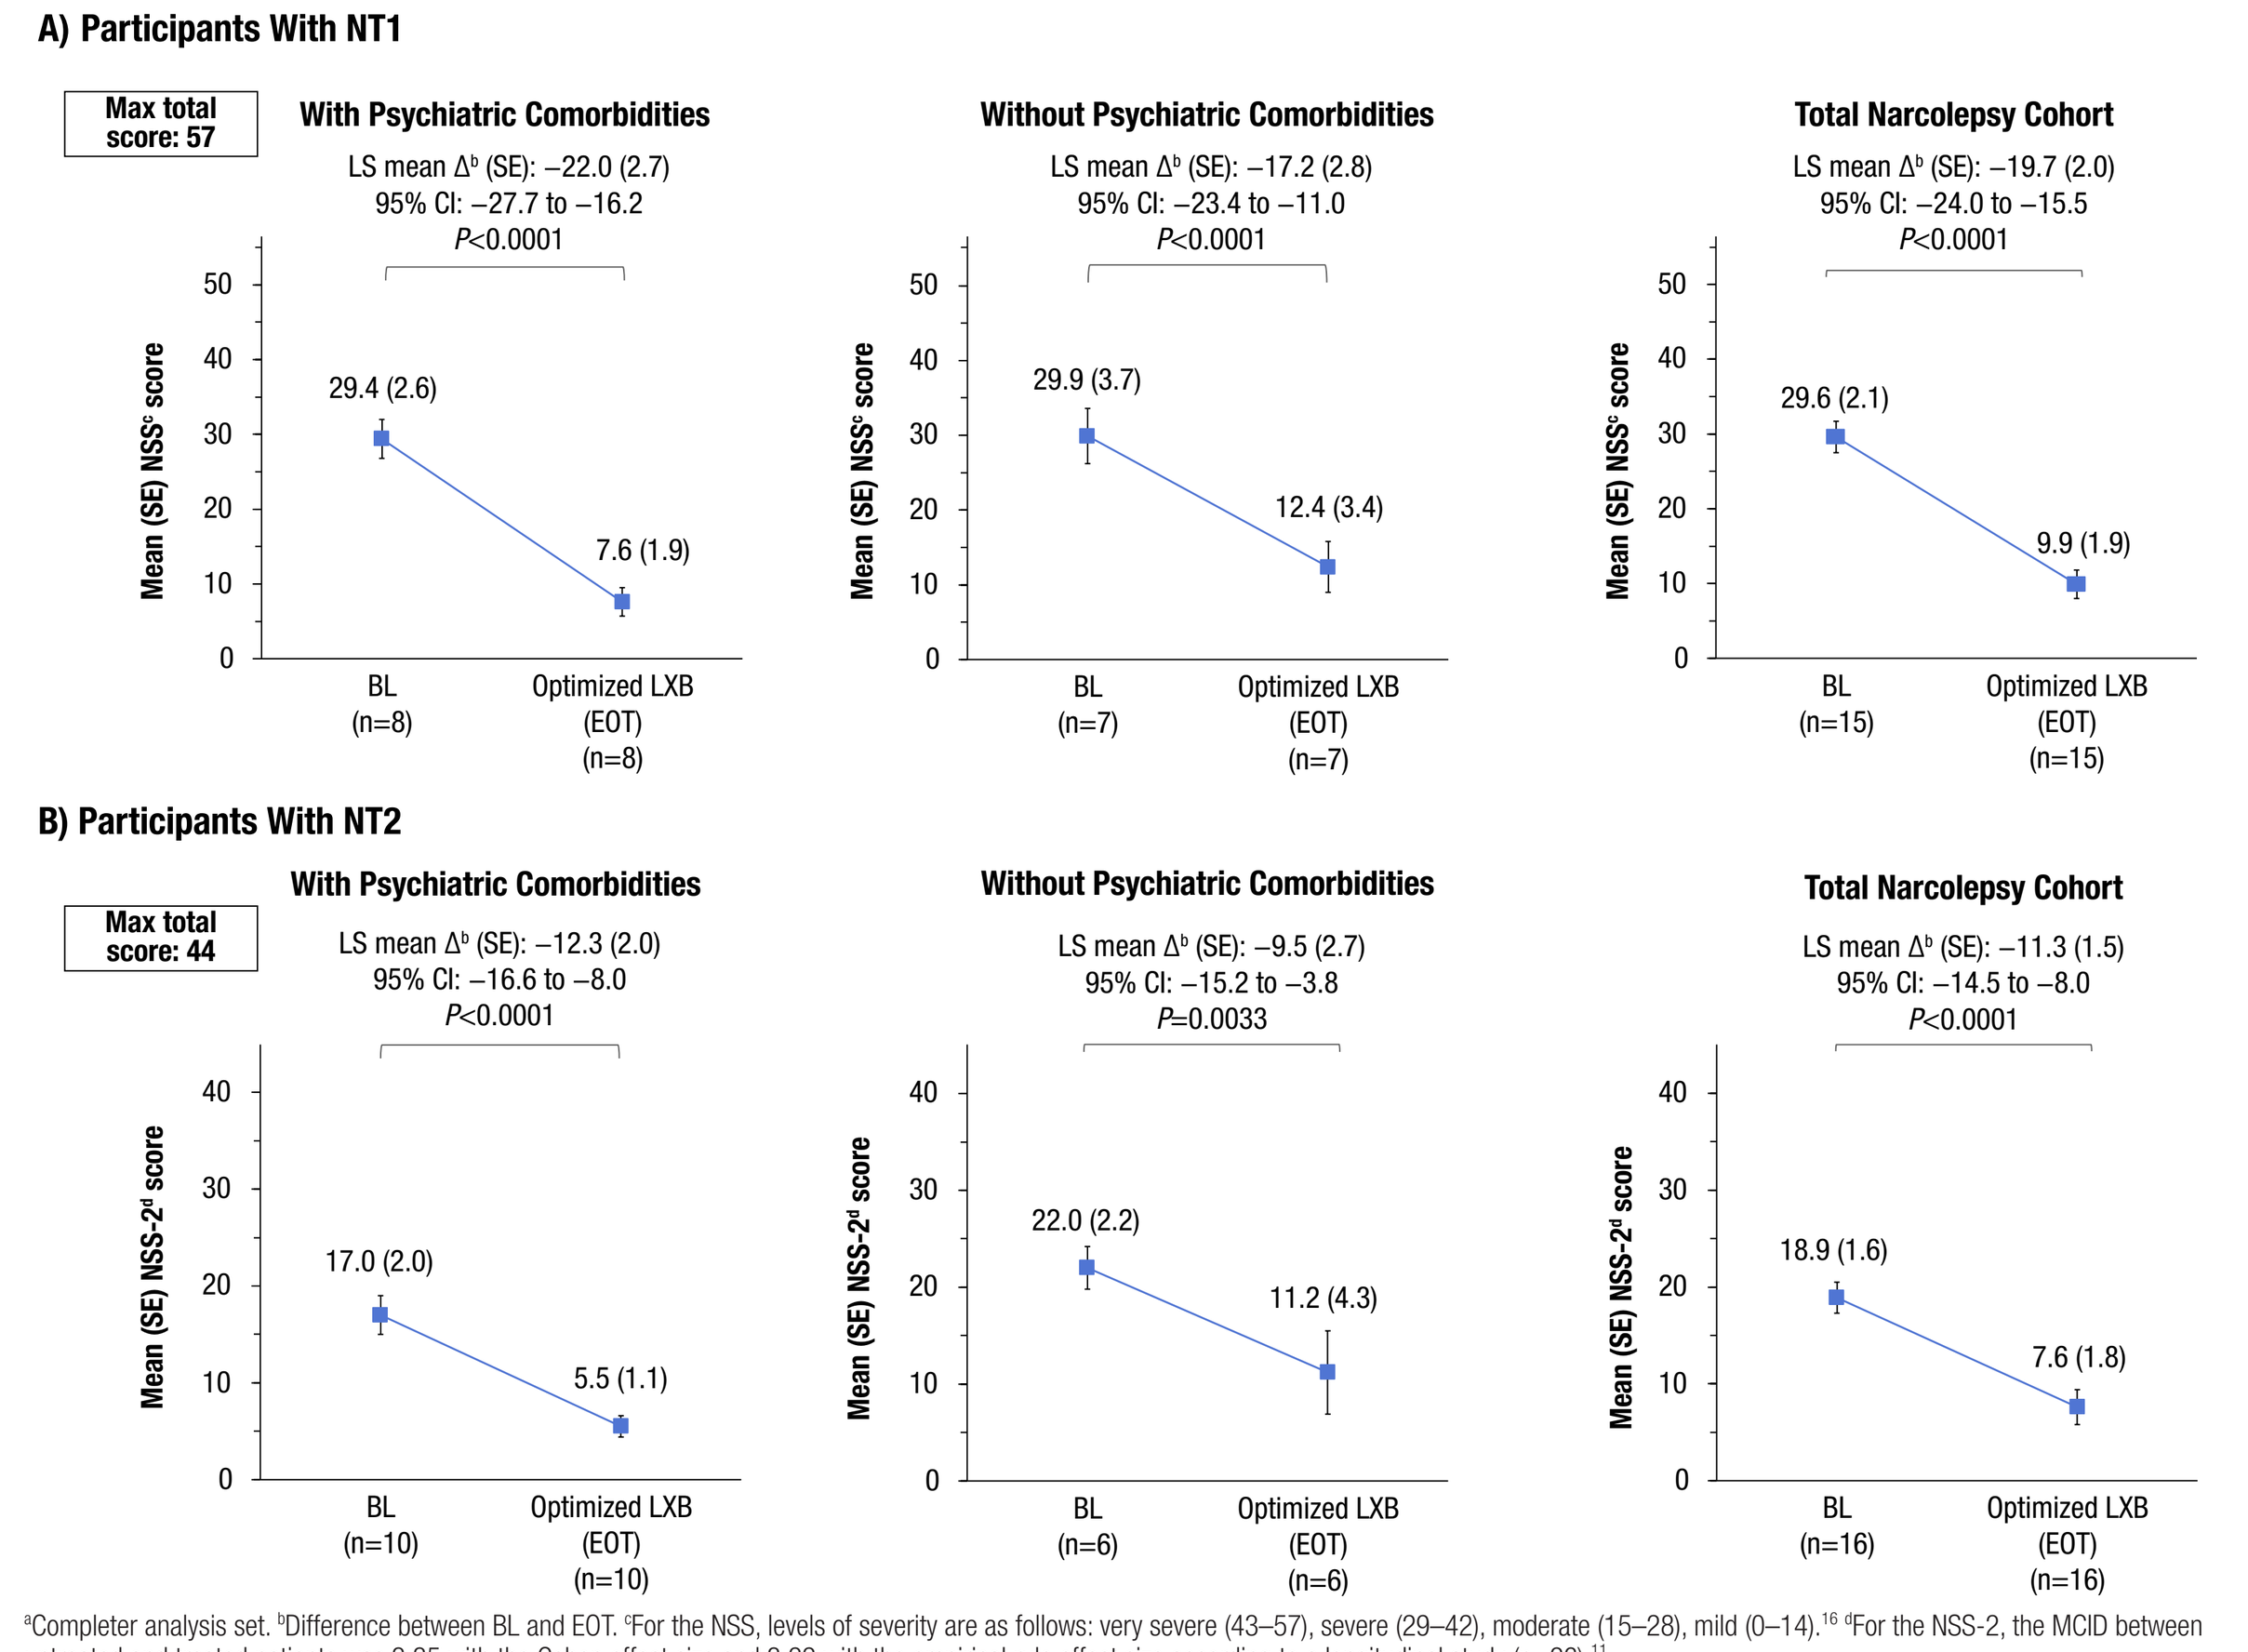

A) Participants With NT1
Max total score: 57
With Psychiatric Comorbidities
LS mean Δ[b] (SE): −22.0 (2.7)
95% CI: −27.7 to −16.2
P<0.0001
29.4 (2.6)
7.6 (1.9)
BL (n=8)
Optimized LXB (EOT) (n=8)
Without Psychiatric Comorbidities
LS mean Δ[b] (SE): −17.2 (2.8)
95% CI: −23.4 to −11.0
P<0.0001
29.9 (3.7)
12.4 (3.4)
BL (n=7)
Optimized LXB (EOT) (n=7)
Total Narcolepsy Cohort
LS mean Δ[b] (SE): −19.7 (2.0)
95% CI: −24.0 to −15.5
P<0.0001
29.6 (2.1)
9.9 (1.9)
BL (n=15)
Optimized LXB (EOT) (n=15)
Mean (SE) NSS[c] score
B) Participants With NT2
Max total score: 44
With Psychiatric Comorbidities
LS mean Δ[b] (SE): −12.3 (2.0)
95% CI: −16.6 to −8.0
P<0.0001
17.0 (2.0)
5.5 (1.1)
BL (n=10)
Optimized LXB (EOT) (n=10)
Without Psychiatric Comorbidities
LS mean Δ[b] (SE): −9.5 (2.7)
95% CI: −15.2 to −3.8
P=0.0033
22.0 (2.2)
11.2 (4.3)
BL (n=6)
Optimized LXB (EOT) (n=6)
Total Narcolepsy Cohort
LS mean Δ[b] (SE): −11.3 (1.5)
95% CI: −14.5 to −8.0
P<0.0001
18.9 (1.6)
7.6 (1.8)
BL (n=16)
Optimized LXB (EOT) (n=16)
Mean (SE) NSS-2[d] score

[a]Completer analysis set. [b]Difference between BL and EOT. [c]For the NSS, levels of severity are as follows: very severe (43–57), severe (29–42), moderate (15–28), mild (0–14).[16] [d]For the NSS-2, the MCID between untreated and treated patients was 3.35 with the Cohen effect size and 3.22 with the empirical rule effect size according to a longitudinal study (n=26).[11]
BL, baseline; CI, confidence interval; EOT, end of treatment; LS, least squares; LXB, low-sodium oxybate; NSS, narcolepsy severity score; NT1, narcolepsy type 1; NT2, narcolepsy type 2; SE, standard error.

Table 3. Mean Nightly LXB Dosage During Stable-Dose Period[a]

	With Psychiatric Comorbidities (n=20)[a]	Without Psychiatric Comorbidities (n=16)[a]	Total Narcolepsy Cohort (n=36)[a]
Total twice-nightly dosage, g/night			
Mean (SD)	7.1 (1.8)	7.0 (1.5)	7.0 (1.6)
First nightly LXB dose	3.7 (0.9)	3.6 (0.9)	3.7 (0.9)
Second nightly LXB dose	3.3 (1.0)	3.4 (0.7)	3.4 (0.9)

[a]Includes participants from the safety analysis set who reached the SDP.
LXB, low-sodium oxybate; SD, standard deviation; SDP, stable-dose period.

- All participants with narcolepsy took LXB twice nightly

Figure 5. PHQ-9 (A) and PHQ-2 (B) Scores Improved From BL to EOT in Participants With or Without Psychiatric Comorbidities[a,b]

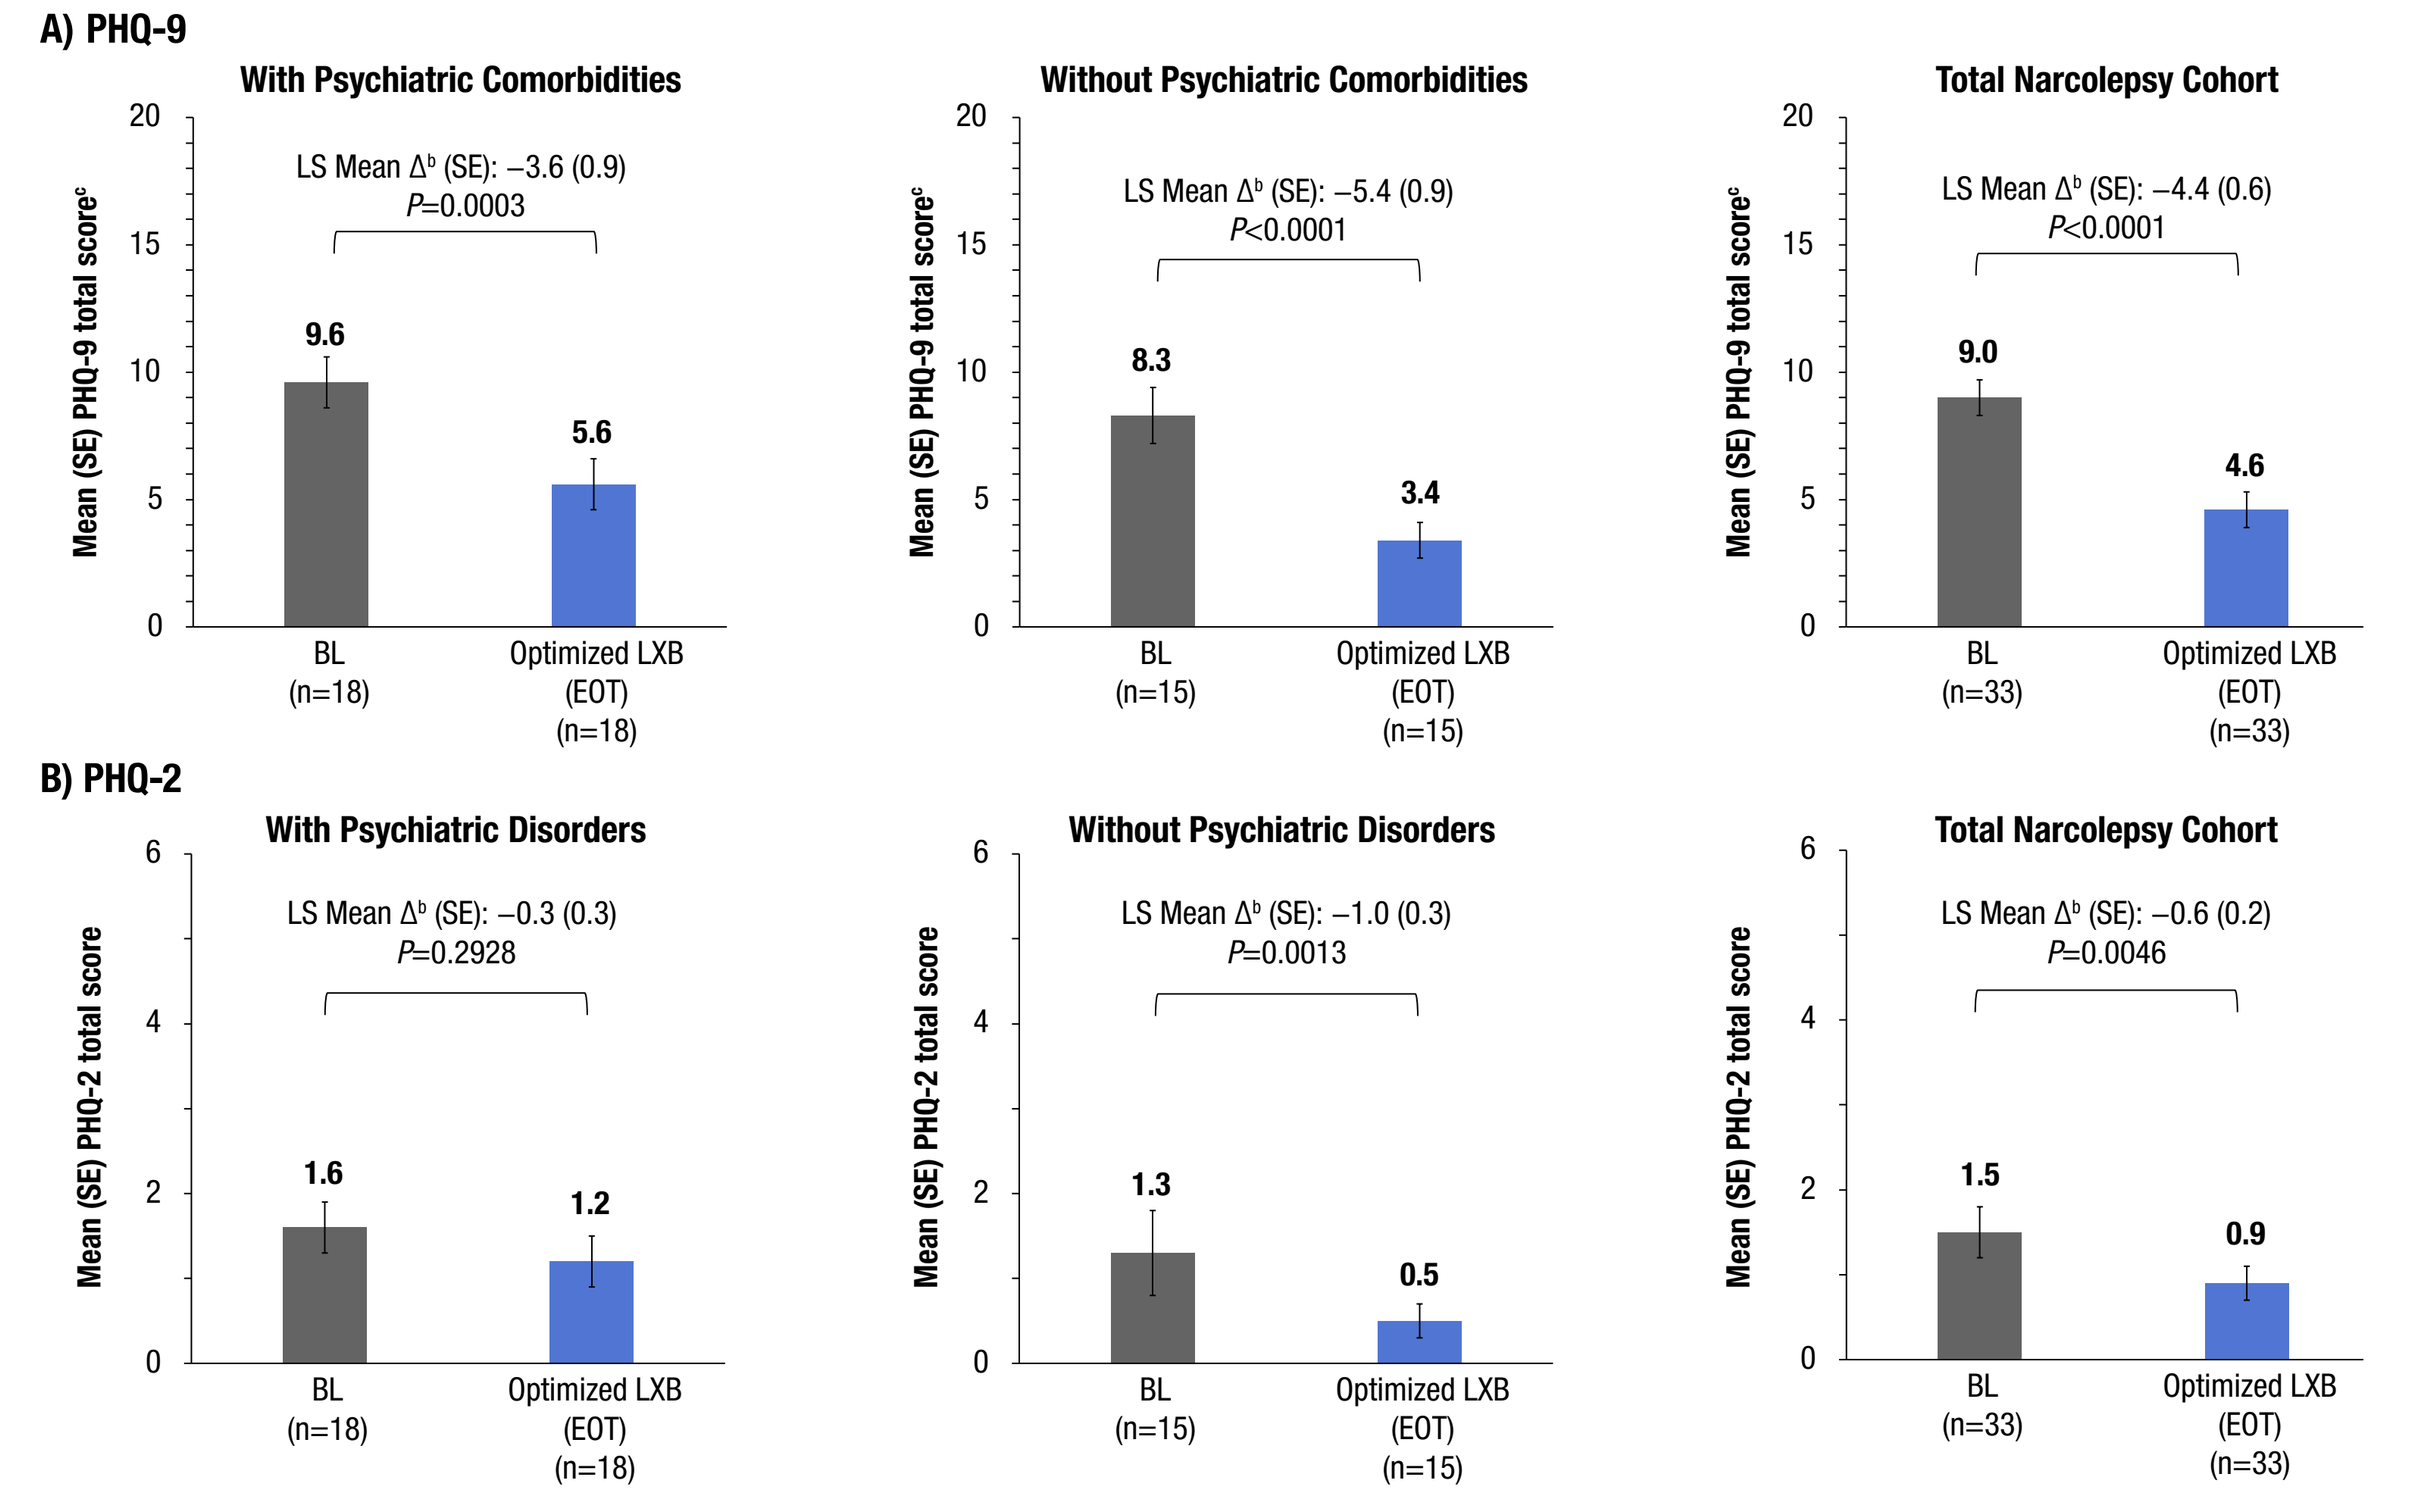

A) PHQ-9
With Psychiatric Comorbidities
LS Mean Δ[b] (SE): −3.6 (0.9)
P=0.0003
9.6
5.6
BL (n=18)
Optimized LXB (EOT) (n=18)
Without Psychiatric Comorbidities
LS Mean Δ[b] (SE): −5.4 (0.9)
P<0.0001
8.3
3.4
BL (n=15)
Optimized LXB (EOT) (n=15)
Total Narcolepsy Cohort
LS Mean Δ[b] (SE): −4.4 (0.6)
P<0.0001
9.0
4.6
BL (n=33)
Optimized LXB (EOT) (n=33)
Mean (SE) PHQ-9 total score[c]
B) PHQ-2
With Psychiatric Disorders
LS Mean Δ[b] (SE): −0.3 (0.3)
P=0.2928
1.6
1.2
BL (n=18)
Optimized LXB (EOT) (n=18)
Without Psychiatric Disorders
LS Mean Δ[b] (SE): −1.0 (0.3)
P=0.0013
1.3
0.5
BL (n=15)
Optimized LXB (EOT) (n=15)
Total Narcolepsy Cohort
LS Mean Δ[b] (SE): −0.6 (0.2)
P=0.0046
1.5
0.9
BL (n=33)
Optimized LXB (EOT) (n=33)
Mean (SE) PHQ-2 total score

[a]Completer analysis set. [b]Difference between BL and EOT. [c]Level of depressive symptom severity per PHQ-9 score: minimal, 0–4; mild, 5–9; moderate, 10–14; moderately severe, 15–19; severe, 20–27.[17]
BL, baseline; CI, confidence interval; EOT, end of treatment; LS, least squares; LXB, low-sodium oxybate; PHQ, Patient Health Questionnaire; SE, standard error.

Figure 6. Proportions of Participants Reporting "None to Minimal" Severity of Depressive Symptoms on the PHQ-9 Increased From BL to EOT in Participants With or Without Psychiatric Comorbidities[a]

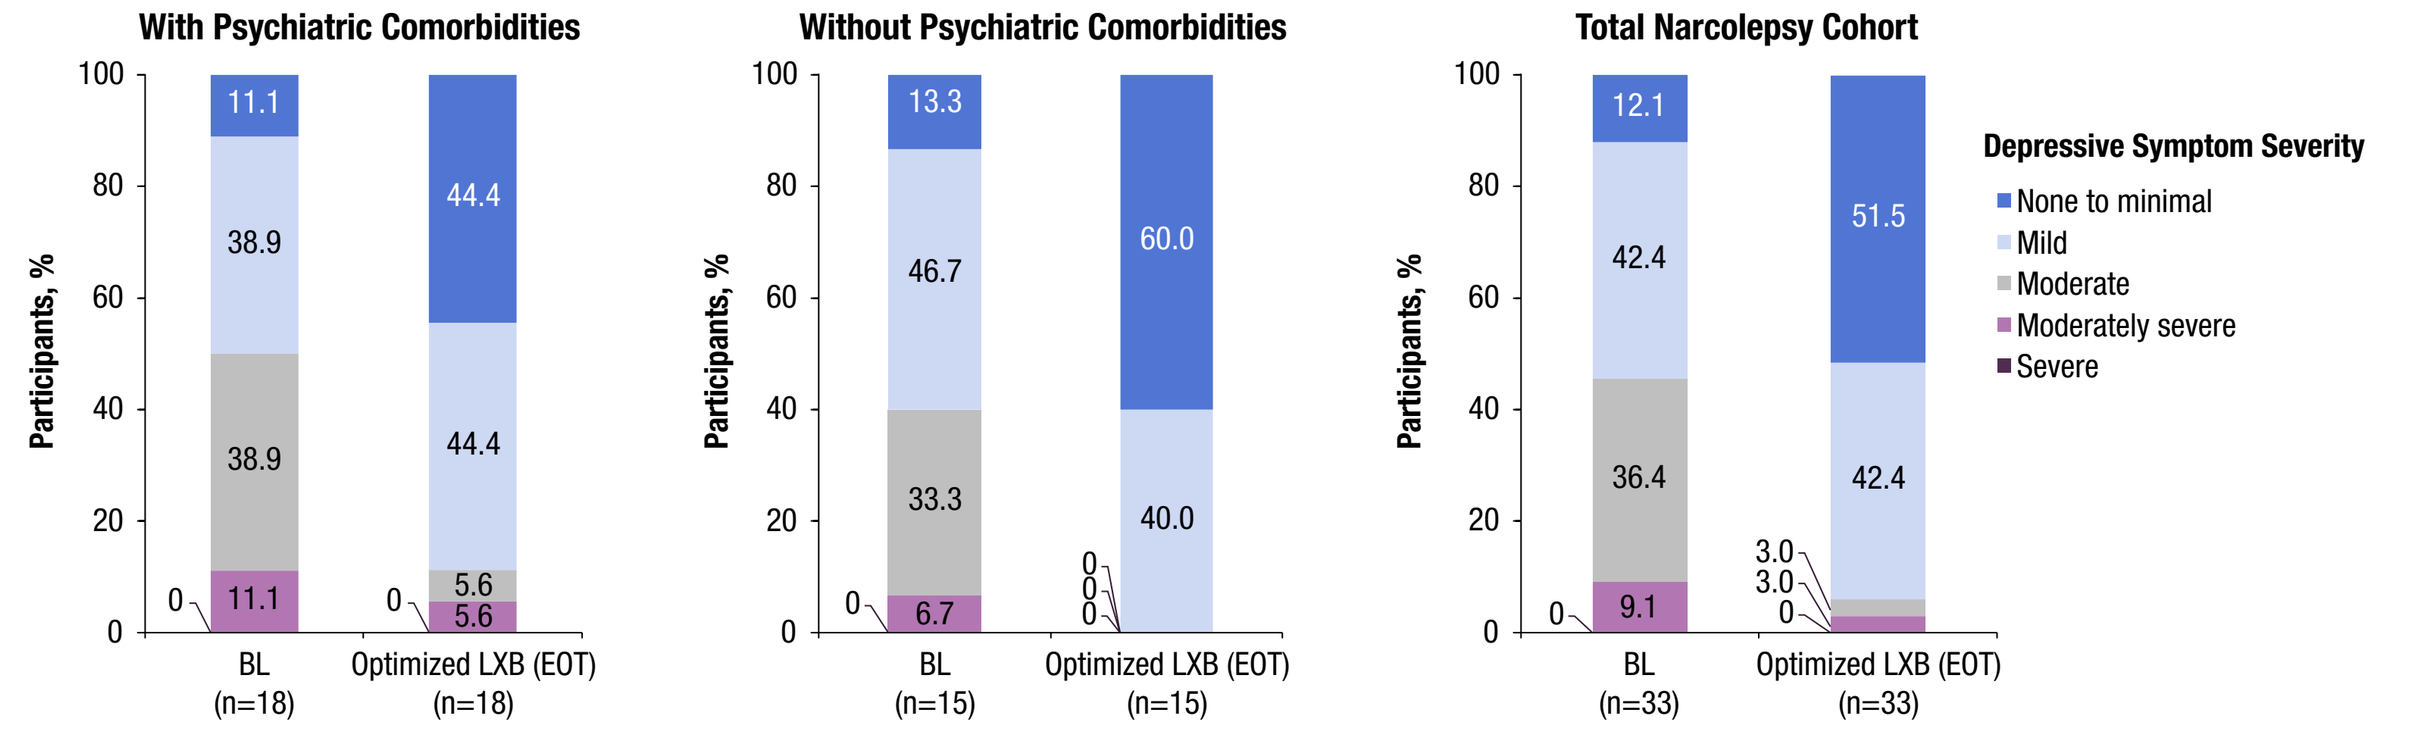

With Psychiatric Comorbidities
Without Psychiatric Comorbidities
Total Narcolepsy Cohort
Participants, %
BL (n=18)
Optimized LXB (EOT) (n=18)
BL (n=15)
Optimized LXB (EOT) (n=15)
BL (n=33)
Optimized LXB (EOT) (n=33)
Depressive Symptom Severity
None to minimal
Mild
Moderate
Moderately severe
Severe

[a]Completer analysis set.
BL, baseline; EOT, end of treatment; LXB, low-sodium oxybate.

Figure 7. Participants With or Without Psychiatric Comorbidities Reported Improvement in Overall Narcolepsy Disease on the Patient Global Impression of Severity (A) and the Patient Global Impression of Change (B) Scales[a]

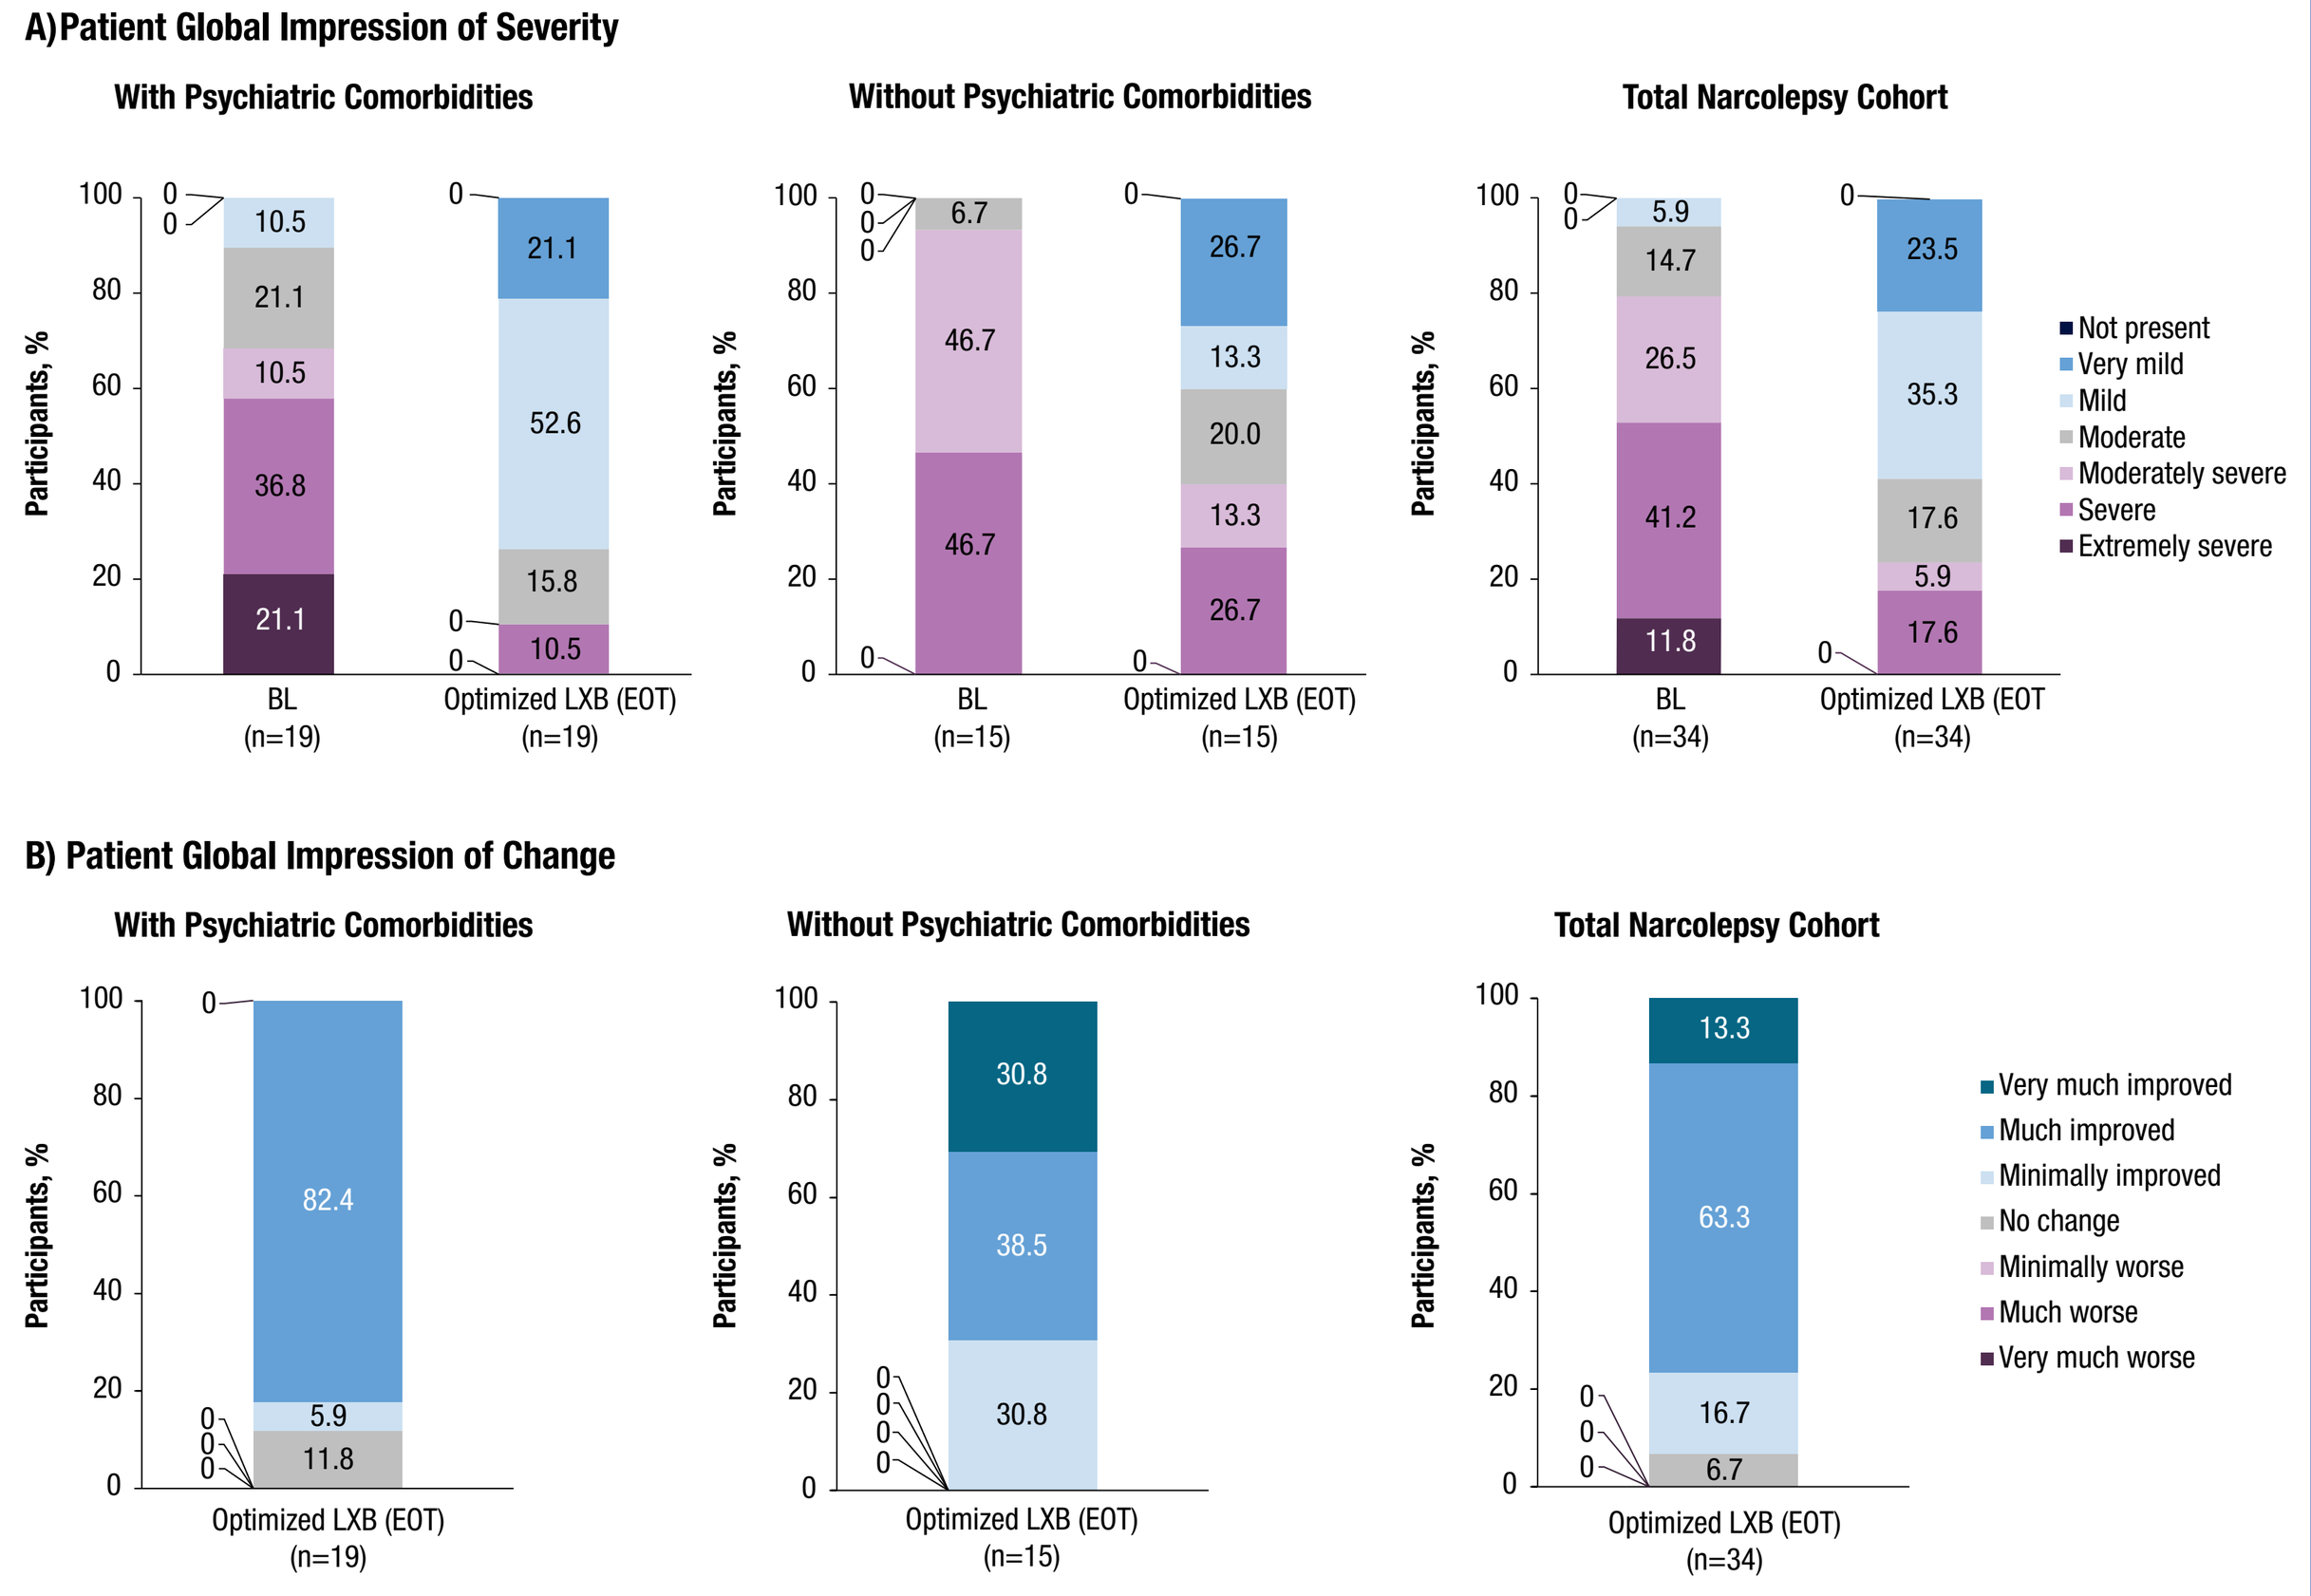

A) Patient Global Impression of Severity
With Psychiatric Comorbidities
Without Psychiatric Comorbidities
Total Narcolepsy Cohort
Participants, %
BL (n=19)
Optimized LXB (EOT) (n=19)
BL (n=15)
Optimized LXB (EOT) (n=15)
BL (n=34)
Optimized LXB (EOT (n=34)
Not present
Very mild
Mild
Moderate
Moderately severe
Severe
Extremely severe
B) Patient Global Impression of Change
Optimized LXB (EOT) (n=19)
Optimized LXB (EOT) (n=15)
Optimized LXB (EOT) (n=34)
Very much improved
Much improved
Minimally improved
No change
Minimally worse
Much worse
Very much worse

[a]Completer analysis set.
BL, baseline; EOT, end of treatment; LXB, low-sodium oxybate.

Table 4. Treatment-Emergent Adverse Events[a]

Participants, n (%)	With Psychiatric Comorbidities (n=26)	Without Psychiatric Comorbidities (n=29)	Total Narcolepsy Cohort (N=55)
With ≥1 TEAE	20 (76.9)	14 (48.3)	34 (61.8)
With ≥1 serious TEAE	0	0	0
With ≥1 TEAE related to treatment	18 (69.2)	12 (41.4)	30 (54.4)
With ≥1 TEAE leading to discontinuation	0	4 (13.8)	4 (7.3)
TEAEs occurring in ≥5% of participants			
Nausea	7 (26.9)	6 (20.7)	13 (23.6)
Dizziness	4 (15.4)	4 (13.8)	8 (14.5)
Headache	4 (15.4)	3 (10.3)	7 (12.7)
Vomiting	5 (19.2)	1 (3.4)	6 (10.9)
Somnolence	4 (15.4)	2 (6.9)	6 (10.9)
Anxiety	3 (11.5)	1 (3.4)	4 (7.3)
Oropharyngeal pain	3 (11.5)	1 (3.4)	4 (7.3)
Nasal congestion	1 (3.8)	3 (10.3)	4 (7.3)
Hypoesthesia	3 (11.5)	0	3 (5.5)
Decreased appetite	2 (7.7)	1 (3.4)	3 (5.5)
Brain fog	1 (3.8)	2 (6.9)	3 (5.5)
Enuresis	1 (3.8)	2 (6.9)	3 (5.5)
Cough	1 (3.8)	2 (6.9)	3 (5.5)
COVID-19	2 (7.7)	0	2 (3.6)
Sinusitis	2 (7.7)	0	2 (3.6)
Paraesthesia	0	2 (2.9)	2 (3.6)

[a]Safety analysis set.
TEAE, treatment-emergent adverse event.

- Overall, 20 participants (76.9%) with psychiatric comorbidities and 14 (48.3%) of participants without psychiatric comorbidities reported a TEAE
- Four participants discontinued due to a TEAE, which included nausea, anxiety, dysphoria, and irritability
- No participants endorsed suicidal ideation or behavior on the C-SSRS at any point during the study

Conclusions

- Results demonstrated effectiveness of LXB in participants with narcolepsy, both with and without psychiatric comorbidities
 - Participants in both subgroups showed improvement in daytime sleepiness (mean ESS scores from severe EDS to normal range), narcolepsy symptoms (mean NSS scores for NT1 from severe to mild), and overall disease (PGI-C and PGI-S scores) with optimized LXB treatment compared with BL (off-LXB)
 - Participants in both subgroups showed improvements in PHQ-9 scores with optimized LXB treatment compared with BL, demonstrating reduced depressive symptoms
- TEAEs were consistent with the known LXB safety profile
- Limitations include the open-label and single-arm design; causality cannot be established. Analyses were based on the completer analysis set of participants who reached a stable LXB dosage and may not represent the experience of all individuals starting LXB treatment

References: **1.** Xywav® (calcium, magnesium, potassium, and sodium oxybates) oral solution, CIII [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **2.** Szarfman A, et al. *N Engl J Med.* 1995;333(19):1291. **3.** US Food and Drug Administration. Clinical review for Binosto, NDA 202344. 2012. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2012/202344Orig1s000MedR.pdf. **4.** US Food and Drug Administration. Quantitative labeling of sodium, potassium, and phosphorus for human over-the-counter and prescription drug products. Guidance for industry. 2022. https://www.fda.gov/regulatory-information/search-fda-guidance-documents/quantitative-labeling-sodium-potassium-and-phosphorus-human-over-counter-and-prescription-drug. **5.** Ruoff CM, et al. *J Clin Psychiatry.* 2017;78(2):171-176. **6.** Ohayon MM. *Sleep Med.* 2013;14(6):488-492. **7.** Morse AM, Sanjeev K. *Med Sci (Basel).* 2018;6(1):16. **8.** Barateau L, et al. *Curr Psychiatry Rep.* 2017;19(2):13. **9.** Jennum P, et al. *Sleep.* 2013;36(6):835-840. **10.** Black J, et al. *Sleep Med.* 2017;33:13-18. **11.** Barateau L, et al. *Sleep.* 2024;47(5):zsad323. **12.** Kroenke K, et al. *Medical Care.* 2003;41(11):1284-1292. **13.** Berry RB, et al. *The AASM Manual for the Scoring of Sleep and Associated Events: Rules, Terminology and Technical Specifications, Version 3.* Darien, IL: American Academy of Sleep Medicine; 2023. **14.** Johns MW. *Sleep.* 1991;14(6):540-545. **15.** Maski K, et al. *J Clin Sleep Med.* 2021;17(9):1895-1945. **16.** Dauvilliers Y, et al. *Sleep.* 2020;43(6):zsaa009. **17.** Kroenke K, et al. *J Gen Intern Med.* 2001;16(9):606-613.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees Martina Kusi-Mensah, PharmD, and Emily Bruggeman, PhD, provided medical writing support and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: C Ruoff has served as an advisory board member for Alkermes, Eisai, Jazz Pharmaceuticals, and Takeda, and has received grant funding from Jazz Pharmaceuticals. **DT Plante** is a consultant and advisory board member for Jazz Pharmaceuticals; an advisory board member for Apnimed; a consultant for Aditum Bio and Teva Pharmaceuticals (Australia); and has served as a consultant/advisory board member for Alkermes, Centessa, Harmony Biosciences, and Takeda. **DA Nichols, J Dai, MT Kirby,** and **M Whalen** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **TL Steininger** is a former full-time employee and current contract worker for Jazz Pharmaceuticals who has received shares of Jazz Pharmaceuticals, plc. **A Cairns** is a full-time contract worker and advisory board member for Jazz Pharmaceuticals. **LD Schneider** has received personal compensation for serving on advisory boards and speakers bureaus for Avadel, Eisai, and Jazz Pharmaceuticals.

Scan this code to access this poster online.
This code is not for promotional purposes.

Presented at Psych Congress 2025; September 17–21, 2025; San Diego, CA, USA