Response to Dordaviprone (ONC201) by Prior Radiotherapy Regimen in Recurrent H3 K27M-Mutant Diffuse Midline Glioma

Isabel Arrillaga-Romany[1], Susan L. McGovern[2], Joshua E. Allen[3], Sharon L. Gardner[4], Aya M. Haggiagi[5], Yazmin Odia[6], Pablo Lee[3], Samuel C. Ramage[3], Patrick Y. Wen[7]

[1]Massachusetts General Hospital, Boston, MA, USA; [2]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [3]Chimerix, Inc, Durham, NC; [4]New York University, Grossman School of Medicine, New York, NY, USA; [5]Columbia University Irving Medical Center, New York, NY, USA; [6]Miami Cancer Institute, part of Baptist Health South Florida, Miami, FL, USA; [7]Dana-Farber/Brigham and Women's Cancer Center, Boston, MA, USA

Introduction

- The H3 K27M mutation is relatively common in diffuse midline glioma (DMG), occurring in up to 80% of pediatric patients and 15-58% of adult patients.[1-3]
- Survival is exceptionally poor; median overall survival is 13.4 months (95%CI, 10.5-15.5), with no substantial difference in 1-year OS when evaluated by age at diagnosis. In contrast, the median OS of patients with H3 wild-type tumors is 4.59 years.[4-5]
- In 2016, the World Health Organization classified H3 K27M-mutant DMG as a distinct form of Grade IV glioma, regardless of histological features.[6]
- Dordaviprone (ONC201) is an oral, blood-brain barrier penetrating, selective small molecule antagonist of dopamine receptor D2 and agonist of the mitochondrial protease caseinolytic mitochondrial matrix peptidase proteolytic subunit.[7-11]
- An integrated analysis of adult (n=46) and pediatric (n=4) patients with recurrent H3 K27M-mutant DMG who received dordaviprone monotherapy in one of five open-label studies was previously conducted (**Table 1 & Figure 1**).[12]
 - Grade 3 treatment-related treatment-emergent adverse events occurred in 20.0% of patients; the most common was fatigue (n=5; 10%); no Grade 4 TR-TEAEs, deaths, or discontinuations occurred.

Table 1. Dordaviprone ORR in Patients with Recurrent H3 K27M DMG

	Total Population (N=50)		
	RANO-HGG[1]	RANO-LGG[2]	Combined HGG/LGG[3]
Overall response rate, n (%) [95%CI]	**10 (20) [10–34]**	**13 (26) [15–40]**	**15 (30) [18–45]**
Complete response, n (%)	1 (2)	0	1 (2)
Partial response, n (%)	9 (18)	6 (12)	9 (18)
Minor response, n (%)	NA	7 (14)	5 (10)
Stable disease, n (%)	10 (20)	8 (16)	7 (14)
Not evaluable, n (%)	8 (16)[4]	11 (22)[5]	11 (22)[5]
Progressive disease, n (%)	18 (36)	14 (28)	13 (26)
Non-applicable, n (%)	4 (8)[6]	4 (8)[6]	4 (8)[6]
Disease control rate, n (%) [95%CI]	**20 (40) [26–55]**	**21 (42) [28–57]**	**22 (44) [30–59]**

[1]Integrated RANO-HGG criteria assessment by dual reader BICR.
[2]Integrated RANO-LGG criteria assessment by dual reader BICR.
[3]Incorporates the best response by RANO-HGG or -LGG criteria for each patient.
[4]Five overall radiographic stable disease accompanied by increase in corticosteroids; three overall radiographic progressive disease accompanied by decrease in corticosteroids.
[5]Eight overall radiographic stable disease accompanied by increase in corticosteroids; three overall radiographic progressive disease accompanied by decrease in corticosteroids.
[6]Three patients did not have on-treatment monotherapy MRIs available for BICR; one patient censored prior to first on-treatment MRI. BICR, blind independent central review.
HGG, high grade glioma; LGG, low grade glioma; RANO, response assessment in neuro-oncology.

Figure 1. Waterfall Plot of Dordaviprone-treated Patients with Recurrent H3 K27M-mutant DMG (RANO-HGG)

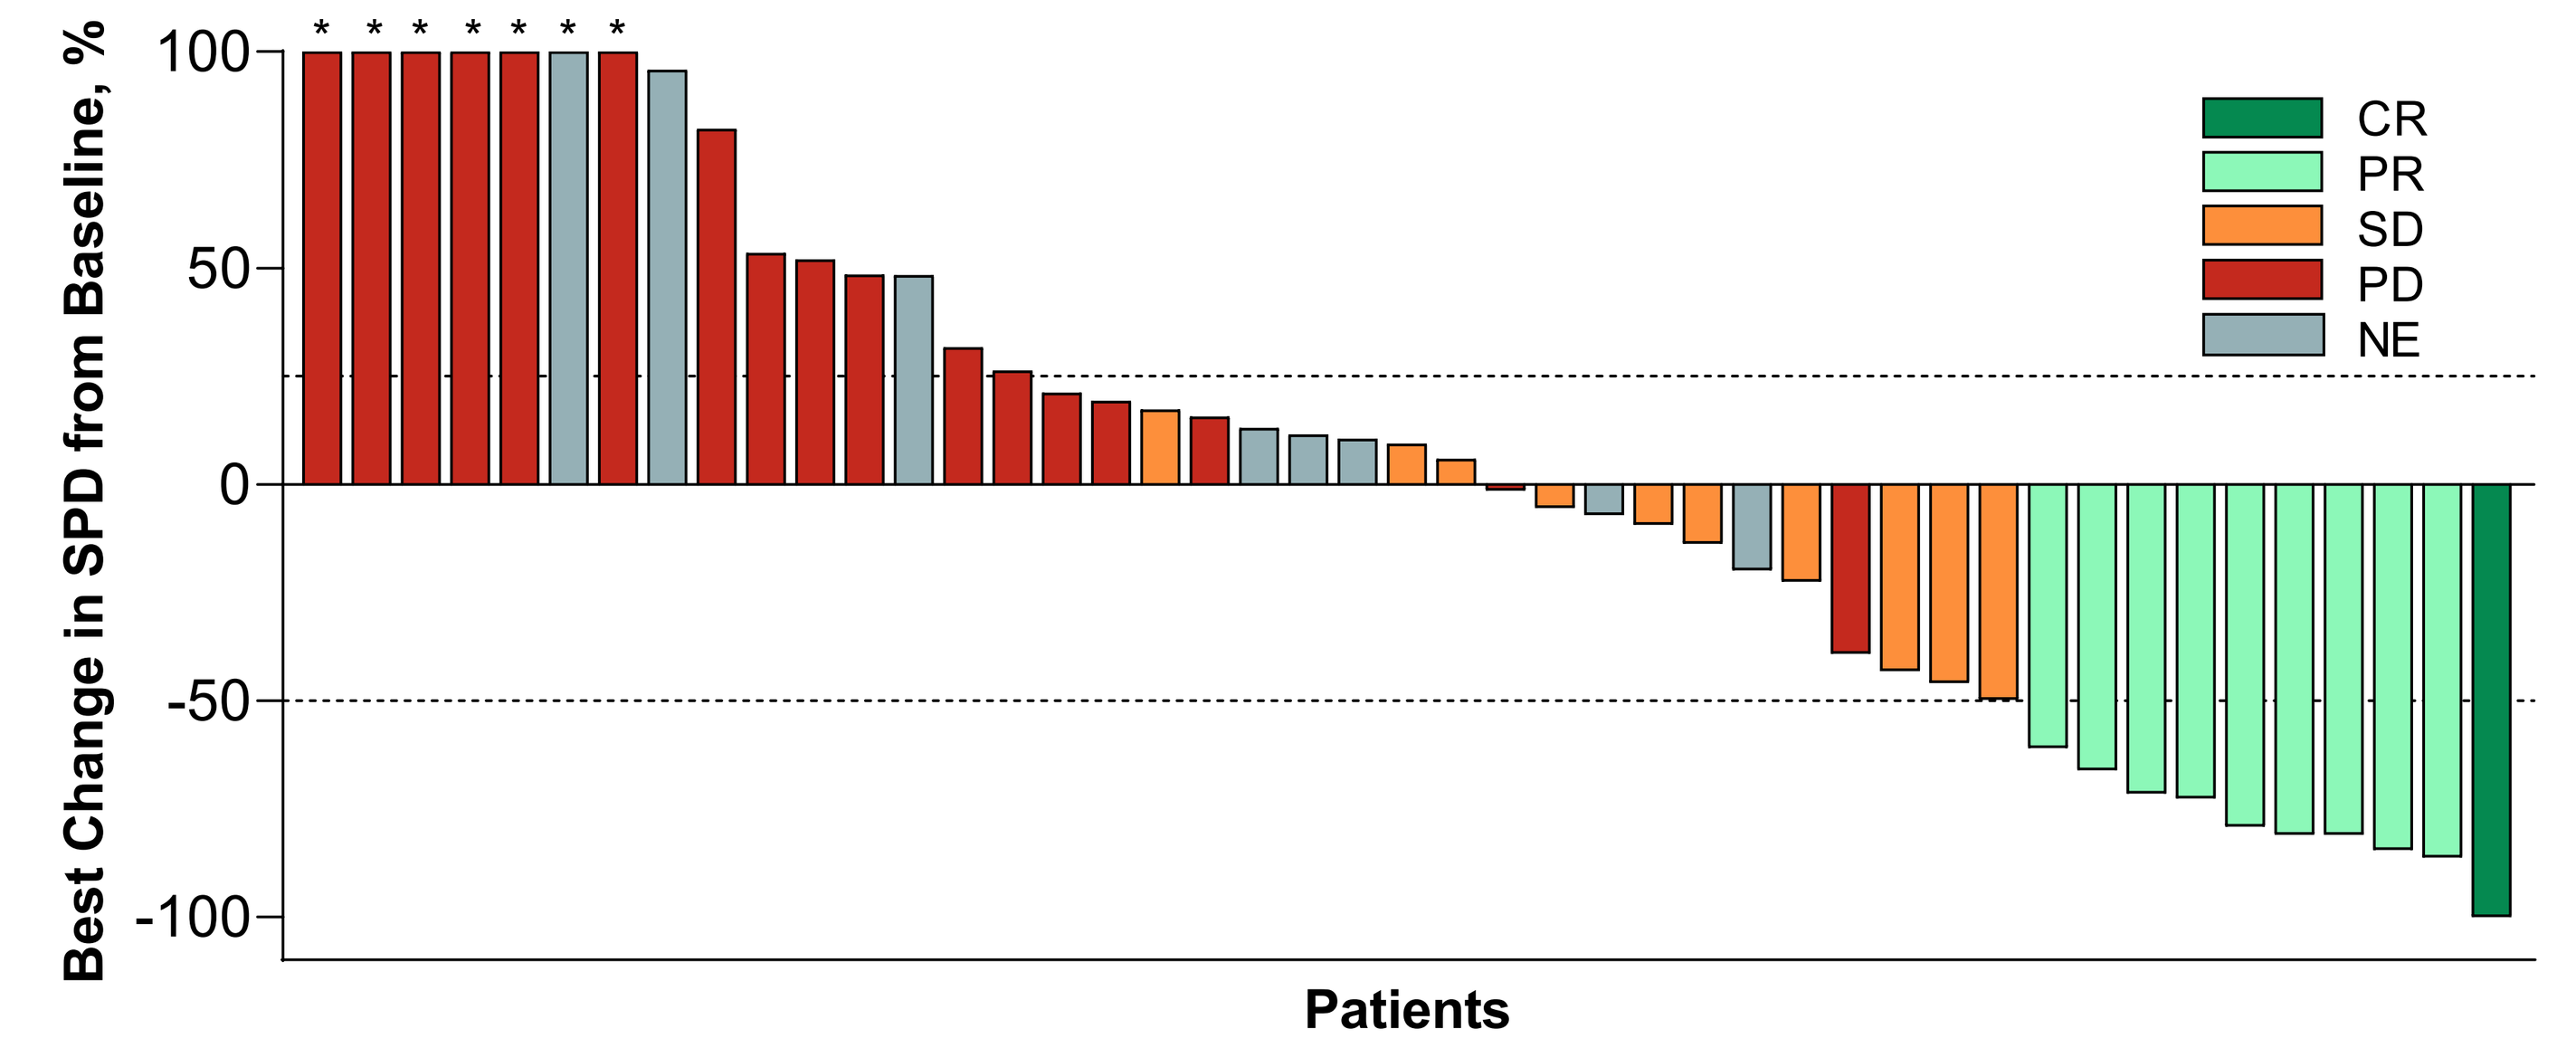
Best Change in SPD from Baseline, %
100
50
0
-50
-100
Patients
CR
PR
SD
PD
NE

CR, complete response; DMG, diffuse midline glioma; DOR, duration of response – defined as from onset of response to progression, defined ; NE, not evaluable; NR, not reached; PD, progressive disease; PR, partial response; RANO–HGG, Response Assessment in Neuro-Oncology–High Grade Glioma; SD, stable disease; SPD, sum product of diameters; TTR, time to response.

- **Corticosteroid response** (≥50% reduction in average daily corticosteroid dose compared to baseline with stable or improved performance score): 46.7% (7/15; 95%CI, 21.3-73.4)[12]
- **Performance score response** (increase in KPS/LPS compared to baseline with stable or reduced corticosteroid use): 20.6% (7/34; 95%CI, 8.7-37.9)[12]
- This sub-analysis of the integrated analysis population was conducted to determine the efficacy of dordaviprone monotherapy in patients with recurrent, H3 K27M-mutant DMG by prior front-line radiotherapy regimen.

Response to ONC201 monotherapy among patients with recurrent H3 K27M-mutant DMG was achieved in patients with diverse front-line radiotherapy regimens.

Scan here to download the poster

Methods

- The treatment regimen and pooled analysis design for this population has previously been reported.[12] Briefly, this integrated analysis included 50 patients (pediatric, n=4; adult, n=46) with recurrent H3 K27M–mutant DMG who received oral dordaviprone monotherapy in four clinical trials or one expanded access protocol.
- Eligible patients had measurable disease by Response Assessment in Neuro-Oncology (RANO) high-grade glioma (HGG) criteria and performance score (PS) ≥60 and were ≥90 days from radiation; pontine and spinal tumors were ineligible
- The primary endpoint was overall response rate (ORR) by RANO-HGG criteria.
- Radiographic end points were assessed by dual-reader, blinded independent central review.

Results

- Most patients received concurrent chemoradiotherapy (CCRT) + adjuvant (70.0%; **Table 2**).
 - Among patients who previously received adjuvant and/or CCRT, most had received temozolomide (92.8%).
 - Most patients received a standard radiotherapy dose (54-60 Gy) over six weeks.
- Fifteen patients had an objective response by combined RANO-HGG/LGG criteria (**Table 1**).
 - Responses occurred in patients who had previously received CCRT+adjuvant, CCRT, and RT+adjuvant (**Table 3**).
- At least two responders had received proton therapy.
 - The first was an adult patient who received CCRT + Adjuvant (dasatinib) and had a PR by both RANO-HGG and -LGG criteria.
 - The second was a pediatric patient who received RT + Adjuvant (dasatinib, bevacizumab, and everolimus) and had a PR by both RANO-HGG and -LGG criteria.

Results

Table 2. Front-line Radiotherapy Regimen and Dose (All Patients)

	Pediatric Patients (n=4)	Adult Patient (n=46)	All Patients (N=50)
Radiotherapy Dosage, n (%)[a]			
Patients with data available, n	4	38	42
<54 Gy	0	2 (5.2)	2 (4.8)
≥54 Gy	4 (100.0)	36 (94.7)	40 (95.2)
Regimen Received, n (%)			
CCRT + Adjuvant	1 (25.0)	34 (73.9)	35 (70.0)
CCRT	0	8 (17.4)	8 (16.0)
RT + Adjuvant	1 (25.0)	3 (6.5)	4 (8.0)
RT Alone	2 (50.0)	1 (2.2)	3 (6.0)
Prior temozolomide, n^1/n^2 (%)	0/2	43/45 (95.6)	43/47 (91.2)

[a]Percent shown out of total number of patients with dosage data available.
[b]Patient received hypofractionated therapy.
CCRT, concurrent chemoradiotherapy; n^1, total number of patients who received prior temozolomide; n^2, total number of patients who received CCRT and/or adjuvant; RT, radiotherapy.

References

1. Lulla RR, et al. *Sci Adv*. 2016;2(3):e1501354.
2. Shereck EB, et al. *Pediatr Blood Cancer.* 2007;49(3):306-12.
3. Qiu FZ, et al. *Virol J*. 2018;15(1):81.
4. Khuong-Quang DA, et al. *Acta Neuropathol*. 2012;124(3):439-47.
5. Ostrom, et al. *Neuro-Oncol.* 2023;25:799-807.
7. Louis DN, et al. *Acta Neuropathol*. 2016;131(6):803-20.
7. Allen JE, et al. *Sci Transl Med.* 2013;5(171):171ra17.
8. Free RB, et al. *Mol Pharmacol.* 2021;100(4):372-387.
9. Madhukar NS, et al. *Nat Commun.* 2019;10(1):5221.
10. Ishizawa J, et al. *Cancer Cell.* 2019;35(5):721-737 e9.
11. Graves PR, et al. *ACS Chem Biol.* 2019;14(5):1020-1029.
12. Arrillaga-Romany I, et al. *J Clin Oncol.* 2024;42(13):1542-1552.

Results

Table 3. Response by Front-line Radiation Regimen[a]

	HGG Criteria		LGG Criteria		HGG/LGG Criteria	
	Responder (n=10)	Non Responder (n=40)	Responder (n=13)	Non Responder (n=37)	Responder (n=15)	Non Responder (n=35)
Regimen Received, n (%)						
CCRT + Adjuvant	7 (70.0)	28 (70.0)	7 (53.8)	28 (75.6)	9 (60.0)	26 (74.3)
CCRT	2 (20.0)	6 (15.0)	3 (23.1)	5 (13.5)	3 (20.0)	5 (14.3)
RT + Adjuvant	1 (10.0)	3 (7.5)	3 (23.1)	1 (2.7)	3 (20.0)	1 (2.9)
RT Alone	0	3 (7.5)	0	3 (8.0)	0	3 (8.6)
Prior temozolomide, n^1/n^2 (%)	8/10 (80.0)	29/37 (78.4)	10/13 (76.9)	27/34 (79.4)	12/15 (80.0)	25/32 (78.1)

[a]Non-responders were patients who did not achieve a MR, PR, or CR. For combined HGG/LGG, non-responders did not have a MR, PR, or CR by both HGG and LGG criteria.
[b]N indicates number of patients who had received a front-line regimen including adjuvant and/or CCRT.
CCRT, concurrent chemoradiotherapy; HGG, high-grade glioma; LGG, low-grade glioma; n^1, total number of patients who received prior temozolomide; n^2, total number of patients who received CCRT and/or adjuvant; NA, non-applicable; NE, non-evaluable; MR, minor response; PD, progressive disease; RT, radiotherapy; SD, stable disease.

- Duration of response appeared similar for patients who had received front-line CCRT, CCRT+Adjuvant, and RT+Adjuvant regimens (**Figure 2**).

Figure 2. Duration of Response by Front-line Radiotherapy Regimen Among Responders by (A) RANO-HGG and (B) RANO-LGG Criteria

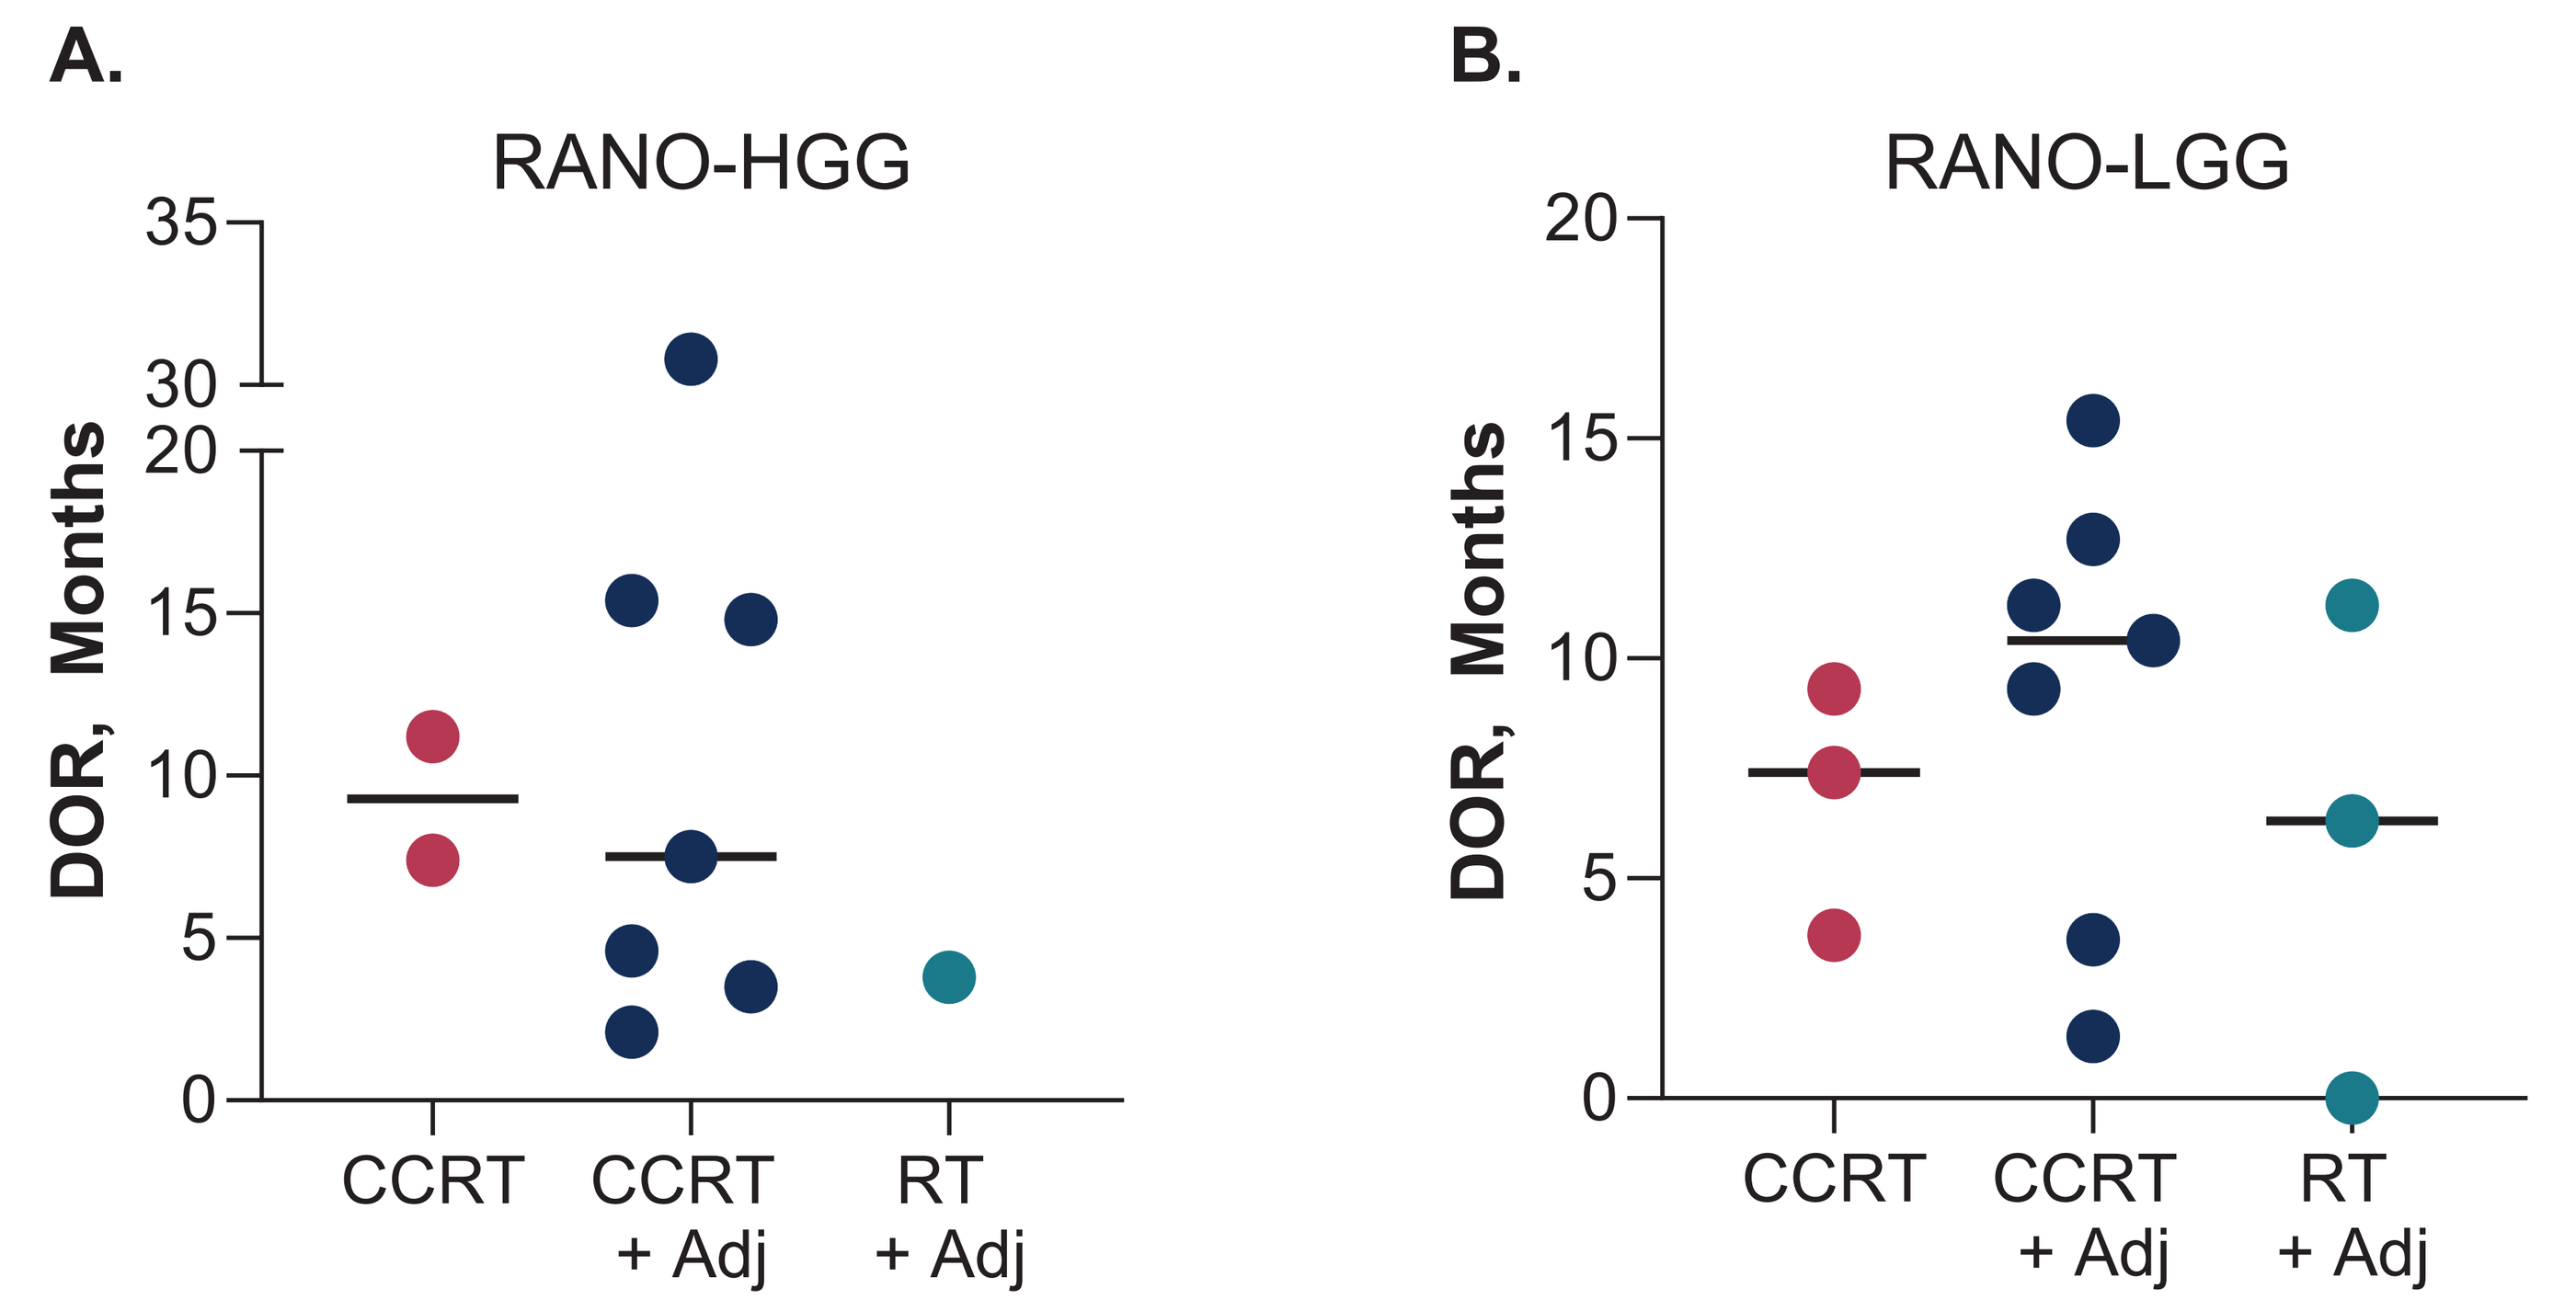
A.
RANO-HGG
DOR, Months
35
30
20
15
10
5
0
CCRT
CCRT + Adj
RT + Adj
B.
RANO-LGG
DOR, Months
20
15
10
5
0
CCRT
CCRT + Adj
RT + Adj

Adj, adjuvant; CCRT, concurrent chemoradiotherapy; DOR, duration of response; HGG, high-grade glioma; LGG, low-grade glioma; RANO, Response Assessment in Neuro-Oncology; RT, radiotherapy.

Conclusions

- Radiographic responses to dordaviprone in patients with H3 K27M-mutant DMG were observed across various front-line radiotherapy regimens.
- Responses to dordaviprone were observed both in patients receiving front-line photon and proton radiation therapy in both pediatric and adult patients.
- This analysis has limitations, as data on prior front-line radiotherapy treatment modality, dose, and duration were incomplete for some patients.
- The phase 3 ACTION trial (NCT05580562) is currently evaluating dordaviprone in newly diagnosed H3 K27M-mutant diffuse glioma following standard front-line radiotherapy; patients previously treated with temozolomide are eligible provided they have sufficient washout (3 weeks).

Conflicts of Interest

IA-R and **AMH** have received research funding from Chimerix, Inc. **JEA**, **PL**, and **SCR** are employees of and have stock ownership in Chimerix, Inc. **PYW** is a consultant for and has received research funding from Chimerix, Inc. **SLM** has received speakers fees from Chimerix, Inc. **YO** and **SLG** have no relevant COI to declare.

Acknowledgments

We would like to express our sincere gratitude to Sharon L. Garner, MD (recently retired from NYU Grossman School of Medicine), whose significant contributions to data collection and analysis have been invaluable to this work. Medical writing and editorial assistance was provided by Meghan Sullivan, PhD, CMPP, an employee of Chimerix, Inc.

American Society for Radiation Oncology (ASTRO) 2024 Annual Meeting; September 29-October 1, 2024; Washington, DC