Assessing Cytochrome P450 Drug Interaction Risk for ONC201 Using Physiologically Based Pharmacokinetic Modeling

Swati Jaiswal[1], Nikunjkumar K Patel[1], Hannah M. Jones[1], Savannah McFeely[1], Shamia L. Faison[2], Tim Tippin[3], Odin Naderer[3]

[1]Certara Simcyp Division, Certara Inc; [2]Certara Strategic Consulting, Certara Inc; [3]Chimerix, Inc.

Introduction

- ONC201 (dordaviprone) is a novel, orally administered, anti-cancer imipridone with demonstrated antitumor effects in patients with glioma,[1-3] and is currently in Phase III testing.[4]
- CYP3A4 is a key elimination pathway for ONC201 accounting for >50% of its clearance.
- *In vitro* inhibition of CYP3A4, CYP2C8, CYP2D6 by ONC201 suggests potential for clinical interaction.

Model Development

- A combination of *in vitro* and clinical data was used to develop the PBPK model using Simcyp™ Population-Based Simulator (V21). Clinical data was from three single-dose ONC201 studies in healthy subjects.
- The ONC201 fraction metabolized by CYP3A4 was set at 0.8 based on the observed data from an itraconazole DDI study in healthy adults.

Figure 1. Minimal PBPK Model with Single Adjusting Compartment (SAC) and ADAM Model

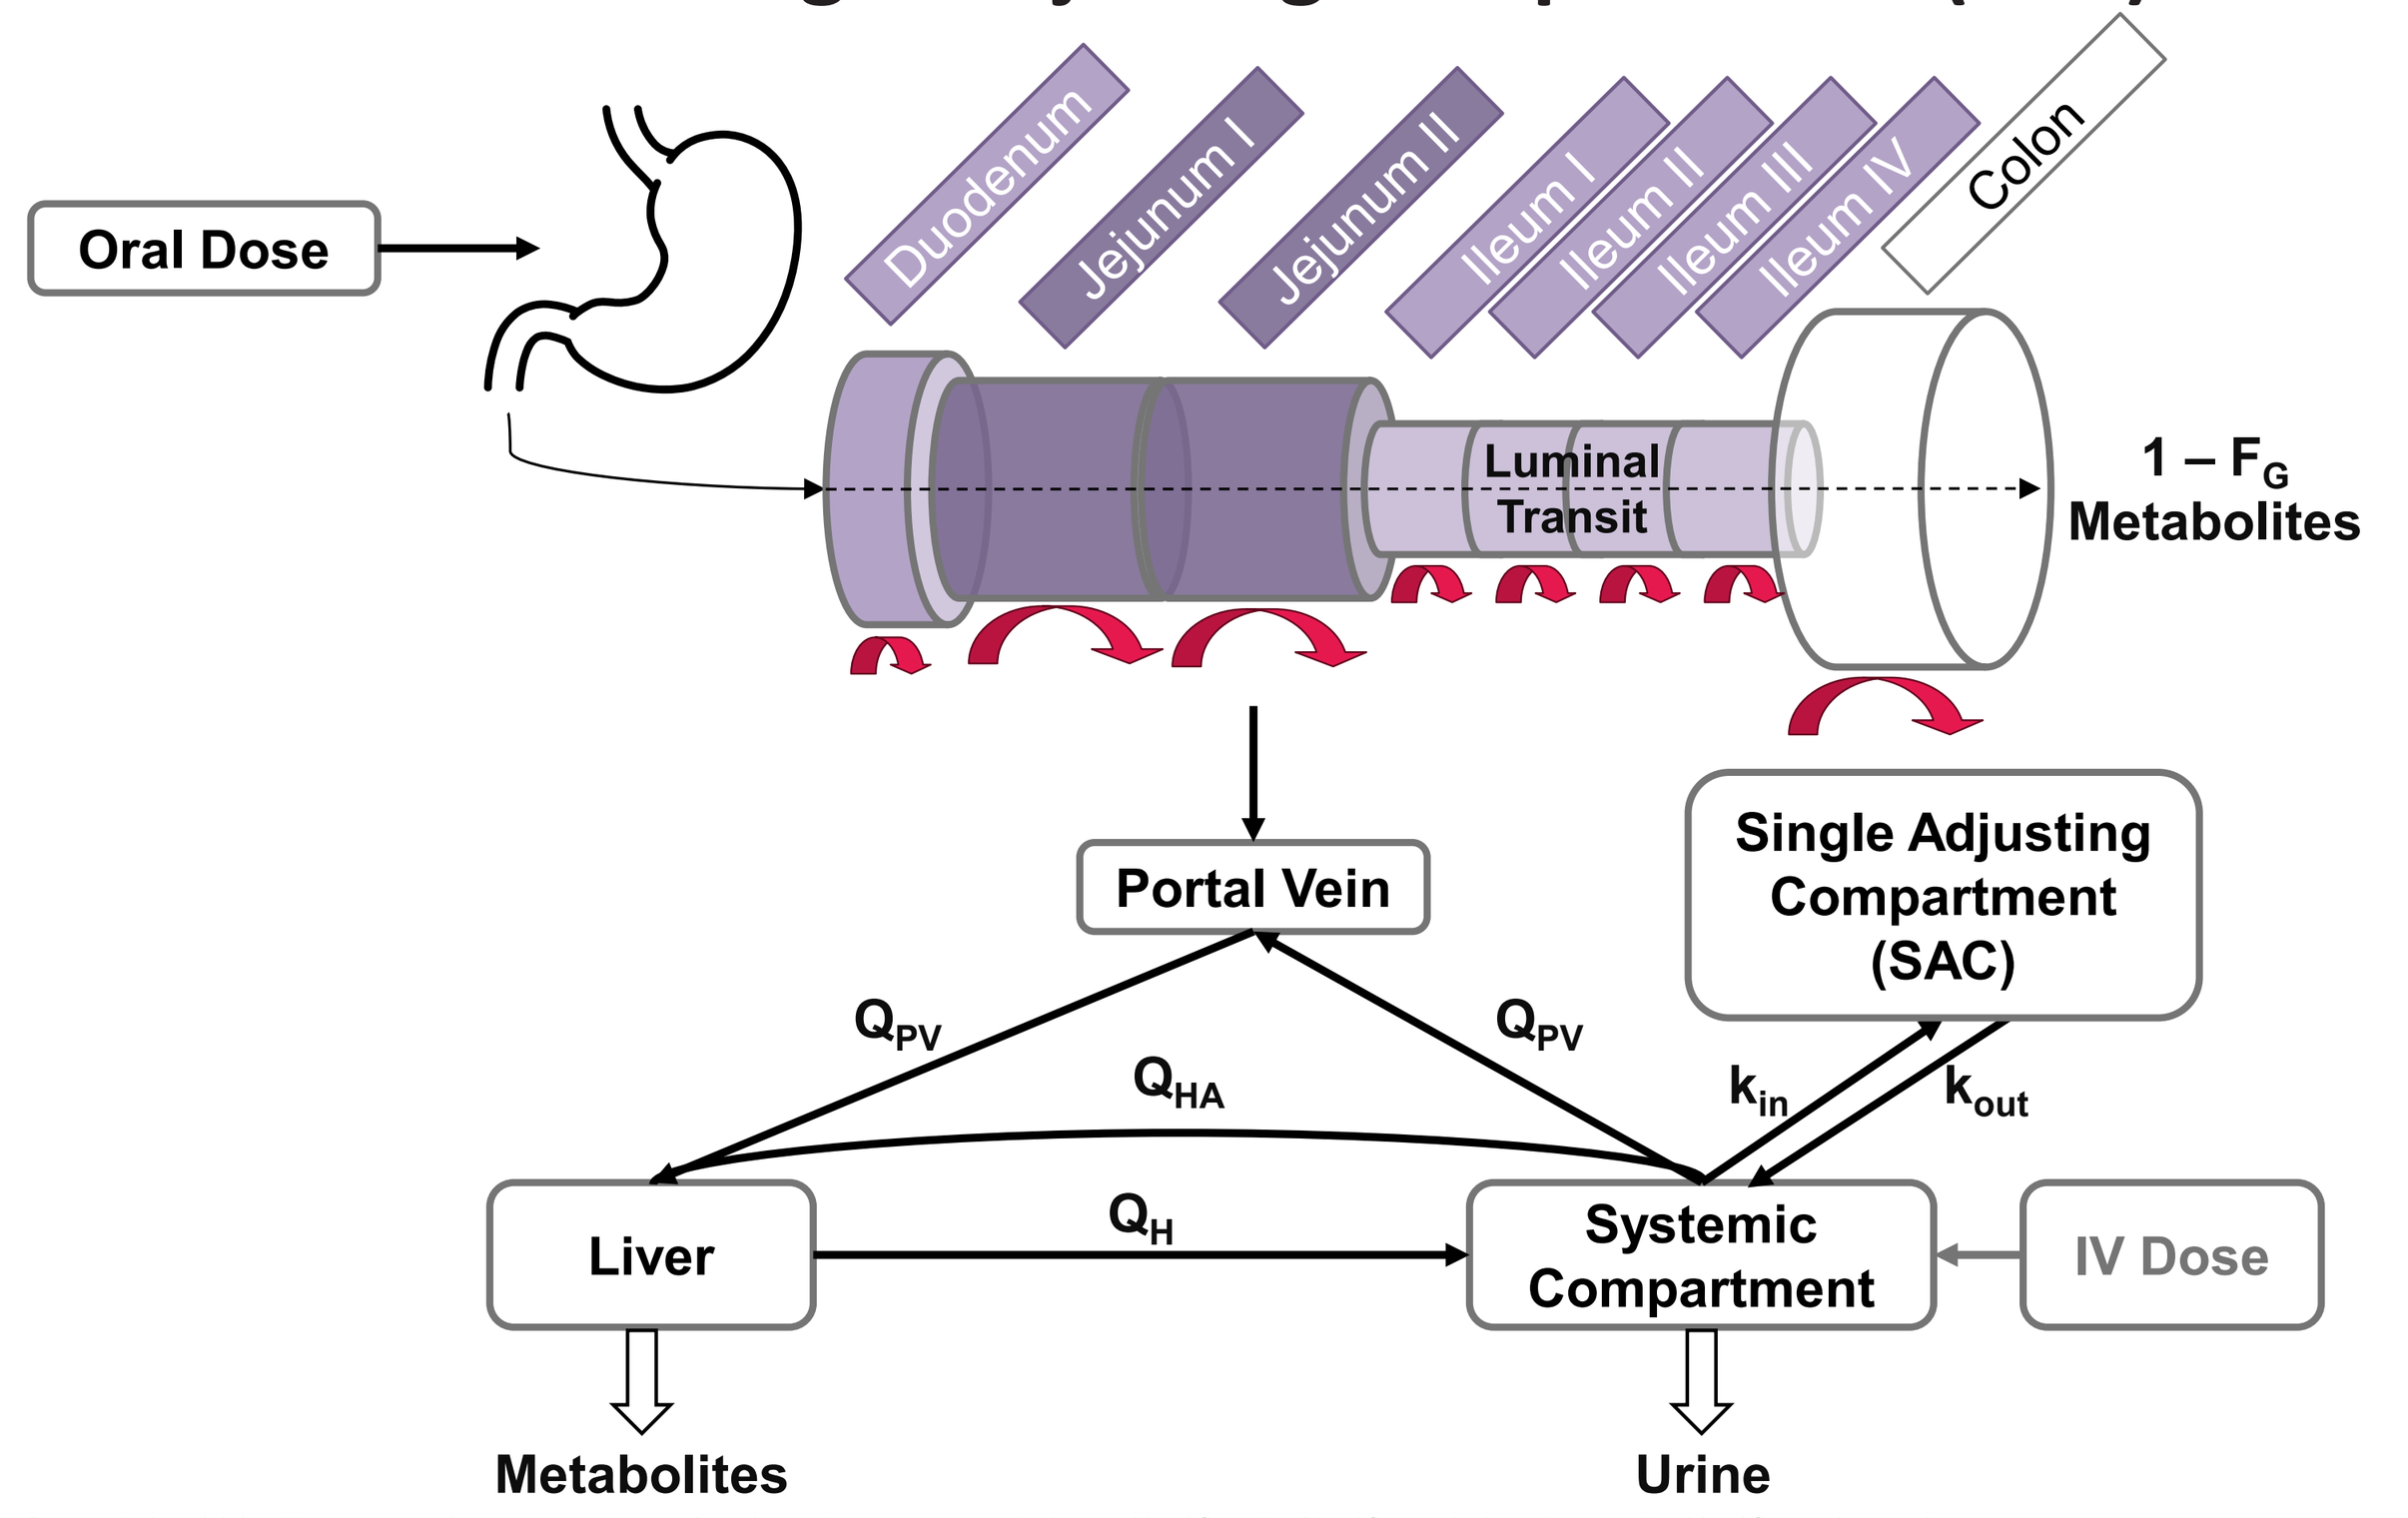
Oral Dose
Duodenum
Jejunum I
Jejunum II
Ileum I
Ileum II
Ileum III
Ileum IV
Colon
Luminal Transit
1 – F_G Metabolites
Portal Vein
Single Adjusting Compartment (SAC)
Q_PV
Q_HA
Q_PV
k_in
k_out
Liver
Q_H
Systemic Compartment
IV Dose
Metabolites
Urine

F_G, intestinal availability; IV, intravenous; k_{in}, input rate constant; k_{out}, elimination rate constant; Q_H, hepatic blood flow; Q_{HA}, Blood flow in the hepatic artery; Q_{PV}, blood flow in the portal vein.

Model Verification

- Model simulated (S) C_{max} and AUC of the three single-dose ONC201 clinical studies used to qualify the PBPK model were within 1.5-fold of observed (O) exposures (n=61 subjects)

Model Application to Predict CYP DDIs

- All CYP3A4 modulators were predicted to change ONC201 plasma exposure, consistent with their CYP3A4 potency classification.
- No change in exposure was predicted for probe substrates of CYP3A4, CYP2C8 and CYP2D6 after coadministration with ONC201.

Conclusions

- A PBPK model for ONC201 was successfully developed and verified. When co-administered with CYP3A4 inducers and inhibitors, changes in ONC201 plasma exposure were predicted by the model. ONC201 was not predicted to perpetrate an interaction with CYP enzymes.

PBPK model of ONC201 predicts no perpetrator interactions with CYP3A4, 2C8, or 2D6 substrates.

ONC201 is predicted to be a victim of DDIs when co-administered with CYP3A4 inhibitors and inducers.

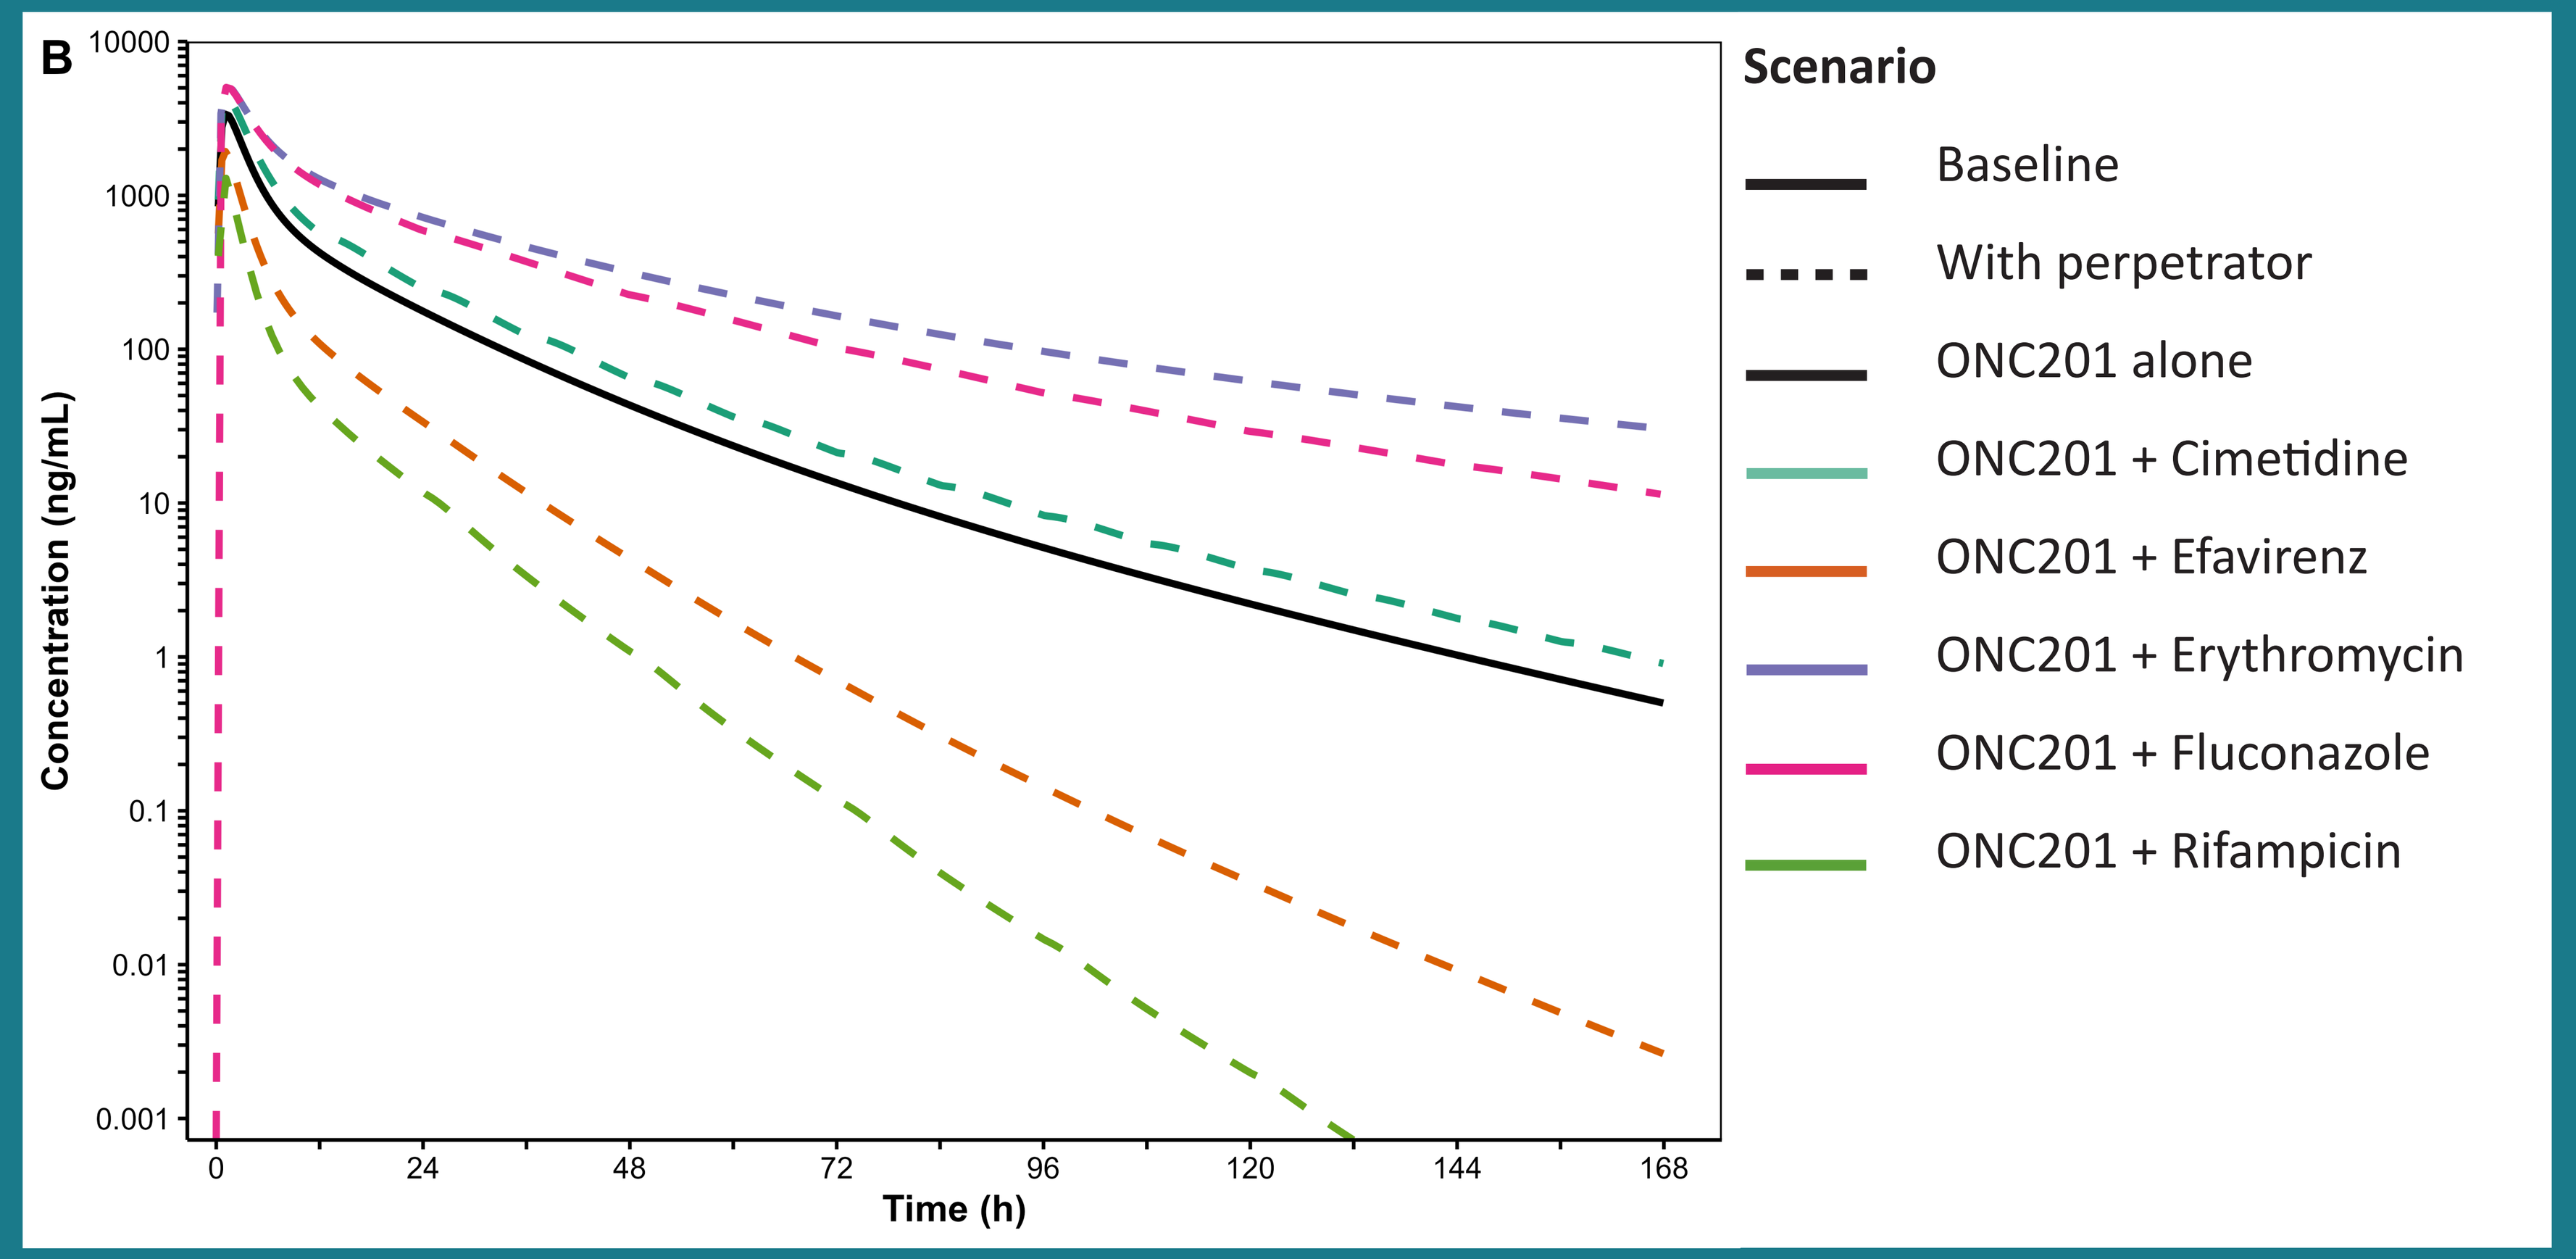
B
10000
1000
100
10
1
0.1
0.01
0.001
Concentration (ng/mL)
0
24
48
72
96
120
144
168
Time (h)
Scenario
Baseline
With perpetrator
ONC201 alone
ONC201 + Cimetidine
ONC201 + Efavirenz
ONC201 + Erythromycin
ONC201 + Fluconazole
ONC201 + Rifampicin

Scan here to download the poster and supplementary data.

Table 1. Final Input Parameters for ONC201

Parameter	Value
Absorption Model – ADAM Model	
fu_{gut}	0.993
Caco 2 P_{app} (x10^{-6} cm/s)	27.01
Calibrator (Metoprolol) P_{app} (x10^{-6} cm/s)	33.27
$P_{eff,man}$ (pred) (x10^{-4} cm/s)	3.74
Formulation type	Immediate Release
Solubility data	User-defined pH-solubility profile
Solubility (mg/mL) pH 1	62.2
Solubility (mg/mL) pH 4	92.1
Solubility (mg/mL) pH 4.5	98.6
Solubility (mg/mL) pH 5	6.02
Solubility (mg/mL) pH 6	0.26
$K_{m:w,neutral}$	4.58
$K_{m,w,ion}$	2.58
Precipitation Model	Model 2
Precipitation Rate Constant (1/h)	4
Critical Supersaturation Ratio	10
Interaction Parameters	
CYP3A4 K_i (µM)	20.24
CYP2C8 K_i (µM)	31.2
CYP2D6 K_i (µM)	27.12
fu_{mic}	0.93
P-gp K_i (µM)	32.2
Physicochemical and Binding Parameters	
MW (g/mol)	386.49
Log P	3.09
Compound type	Diprotic Base
pKa	6.76, 4.99
B:P	0.778
fu – 0.193 mg/L	0.0294
fu – 0.386 mg/L	0.0303
fu – 1.932 mg/L	0.0389
fu – 3.865 mg/L	0.0412
fu – 7.730 mg/L	0.0492
Main binding protein	AAG
Distribution Model – Minimal PBPK Model	
V_{SS} (L/kg)	1.11
k_{in} (1/h)	0.11
k_{out} (1/h)	0.15
V_{SAC} (L/kg)	6.60E-05
Elimination Parameters	
CL/F (L/h)	29
fm_{CYP3A4}	0.8
fm_{CYP2D6}	0.2
CYP3A4 CL_{int} (µL/min/pmol)	1.17
CYP2D6 CL_{int} (µL/min/pmol)	6.93
CL_R (L/h)	0.00

B:P, blood to plasma ratio; Caco 2, human colon adenocarcinoma cells (clone 2); CL/F, apparent oral clearance; Clint, intrinsic metabolic clearance; CIR, renal clearance; CYP, Cytochrome P450; fm, fraction of drug metabolized; fugut, fraction unbound in the gut, i.e., within the enterocyte; fumic, free fraction of drug in an in vitro microsomal preparation; Ki, enzyme competitive inhibition constant; Kin, input rate constant; Kout, elimination rate constant; MW, molecular weight; Papp, apparent permeability coefficient; PBPK, physiologically-based pharmacokinetic; Peff,man, effective human jejunum permeability; P-gp, P-glycoprotein; pKa, acid dissociation constant; VSAC, volume of single adjusting compartment; VSS, volume of distribution at steady state.

Table 2. Model Development: Simulated and Observed ONC201 Parameters with and without Itraconazole

	ONC201		ONC201 + Itraconazole		GMR	
	AUC_{0-t} (h.ng/mL)	C_{max} (ng/mL)	AUC_{0-t} (h.ng/mL)	C_{max} (ng/mL)	AUC_{0-t}	C_{max}
Simulated	4185	695	19356	1191	4.62	1.71
CV%	60.5	51.7	100	52.3	57.0	18.0
90% CI – Lower	3907	655	17463	1121	4.33	1.68
90% CI – Upper	4483	738	21453	1266	4.94	1.75
Observed	4119	763	17663	1475	4.29	1.93
CV%	35.1	37.2	34.0	27.8	-	-
90% CI – Lower	3476	638	14985	1287	3.86	1.76
90% CI – Upper	4881	913	20820	1690	4.76	2.12
S/O	**1.02**	**0.91**	**1.10**	**0.81**	**1.08**	**0.89**

125 mg ONC201 alone and 200 mg itraconazole daily for Days 1-8, single dose 125 mg ONC201 on Day 5.
AUC_{0-t}, area under the curve from time zero to t; GMR, geometric mean ratio; C_{max}, maximal drug concentration; CV, coefficient of variation; GMR, geometric mean ratio.

References
(1) Venneti S, et al. Cancer Discov. 2023; DOI: 10.1158/2159-8290.CD-23-0131 (2) Arrillaga-Romany I, et al. Neuro Oncol. 2021;23(Supplement_6):vi230-vi230; (3) Arrillaga-Romany I, et al. Neuro Oncol. 2020;22(1):94-102; (4) Arrillaga-Romany I, et al. Neuro Oncol. 2023;25(Supplement_2):ii84-ii84.

Conflicts of Interest
SJ, NKP, HMJ, SM, and SLF are paid consultants of Chimerix, Inc. TT and ON are employees of and have stock options in Chimerix, Inc.
Acknowledgements
Medical writing and editorial assistance was provided by Meghan Sullivan, PhD, CMPP, an employee of Chimerix, Inc.

Table 3. Model Verification: Simulated and Observed ONC201 PK Parameters (Dose Escalation)

	AUC_{0-inf} (h.ng/mL)	C_{max} (ng/mL)	t_{max}[a] (h)
125 mg Dose			
Simulated	3971	620	1.25
CV%	63.3	45.2	-
90% CI – Lower	3672	585	0.75
90% CI – Upper	4295	657	2.05
Observed	2946	829	0.75
CV%	35.2	29.3	-
S/O	**1.35**	**0.748**	**1.67**
375 mg Dose			
Simulated	10880	1771	1.25
CV%	62.1	44.1	-
90% CI – Lower	10072	1673	0.75
90% CI – Upper	11753	1875	2.00
Observed	10564	2501	0.75
CV%	30.5	35.0	-
S/O	**1.03**	**0.708**	**1.67**
625 mg Dose			
Simulated	17123	2810	1.20
CV%	61.9	44.6	-
90% CI – Lower	15855	2653	0.70
90% CI – Upper	18492	2976	2.15
Observed	17458	3830	0.750
CV%	29.8	25.6	-
S/O	**0.981**	**0.734**	**1.60**

[a]t_{max}, median, min, max.
AUC_{0-inf}, area under the curve from time zero to infinity; CV, coefficient of variation; C_{max}, maximal drug concentration; S/O, simulated/observed, t_{max}, time of maximum drug plasma concentration.

Table 4. Model Verification: Simulated and Observed ONC201 PK Parameters (Fed vs Fasted)[a]

	Fed			Fasted			GMR	
	AUC_{0-inf} (h.ng/mL)	C_{max} (ng/mL)	t_{max}[b] (h)	AUC_{0-inf} (h.ng/mL)	C_{max} (ng/mL)	t_{max}[b] (h)	AUC_{0-inf}	C_{max}
Simulated	18515	2520	2.05	18474	3372	1.20	1.00	0.75
CV%	56.6	45.8		56.4	44.6	-	-	-
90% CI – Lower	17608	2417	1.15	17571	3238	0.700	-	-
90% CI – Upper	19470	2627	4.65	19423	3512	2.50	-	-
Observed	18794	2522	2.25	18715	4148	0.750	1.00	0.61
CV%	36.3	33.3		40.7	45.1	-	-	-
S/O	**0.985**	**0.999**	**0.911**	0.987	0.813	1.60	1.00	1.23

[a]ONC201 was administered as a single 625 mg dose. Predictions assumed no blood flow change in the fed condition.
[b]t_{max}, median, min, max.
AUC_{0-inf}, area under the curve from time zero to infinity; CV, coefficient of variation; C_{max}, maximal drug concentration; GMR, geometric mean ratio; S/O, simulated/observed, t_{max}, time of maximum drug plasma concentration.

Table 5. Model Application: Predicted Change in ONC201 PK with CYP3A4 Modulators

	CYP3A4 Modulator (Potency)	Single Dose (625 mg) Geometric Mean Ratio (CV%) [90%CI] AUC_{0-inf}	C_{max}
Inhibitor	Erythromycin (500 mg QID) (moderate)	2.68 (43.6) [2.50-2.87]	1.51 (14.2) [1.47-1.54]
	Fluconazole (200 mg QD) (moderate)	2.48 (27.2) [2.38-2.60]	1.50 (13.4) [1.47-1.54]
	Cimetidine (400 mg BID) (weak)	1.42 (10.4) [1.39-1.44]	1.28 (8.14) [1.26-1.30]
Inducer	Efavirenz (600 mg QD) (moderate)	0.349 (43.2) [0.326-0.374]	0.565 (28.5) [0.539-0.592]
	Rifampicin (600 mg QD) (strong)	0.167 (74.4) [0.149-0.186]	0.328 (58.2) [0.300-0.359]

AUC_{0-inf}, area under the curve from time zero to infinity; BID, twice daily; C_{max}, maximal drug concentration, CV, coefficient of variation; QD, once-daily; QID, four times daily.
ONC201 with modulator dose regimens: ONC201 administered on the 7th day of 20 days of erythromycin dosing; ONC201 administered on the 10th day of 25 days of fluconazole dosing; ONC201 administered on the 10th day 18 days of cimetidine dosing; ONC201 administered on the 10th day of 18 days of efavirenz dosing; ONC201 administered on the 28th day of 38 days of rifampicin dosing.

Table 6. Model Application: Simulated CYP Probe Substrate PK Parameters with and without ONC201

	Probe Substrate		ONC201 + Probe Substrate		GMR	
	AUC_{0-t} (h.ng/mL)	C_{max} (ng/mL)	AUC_{0-t} (h.ng/mL)	C_{max} (ng/mL)	AUC_{0-t}	C_{max}
5 mg Midazolam (CYP3A4 Substrate)						
Geometric Mean	55.7	19.3	55.7	19.3	1.00	1.00
CV%	86.1	81.0	86.2	81.0	0.03	0.03
90% CI – Lower	49.2	17.1	49.2	17.1	1.00	1.00
90% CI – Upper	63.0	21.7	63.0	21.7	1.00	1.00
0.25 mg Repaglinide (CYP2C8 Substrate)						
Geometric Mean	4.78	4.59	4.78	4.59	1.00	1.00
CV%	92.6	65.2	92.6	65.2	0.01	0.01
90% CI – Lower	4.19	4.15	4.19	4.15	1.00	1.00
90% CI – Upper	5.45	5.06	5.45	5.06	1.00	1.00
50 mg Desipramine (CYP2D6 Substrate)						
Geometric Mean	821	16.0	821	16.0	1.00	1.00
CV%	1.06	52.9	1.06	52.9	0.02	0.00
90% CI – Lower	710	14.8	711	14.8	1.00	1.00
90% CI – Upper	948	17.4	948	17.4	1.00	1.00

GMRs for probe substrate in the absence (Day 1) and presence (Day 21) of 625 mg ONC201 administered once-weekly over 29 days in healthy subjects with 10 fold (CYP3A4, CYP2C8 and CYP2D6) and 15 fold (P-gp) reduction in *in vitro* Ki,u values.
AUC_{0-t}, area under the curve from time zero to t; GMR, geometric mean ratio; C_{max}, maximal drug concentration; CV, coefficient of variation.

Presented at the American Society for Clinical Pharmacology & Therapeutics (ASCPT) 2024 Annual Meeting, March 27-29, 2024, Colorado Springs, CO, USA