125

Greater Than 9 Gram Dosage of Low-Sodium Oxybate in Study Participants With Narcolepsy: Effectiveness and Safety Results From the DUET Study

Jerald H. Simmons, MD[1,2]; Logan D. Schneider, MD[3]; Chad M. Ruoff, MD[4]; David T. Plante, MD, PhD[5]; Deborah A. Nichols, MS[6]; Teresa L. Steininger, PhD[6]; Marisa Whalen, PharmD[7]; M. Todd Kirby, PhD[7,8]; Jing Dai, PhD[6]; Alyssa Cairns, PhD[7]; Richard K. Bogan, MD[9]

[1]Comprehensive Sleep Medicine Associates, Houston, TX, USA; [2]Sleep Education Consortium, Sugar Land, TX, USA; [3]Stanford University Center for Sleep Sciences and Medicine, Stanford, CA, USA; [4]Mayo Clinic, Scottsdale, AZ, USA; [5]Department of Psychiatry, University of Wisconsin–Madison, Madison, WI, USA; [6]Jazz Pharmaceuticals, Palo Alto, CA, USA; [7]Jazz Pharmaceuticals, Philadelphia, PA, USA; [8]Department of Psychiatry & Health Behavior, Medical College of Georgia at Augusta University, Augusta, GA, USA; [9]University of South Carolina School of Medicine, Columbia, SC, USA

Introduction

- Low-sodium oxybate (LXB, Xywav®) is approved by the US Food and Drug Administration to treat excessive daytime sleepiness (EDS) or cataplexy in patients ≥7 years of age with narcolepsy and for the treatment of idiopathic hypersomnia in adults[1-4]
- The recommended dosage range is 6 g to 9 g/night, gradually titrated based on efficacy and tolerability[1]
- Some patients may achieve better responses with unequal doses at bedtime and 2.5 to 4 hours later[1]
- Dosages higher than 9 g/night have not been studied and ordinarily should not be administered
 - Real-world data from the Nexus Narcolepsy Registry reported that 4.1% of patients taking sodium oxybate were taking a dosage of >9 g/night[5]
- Jazz DUET (**D**evelop hypersomnia **U**nderstanding by **E**valuating low-sodium oxybate **T**reatment) was a prospective, open-label study (NCT05875974) of LXB treatment in participants with narcolepsy (type 1 [NT1] or type 2 [NT2]) or idiopathic hypersomnia
- DUET included a third cohort in which participants with narcolepsy who were currently taking 9 g of oxybate and, in the opinion of the investigator, would likely benefit from continued titration could enter the study and titrate from 9 g to an optimized dosage up to 12 g (twice nightly; maximum 4.5 g for second dose)

Objective

- To evaluate the effectiveness and safety of LXB in participants with narcolepsy taking a dosage of >9 g/night

Methods

Figure 1. Study Design

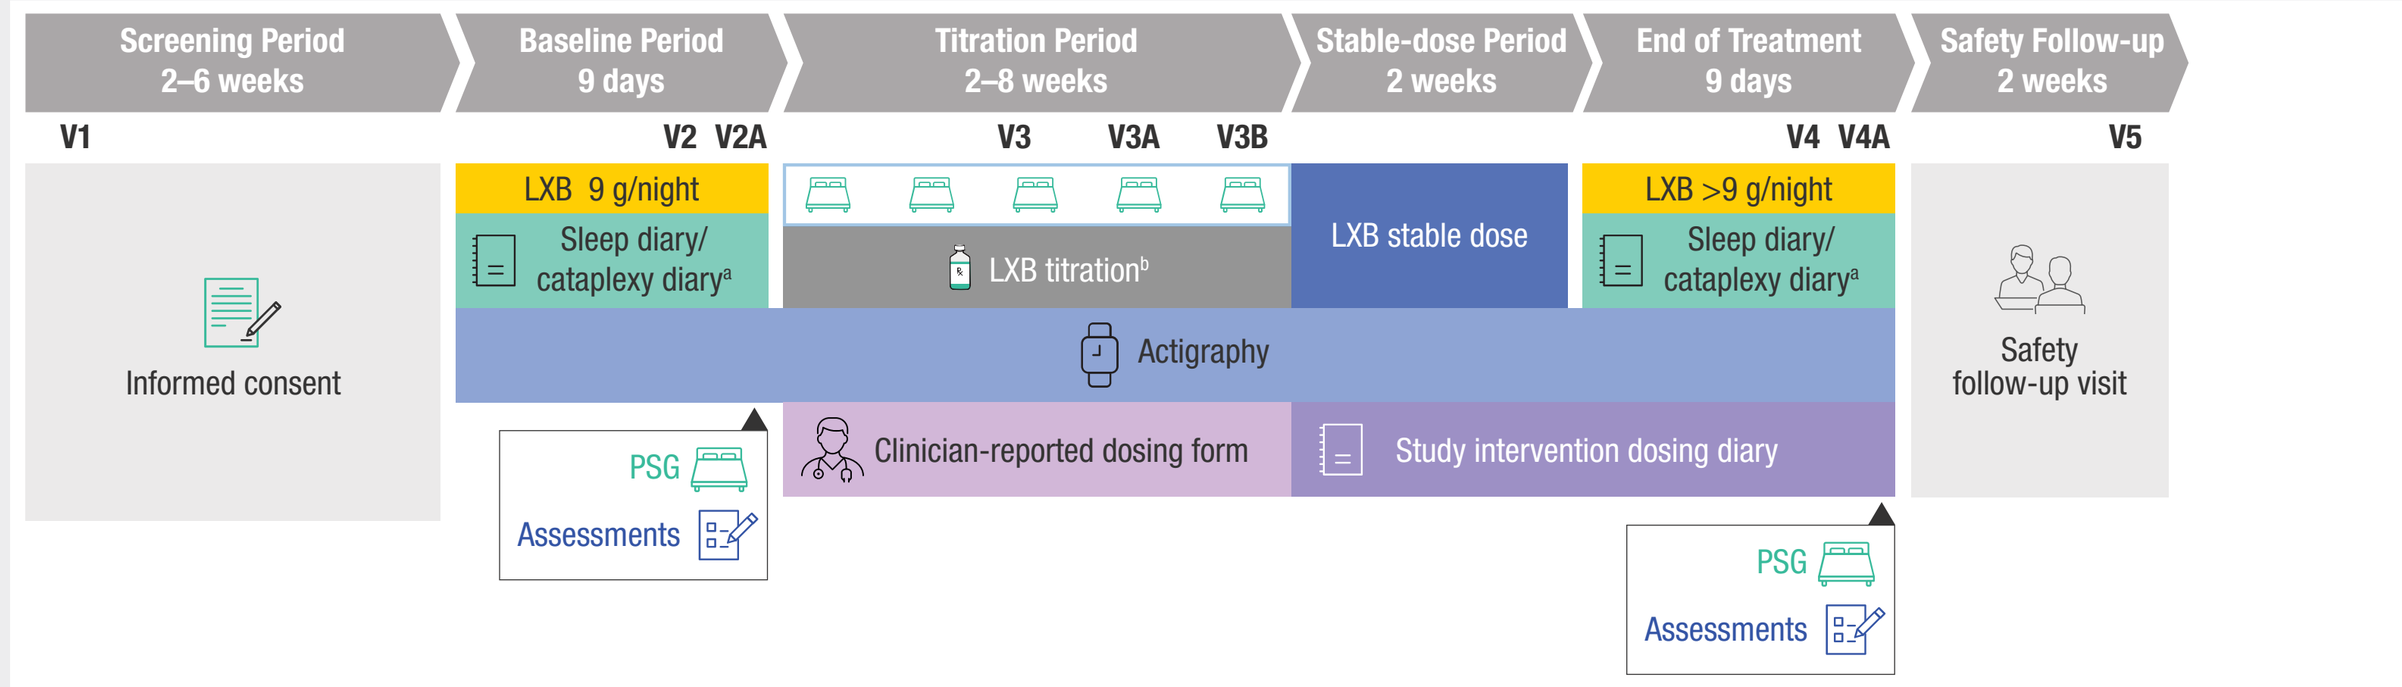
Screening Period
2–6 weeks
Baseline Period
9 days
Titration Period
2–8 weeks
Stable-dose Period
2 weeks
End of Treatment
9 days
Safety Follow-up
2 weeks
V1
V2 V2A
V3 V3A V3B
V4 V4A
V5
Informed consent
LXB 9 g/night
Sleep diary/ cataplexy diary[a]
LXB titration[b]
LXB stable dose
LXB >9 g/night
Sleep diary/ cataplexy diary[a]
Actigraphy
Clinician-reported dosing form
Study intervention dosing diary
PSG
Assessments
PSG
Assessments
Safety follow-up visit

[a]Cataplexy diary in NT1 only. [b]Weekly titration visits with a dose escalation were performed as an overnight PSG visit for safety monitoring; visits without a dose escalation could be via teleconference. Titration could take between 2 and 8 weeks. V3 occurred on titration day 14. Additional in-clinic visits were scheduled for day 35 (V3A) and day 56 (V3B), as needed. Investigator could optimize participant dosage and move participant to SDP at V3, V3A, or V3B, but not during intervening weekly teleconferences.
LXB, low-sodium oxybate; PSG, polysomnography; NT1, narcolepsy type 1; SDP, stable-dose period; V, visit.

- DUET included a screening period, a 9-day baseline (BL) period on LXB 9 g/night (8 days of daily assessments ending with an overnight BL polysomnography [PSG] with additional assessments, followed by an optional overnight pharmacokinetic [PK] visit), a 2- to 8-week LXB titration period (with an overnight PSG visit for all dose escalations, for safety monitoring), a 2-week stable-dose period (SDP), a 9-day end-of-treatment (EOT) period while participants were taking their optimized stable dose of LXB (daily assessments ending with an overnight EOT PSG with additional assessments, followed by an optional overnight PK visit), and a 2-week safety follow-up; PK visits were conducted at select study sites at V2A and V4A

Figure 2. Key Inclusion and Exclusion Criteria

Key Inclusion Criteria
- Adults aged 18 to 75 with a primary diagnosis of NT1 or NT2[a]
- Stable use of concomitant anticataplectics or alerting agents[b] allowed
- On 9 g of LXB at screening

Key Exclusion Criteria
- Untreated or inadequately treated OSA at baseline PSG (AHI >10)[c]
- History/presence of:
 - Unstable or clinically significant medical condition
 - Behavioral, psychiatric, neurologic disorder[d]

[a]Meeting the *International Classification of Sleep Disorders – Third Edition* or *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition* criteria. [b]Alerting agents were defined as stimulants or wake-promoting agents. [c]Defined as AHI >10, with hypopnea definition including a ≥4% desaturation as per Rule 1B of *The AASM Manual for the Scoring of Sleep and Associated Events*.[6] [d]As determined by the investigator.
AHI, apnea-hypopnea index; AASM, American Academy of Sleep Medicine; LXB, low-sodium oxybate; NT1, narcolepsy type 1; NT2, narcolepsy type 2; OSA, obstructive sleep apnea; PSG, polysomnography.

- Participants could enter the >9 g narcolepsy cohort 1 of 2 ways:
 - Potential participants with narcolepsy taking LXB at a dosage of 9 g/night at screening who, in the opinion of the investigator, would likely benefit from continued titration to dosages >9 g/night and who met eligibility criteria
 - Participants with narcolepsy already enrolled in the narcolepsy cohort, who had titrated up to 9 g and who, in the opinion of the investigator, could benefit from a higher LXB dose, could transfer to the >9 g cohort
- Participants adequately treated for OSA (apnea-hypopnea index [AHI] ≤10) were eligible and asked to maintain any treatment regimen (eg, positive airway pressure, oral appliance) for the entirety of the study
- There was no Epworth Sleepiness Scale (ESS) score eligibility requirement at screening for the >9 g cohort

LXB Titration Requirements

Investigators titrated LXB to an optimal dosage for participants in the >9 g cohort under the following requirements:
- The first night for all LXB dose escalations above 9 g were conducted in the sleep laboratory clinic with PSG and appropriate in-clinic safety monitoring
- Each dose-escalation step during titration was limited to 0.5 g/night per week up to a maximum of 12 g nightly
- All participants took twice-nightly LXB
 - The first nightly maximal dose was 7.5 g
 - The second nightly maximal dose was 4.5 g and was administered approximately 4 hours after the first dose

LXB, low-sodium oxybate; PSG, polysomnography

- Key endpoints included change from BL to EOT in ESS score, Narcolepsy Severity Scale (NSS) score (NT1; the NSS-2 version, excluding questions on cataplexy, was administered to participants with NT2), and frequency of weekly cataplexy attacks (NT1 only; self-reported via eDiary)
- Safety endpoints included incidence and severity of treatment-emergent adverse events (TEAEs), Columbia-Suicide Severity Rating Scale (C-SSRS), and PSG-measured sleep-related respiratory assessments, including AHI, number of central sleep apnea events, mean oxygen saturation (SpO_2), and percentage of total sleep time spent with SpO_2 <90%
- The safety analysis set includes all participants who enrolled in the study and took their prescribed LXB regimen for ≥1 night after the BL period (N=48); the completer analysis set includes all participants who enrolled in the study, took their prescribed LXB regimen for ≥1 night after the BL period, completed the SDP, and completed the PSG EOT visit (n=44)
- Observed values and change from BL were summarized as continuous variables; no formal hypothesis testing was planned for the >9 g cohort
 - Least squares (LS) mean differences were calculated using mixed model with repeated measures of change from BL to EOT, adjusted for the BL value

Results

Table 1. Demographics and Baseline Characteristics[a]

Characteristic	Narcolepsy Type 1 (n=30)	Narcolepsy Type 2 (n=18)	Total Narcolepsy Cohort (N=48)
Age, years			
Mean (SD)	38.2 (11.5)	40.8 (10.0)	39.2 (10.9)
Median (min, max)	37.0 (18, 65)	39.0 (20, 62)	38.0 (18, 65)
Sex at birth, n (%)			
Male	10 (33.3)	8 (44.4)	18 (37.5)
Female	20 (66.7)	10 (55.6)	30 (62.5)
Gender identity, n (%)			
Male (including transgender man)	10 (33.3)	8 (44.4)	18 (37.5)
Female (including transgender woman)	20 (66.7)	10 (55.6)	30 (62.5)
Participant of childbearing potential, n (%)[b]	16 (80.0)	7 (70.0)	23 (76.7)
Race, n (%)			
White	24 (80.0)	13 (72.2)	37 (77.1)
Black or African American	3 (10.0)	4 (22.2)	7 (14.6)
Asian	1 (3.3)	0	1 (2.1)
Multiple[c]	0	1 (5.6)	1 (2.1)
Ethnicity, n (%)			
Hispanic or Latino	1 (3.3)	0	1 (2.1)
Not Hispanic or Latino	29 (96.7)	18 (100)	47 (97.9)
Body mass index, kg/m²			
Mean (SD)	30.0 (8.0)	29.7 (10.5)	29.9 (8.9)
Median (min, max)	28.6 (20, 52)	27.2 (19, 59)	27.4 (19, 59)
Diagnosed obstructive sleep apnea,[d] n (%)	6 (20.0)	6 (33.3)	12 (25.0)

[a]Safety analysis set. [b]Percentage calculated only for female sex at birth. [c]Participant reported >1 race. [d]Per medical history at screening V1.
max, maximum; min, minimum; SD, standard deviation; V, visit.

- Most participants were female (63%) and White (77%)

Table 2. Mean Nightly LXB Dosage During Stable-Dose Period[a,b]

LXB Dose, g	Narcolepsy Type 1 (n=28)	Narcolepsy Type 2 (n=17)	Total Narcolepsy Cohort (N=45[c])
Overall total nightly LXB dosage			
Mean (SD)	11.1 (1.0)	11.1 (1.0)	11.1 (1.0)
Median (min, max)	11.5 (9.0, 12.0)	11.5 (9.0, 12.0)	11.5 (9.0, 12.0)
Twice-nightly LXB dosage[d]			
First nightly LXB dose			
Mean (SD)	6.6 (1.1)	6.6 (1.1)	6.6 (1.1)
Median (min, max)	7.0 (4.5, 7.5)	7.0 (4.5, 7.5)	7.0 (4.5, 7.5)
Second nightly LXB dose			
Mean (SD)	4.4 (0.2)	4.5 (0.1)	4.5 (0.2)
Median (min, max)	4.5 (3.5, 4.8)	4.5 (4.5, 5.0)	4.5 (3.5, 5.0)

[a]Includes all participants from the safety analysis set who reached the SDP. [b]Once a participant reached an optimized dosage based on investigator judgment, they continued this dosage as a stable regimen during the SDP and EOT periods. One participant downtitrated to 9.0 g/night and later withdrew due to an AE of nausea. [c]All participants took LXB twice nightly.
AE, adverse event; LXB, low-sodium oxybate; max, maximum; min, minimum; SD, standard deviation.

- 46.7% (21/45) of participants reached a stable total LXB dosage of 12 g/night

Figure 3. ESS Scores Improved From BL to EOT in Participants Taking >9 g/night LXB[a]

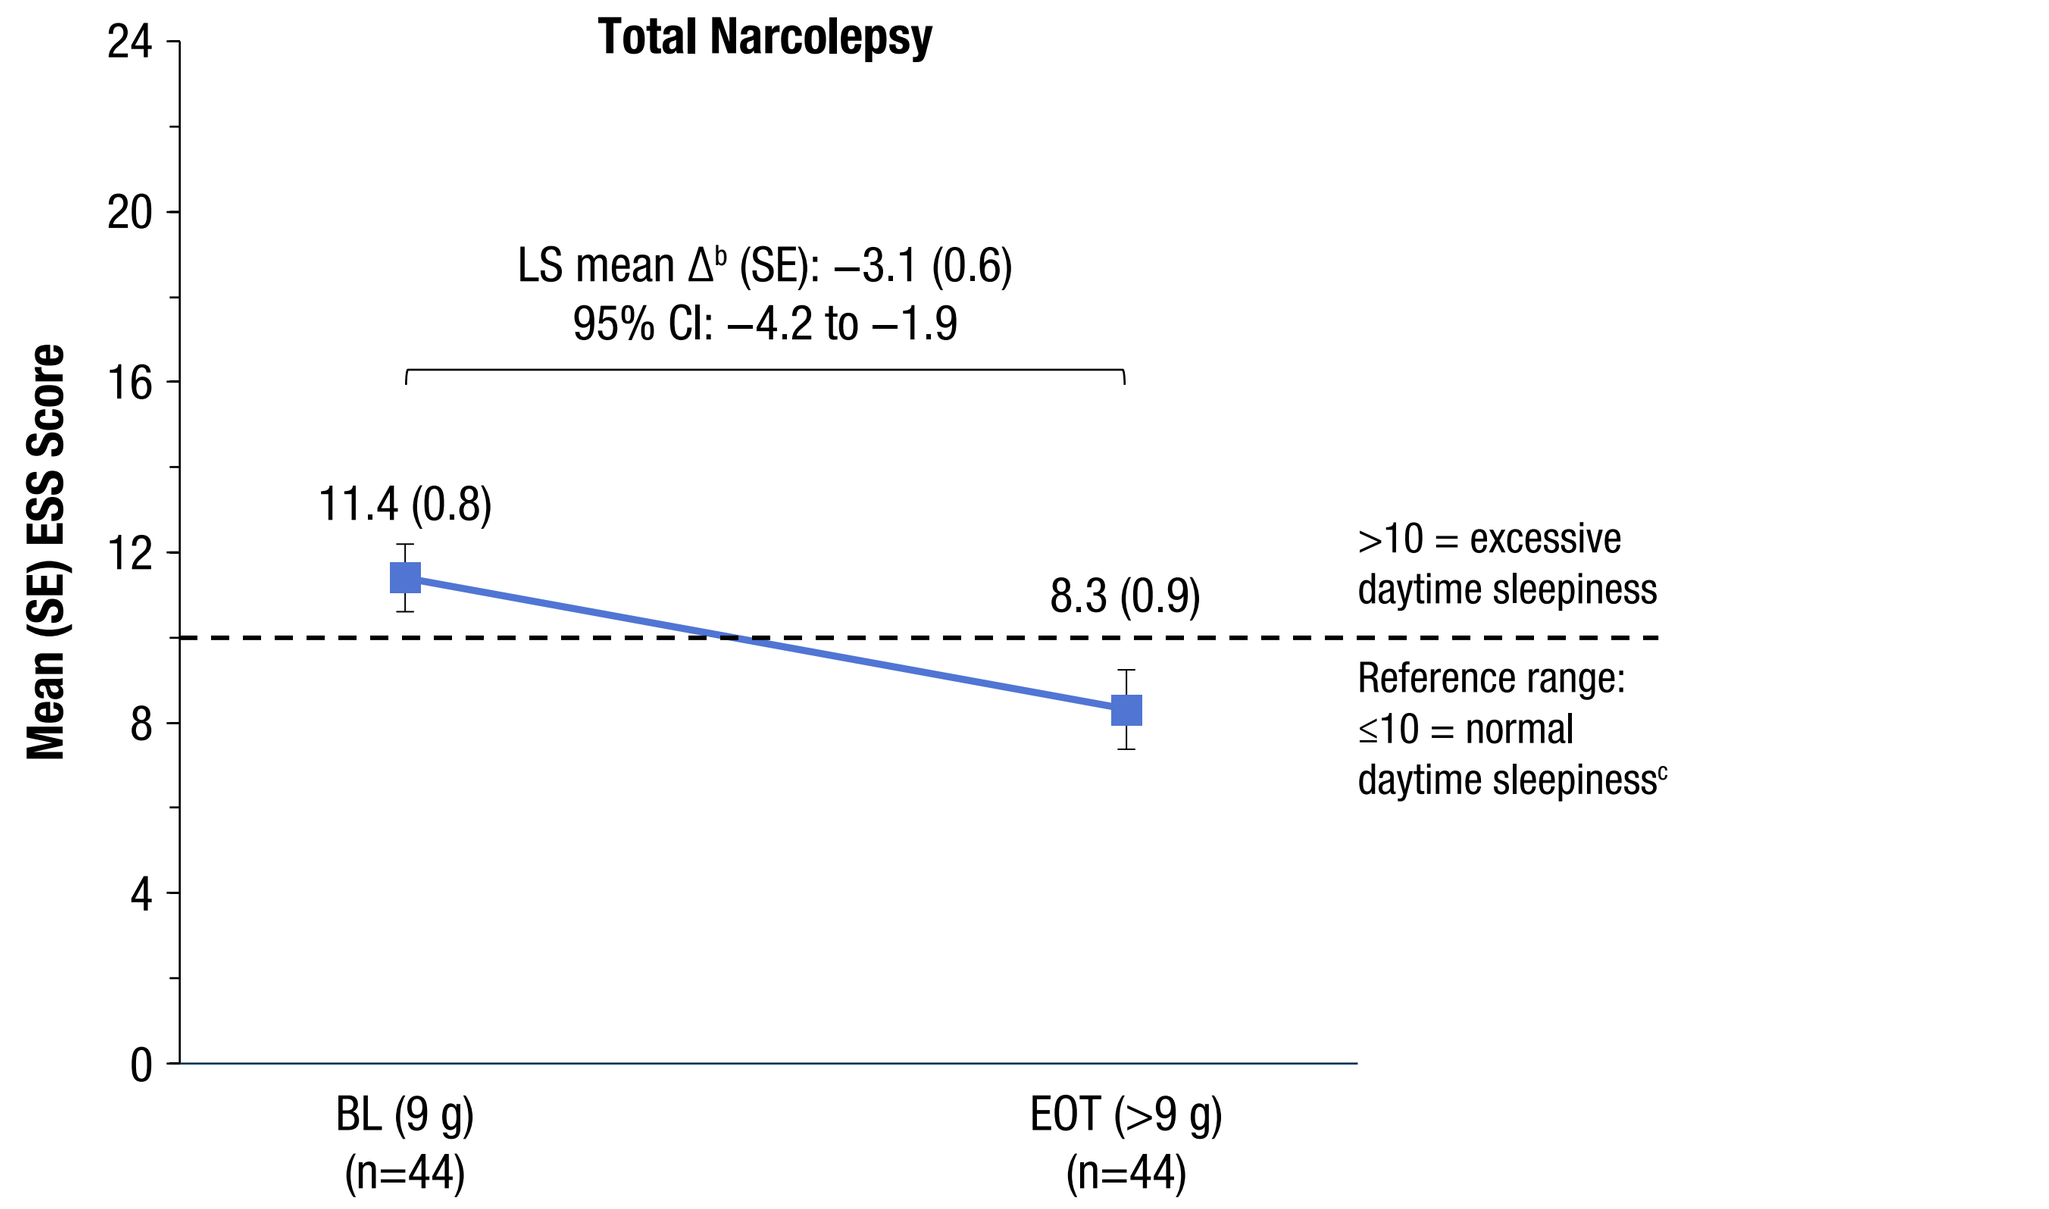
Total Narcolepsy
Mean (SE) ESS Score
24
20
16
12
8
4
0
LS mean Δ[b] (SE): −3.1 (0.6)
95% CI: −4.2 to −1.9
11.4 (0.8)
8.3 (0.9)
>10 = excessive daytime sleepiness
Reference range: ≤10 = normal daytime sleepiness[c]
BL (9 g) (n=44)
EOT (>9 g) (n=44)

[a]Completer analysis set. [b]Difference between EOT and BL. [c]Established categories consider scores up to 10 normal.[7] The MCID for ESS was defined as 2 points according to an AASM systematic review and meta-analysis.[8]
AASM, American Academy of Sleep Medicine; BL, baseline; CI, confidence interval; EOT, end of treatment; ESS, Epworth Sleepiness Scale; LS, least squares; LXB, low-sodium oxybate; MCID, minimal clinically important difference; SE, standard error.

- Mean (SE) ESS score at BL was 11.4 (0.8), indicating EDS despite treatment with LXB 9 g/night and the use of alerting agents by 58% of participants; at EOT, this decreased to 8.3 (0.9)
- Mean (SE) ESS scores also decreased from BL to EOT for NT1 and NT2
 - NT1: 12.0 (1.1) to 9.2 (1.3); LS mean (95% CI) change, −2.8 (−4.3, −1.4)
 - NT2: 10.3 (1.1) to 6.9 (1.3); LS mean (95% CI) change, −3.5 (−5.4, −1.6)

Figure 4. Participants Taking >9 g/night Reported Improvements in Narcolepsy Symptoms on the NSS (A) and NSS-2 (B)[a]

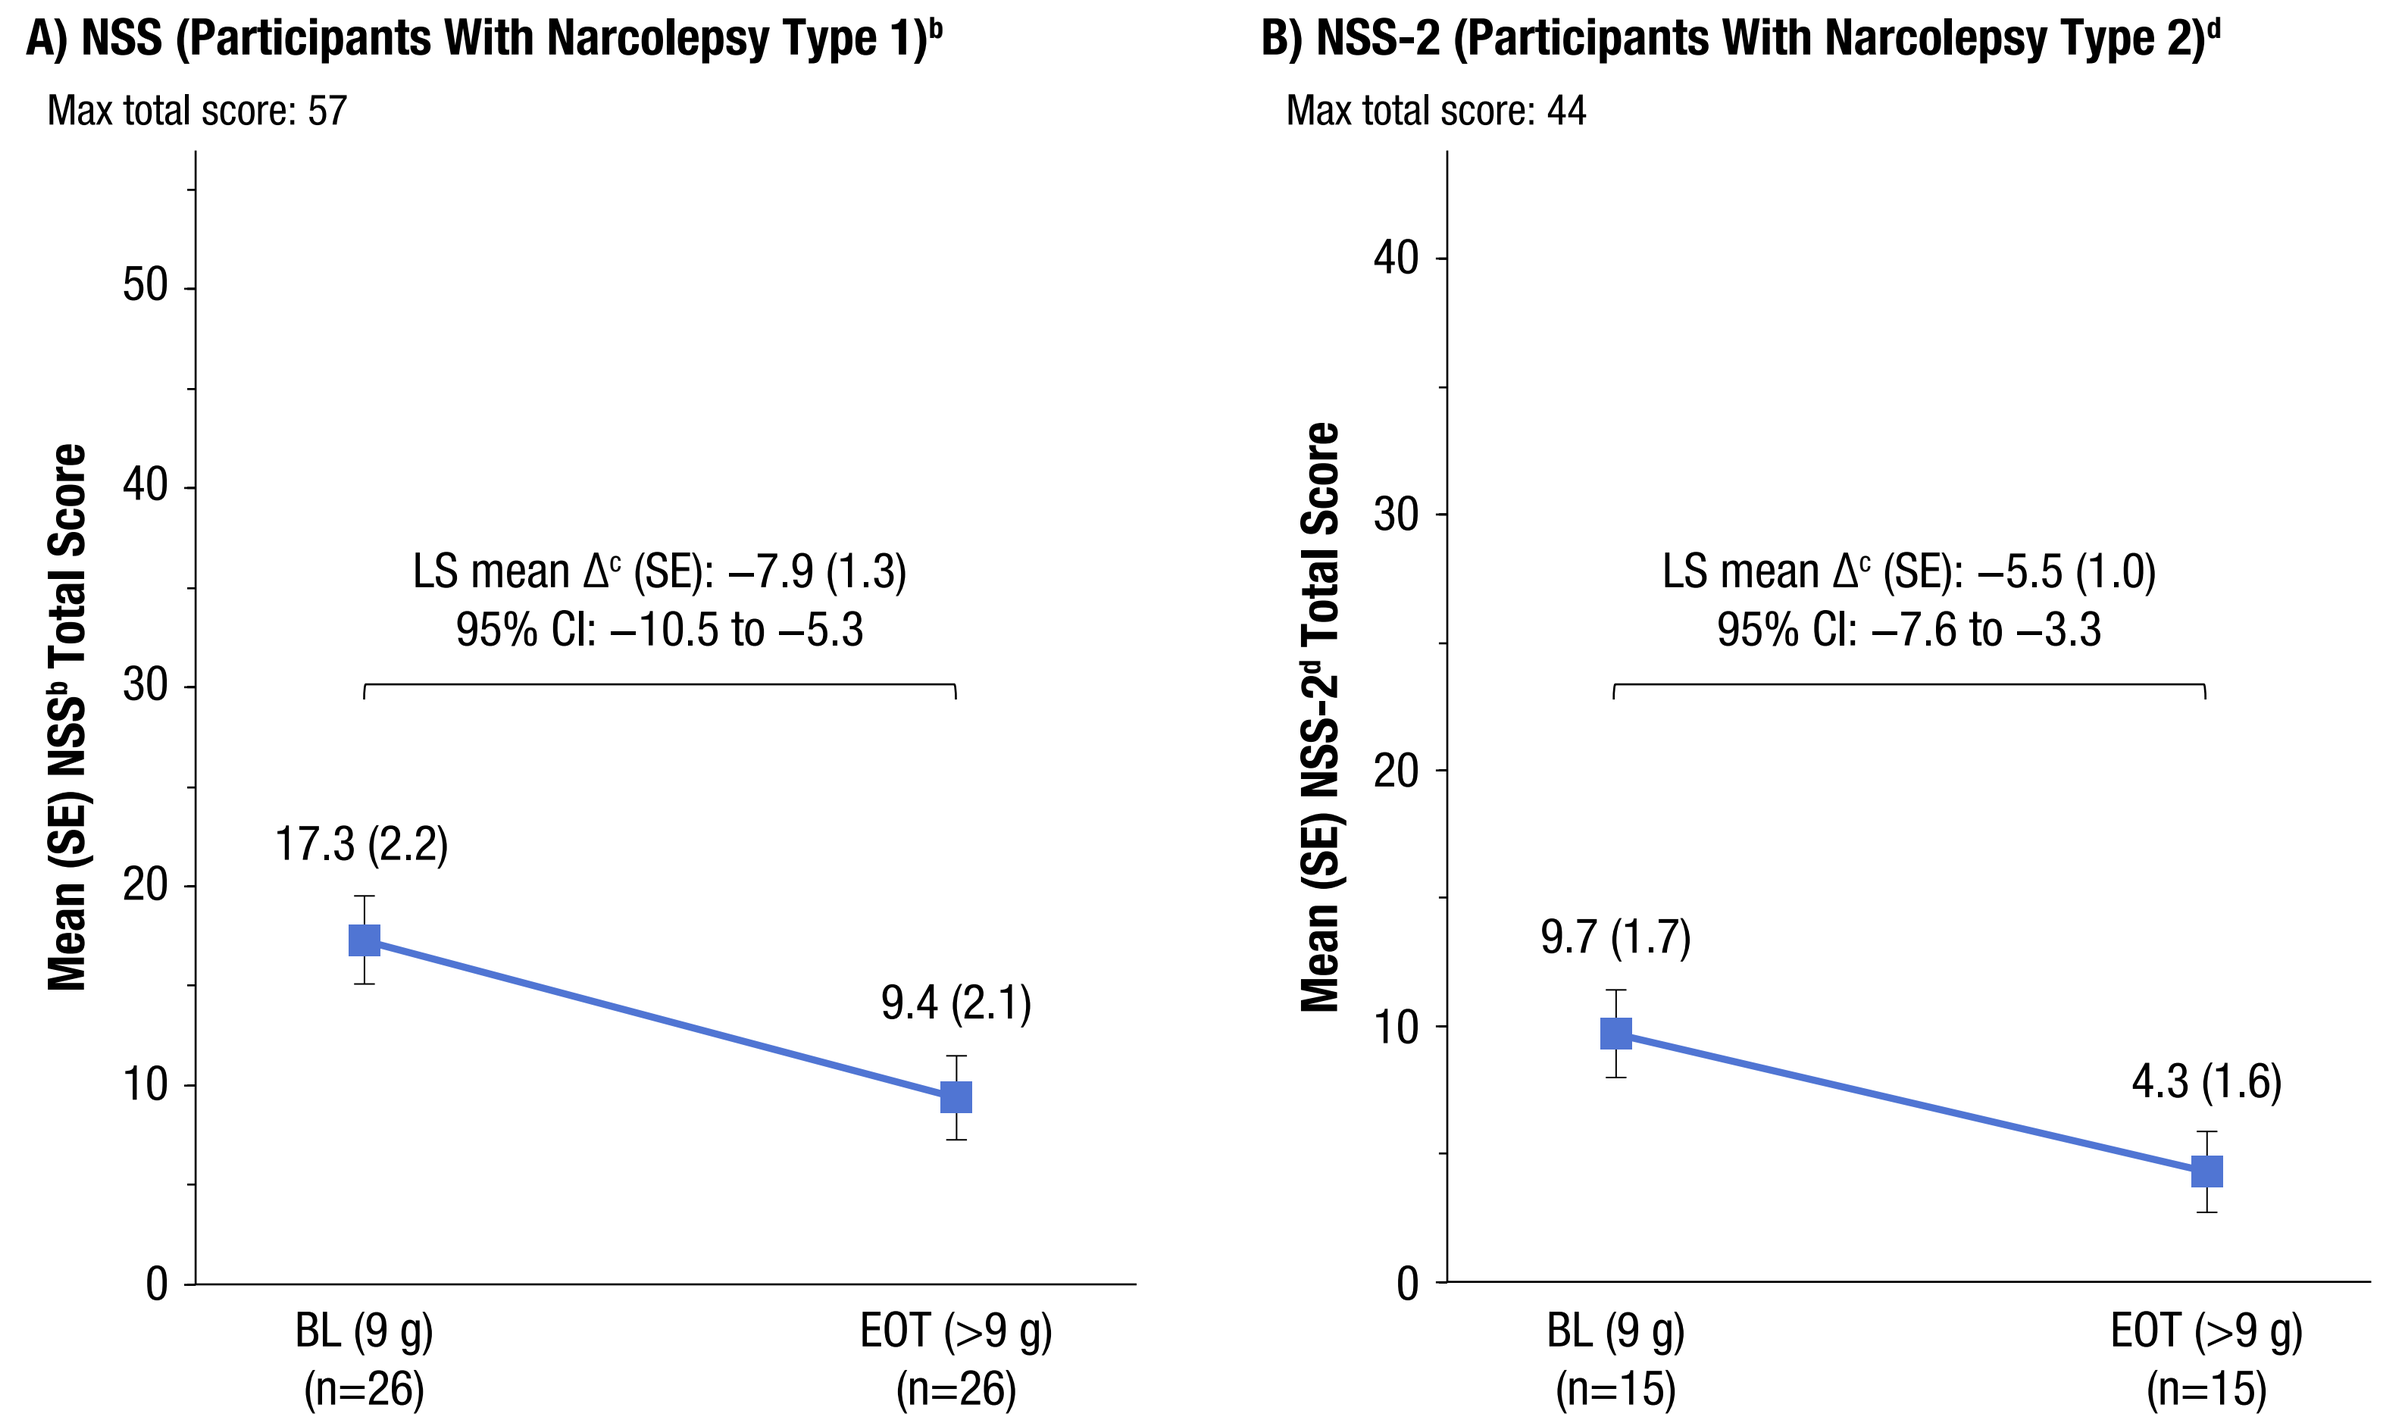
A) NSS (Participants With Narcolepsy Type 1)[b]
Max total score: 57
Mean (SE) NSS[b] Total Score
50
40
30
20
10
0
LS mean Δ[c] (SE): −7.9 (1.3)
95% CI: −10.5 to −5.3
17.3 (2.2)
9.4 (2.1)
BL (9 g) (n=26)
EOT (>9 g) (n=26)
B) NSS-2 (Participants With Narcolepsy Type 2)[d]
Max total score: 44
Mean (SE) NSS-2[d] Total Score
40
30
20
10
0
LS mean Δ[c] (SE): −5.5 (1.0)
95% CI: −7.6 to −3.3
9.7 (1.7)
4.3 (1.6)
BL (9 g) (n=15)
EOT (>9 g) (n=15)

[a]Completer analysis set. [b]For the NSS, levels of severity are as follows: very severe (43–57), severe (29–42), moderate (15–28), mild (0–14).[9] [c]Difference between EOT and BL. [d]For the NSS-2, the MCID between untreated and treated patients was 3.35 with the Cohen effect size and 3.22 with the empirical rule effect size according to a longitudinal study (n=26).[10]
BL, baseline; CI, confidence interval; EOT, end of treatment; LS, least squares; LXB, low-sodium oxybate; Max, maximum; MCID, minimal clinically important difference; NSS, Narcolepsy Severity Scale; NSS-2, Narcolepsy Severity Scale-2; SE, standard error.

- For NT1, mean (SE) NSS score at BL was 17.3 (2.2), indicating participants' symptoms were moderate in severity despite treatment with LXB 9 g/night; at EOT, this decreased to 9.4 (2.1), indicating symptoms improved from moderate to mild[9]
- For NT2, NSS-2 score decreased by 5.5 points from BL to EOT (LS mean difference), which is greater than the minimal clinically important difference of 3.35 points[10]

Figure 5. Weekly Cataplexy Attack Frequency (eDiary) Decreased From BL to EOT in Participants With Narcolepsy Type 1 Taking >9 g/night LXB[a]

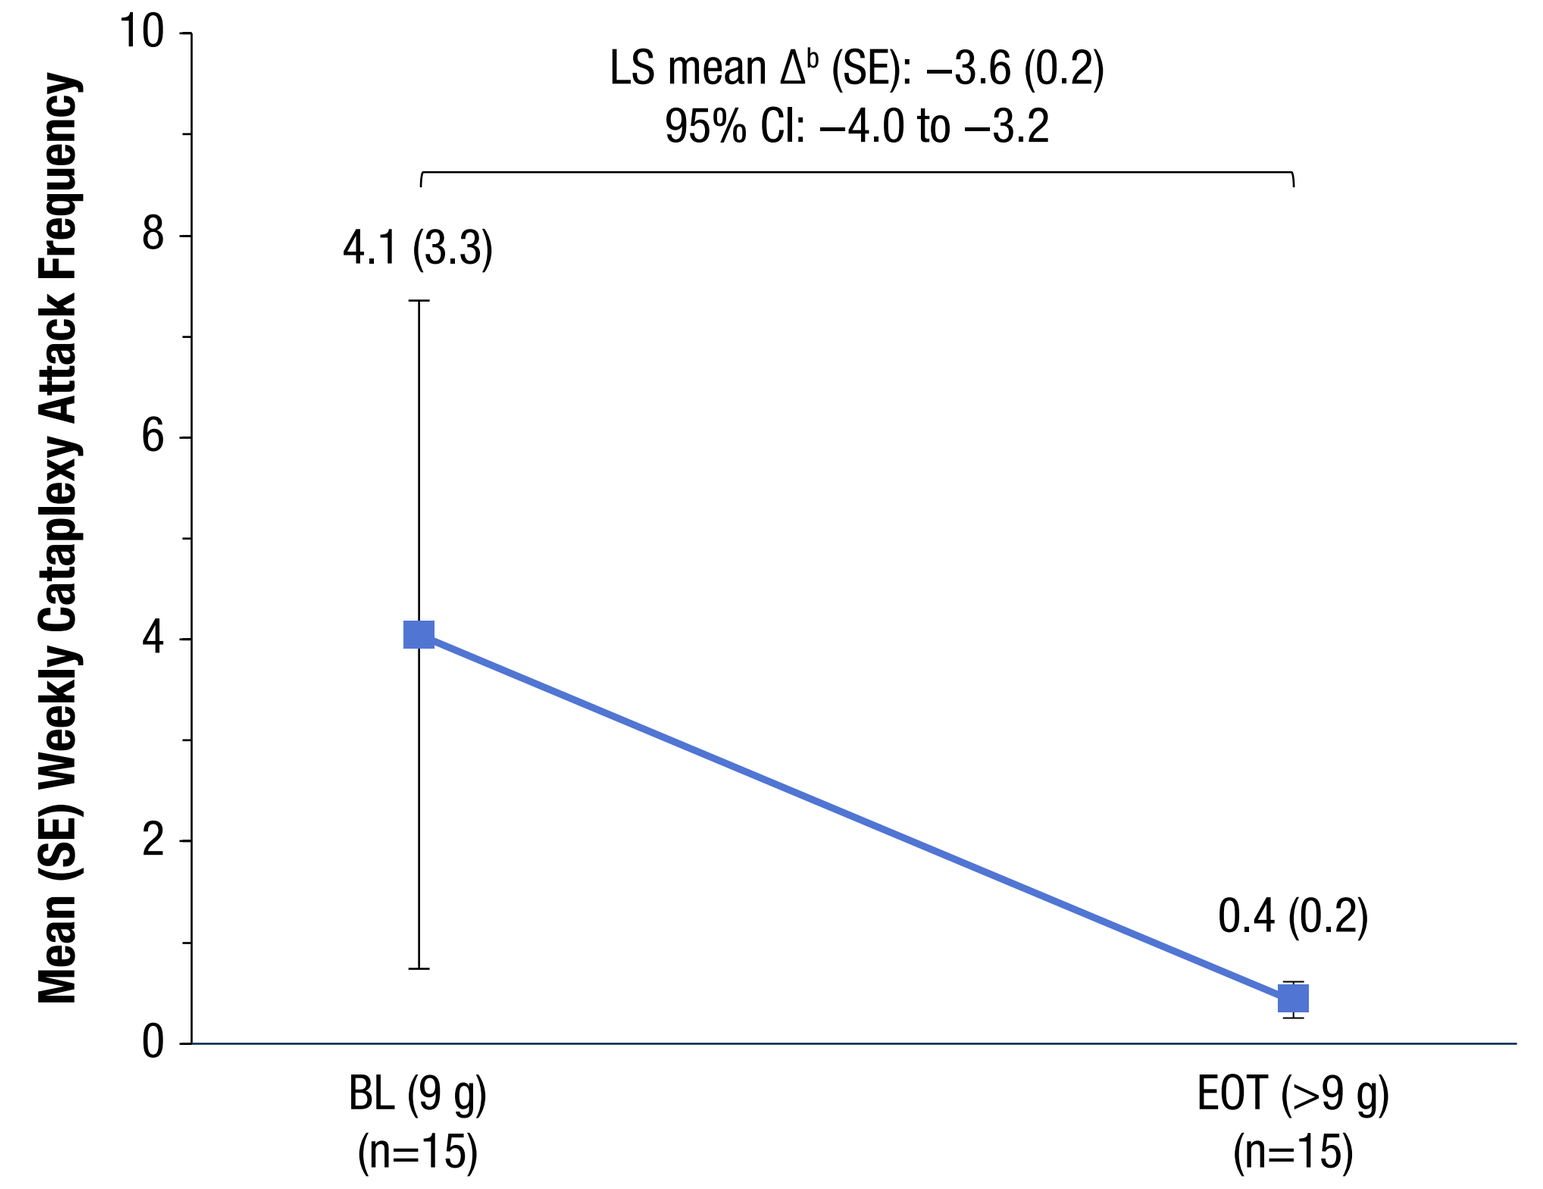
Mean (SE) Weekly Cataplexy Attack Frequency
10
8
6
4
2
0
LS mean Δ[b] (SE): −3.6 (0.2)
95% CI: −4.0 to −3.2
4.1 (3.3)
0.4 (0.2)
BL (9 g) (n=15)
EOT (>9 g) (n=15)

[a]Completer analysis set with 1 participant excluded from the analysis due to being an extreme outlier. [b]Difference between EOT and baseline.
BL, baseline; CI, confidence interval; EOT, end of treatment; LS, least squares; LXB, low-sodium oxybate; NT1, narcolepsy type 1; SE, standard error.

- Median (Q1, Q3) weekly cataplexy attack frequency was 0 (0, 1.0) at BL and 0 (0, 1.0) at EOT

Table 3. Concomitant Alerting Agent Medications[a,b]

ATC Level 4 Term, n (%) Preferred Term, n (%)	Narcolepsy Type 1 (n=30)	Narcolepsy Type 2 (n=18)	Total Narcolepsy Cohort (N=48)
Participants taking a concomitant alerting agent,[c,d] n (%)	16 (53.3)	12 (66.7)	28 (58.3)
Centrally acting sympathomimetics	14 (46.7)	10 (55.6)	24 (50.0)
Amphetamine aspartate, amphetamine sulfate, dexamphetamine saccharate, dexamphetamine sulfate	9 (30.0)	6 (33.3)	15 (31.3)
Solriamfetol hydrochloride	2 (6.7)	2 (11.1)	4 (8.3)
Methylphenidate	3 (10.0)	1 (5.6)	4 (8.3)
Armodafinil	1 (3.3)	1 (5.6)	2 (4.2)
Lisdexamphetamine mesilate	1 (3.3)	1 (5.6)	2 (4.2)
Amphetamine sulfate	0	1 (5.6)	1 (2.1)
Other antidepressants	0	1 (5.6)	1 (2.1)
Bupropion hydrochloride	0	1 (5.6)	1 (2.1)
Other nervous system drugs	5 (16.7)	3 (16.7)	8 (16.7)
Pitolisant hydrochloride	5 (16.7)	3 (16.7)	8 (16.7)

[a]Safety analysis set. [b]Participants could have been taking multiple different alerting medications. [c]It is not known whether these agents were prescribed for excessive sleepiness, narcolepsy, idiopathic hypersomnia, or another condition. [d]Concomitant medications were started prior to the first dose of LXB and were ongoing throughout the study. Concomitant medications for transfer participants were programmatically imputed from the main narcolepsy cohort if the medication was ongoing.
ATC, anatomical therapeutic chemical.

- At study entry, 28 participants (58%) were taking alerting agents

Table 4. Respiratory Assessment Measures Remained Stable From BL to EOT in Participants Taking >9 g/night LXB[a]

Characteristic	Narcolepsy Type 1 (n=28) BL	Narcolepsy Type 1 (n=28) EOT (>9 g)	Narcolepsy Type 2 (n=16) BL	Narcolepsy Type 2 (n=16) EOT (>9 g)	Total Narcolepsy Cohort (N=44) BL	Total Narcolepsy Cohort (N=44) EOT (>9 g)
Apnea-hypopnea index score (full night; events/hour)						
Mean (SE)	2.0 (0.6)	1.6 (0.4)	1.3 (0.2)	1.6 (0.3)	1.8 (0.4)	1.6 (0.3)
LS mean Δ[b] (SE)	−0.2 (0.4)		−0.1 (0.5)		−0.2 (0.3)	
95% CI	−0.9, 0.5		−1.0, 0.8		−0.7, 0.4	
Central apnea events (full night; number)						
Mean (SE)	6.2 (3.8)	5.2 (1.8)	2.8 (0.8)	4.1 (1.5)	5.0 (2.4)	4.8 (1.3)
LS mean Δ[b] (SE)	0.2 (1.6)		−0.7 (2.1)		−0.2 (1.3)	
95% CI	−3.2, 3.5		−5.0, 3.6		−2.8, 2.4	
Mean SpO_2 (full night; %)						
Mean (SE)	95.4 (0.3)	95.6 (0.2)	96.6 (0.3)	96.2 (0.2)	95.8 (0.2)	95.8 (0.1)
LS mean Δ[b] (SE)	−0.1 (0.2)		0.1 (0.2)		0 (0.1)	
95% CI	−0.4, 0.3		−0.3, 0.5		−0.2, 0.2	
Percentage of total sleep time spent with SpO_2 <90% (full night; %)						
Mean (SE)	0.6 (0.3)	0.1 (0.1)	0.1 (0.1)	0.2 (0.2)	0.4 (0.2)	0.1 (0.1)
LS mean Δ[b] (SE)	−0.3 (0.1)		−0.1 (0.1)		−0.3 (0.1)	
95% CI	−0.5, −0.2		−0.4, 0.1		−0.4, −0.1	

[a]Completer analysis set. [b]Difference between EOT and BL.
BL, baseline; CI, confidence interval; EOT, end of treatment; LS, least squares; LXB, low-sodium oxybate; SE, standard error; SpO_2, oxygen saturation.

- No participants taking LXB dosages >9 g/night had AHI >10 during the EOT PSG

Table 5. Treatment-Emergent Adverse Events[a]

TEAEs occurring in ≥5% of participants, n (%)	Narcolepsy Type 1 (n=30)	Narcolepsy Type 2 (n=18)	Total Narcolepsy Cohort (N=48)
Nausea	2 (6.7)[b]	4 (22.2)[c]	6 (12.5)
Vomiting	3 (10.0)[b]	3 (16.7)[b]	6 (12.5)
Headache	3 (10.0)[c]	2 (11.1)[d]	5 (10.4)
Enuresis	4 (13.3)[e]	0	4 (8.3)
Hyperhidrosis	3 (10.0)[b]	1 (5.6)[d]	4 (8.3)
Somnolence	2 (6.7)[d]	2 (11.1)[b]	4 (8.3)
Diarrhea	2 (6.7)	1 (5.6)[d]	3 (6.3)
Fall	2 (6.7)[b]	1 (5.6)	3 (6.3)
Hypotension	2 (6.7)[b]	1 (5.6)[d]	3 (6.3)
Pollakiuria	2 (6.7)[b]	1 (5.6)[d]	3 (6.3)
Upper respiratory tract infection	2 (6.7)	1 (5.6)	3 (6.3)

[a]Safety analysis set. [b]Considered treatment-related, n=2. [c]Considered treatment-related, n=3. [d]Considered treatment-related, n=1. [e]Considered treatment-related, n=4.
TEAE, treatment-emergent adverse event.

- 75% (36/48) of participants reported a TEAE
- TEAEs were mostly mild or moderate in severity
 - One serious TEAE was reported (NT2, syncope)
 - Three severe TEAEs occurred (NT1, hypnagogic hallucinations; NT2, nausea, syncope)
 - Three TEAEs led to discontinuation (NT1, fall, anxiety; NT2, nausea)
- One participant with NT1 reported suicidal ideation on the C-SSRS during the titration period (day 35; depressive symptoms related to election), which resolved by the next visit; no suicidal ideation was present for the rest of the study

Conclusions

- Participants with narcolepsy treated with an LXB dosage >9 g/night (mean [SD] 11.1 [1.0] g/night) experienced additional symptom benefit compared with treatment with 9 g/night (at baseline), with reductions in ESS scores, NSS scores, and frequency of cataplexy attacks
- Limitations include the open-label design and lack of a control cohort, which limits the ability to causally attribute findings to LXB
- No respiratory safety signals were observed in this study with LXB treatment at dosages >9 g/night
- Overall, TEAEs were consistent with the known safety profile of LXB at dosages ≤9 g/night, supporting individualized clinical decision-making

References: **1.** Xywav® (calcium, magnesium, potassium, and sodium oxybates) oral solution, CIII [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **2.** Szarfman A, et al. *N Engl J Med.* 1995;333(19):1291. **3.** US Food and Drug Administration. Clinical review for Binosto, NDA 202344. 2012. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2012/202344Orig1s000MedR.pdf. **4.** US Food and Drug Administration. Quantitative labeling of sodium, potassium, and phosphorus for human over-the-counter and prescription drug products. Guidance for industry. 2022. https://www.fda.gov/regulatory-information/search-fda-guidance-documents/quantitative-labeling-sodium-potassium-and-phosphorus-human-over-counter-and-prescription-drug. **5.** Ohayon MM, et al. Presented at: Annual Scientific Meeting of the Associated Professional Sleep Societies; June 8-12, 2019; San Antonio, TX. **6.** Berry RB, et al. *The AASM Manual for the Scoring of Sleep and Associated Events: Rules, Terminology and Technical Specifications, Version 3.* Darien, IL: American Academy of Sleep Medicine; 2023. **7.** Johns MW. *Sleep.* 1991;14(6):540-545. **8.** Maski K, et al. *J Clin Sleep Med.* 2021;17(9):1895-1945. **9.** Dauvilliers Y, et al. *Sleep.* 2020;43(6):zsaa009. **10.** Barateau L, et al. *Sleep.* 2024;47(5):zsad323.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Under the direction of the authors, Peloton Advantage, LLC (an OPEN Health company), employees Hannah Ritchie, PhD, and Stephanie Phan, PharmD, provided medical writing support and an editor provided editorial support for this poster, which were funded by Jazz Pharmaceuticals.

Disclosures: **JH Simmons** has received grant funding from Jazz Pharmaceuticals, Eisai, Alkermes, Takeda, and Harmony Biosciences and speaker honorarium from Avadel. **LD Schneider** has received personal compensation for serving on advisory boards and speakers bureaus for Avadel, Eisai, and Jazz Pharmaceuticals. **CM Ruoff** has served as an advisory board member for Alkermes, Eisai, Jazz Pharmaceuticals, and Takeda, and has received grant funding from Jazz Pharmaceuticals. **DT Plante** is a consultant and advisory board member for Jazz Pharmaceuticals. He has also served as an advisory board member for Apnimed; consultant/advisory board member for Alkermes, Centessa, Harmony Biosciences, and Takeda; and a consultant for Aditum Bio and Teva Pharmaceuticals (Australia). **DA Nichols, M Whalen, MT Kirby,** and **J Dai** are full-time employees of Jazz Pharmaceuticals who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **TL Steininger** is a former full-time employee and current contract worker for Jazz Pharmaceuticals who has received shares of Jazz Pharmaceuticals, plc. **A Cairns** is a full-time contract employee and advisory board member for Jazz Pharmaceuticals. **RK Bogan** is a shareholder of Watermark Medical and Healthy Humming; serves on the board of directors for Watermark; is a medical consultant to Alkermes, Avadel, Harmony Biosciences, Jazz Pharmaceuticals, Oventus, and Takeda; has conducted industry-funded research for Avadel, Axsome, Balance, Bayer, Bresotec, Eisai, Fresca, Idorsia, Jazz Pharmaceuticals, LivaNova, Merck, NLS, Noctrix, Philips, Roche, Sanofi, Sommetrics, Suven, Takeda, and Vanda; and is on speakers bureaus for Axsome, Eisai, Harmony Biosciences, and Jazz Pharmaceuticals.

Scan this code to access this poster online.
This code is not for promotional purposes.

Presented at Psych Congress 2025; September 17–21, 2025; San Diego, CA, USA