143

The Effect of Severe Renal Impairment on Dordaviprone (ONC201) Pharmacokinetics

Shamia L. Faison[1], Joelle Batonga[1], Maggie Anderson[2], Thangam Arumugham[2], Angela Bartkus[3], Marion Morrison[3], Mark J Mullin[3], Tim Tippin[3], Odin Naderer[3]

[1]Certara Strategic Consulting; [2]Consultant; [3]Chimerix Inc

Introduction

- Dordaviprone (ONC201) is a novel, orally administered, anti-cancer imipridone with demonstrated antitumor effects in patients with glioma.[1-2] Dordaviprone is currently being evaluated in ACTION, a global Phase 3 trial of patients with newly-diagnosed diffuse glioma.[3]
- Metabolic clearance via CYP3A4 has been confirmed as a major elimination pathway of dordaviprone; renal clearance of dordaviprone has been identified as a minor elimination pathway.
- This study was designed to assess the effect of severe renal impairment (RI) on dordaviprone pharmacokinetics (PK).

Methods

- This was a single-center, open-label, parallel-group, single-period, single-dose study (**Figure 1**).

Figure 1. Study Design

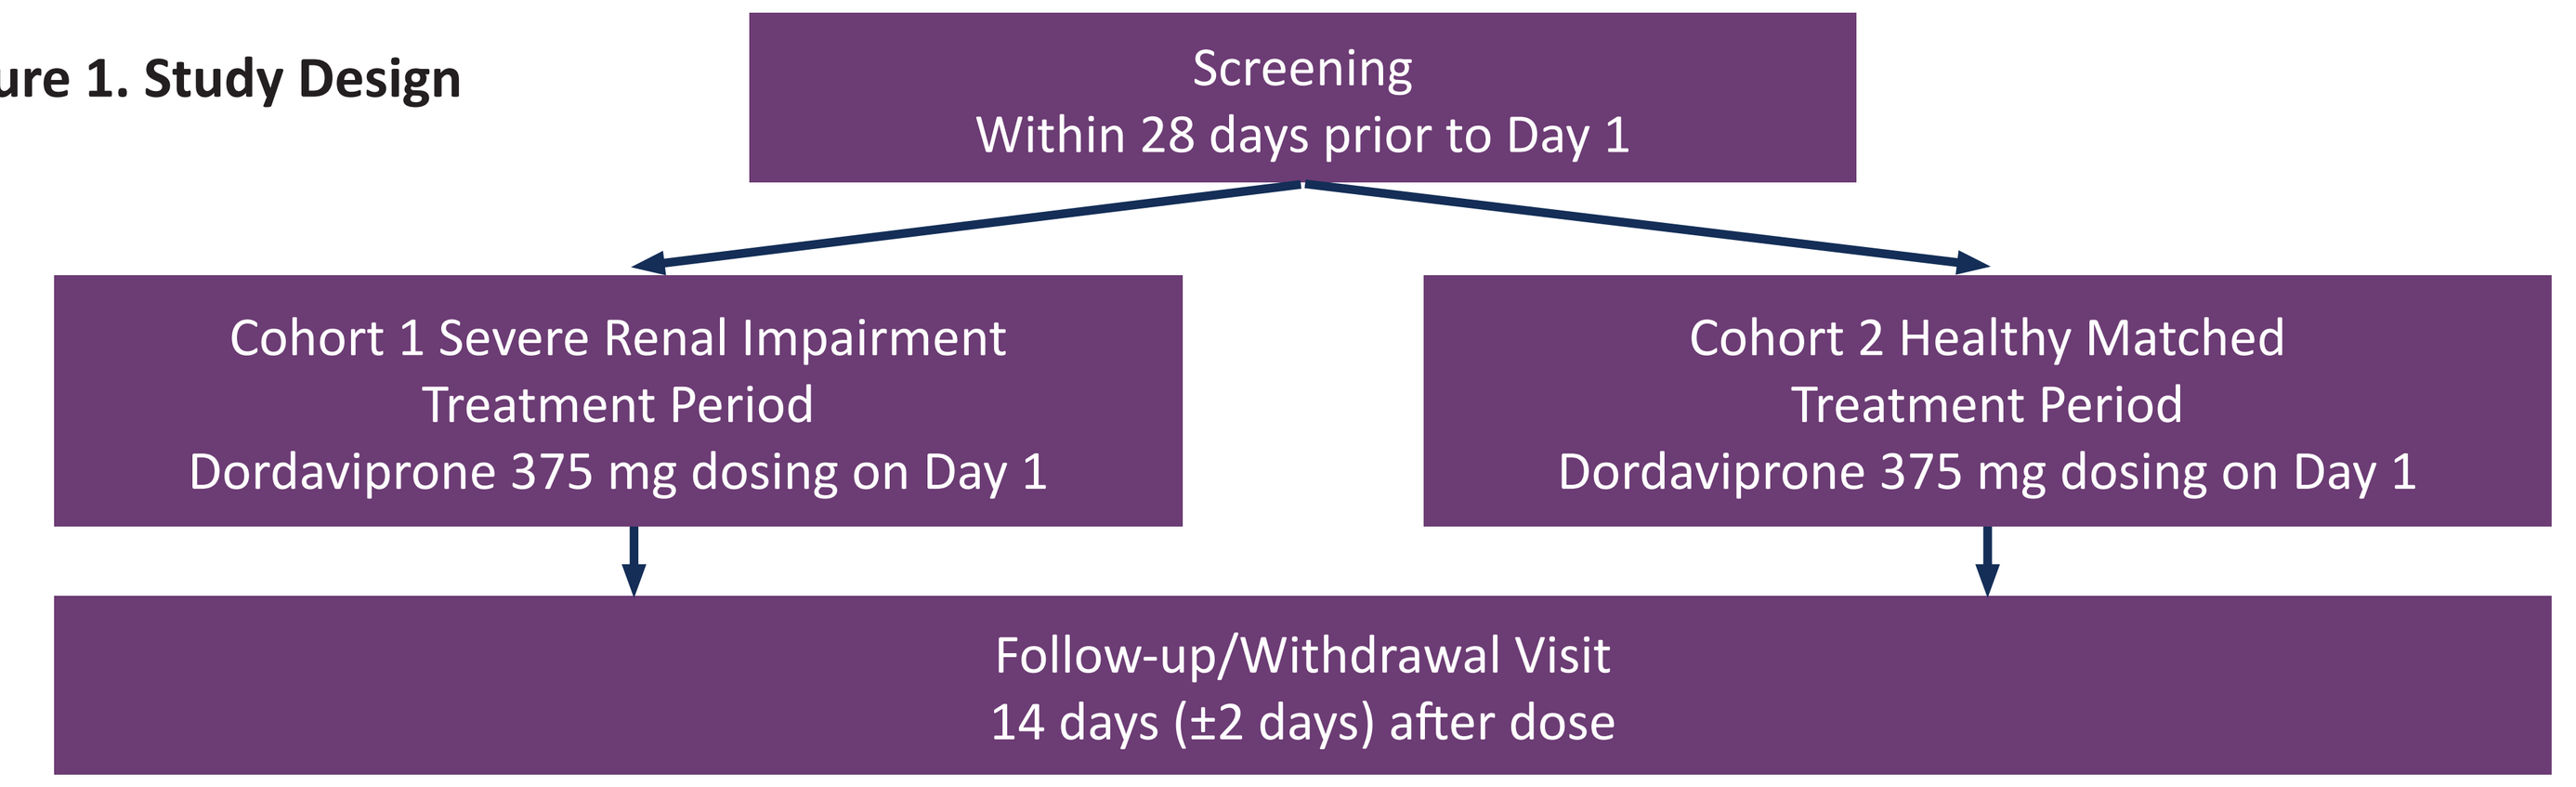
Screening
Within 28 days prior to Day 1
Cohort 1 Severe Renal Impairment
Treatment Period
Dordaviprone 375 mg dosing on Day 1
Cohort 2 Healthy Matched
Treatment Period
Dordaviprone 375 mg dosing on Day 1
Follow-up/Withdrawal Visit
14 days (±2 days) after dose

- Eight participants with severe RI (eGFR < 30mL/min), and 8 participants matched for age (±10 years), body mass index (±20%), and sex, with normal renal function, received a single 375 mg dose of dordaviprone.
- Plasma PK samples were collected at predose and at 0.25, 0.5, 0.75, 1, 1.5, 2, 2.5, 3, 4, 6, 8, 10, 12, 14, 16, 24, 36, 48, 72, and 168 hours post dose.
- Urine PK samples were collected predose and during 0-4, 4-8, 8-12, and 12-24 hours post dose.
- Plasma and urine samples were analyzed for dordaviprone and metabolite ONC207 using validated liquid chromatography tandem mass spectrometry methods.
 - The plasma analytical range for dordaviprone was 5 to 5000 ng/mL
 - The plasma analytical range for ONC207 was 3 to 3000 ng/mL
 - The urine analytical range for dordaviprone and ONC207 was 10 to 2000 ng/mL
- PK and safety profiles were evaluated between cohorts.
- To assess the effect of severe RI on dordaviprone exposure, point estimates and 90%CIs were calculated for the ratio of geometric means for PK parameters between cohorts (severe renal impairment-test, health match- reference).

Results

Table 1. Baseline Characteristics, Demographics, and Screening Characteristics

	Severe Renal Impairment (n=8)	Healthy Matched Participants (n=8)	Overall (N=16)
Demographics			
Age, mean (SD), years	62.9 (8.43)	57.6 (8.11)	60.3 (8.43)
Sex, n (%)			
Female	1 (12.5)	1 (12.5)	2 (12.5)
Male	7 (87.5)	7 (87.5)	14 (87.5)
Race, n (%)			
Native Hawaiian or Pacific Islander	1 (12.5)	0	1 (6.3)
White	7 (87.5)	8 (100.0)	15 (93.8)
Ethnicity, n (%)			
Not Hispanic/Latino	8 (100.0)	8 (100.0)	16 (100.0)
Weight, median (range), kg	82.1 (66.4, 108.5)	82.6 (72.0, 91.7)	82.6 (66.4, 108.5)
Height, median (range), cm	168.4 (154.5, 186.4)	172.5 (163.5, 180.5)	170.9 (154.5, 186.4)
BMI, median (range), kg/m²	28.3 (24.2, 36.0)	27.9 (25.2, 34.3)	28.0 (24.2, 36.0)
eGFR[a], mean (SD), mL/min	22.1 (4.5)	107.4 (15.3)	64.8 (45.4)
Disposition			
Participants enrolled, n (%)	8 (100.0)	8 (100.0)	16 (100.0)
Analysis sets, n (%)			
Safety analysis set	8 (100.0)	8 (100.0)	16 (100.0)
PK analysis set	8 (100.0)	8 (100.0)	16 (100.0)
Completed participants, n (%)	8 (100.0)	8 (100.0)	16 (100.0)

[a]Baseline eGFR was calculated by the Cockcroft-Gault equation based on Day -1 assessment.
BMI, body mass index; eGFR, estimated glomerular filtration rate; PK, pharmacokinetic.

- Dordaviprone was absorbed into circulation and then eliminated in an apparent multiphasic manner in both cohorts (**Figure 2**)

Figure 2. Mean ±SD Plasma Dordaviprone Concentration-Time Profiles in Participants with Severe Renal Impairment and Healthy Matched Participants after a Single Oral Dose of 375 mg Dordaviprone

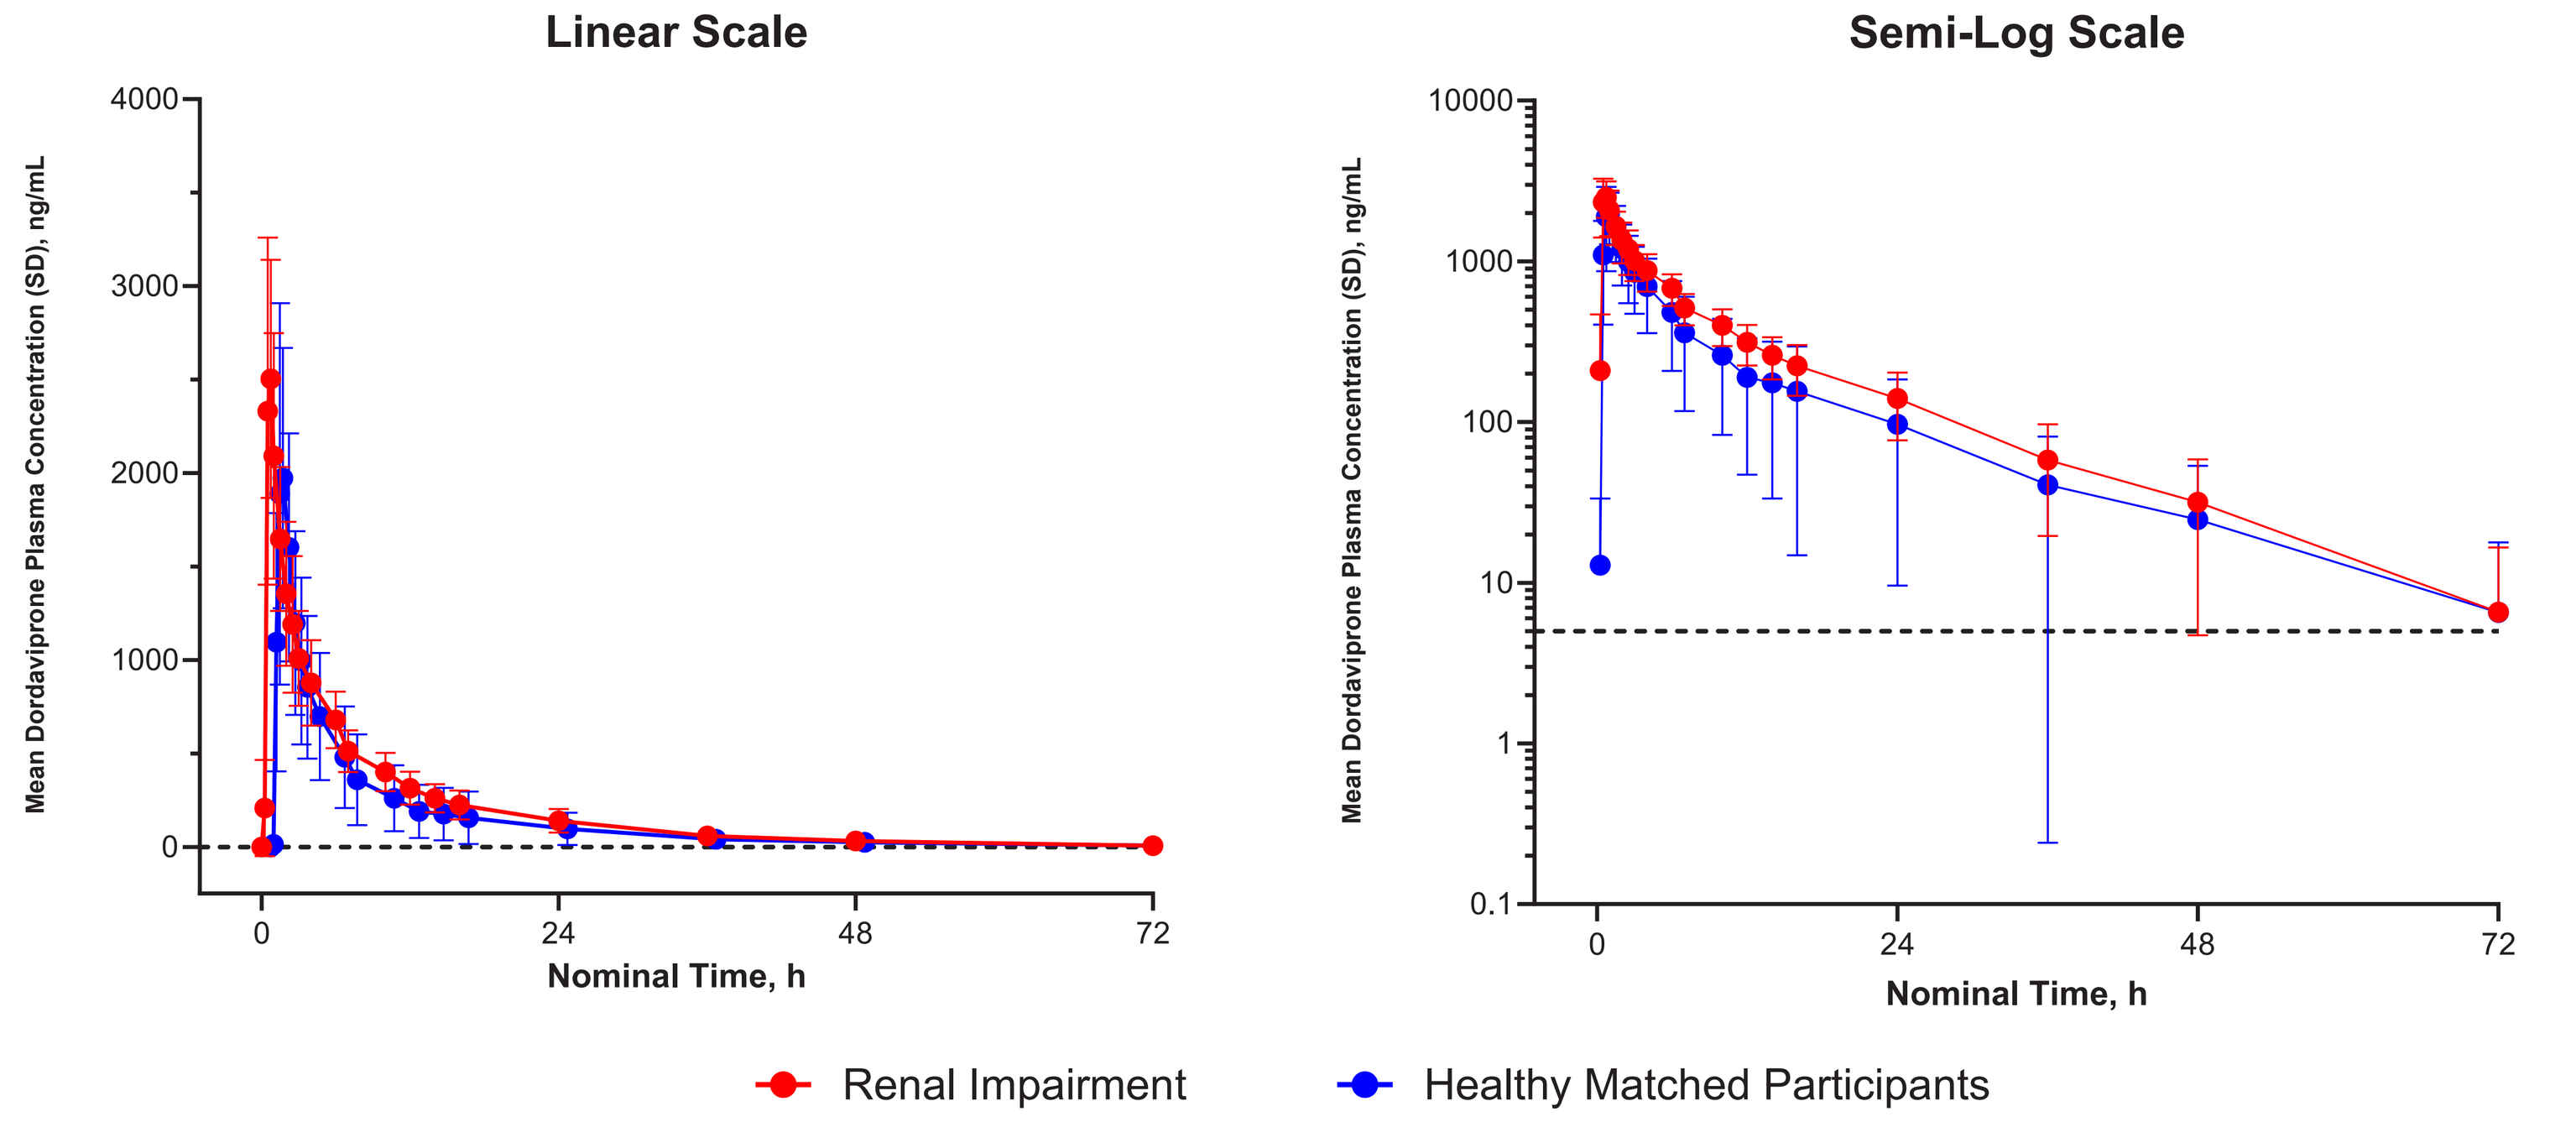
Linear Scale
Semi-Log Scale
Mean Dordaviprone Plasma Concentration (SD), ng/mL
Nominal Time, h
Renal Impairment
Healthy Matched Participants

- Dordaviprone exposure parameters in the severe renal impairment cohort trended higher (up to ~50%) and CL/F was lower (~30%) compared to dordaviprone exposure parameters in the healthy match cohort (**Table 2**).

Table 2. Plasma PK Parameters of Dordaviprone After a Single Oral Dose of 375 mg Dordaviprone in Participants with Severe Renal Impairment and Healthy Matched Participants

	Severe Renal Impairment (n=8)	Healthy Matched Participants (n=8)
Cmax (ng/mL) Geometric mean (95%CI) [CV%]	2621 (2158, 3185) [23.6]	2321 (1901, 2834) [24.2]
Tmax (h) Median (range)	0.75 (0.50, 2.00)	0.75 (0.75, 1.50)
AUClast (h*ng/mL) Geometric mean (95%CI) [CV%]	13522 (10324, 17711) [33.1]	9147 (5659, 14784) [62.5]
AUCinf (h*ng/mL) Geometric mean (95%CI) [CV%]	13733 (10442, 18061) [33.7]	9373 (5794, 15163) [62.6]
t1/2 Geometric mean (95%CI) [CV%]	9.9 (7.9, 12.4) [27.7]	10.9 (8.1, 14.8) [37.5]
CL/F (L/h) Geometric mean (95%CI) [CV%]	27.3 (20.8, 35.9) [33.7]	40.0 (24.7, 64.7) [62.6]
Vz/F (L) Geometric mean (95%CI) [CV%]	391 (325, 469) [22.2]	629 (505, 784) [26.7]
Clast (ng/mL) Geometric mean (95%CI) [CV%]	13.9 (9.16, 21.1) [53.3]	13.6 (9.12, 20.1) [50.2]
Tlast (h) Median (range)	48.06 (48.00, 72.00)	48.02 (24.00, 72.00)

AUCinf, area under the plasma concentration versus time curve from time zero to infinity; AUClast, area under the plasma concentration-time curve from time zero to time of last measurable plasma concentration; CL/F, apparent clearance of drug from plasma after extravascular administration; Cmax, peak (maximum) plasma concentration of the drug; CV%, percentage coefficient of variation (geometric); t½, half-life associated with the terminal slope of the semi-logarithmic drug concentration-time curve; Tmax, time to peak (maximum) plasma concentration; Vz/F, apparent volume of distribution during the terminal (λz) phase after extravascular administration.

- Geometric mean ratios and 90%CIs of dordaviprone exposure for Cmax, AUClast, and AUCinf in the severe RI cohort compared to the healthy matched cohort were 1.13 (0.92 to 1.39), 1.48 (0.98 to 2.23), and 1.47 (0.97 to 2.21), respectively (**Table 3**).

Table 3. Comparison of Dordaviprone PK in Participants with Severe Renal Impairment Compared to Healthy Matched Participants with Normal Renal Function

	Severe Renal Impairment (Test)		Healthy Matched Participants (Reference)		Test/Reference Ratio (90%CI)
	n	LSMean	n	LSMean	
Cmax	8	2621.32	8	2321.01	1.13 (0.92, 1.39)
AUClast	8	13521.85	8	9147.22	1.48 (0.98, 2.23)
AUCinf	8	13732.91	8	9372.74	1.47 (0.97, 2.21)

AUCinf, area under the plasma concentration versus time curve from time zero to infinity; AUClast, area under the plasma concentration-time curve from time zero to time of last measurable plasma concentration; Cmax, peak (maximum) plasma concentration of the drug; LSMean, least squares mean.

- ONC207 was rapidly formed following oral administration of dordaviprone in both cohorts and declined in an apparent exponential manner (**Figure 3**).

Figure 3. Mean ±SD Plasma ONC207 Concentration-Time Profiles in Participants with Severe Renal Impairment and Healthy Matched Participants after a Single Oral Dose of 375 mg Dordaviprone

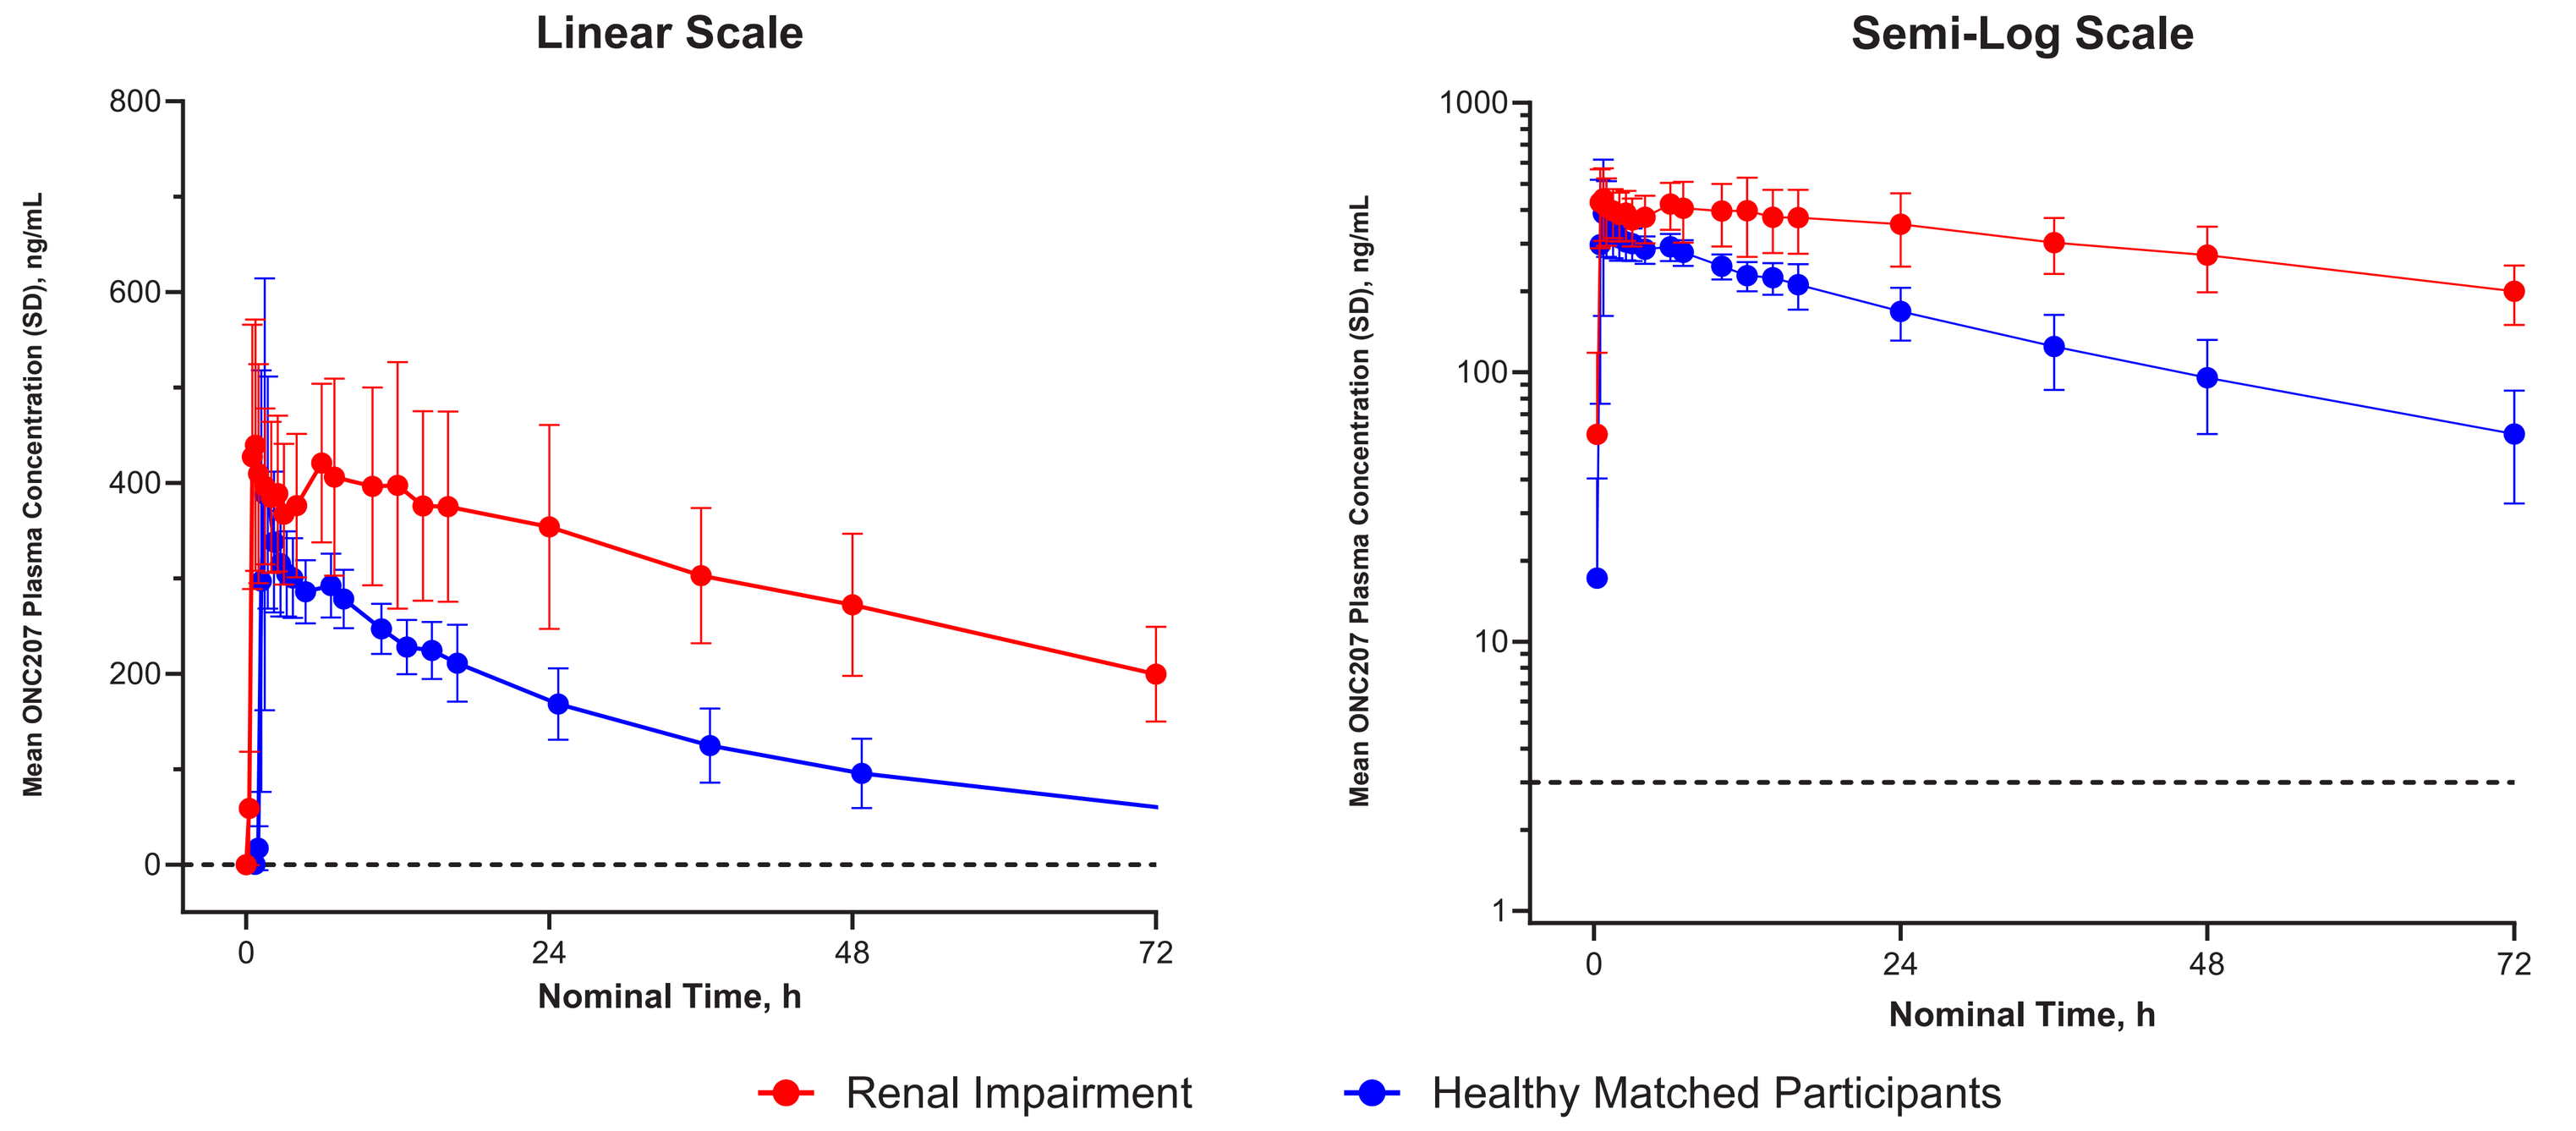
Linear Scale
Semi-Log Scale
Mean ONC207 Plasma Concentration (SD), ng/mL
Nominal Time, h
Renal Impairment
Healthy Matched Participants

- For metabolite ONC207, greater Cmax (~13%) and AUC (~2.9-fold)were observed in the severe RI cohort (**Table 4**).

Table 4. Plasma PK Parameters of ONC207 After a Single Oral Dose of 375 mg Dordaviprone in Participants with Severe Renal Impairment and Healthy Matched Participants

	Severe Renal Impairment (n=8)	Healthy Matched Participants (n=8)
Cmax (ng/mL) Geometric mean (95%CI) [CV%]	507 (423, 608) [21.9]	447 (349, 574) [30.4]
Tmax (h) Median (range)	0.75 (0.50, 24.02)	0.75 (0.75, 8.00)
AUClast (h*ng/mL) Geometric mean (95%CI) [CV%]	33461 (27993, 39998) [21.6]	12784 (10157, 16090) [28.0]
AUCinf (h*ng/mL) Geometric mean (95%CI) [CV%]	38817 (30711, 49064) [22.6]	13458 (10700, 16928) [28.0]
t1/2 Geometric mean (95%CI) [CV%]	59.3 (51.7, 67.9) [13.1]	35.7 (29.9, 42.6) [21.5]
M/P Ratio Cmax Geometric mean (95%CI) [CV%]	0.194 (0.138, 0.272) [42.5]	0.193 (0.136, 0.274) [43.8]
M/P Ratio AUClast Geometric mean (95%CI) [CV%]	2.47 (1.95, 3.13) [28.9]	1.40 (1.06, 1.84) [34.0]
M/P Ratio AUCinf Geometric mean (95%CI) [CV%]	3.18 (2.48, 4.08) [24.0]	1.44 (1.09, 1.90) [34.5]
Clast (ng/mL) Geometric mean (95%CI) [CV%]	71.8 (57.5, 89.5) [26.9]	11.8 (7.09, 19.6) [66.8]
Tlast (h) Median (range)	167.92 (167.25, 168.28)	168.02 (72.27, 168.28)

AUCinf, area under the plasma concentration versus time curve from time zero to infinity; AUClast, area under the plasma concentration-time curve from time zero to time of last measurable plasma concentration; Cmax, peak (maximum) plasma concentration of the drug; CV%, percentage coefficient of variation (geometric); M/P, metabolite/parent (unadjusted for molecular weight difference); t½, half-life associated with the terminal slope of the semi-logarithmic drug concentration-time curve; Tmax, time to peak (maximum) plasma concentration.

- Dordaviprone was minimally recovered in urine; geometric mean percent of dordaviprone dose recovered over the 0-24 hours postdose in severe RI and healthy matched cohorts were 0.158% and 0.173%, respectively (**Table 5**).
- Geometric mean CLr was similarly low in severe RI (0.0430 L/h) and healthy-matched cohorts (0.0694 L/h).

Table 5. Dordaviprone Urine PK Parameters

	Severe Renal Impairment (n=8)	Healthy Matched Participants (n=8)
Ae (mg) Geometric mean (95%CI) [CV%]	0.591 (0.265, 1.32) [123.0]	0.651 (0.460, 0.921) [43.4]
Ae (%) Geometric mean (95%CI) [CV%]	0.158 (0.076, 0.352) [123.0]	0.173 (0.123, 0.246) [43.4]
CLr (L/h) Geometric mean (95%CI) [CV%]	0.0430 (0.0198, 0.0935) [117.0]	0.0694 (0.0537, 0.0897) [31.4]

Ae, cumulative amount of drug excreted unchanged in urine from time 0-24 hr post-dose; CLr, renal clearance.

- Treatment-emergent adverse events (TEAEs) were reported in 6/8 (75%) of severe RI participants and 5/8 (62.5%) of healthy-matched participants; all TEAEs were mild (**Table 6**).
- Treatment-related AEs occurred in 50% of severe RI participants compared to 37.5% of healthy-matched participants.
- The most common treatment-related AE was headache, occurring in 2 (25%) participants from each cohort; no other treatment-related AEs occurred in more than 1 participant.

Table 6. Treatment-emergent Adverse Events

TEAE, n (%)	Severe Renal Impairment (n=8)	Healthy Matched Participants (n=8)	Overall (N=16)
Any TEAE	**6 (75.0)**	**5 (62.5)**	**11 (68.8)**
Headache	2 (25.0)	2 (25.0)	4 (25.0)
COVID-19	1 (12.5)	1 (12.5)	2 (12.5)
Dizziness	0	1 (12.5)	1 (6.3)
Muscle contractions, involuntary	0	1 (12.5)	1 (6.3)
Nausea	1 (12.5)	0	1 (6.3)
Dry mouth	0	1 (12.5)	1 (6.3)
Back pain	1 (12.5)	0	1 (6.3)
Neck pain	1 (12.5)	0	1 (6.3)
Nasal congestion	0	1 (12.5)	1 (6.3)
Oropharyngeal discomfort	0	1 (12.5)	1 (6.3)
Erythema	0	1 (12.5)	1 (6.3)
Pruritus	1 (12.5)	0	1 (6.3)
Eye irritation	0	1 (12.5)	1 (6.3)
Feeling abnormal	1 (12.5)	0	1 (6.3)
Dysuria	0	1 (12.5)	1 (6.3)

Conclusions

- The ratio of dordaviprone exposure between severe RI and healthy-matched cohorts was less than 2-fold.
- Dordaviprone was minimally recovered in urine in both severe RI and healthy-matched cohorts confirming renal clearance is a minor elimination pathway of dordaviprone.
- The observed decrease in dordaviprone CL/F in severe RI may be due to changes in CYP3A4 activity due to renal disease.
- Single-dose dordaviprone (375 mg) was well tolerated in both cohorts.
- The results of this study will be used to inform dosing in patients with RI.

References

(1) Venneti S, et al. *Cancer Discov*. 2023; DOI: 10.1158/2159-8290.CD-23-0131.
(2) Arrillaga-Romany I, et al. *J Clin Oncol*. 2024;42(13): 1542-1552.
(3) Arrillaga-Romany I, et al. *Neuro Oncol.* 2020;22(1):94-102.

Conflicts of Interest

SLF, JB, MA and TA are paid consultants of Chimerix, Inc. TA owns stock in Chimerix, Inc. AB, MM, MJM, TT, and ON are employees of and have stock options in Chimerix, Inc.

Poster PDF

Acknowledgments

Bioanalysis was performed by Q² Solutions (Durham, NC). Medical writing and editorial assistance was provided by Meghan Sullivan, PhD, CMPP, an employee of Chimerix, Inc.

The American College of Clinical Pharmacology (ACCP) 2024 Annual Meeting; September 8-10, 2024; N Bethesda, MD