Metabolism and Disposition of [14C]-ONC201 (Dordaviprone) in Healthy Adult Participants

Yohannes Teffera[1], Tim Tippin[1], Francine A. de Castro[2], Joelle Batonga[2], Shamia L. Faison[2], Maggie Anderson[3], Mark J. Mullin[1], Angela Bartkus[1], Marion Morrison[1], Odin Naderer[1]

[1]Chimerix Inc; [2]Certara Strategic Consulting; [3]Consultant

Introduction

- ONC201 (dordaviprone) is a novel, orally administered, anti-cancer imipridone with demonstrated antitumor effects in patients with glioma,[1-3] and is currently in Phase III testing.[4]
- The inactive, N-dealkylation metabolite, ONC207, was the predominant metabolite formed in vitro in human, dog, and rat hepatocytes. However, in vivo in rats, ONC207 was a minor metabolite due to subsequent metabolism to other products.
- This study evaluated the absorption, metabolism, and excretion of ONC201 in humans.

Methods

- Six healthy male participants received a single 625 mg (100 µCi) dose of [14C]-ONC201.
- Blood, plasma, urine and feces were collected post dose throughout confinement (up to 288 h) and analyzed by liquid scintillation counting.
- Metabolite profiles were evaluated using liquid chromatography (LC) with radioactivity detection, and metabolite identification was assessed by LC with tandem mass spectroscopy.

Results

- Concentrations of drug-derived radioactivity in blood and plasma peaked 1-hour postdose and were below the limit of quantitation by 72 to 96 h postdose. Low distribution of radioactivity in blood cells was observed (blood to plasma ratios 0.568 to 0.796 from 0.5 to 36 hours postdose).
- The amount of administered radioactivity recovered in urine and feces combined was 91%. The majority of dosed radioactivity was recovered in the urine (71%), with 20% in feces. Most of the radioactivity (approximately 85%) was excreted within the first 96 hours post dose.
- Unchanged ONC201 was a minor component (<0.3%) in urine and feces; 24 metabolites were identified in urine and 17 were identified in feces.
- The primary metabolic pathways for ONC201 were oxidative.
- In plasma, the major circulating compounds were ONC201 and ONC207, contributing 32.8% and 31.5% of the total radioactivity (TRA) AUC, respectively. Of the 19 metabolites identified in plasma, no other single metabolite contributed >10% of the TRA AUC.
- Phase 2 conjugation and excretion of conjugates into feces via bile represented minor pathways in humans.

Discussion and Conclusions

- [14C]-ONC201 was well-absorbed after oral administration and largely cleared as oxidative metabolites in urine and feces. The major circulating metabolite was ONC207. Adequate toxicology coverage for ONC207 was achieved in dogs.

ONC201 was well-absorbed after oral administration and largely cleared as metabolites. The major metabolite was ONC207.

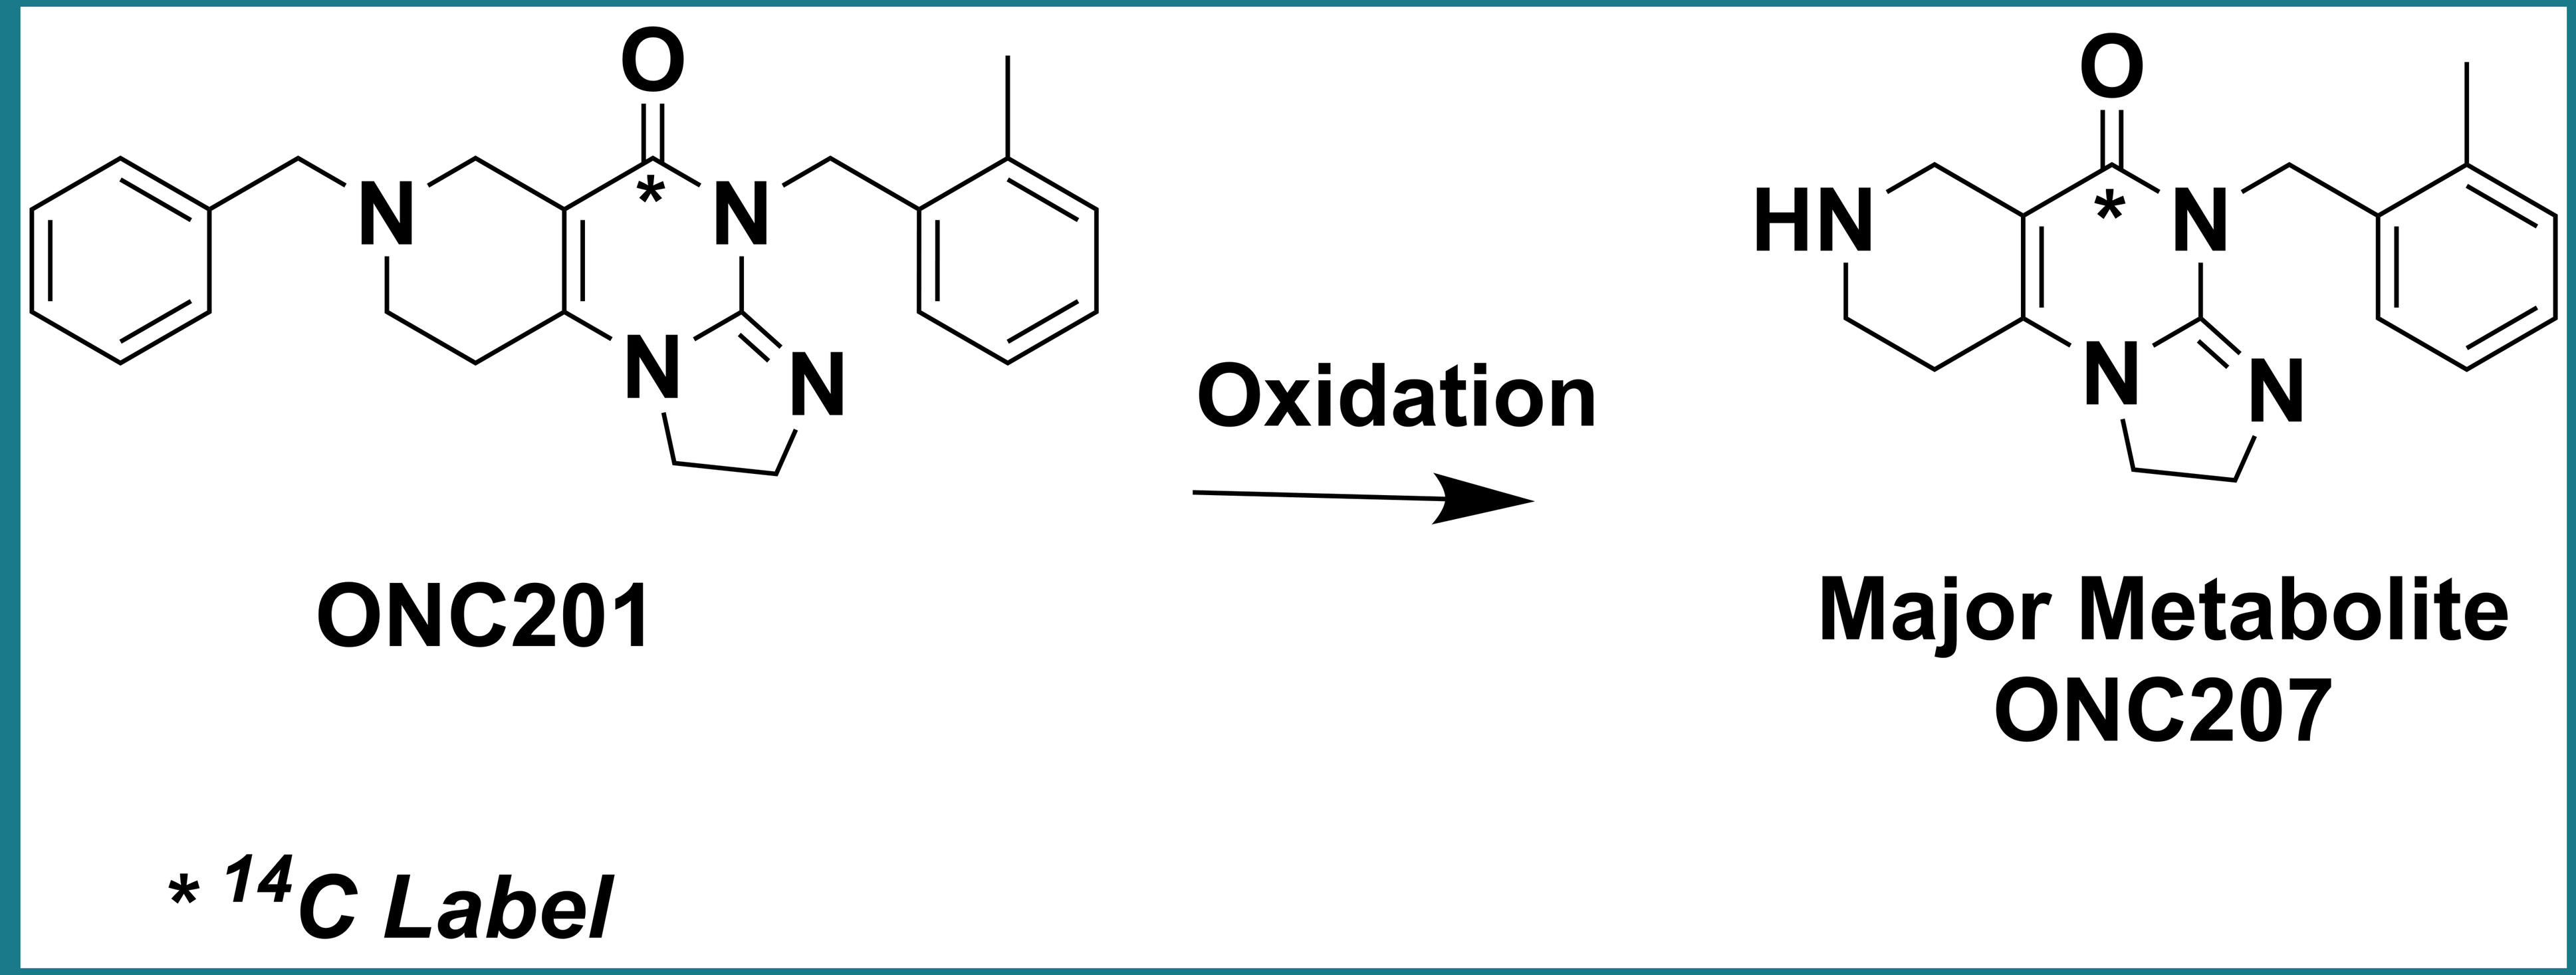
ONC201
Oxidation
Major Metabolite
ONC207
* 14C Label

Scan here to download the poster

Figure 1. Total Radioactivity Concentration-Time Profiles in Plasma and Whole Blood After a Single 625 mg Dose of [14C]-ONC201

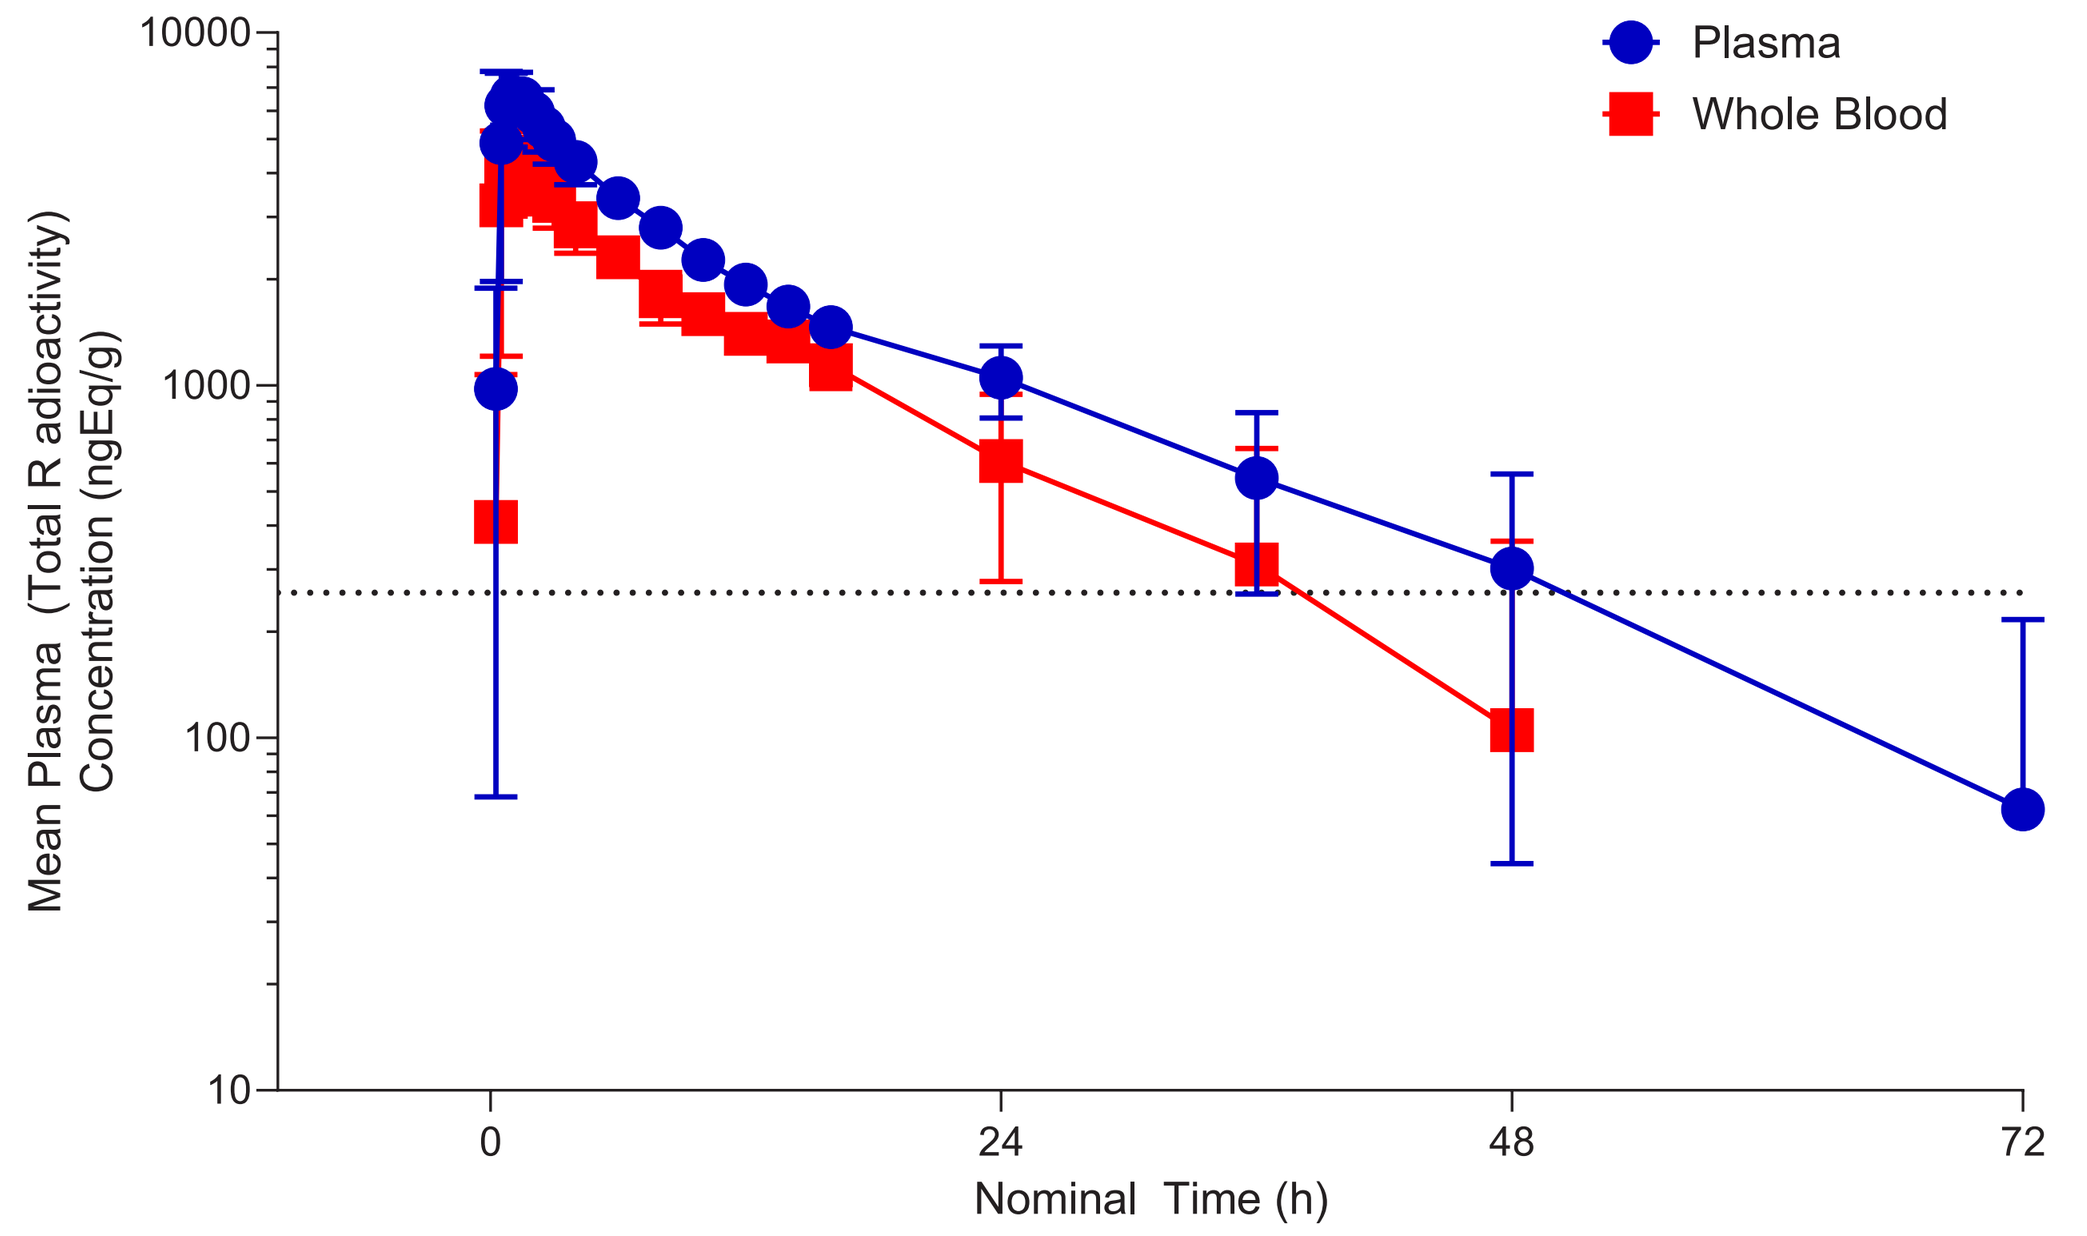
Plasma
Whole Blood
Mean Plasma (Total R adioactivity) Concentration (ngEq/g)
10000
1000
100
10
0
24
48
72
Nominal Time (h)

Figure 2. Cumulative Amount of Total Radioactivity Excreted in Urune + Feces

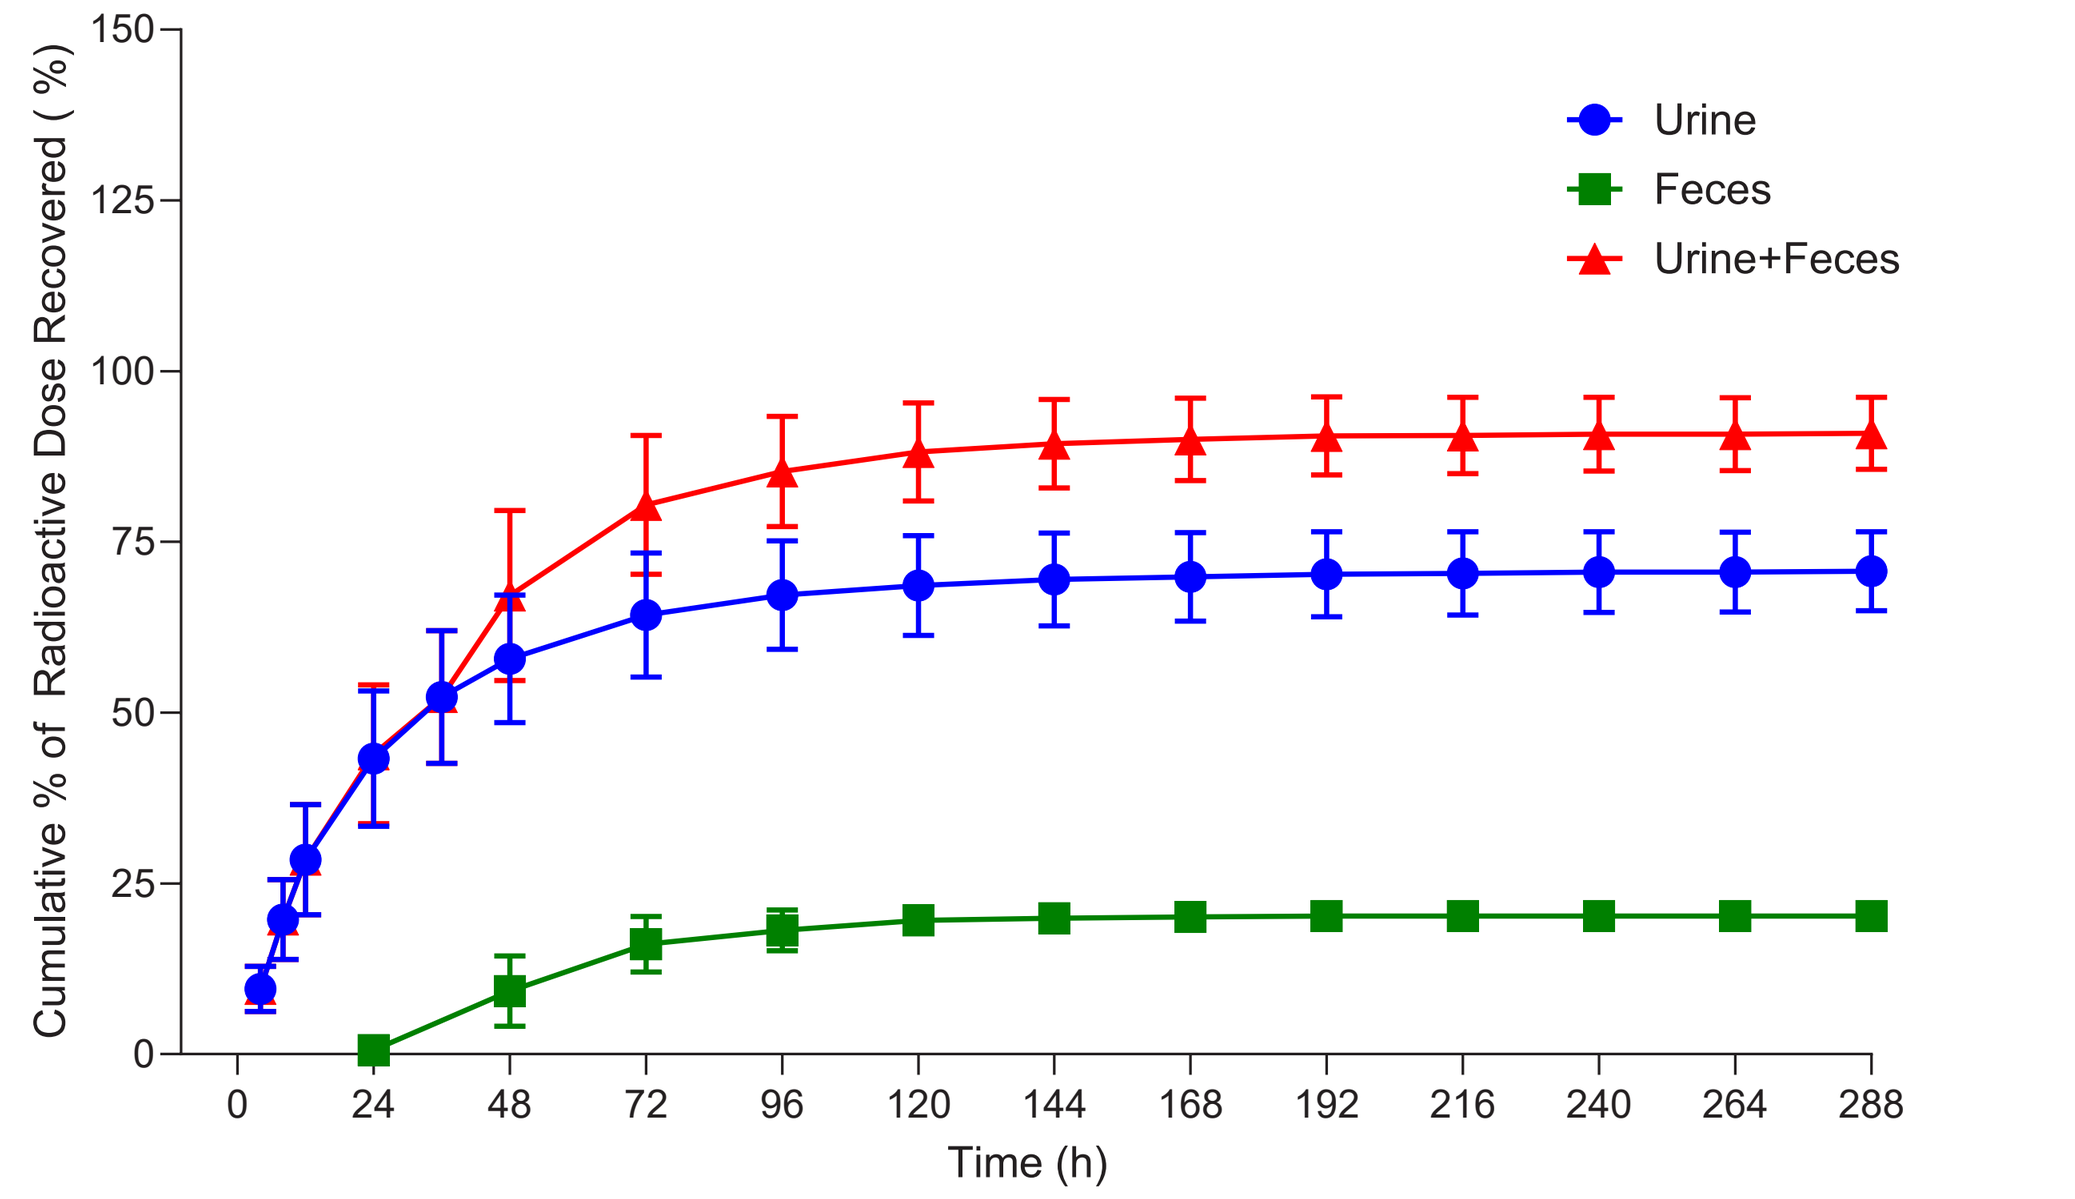
Urine
Feces
Urine+Feces
Cumulative % of Radioactive Dose Recovered (%)
150
125
100
75
50
25
0
0 24 48 72 96 120 144 168 192 216 240 264 288
Time (h)

Table 1. Summary of Urine and Fecal Radiokinetic Parameters Following a Single 625 mg Dose of [14C]-ONC201

	Urine		Fecal		Total	
	Ae (mgEq)	% Dose	Ae (mgEq)	% Dose	Ae (mgEq)	% Dose
N	6	6	6	6	6	6
Geometric Mean (%CV)	442 (8.5)	70.5 (8.3)	127 (8.5)	20.2 (8.8)	570 (5.9)	90.8 (5.7)
Minimum	386	61.7	109	17.3	525	83.9
Median	442	70.3	126	20.1	566	90.4
Maximum	503	79.8	138	22.1	629	99.7

Ae, total amount of drug-equivalents excreted; %CV, percentage coefficient of variation.

Figure 3. Putative Structures of ONC201 Metabolites in Plasma, Urine, and Feces

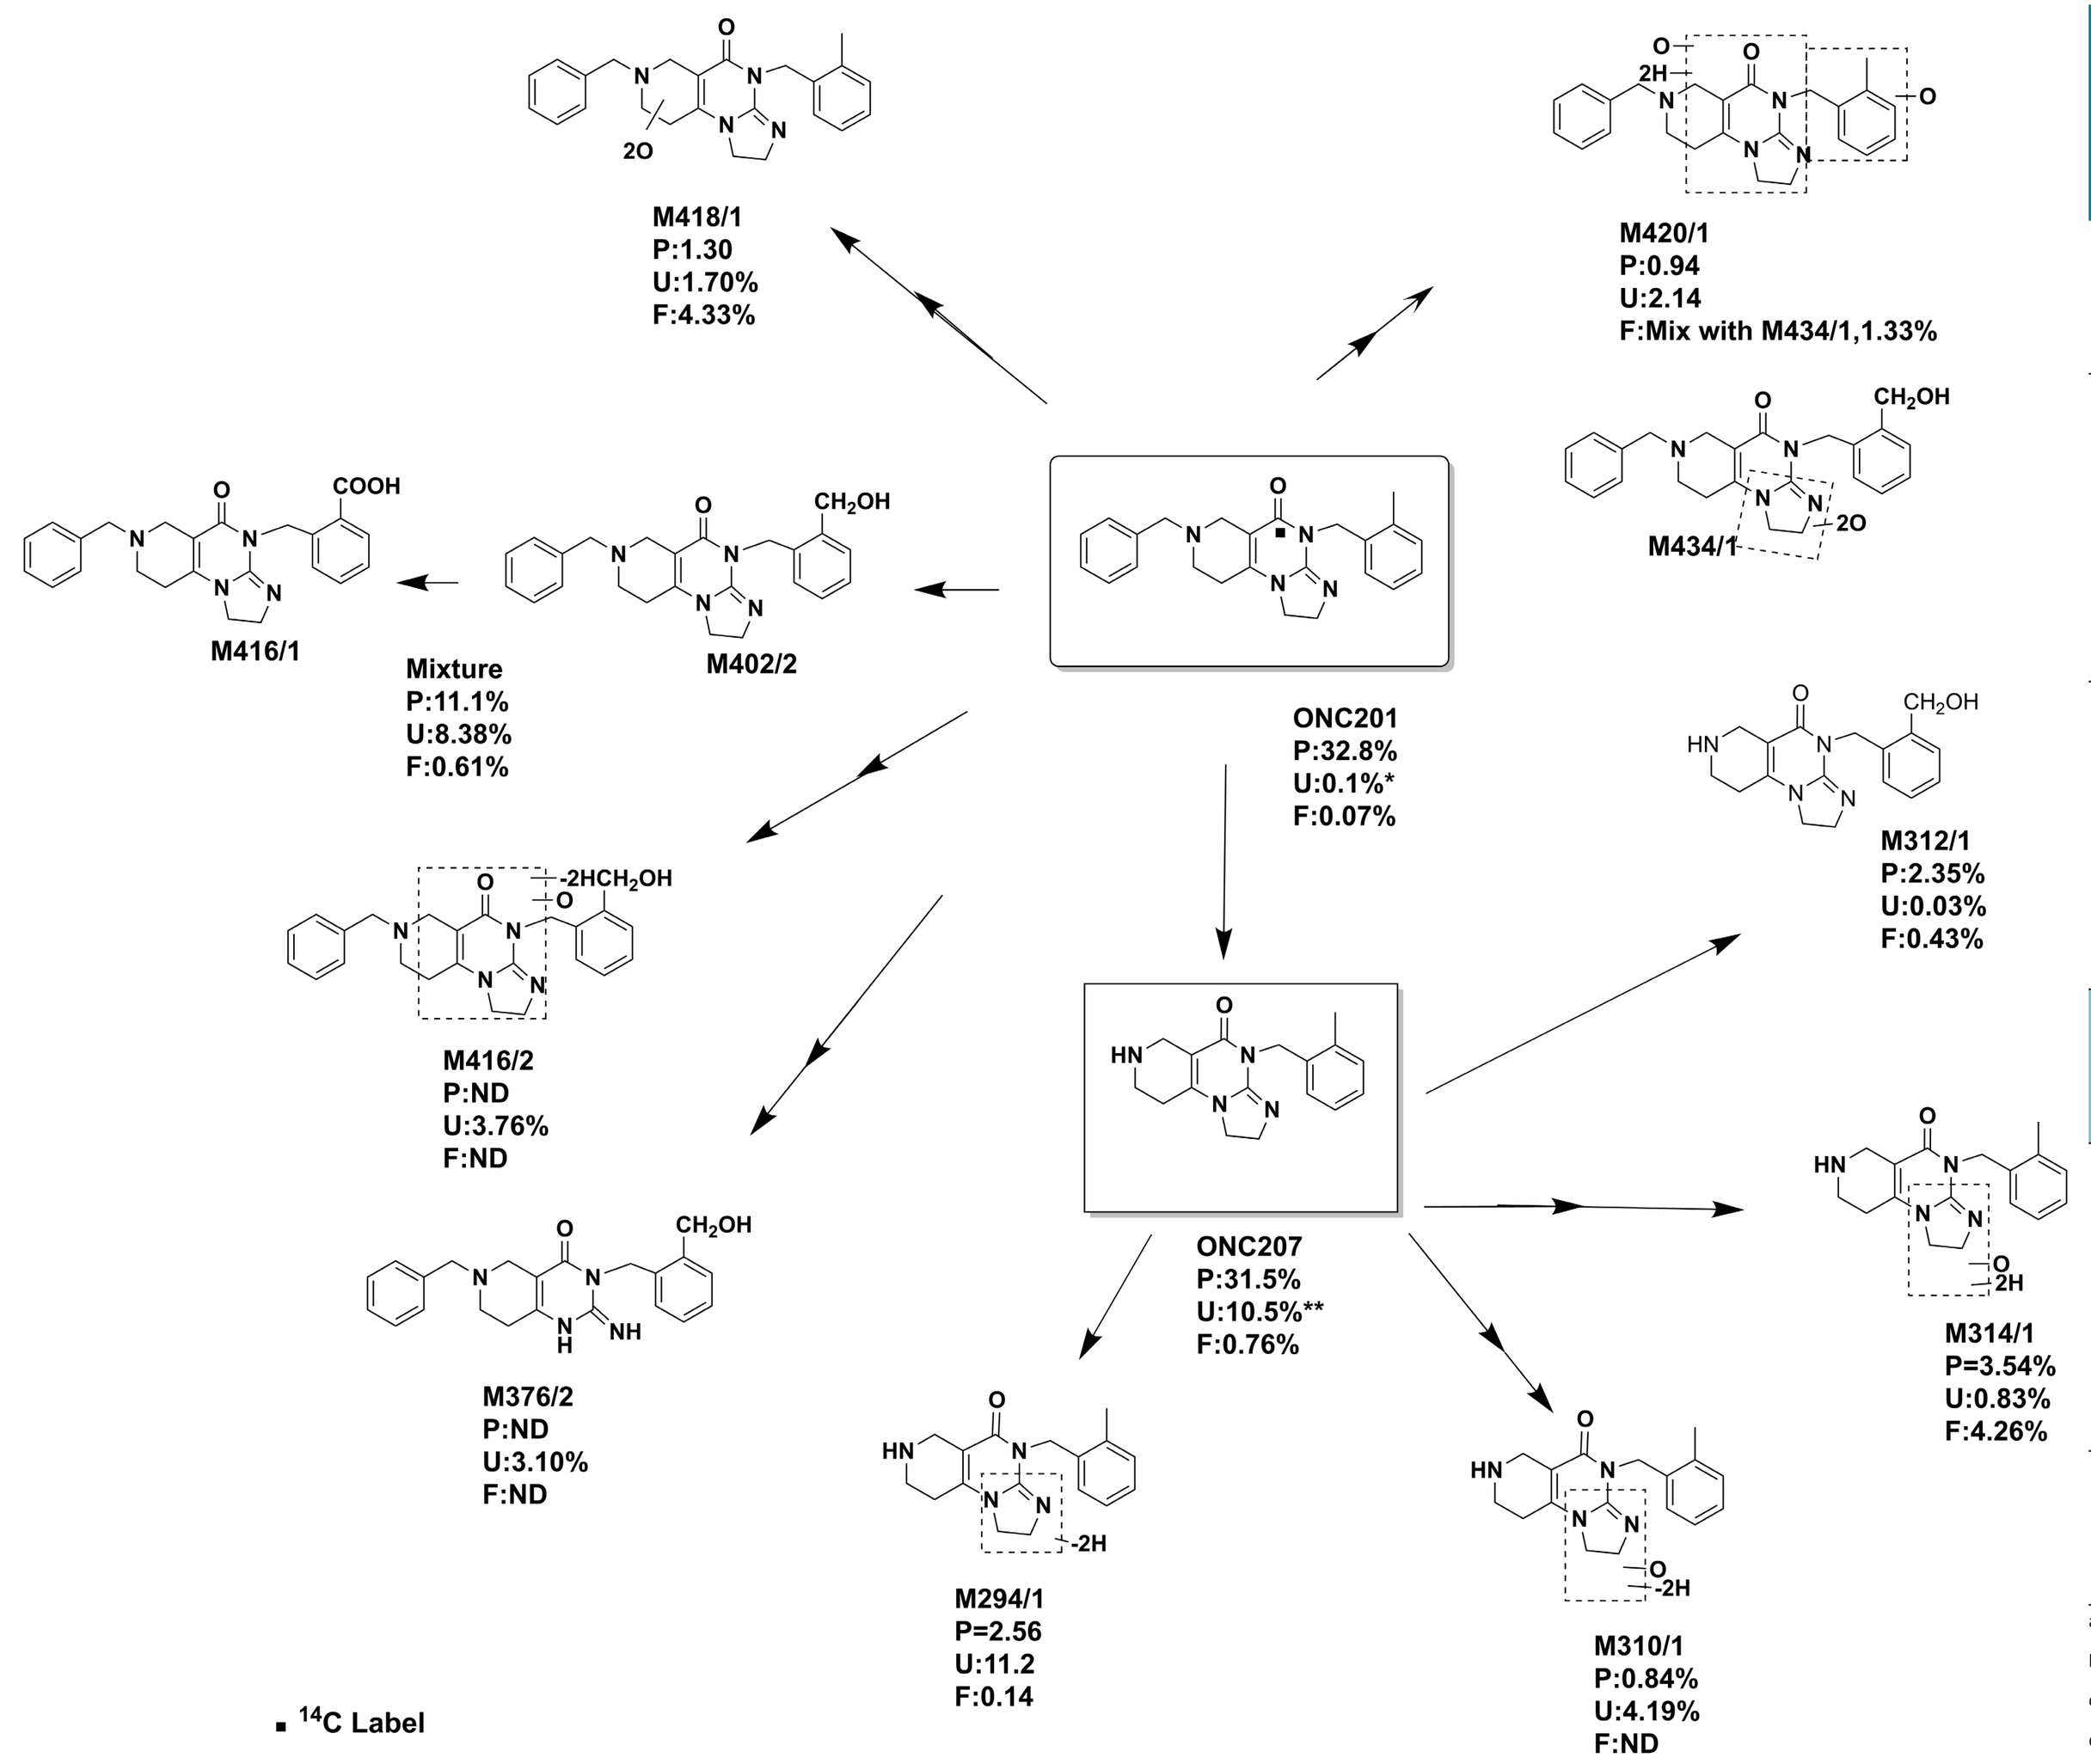
M418/1
P:1.30
U:1.70%
F:4.33%
M420/1
P:0.94
U:2.14
F:Mix with M434/1,1.33%
M434/1
M416/1
Mixture
P:11.1%
U:8.38%
F:0.61%
M402/2
ONC201
P:32.8%
U:0.1%*
F:0.07%
M312/1
P:2.35%
U:0.03%
F:0.43%
M416/2
P:ND
U:3.76%
F:ND
ONC207
P:31.5%
U:10.5%**
F:0.76%
M314/1
P=3.54%
U:0.83%
F:4.26%
M376/2
P:ND
U:3.10%
F:ND
M294/1
P=2.56
U:11.2
F:0.14
M310/1
P:0.84%
U:4.19%
F:ND
14C Label

*The reported amount of ONC201 in urine is from the LC-MS/MS analysis.
**The amount of ONC207 excreted in urine determined by LCRadiometry (10.5% of dose) was lower than from the validated LC-MS/MS analysis (36% of the dose), likely due to degradation during processing to M294/1.

Table 2. Cross Species Comparison of Exposure to ONC207 in Plasma Following Oral Administration of ONC201

Species	Dose	Sex	Dose Regimen	ONC207 End of Study AUC (ng*hr/mL plasma)[a]
Human[b]	625 mg	Male	Single	26031
Rat[c]	125 mg/kg	Male	Repeat once weekly for 13 weeks	1650
		Female		2150
Dog[d]	30 mg/kg	Male+Female	Repeat once weekly for 13 weeks	25500
				Exposure Ratio
Rat:Human		Male	(see above)	0.06
		Female	(see above)	0.08
Dog:Human		Male+Female	(see above)	0.98

[a]Values are AUClast at end of study for rats and dogs (AUC_{0-48}); AUCinf for human.
[b]Study ONC-201-106.
[c]Study ONC201-TX-100.
[d]Study ONC201TX101; values for dog are for male and female animals combined.

References

(1) Venneti S, et al. Cancer Discov. 2023; DOI: 10.1158/2159-8290.CD-23-0131 (2) Arrillaga-Romany I, et al. Neuro Oncol. 2021;23(Supplement_6):vi230-vi230; (3) Arrillaga-Romany I, et al. Neuro Oncol. 2020;22(1):94-102; (4) Arrillaga-Romany I, et al. Neuro Oncol. 2023.

Conflicts of Interest

FAd, JB, SLF, and MA are paid consultants of Chimerix, Inc. YT, TT, MJM, AB, MM, and ON are employees of and have stock options in Chimerix, Inc.

Acknowledgements

Medical writing and editorial assistance was provided by Meghan Sullivan, PhD, CMPP, an employee of Chimerix, Inc.

Presented at the American Society for Clinical Pharmacology & Therapeutics (ASCPT) 2024 Annual Meeting, March 27-29, 2024, Colorado Springs, CO, USA