The Effect of Dordaviprone (ONC201) on Cardiac Repolarization in Healthy Adult Participants

Shamia L. Faison[1], Joelle Batonga[1], Maggie Anderson[2], Thangam Arumugham[2], Angela Bartkus[2], Marion Morrison[2], Mark J Mullin[2], Tim Tippin[2], Robert Kleiman[3], Odin Naderer[2]

[1]Certara Strategic Consulting; [2]Chimerix, Inc., a Subsidiary of Jazz Pharmaceuticals; [3]Clario

Introduction

- Dordaviprone (ONC201) is a novel, orally administered, anti-cancer imipridone with demonstrated antitumor effects in patients with glioma,[1-2] and is currently being evaluated in a Phase III clinical trial.[3]
- As dordaviprone is a novel compound, it was evaluated for potential effects on heart rate corrected QT duration (QTc) compared to a positive (moxifloxacin) and negative (placebo) control.

Methods

- This was a single-center, randomized, partially blinded, positive- and placebo-controlled, 3-period crossover study (**Figure 1**)
- Thirty (30) healthy adult participants received a single-dose of 750 mg dordaviprone, 750 mg matched-placebo, or 400 mg moxifloxacin (not blinded) per treatment period according to their randomized sequence.
- Time-matched PK blood draws and replicate 12-lead ECGs via holter were collected at predose and 0.25, 0.5, 0.75, 1, 2, 3, 4, 5, 6, 8, 12, 16, and 24 hours following treatment.
- ECG extractions occurred prior to PK sampling to avoid changes in autonomic tone.
- ECG extractions were analyzed by a blinded central cardiologist.
- A repeated measures mixed model analysis of the change from baseline in QT interval corrected for heart rate by Fridericia's formula (QTcF) was used to estimate the difference in mean change from baseline between dordaviprone and placebo; a similar analysis for moxifloxacin was used for assay sensitivity.

Results

- Thirty (30) participants were included in the statistical analysis (**Table 1**).
 - Data for one participant during the dordaviprone treatment period and two participants during the placebo treatment period were excluded from the analysis. However, data for these participants were included for the other treatment periods.

Cardiac Repolarization

- The placebo-corrected mean change from baseline in QTcF for dordaviprone from 0.75 to 8 hr post dose ranged from 9.2 to 13.8 msec with a peak effect of 13.8 msec occurring at 5 hrs postdose. The upper bounds of the 90%CIs ranged from 11.78 to 16.48 msec for these time points (**Table 2**)
- Moxifloxacin demonstrated a 10.4 msec (90% CI: 8.66 to 12.16) peak increase in QT interval at 3 hrs postdose, demonstrating assay sensitivity (**Table 2**)

Safety

- Dordaviprone treatment-related AEs occurred in 37% of dordaviprone recipients (**Table 3**).
- The most common dordaviprone treatment-related AEs were mild dizziness in five participants (17%) and mild nausea in two participants (7%); no other dordaviprone treatment-related AEs occurred in more than one participant.
- One participant experienced a serious event of deep vein thrombosis (Grade 2), which was considered possibly related to dordaviprone.

Following dordaviprone administration, the mean placebo-corrected change from baseline QTcF was modest (<15 msec). Concomitant use of dordaviprone with drugs known to prolong QT interval should be done with caution.

Figure 2. Placebo-corrected change from baseline QTcF (ΔΔQTcF) across time points with statistical modeling

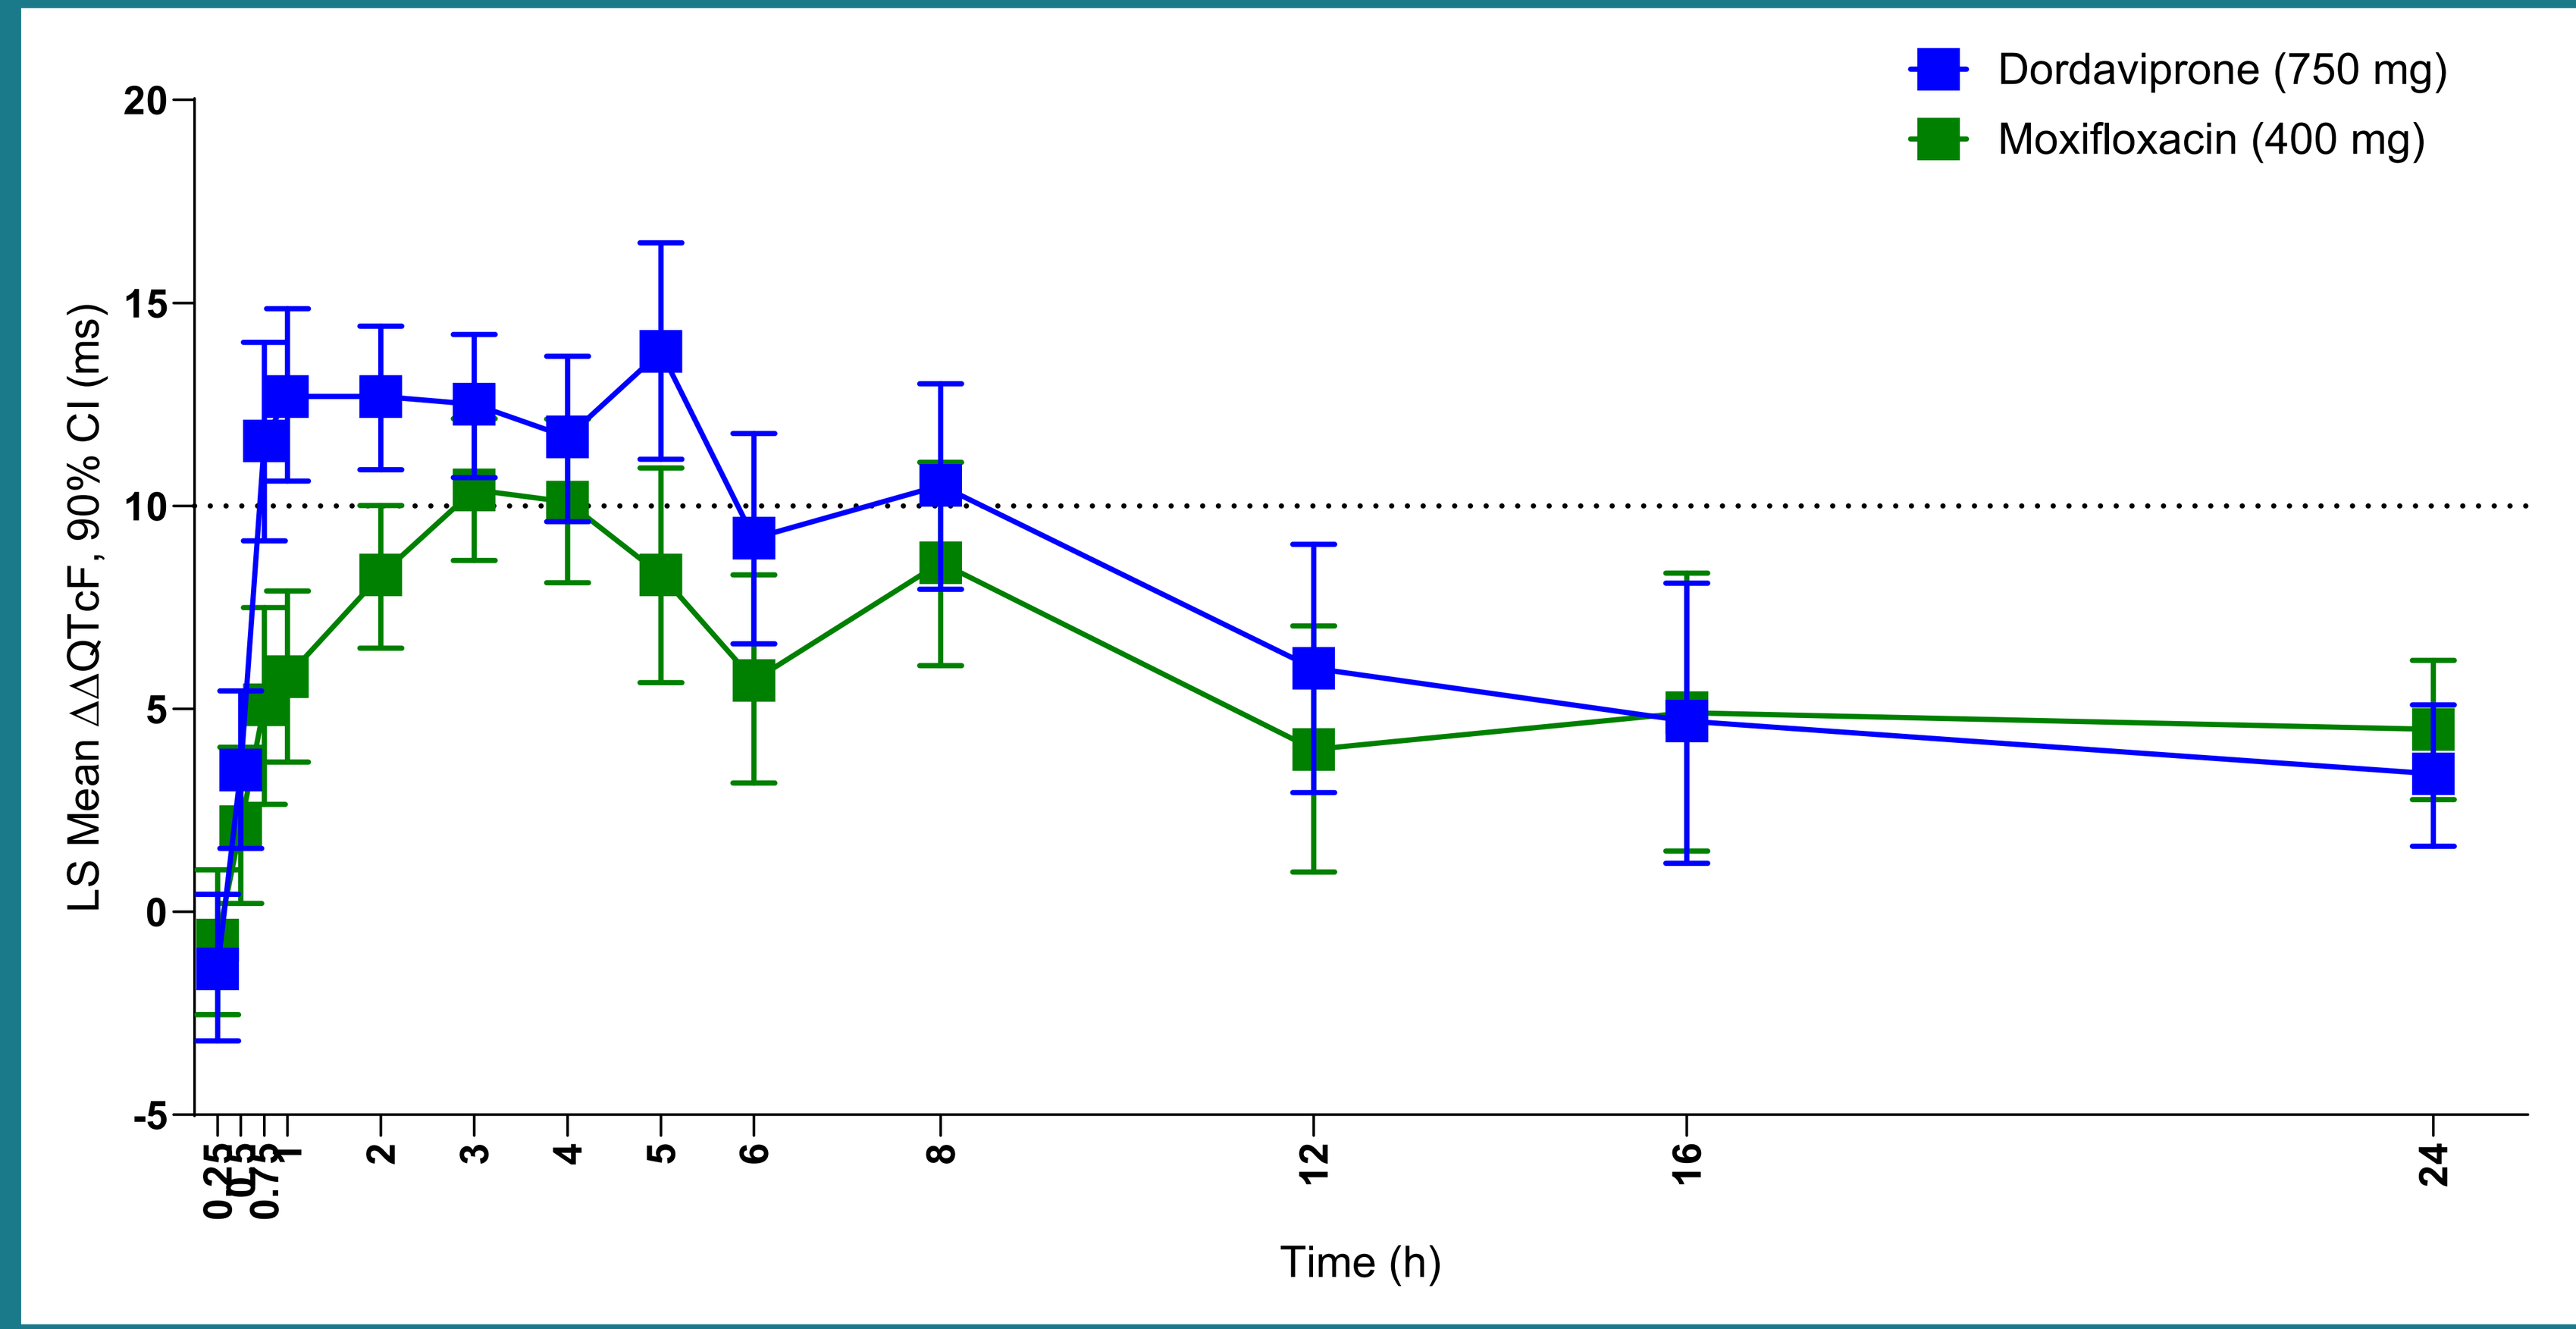
Dordaviprone (750 mg)
Moxifloxacin (400 mg)
LS Mean ΔΔQTcF, 90% CI (ms)
20
15
10
5
0
-5
0.25
0.5
0.75
1
2
3
4
5
6
8
12
16
24
Time (h)

LS mean ΔΔQTcF and 90% CI based on a linear mixed-effects model: ΔQTcF = Baseline QTcF + Treatment + Time + Treatment×Time + Period + Sequence. An unstructured covariance structure was used to specify the repeated measures (time within participant and period), and an unstructured covariance structure was used to specify the participant-specific random effect.
Median dordaviprone Tmax, 0.75 hrs (range, 0.50-3.00); median ONC207 Tmax, 3.00 hrs (0.50-8.00); median moxifloxacin Tmax, 2.00 hrs (0.50, 6.00).
Number of participants included in statistical analysis, N=30. Data for one participant during dordaviprone treatment period was excluded from the analysis while data for the same participant during placebo treatment period was included.

Scan here to download the poster.

Figure 1. Study design

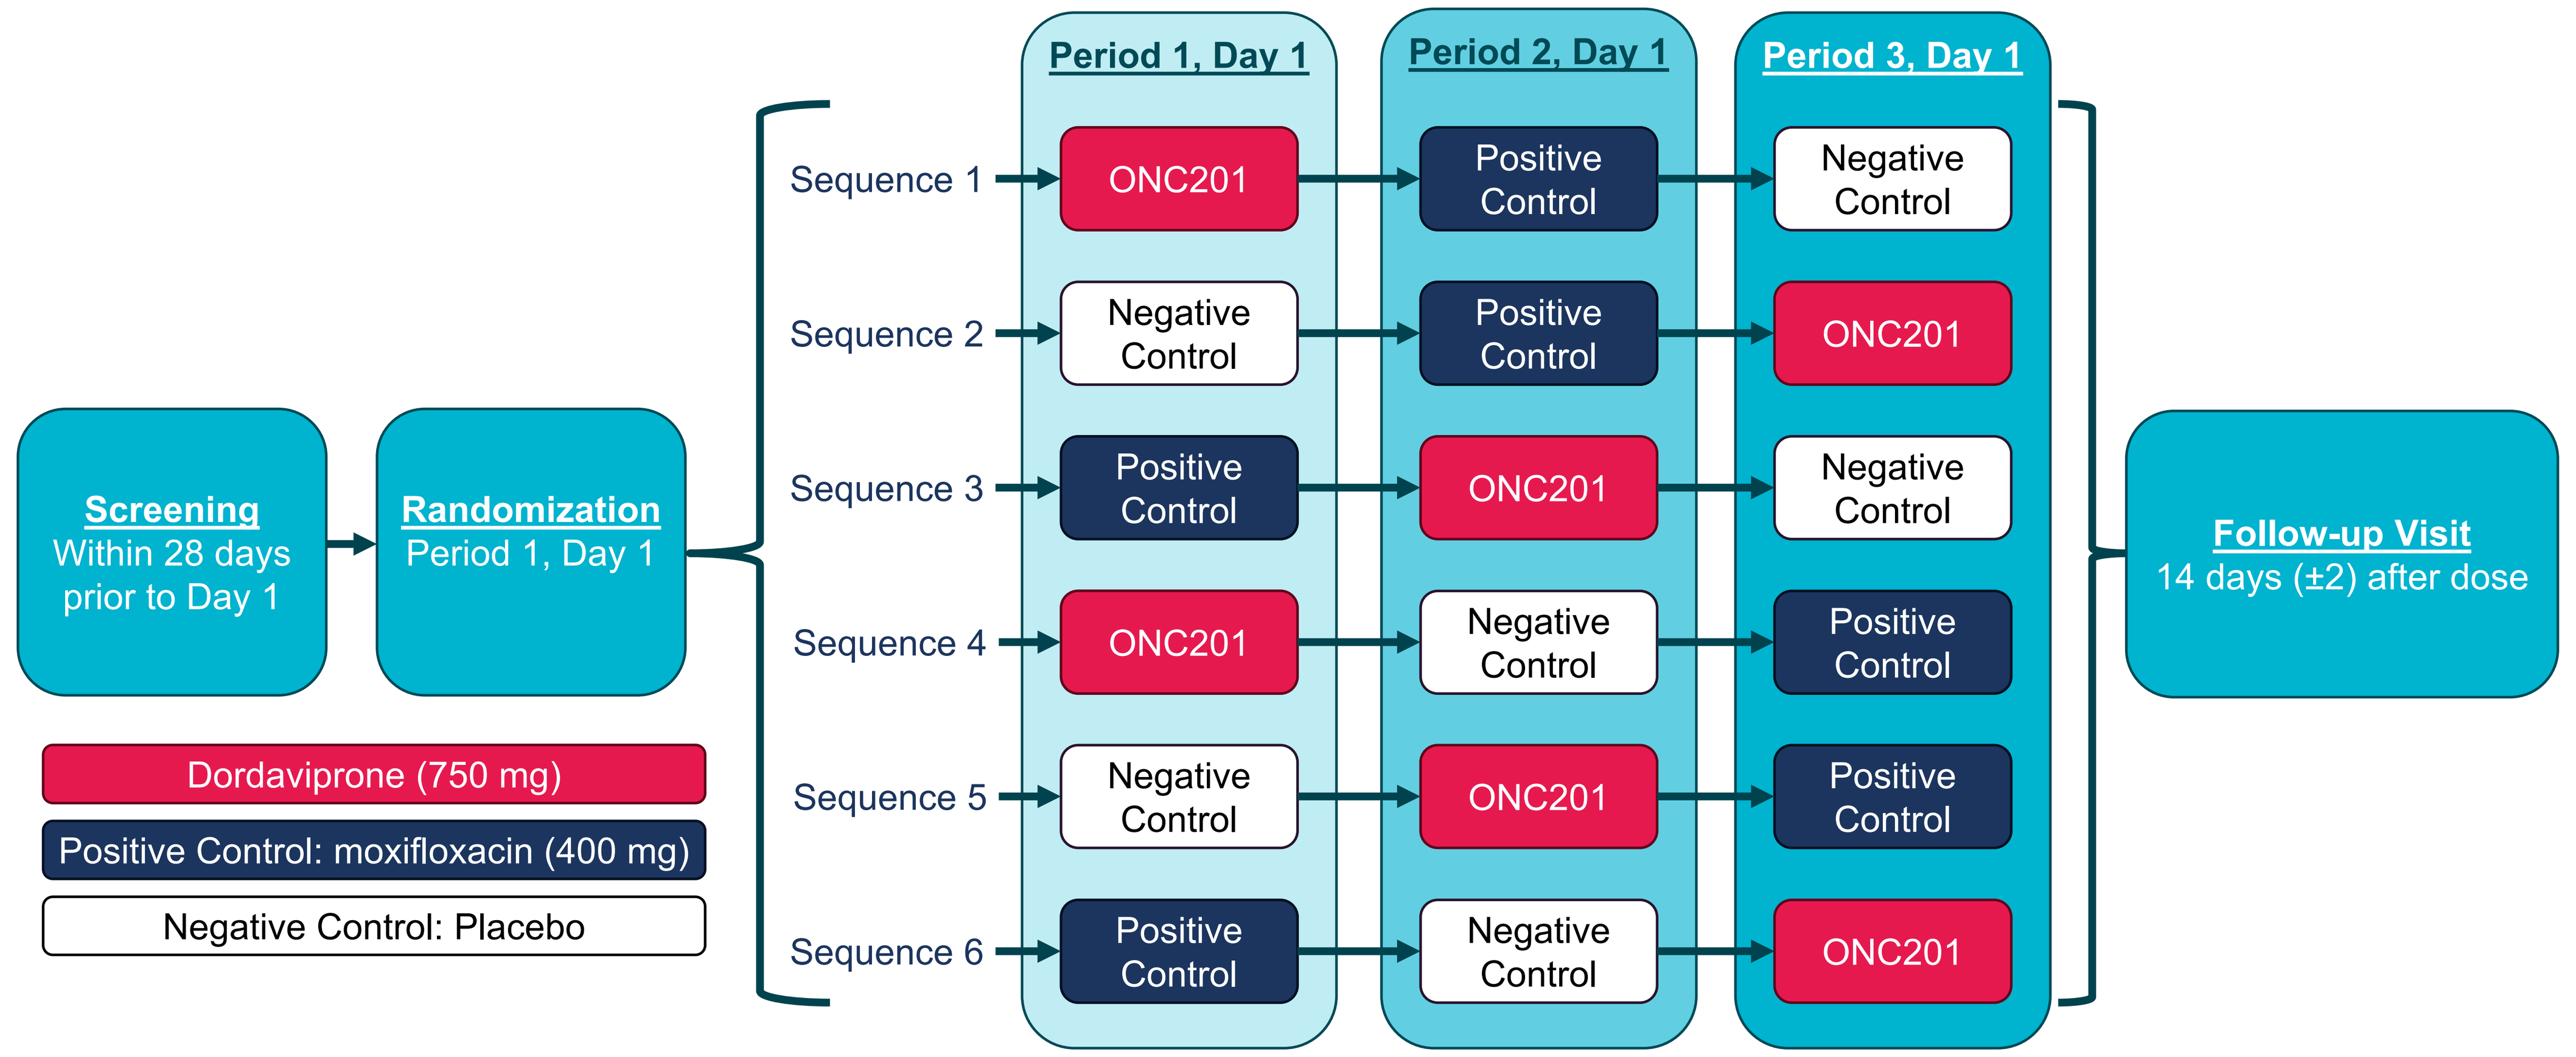
Screening
Within 28 days prior to Day 1
Randomization
Period 1, Day 1
Period 1, Day 1
Period 2, Day 1
Period 3, Day 1
Sequence 1
ONC201
Positive Control
Negative Control
Sequence 2
Negative Control
Positive Control
ONC201
Sequence 3
Positive Control
ONC201
Negative Control
Sequence 4
ONC201
Negative Control
Positive Control
Sequence 5
Negative Control
ONC201
Positive Control
Sequence 6
Positive Control
Negative Control
ONC201
Follow-up Visit
14 days (±2) after dose
Dordaviprone (750 mg)
Positive Control: moxifloxacin (400 mg)
Negative Control: Placebo

Treatments were separated by at least a 7-day washout period.

Table 1. Patient disposition

n, (%)	Seq 1	Seq 2	Seq 3	Seq 4	Seq 5	Seq 6	Overall
Randomized	5	5	5	5	5	5	30
Participants Dosed	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	30 (100.0)
Placebo	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	30 (100.0)
Positive Control	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	30 (100.0)
Dordaviprone	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	30 (100.0)
Completed Study	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	5 (100.0)	30 (100.0)
Discontinued Study	0	0	0	0	0	0	0

Positive control, moxifloxacin (400 mg); dordaviprone (750 mg).

Table 3. Treatment-emergent adverse events

	Treatment			Overall (N=30)
	Placebo (N=30)	Positive Control (N=30)	Dordaviprone (N=30)	
TEAEs, n (%)	12 (40.0)	14 (46.7)	19 (63.3)	26 (86.7)
TR-AEs, n (%)	6 (20.0)	5 (16.7)	11 (36.7)	14 (46.7)
SAEs, n (%)	0	0	1 (3.3)	1 (3.3)
AEs leading to discontinuation, n (%)	0	0	0	0
≥Grade 3 TEAEs, n (%)	0	0	0	0
Maximum severity (all TEAEs), n (%)				
Grade 1	12 (40.0)	13 (43.4)	18 (60.0)	24 (80.0)
Grade 2	0	1 (3.3)	1 (3.3)	2 (6.6)
Grade 3	0	0	0	0
Grade 4	0	0	0	0
Grade 5	0	0	0	0
TEAEs occurring in ≥2 pts, n (%)				
Dizziness	1 (3.3)	2 (6.7)	6 (20)	8 (26.7)
Headache	2 (6.7)	4 (13.3)	1 (3.3)	6 (20)
Catheter site bruise	1 (3.3)	0	2 (6.7)	3 (10)
Contusion	0	2 (6.7)	1 (3.3)	3 (10)
Medical device site erythema	0	0	3 (10)	3 (10)
Nausea	0	1 (3.3)	2 (6.7)	3 (10)
Vessel puncture site bruise	2 (6.7)	0	1 (3.3)	3 (10)
Back pain	0	2 (6.7)	0	2 (6.7)
Blood creatine phosphokinase increased	1 (3.3)	1 (3.3)	0	2 (6.7)
Catheter site swelling	1 (3.3)	0	1 (3.3)	2 (6.7)
Epistaxis	1 (3.3)	1 (3.3)	0	2 (6.7)
Fatigue	1 (3.3)	0	1 (3.3)	2 (6.7)
Medical device site irritation	0	1 (3.3)	1 (3.3)	2 (6.7)
Upper respiratory tract infection	2 (6.7)	0	0	2 (6.7)

Positive control, moxifloxacin (400 mg); dordaviprone (750 mg).
AE, adverse event; TEAE, treatment-emergent adverse event; TR-AE, treatment-related adverse event; pts. patients; SAE, serious adverse event.

References
(1) Venneti S, et al. *Cancer Discov.* 2023;13(11):2370-2393; **(2)** Arrillaga-Romany I, et al. *J Clin Oncol.* 2024;42(13):1542-1552; **(3)** Arrillaga-Romany I, et al. Neuro Oncol. 2024;26(Supplement_2):S173-S181.

Conflicts of Interest
SLF, JB, RK are paid consultants of Chimerix, Inc., a subsidiary of Jazz Pharmaceuticals. MA, TA, AB, MM, MJM, TT, and ON are employees of Chimerix, Inc., a subsidiary of Jazz Pharmaceuticals.

Acknowledgments
Medical writing and editorial assistance was provided by Meghan Sullivan, PhD, CMPP, an employee of Chimerix, Inc., a subsidiary of Jazz Pharmaceuticals.

Table 2. Placebo-corrected change from baseline QTcF (ΔΔQTcF) at each time point with statistical modeling

Parameter: ΔΔQTcF (ms)		Treatment	
Time point	Statistics	Dordaviprone (750 mg)	Moxifloxacin (400 mg)
0.25 h Post-dose	n	29	30
	LS Mean	-1.4	-0.7
	SE	1.08	1.07
	90% CI	(−3.18; 0.43)	(−2.53; 1.04)
0.5 h Post-dose	n	29	30
	LS Mean	3.5	2.1
	SE	1.16	1.15
	90% CI	(1.56; 5.44)	(0.21; 4.05)
0.75 h Post-dose	n	29	30
	LS Mean	11.6	5.1
	SE	1.47	1.45
	90% CI	(9.14; 14.03)	(2.65; 7.49)
1 h Post-dose	n	29	30
	LS Mean	12.7	5.8
	SE	1.27	1.26
	90% CI	(10.61; 14.86)	(3.69; 7.90)
2 h Post-dose	n	29	29
	LS Mean	12.7	8.3
	SE	1.06	1.05
	90% CI	(10.89; 14.43)	(6.50; 10.01)
3 h Post-dose	n	29	30
	LS Mean	12.5	10.4
	SE	1.05	1.04
	90% CI	(10.70; 14.22)	(8.66; 12.15)
4 h Post-dose	n	29	30
	LS Mean	11.7	10.1
	SE	1.22	1.20
	90% CI	(9.61; 13.69)	(8.11; 12.14)
5 h Post-dose	n	29	30
	LS Mean	13.8	8.3
	SE	1.60	1.58
	90% CI	(11.15; 16.48)	(5.65; 10.93)
6 h Post-dose	n	29	30
	LS Mean	9.2	5.7
	SE	1.55	1.54
	90% CI	(6.60; 11.78)	(3.17; 8.30)
8 h Post-dose	n	29	30
	LS Mean	10.5	8.6
	SE	1.52	1.50
	90% CI	(7.95; 13.01)	(6.06; 11.07)
12 h Post-dose	n	29	30
	LS Mean	6.0	4.0
	SE	1.83	1.82
	90% CI	(2.94; 9.05)	(0.98; 7.04)
16 h Post-dose	n	29	30
	LS Mean	4.7	4.9
	SE	2.07	2.05
	90% CI	(1.20; 8.10)	(1.50; 8.34)
24 h Post-dose	n	28	30
	LS Mean	3.4	4.5
	SE	1.04	1.02
	90% CI	(1.62; 5.10)	(2.77; 6.19)

LS mean ΔΔQTcF based on a linear mixed-effects model: ΔQTcF = Baseline QTcF + Treatment + Time + Treatment×Time + Period + Sequence. An unstructured covariance structure was used to specify the repeated measures (time within participant and period), and an unstructured covariance structure was used to specify the participant-specific random effect. Number of participants included in statistical analysis, N=30. Data for one participant during the dordaviprone treatment period was excluded from the analysis while data for the same participant during the placebo treatment period was included.

Presented at the American Society for Clinical Pharmacology and Therapeutics (ASCPT) Annual Meeting, May 28-31 2025